Real-World Data to support surrogate endpoint evaluation for health technology assessment

J. Dupin, K. Thokagevistk, L. Emonnot, S. Micon, K. Benyounes, D. Havet, A. Civet, D. Pau, C. Le Dissez, S. Gally, C. Esnault

Roche, France

Background

A **surrogate endpoint** (SE) is a clinical endpoint used as a substitute of a more relevant clinical criterion (e.g. Overall Survival (OS) in oncology) supposed to be the gold standard endpoint (GSE). When used as primary outcomes, surrogate endpoints enable clinical trials of smaller sample size, shorter duration, and lower cost than trials with GSE. For example, surrogate endpoints are used when the clinical outcomes, like OS, might take a very long time to evaluate.

Acceptability of SE by Health Technology Agencies (HTA) depends mainly on the level of **SE validity** when evidence on the GSE is lacking.

In recent years, **regulatory agencies**, including the European Medicines Agency (EMA) and the Food and Drug Administration (FDA), have increasingly approved drugs and biologics on the basis of surrogate endpoints, although there are some uncertainties in this assessment.

Real-World Data (RWD) can help to evaluate relationship between SE and GSE before HTA assessment and confirm effectiveness on GSE after assessment. In France, a lot of qualitative RWD are available (including cohorts, claims databases & chart reviews) allowing to support SE assessment.

Objective

To discuss the current knowledge on the validity of SE, the potential contribution of RWD in this context, and the limits to their full exploitation.

The value of RWD in supporting SE validity is illustrated through two examples based on RWD studies initiated in early Renal Cell Carcinoma and Alzheimer disease. First recommendations are also provided.

What is a good surrogate endpoint?

It must be **clinically relevant, valid and reproducible.**

Based on regulators (mainly NICE & IQWIG), EUnetHTA defined **3 validity levels** depending on SE clinical relevance, correlation with the GSE and ability to predict treatment effect size on GSE supported by a clear mechanistic rationale and clinical data.

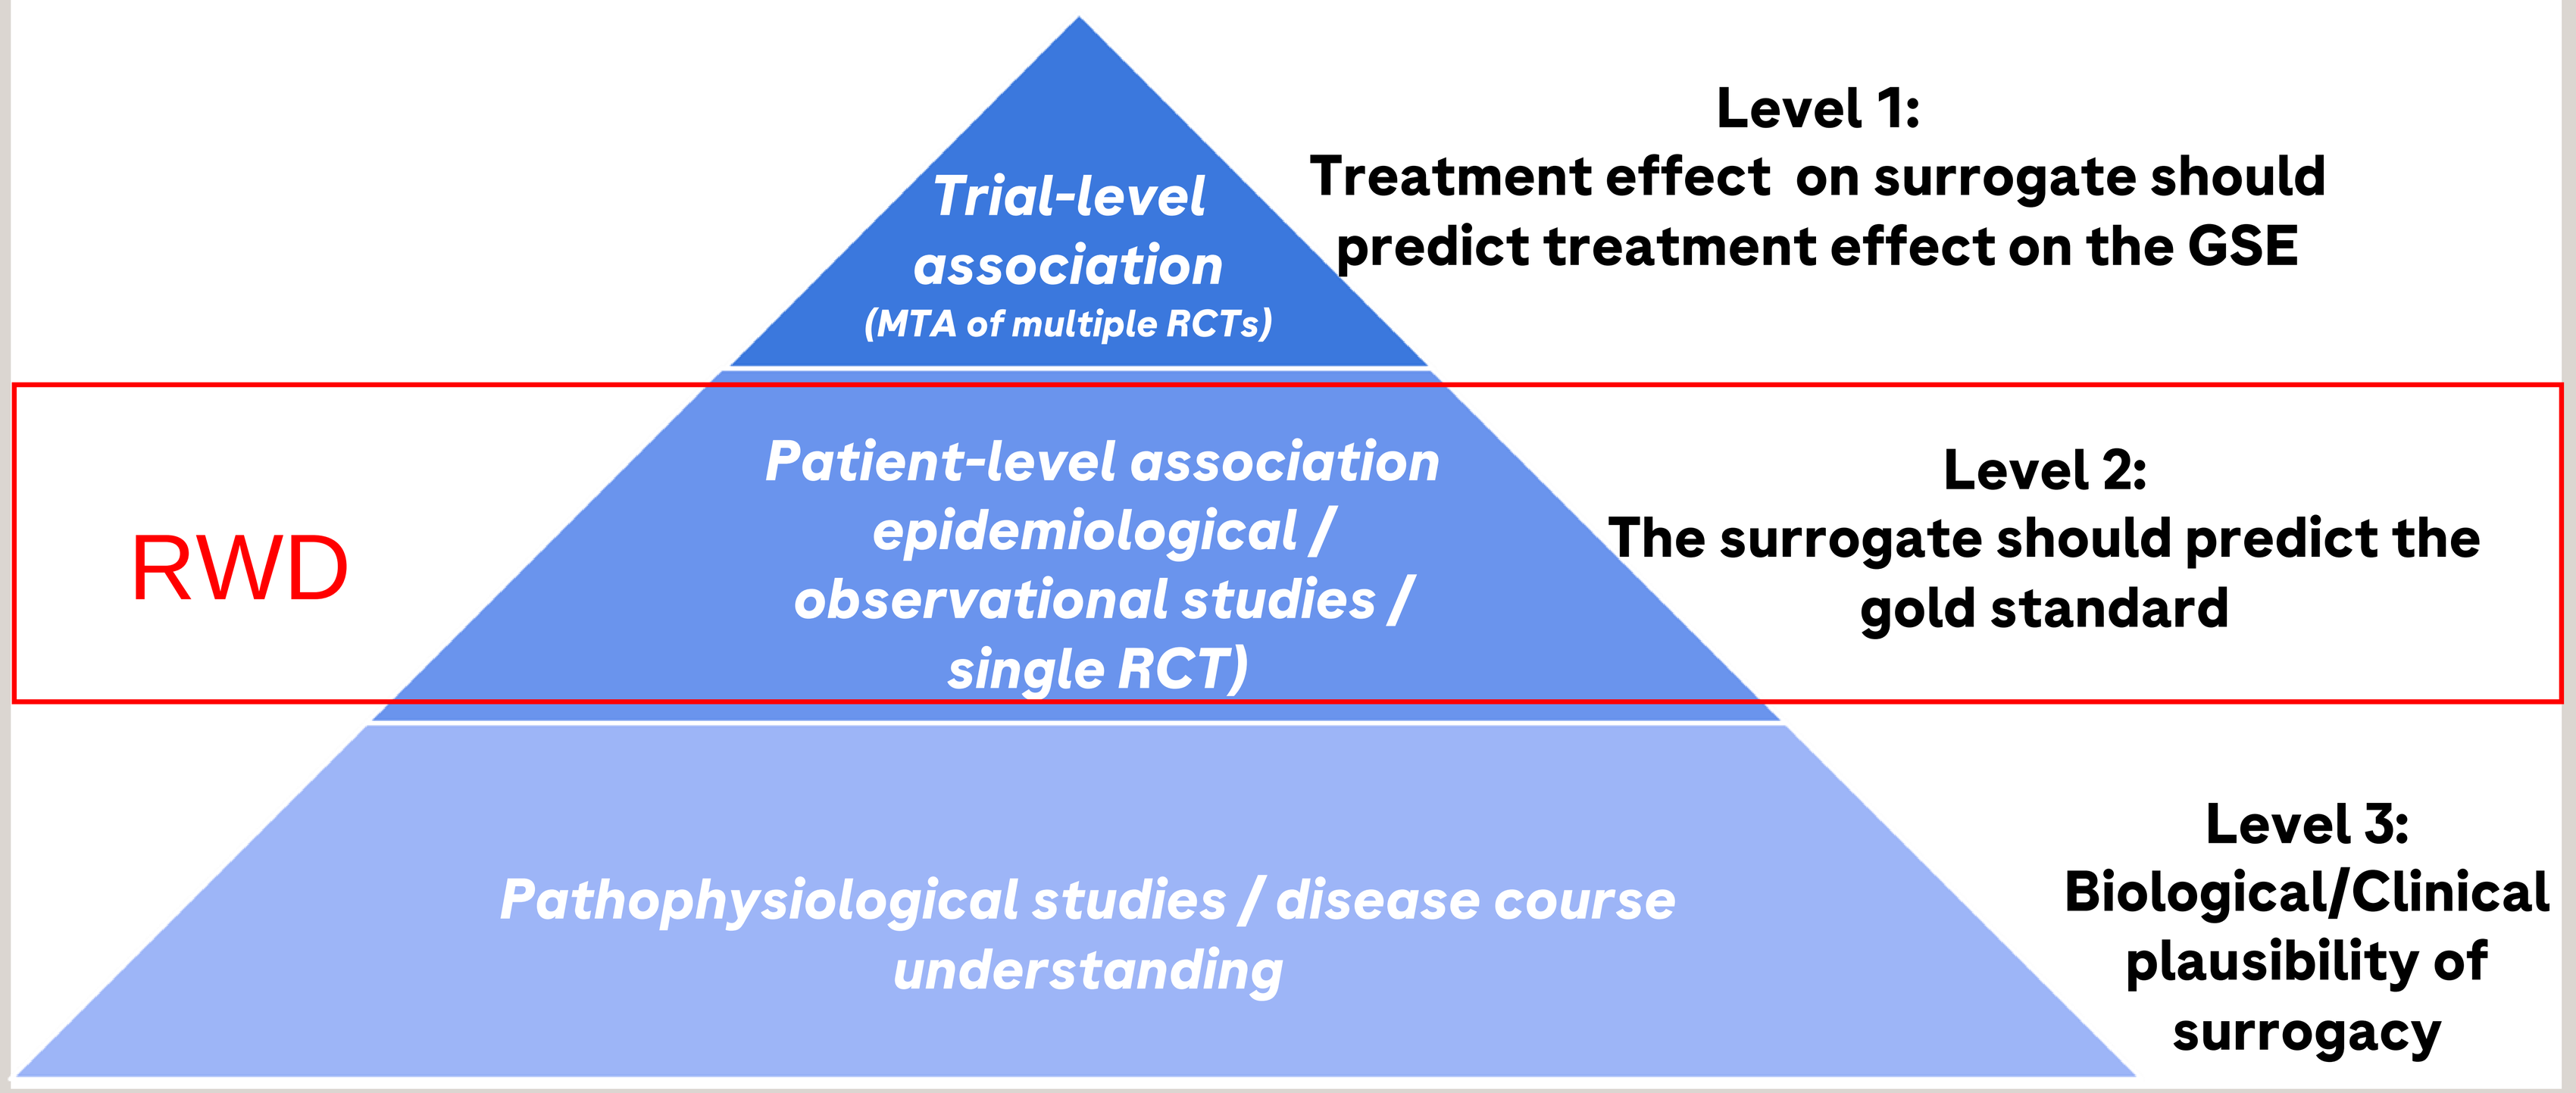
Trial-level association
(MTA of multiple RCTs)
Level 1:
Treatment effect on surrogate should predict treatment effect on the GSE
RWD
Patient-level association epidemiological / observational studies / single RCT)
Level 2:
The surrogate should predict the gold standard
Pathophysiological studies / disease course understanding
Level 3:
Biological/Clinical plausibility of surrogacy

How to evaluate surrogate endpoint acceptance?

Strict **demonstration of validity**, performed by meta-analysis (MTA) on clinical trials, is time consuming and often not feasible.

Acceptance is rarely straightforward and its evaluation depends of the clinical context on available evidence:

- ✓ In the indication of interest (or other indications)
- ✓ In the product of interest or therapeutic class or other products, e.g. a therapeutic class close to the product of interest

In indication > in other close indications > in other more distant indications
on targeted product > on other similar products > on other products

Acceptance level

Level 1: Has the correlation of the treatment effect between the SE and the "gold standard" endpoint already been demonstrated?

Level 2: Has the correlation between the SE and the "gold standard" endpoint already been demonstrated?

Level 3: Have experts (methodologists and clinicians) established evidence of biological plausibility of relationship between SE and the "gold standard" endpoint?

Illustrative examples	Early phase oncology setting ➔ Renal Cell Carcinoma in adjuvant	Neurodegenerative disease setting ➔ Alzheimer Disease
What are the Hurdles of acceptance in this specific indication?	Validation of SE defined as time to disease recurrence [Disease Free Survival (DFS), Recurrence Free Survival (RFS), Event Free Survival,...] is challenging, often affected by the delayed evaluation of GSE (Overall Survival) as well as the lack of data available for the related indication or therapeutic class	In certain neurodegenerative diseases, the innovative and subjective aspect of SE (as cognitive score) or the multiplicity and non-consensus of GSE (as institutionalization, death or other cognitive score) affect the assessment of SE validity.
What is the SE identified in the pivotal clinical trial?	DFS defined as local or distant recurrence or death	Change from baseline in Clinical Dementia Rating Scale Sum of Boxes (CDR-SOB) score at week 127
What is/are the relevant(s) GSE in this indication?	OS defined as death from any cause	Long-term outcomes: Disease stage progression, loss of autonomy, institutionalization and/or premature death
What is/are qualitative and long term follow-up RWD source(s) in this indication?	Linkage between a French national cohort and a quasi-exhaustive French claim database with 10 years of patients follow-up	French national cohort with 17 years of patient follow-up
Identify the research question and ad-hoc methodology to evaluate SE/GSE relationship	To describe DFS and OS outcomes in RWD and evaluate if a longer time to local or distant disease recurrence predicts a longer time to death	To describe the 2/3-year evolution of the CDR-SB and to analyze its relationship with short, medium and long (10 years) term risk of a combined indicator of moderate or more severe stage of dementia, severe AD dependency, institutionalization or death

Recommendations to support SE evaluation with RWD

- Perform literature and scientific advice review to evaluate level of SE validity and acceptability in the concerned indication and therapeutic class.
- Identify RWD sources available in your country with long term follow up and qualitative data at least on patients clinical characteristics and relevant outcomes.
- Before HTA evaluation: Possibility to set-up a RWD study to evaluate association between surrogate endpoint and GSE (to support level 2 of SE validation).
- After HTA evaluation: Possibility to set-up a RWD study to confirm SE association in the therapeutic class and/or effectiveness on the GSE.

Conclusion

Strict methodological validation of SE is rarely feasible, making the demonstration incomplete for industry and decision making complex for HTAs. RWD are a lever of particular interest to better support the demonstration of SE validity and to reinforce the evidence before HTA assessment or confirm the long-term treatment effect on GSE.
Clear guidelines from HTA are essential to unleash RWD potential to generate evidence, reduce uncertainties and optimize time and cost of demonstration.

ISPOR Europe 2022, 6-9 November 2022, Vienne