Daniel Brás
daniel.bras@novartis.com

Poster 15075

Recommendations for the design and implementation of a multidisciplinary consensus guidance for clinical practice on advanced breast cancer in Portugal

Rosa, Patricia Rosa,[1] Marco Domingues,[1] Daniel Brás,[1] Daniela Brandão,[2] Ângela Dias,[3] Vanessa Leomaro,[4] Maria Rita Dionísio,[1]

[1] Novartis Pharmaceuticals Corporation, Lisboa, Portugal
[2] Pharmacy Service, Portuguese Institute of Oncology, 4200-072 – Porto, Portugal
[3] Medical Oncology, Hospital Garcia de Orta, 2805-267 – Almada, Portugal
[4] Medical Oncology, Hospital Prof. Dr. Fernando da Fonseca, 2720-276 Amadora, Portugal, Portugal

KEY FINDINGS

- A multidisciplinary core group established the need of a guidance document to gather the best clinical practice considering the patient's journey of ABC
- Two parallel workstreams were constituted, composed of at least one element of each identified specialty/professional category:
 - ✓ Each group was focused on specific subject of the guidance according to their expertise and there were moments of broader group discussion for an integration of the multidisciplinary vision
- The final revision of the guidance was performed by two specific healthcare professionals who were responsible for compiling all pending topics
- The final draft will be sent to local scientific societies for assessment and possible scientific endorsement

INTRODUCTION

Consensus statements are being promoted to provide high-quality healthcare guidance.[1] The recommendations for the design and implementation of a multidisciplinary consensus guidance for clinical practice on advanced breast cancer (ABC) in Portugal emerged from the realization that the Portuguese Health System is not prepared to provide the best response to the specific needs of patients with advanced breast cancer throughout their continuum of care. These patients are dealing with numerous challenges along their journey and need a multidisciplinary and integrated approach that allows them to benefit from the skills of a wide range of specialties to obtain the best health care results. On the other hand, the increasing use of digital resources and online information research by patients makes them increasingly seek a multidisciplinary follow-up of their disease.

AIMS

This consensus guidance aims to establish new procedures and improve existing ones on ABC patient's treatment journey, based on expert opinions, that will ultimately improve healthcare, treatment outcomes and enhance the patient navigation within healthcare systems, specifically:

- Optimize the multidisciplinary approach to the patient with advanced breast cancer from the moment they enter the health system and throughout their journey.
- Promote and optimize networking and educate about best practices in multidisciplinary care for these patients
- Define a set of strategic lines of action that can be implemented and followed by health institutions to standardize the approach to patients with advanced breast cancer in Portugal.

METHODOLOGY

A multidisciplinary team (MDT) composed of patient association representatives and experts from nine different specialties, involved in the diagnosis and treatment of BC, used their knowledge and experience to address gaps within patient's journey of ABC.

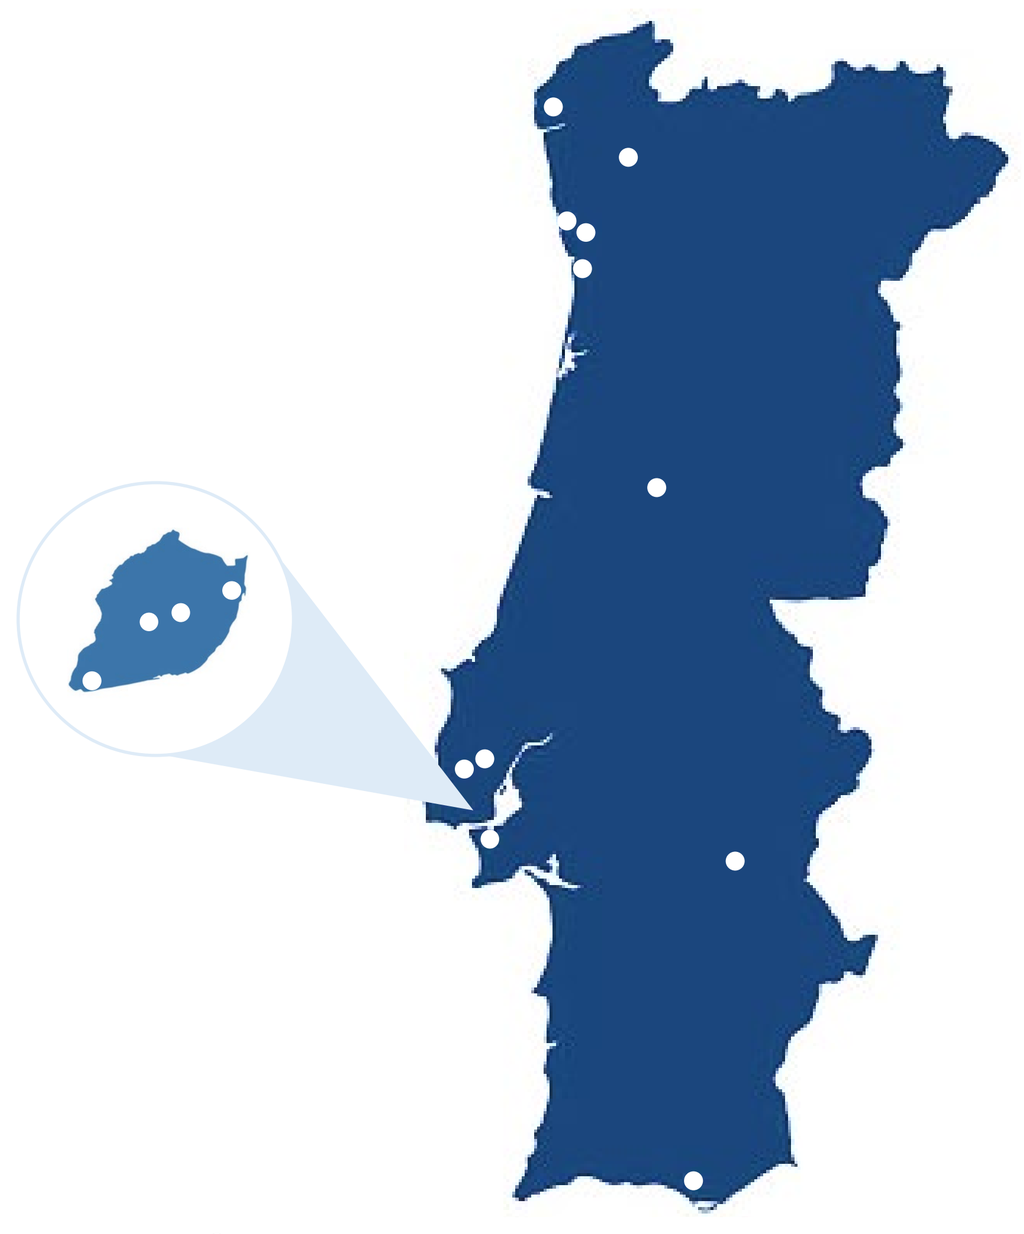

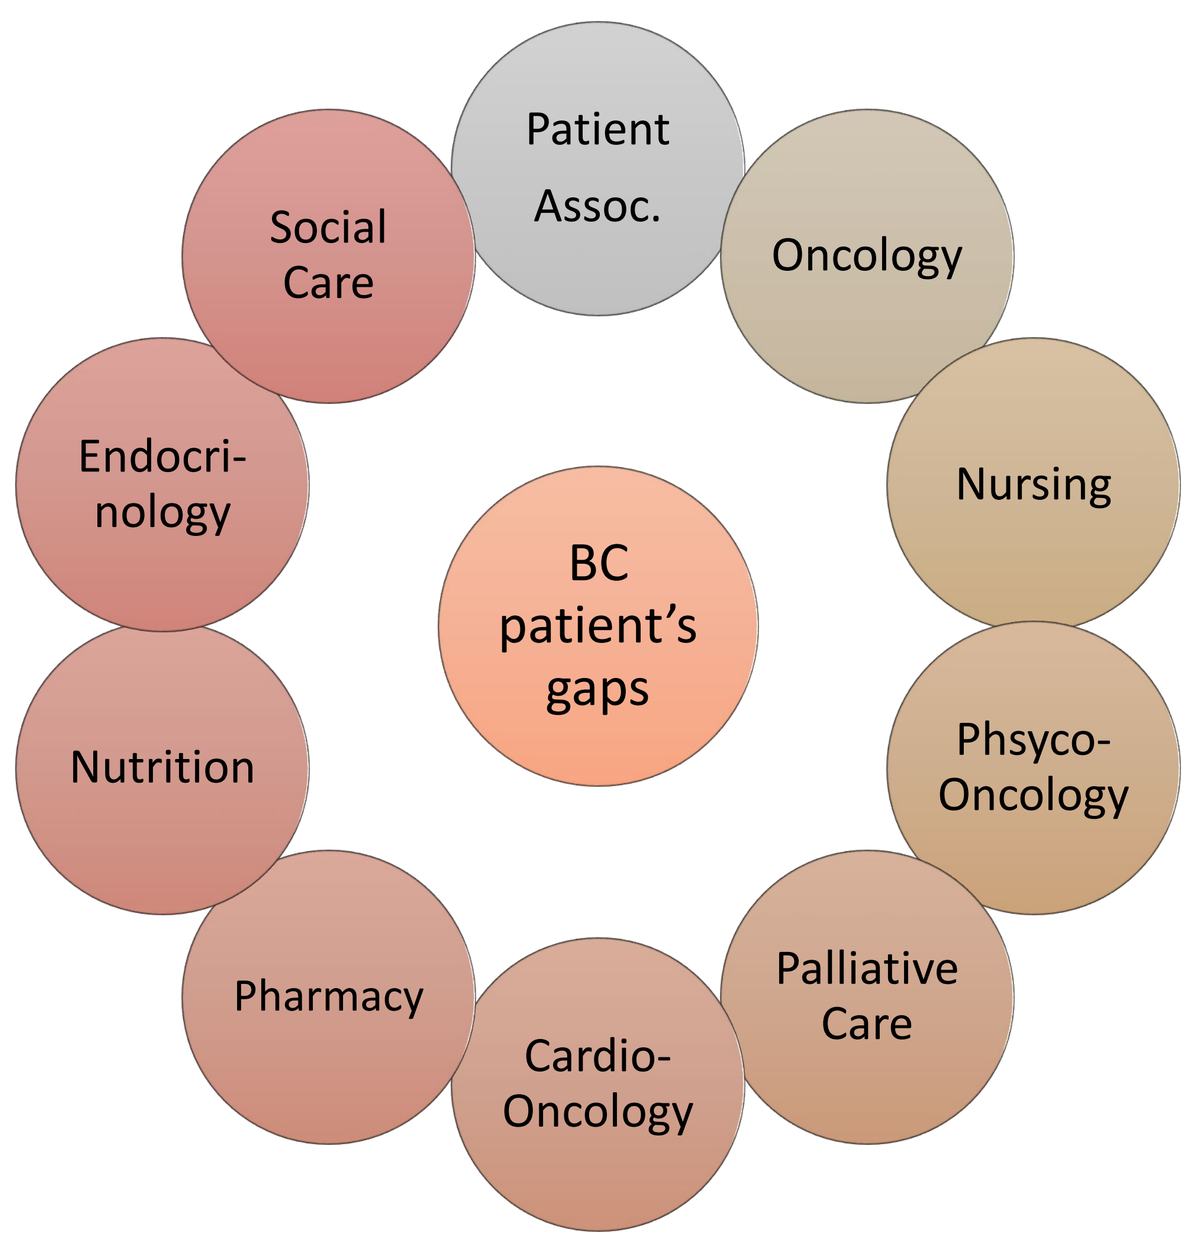
Patient Assoc.
Oncology
Nursing
Phsyco-Oncology
Palliative Care
Cardio-Oncology
Pharmacy
Nutrition
Endocri-nology
Social Care
BC patient's gaps

Figure 1 - The ABC patient journey

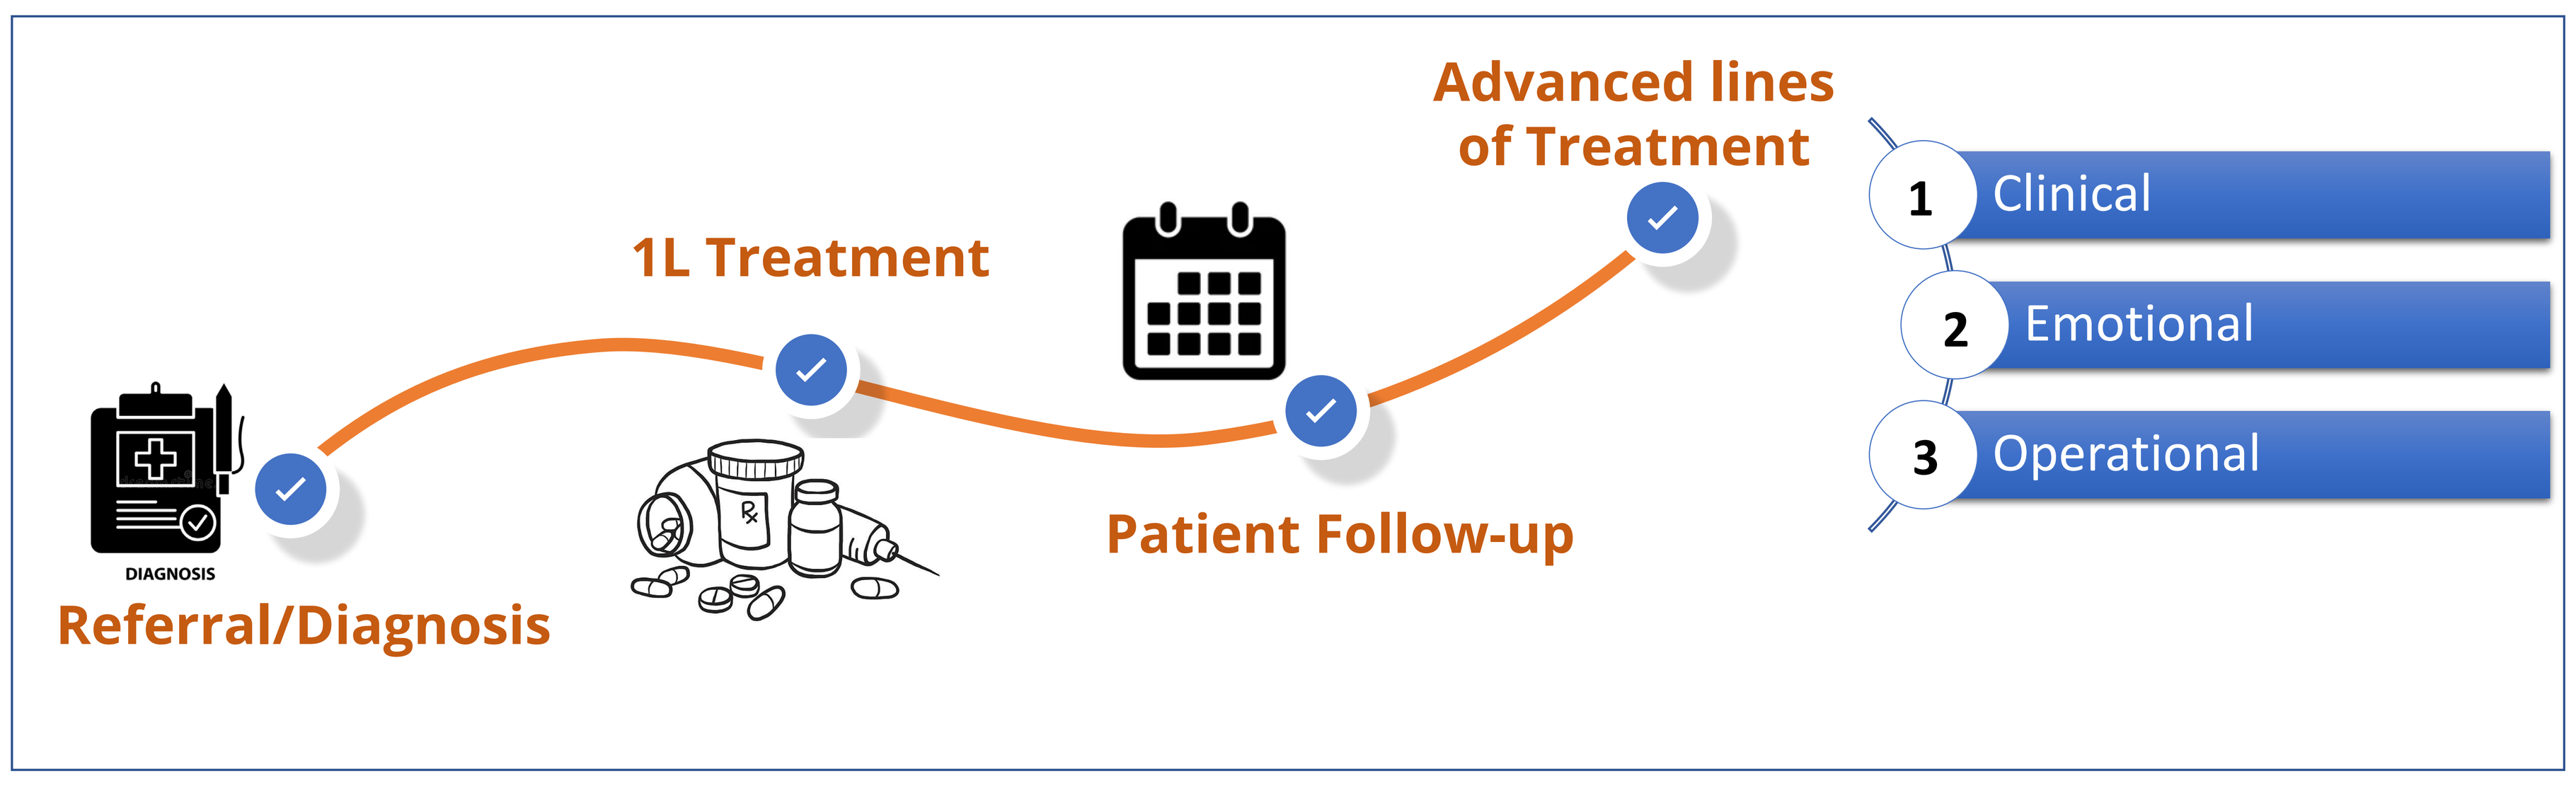
DIAGNOSIS
Referral/Diagnosis
1L Treatment
Patient Follow-up
Advanced lines of Treatment
1 Clinical
2 Emotional
3 Operational

RESULTS

After some brainstorming meetings, the multidisciplinary core group identified the need to gather in a guidance document the best clinical practice considering the patient's journey of ABC. Thus, two parallel workstreams were constituted, composed of at least one element of each identified specialty/professional category. Each group focused on specific subjects of the guidance, according to their expertise, had several moments of broader group discussion for an integration of the multidisciplinary vision. The final revision of the guidance was performed by two specific HCPs that compiled all pending topics. The final draft will be sent to local scientific societies for assessment and possible scientific endorsement.

Figure 2 – Guidance ellaboration process

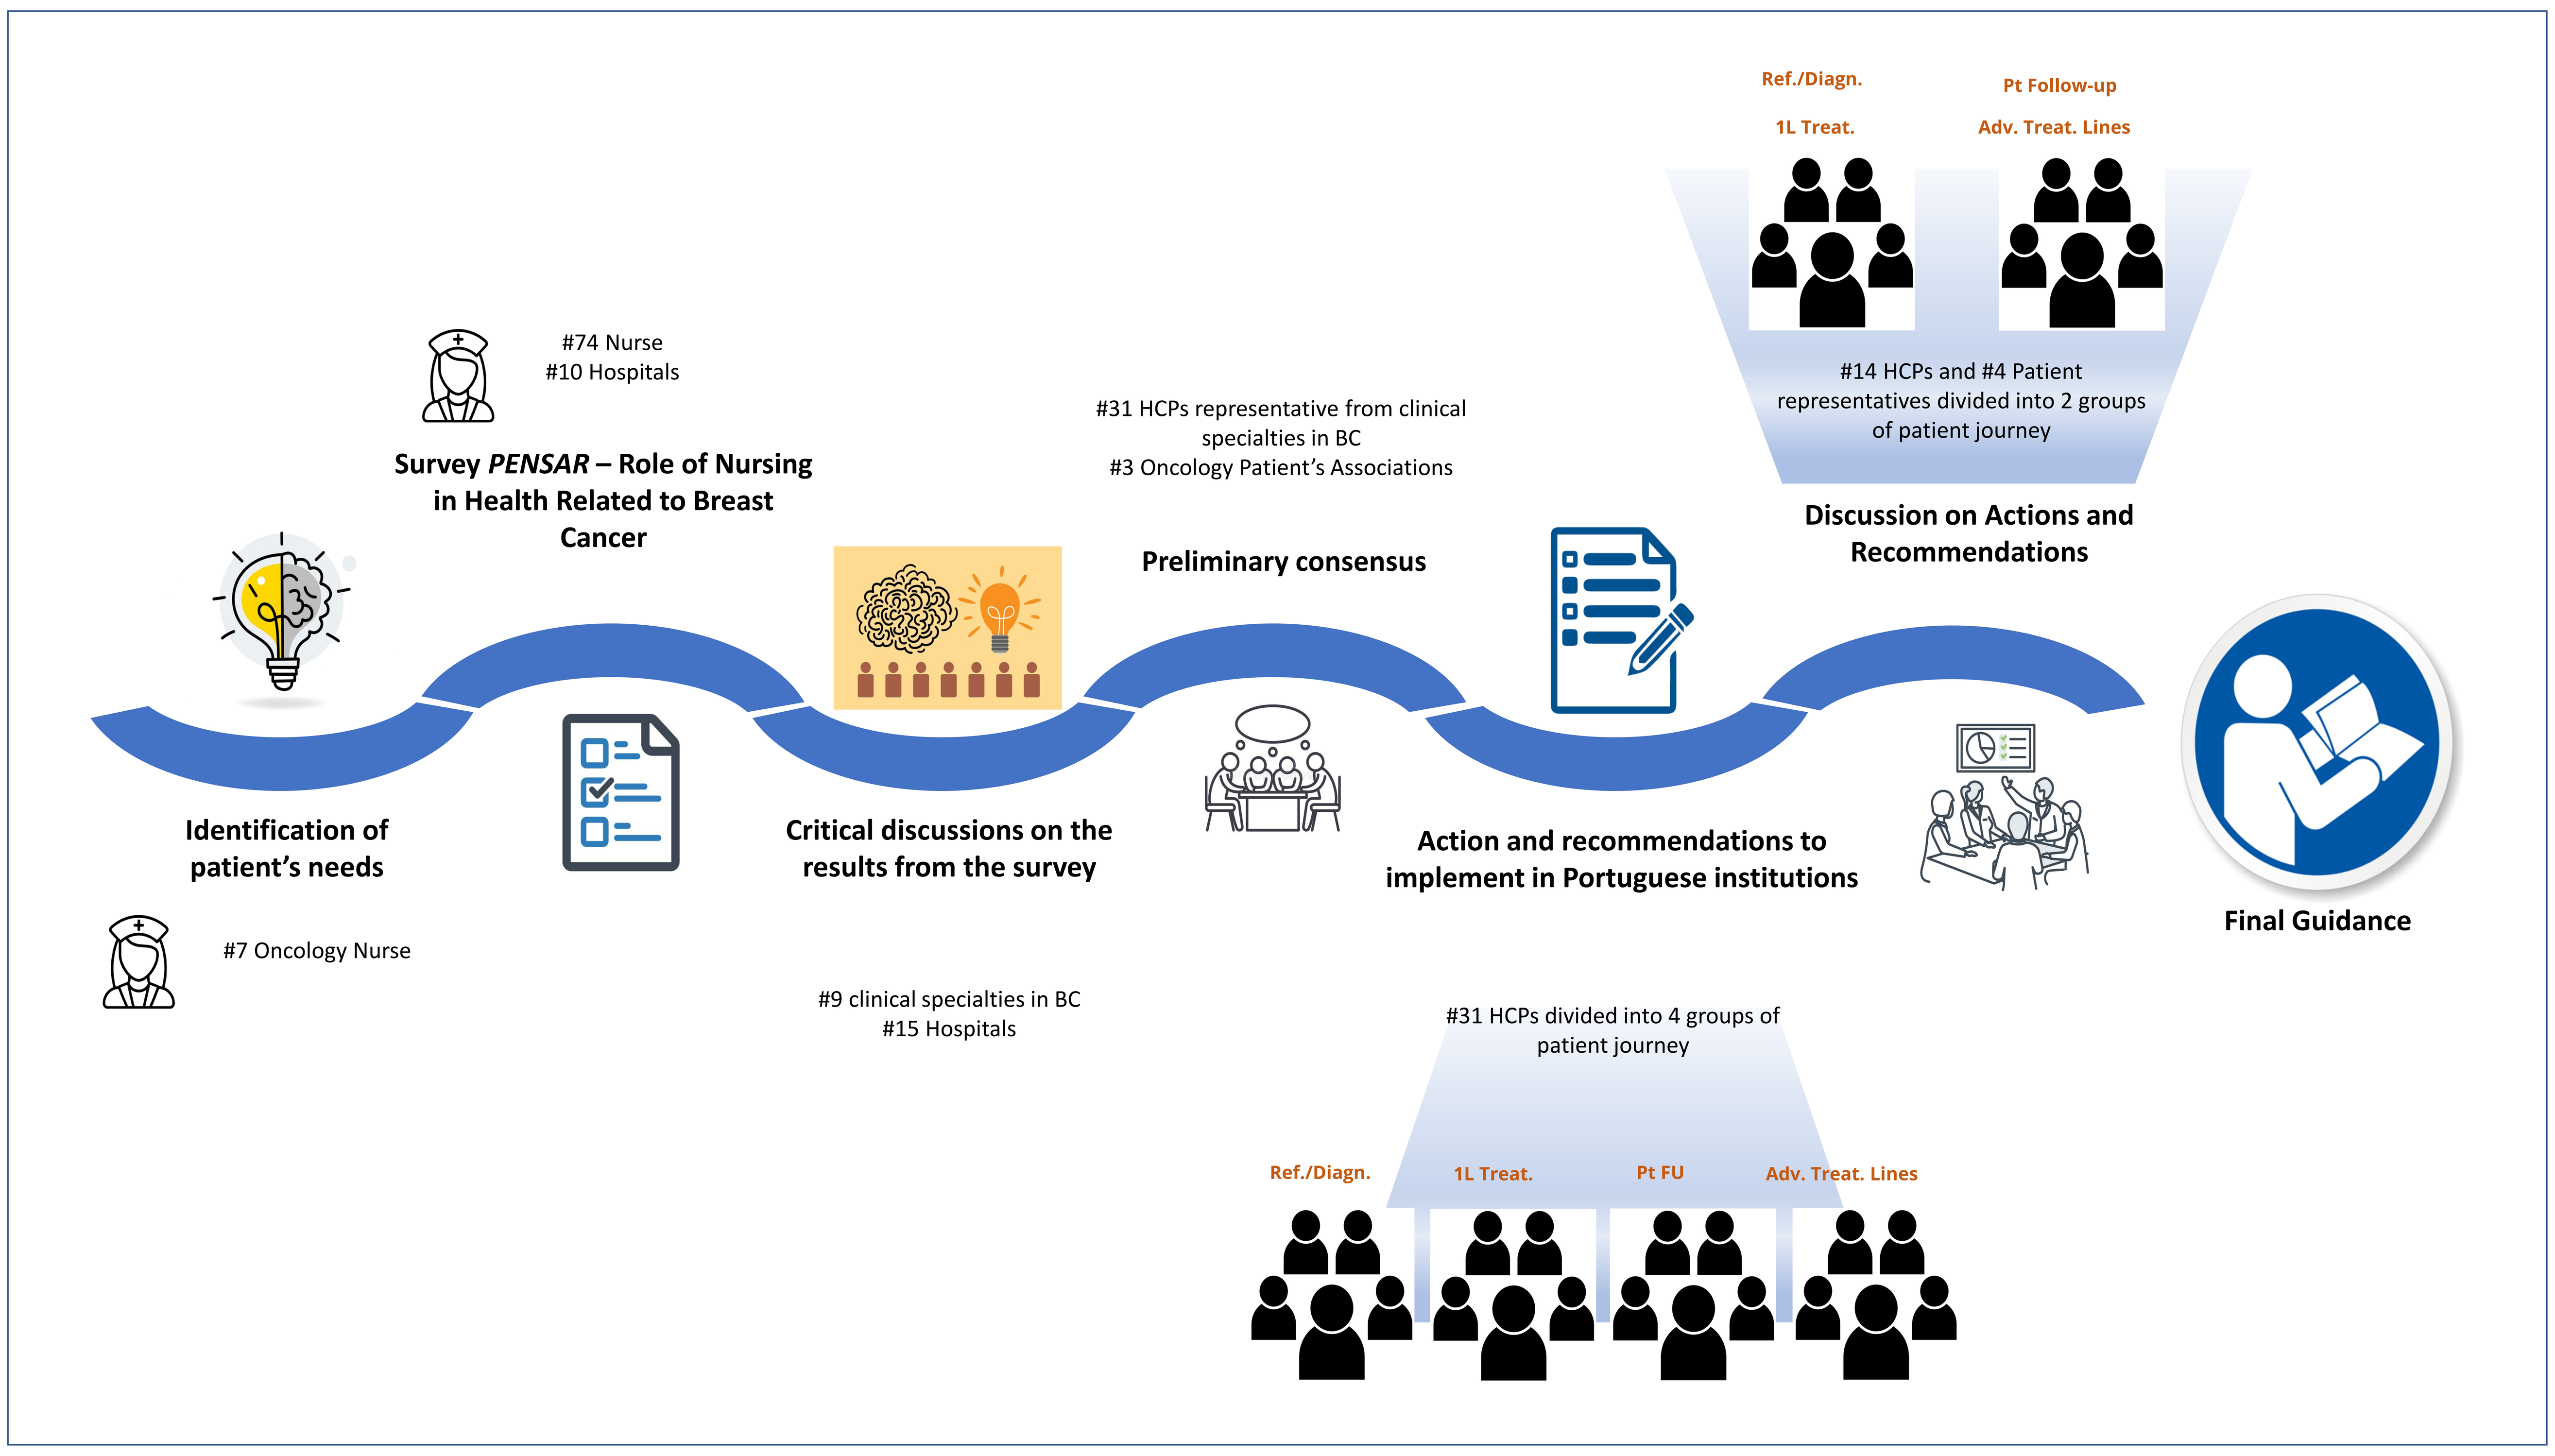
Identification of patient's needs
#7 Oncology Nurse
#74 Nurse
#10 Hospitals
Survey PENSAR – Role of Nursing in Health Related to Breast Cancer
Critical discussions on the results from the survey
#9 clinical specialties in BC
#15 Hospitals
#31 HCPs representative from clinical specialties in BC
#3 Oncology Patient's Associations
Preliminary consensus
#31 HCPs divided into 4 groups of patient journey
Ref./Diagn.
1L Treat.
Pt FU
Adv. Treat. Lines
Action and recommendations to implement in Portuguese institutions
Ref./Diagn.
1L Treat.
Pt Follow-up
Adv. Treat. Lines
#14 HCPs and #4 Patient representatives divided into 2 groups of patient journey
Discussion on Actions and Recommendations
Final Guidance

Aknowledgements

The authors would like to thank the contribution of all the team members involved in the guidance.
Medical writing assistance was provided by LaTM – Life Sciences Consultants and was funded by Novartis Pharmaceuticals Corporation.
The authors had final responsibility for the poster.

Disclosures

Patrícia Rosa, **Marco Domingues**, **Daniel Brás**, **Maria Rita Dionísio**: employment. **Daniela Brandão**: , **Ângela Dias**:, **Vanessa Leomaro**: nothing to disclose.

References

1. Maes-Carballo, M., et al (2022). European journal of cancer care, 31(2), e13540.