Diana Tran, **Email:** diana.tran@eversana.com

Poster EE360

Health Care Resource Utilization With Asciminib and Bosutinib Among Adults With Chronic Myeloid Leukemia in Chronic Phase Previously Treated With ≥2 Tyrosine Kinase Inhibitors: Week 48 and Week 96 Results From ASCEMBL Trial

Jorge E Cortes,[1] Koji Sasaki,[2] Delphine Rea,[3] Michael J Mauro,[4] Diana Tran,[5] Pearl Wang,[5] Kejal Jadhav,[6] Aurore Yocolly[7]

[1]Georgia Cancer Center, Augusta University, Augusta, GA, USA; [2]MD Anderson Cancer Center, The University of Texas, Houston, TX, USA; [3]Hopital Saint-Louis, APHP, Paris, France; [4]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [5]EVERSANA, Burlington, Ontario, Canada; [6]Novartis Pharmaceuticals, East Hanover, New Jersey, USA; [7]Novartis Services Inc., East Hanover, New Jersey, USA

Scan to obtain a copy of the poster

https://www.medicalcongress.novartisoncology.com/ISPOREU22/CML/cml.html#DianaEE360

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- Findings similar to the Week 24 analysis were observed at the Week 48 analysis and the Week 96 analysis.
- As HCRU stabilized over time, asciminib maintained a consistently lower overall resource utilization over the long-term compared to bosutinib in patients with CML-CP from the ASCEMBL trial.

This study is sponsored by Novartis Services Inc.
Poster presented at the ISPOR Europe 2022, Vienna, Austria, 6–9 November 2022

INTRODUCTION

- Chronic myeloid leukemia (CML) is a hematological neoplasm that is typically present with the abnormal Philadelphia chromosome.[1]
- The pivotal ASCEMBL phase 3, multi-center, open label, randomized controlled trial compared asciminib, a first-in-class agent Specifically Targeting the ABL Myristoyl Pocket (STAMP) to inhibit the BCR::ABL1 oncoprotein, to bosutinib, among 3L+ CML in chronic phase (CML-CP) patients.[2,3]
- Primary endpoint was achieved in the ASCEMBL trial with 25.5% of patients receiving asciminib achieving major molecular response at Week 24 compared to 13.2% of patients receiving bosutinib.[2] Asciminib also demonstrated a better safety and tolerability profile as reflected by the low discontinuation rate due to adverse events (AEs) compared to bosutinib at Week 24.
- Funding decision-makers are interested in assessing how the efficacy and safety benefits of asciminib translate into health care resource utilization (HCRU) reduction.
- A preliminary analysis at the time of the assessment of the primary endpoint for ASCEMBL (i.e., Week 24) suggested a lower HCRU compared to bosutinib among patients with 3L+ CML-CP.[4] It is of interest to decision-makers to assess whether the potential economic benefits of asciminib demonstrated in the Week 24 analysis are still maintained in the long-term follow-up.

OBJECTIVES

- To assess and compare HCRU rates of asciminib and bosutinib at the Week 24, Week 48, and Week 96 cut-offs.

METHODS

- HCRU was assessed in adult patients with CML-CP who had previously been treated with ≥2 tyrosine kinase inhibitors as:
 - frequency and duration of hospitalization from baseline up to end of treatment,
 - frequency of emergency room (ER) visits from baseline up to end of treatment, and
 - frequency of additional (unplanned) outpatient office visits for general practitioner, specialist, and urgent care from baseline up to end of treatment.
- For hospitalizations, type of ward (hospital unit), length of hospital stay (number of days in ward), reasons for hospitalization, and discharge status were also captured.
- HCRU assessments were completed by investigators at each scheduled clinical visit. Clinical visits were scheduled at Week 1, followed by visits every two weeks from Week 2 to Week 16, and every four weeks thereafter from Week 20 to Week 96.
- The number of ER (<24 hours) visits, general practitioner visits, specialist visits, and urgent care visits were self-reported by the patients.
- For each category of resource and overall, the proportion of patients with any HCRU, the frequency of HCRU, the rate and corresponding 95% confidence intervals (CIs) of HCRU per patient-year on randomized treatment, and length of hospital stay by ward type were summarized using descriptive statistics (mean, standard deviation, median, and range for quantitative variables, and count and percentage for qualitative variables) and compared between 157 patients receiving asciminib 40 mg twice daily and 76 patients receiving bosutinib 500 mg once daily.

RESULTS

- Of the 157 and 76 patients recruited in asciminib and bosutinib arms, the respective number of drop-outs were 60 (38.2%) and 54 (71.1%) at the Week 24 analysis, 68 (43.3%) and 59 (77.6%) at the Week 48 analysis, and 73 (46.5%) and 61 (80.3%) at the Week 96 analysis.
- Respective median duration of treatment exposure for asciminib and bosutinib arms were 43.4 weeks and 29.2 weeks at the Week 24 analysis (data cut-off: 25 May 2020), 66.9 weeks and 32.6 weeks at the Week 48 analysis (data cut-off: 06 Jan 2021), and 103.1 weeks and 34.4 weeks at the Week 96 analysis (data cut-off: 06 October 2021).
- The proportions of patients with any HCRU (overall and by category) are presented in **Figure 1**. Despite longer median treatment duration, a lower proportion of patients in the asciminib arm compared to the patients in the bosutinib arm used at least one health care resource (i.e., hospitalizations, ER visits, general practitioner visits, specialist visits, or urgent care visits).
 - Hospitalization was the most common resource used for asciminib and bosutinib at all timepoints.

Figure 1. Proportions of patients with HCRU at Week 24, Week 48 and Week 96 analysis, overall and by category

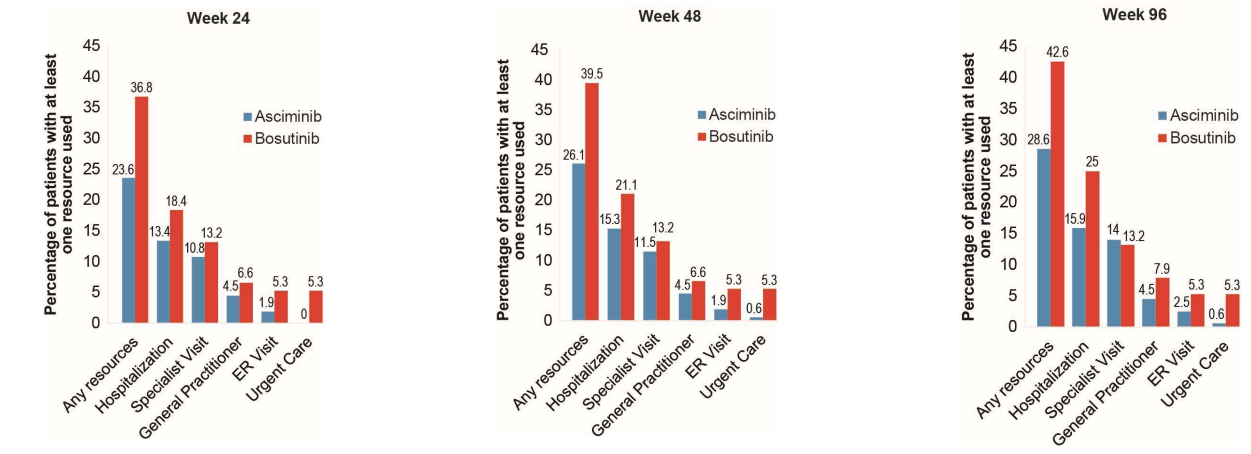
Week 24
Week 48
Week 96
Percentage of patients with at least one resource used
Asciminib
Bosutinib
23.6 36.8
13.4 18.4
10.8 13.2
4.5 6.6
1.9 5.3
0 5.3
26.1 39.5
15.3 21.1
11.5 13.2
4.5 6.6
1.9 5.3
0.6 5.3
28.6 42.6
15.9 25
14 13.2
4.5 7.9
2.5 5.3
0.6 5.3
Any resources
Hospitalization
Specialist Visit
General Practitioner
ER Visit
Urgent Care

Proportions for specific resources may not add up exactly to the proportion for "any resources" use because some patients used more than one resource.
Abbreviations: ER, emergency room; HCRU, health care resource utilization.

- Since the median duration of treatment with asciminib was much longer than with bosutinib, the data showed higher resource utilization in the asciminib arm (**Table 1**), particularly for general practitioner and specialist visits.
- Despite this, asciminib was associated with less hospitalizations, ER visits, and urgent care visits due to AEs compared to bosutinib.

Table 1. Frequency and reasons for HCRU

Hospitalization and Clinical Visits		Week 24 Asciminib (n = 157)	Week 24 Bosutinib (n = 76)	Week 48 Asciminib (n = 157)	Week 48 Bosutinib (n = 76)	Week 96 Asciminib (n = 157)	Week 96 Bosutinib (n = 76)
All Hospitalizations							
Number of subjects with at least one visit – n (%)		21 (13.4)	14 (18.4)	24 (14.6)	16 (21.1)	25 (15.9)	19 (25.0)
Frequency of visit (median duration of exposure, weeks)		37 (58.1)	17 (21.4)	44 (78.1)	24 (33.3)	49 (129.4)	30 (43.4)
Number per subject	Mean (SD)	1.8 (0.9)	1.2 (0.6)	1.8 (1.2)	1.5 (1.1)	2.0 (1.2)	1.6 (1.1)
	Median (range)	1 (1, 4)	1 (1, 3)	1 (1, 5)	1 (1, 2)	1 (1, 5)	1 (1, 2)
Reason – n (%)	AE related to CML therapy	3 (8.6)	6 (35.3)	3 (6.8)	8 (38.1)	3 (6.3)	9 (39.1)
	CML	1 (2.9)	0 (0)	1 (2.3)	0 (0)	1 (2.1)	0 (0)
	Other reason	32 (88.9)	11 (64.7)	40 (90.9)	13 (61.9)	45 (91.7)	14 (60.9)
ER Visits (< 24 hours)							
Number of subjects with at least one visit – n (%)		3 (1.9)	4 (5.3)	3 (1.9)	4 (5.3)	4 (2.5)	4 (5.3)
Frequency of visit (median duration of exposure, weeks)		4 (60.1)	4 (22.7)	4 (92.4)	4 (19.4)	6 (162.0)	4 (23.0)
Number per subject	Mean (SD)	1.3 (0.6)	1.0 (0)	1.3 (0.6)	1.0 (0)	1.5 (0.6)	1.0 (0)
	Median (range)	1 (1, 2)	1 (1, 1)	1 (1, 2)	1 (1, 1)	1.5 (1, 2)	1 (1, 1)
Reason – n (%)	AE related to CML therapy	2 (50.0)	3 (75.0)	2 (50.0)	3 (75.0)	3 (50.0)	3 (75.0)
	CML	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)
	Other reason	2 (50.0)	1 (25.0)	2 (50.0)	1 (25.0)	3 (50.0)	1 (25.0)
General Practitioner Visits							
Number of subjects with at least one visit – n (%)		7 (4.5)	5 (6.6)	7 (4.5)	5 (6.6)	7 (4.5)	6 (7.9)
Frequency of visit (median duration of exposure, weeks)		29 (54.6)	5 (69.1)	30 (56.9)	6 (101.0)	30 (56.9)	8 (114.0)
Number per subject	Mean (SD)	4.1 (6.6)	1.0 (0)	4.3 (6.6)	1.2 (0.5)	4.3 (6.6)	1.3 (0.8)
	Median (range)	2 (1, 19)	1 (1, 1)	2 (1, 19)	1 (1, 2)	2 (1, 19)	1 (1, 3)
Reason – n (%)	AE related to CML therapy	2 (6.9)	0 (0)	2 (6.7)	0 (0)	2 (6.7)	0 (0)
	CML	20 (69.0)	0 (0)	20 (66.7)	0 (0)	20 (66.7)	0 (0)
	Other reason	7 (24.1)	5 (100.0)	8 (26.7)	6 (100.0)	8 (26.7)	8 (100.0)
Specialist Visits							
Number of subjects with at least one visit – n (%)		17 (10.8)	10 (13.2)	18 (11.5)	10 (13.2)	22 (14.0)	10 (13.2)
Frequency of visit (median duration of exposure, weeks)		90 (60.1)	25 (50.9)	97 (76.1)	26 (55.2)	110 (117.0)	28 (80.3)
Number per subject	Mean (SD)	5.3 (6.1)	2.5 (2.4)	5.4 (6.3)	2.6 (2.3)	5.0 (6.1)	2.8 (2.3)
	Median (range)	4 (1, 24)	1 (1, 8)	3.5 (1, 25)	1.5 (1, 8)	3 (1, 25)	2.5 (1, 8)
Reason – n (%)	AE related to CML therapy	9 (10.0)	2 (8.0)	9 (9.3)	2 (7.7)	13 (11.8)	3 (10.7)
	CML	1 (1.1)	0 (0)	1 (1.0)	0 (0)	1 (0.9)	0 (0)
	Other reason	80 (88.9)	23 (92.0)	87 (89.7)	24 (92.3)	96 (87.2)	25 (89.3)
Urgent Care Visits							
Number of subjects with at least one visit – n (%)		0 (0)	4 (5.3)	1 (0.6)	4 (5.3)	1 (0.6)	4 (5.3)
Frequency of visit (median duration of exposure, weeks)		0 (0)	4 (24.1)	1 (92.4)	4 (19.3)	1 (131.0)	4 (19.3)
Number per subject	Mean (SD)	0 (0)	1.0 (0)	1.0 (0)	1.0 (0)	1.0 (0)	1.0 (0)
	Median (range)	0 (0, 0)	1 (1, 1)	1 (1, 1)	1 (1, 1)	1 (1, 1)	1 (1, 1)
Reason – n (%)	AE related to CML therapy	0 (0)	2 (50.0)	0 (0)	2 (50.0)	0 (0)	2 (50.0)
	CML	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)
	Other reason	0 (0)	2 (50.0)	1 (100.0)	2 (50.0)	1 (100.0)	2 (50.0)

Abbreviations: AE, adverse event; CML, chronic myeloid leukemia; HCRU, health care resource utilization; SD, standard deviation.

- Adjusting for differential treatment exposure, patients receiving asciminib had significantly lower rates of overall HCRU per patient-year compared to those receiving bosutinib at all three data cut-offs (**Figure 2**).
 - Lower rates of HCRU were observed for all of the specific resources among patients in the asciminib arm compared to those in bosutinib arm; however, the differences in rates were not statistically significant.

Figure 2. Rates of HCRU per patient-year at Week 24, Week 48 and Week 96 analysis

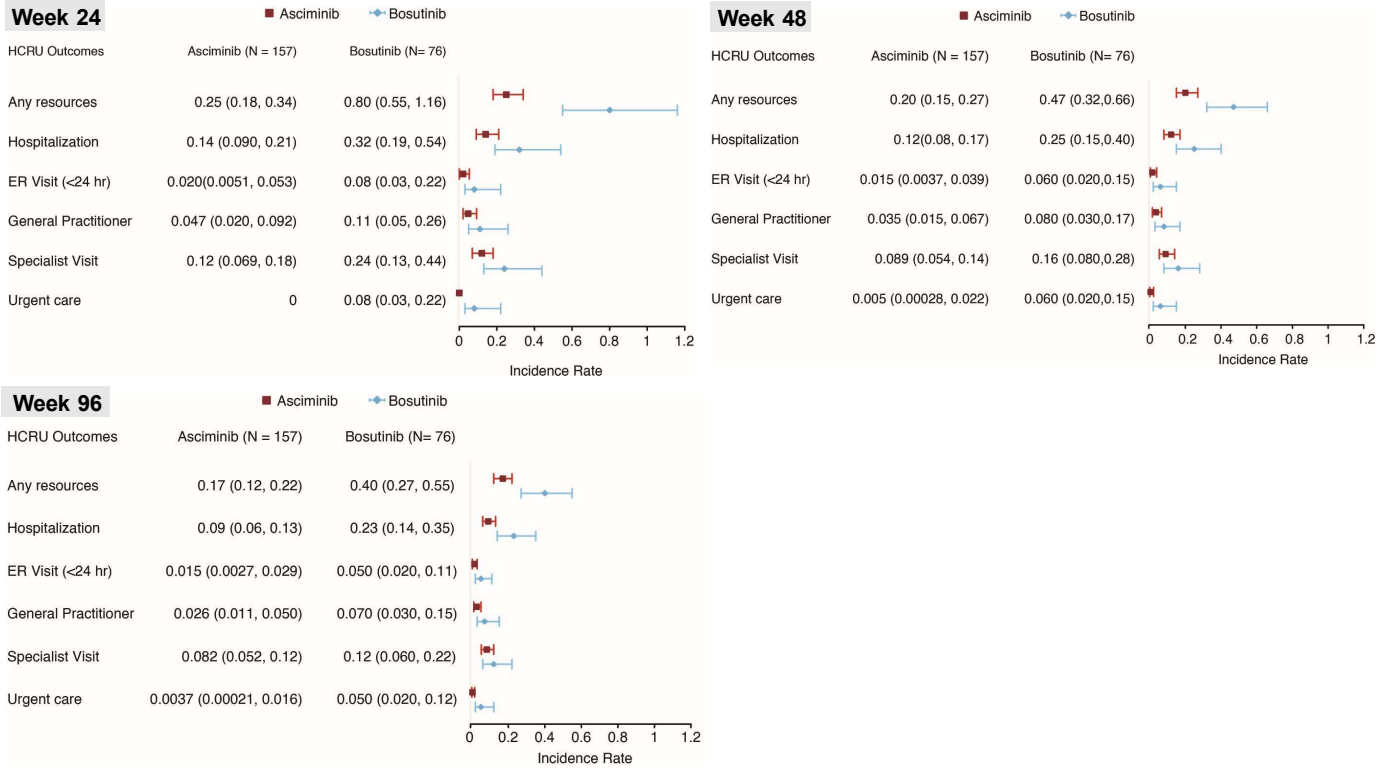
Week 24
Asciminib
Bosutinib
HCRU Outcomes | Asciminib (N = 157) | Bosutinib (N= 76)
Any resources | 0.25 (0.18, 0.34) | 0.80 (0.55, 1.16)
Hospitalization | 0.14 (0.090, 0.21) | 0.32 (0.19, 0.54)
ER Visit (<24 hr) | 0.020(0.0051, 0.053) | 0.08 (0.03, 0.22)
General Practitioner | 0.047 (0.020, 0.092) | 0.11 (0.05, 0.26)
Specialist Visit | 0.12 (0.069, 0.18) | 0.24 (0.13, 0.44)
Urgent care | 0 | 0.08 (0.03, 0.22)
0 0.2 0.4 0.6 0.8 1 1.2
Incidence Rate
Week 48
Asciminib
Bosutinib
HCRU Outcomes | Asciminib (N = 157) | Bosutinib (N= 76)
Any resources | 0.20 (0.15, 0.27) | 0.47 (0.32,0.66)
Hospitalization | 0.12(0.08, 0.17) | 0.25 (0.15,0.40)
ER Visit (<24 hr) | 0.015 (0.0037, 0.039) | 0.060 (0.020,0.15)
General Practitioner | 0.035 (0.015, 0.067) | 0.080 (0.030,0.17)
Specialist Visit | 0.089 (0.054, 0.14) | 0.16 (0.080,0.28)
Urgent care | 0.005 (0.00028, 0.022) | 0.060 (0.020,0.15)
0 0.2 0.4 0.6 0.8 1 1.2
Incidence Rate
Week 96
Asciminib
Bosutinib
HCRU Outcomes | Asciminib (N = 157) | Bosutinib (N= 76)
Any resources | 0.17 (0.12, 0.22) | 0.40 (0.27, 0.55)
Hospitalization | 0.09 (0.06, 0.13) | 0.23 (0.14, 0.35)
ER Visit (<24 hr) | 0.015 (0.0027, 0.029) | 0.050 (0.020, 0.11)
General Practitioner | 0.026 (0.011, 0.050) | 0.070 (0.030, 0.15)
Specialist Visit | 0.082 (0.052, 0.12) | 0.12 (0.060, 0.22)
Urgent care | 0.0037 (0.00021, 0.016) | 0.050 (0.020, 0.12)
0 0.2 0.4 0.6 0.8 1 1.2
Incidence Rate

Abbreviations: ER, emergency room; HCRU, health care resource utilization; hr, hour; N, number of patients.

- Length of hospital stay was lower with asciminib compared to bosutinib in most ward types at all data cut-offs (**Table 2**).
 - Similar to the results seen at Week 24 analysis, most hospitalizations had time spent in general ward for both asciminib and bosutinib.
 - After general ward, hospitalizations for asciminib had time spent in 'other care unit' ward, whereas hospitalizations for bosutinib had time spent in the emergency room ward.

Table 2. Length of hospital stay by ward type for hospitalized patients

Ward Type		Week 24 Asciminib (n = 157)	Week 24 Bosutinib (n = 76)	Week 48 Asciminib (n = 157)	Week 48 Bosutinib (n = 76)	Week 96 Asciminib (n = 157)	Week 96 Bosutinib (n = 76)
All Hospitalizations	Number of patients – n (%)	21 (13.4)	14 (18.4)	24 (15.3)	16 (21.1)	25 (15.9)	19 (25.0)
	Mean (SD)	9.5 (10.7)	10.7 (10.2)	9.7 (12.7)	9.0 (9.7)	9.1 (11.9)	8.5 (9.2)
	Median (Range)	5 (1, 47)	7 (1, 35)	5 (1, 60)	5 (1, 35)	5 (1, 60)	5 (1, 35)
ER	Number of patients – n (%)	1 (0.6)	5 (6.6)	3 (1.9)	5 (6.6)	4 (2.5)	8 (10.5)
	Hospitalization with time spent in ER – n (%)	1 (2.7)	6 (28.6)	3 (6.8)	6 (25.0)	4 (8.2)	9 (30.0)
	Mean (SD)	1.0 (NA)	1.5 (1.2)	2.0 (1.7)	1.5 (1.2)	1.8 (1.5)	3.3 (6.0)
	Median (Range)	1 (1, 1)	1 (1, 4)	1 (1, 4)	1 (1, 4)	1 (1, 4)	1 (1, 19)
General Ward	Number of patients – n (%)	15 (9.6)	10 (13.2)	15 (9.6)	12 (15.8)	17 (10.8)	14 (18.4)
	Hospitalization with time spent in general ward – n (%)	26 (70.3)	12 (57.1)	29 (65.9)	15 (62.5)	32 (65.3)	17 (56.7)
	Mean (SD)	9.2 (10.3)	10.2 (9.2)	9.0 (9.8)	10.4 (8.8)	8.6 (9.4)	9.7 (8.4)
	Median (Range)	6 (1, 47)	6.5 (2, 32)	6 (1, 47)	7 (2, 32)	6 (1, 47)	7 (2, 32)
Intensive Care Unit	Number of patients – n (%)	3 (1.9)	2 (2.6)	3 (1.9)	2 (2.6)	4 (2.5)	2 (2.6)
	Hospitalization with time spent in intensive care unit – n (%)	3 (8.1)	2 (9.5)	3 (6.8)	2 (8.3)	5 (10.2)	2 (6.7)
	Mean (SD)	13.0 (15.6)	19.0 (22.6)	13.0 (15.6)	19.0 (22.6)	10.2 (11.9)	19.0 (22.6)
	Median (Range)	5 (3, 31)	19 (3, 35)	5 (3, 31)	19 (3, 35)	5 (3, 31)	19 (3, 35)
Other Care Unit	Number of patients – n (%)	5 (3.2)	1 (1.3)	5 (3.2)	1 (1.3)	5 (3.2)	1 (1.3)
	Hospitalization with time spent in other care unit – n (%)	7 (18.9)	1 (4.8)	8 (18.2)	1 (4.2)	7 (14.3)	1 (3.3)
	Mean (SD)	7.3 (10.6)	12.0 (NA)	6.5 (10.0)	12.0 (NA)	7.3 (10.6)	12.0 (NA)
	Median (Range)	3 (2, 31)	12 (12, 12)	3 (1, 31)	12 (12, 12)	3 (2, 31)	12 (12, 12)
Rehabilitation Unit	Number of patients – n (%)	0 (0)	0 (0)	1 (0.6)	0 (0)	1 (0.6)	1 (1.3)
	Hospitalization with time spent in rehabilitation unit – n (%)	0 (0)	0 (0)	1 (2.3)	0 (0)	1 (2.0)	1 (3.3)
	Mean (SD)	0 (0)	0 (0)	60.0 (NA)	0 (0)	60.0 (NA)	12.0 (NA)
	Median (Range)	0 (0, 0)	0 (0, 0)	60 (60, 60)	0 (0, 0)	60 (60, 60)	12 (12, 12)

Patients who had duration of 0 hospitalization days at the ward they stayed at (i.e., date of admission is the same as date of discharge) were excluded from the calculation for length of hospital stay.
Abbreviations: ER, emergency room; SD, standard deviation.

LIMITATIONS

- The exploratory nature of the HCRU endpoint, the small sample size, and the low number of HCRU events from ASCEMBL prevented any robust conclusions about the economic impact of asciminib use from being made.
- The relative frequency of HCRU and mean length of stay in hospital were heavily influenced by select patients due to small number of observations.
- Self-reported HCRU data by the patients can result in underestimation of the number of visits or introduce recall bias.
- The patterns of HCRU as observed in the trial setting might not reflect routine clinical practice.

Acknowledgements

This study was supported by Novartis Services Inc. The authors thank Parash Mani Bhandari, MSc (EVERSANA™) for medical writing assistance. All authors contributed to, critically reviewed for intellectual content, and approved the presentation.

Disclosures

JEC: Consultant for Novartis, Pfizer, Takeda and Sun Pharma; KS: Received research funding and advisory board payments from Novartis; DR: Received honoraria and/or advisory board payments from Incyte, Novartis and Pfizer; MJM: Consultant for Novartis, BMS, Takeda and Pfizer; DT and PW: Employees of EVERSANA™, Inc who were paid consultants to Novartis; KJ and AY: Employees and shareholders of Novartis

References

1. American Cancer Society (2022) Key Statistics for Chronic Myeloid Leukemia.

2. Réa D, Mauro MJ, Boquimpani C, et al. (2021) A phase 3, open-label, randomized study of asciminib, a STAMP inhibitor, vs bosutinib in CML after 2 or more prior TKIs. Blood 138 (21): 2031-2041.

3. Réa D, Mauro MJ, Hochhaus A, et al. (2022) Efficacy and safety results from ASCEMBL, a phase 3 study of asciminib versus bosutinib (BOS) in patients (pts) with chronic myeloid leukemia in chronic phase (CML-CP) after ≥2 prior tyrosine kinase inhibitors (TKIs): Week 96 update. J Clin Oncol 40 (16_suppl): 7004.

4. Smith BD, Cortes JE, Réa D, et al. (2021) CML-353: Health Care Resource Utilization (HCRU) with Asciminib and Bosutinib among Patients with Chronic Myeloid Leukemia in Chronic Phase (CML-CP) Previously Treated with ≥2 Tyrosine Kinase Inhibitors (TKIs): Results from the Multicenter, Open-Label Phase 3 ASCEMBL Trial. Clin Lymphoma Myeloma Leuk 21 S334.