Clinical, Humanistic, and Economic Burden in Fibrodysplasia Ossificans Progressiva (FOP): A Systematic Literature Review

SA20

Juliette C. Thompson[1], Qian Xin[1], Elaine A. Böing[2]

[1]Visible Analytics, Oxford, UK; [2]Ipsen, Cambridge, MA, USA

Background

- Fibrodysplasia ossificans progressiva (FOP) is an ultra-rare, severely debilitating genetic disorder characterised by congenital skeletal malformations and irreversible heterotopic ossification (HO) in soft and connective tissues.[1]

Objectives

This review identified clinical, humanistic, and economic published literature to elucidate the burden of illness associated with FOP.

Methods

- Searches were conducted in MEDLINE, Embase, and Cochrane (2000 to 09-Jun-2022); related conferences were manually searched. Dual screening, data extraction, and qualitative analysis were conducted.
- The eligibility criteria are summarised in **Table 1.**

Results

- From 1,429 abstracts identified, 53 publications were included (**Figure 1**); most reported clinical data (42/53), however humanistic (5/53) and economic (11/53) data were also identified.
- Information identified was primarily derived from three relatively new sources that have collated data from large numbers of individuals with FOP:
 - the FOP Natural History Study (NCT02322255),[2–12]
 - the FOP Registry (~34% of known FOP population internationally [2019]),[13–18] and
 - the FOP International Burden of Illness (BoI) Survey (NCT04665323).[19–21]
- The following clinical trials also provided relevant information:
 - Phase II[22] and III[23,24] (MOVE) trials for palovarotene,
 - Phase II trial (LUMINA-1) for garetosmab.[25]

Epidemiology

- Prevalence of FOP per million inhabitants was estimated between 0.36 in Spain,[26] 0.88 in the USA,[27] and 1.36 (95% CI: 1.1-1.68) in France.[28]

Clinical data

- FOP is progressive, with the neck and upper back earliest affected (at age 8.5 and 9.5 years, respectively) and wrists and feet affected later in life (54.5 and 58.5 years).[15]
- Over 12 months, 48% of individuals reported >1 flare-up frequently characterised by swelling, pain and decreased movement.[4]
- Consistent with the progressive nature of the disease, the number of regions affected by HO increased from a mean of 3.1 in those aged <8 years to 8.8 in those aged 25-65 years;[2] the HO volume also increased with age.[2]
- The median age of death regardless of cause was 56 years, with excess mortality becoming particularly apparent in patients from 30 years of age. Cardiorespiratory failure from thoracic insufficiency syndrome was the most common cause of death (54%).[29]
 - The median age at which the chest was affected by HO was 38.5 years (95%CI: 30.5 to not reached), which reflects the increased mortality observed in patients over the age of 30.[15]

Humanistic data

- Patients with FOP experiencing flare-ups consistently reported poorer mental health and a higher degree of emotional problems compared with those patients without flare-up.[14]
- Adults (15 to <25 years and 25-65 years) reported similar PROMIS scores, whereas older children (8-14 years) reported worse PROMIS scores than younger children (<8 years).[2,20]
- Individuals with FOP with the most severe degree of joint impairment (PRMA level 4) had very poor quality of life (EQ-5D-5L 0.05); the EQ-5D-5L score for the whole population was influenced by the higher number of patients in the PRMA level 4 category (mean 0.24; SD 0.36) (**Table 2**).[20]

Economic data

- Over 12 months, 86% of individuals with FOP required ≥1 physician visit and 27% were hospitalised (**Figure 2**).[13]
- In older age groups (≥15 years vs <15 years), there was increased use of assistive devices and number of hospitalisations, which limited patients' ability to attend school and thus become employed; 67% of patients did not have a paid occupation.[19-21]
- Most people with FOP required caregivers who were usually patients' direct relatives;[30] 51.3% of primary caregivers felt that caring for someone with FOP impacted their career decisions.[19]

Table 1. Eligibility criteria

	Epidemiology	Clinical	Humanistic	Economic
Patients	Individuals (any age) with a diagnosis of fibrodysplasia ossificans progressiva (FOP)			
Interventions/ Comparators	No restriction	Pharmacological and non-pharmacological interventions	No restriction	No restriction
Outcomes	• Incidence • Prevalence	• Heterotopic ossification (HO) outcomes • Cumulative analogue joint scale (CAJIS) • FOP Physical Function Questionnaire (FOP-PFQ) • Pain • Activities of daily living • Safety • Mortality	• Utility scores • Disutility scores • Health-related quality of life measures (e.g., PROMIS)	• Direct costs • Indirect costs • Resource use • Economic model (including, CEA, CUA, BIA, CMA) results (e.g., ICER, LYG, cost per QALY)
Study design	No restriction	• Clinical trials (RCTs, NRS) • Observational studies	No restriction	No restriction
Exclusion criteria	• Studies not meeting the inclusion criteria outlined above • Observational case reports and small case series (<5 people)		• Published <2000 • Non-English language publications	

Figure 1. PRISMA flow diagram

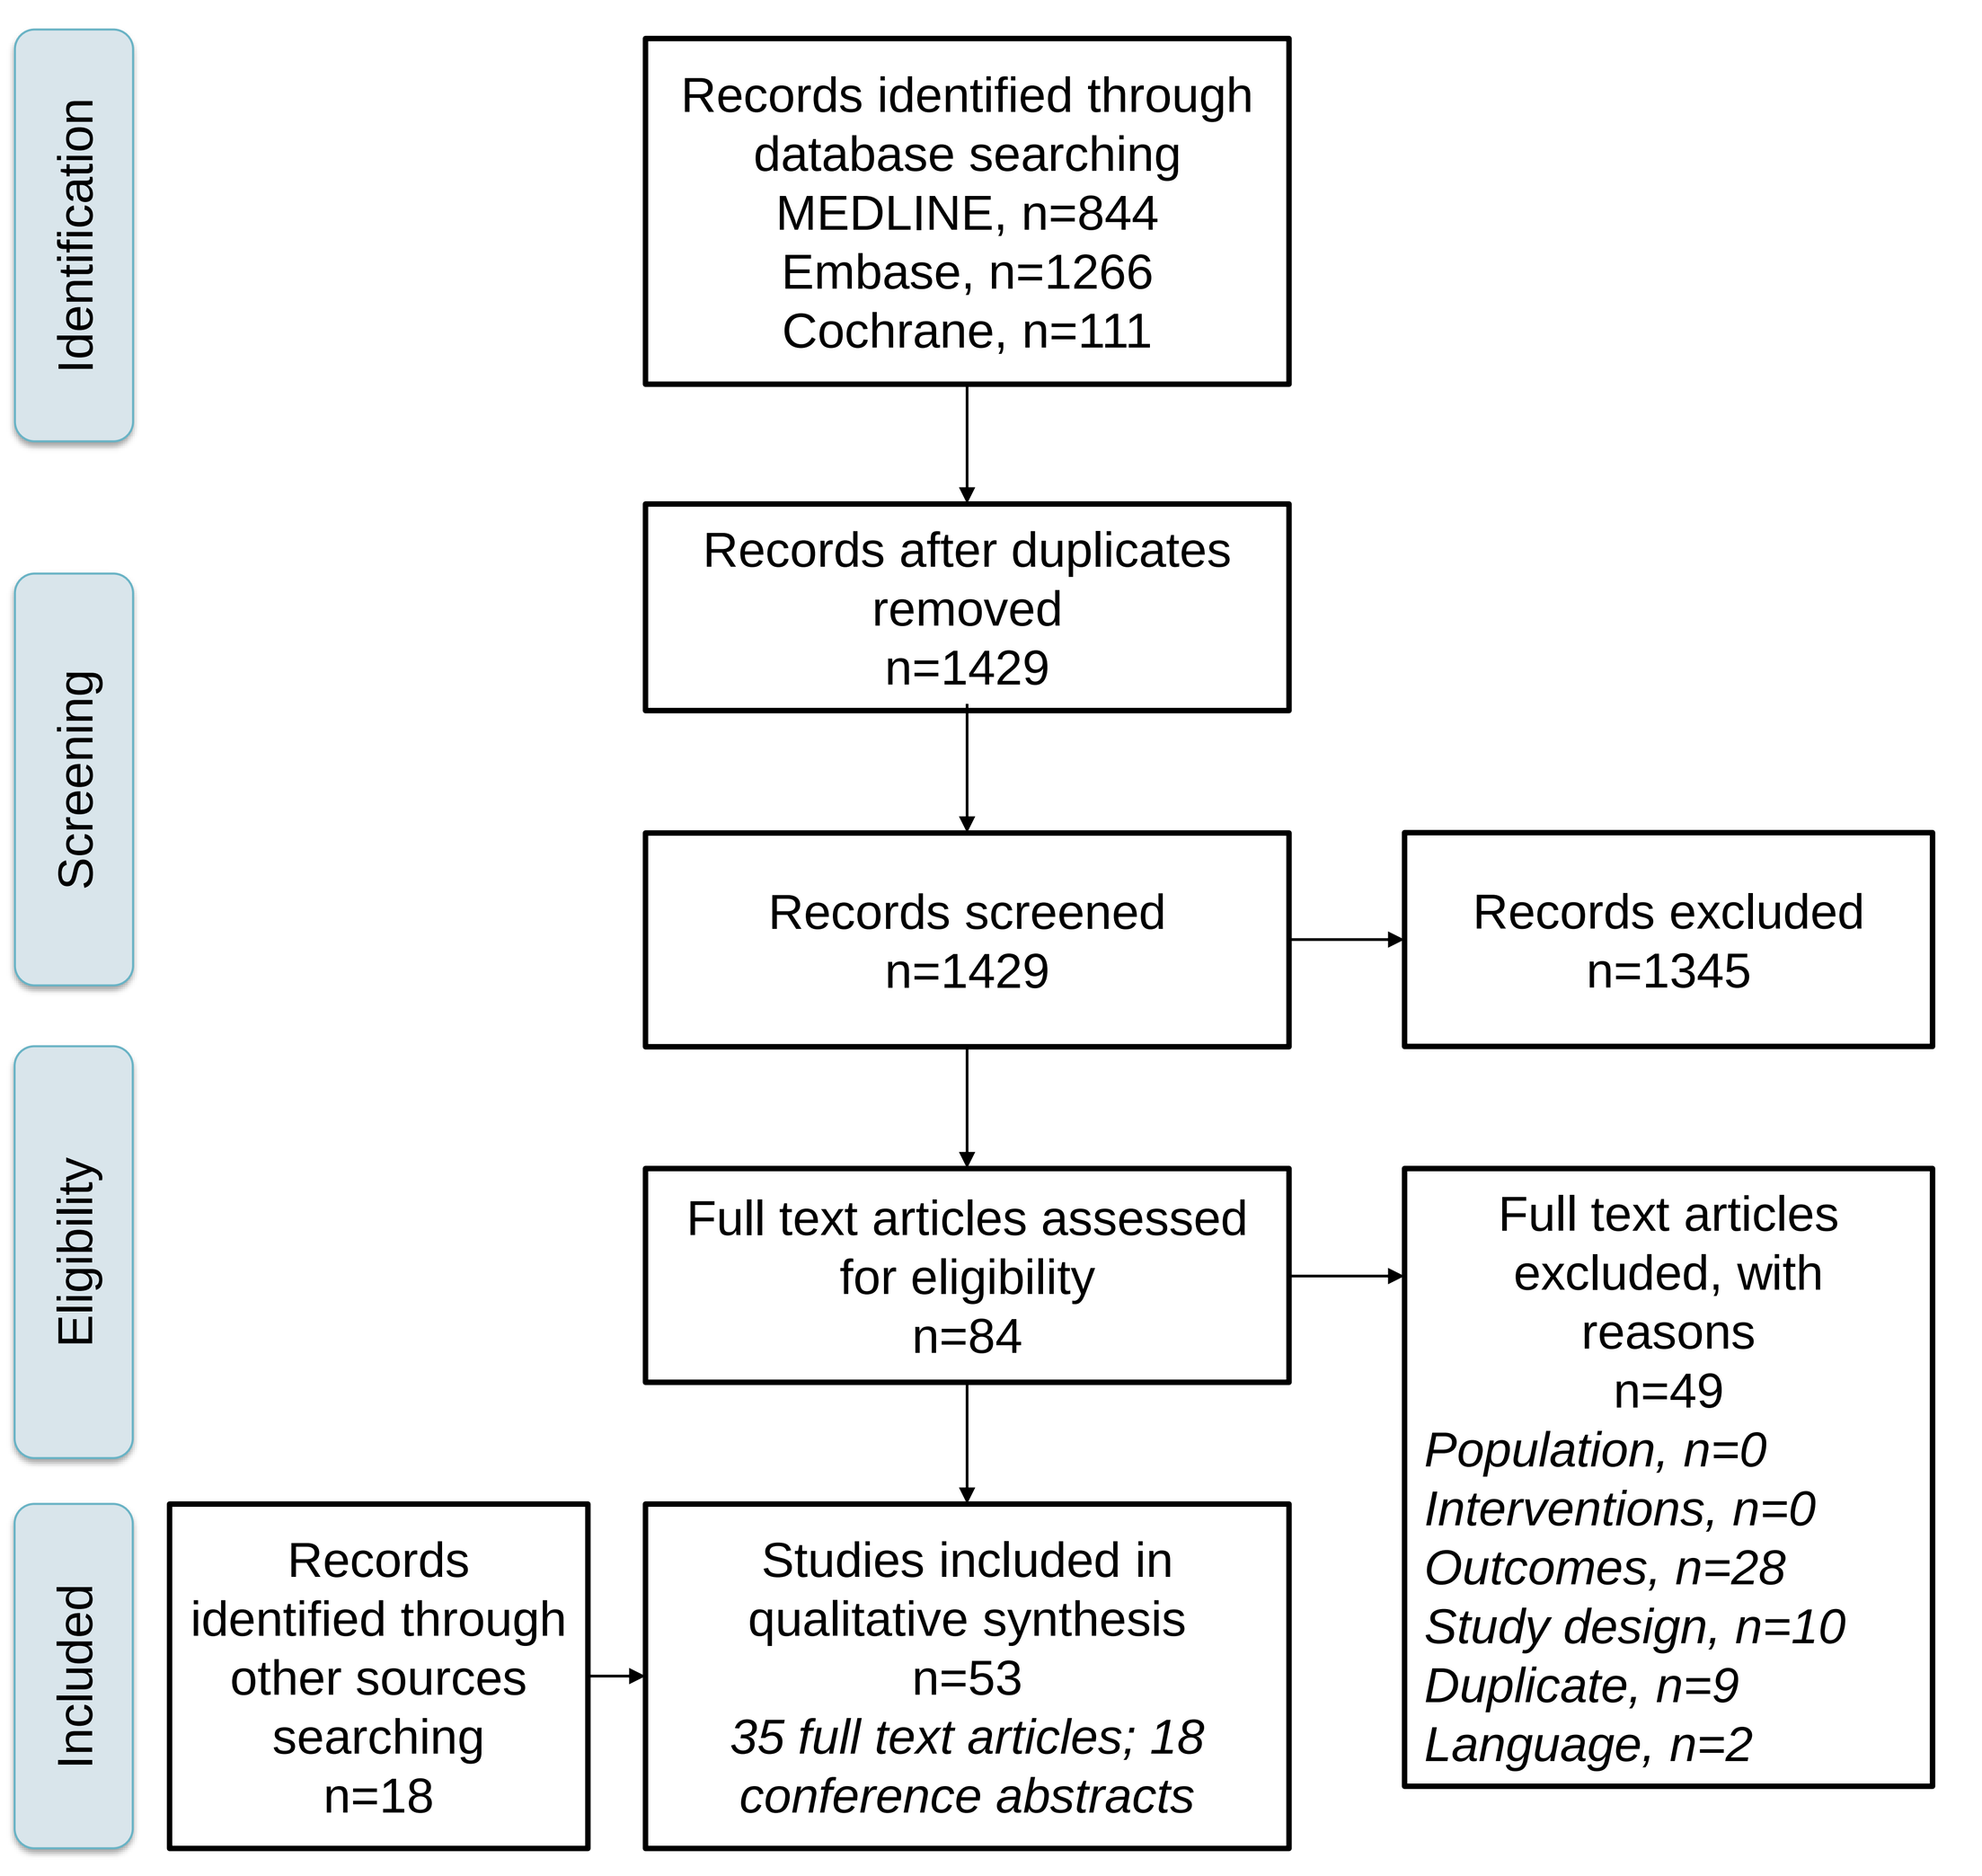
Identification
Screening
Eligibility
Included
Records identified through database searching MEDLINE, n=844 Embase, n=1266 Cochrane, n=111
Records after duplicates removed n=1429
Records screened n=1429
Records excluded n=1345
Full text articles assessed for eligibility n=84
Full text articles excluded, with reasons n=49 Population, n=0 Interventions, n=0 Outcomes, n=28 Study design, n=10 Duplicate, n=9 Language, n=2
Records identified through other sources searching n=18
Studies included in qualitative synthesis n=53 35 full text articles; 18 conference abstracts

Figure 2. Encounters with medical and dental care providers in the 12 months prior to enrolment (n=299) (FOP Registry)[13]

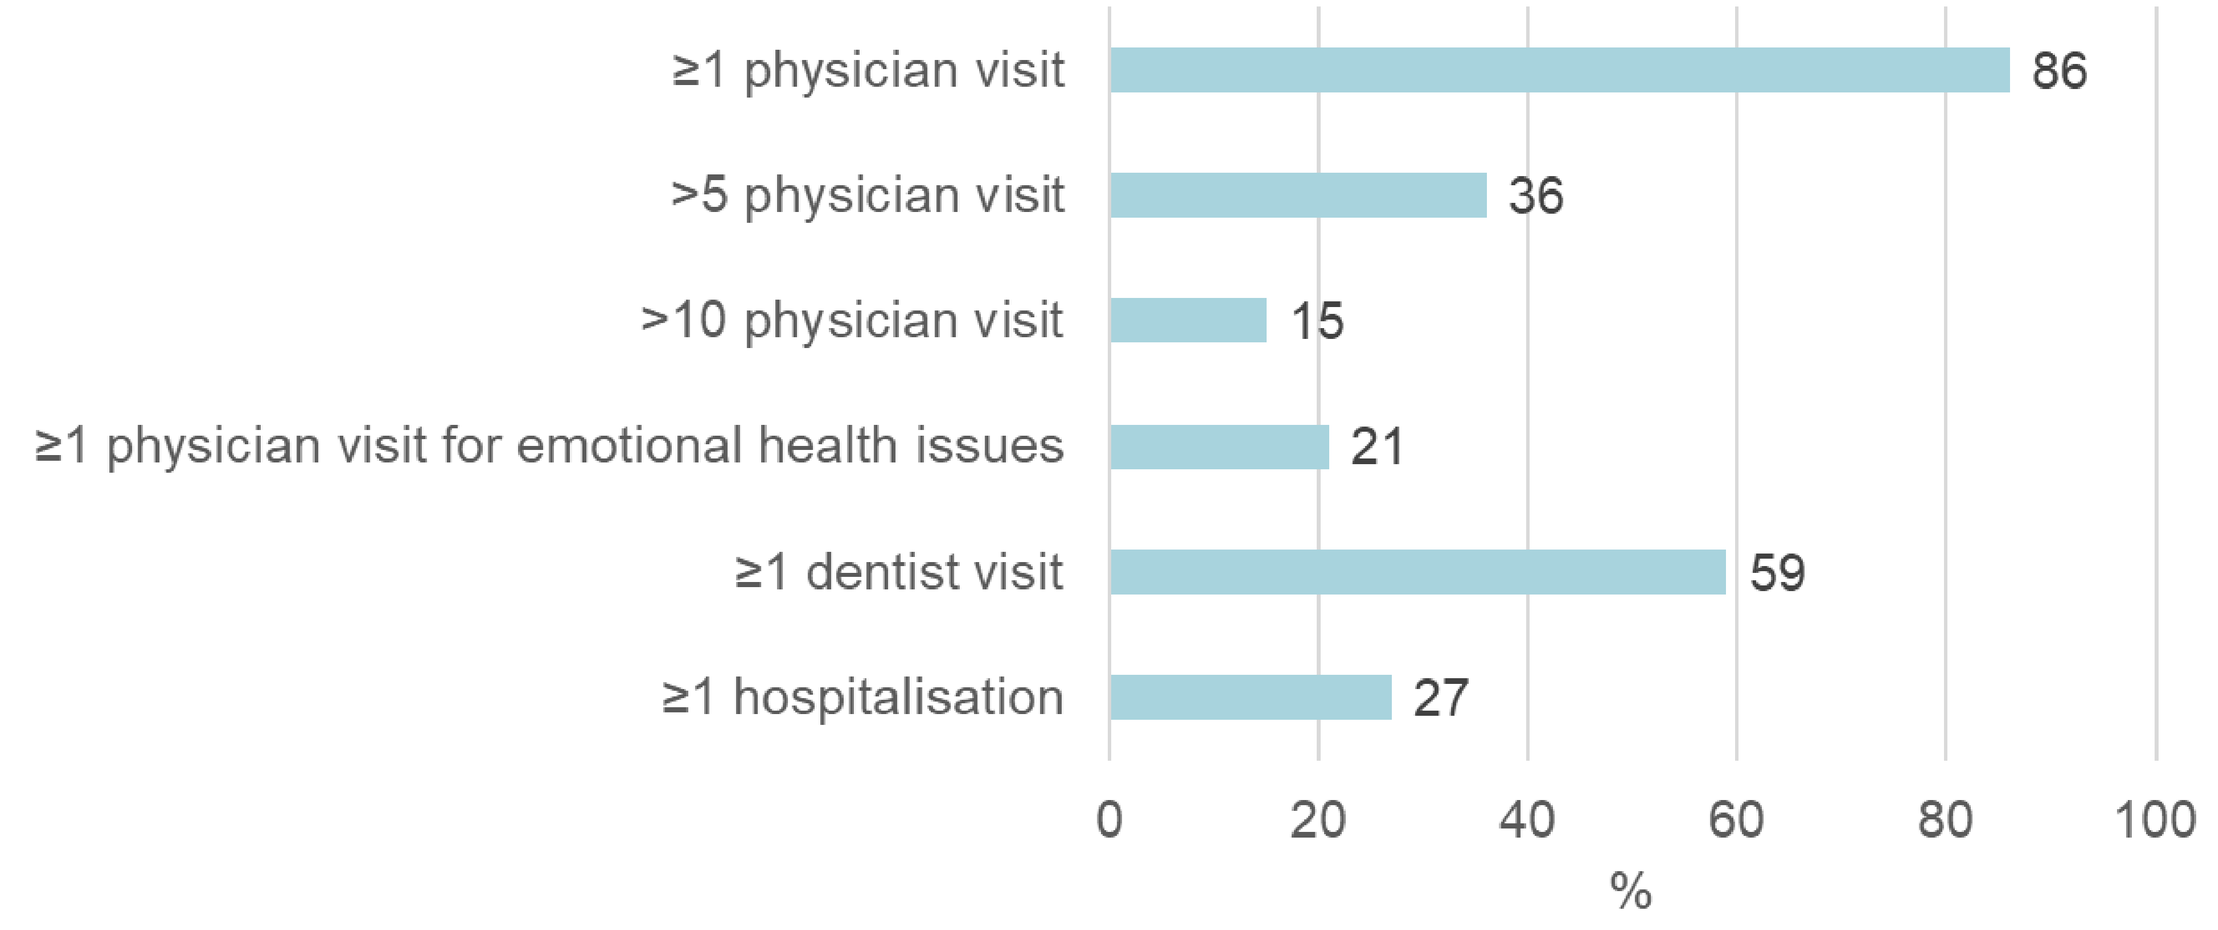
≥1 physician visit
86
>5 physician visit
36
>10 physician visit
15
≥1 physician visit for emotional health issues
21
≥1 dentist visit
59
≥1 hospitalisation
27
0
20
40
60
80
100
%

Table 2. EQ-5D-5L from the International BoI Survey[20]

Participant group	Degree of mobility limitations	n	Mean (SD)
Individuals with FOP (≥13 years)	PRMA Level 1 (total score: 0-6)	20	0.61 (0.26)
	PRMA Level 2 (total score: 7-12)	31	0.46 (0.34)
	PRMA Level 3 (total score: 13-18)	33	0.21 (0.30)
	PRMA Level 4 (total score: ≥19)	62	0.05 (0.26)
	All	161	0.24 (0.36)
All family members	-	238	0.85 (0.21)

The PRMA scores 12 joints and three body regions as 0 (not limited at all), 1 (moderately limited), or 2 (extremely limited); total scores range from 0–30, with higher scores representing more severe limitations in mobility

CONCLUSIONS

- **The estimated prevalence of FOP varies by geography.**
- **FOP is progressive, with the number of regions affected and volume of HO increasing with age.**
- **There is still limited information on the humanistic and economic burden associated with FOP, especially related to resource use and costs.**
- **Recent, and ongoing, trials indicate that research efforts have increased to address an unmet need for more therapeutic options for patients with FOP.**

References available with poster download

Author contributions Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: JCT, QX, EAB; Drafting of the publication, or revising it critically for important intellectual content: JCT, QX, EAB; Final approval of the publication: JCT, QX, EAB

Disclosures JCT, QX: Employees of Visible Analytics; EAB: Employee and shareholder of Ipsen.

Abbreviations

BIA: budget impact analysis; BoI: Burden of Illness; CEA: cost-effectiveness analysis; CMA: cost-minimisation analysis; CT: computed tomography; CUA: cost-utility analysis; FOP: Fibrodysplasia ossificans progressiva; HO: heterotopic ossification; ICER: incremental cost-effectiveness ratio; LYG: life-years gained; NRS: non-randomised study; PRMA: Patient-Reported Mobility Assessment; PROMIS: Patient-Reported Outcomes Measurement Information System; QALY: quality-adjusted life-year; RCT: randomised controlled trial

For further information, please send your question(s) to **Juliette.Thompson@visibleanalytics.co.uk**; **Elaine.Boing@ipsen.com**

Presented at ISPOR Europe | Vienna, Austria | 06–09 November 2022

This study was sponsored by Ipsen