Inpatient Burden of Respiratory Syncytial Virus in Children ≤2 Years of Age in Germany: A Retrospective Analysis of Nationwide Hospitalization Data, 2019-2021

Moritz Wick[1], Oliver Damm[1], Rolf Kramer[1], Mathieu Bangert[1]

[1]Sanofi-Aventis Deutschland GmbH, Berlin, Germany

Scan here to download e-poster
EPH182

INTRODUCTION

Respiratory syncytial virus (RSV) is a pathogen that causes respiratory tract disease, such as pneumonia, bronchitis, and bronchiolitis in seasonal waves. Infants and young children are primarily affected. However, there is limited data describing the epidemiological and economic burden of RSV at national level in Germany.

Objective

This study aims to quantify the number of RSV-associated hospitalizations in children ≤2 years of age, and related inpatient resource use and costs in Germany.

METHODS

- We retrospectively analyzed publicly available nationwide hospitalization data from the Institute for the Hospital Remuneration System (InEK, so-called *§21-dataset*) for the calendar years 2019 to 2021.
- All hospital inpatient stays with a principal diagnosis of severe acute respiratory infection (SARI, ICD-10 codes J09-J22) were extracted.
- RSV cases were defined as cases with a principal RSV-specific ICD-10 diagnosis of either J12.1 (RSV-pneumonia), J20.5 (RSV-bronchitis), or J21.0 (RSV-bronchiolitis).
- Mean length of stay (LOS), intensive care unit (ICU) admissions, mechanical ventilation, in-hospital deaths and the distribution of allocated diagnosis related groups (DRG) were retrieved for RSV-related hospitalizations.
- Mean hospitalization costs per RSV case were estimated using the age group-specific distribution of DRGs and corresponding relative weights, multiplied with federal base rates. For 2020 and 2021, nursing care revenues were considered due to changes in the German DRG system.

Figure 1: SARI hospitalizations and proportion of RSV in children aged ≤2 years, by age group and year

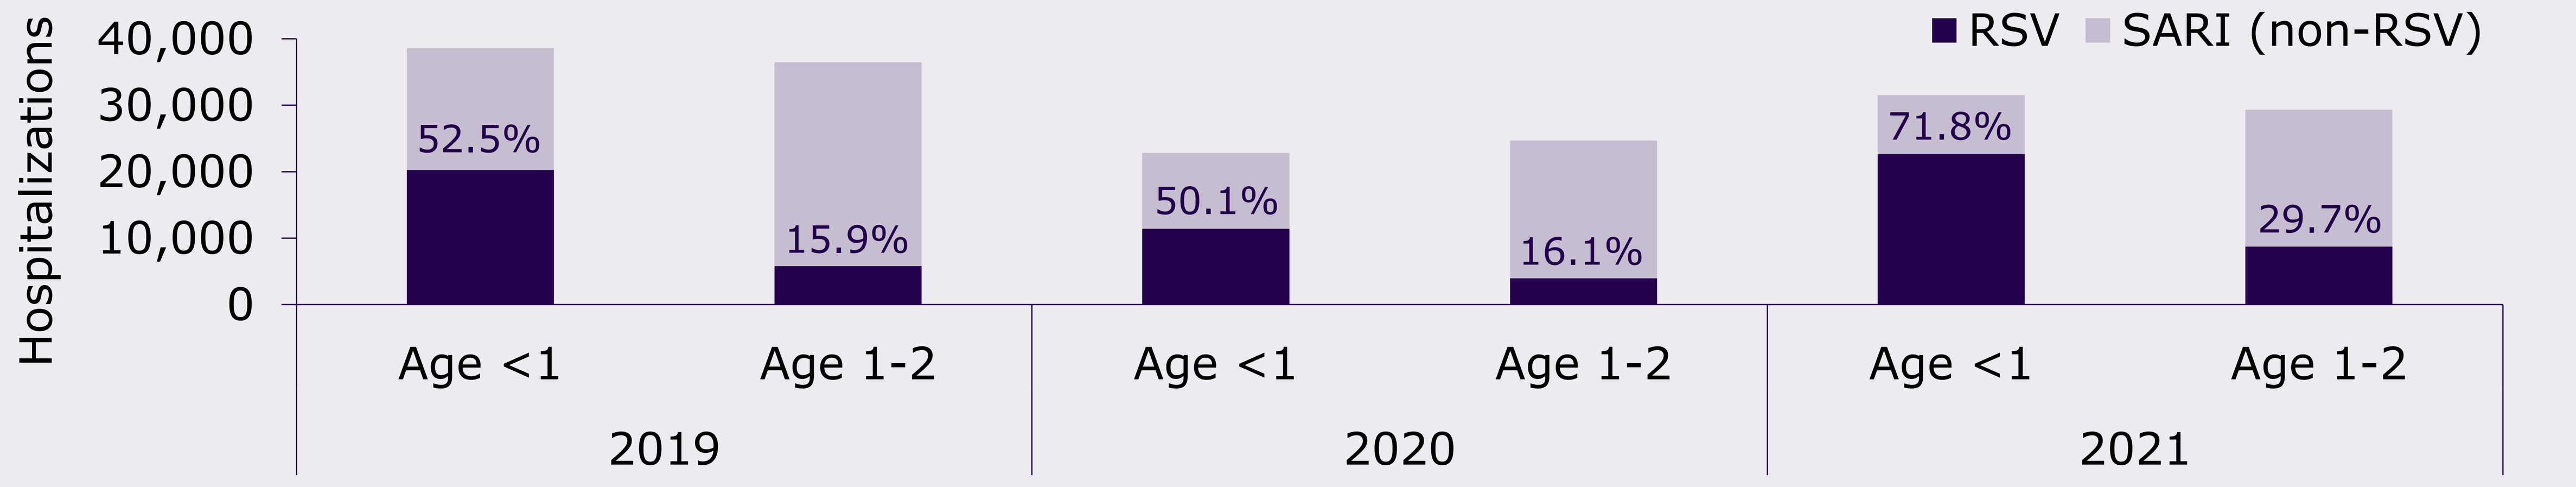
Hospitalizations
40,000
30,000
20,000
10,000
0
RSV
SARI (non-RSV)
52.5%
15.9%
50.1%
16.1%
71.8%
29.7%
Age <1
Age 1-2
2019
2020
2021

SARI: Severe acute respiratory infection, RSV: Respiratory syncytial virus

Figure 2: Principal diagnoses of RSV hospitalizations in children ≤2 years of age, by age group and year

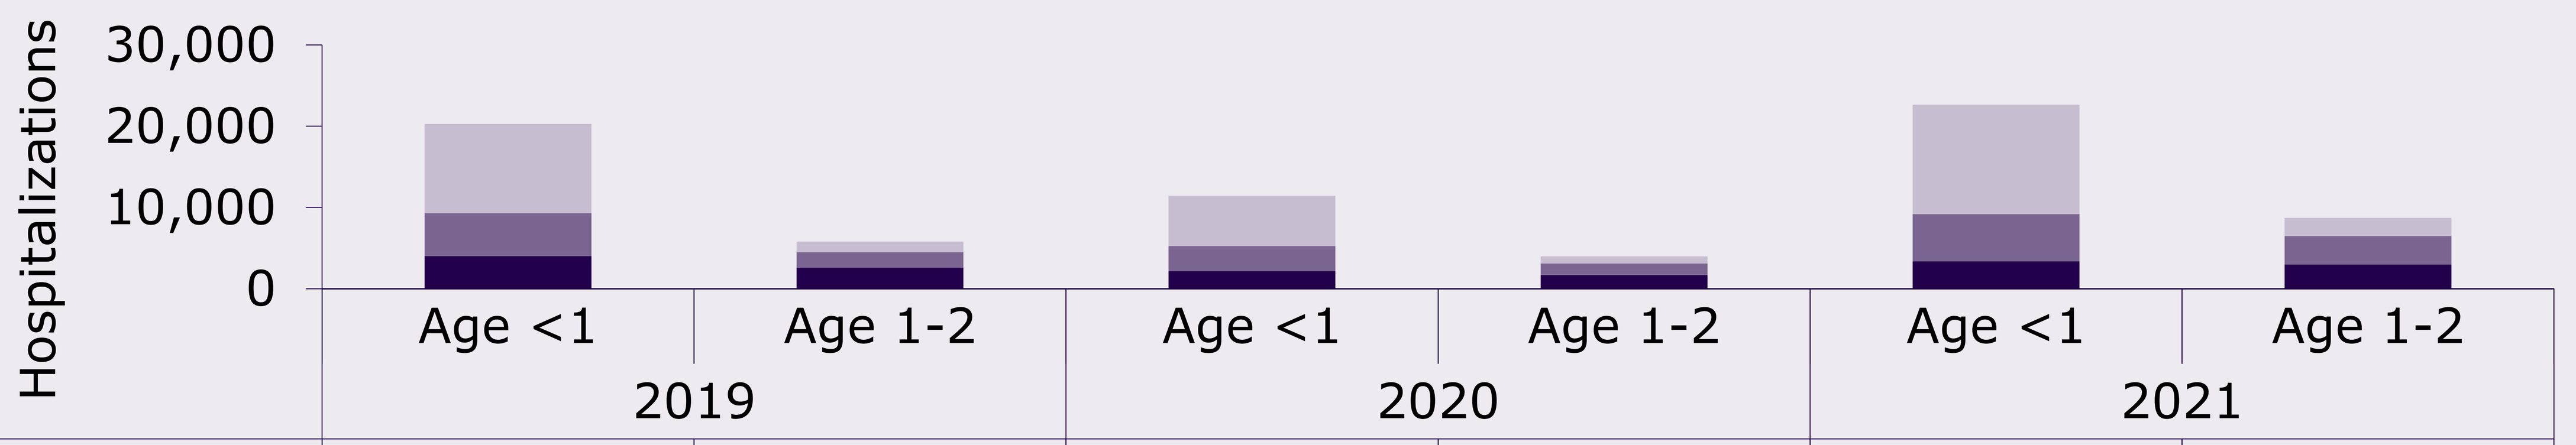
Hospitalizations
30,000
20,000
10,000
0

	2019 Age <1	2019 Age 1-2	2020 Age <1	2020 Age 1-2	2021 Age <1	2021 Age 1-2
J21.0 (bronchiolitis)	10,967	1,275	6,192	880	13,458	2,247
J20.5 (bronchitis)	5,300	1,917	3,067	1,405	5,808	3,484
J12.1 (pneumonia)	4,002	2,591	2,170	1,693	3,378	2,987

RSV: Respiratory syncytial virus

RESULTS

- From 2019 to 2021, there were 72,821 hospitalizations (26,052, 15,407 and 31,362 in 2019, 2020 and 2021, respectively) with a principal diagnosis of RSV in children ≤2 years of age in Germany (see Table 1 for details).
- Hospitalization incidence between 2019 and 2021 ranged from 14.9 to 28.6 per 1,000 population and year in children aged <1, and from 2.5 to 5.6 in children aged 1-2 years.
- With 54,342 RSV hospitalizations over the study period, infants <1 year of age were most affected. Figure 1 shows that RSV caused 58.5% of SARI hospitalizations in this age group.
- In children ≤2 years of age, mean LOS was 4.5 days, 5.6% of cases were admitted to ICU, and 4.6% were mechanically ventilated over the study period.
- 48.1% of RSV hospitalizations among children aged ≤2 years were associated with bronchiolitis, followed by bronchitis (28.8%) and pneumonia (23.1%). In infants <1 year of age, bronchiolitis was the clinical manifestation in 56.3% of RSV cases (see Figure 2).
- In 2019 and 2020, seasonal peaks of RSV hospitalizations were reached in calendar weeks (CW) 6 and 7, while the RSV season in 2021 was shifted, culminating in CW43 (see Figure 3).
- The mean hospitalization costs of a child ≤2 years of age with RSV were €3,458 (SD €3,216), €3,001 (SD €2,775), and €3,444 (SD €3,389) in 2019, 2020, and 2021, respectively.
- Severe RSV hospitalization cases of children ≤2 years of age involving ICU admission were associated with average costs of €8,652 (SD €9,382), €7,825 (SD €7,287), and €9,577 (SD €8,436), for 2019, 2020, and 2021, respectively.

Table 1: Hospitalization incidence, health care resource use, and case-fatality associated with RSV hospitalizations in children ≤2 years of age, by age group and year

Year	Age Group (years)	Hospitali-zations	Incidence (n/1,000)	Gender (% male)	Mean LOS (SD)	ICU admission rate (%)	Ventilation rate (%)	Deaths (CFR%)
2019	<1	20,269	26.2	56.5	4.7 (3.2)	5.3	3.8	1 (<0.01)
	1-2	5,783	3.6	53.7	4.6 (3.7)	3.6	1.4	2 (0.03)
	Total ≤2	**26,052**	**11.0**	**55.9**	**4.7 (3.3)**	**4.9**	**3.3**	**3 (0.01)**
2020	<1	11,429	14.9	56.7	4.7 (3.1)	7.1	6.8	2 (0.02)
	1-2	3,978	2.5	53.5	4.4 (2.8)	4.7	1.3	2 (0.05)
	Total ≤2	**15,407**	**6.6**	**55.9**	**4.6 (3.0)**	**6.4**	**5.4**	**4 (0.03)**
2021	<1	22,644	28.6	56.3	4.4 (2.9)	6.5	6.9	6 (0.03)
	1-2	8,718	5.6	54.2	4.2 (2.7)	4.0	0.9	1 (0.01)
	Total ≤2	**31,362**	**13.3**	**55.7**	**4.3 (2.8)**	**5.8**	**5.3**	**7 (0.02)**

CFR: case-fatality rate, ICU: Intensive-care unit, LOS: Length of stay, RSV: Respiratory syncytial virus, SD: Standard deviation

Conclusions

- RSV causes substantial disease burden and is a leading cause of SARI-associated hospital admissions in children aged ≤2 years in Germany.
- Infants <1 years of age are most affected and are associated with higher health care resource use than children aged 1-2 years.
- Our results confirm the need to explore and evaluate strategies to prevent RSV in infants and young children.

Figure 3: RSV hospitalizations of children ≤2 years of age, by calendar week of hospital discharge, 2019-2021

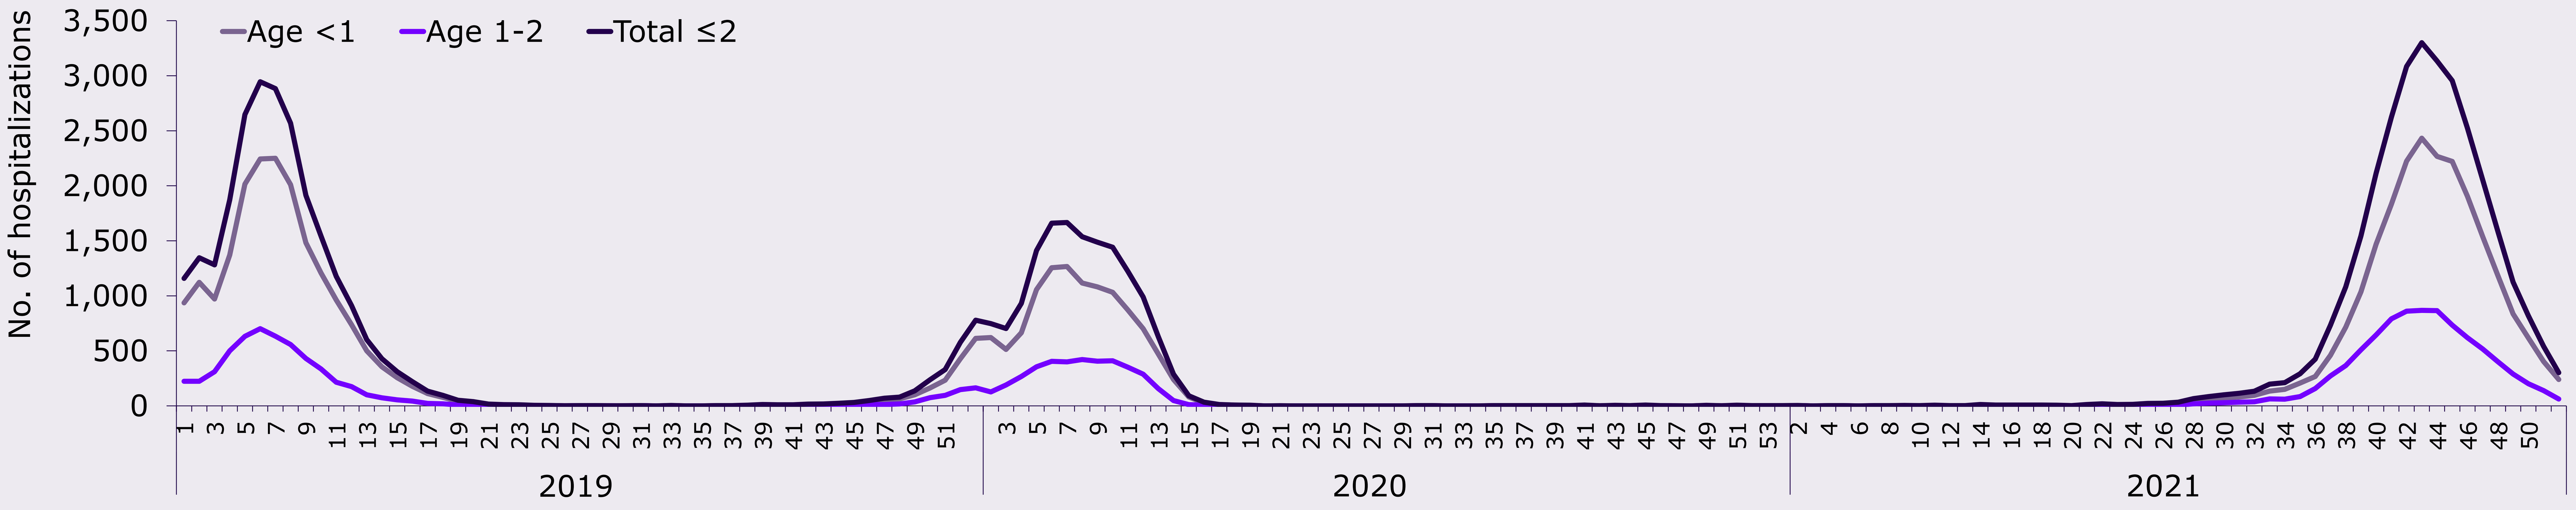
No. of hospitalizations
3,500
3,000
2,500
2,000
1,500
1,000
500
0
Age <1
Age 1-2
Total ≤2
2019
2020
2021

RSV: Respiratory syncytial virus

Funding: Sanofi-Aventis Deutschland GmbH

Contact: moritz.wick@sanofi.com

Poster presented at ISPOR Europe 2022, Vienna, 6-9 November 2022