The cost-utility results presented in this poster are based on the costs inputs from 1st March 2022 in Switzerland

ISPOR EU 2022
Vienna, Austria

COST-UTILITY OF NABIXIMOLS FOR THE MANAGEMENT OF SPASTICITY DUE TO MULTIPLE SCLEROSIS IN SWITZERLAND

Poster Session IV
EE411

Schwander B[1], Dykukha I[2], Merinova M[3], Akmaz B[2]

[1]AHEAD GmbH, Bietigheim-Bissingen, Germany; [2]Almirall Hermal GmbH, Reinbek, Germany; [3]Almirall SA, Barcelona, Spain

OBJECTIVES

- Nabiximols oromucosal spray (Sativex®), a complex botanical containing THC and CBD as well as other cannabinoid and non-cannabinoid components, was approved by Swissmedic in 2013 for treatment of moderate-to-severe spasticity due to multiple sclerosis (MS).
- We investigated cost-utility of nabiximols in Switzerland using the Markov model developed by Oppe et al. 2021 [1].

METHODS

- The Markov model-based analysis uses a probabilistic bootstrapping sensitivity analysis approach to evaluate the cost–utility of nabiximols as an add-on therapy compared to standard-of-care (SoC) alone.
- Healthcare costs and quality adjusted-life years (QALY) were simulated in an Excel-based model including four health states.
- As presented in **Figure 1**, three health states related to spasticity severity and one to death were simulated over a five-year time horizon.
- The 28-day model cycles matched study design.

Figure 1: Schematic Representation of the Markov Model

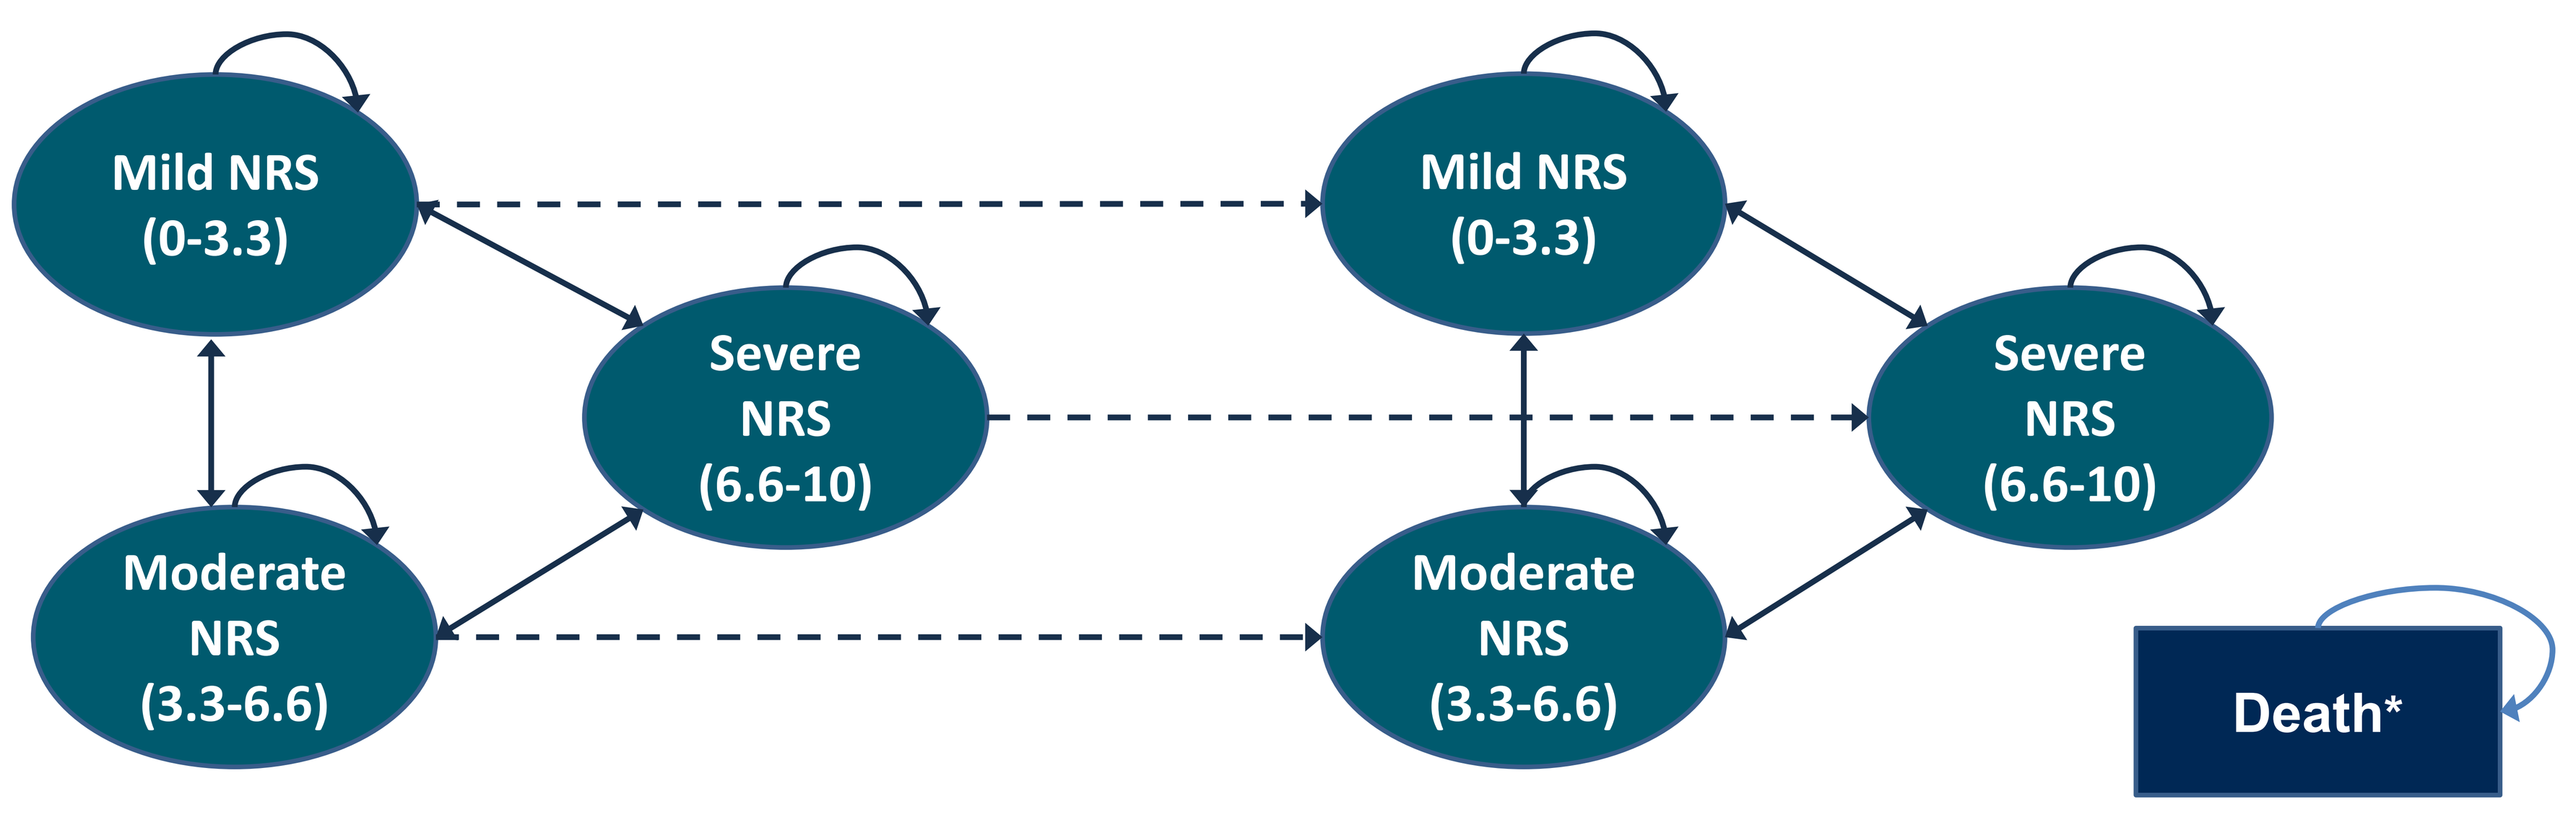

SoC + Nabiximols
SoC
Mild NRS (0-3.3)
Severe NRS (6.6-10)
Moderate NRS (3.3-6.6)
Mild NRS (0-3.3)
Severe NRS (6.6-10)
Moderate NRS (3.3-6.6)
Death*

discontinuation from Nabiximols; *patients may transition to death from all states;
NRS: spasticity severity Numerical Rating Scale; SoC: Standard-of-Care
Reproduced from Oppe et al. 2021 [1]

- The transition probabilities between the mild, moderate, and severe states for the SoC + nabiximols arm for cycles 1–5 were calculated using the data from the SAVANT trial (EudraCT 2015-004451-40) [2]. In the base case scenario, transition probabilities for cycle 6 and subsequent cycles were assumed to be the same as those for cycle 5. Hence active treatment effects were assumed to be maintained after the end of the trial period, which is realistic according to published literature [3, 4].
- Transition probabilities between the mild, moderate, and severe spasticity state for the SoC arm were based on the retrospective observational study by Arroyo et al. 2011 [5]. Rationale for not choosing the SAVANT trial [2] data was the much longer time span (> 2 years) of the Arroyo et al. study [5]. We included a scenario (scenario 1) where the transition probabilities for the SoC arm were based on the SAVANT trial [2] in the sensitivity analyses (see **Figure 2** below).
- Transition probabilities for death were based on Swiss life tables [6].

Figure 2: Source of the Data used to Determine the Transition Probabilities for the Three Scenarios

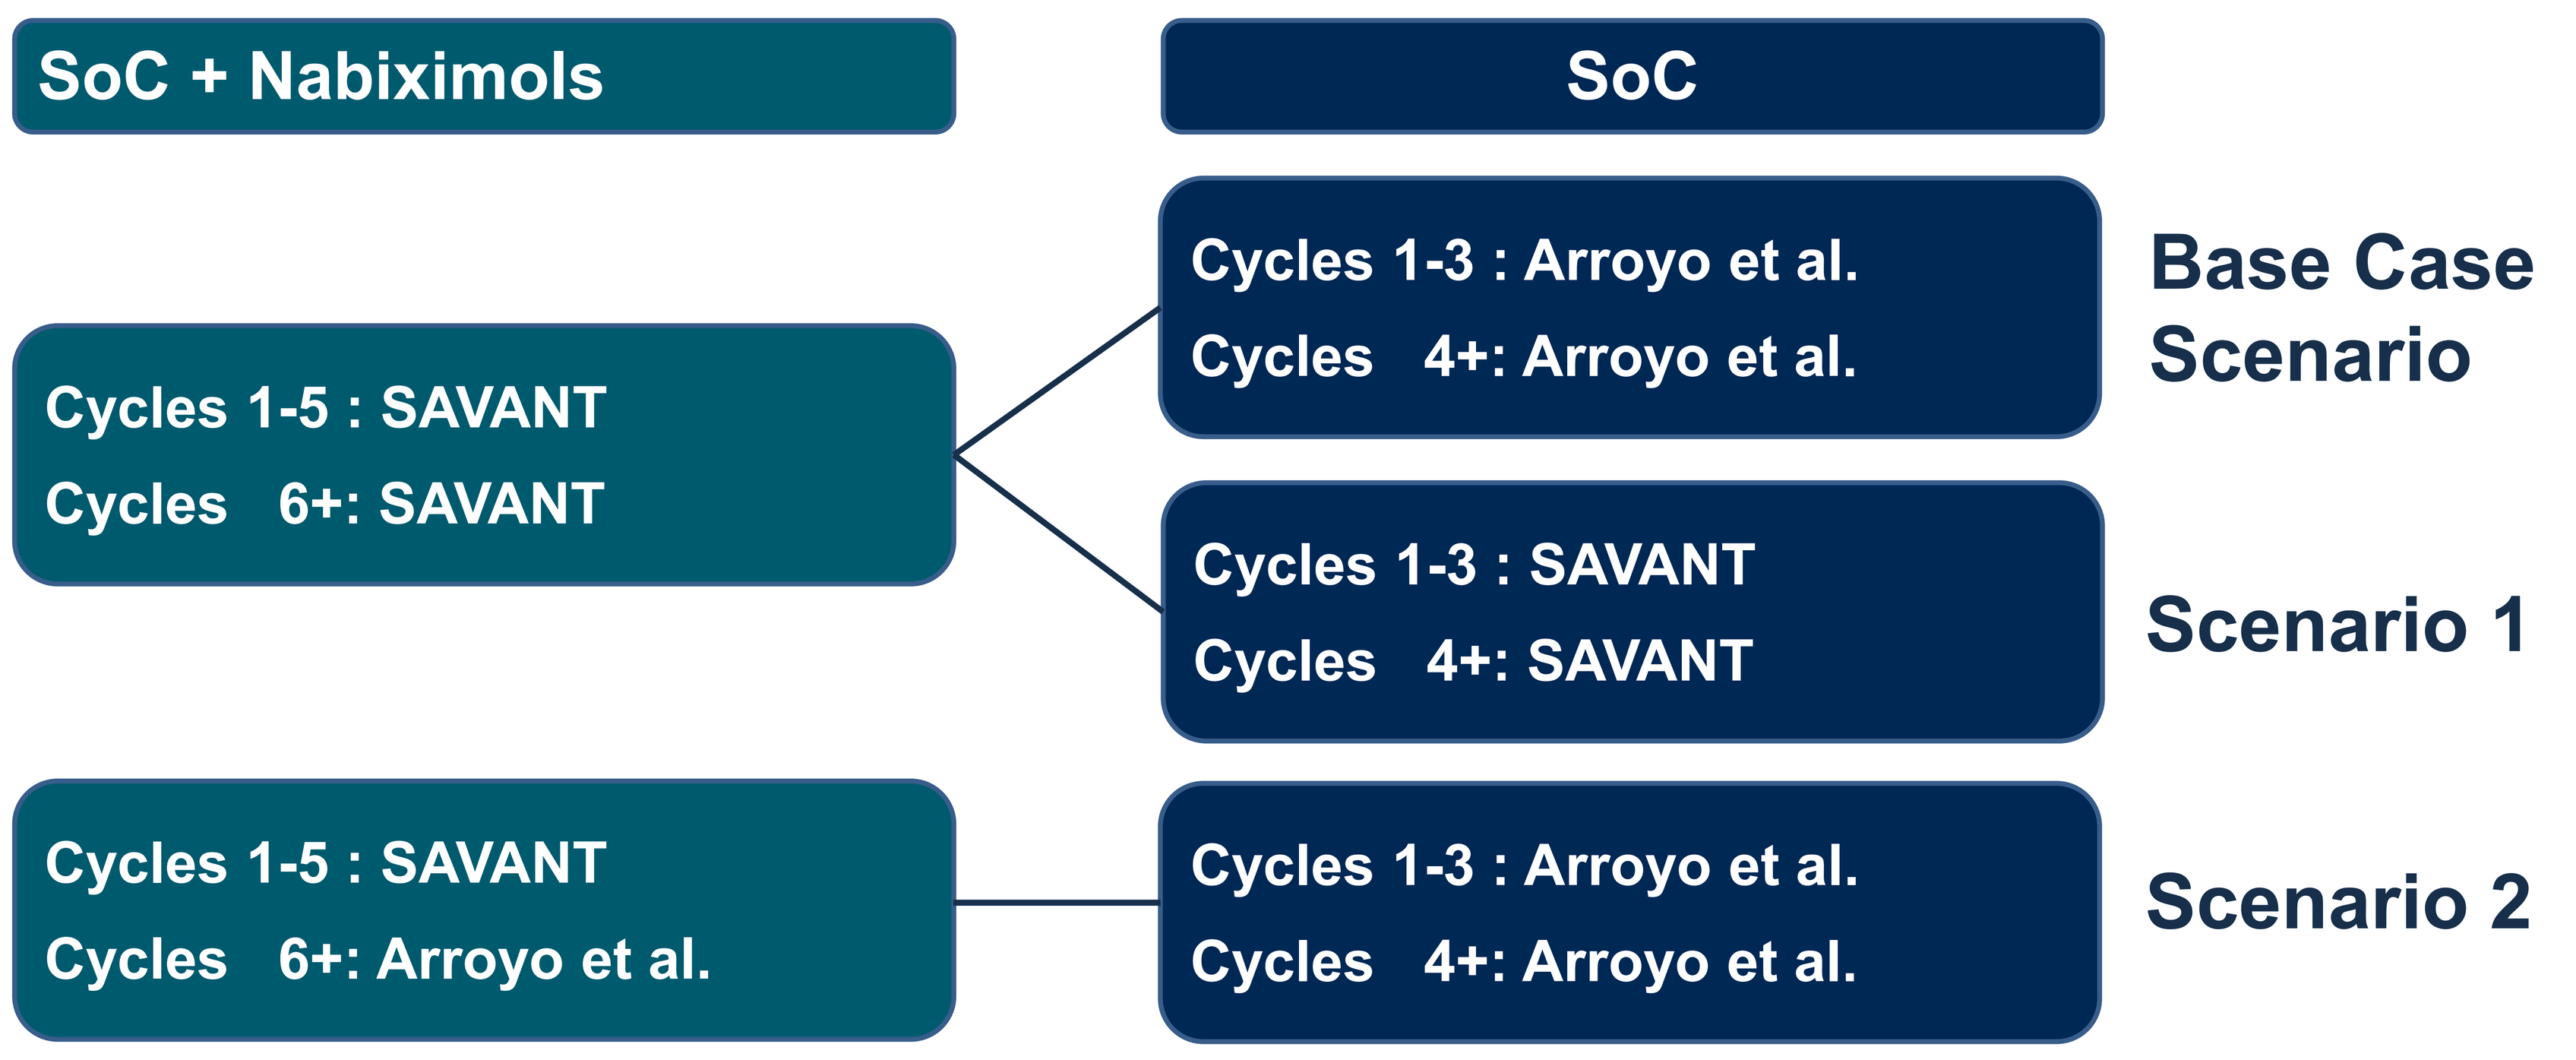

SoC + Nabiximols
SoC
Cycles 1-5 : SAVANT
Cycles 6+: SAVANT
Cycles 1-3 : Arroyo et al.
Cycles 4+: Arroyo et al.
Base Case Scenario
Cycles 1-3 : SAVANT
Cycles 4+: SAVANT
Scenario 1
Cycles 1-5 : SAVANT
Cycles 6+: Arroyo et al.
Cycles 1-3 : Arroyo et al.
Cycles 4+: Arroyo et al.
Scenario 2

Reproduced from Oppe et al. 2021 [1]

RESULTS

- After one year of treatment with nabiximols, nabiximols dominated SoC with per patient mean cost savings of CHF 41.54 and a QALY gain of 0.038.
- This dominance became more pronounced over time; in the fifth year the per patient mean cost savings were CHF 15,642.67 and a QALY gain of 0.169. The probability of dominance increased from 47% in the first year to 94% in the fifth year.
- The cost-utility results for all scenarios analyzed are presented in **Table 1**.
- The probabilistic sensitivity results for the base are presented in **Figure 3** for Year 5. which show the cost saving potential of nabiximols

Table 1: Overview of Cost-Utility Results by Analysis Scenario

Scenario	Time Horizon	Incremental Costs	Incremental QALYs	ICER	Dominance Probability
Base Case Scenario	Year 1	-41.54 CHF	0.038	-1,098.43 CHF	47.3%
	Year 2	-3,637.84 CHF	0.079	-45,935.94 CHF	78.5%
	Year 3	-7,749.92 CHF	0.116	-66,546.26 CHF	88.3%
	Year 4	-11,644.90 CHF	0.149	-78,399.01 CHF	90.9%
	Year 5	-15,642.67 CHF	0.169	-92,770.71 CHF	94.2%
Scenario 1	Year 1	748.78 CHF	0.031	24,100.30 CHF	32.1%
	Year 2	387.37 CHF	0.055	7,092.83 CHF	41.1%
	Year 3	-115.16 CHF	0.069	-1,658.75 CHF	46.9%
	Year 4	-435.71 CHF	0.079	-5,517.32 CHF	49.9%
	Year 5	-507.82 CHF	0.085	-5,942.76 CHF	51.6%
Scenario 2	Year 1	957.09 CHF	0.022	43,848.04 CHF	32.3%
	Year 2	-2,113.89 CHF	0.054	-39,486.39 CHF	65.4%
	Year 3	-6,086.24 CHF	0.087	-70,055.22 CHF	80.1%
	Year 4	-9,748.61 CHF	0.111	-87,552.86 CHF	87.0%
	Year 5	-14,143.66 CHF	0.133	-106,417.44 CHF	90.5%

QALY = quality adjusted-life years; ICER = Incremental Cost-Effectiveness Ratio

Figure 3: Incremental Cost-Effectiveness Scatterplots reflecting the Base Case Analyses for Year 5

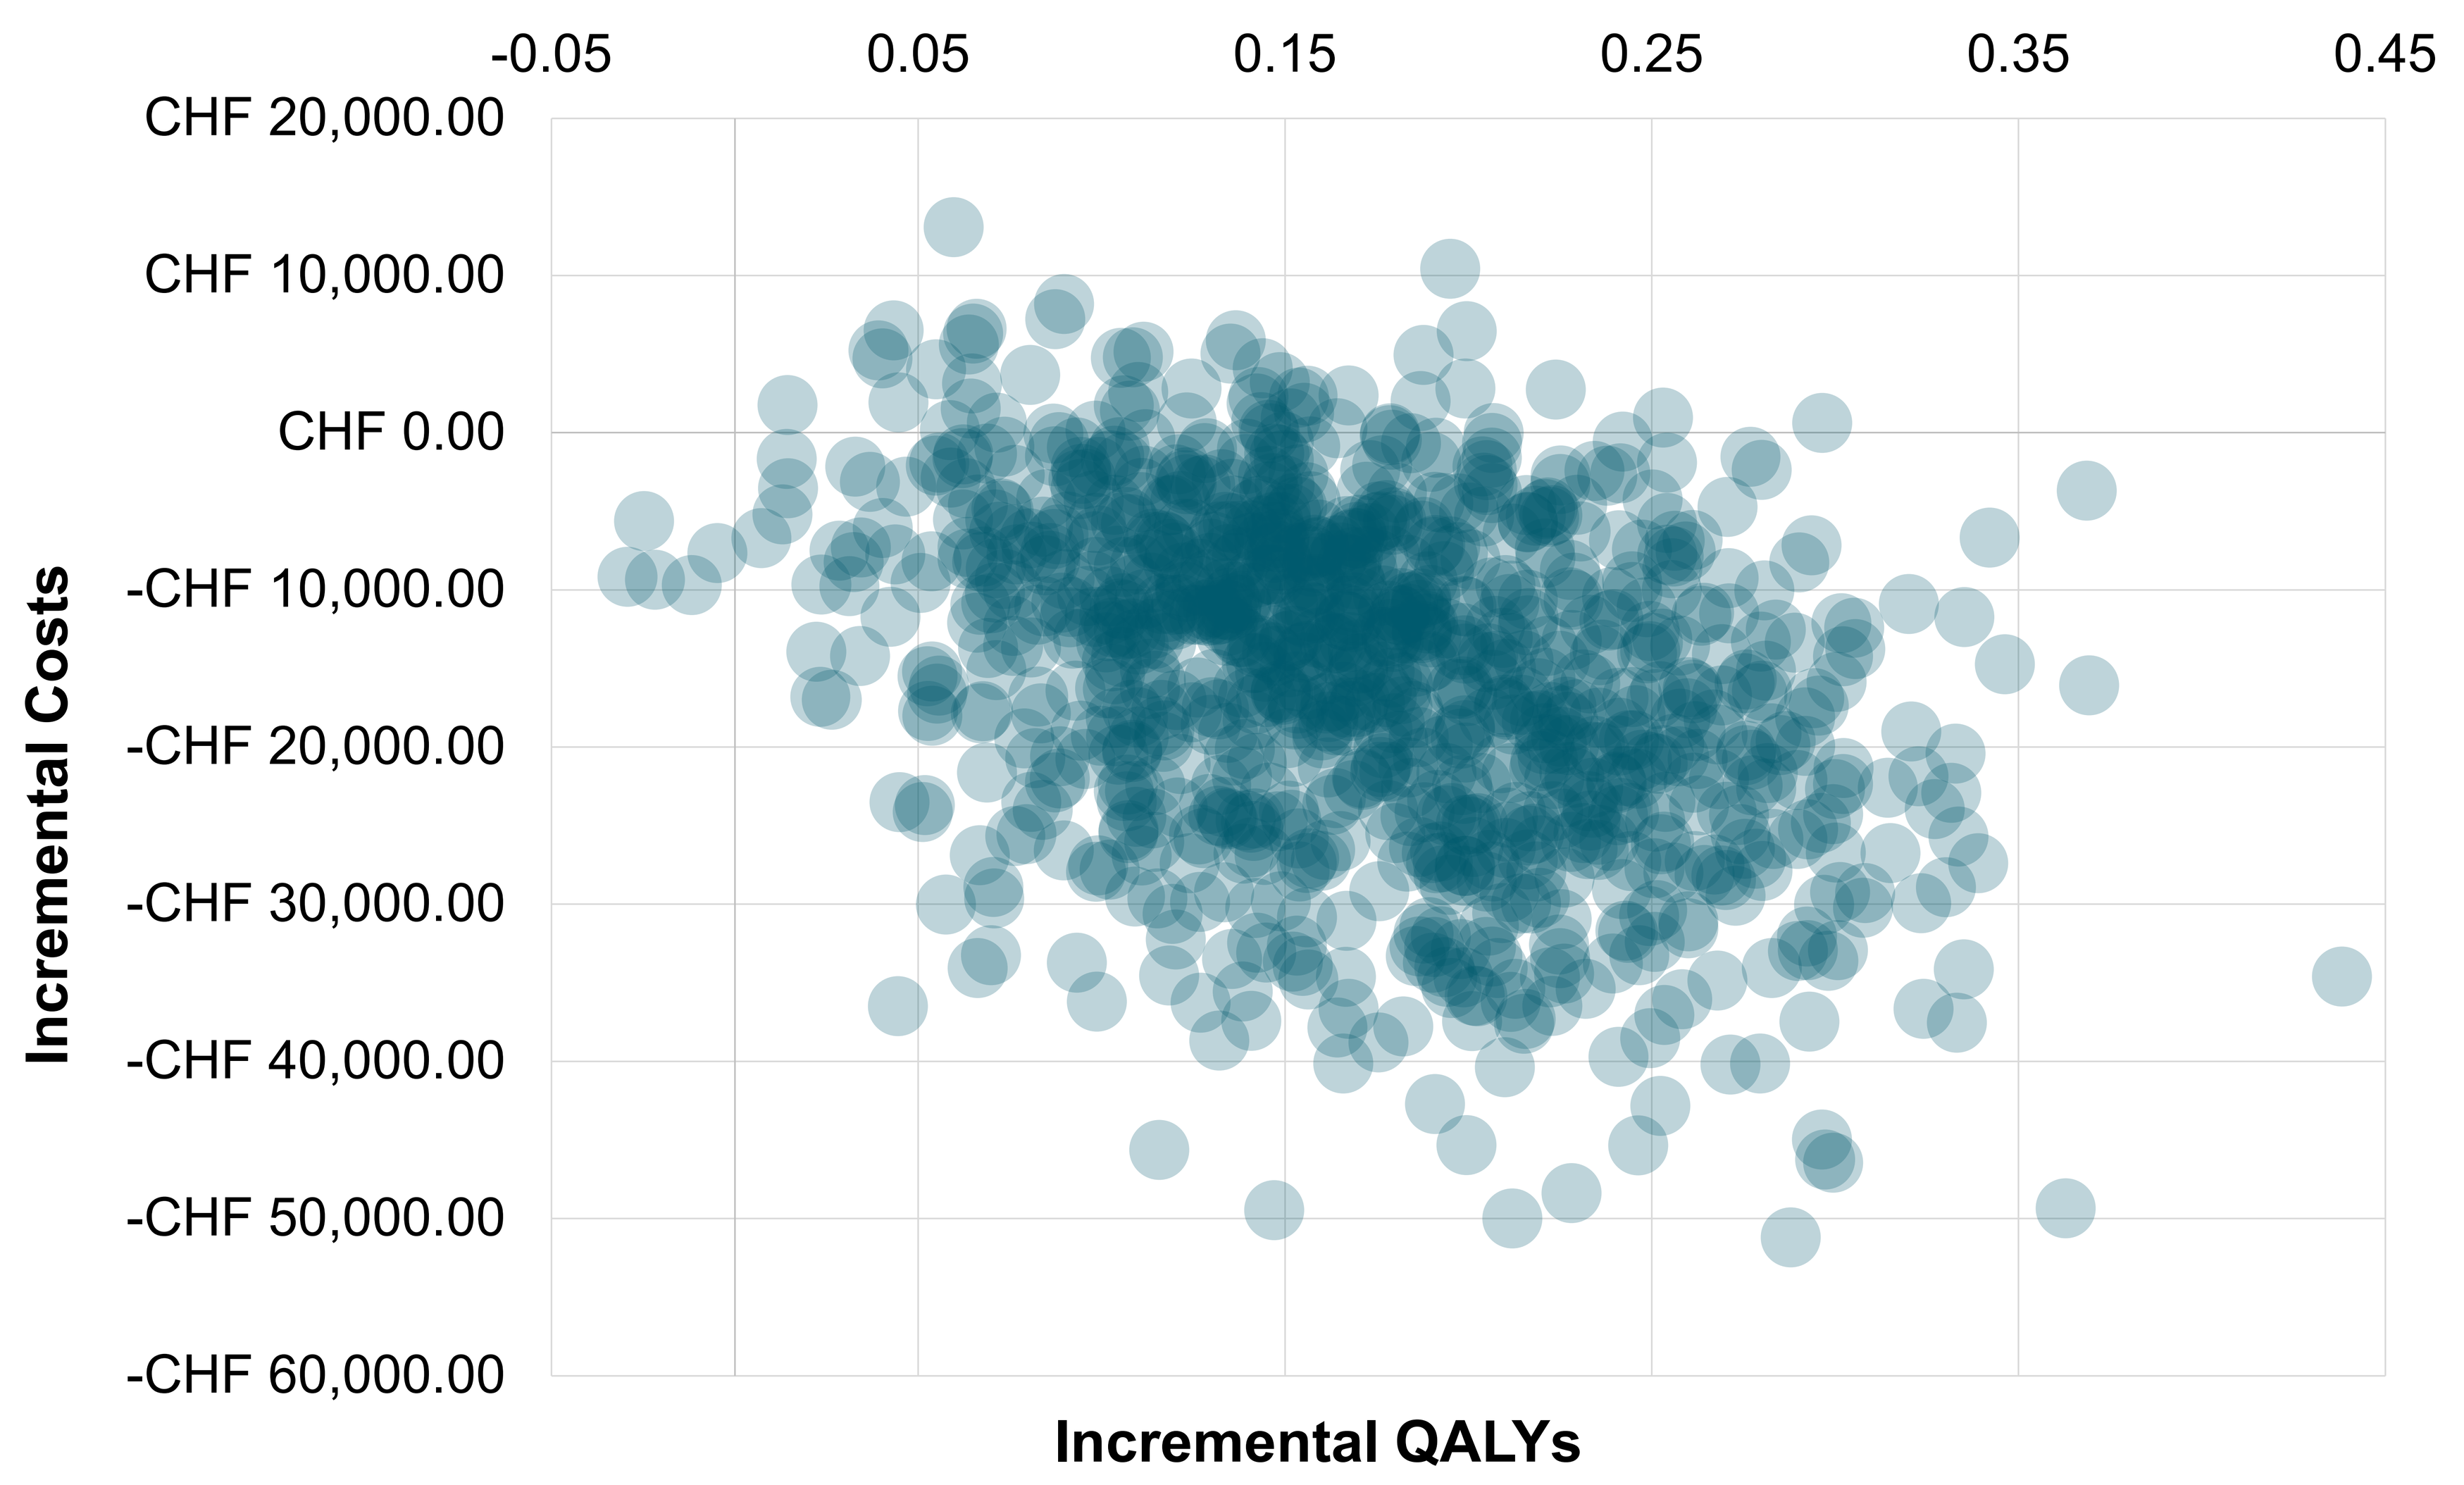

-0.05
0.05
0.15
0.25
0.35
0.45
CHF 20,000.00
CHF 10,000.00
CHF 0.00
-CHF 10,000.00
-CHF 20,000.00
-CHF 30,000.00
-CHF 40,000.00
-CHF 50,000.00
-CHF 60,000.00
Incremental Costs
Incremental QALYs

CONCLUSIONS

- In the Swiss healthcare setting, treatment of moderate-to-severe spasticity due to MS with nabiximols according to the approved label was found to be cost-saving and providing QALY gains comparing to SoC alone.

LIMITATIONS

- The cost-utility results presented in this poster are based on the costs inputs from March 2022 in Switzerland.

REFERENCES

[1] Oppe M, Ortín-Sulbarán D, Vila Silván C, et al. Cost-effectiveness of adding Sativex® spray to spasticity care in Belgium: using bootstrapping instead of Monte Carlo simulation for probabilistic sensitivity analyses. Eur J Health Econ. 2021 Jul;22(5):711-721.
[2] Markovà J, Essner U, Akmaz B, et al. Sativex® as add-on therapy vs. further optimized first-line ANTispastics (SAVANT) in resistant multiple sclerosis spasticity: a double-blind, placebo-controlled randomised clinical trial. Int J Neurosci. 2019 Feb;129(2):119-128.
[3] Flachenecker P, Henze T, Zettl UK. Long-term effectiveness and safety of nabiximols (tetrahydrocannabinol/cannabidiol oromucosal spray) in clinical practice. Eur Neurol. 2014;72(1-2):95-102.
[4] Oreja-Guevara, C., Casanova, B., Ordás, C.M., et al. Observational safety study of THC:CBD Oromucosal Spray (Sativex) in multiple sclerosis patients with spasticity. Clin. Exp. Pharmacol. 2015; 5(184):2161-1459.
]5] Arroyo R, Vila C, Clissold S. Retrospective observational study of the management of multiple sclerosis patients with resistant spasticity in Spain: the '5E' study. Expert Rev Pharmacoecon Outcomes Res. 2011 Apr;11(2):205-13.
[6] Swiss Federal Statistical Office. Life Tables for Males and Females 2020. URL: https://www.bfs.admin.ch/bfs/de/home/statistiken/bevoelkerung/geburten-todesfaelle/lebenserwartung, last accessed on 13th September 2022
[7] Kobelt G, Berg J, Lindgren P, et al. Costs and quality of life of multiple sclerosis in Switzerland. Eur J Health Econ. 2006 Sep;7 Suppl 2:S86-95
[8] Swiss Drug Compendium. Costs for MS Drugs included in the CE Model. https://compendium.ch/
[9] Tarmed – Swiss Physician Fee Schedule. Administration Costs for MS Drugs included in the CE Model. https://www.tarmed-browser.ch/de
[10] Analysis List - Swiss Federal Office for Health. Diagnostic Testing Costs for MS Therapies included in the CE Model. https://www.bag.admin.ch/bag/de/home/versicherungen/krankenversicherung/krankenversicherung-leistungen-tarife/Analysenliste.html
[11] Swiss DRG Grouper. Surgical MS Therapy Costs included in the CE Model. https://grouper.swissdrg.org/swissdrg/single, last accessed on 13th September 2022

ISPOR Europe 2022, 6-9 November 2022, Vienna, Austria