PREFERENCES FOR TOENAIL ONYCHOMYCOSIS TREATMENTS AMONG CANADIANS WITH AND WITHOUT DIABETES MELLITUS: A DISCRETE CHOICE EXPERIMENT

PCR146

Lozano-Ortega G[1], Mickle AT[1], Gaudet V[2], Popoff E[1], Barbeau M[2], Mathieu S[3]

[1]Broadstreet Health Economics & Outcomes Research, Vancouver, BC, Canada; [2] Bausch Health, Canada Inc., Laval, QC, Canada; [3]Centre Hospitalier Universitaire, Quebec, QC, Canada

BACKGROUND & OBJECTIVE

- Toenail onychomycosis (fungal infection) affects approximately 7% of Canadians.[1,2]
- Comorbid type I/II diabetes mellitus is associated with both an increased risk of onychomycosis infection and of experiencing associated complications.[3,4]
- Pharmacotherapies that have received a Notice of Compliance by Health Canada include topicals (ciclopirox, efinaconazole) and oral agents (terbinafine, itraconazole).
- Toenail onychomycosis treatment selection is multifactorial; considerations may include severity of nail involvement, mode of administration, efficacy, and side effects. However, little is known about preferences from patients in the management of onychomycosis.
- Presence of diabetes may also affect an individual's preferences for toenail onychomycosis treatment given their need to manage comorbidities, challenges with adverse events (AEs) and polypharmacy, and that such individuals are at higher risk of onychomycosis-related complications.[5]
- **The objective was to understand patient preferences regarding onychomycosis treatment features and how preferences vary among those with and without diabetes.**

METHODS

- A discrete choice experiment (DCE) was conducted; attributes and levels were informed by: a targeted literature review, qualitative interviews with 14 toenail onychomycosis patients (7 with diabetes), and consultation with one clinical expert.
- The DCE comprised 16 choice scenarios developed using the Hahn Shapiro catalogue.[6] Both, choice questions and attribute order within choices were randomized to minimize impact of respondent fatigue.
- Adults residing in Canada with self-reported, physician-diagnosed toenail onychomycosis were recruited online. The sample was stratified according to self-reported diabetes status and onychomycosis treatment experience.
- Clinical and demographic characteristics were summarized using frequency and percent, or mean and standard deviation (SD), as relevant. Data from the DCE were analyzed using conditional logit regression (presented as odds ratios [ORs] with 95% confidence intervals [CIs]) and stratified by diabetes status.
- Ethics approval was provided by Veritas IRB (qualitative: #16575-15:38:275-06-2020; DCE: #2022-2743-9764-1).

RESULTS

- Six attributes (one with 4, and five with 2 levels) were constructed (**Figure 1**).

RESULTS

Figure 1. Discrete choice experiment - attributes and associated levels

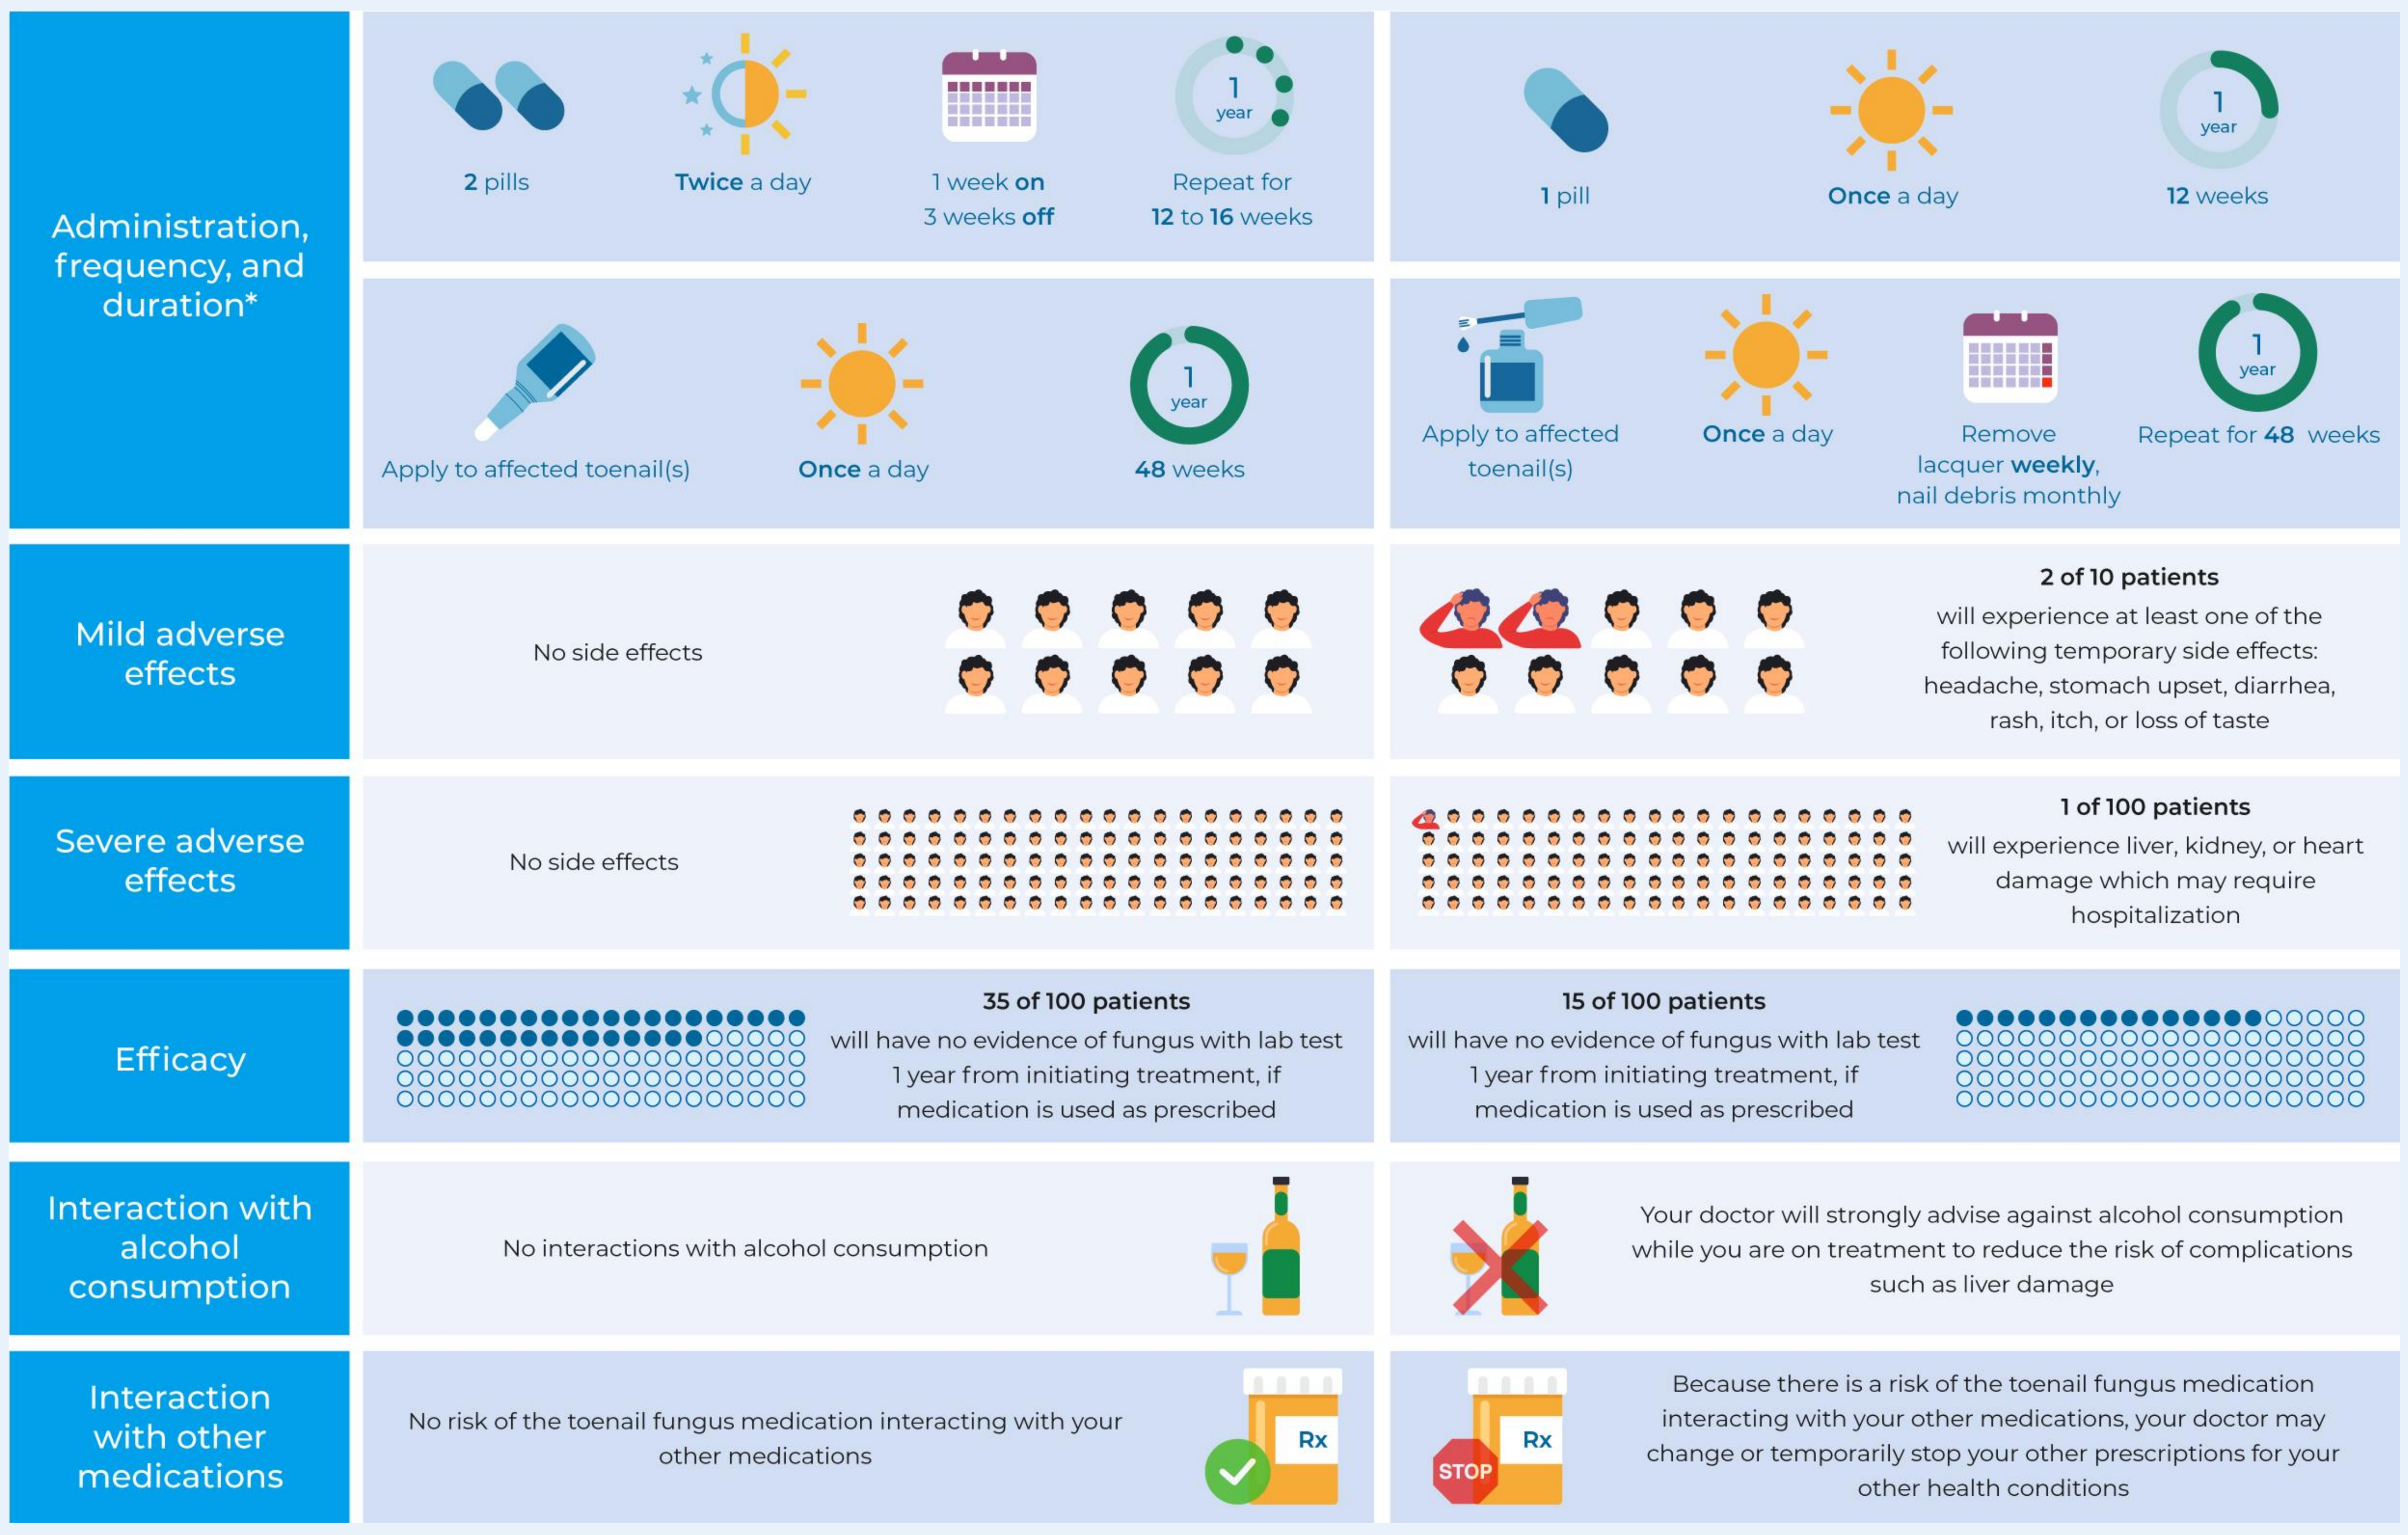
Administration, frequency, and duration*
2 pills
Twice a day
1 week on 3 weeks off
Repeat for 12 to 16 weeks
1 year
1 pill
Once a day
12 weeks
Apply to affected toenail(s)
Once a day
48 weeks
Apply to affected toenail(s)
Once a day
Remove lacquer weekly, nail debris monthly
Repeat for 48 weeks
Mild adverse effects
No side effects
2 of 10 patients
will experience at least one of the following temporary side effects: headache, stomach upset, diarrhea, rash, itch, or loss of taste
Severe adverse effects
No side effects
1 of 100 patients
will experience liver, kidney, or heart damage which may require hospitalization
Efficacy
35 of 100 patients
will have no evidence of fungus with lab test 1 year from initiating treatment, if medication is used as prescribed
15 of 100 patients
will have no evidence of fungus with lab test 1 year from initiating treatment, if medication is used as prescribed
Interaction with alcohol consumption
No interactions with alcohol consumption
Your doctor will strongly advise against alcohol consumption while you are on treatment to reduce the risk of complications such as liver damage
Interaction with other medications
No risk of the toenail fungus medication interacting with your other medications
Rx
STOP
Because there is a risk of the toenail fungus medication interacting with your other medications, your doctor may change or temporarily stop your other prescriptions for your other health conditions

** Descriptions represent Canada-approved products and recommended use: 2 pills = itraconazole, 1 pill = terbinafine, topical applicator = efinaconazole, lacquer = ciclopirox*

- Three hundred and nine participants completed the DCE; 160 had comorbid diabetes. The diabetes group included a slightly higher proportion of males and was slightly older (**Table 1**).
- More patients with diabetes were currently using prescription medication for their toenail fungus (either topical or oral medications) than those without diabetes.
- Severe toenail fungus was more common among those with diabetes (70%) vs. those without (51%).
- Differences in treatment efficacy; treatment administration, frequency and duration; and risk of severe AEs, medication interactions, and interaction with alcohol had statistically significant impacts on patient preferences (**Figure 2**).
- Everything else being equal:
 - A 1% increase in efficacy increased the odds of the treatment being preferred by 4% (OR = 1.04 [95% CI: 1.02, 1.05]).
 - A treatment with 1% risk of severe AEs was 15% less likely to be preferred than one without risk of severe AEs (OR = 0.85 [0.80, 0.90]).
- Preferences estimated among those with diabetes were broadly similar to those from the overall sample.
- Those with diabetes had stronger preferences regarding treatment efficacy, the risk of severe AEs, and interactions with other medications and alcohol, while those without diabetes had stronger preferences regarding the route and frequency of treatment administration.

DISCUSSION

- Results from this DCE provide novel data on patient preferences regarding onychomycosis treatments, and how these differ by diabetes status. Similar data from patients with toenail fungus were previously unavailable.
- Another strength is the study's large sample size, overall and by diabetes status.
- Limitations to this study include self, rather than physician-confirmed onychomycosis and diabetes diagnosis and the recruitment of a convenience sample; however, effort was made to recruit patients from across Canada.
- Utilities were elicited alongside the DCE (results presented at ISPOR Europe 2022 [PCR28])

Table 1. Clinical and demographic characteristics of discrete choice experiment respondents

	Overall	Diabetes status No	Diabetes status Yes
	N = 309	N = 149 (48%)	N = 160 (52%)
Demographic characteristics			
Sex, male	174 (56.3)	77 (51.7)	97 (60.6)
Age			
Mean, SD	57.7 (14.6)	56.1 (16.5)	59.3 (12.4)
Median, IQR	59.0 (50.0, 67.0)	59.0 (45.0, 67.0)	60.0 (52.0, 67.0)
Clinical characteristics			
Currently taking medication for diabetes	151 (48.9)	0 (0.0)	151 (94.4)
Currently using toenail fungus prescription medication	112 (36.2)	42 (28.2)	70 (43.8)
Oral prescription	26 (8.4)	1 (0.7)	25 (15.6)
Topical prescription	101 (32.7)	41 (27.5)	60 (37.5)
Time since diagnosis (in years) - Mean, SD	11.0 (12.3)	12.6 (13.8)	9.6 (10.7)
Number of toenails affected - Mean, SD	3.6 (2.6)	3.4 (2.4)	3.8 (2.7)
Big toenail most affected	261 (84.5)	116 (77.9)	145 (90.6)
Severity based on percent area affected of most affected toenail			
Mild onychomycosis (0 to 20%)	7 (2.3)	5 (3.4)	2 (1.2)
Moderate onychomycosis (21% to 60%)	114 (36.9)	68 (45.6)	46 (28.7)
Severe onychomycosis (61%+)	188 (60.8)	76 (51.0)	112 (70.0)

IQR = interquartile range; SD = standard deviation.

Figure 2. Odds ratios and 95% CIs estimated from the results of the DCE

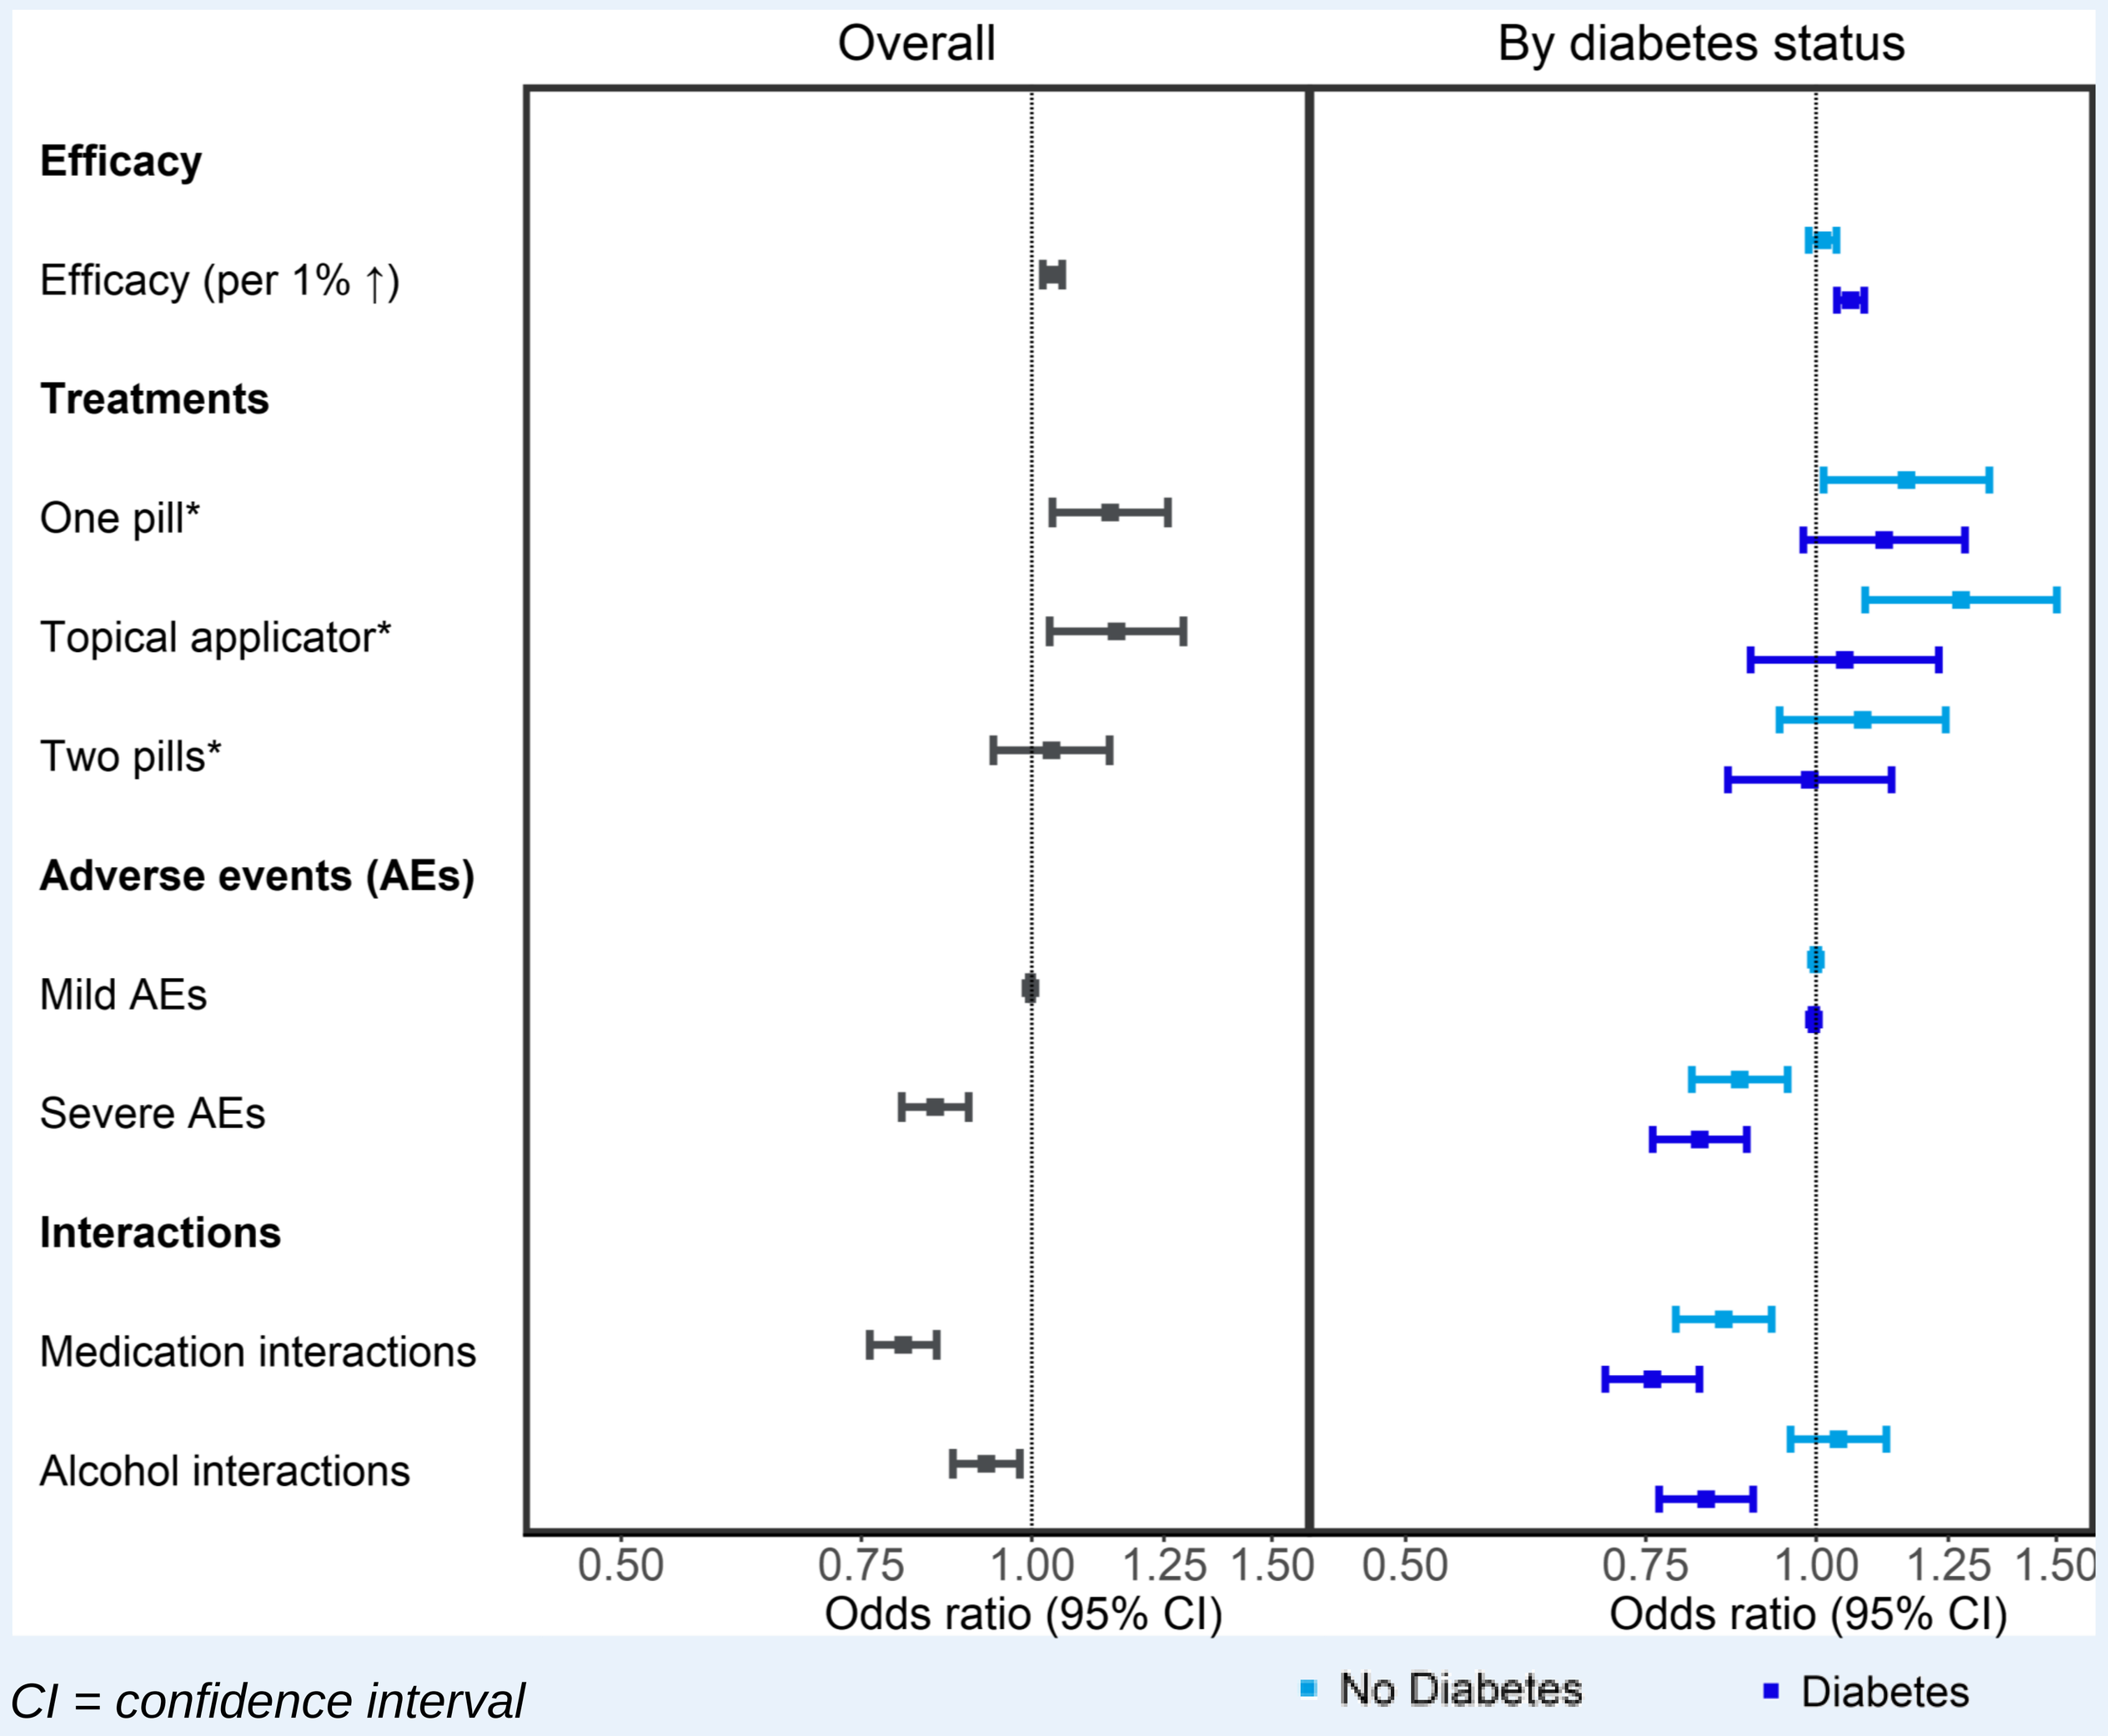
Overall
By diabetes status
Efficacy
Efficacy (per 1% ↑)
Treatments
One pill*
Topical applicator*
Two pills*
Adverse events (AEs)
Mild AEs
Severe AEs
Interactions
Medication interactions
Alcohol interactions
0.50
0.75
1.00
1.25
1.50
Odds ratio (95% CI)
No Diabetes
Diabetes

CI = confidence interval
** reference: lacquer*

CONCLUSION

Among patients with toenail onychomycosis from Canada, the presence of diabetes was associated with differences in treatment-related preferences.

Those with diabetes were more likely to select a treatment with higher efficacy, no risk of severe AEs, medication or alcohol interactions; those without diabetes were more concerned with mode of administration.

REFERENCES

1. Gupta AK, et al. *J Cutan Med Surg.* 2017;21(6):525-539.
2. Gupta AK, et al. *J Eur Acad Dermatol Venereol.* 2016;30(9):1567-1572.
3. Christenson JK, et al. *J Fungi (Basel).* 2018;4(3):87.
4. Elewski BE, et al. *J Clin Aesthet Dermatol.* 2015;8(11):38-42.
5. Mayser P. et al. *Am J Clin Dermatol.* 2009;10(4):211-220
6. Hahn GJ Shapiro SS et al. *A Catalog and Computer Program for the Design and Analysis of Orthogonal Symmetric and Asymmetric Fractional Factorial Experiments.* 1966.

Sources of Support: The study was funded by Bausch Health, Canada Inc. and conducted by Broadstreet Health Economics and Outcomes Research.

BAUSCH Health

Presented at ISPOR Europe 2022 ● November 6-9, 2022