Hospitalisation and Healthcare Costs in Adults With Systemic Lupus Erythematosus in Sweden: A Real-World Observational Cohort Study

EE553

Thomas Stephens[1], Martina Frodlund[2], Dag Leonard[3], Fabian Söderdahl[4], Gunilla Telg[5], Andreas Jönsen[6]

1. AstraZeneca AS, Oslo, Norway **2.** Department of Biomedical and Clinical Sciences, Division of Inflammation and Infection/Rheumatology, Linköping University, Linköping, Sweden
3. Department of Medical Sciences, Section of Rheumatology, Uppsala University, Uppsala, Sweden **4.** Statisticon AB, Uppsala, Sweden **5.** AstraZeneca AB, Stockholm, Sweden
6. Department of Clinical Sciences, Section of Rheumatology, Lund University, Skåne University Hospital, Lund, Sweden

BACKGROUND

- Systemic lupus erythematosus (SLE) is a chronic multi-organ autoimmune disease which causes significant morbidity and mortality.[1,2]
- SLE is characterised by periods of increased disease activity (known as flares) and remission.
- SLE is associated with high healthcare resource utilisation and costs, with inpatient care documented as the highest driver of medical care costs, as well as outpatient visits.
- There is limited contemporary evidence of the direct costs of treating SLE and disease flares in Sweden.

OBJECTIVES

- To describe resource utilisation in secondary care in adults with SLE in Sweden and estimate the excess healthcare costs of SLE.
- To estimate the costs associated with SLE disease flares over time and by severity in Sweden.

METHODS

STUDY DESIGN

- This was a real-world, observational cohort study using Swedish national registers.
- Adult patients with ≥2 hospital visits (inpatient or outpatient) with a primary diagnosis of SLE between July 2005 to December 2019 were identified in the Swedish National Patient Registry – a sensitive and specific method for identifying SLE cases from Swedish registers.[3]
- Patients were followed from the first recorded hospital visit for SLE (index date) until death, loss to follow-up, or 31 December 2020.
- A subset of patients were identified with presumed incident SLE, defined as having no recorded inpatient or outpatient visits with a primary or secondary diagnosis code of SLE between January 2001 and their index date.
- Population controls (without current or prior SLE) were matched 5:1 from the National Patient Registry to each SLE case on their index date based on age, sex, and county of residence.

MEASURES

- The annual number of outpatient visits, inpatient stays, and days spent as an inpatient per year were derived for SLE patients and controls from the Swedish National Patient Registry.
- Average annual total costs of secondary care and prescription medications (obtained from the Prescribed Drug Register) were estimated.
 - Data on hospital-administered medications, primary care visits, and indirect costs were not available.
 - Healthcare costs were derived using in 2021 DRG weights and reported in Swedish kronor (1 SEK = 0.0985 EUR = 0.1165 USD). Medication costs were included at sales price at the time of use and not inflated as product prices were relatively stable over the follow-up period.
- Flares were identified using an algorithm integrating medications, including increases in steroid dose, and healthcare for specified diagnoses, and were classified as mild, moderate, or severe.[4] Flare rates were calculated as the average number of flares per SLE patient per year.
- Costs per flare by severity were estimated based on hospital visits and prescription medications dispensed within 45 days of the start of the flare, assumed to be the average duration of a typical SLE flare.

STATISTICAL ANALYSES

- Descriptive analysis of the identified SLE populations was performed.
- The estimated annual costs of SLE flares were assessed over time since diagnosis for incident SLE patients by multiplying the average cost per flare and the number of flares per patient.
- Differences in healthcare utilisation and costs between SLE patients and controls were assessed using Mann-Whitney U tests.

Figure 1. Estimated average costs per patient per year for disease flares in the years following SLE diagnosis.

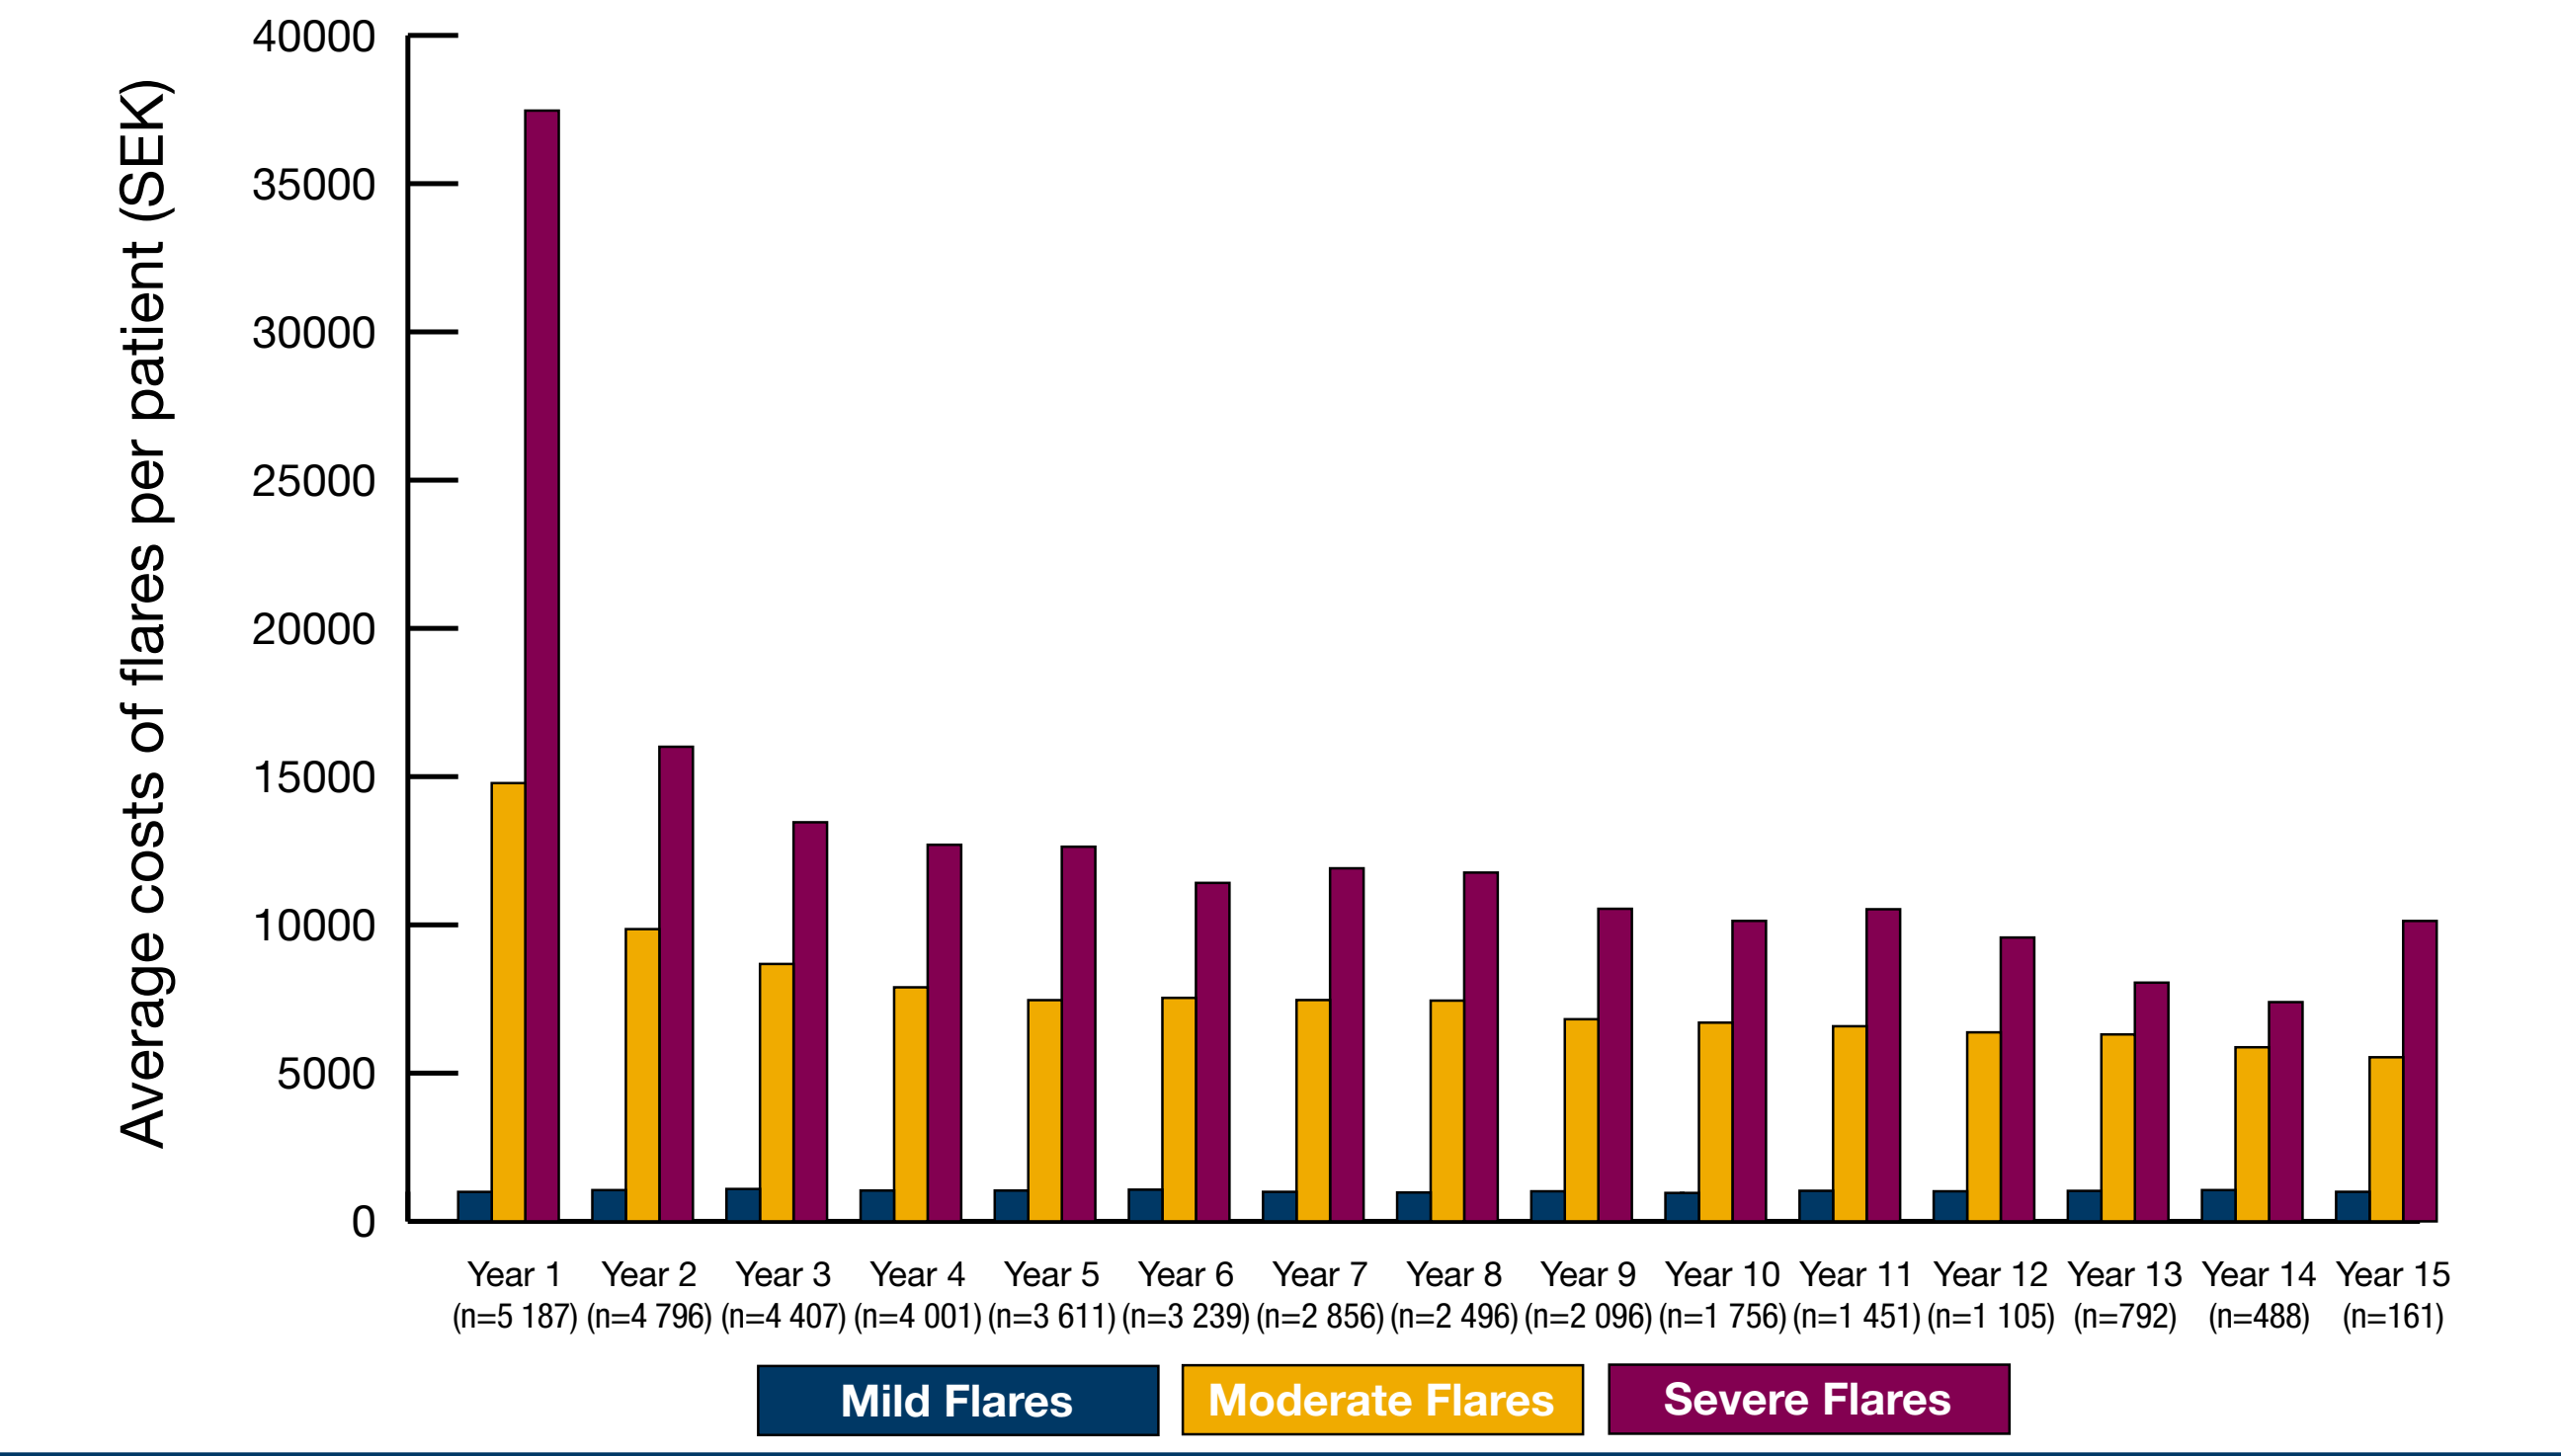
Average costs of flares per patient (SEK)
40000
35000
30000
25000
20000
15000
10000
5000
0
Year 1 (n=5 187)
Year 2 (n=4 796)
Year 3 (n=4 407)
Year 4 (n=4 001)
Year 5 (n=3 611)
Year 6 (n=3 239)
Year 7 (n=2 856)
Year 8 (n=2 496)
Year 9 (n=2 096)
Year 10 (n=1 756)
Year 11 (n=1 451)
Year 12 (n=1 105)
Year 13 (n=792)
Year 14 (n=488)
Year 15 (n=161)
Mild Flares
Moderate Flares
Severe Flares

RESULTS

- A total of 10 186 patients with SLE were identified in the national registers, of which 5 309 are assumed to have been diagnosed after July 2005 and have incident SLE. Patient characteristics at first visit (presumed diagnosis for incident cases) are shown in **Table 1**.
- SLE patients had significantly more outpatient visits, inpatient stays, and more inpatient days per year compared to controls **(Table 2)**.
- This led to significantly higher average annual healthcare costs for SLE patients **(Table 2)**. Mean costs were heavily skewed, but median annual healthcare costs were still substantially higher for SLE patients.
- Across the whole follow-up of the study, patients had an average of 1.7 flares per year, of which the majority were of moderate severity **(Table 3)**.
- The average cost per flare ranged from 3 029 SEK to 49 434 SEK, depending on severity **(Table 3)**.
- Based on the number of observed flares for incident patients, the estimated total costs of flares in the year after diagnosis was 53 240 SEK, of which 70% was attributable to severe flares **(Figure 1)**. By 10 years after diagnosis this decreased to 17 796 SEK due to patients having fewer moderate and severe flares, though the cost of mild flares remained similar over time.

Table 1. Characteristics of the SLE cohort

	All SLE Patients (N = 10 186)	Incident Cases (N = 5 309)
Age at baseline (years), mean (SD)	50.2 (17.2)	49.9 (18.2)
Female, n (%)	8 712 (85.5)	4 491 (84.6)
Follow-up (years), mean (SD)	11.3 (6.2)	7.7 (4.3)

Table 2. Difference between annual secondary care visits, inpatient days, and costs of healthcare for SLE patients and controls

	Controls (N = 48 985)	SLE Patients (N = 9 672)
Outpatient visits per year, mean (SD)	0.26 (1.83)	2.75 (6.60)
Median (IQR)	0 (0-0)	1 (0-3)
Inpatient stays per year, mean (SD)	0.08 (0.39)	0.43 (1.18)
Median (IQR)	0 (0-0)	0 (0-0)
Inpatient days per year, mean (SD)	0.49 (3.95)	2.86 (12.69)
Median (IQR)	0 (0-0)	0 (0-0)
Total costs of healthcare (SEK), mean (SD)	7 898 (40 304)	43 758 (105 002)
Median (range)	50 (0-2 849 215)	9478 (0-3 714 632)

All differences between SLE patients and controls were statistically significant ($p < 0.0001$). IQR, interquartile range

Table 3. Average cost of SLE flares by severity

Flare severity	Average flares per patient per year	Average cost per flare (SEK)
Mild	0.33	3 029
Moderate	1.10	7 683
Severe	0.27	49 434
Total	1.70	14 389

CONCLUSIONS

- **This nationwide study showed that patients with SLE in Sweden use 5-10 times more hospital care than controls.**
- **The direct costs of healthcare for patients with SLE in Sweden are substantially higher than for patients without SLE.**
- **Managing disease flares is a key driver of healthcare costs in SLE, representing a large proportion of annual per patient expenditure.**
- **The cost burden associated with disease flares is still considerable several years after diagnosis, at a time when patients' disease activity typically is better controlled.**
- **These results do not consider the indirect costs of SLE, which can be substantial in patients with active disease.[5]**
- **Effective treatments reducing flare rates could reduce the economic burden of SLE.**

REFERENCES

1. Justiz Vaillant A, Goyal A, Bansal P, Varacallo M. Systemic Lupus Erytheatosus. Treasure Island, FL: StatPearls Publishing; 2020.
2. Yurkovich M, Vostretsova K, Chen W, Avina-Zubieta JA. Overall and cause-specific mortality in patients with systemic lupus erythematosus: a meta-analysis of observational studies. Arthritis Care Res (Hoboken) 2014; 66(4): 608-16.
3. Arkema EV, Jonsen A, Ronnblom L, Svenungsson E, Sjowall C, Simard JF. Case definitions in Swedish register data to identify systemic lupus erythematosus. BMJ Open 2016; 6(1): e007769.
4. Garris C, Jhingran P, Bass D, Engel-Nitz NM, Riedel A, Dennis G. Healthcare utilization and cost of systemic lupus erythematosus in a US managed care health plan. J Med Econ 2013; 16(5): 667-77.
5. Jonsen A, Hjalte F, Willim M, et al. Direct and indirect costs for systemic lupus erythematosus in Sweden. A nationwide health economic study based on five defined cohorts. Semin Arthritis Rheum 2016; 45(6): 684-90.

The study was sponsored by AstraZeneca.
Presented at ISPOR Europe 2022, November 6-9, Vienna, Austria