Indirect Costs of Multiple Myeloma in Portugal

Miguel, S.[1]; Bernardo, P.[2]; Boaventura, C.[3]; Carrilho, M.[4]; Constanço, C.[5]; Damião, C.[6]; Leal da Costa, F.[7]; Martins, H.[8]; Martins, J.[9]; Parreira, S.[10]; Pedrosa H.[4]; Ponte, S.[11]; Santos, S[1]

[1]Janssen-Cilag Farmacêutica, Oeiras, Portugal; [2]Hematology Department, Hospital da Luz, Portugal; [3]Nursing Department, Centro Hospitalar Universitário de Coimbra, Portugal; [4]IQVIA Solutions, Oeiras, Portugal; [5]Hematology Department, Centro Hospitalar Tondela-Viseu, Portugal; [6]Nursing Department, Centro Hospitalar Universitário Lisboa Central, Portugal; [7]Hematology Department, Instituto Português de Oncologia de Lisboa - IPO Lisboa, Portugal; [8]Hematology Department, Centro Hospitalar Universitário de Lisboa Norte, Portugal; [9]Hematology Department, Unidade Local de Saúde de Matosinhos, Portugal; [10]Oncology Nursing Department, CUF Tejo, Portugal; [11]Nursing Department, Centro Hospitalar de Lisboa Ocidental, Portugal

ISPOR Europe 2022
6th - 9th November 2022
Vienna, Austria and Virtual

Poster Code: EE287

Background and Objectives

BACKGROUND

- Multiple Myeloma is a hematological cancer that develops in plasma cells in bone marrow. In Europe, it is the third most common malignancy of all hematological cancers. Global Cancer Observatory 2020 estimates 3,2/100.000 incident cases and 1,9 deaths/100.000 per year for MM in Portugal[1].
- Despite recent improvements in treatment options, which have increased overall survival, MM remains incurable. The burden associated with its management is considerable, both for patients and their caregivers.
- Economic data at country level with respect to this disease is limited and mainly focused on direct healthcare costs[2]. Indirect costs are poorly quantified and are not considered in economic evaluation of medicines and seem to have limited impact on physicians' judgment.

OBJECTIVES

- The present work aims to assess and quantify the indirect costs on Multiple Myeloma for both patients and caregivers, in Portugal.

Methods

- Targeted literature review was conducted to determine the relevant dimensions to consider in indirect costs, epidemiological data and country-specific indicators.
- Data was collected from national public data sources and complemented with semi-structured interviews to 2 MM patients and 1 caregiver, from a Portuguese Patient Association (*Associação Portuguesa Contra a Leucemia - APCL*). Information collected from literature review and interviews was discussed and validated with two expert panels of 5 Portuguese Hematologists and 4 specialized Nurses.
- The analysis considered the prevalence of the disease based on GLOBOCAN 2020 data for Portugal which includes all patients diagnosed with MM in Portugal.
- Key data sources were computed in an Excel model. Costs were valued according to official public data sources considering the most recent data available.
- MM indirect costs in Portugal were evaluated from third-party payer perspective (patient/caregivers and National Social Security), which includes absenteeism (patient and caregiver), presenteeism and early retirement. Presenteeism was not considered in the final model as it represented a residual impact. Additionally, the cost of premature death was assessed to understand the impact from a societal perspective.

Indirect Costs in Multiple Myeloma, Portugal

	Patient Absenteeism	Caregiver Absenteeism	Early Retirement	Premature Death
Calculation method	Number of patients on active age **x** national daily wage **x** number of days lost due to MM (outpatient visits + sick leave)	Number of caregivers **x** number of caregivers employed **x** number of days lost as a caregiver **x** national daily wage	Number of patients on active age that do not return to work **x** average years of productivity loss due to MM **x** annual disability living allowance	Number of patients on active age **x** number of contributing years lost due to MM **x** average national income per capita/year

Results

	Patient Absenteeism	Caregiver Absenteeism	Early Retirement	Premature Death
	Outpatient visits: • 4 hospital visits/month (including treatments, appointments and exams) • 30 working days lost/year; ***Sick Leave***: • 10% of patients take 1 year of sick leave and return to work • 90% take 3 years of sick leave (maximum time allowed by law) and do not return to work	• Only informal caregivers were considered since in Portuguese reality these are the great majority • 70% of patients have a caregiver, and most are informal (e.g., spouses and descendants) • 52% of caregivers are still employed • Legally, caregivers are allowed a maximum of 15 absent days per year[3] • € 75,31 is the average daily wage in Portugal (2020)[4]	• 97% of active patients retire earlier after being on sick leave • 4,3 average years of productivity loss estimated until death • € 5.020/year is the disability living allowance in Portugal (2020)[5]	• Considered only patients on active age • 66,7 years old as the average retirement age[6] • € 19.126 average national income per capita/year (2020)[4]
Cost per patient/year	***€ 1.591***	***€ 415***	***€ 5.036***	***€ 22.630***
Total cost/year	***€ 3.599.700***	***€ 938.739***	***€ 11.391.018***	***€ 51.188.633***
	23%	**6%**	**71%**	
	€ 15.929.457 (18%)			

Direct Costs[2]
(Diagnosis, HCP appointments, surgery, ER visits, drugs and hospital transportation)
€ 71.137.638 (82%)

Total cost in MM (direct[2] + indirect costs) ***€ 87.067.095***

Multiple Myeloma Epidemiology, Portugal

2.262[1]
MM Prevalence in 2020

67,7
average age at diagnosis[2]

7,3
Overall Survival[2]

75,0
average age at death[2]

Figure 1. Proportion of MM patients in active age (less than 66,7 years old)[2]

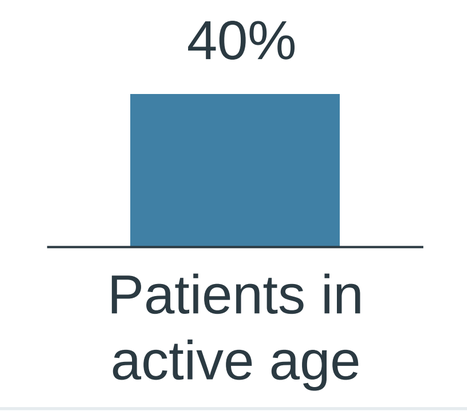
40%
Patients in active age

Discussion

- On a third-party payers' perspective, the total annual indirect costs associated with MM patients in Portugal, in 2020/2021, were estimated to be € 15.929.457, with an average of € 7.042 per patient.
- Based on a study from 2019, in which direct costs associated with MM amounted to a total of € 71.137.638[2], it can be estimated a total annual cost of € 87.067.095, an average of € 38.491 per patient, corresponding to 82% and 18% in direct and indirect costs, respectively. If the model had considered premature death, the total annual indirect costs would increase to € 67.118.090.
- Of the indirect costs' dimensions considered, early retirement had the most relevant weight. This was expected, since the large majority of patients on active age retire earlier due to the disease burden. Caregivers' work absence had the lowest cost associated since these patients are normally accompanied by informal caregivers whose number of absent days is limited by law.
- Overall, results are aligned with other MM cost of illness studies, as for example in the study conducted by Petrucci's et al. (2013) in MM patient population in Italy, showing a similar proportion between direct and indirect costs (84%/16%[7]). Petrucci's[7] study does not consider early retirement in his calculations. Thus, considering only absenteeism, the cost per patient of the present study is very similar to the comparator (€ 3.118).

Conclusion

- This study provides updated insights into the indirect costs involved in MM for Portuguese population revealing the impact to patients/caregivers and to the National Social Security. Indirect costs represent 18% of MM related costs being mainly associated to early retirement.
- These results will hopefully raise awareness to the disease burden especially concerning indirect costs impact, which are usually more difficult to quantify, due to their nature, and often under valued.
- With the increasing prevalence of MM in recent years and the significant impact of this disease, it is essential to further generate relevant evidence at national level, capable of supporting future healthcare decisions.

Limitations

- For calculation purposes, some assumptions have been made: 66,7 years old as the age of retirement[6], an overall survival of 7,3 years[2], regardless of the age group, and an average of 4,3 years of productivity loss estimated until death.
- Only informal caregivers were considered as they represent the majority in Portugal, for this specific disease.
- The model does not account for specific patients' subgroups (e.g., patients on dialysis), who end up being more frequently absent from work.

Funding and Disclosures

- The authors acknowledge the support of APCL.
- The study was funded by Janssen Portugal. IQVIA Solutions Portugal was contracted to develop and implement the project, including conduct interviews to patients and caregivers, moderate the Expert Panels and medical writing support.

References

[1] GLOBOCAN 2020 – Multiple Myeloma Portugal

[2] Neves M, Trigo F, Rui B. et al. Multiple Myeloma in Portugal: Burden of Disease and Cost of Illness. Pharmacoeconomics. 2021 May;39(5):579-587. doi: 10.1007/s40273-020-00993-5. Epub 2021 Jan 31. PMID: 33517511; PMCID: PMC8079298.

[3] Diário da República, Lei nº7/2009, Artigo 252º Falta para assistência a membro do agregado familiar.

[4] PORDATA 2020, Gross national income per capita

[5] PORDATA 2020, Pensão média anual da Segurança Social: total, de sobrevivência, de invalidez e de velhice

[6] Portaria n.º 30/2020, de 31 de janeiro 2021

[7] Petrucci, M. T., et al. Cost of illness in patients with multiple myeloma in Italy: the CoMiM study. Tumori. 2013 Jul-Aug;99(4):e193-202. doi: 10.1177/030089161309900434. PMID: 24326862.