Correlation and prognostic value of surrogate endpoints and overall survival in multiple myeloma: a systematic literature review

Presentation Number
112609

Emre Yucel,[1] Bill Malcolm,[1] Ibrahim Diakite,[2] Elisabeth F.P. Peterse,[3] Alexa Stewart,[4] Jelle A. Spoorendonk[3]

[1]Bristol Myers Squibb, Princeton, NJ, USA; [2]OPEN Health Group, Bethesda, MD, USA; [3]OPEN Health Group, Rotterdam, Netherlands; [4]OPEN Health Group, Oxford, UK

Introduction

- Multiple myeloma (MM) is the second most common hematologic malignancy worldwide with an estimated 176,000 new cases and 117,000 deaths in 2020[1]
- Improvements in overall survival (OS) have been observed with novel, potent therapeutic advances in MM
 - Longer OS outcomes are extending the time to realization of mature OS data and subsequent health technology assessment (HTA) evaluations
- There is a need to explore the feasibility of adopting surrogate endpoints for interim HTA evaluations while extended OS outcomes remain ongoing

Objective

- To conduct a systematic literature review (SLR) of studies evaluating the predictive value of surrogate endpoints for OS in MM

Methods

Study identification

- The SLR was carried out according to guidelines published by the Centre for Reviews and Dissemination[2] with searches in MEDLINE and EMBASE to identify full-text publications
- Screening of the title, abstract, and full text was conducted by 2 reviewers
 - Discrepancies were resolved by a third reviewer
- A hand search of reports from HTA organizations, including NICE, CADTH, G-BA, IQWiG, ICER, and TLV was performed to assess how surrogate endpoints were interpreted for OS in MM
- Study inclusion and exclusion criteria are shown in **Table 1**

Table 1. Study inclusion and exclusion criteria

PICOS concept	Inclusion criteria	Exclusion criteria
Population	• Patients with MM regardless of treatment line	• Healthy patients • In vitro studies • Patients with MM with comorbid conditions • When MM is not the focus of the research
Interventions/ comparators	• No specific interventions/comparators are of interest	• Not applicable
Outcomes	• OS prediction methodology concepts that discuss surrogate outcomes in the absence of mature OS • Correlation between or extrapolation/ prediction of surrogate endpoints of survival by statistical test (eg, Spearman correlation, Pearson correlation, bivariate analysis, multivariate analysis, regression) of which one parameter should always be OS • Surrogate endpoints for OS: any type of response, such as: CR, PR, VGPR, duration of CR, MRD, PFS, TTP, failure-free survival, EFS, DFS, TTNT, DOR, HRQoL, and clinical relapse/paraprotein relapse	• Studies investigating OS for specific subgroup cohorts (eg, specific biomarker subgroup has higher, or lower median OS compared to overall population) • Correlation between or extrapolation/prediction of survival not investigated in relation to OS • Prognostic factors (without the statistical scrutiny as defined under the inclusion criteria) • Reporting only outcomes not included in the inclusion criteria
Study design	• Clinical trials • Real-world evidence/observational studies/ registry data (with ≥ 100 enrolled patients) • Review and meta-analysis • Studies investigating independent patient-level data	• Phase 1 trials • Trials investigating survival outcomes without specifying the appropriate techniques as listed under outcomes
Publication type	• Reviews • Original research • HTA documentation	• Conference abstracts • Letters to the editor • Errata • Commentary • Expert opinion excluded except if informing HTA decision making • Case study
Language	• English, except for HTA documentation	• Other languages
Time horizon	• Full text: January 1, 2010 – April 21, 2021 • Reviews and HTA documentation: no time restriction	• Full text: before January 1, 2010 • Reviews and HTA documentation: no time restriction

CR, complete response; DFS, disease-free survival; DOR, duration of response; EFS, event-free survival; HRQoL, health-related quality of life; MRD, minimal residual disease; PFS, progression-free survival; PICOS, patient/population, intervention, comparison, outcome, study design; PR, partial response; TTNT, time to next treatment; TTP, time to progression; VGPR, very good partial response.

Study assessment

- Two criteria were used to evaluate if a study had established an endpoint as a surrogate for OS in MM:
 - The endpoint was shown to be correlated with OS
 - The full effect of treatment on OS was shown to be captured by the endpoint
- Selected studies were assessed to determine if they had established an association between the candidate surrogate endpoint and OS at:
 - The individual patient level, using a Pearson correlation coefficient or jointly modeling the endpoints regardless of treatment arm
 - The trial level, using the determination coefficient R^2 of a linear regression or a multivariate regression model weighted by study sample size

Results

Study characteristics

- A total of 2526 records were identified; 2514 were screened, of which 174 (6.9%) were assessed for full-text eligibility (**Figure 1**)
- Five studies (2.9%) analyzed the correlation and predictive relationship between surrogate endpoints and OS, referred to here as surrogate endpoint studies (**Table 2**)
- Forty-six studies evaluated OS in the context of certain endpoints (eg, treatment responses, MRD), referred to here as stratification studies
- Twenty-four appraisals from HTA bodies were identified discussing surrogate endpoints for OS in MM
 - Most considered PFS as a valid surrogate endpoint for OS
 - Less frequently, MRD-negative status was suggested as a surrogate endpoint for OS

Surrogate endpoint studies

- Of the 5 surrogate endpoint studies identified, 3 used aggregated SLR data and 2 used individual patient-level data (**Table 2**)

Figure 1. PRISMA flow chart of included studies

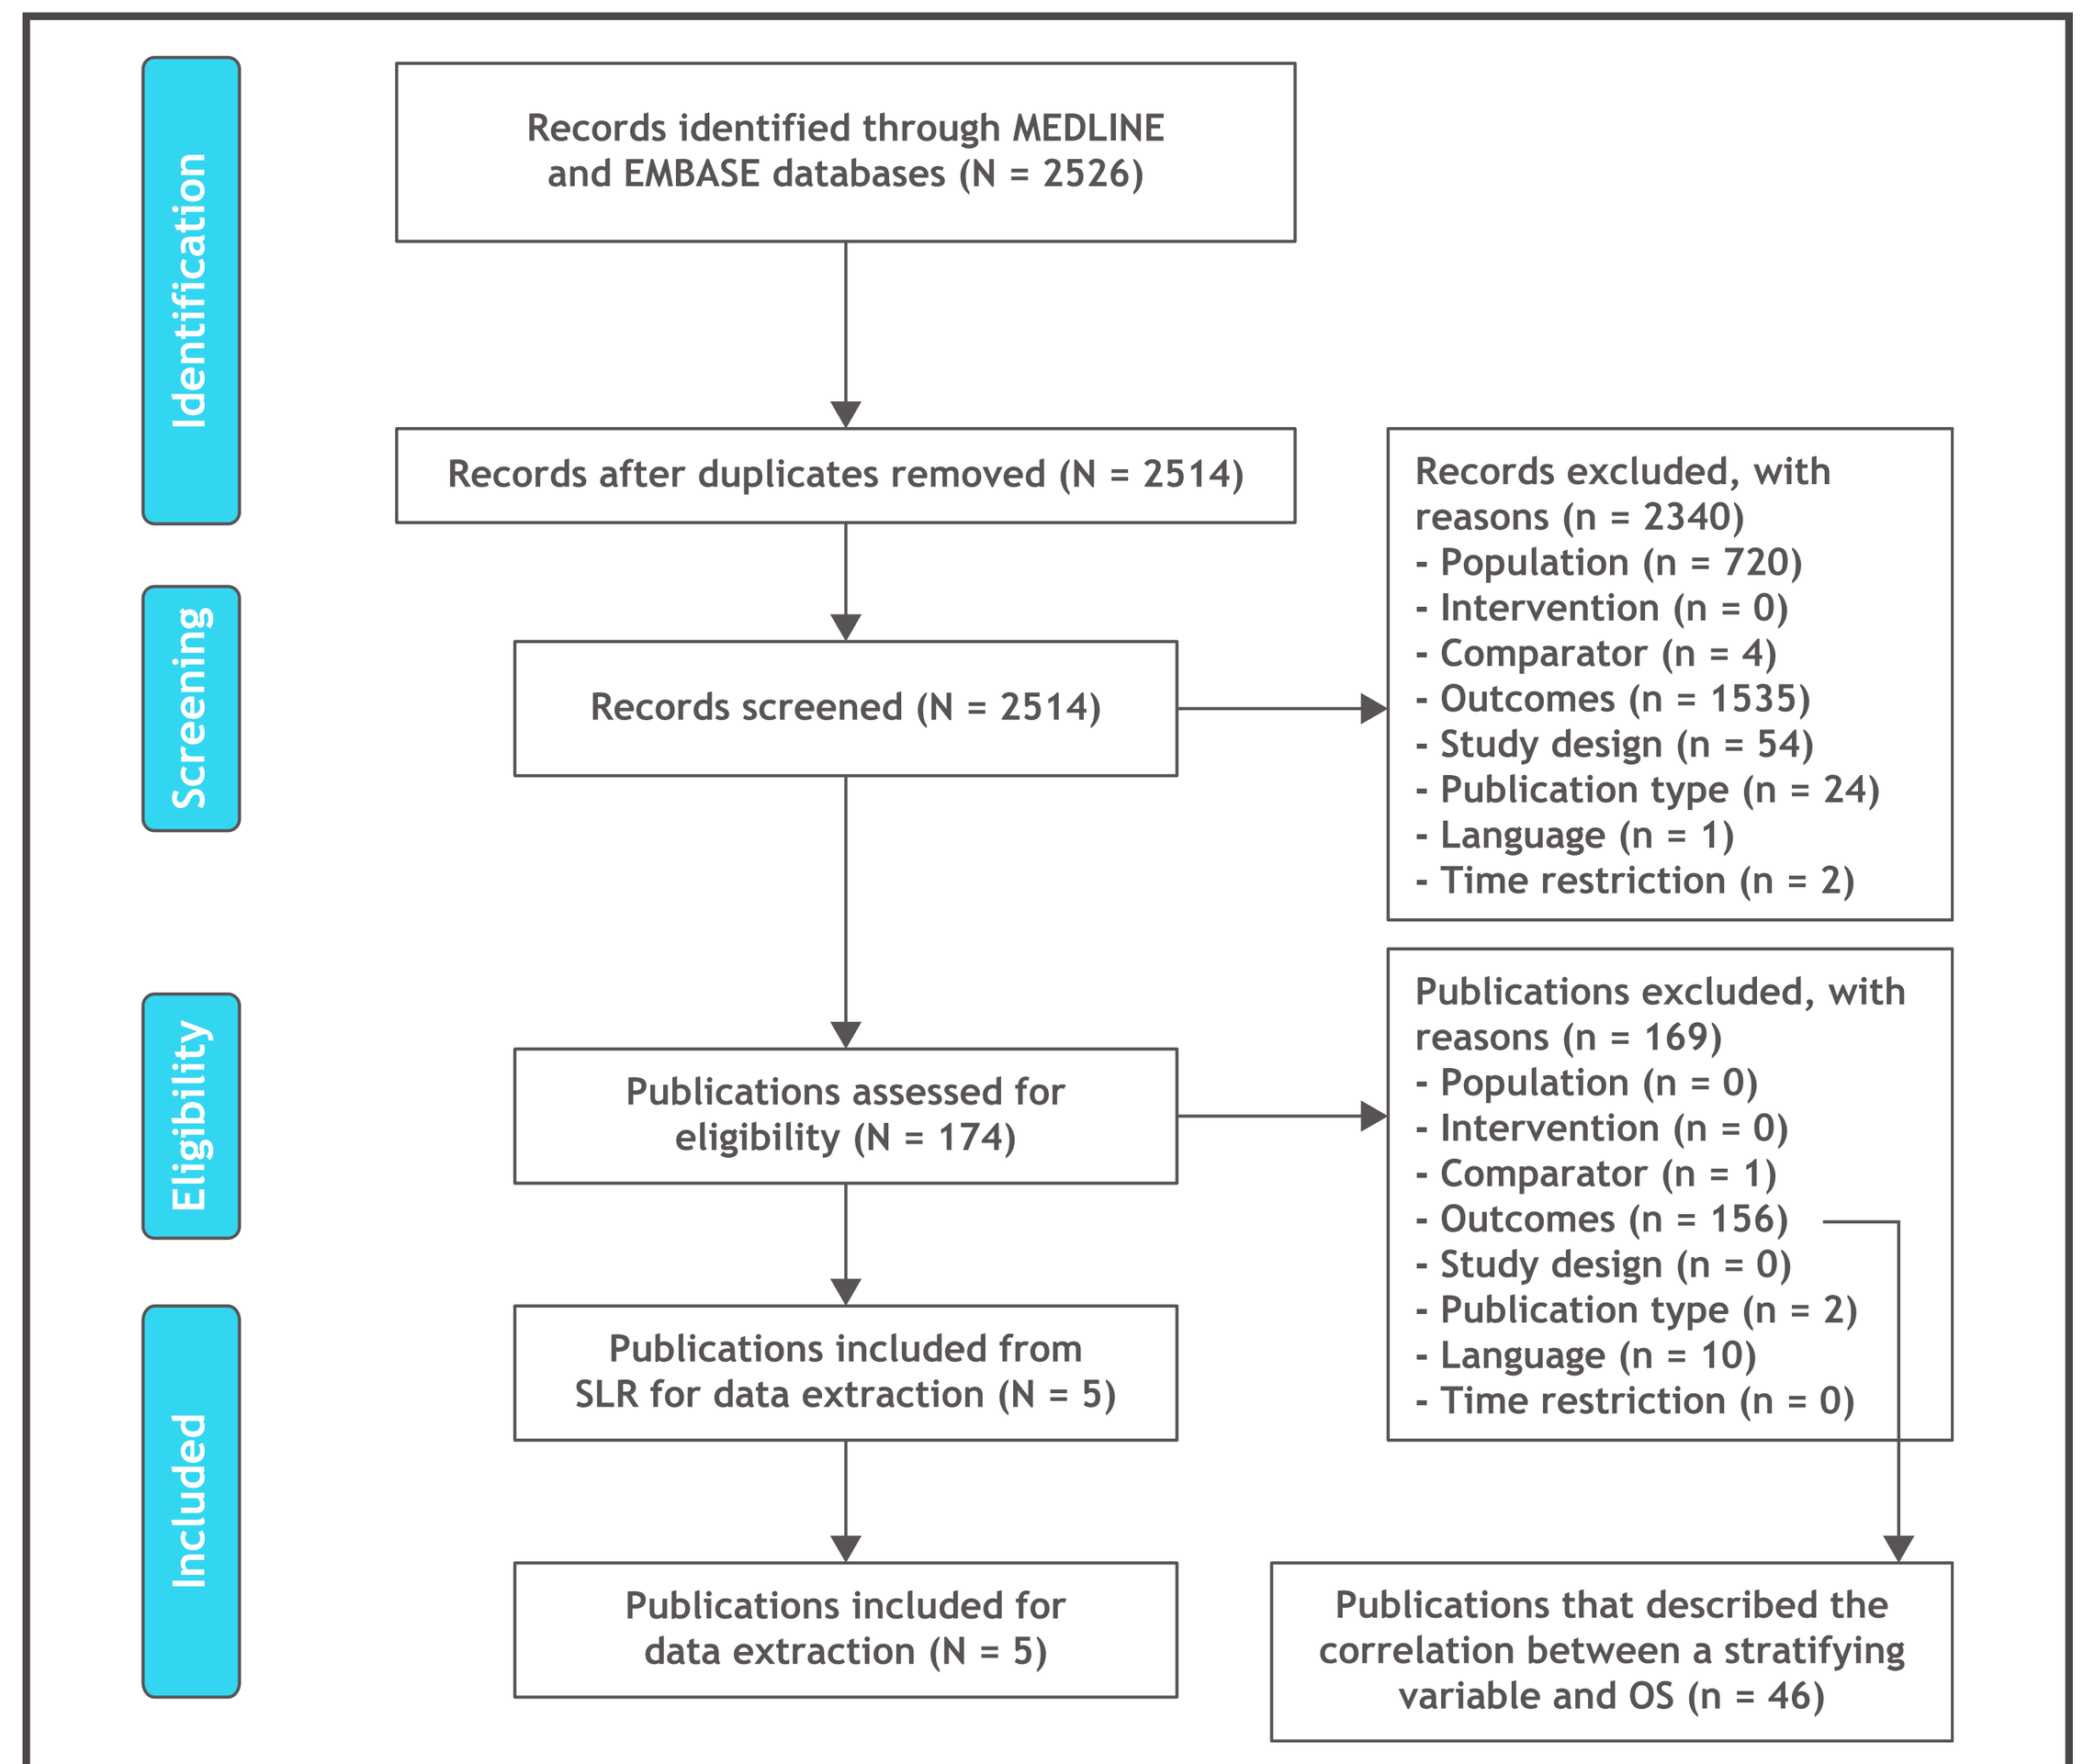

Identification
Screening
Eligibility
Included
Records identified through MEDLINE and EMBASE databases (N = 2526)
Records after duplicates removed (N = 2514)
Records screened (N = 2514)
Records excluded, with reasons (n = 2340)
- Population (n = 720)
- Intervention (n = 0)
- Comparator (n = 4)
- Outcomes (n = 1535)
- Study design (n = 54)
- Publication type (n = 24)
- Language (n = 1)
- Time restriction (n = 2)
Publications assessed for eligibility (N = 174)
Publications excluded, with reasons (n = 169)
- Population (n = 0)
- Intervention (n = 0)
- Comparator (n = 1)
- Outcomes (n = 156)
- Study design (n = 0)
- Publication type (n = 2)
- Language (n = 10)
- Time restriction (n = 0)
Publications included from SLR for data extraction (N = 5)
Publications included for data extraction (N = 5)
Publications that described the correlation between a stratifying variable and OS (n = 46)

PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses.

Table 2. Key findings from surrogate endpoint studies

Publication (author, year)	MM population	Studies/ patients (N)	Data type	Method to establish surrogacy	Surrogate endpoint used for OS	Parameter used for OS and surrogate relationship	Results	Authors' conclusion
Félix et al., 2013[3]	Newly diagnosed, R/R, and advanced patients	152/22,696	Aggregated	Spearman rank correlation, multivariate censored normal regression model	TDEs: TTP, PFS, EFS	Correlation between median OS and median TDE	*P* = 0.51; TTP *P* = 0.75; PFS *P* = 0.84; EFS Each additional month in the observed median TDE correlated to an increase of 2.45 months (95% CI, 1.71-3.20) in median OS	TTP, PFS, and EFS are valid surrogate endpoints of OS
Hari et al., 2018[4]	R/R	1/628	Individual patient	Logistic marginal structural model	DSLT	OR, representing the odds of 1-year mortality for each additional month of contin-ued therapy	OR = 0.78 (95% CI, 0.77-0.83) Each additional month of 2L therapy was associated with a reduced risk of death at 12 months	There is a strong correlation between DSLT and OS
Cartier et al., 2015[5]	Adults: newly diagnosed, maintenance, or R/R	21/12,048	Aggregated	Pearson correlation, R^2 from a linear regression	PFS	HRs for PFS and OS, log (HRs) for PFS and OS	Pearson r = 0.82 Pearson r (log transformed) = 0.80 There is a strong statistically significant (*P* < 0.0001) positive correlation between PFS and OS	PFS is a valid surrogate for OS
Mainou et al., 2017[6]	Newly diagnosed or previously treated with corticosteroids alone	72/24,094	Aggregated	Random effects meta-regression models	CR, VGPR	Regression coefficient between log-transformed relative risk of response (CR or VGPR) and the log-transformed HR of OS or PFS	Patients without ASCT (regression coefficient, 0.02; 95% CI, −0.06 to 0.10) Patients undergoing ASCT (−0.11; 95% CI, −0.44-0.22) Patients with ASCT vs non-ASCT patients (0.04; 95% CI, −0.29-0.38)	No association between response outcomes (CR and VGPR) and OS
Rawstron et al., 2015[7]	Newly diagnosed adults	1/397	Individual patient	Cox proportional model	MRD	Association between log-transformed MRD and OS	MRD ≥ 10%: 1 year MRD 1% to < 10%: 4 years MRD 0.1% to < 1%: 5.9 years MRD 0.01% to < 0.1%: 6.8 years MRD < 0.01%: 7.5 years It was concluded that MRD level predicted OS and PFS with approximately 1-year median OS benefit per log depletion	MRD level is a strong predictor for OS

CI, confidence interval; OR, odds ratio; R/R, relapsed/refractory.

- Correlation was established using Spearman rank or Pearson correlation tests, and predictive value was assessed using meta-regression models
- Key findings from each of the surrogate endpoint studies are summarized below:
 - Félix et al. 2013[3] established a positive monotonic relationship between median time-dependent endpoints (TDEs) and median OS, using Spearman rank correlation and a multivariate censored normal regression
 - Hari et al. 2018[4] reported a strong correlation of OS with duration of second-line (2L) therapy (DSLT), with a reduced risk of death for each additional month of 2L treatment
 - Cartier et al. 2015,[5] used the Pearson coefficient to establish a positive linear relationship between hazard ratios (HRs) for PFS and OS (and between the log-transformed HRs), demonstrating PFS as a valid surrogate of OS
 - Mainou et al. 2017[6] reported a meta-regression coefficient that indicated no association between treatment response outcomes (CR and VGPR) and OS for patients without autologous stem cell transplantation (ASCT), those undergoing ASCT, and those with ASCT
 - Rawstron et al. 2015[7] showed a significant benefit in median OS per log depletion in MRD percentage, although HRs from the multivariate Cox model where log (MRD) was shown to be a statistically significant predictor of OS were not reported

Stratification studies[8-53]

- To measure the association with OS, multiple endpoints were assessed across stratification studies: CR, VGPR, PR, progressive disease (PD), stable disease (SD), positive or negative MRD status
- Type of response was a significant factor prolonging survival, where increasingly better response indicated longer OS (**Figure 2**)
- MRD-negative (MRD-) status was a strong indicator for better survival outcomes compared with MRD-positive (MRD+) status (**Figure 3**)
- Using a Cox proportional hazards model, the effect on OS was stratified by different categories of treatment response or PD
 - The risk of dying was substantially higher for patients with ≤ PR versus patients with CR or VGPR (**Figure 4**)

Surrogacy in HTAs

- Many HTAs acknowledged that increased PFS may be associated with better OS[54-57], and that PFS can serve as a surrogate endpoint for OS, especially in Canada and in the US for regulatory approval by the FDA[58-65]
- MRD-status was associated with improved OS, but to a lesser extent than PFS[54,55,60,65]
- Several HTA bodies (NICE,[66] CADTH,[67] IQWiG[68]) have published multiple guidance documents that provide methods for validation of surrogate endpoints
- Although many of these assessments hint toward a positive association between PFS and OS, and less frequently MRD-status, any supportive statistical analysis or predictive models applied to trial results are lacking
 - Often these claims are made by referring to the earlier published meta-analyses by Félix et al. in 2013[3] and Dimopoulos et al. in 2017,[69] which state a positive association between MRD, treatment response, or PFS, versus OS, shown by stratification or based on clinical experts

- Analyses in other types of cancers show a positive association between PFS and OS, such as metastatic melanoma, metastatic colorectal cancer, and advanced breast cancer[3,4]

Figure 2. Association between OS and clinical response

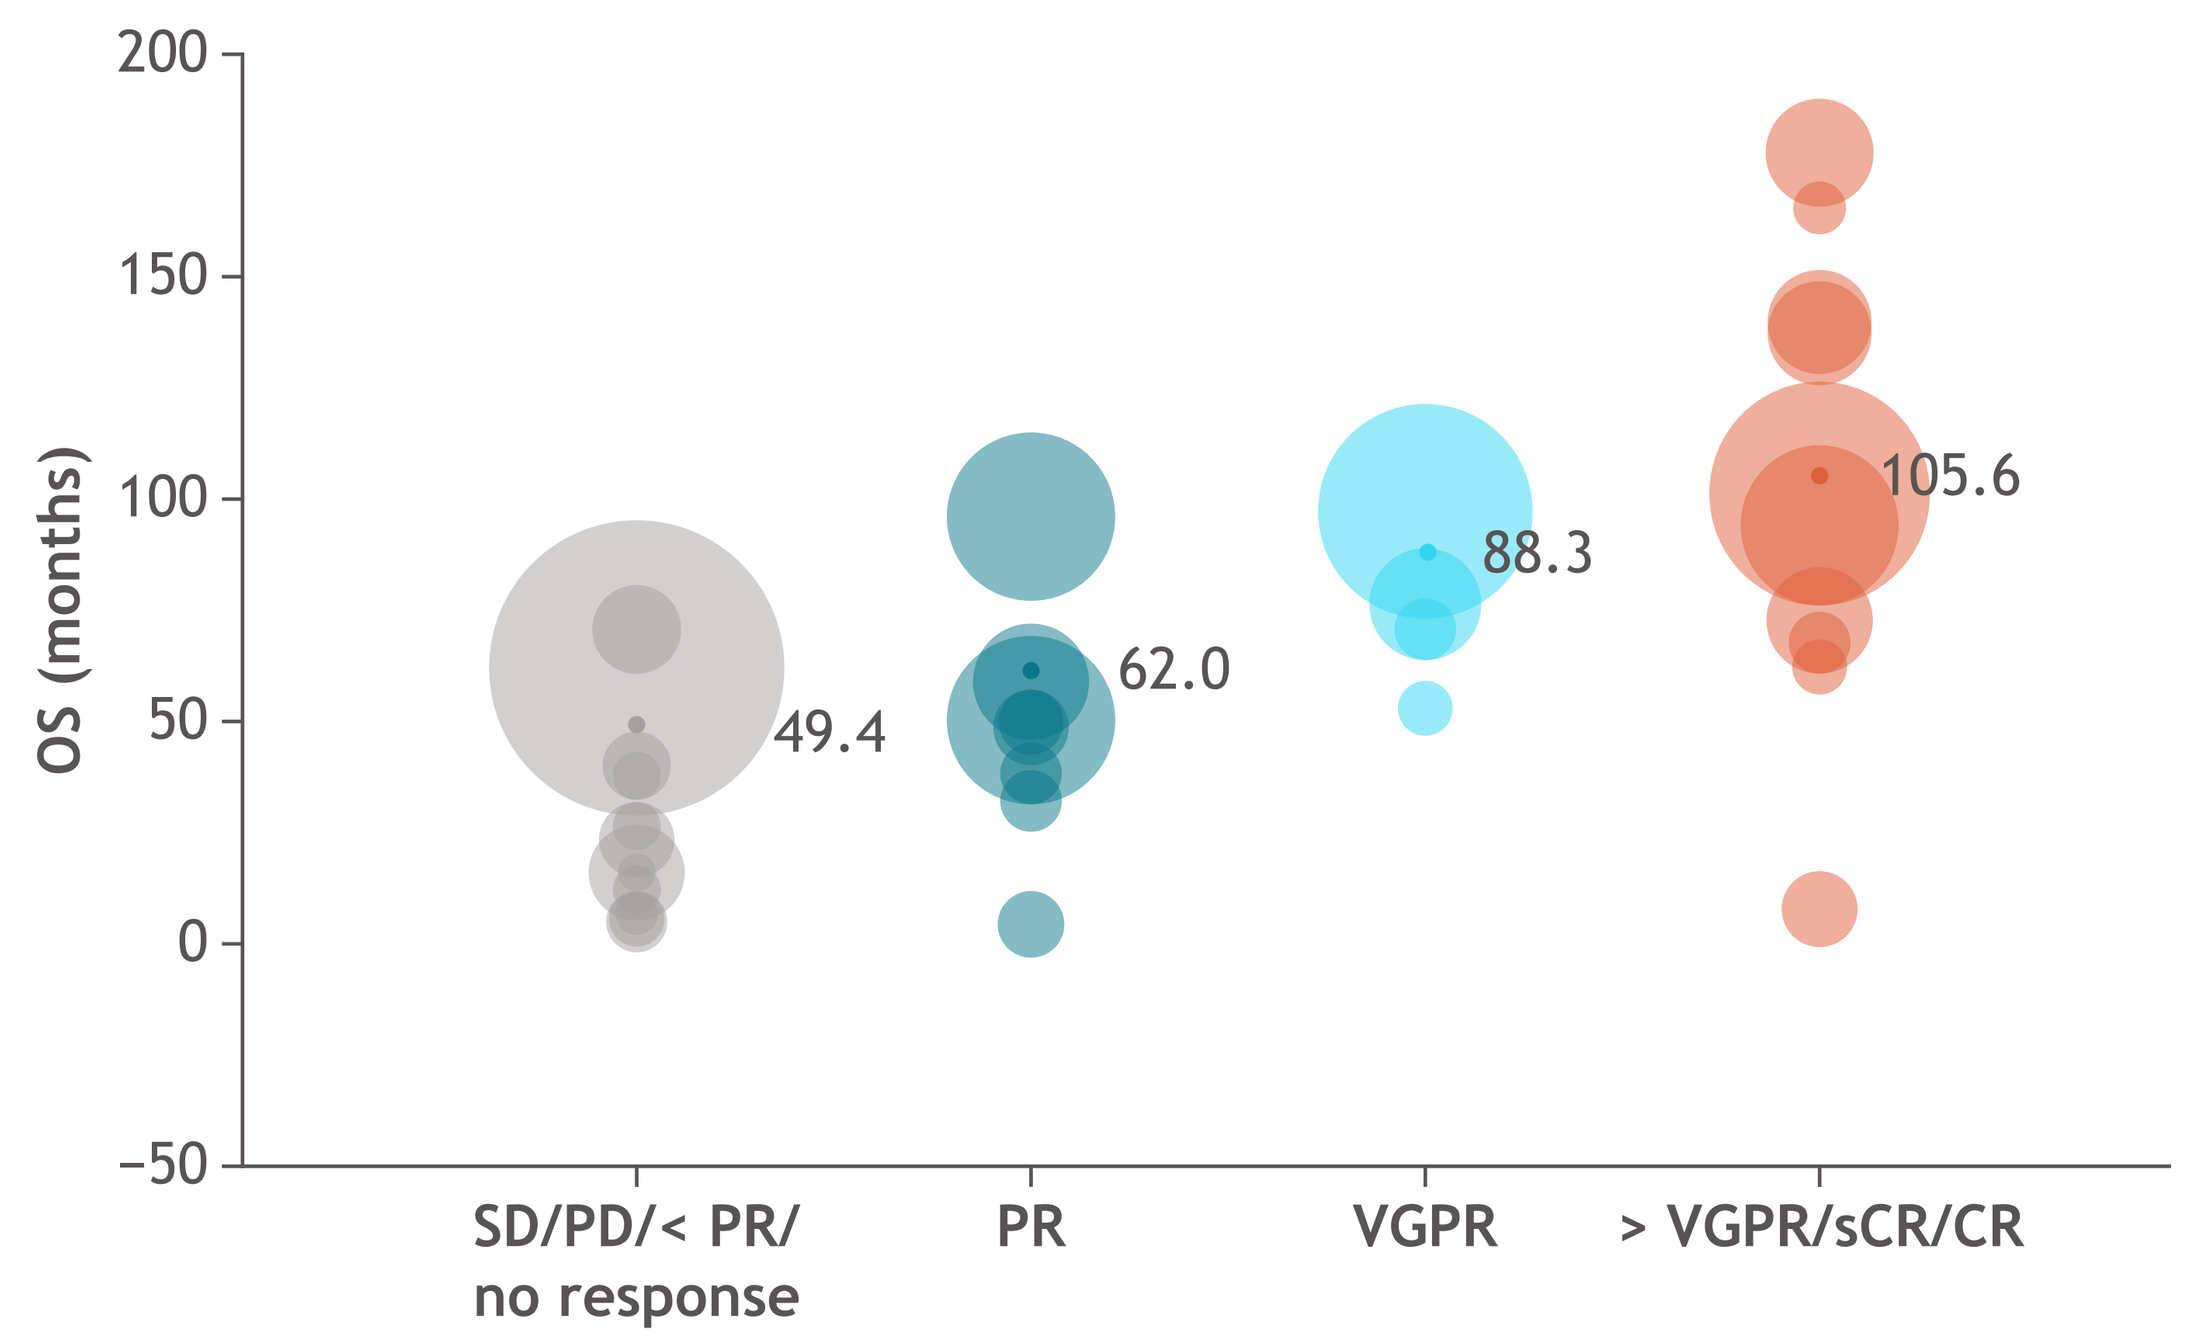

OS (months)
200
150
100
50
0
−50
49.4
62.0
88.3
105.6
SD/PD/< PR/ no response
PR
VGPR
> VGPR/sCR/CR

Dark colored circles indicate mean OS.
These results do not consider disease stage or treatment approach in the MM populations, but are weighted to account for the sample size, where a larger circle indicates a larger patient population.
Additionally, these results reflect treatment response data collected from 16 publications where multiple data points for a single publications may have been provided.[8,14,22,24,27,28,30,32-35,42,46,48,51,52]
sCR, stringent complete response.

Figure 3. Association between OS and MRD status

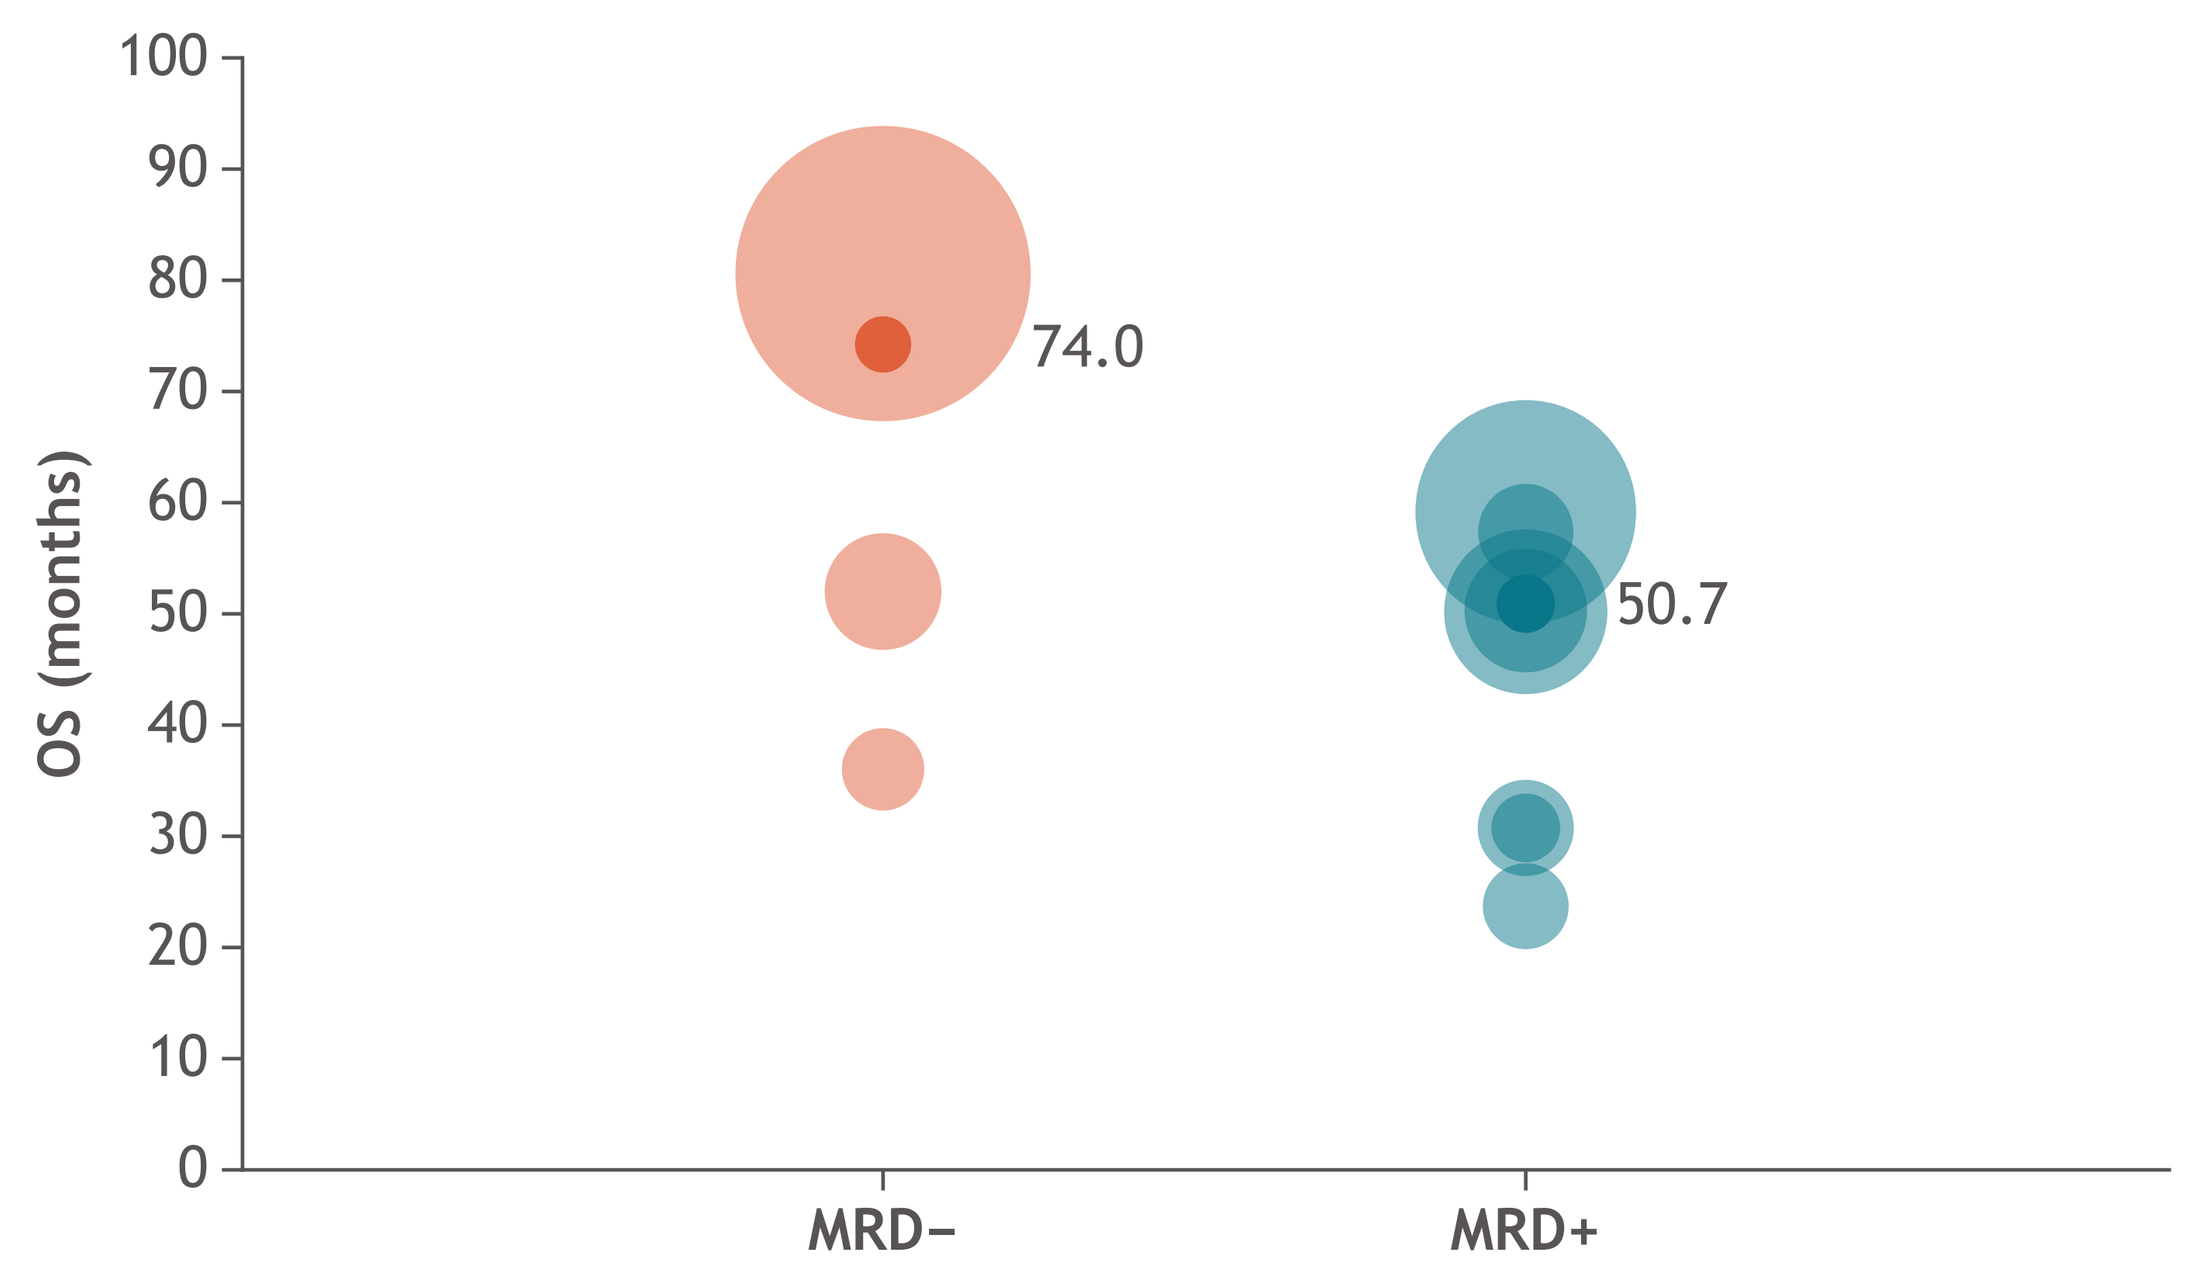

OS (months)
100
90
80
70
60
50
40
30
20
10
0
74.0
50.7
MRD−
MRD+

Dark colored circles indicate mean OS.
These results do not consider disease stage or treatment approach in the MM populations, but are weighted to account for the sample size, where a larger circle indicates a larger patient population.
Additionally, these results reflect MRD data collected from 3 publications where multiple data points for a single publications may have been provided.[12,25,44]

Figure 4. HRs for OS stratified by comparison with clinical response, MRD status, or PD

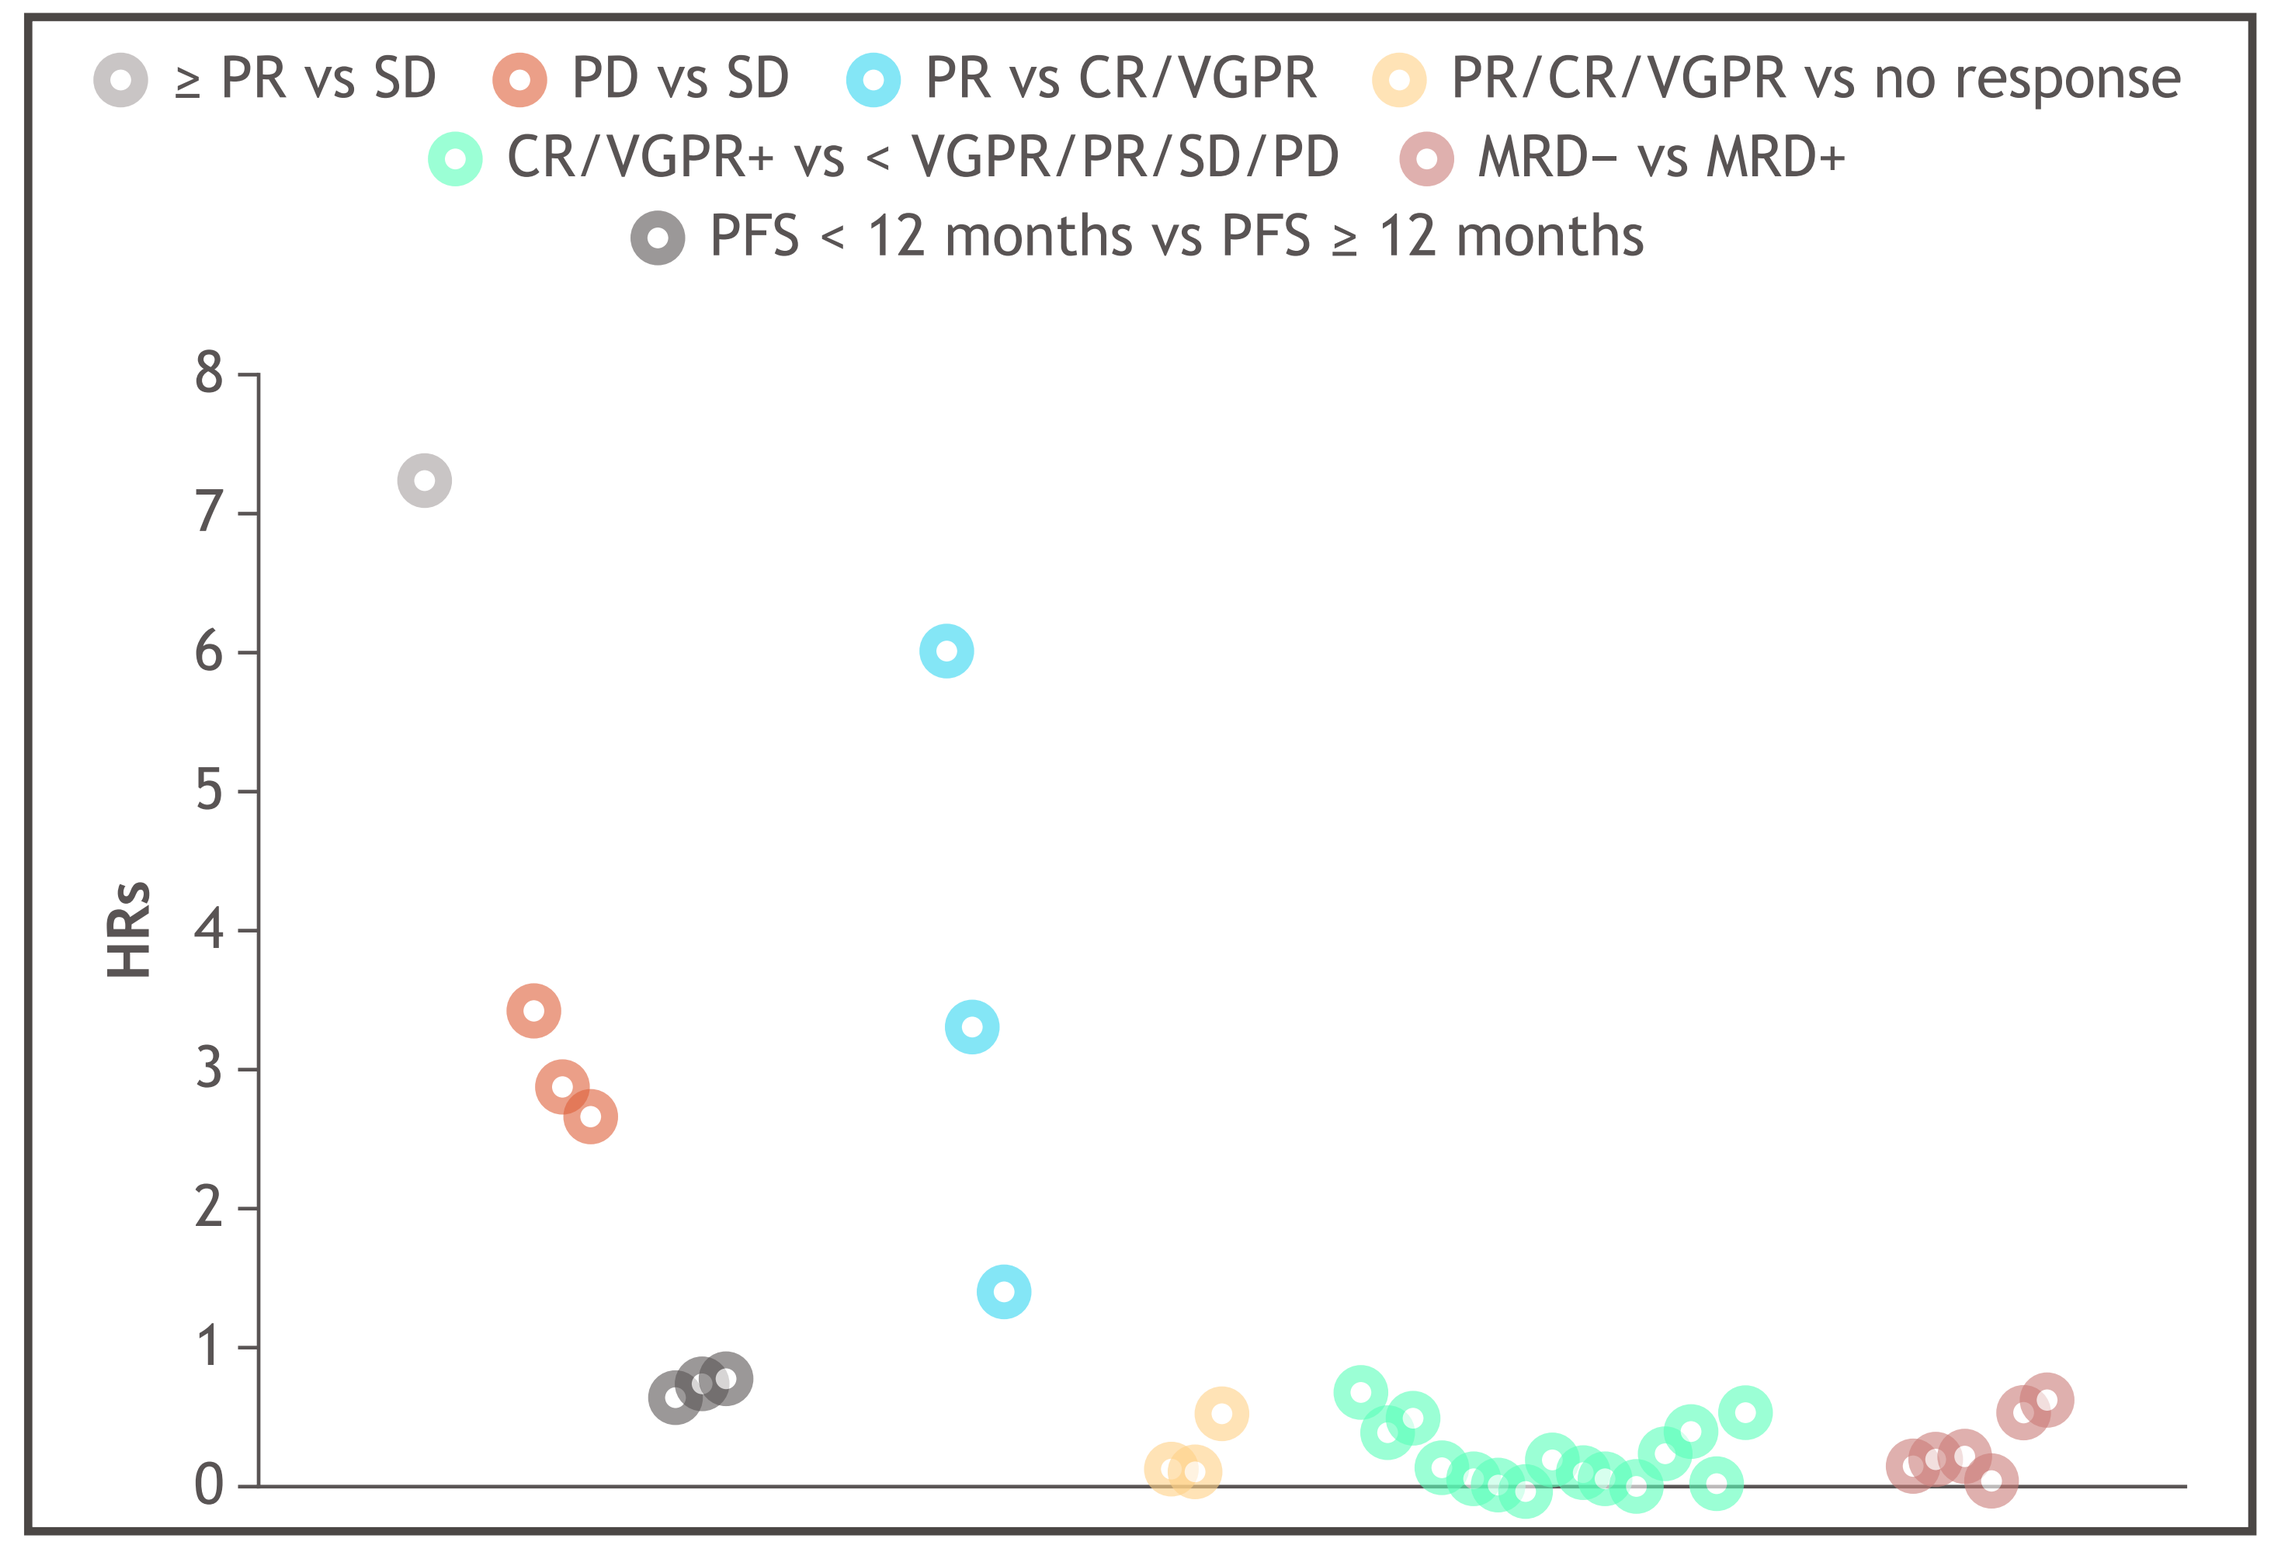

≥ PR vs SD
PD vs SD
PR vs CR/VGPR
PR/CR/VGPR vs no response
CR/VGPR+ vs < VGPR/PR/SD/PD
MRD− vs MRD+
PFS < 12 months vs PFS ≥ 12 months
HRs
8
7
6
5
4
3
2
1
0

Conclusions

- Evidence from the publications identified in this SLR demonstrates the prognostic value of certain endpoints such as CR, VGPR, and MRD on OS; however, only a few publications established PFS, EFS, and TTP as validated surrogate endpoints for OS in MM
- Additional studies are required to validate surrogate endpoints for OS in MM, which will help strengthen the current body of evidence surrounding surrogate endpoints for OS, potentially supporting HTA

References

1. Sung H, et al. *CA Cancer J Clin* 2021;71:209-249.
2. Centre for Reviews and Dissemination (CRD). CRD's guidance for undertaking reviews in health care. University of York website. https://www.york.ac.uk/media/crd/Systematic_Reviews.pdf. Published January 2009. Accessed October 4, 2021.
3. Félix J, et al. *BMC Cancer* 2013;13:122.
4. Hari P, et al. *Clin Lymphoma Myeloma Leuk* 2018;18:152-160.
5. Cartier S, et al. *Oncol Res Treat* 2015;38:88-94.
6. Mainou M, et al. *Eur J Haematol* 2017;98:563-568.
7. Rawstron AC, et al. *Blood* 2015;125:1932-1935.
8. Aggarwal M, et al. *Ann Hematol* 2018;97:1869-1877.
9. D'Agostino M, et al. *Clin Cancer Res* 2020;26:4832-4841.
10. Medina A, et al. *Blood Cancer* J 2020;10:108.
11. Alonso R, et al. *Am J Hematol* 2019;94:853-861.
12. Rawstron AC, et al. *J Clin Oncol* 2013;31:2540-2547.
13. Avivi I, et al. *Am J Hematol* 2019;94:1132-1140.
14. Binder M, et al. *Am J Hematol* 2015;90:888-891.
15. Chanan-Khan AA, Giralt S. *J Clin Oncol* 2010;28:2612-2624.
16. Cohen YC, et al. *Leuk Lymphoma* 2016;57:1382-1388.
17. Decaux O, et al. *Clin Lymphoma Myeloma Leuk* 2012;12:418-422.
18. Gassiot S, et al. *Clin Lymphoma Myeloma Leuk* 2019;19:585-592.e1.
19. Gay F, et al. *Blood* 2011;117:3025-3031.
20. Gertz MA, et al. *Blood* 2010;115:2348-2353.
21. Li H, et al. *Ann Hematol* 2019;98:1185-1195.
22. Iwama K-I, et al. *Eur J Haematol* 2013;90:134-141.
23. Jaglowski SM, et al. *Bone Marrow Transplant* 2014;49:1323-1329.
24. Jimenez-Zepeda VH, et al. *Bone Marrow Transplant* 2015;50:204-208.
25. Gu J, et al. *Biol Blood Marrow Transplant* 2018;24:2568-2574.
26. Kazmi SM, et al. *Clin Lymphoma Myeloma Leuk* 2015;15:687-693.
27. Kumar L, et al. *Cancer Med* 2014;3:939-946.
28. Kumar L, et al. *Natl Med J India* 2016;29:192-199.
29. Lee HS, et al. *J Korean Med Sci* 2013;28:80-86.
30. Majithia N, et al. *Am J Hematol* 2015;90:981-985.
31. Tschautscher MA, et al. *Am J Hematol* 2019;94:751-756.
32. Martino M, et al. *Clin Lymphoma Myeloma Leuk* 2014;14:148-154.
33. Mele G, et al. *Leuk Res* 2015;39:279-283.
34. Mellors PW, et al. *Am J Hematol* 2018;93:889-894.
35. Migkou M, et al. *Eur J Haematol* 2011;87:323-329.
36. Min C-K, et al. *Blood Res* 2013;48:198-205.
37. Moreau P, et al. *Leuk Lymphoma* 2016;57:2839-2847.
38. Morgan GJ, et al. *Blood* 2011;118:1231-1238.
39. Munshi NC, et al. *JAMA Oncol* 2017;3:28-35.
40. Ong SY, et al. *Bone Marrow Transplant* 2016;51:933-937.
41. Perrot A, et al. *Blood* 2018;132:2456-2464.
42. Prica A, et al. *Clin Lymphoma Myeloma Leuk* 2013;13:25-31.
43. Puig N, et al. *Leukemia* 2014;28:391-397.
44. Chakraborty R, et al. *Biol Blood Marrow Transplant* 2017;23:598-605.
45. Rasche L, et al. *Biol Blood Marrow Transplant* 2016;21:1988-1996.
46. Gupta R, et al. *Ann Hematol* 2018;97:2447-2454.
47. Samaras P, et al. *Swiss Med Wkly* 2011;141:w13203.
48. Sinha S, et al. *Br J Haematol* 2010;148:853-858.
49. Spina F, et al. *Leuk Lymphoma* 2011;52:1262-1270.
50. Sidana S, et al. *Leukemia* 2019;33:730-738.
51. Tan D, et al. *Bone Marrow Transplant* 2010;45:1625-1630.
52. Usmani SZ, et al. *Blood Cancer J* 2018;8:123.
53. Munshi NC, et al. *Blood Adv* 2020;4:5988-5999.
54. National Institute for Health and Care Excellence (NICE). Single technology appraisal. NICE website. https://www.nice.org.uk/guidance/ta587/evidence/committee-papers-pdf-6836182237. Accessed May 3, 2021.
55. National Institute for Health and Care Excellence (NICE). Single technology appraisal. NICE website. https://www.nice.org.uk/guidance/ta658/evidence/appraisal-consultation-committee-papers-pdf-8900336989. Accessed May 3, 2021.
56. National Institute for Health and Care Excellence (NICE). Single technology appraisal. NICE website. https://www.nice.org.uk/guidance/ta573/evidence/appraisal-consultation-committee-papers-pdf-6727151053. Accessed May 3, 2021.
57. Pan-Canadian Oncology Drug Review (pCODR). Final clinical guidance report. pCODR website. https://www.cadth.ca/sites/default/files/pcodr/pcodr_carfilzomib_kyprolis_mm_fn_cgr.pdf. Accessed October 20, 2021
58. Pan-Canadian Oncology Drug Review (pCODR). Final clinical guidance report. pCODR website. https://www.cadth.ca/sites/default/files/pcodr/pcodr_daratumumab_darzalex_mm_2ndln_fn_cgr.pdf. Accessed October 20, 2021.
59. Pan-Canadian Oncology Drug Review (pCODR). Final clinical guidance report. pCODR website. https://www.cadth.ca/sites/default/files/pcodr/Reviews2020/10189DaratumumabNDMM_fnCGR_Post05Mar2020_REDACT_Final.pdf. Accessed October 20, 2021.
60. Pan-Canadian Oncology Drug Review (pCODR). Final clinical guidance report. pCODR website. https://www.cadth.ca/sites/default/files/pcodr/Reviews2019/10165PomalidomideBortezomibMM_fnCGR_REDACT_EarlyConv_Post_18Sep2019_final.pdf. Accessed
61. Pan-Canadian Oncology Drug Review (pCODR). Canadian Agency for Drugs and Technologies in Health (CADTH). Initial Clinical Guidance Report. CADTH website. https://www.cadth.ca/sites/default/files/pcodr/Reviews2021/10220IsatuximabMM_inCGR_NOREDACT_Post04Feb2021_final.pdf. Accessed October 20, 2021.
62. Pan-Canadian Oncology Drug Review (pCODR). Canadian Agency for Drugs and Technologies in Health (CADTH). Final Clinical Guidance Report. CADTH website. https://www.cadth.ca/sites/default/files/pcodr/pcodr_carfilzomib_kyprolis_mm_rel_fn_cgr.pdf. Accessed October 20, 2021.
63. Institute for Clinical and Economic Review (ICER). Evidence report. ICER website. https://icer.org/wp-content/uploads/2020/10/MWCEPAC_MM_Evidence_Report_050516-1.pdf. Accessed May 19, 2021.
64. Institute for Clinical and Economic Review (ICER). Draft evidence report. ICER website. https://icer.org/wp-content/uploads/2020/10/ICER_Multiple-Myeloma_Draft-Report_021121.pdf. Accessed May 19, 2021.
65. Der Gemeinsame Bundesausschuss (G-BA). Dossier zur nutzenbewertung. G-BA website. https://www.g-ba.de/downloads/92-975-2623/2018-09-28_Modul4A_Daratumumab.pdf. Accessed October 20, 2021.
66. National Institute for Health and Care Excellence (NICE). Single technology appraisal. NICE website. https://www.nice.org.uk/guidance. Accessed October 4, 2021.
67. Pan-Canadian Oncology Drug Review (pCODR) - Canadian Agency for Drugs and Technologies in Health (CADTH). Clinical guidance - health technology review. CADTH website. https://www.cadth.ca/healthtechnology-review. Accessed October 4, 2021.
68. Institute for Quality and Efficiency in Health Care (IQWiG). IQWiG website. https://www.iqwig.de/en/. Accessed October 4, 2021.
69. Dimopoulos M, et al. Value Health 2017;20:A408.

Disclosures

- E.Y., B.M.: Bristol Myers Squibb - employment; I.D., E.F.P.P., A.S., J.A.S.: OPEN Health Group - employment.

Acknowledgments

- The study was supported by Bristol Myers Squibb
- All authors contributed to and approved the presentation; editorial assistance was provided by Eilish McBurnie, PhD, of Excerpta Medica, funded by Bristol Myers Squibb

Presented at the Virtual International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe 2021 Annual Meeting, November 30 – December 3, 2021.

Email: Emre.Yucel@bms.com

Copies of this poster are for personal use only and may not be reproduced without written permission from the author of this poster.