MEASURES TO EVALUATE QUALITY OF CARE IN HEAD AND NECK CANCER: INTERIM RESULTS OF A DELPHI STUDY

Cátedra UFV-Fundación ECO en Oncología centrada en la persona

Cruz Hernández JJ[1], Arrazubi V[2], Escobar Y[3], García Castaño A[4], Grau JJ[5], Iglesias L[6], Lambea J[7], Pérez Segura P[8], Antón Rodríguez C[9], Caballero F[9], Campos-Lucas FJ[9], Lugo I[10], Monge D[9], Rogado Á[10], Santamaría I[9], Rueda A[11]

[1]Fundación ECO (Excelencia y Calidad de la Oncología). Servicio de Oncología Médica. Hospital Universitario de Salamanca. Universidad de Salamanca. Instituto de Investigación Biomédica de Salamanca (IBSAL), Salamanca, Spain. [2]Servicio de Oncología Médica. Complejo Hospitalario de Navarra, Pamplona, Spain. [3]Fundación ECO (Excelencia y Calidad de la Oncología). Servicio de Oncología Médica. Hospital Universitario Gregorio Marañón, Madrid, Spain. [4]Servicio de Oncología Médica. Hospital Universitario Marqués de Valdecilla, Santander, Spain. [5]Servicio de Oncología Médica. Hospital Clínic de Barcelona, Barcelona, Spain. [6]Servicio de Oncología Médica. Hospital Universitario 12 de Octubre, Madrid, Spain. [7]Servicio de Oncología Médica. Hospital Clínico Universitario Lozano Blesa, Zaragoza, Spain. [8]Fundación ECO (Excelencia y Calidad de la Oncología). Servicio de Oncología Médica. Hospital Universitario Clínico San Carlos, Madrid, Spain. [9]Facultad de Medicina. Universidad Francisco de Vitoria, Madrid, Spain. [10]Fundación ECO (Excelencia y Calidad de la Oncología), Madrid, Spain. [11]Fundación ECO (Excelencia y Calidad de la Oncología). UGCI Oncología Médica. Hospitales Universitarios Regional y Virgen de la Victoria, Málaga, Spain.

OBJECTIVES

This study aimed to develop measures to evaluate the quality of care in head and neck cancer, far which no specific measures are currently used in Spain or in international quality-of-care evaluation programs.

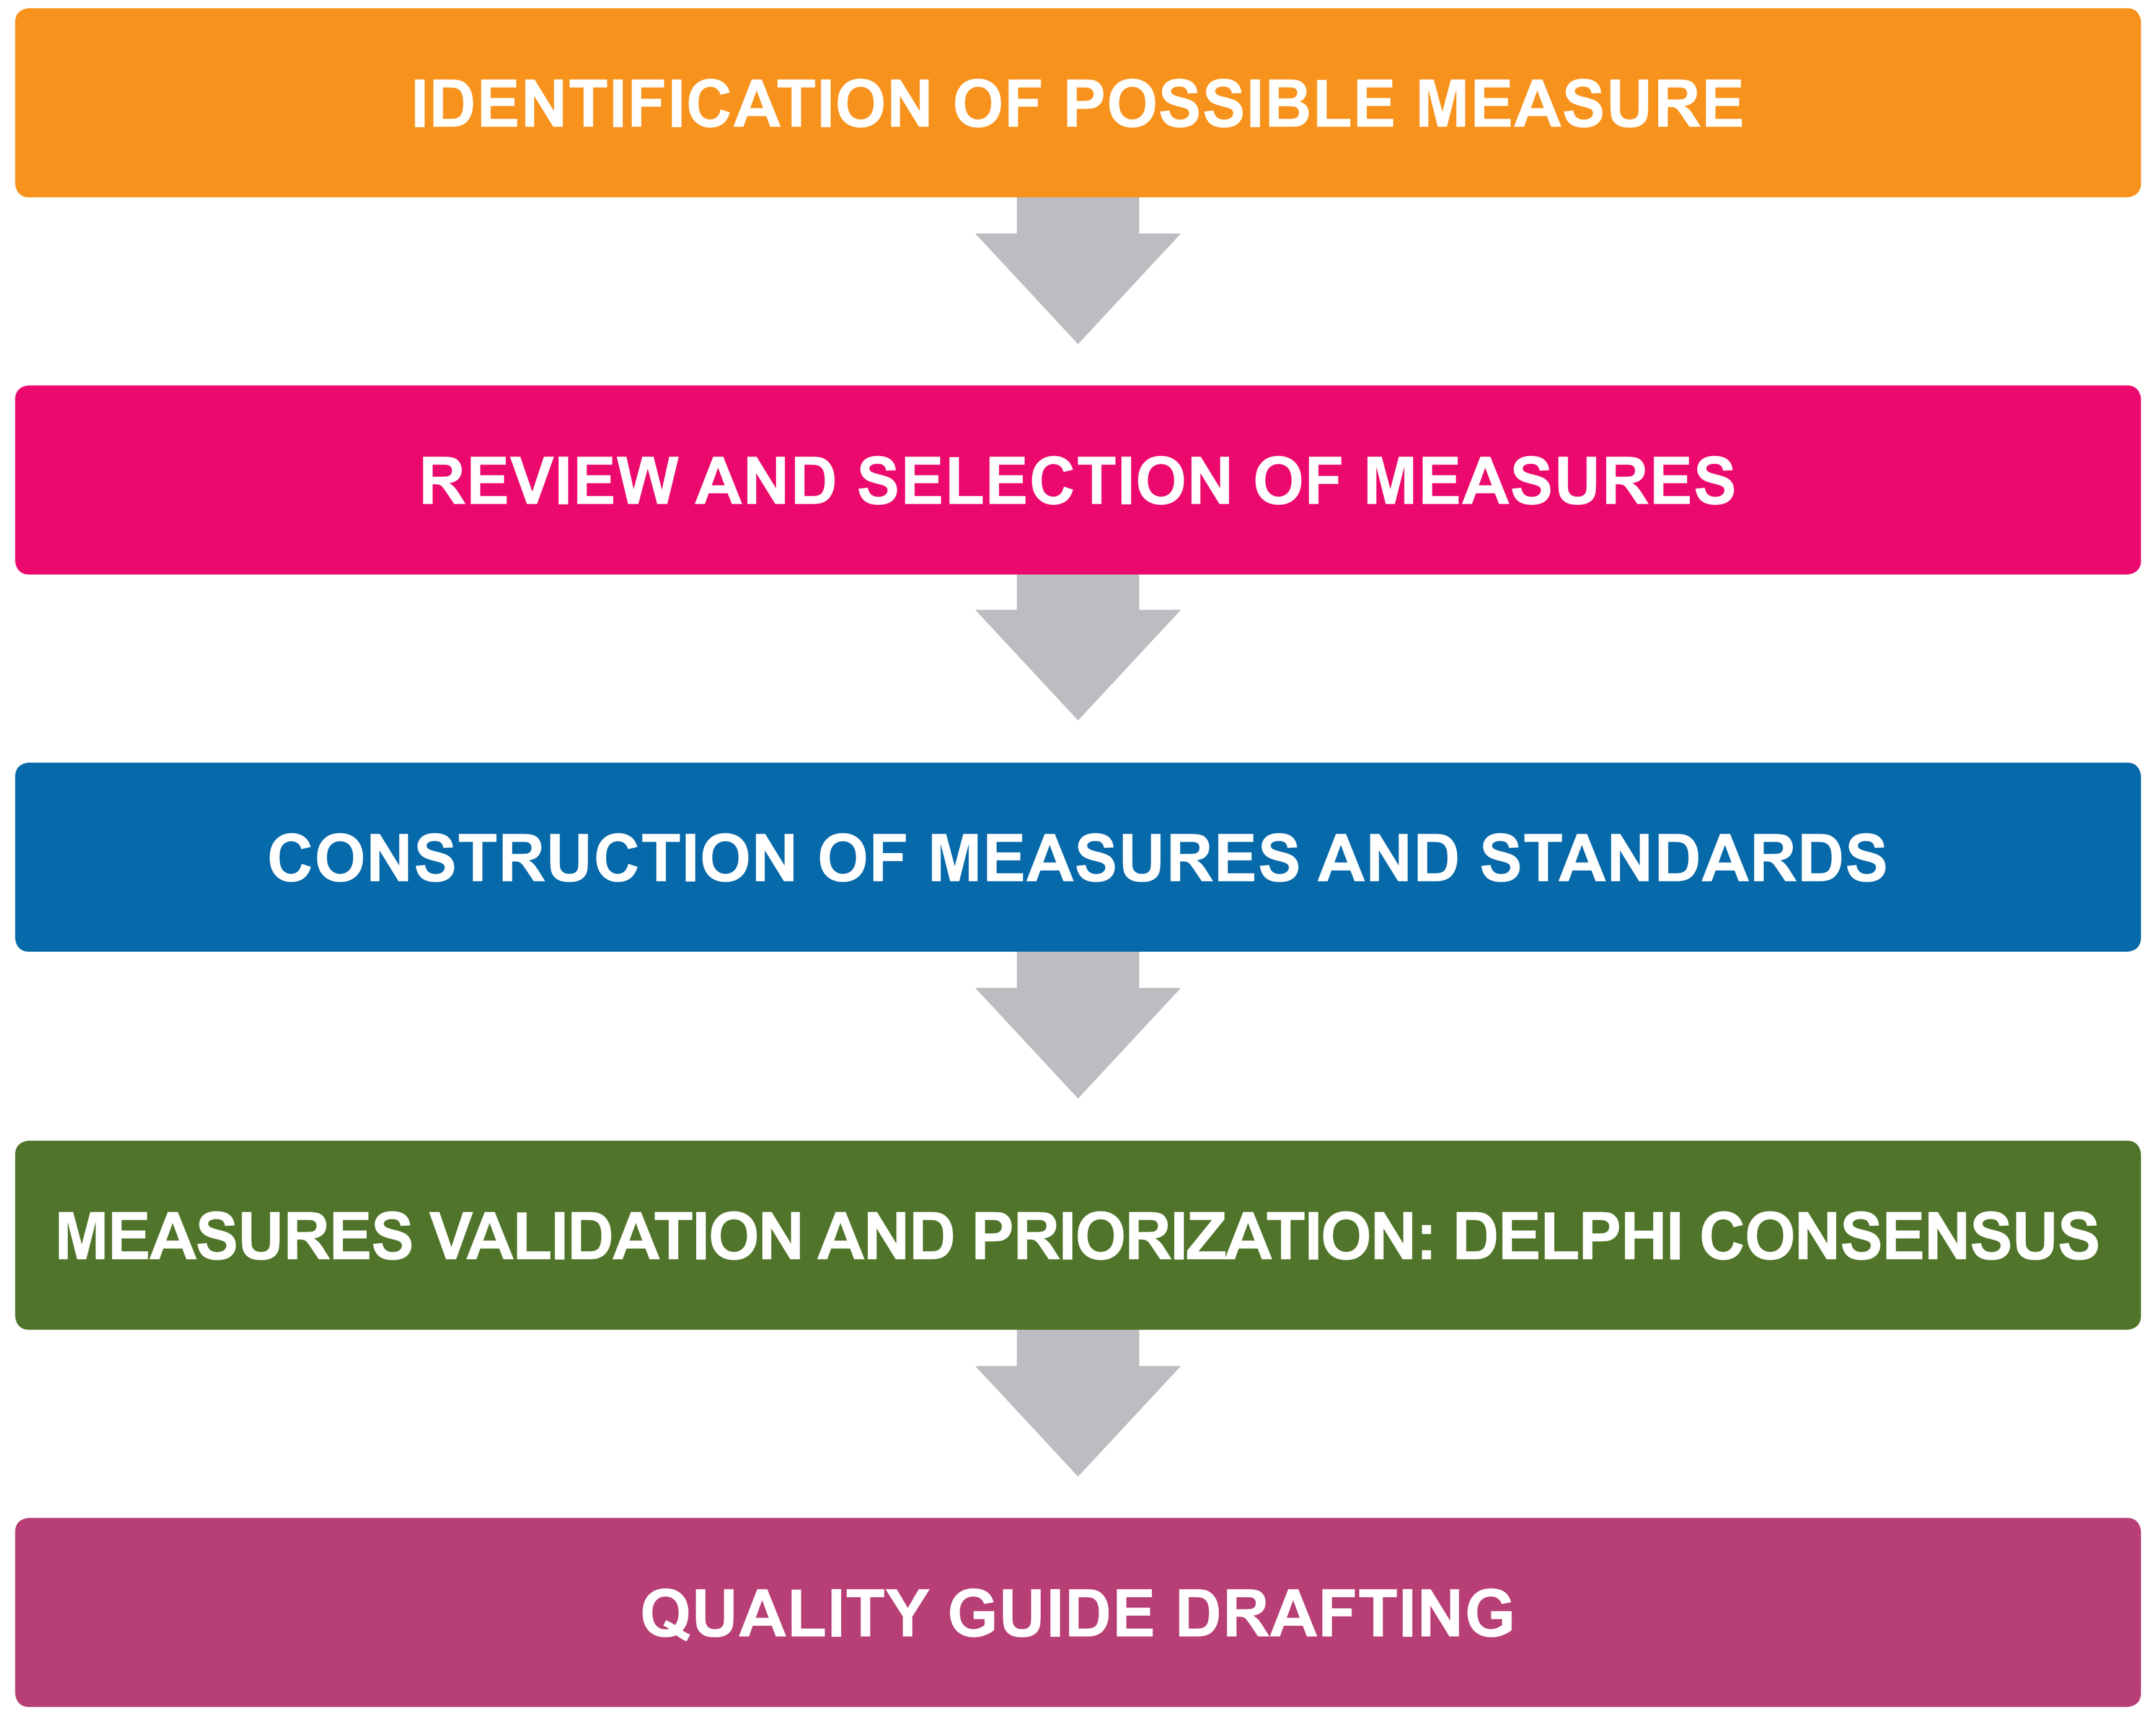
IDENTIFICATION OF POSSIBLE MEASURE
REVIEW AND SELECTION OF MEASURES
CONSTRUCTION OF MEASURES AND STANDARDS
MEASURES VALIDATION AND PRIORIZATION: DELPHI CONSENSUS
QUALITY GUIDE DRAFTING

Figure 1. Phases of the project for the measures' development.

METHODS

A systematic literature review was conducted to identify measures far evaluating quality of care in head and neck cancer. A scientific committee, comprising 9 medical oncologists specialized in head and neck cancer, reviewed the literature findings and defined criteria. Fifty measures were evaluated with a 2-step Delphi method to gather feedback from 52 experts in head and neck cancer in Spain, specialized in medical oncology, radiotherapy oncology, maxillofacial surgery, pathology or otorhinolaryngology. In the Delphi, experts scored the appropriateness of the measures using a 9-point Likert scale (1, extremely inappropriate; 9, extremely appropriate). Consensus was defined as at least two-thirds of Delphi respondents selecting a score sub-category (1-3, 4-6, or 7-9) that encompassed the median score of the group.

RESULTS

Twenty documents were selected from the literature review. Consensus was reached on the appropriateness of all 50 measures-covering diagnosis (13), treatment (28), fallow-up (5), and outcome (4)-far evaluating the quality of care in head and neck cancer. The measures with lowest scores concerned re-hospitalization shortly after surgery or hospitalization time after surgery; those with highest scores regarded correct fallow-up, use of imaging far fallow-up, and complete resection of the tumor.

CONCLUSIONS

The goal of this study is to develop measures to evaluate and improve the quality of care in head and neck cancer. These interim results show unanimous consensus on the proposed measures on diagnosis, treatment, fallow-up and outcome. Next, the scientific committee will prioritize and select the final set of measures and will develop index cards with definitions and instructions on how to use them in clinical practice.

TABLE 1. MEASURE'S TECHNICAL NOTE TEMPLATE

QUALITY MEASURE TECHNICAL NOTE

Measure name	Short descriptive name that designates the measure
Definition	Characteristic or attribute of the care provided, so that it is considered of quality
Formula	Mathematical expression that will reflect the result of the measurement (usually expressed as a percentage, but it can also be done as a mean or absolute number)
Type of measure	Structure, process or outcome
Justification / exclusions clarifications	JUSTIFICATION: Usefulness of the measure as a measure of quality. It is related to validity (regarding what we are going to measure, does it make sense?).
Target level / standard	Desired level of compliance for the measure, proposed by the ECO Foundation.
Information sources	Origin of the information and sequence for obtaining the necessary data to be able to quantify the measure.

Model proposed by the Spanish Society for Healthcare Quality (SECA) in its proposal for quality indicators for hospitals of the Spanish National Health System published in 2012

#112841

First author: jjcruz@usal.es