Budget Impact Analysis Of An Alternative Meningococcal Conjugate Vaccine For High-risk Toddlers In Egypt

Hamouda M[1], Hassan H[2], Elsisi G[3], Ibrahim R[4], Temerik A[5], Hegazy H[6]

[1]GSK Value Evidence and Outcomes, Dubai, United Arab Emirates, [2]National Organization for Drug Control and Research "NODCAR", Maadi, Egypt, [3]HTA office, cairo, Egypt, [4]MOH, Giza, Egypt,[5]Shifa Alorman Oncology Hospital, Luxor,LX, Egypt, [6]Children Cancer Hospital 75375, Cairo Egypt

Background

- Meningitis is a devastating disease with a high case fatality rate and leading to serious long-term complications.[1]
- Meningitis remains a major global public-health challenge.[1]
- Invasive Meningococcal Meningitis has an estimated incidence of 0.1/100,000 in Egypt.[2]
- Rates of disease are highest in children younger than 1 year old.[3]
- The most effective way is the application of preventive mechanisms through meningitis vaccination.[1]
- IMM vaccination has an impact in reducing the disease burden by delivering long-lasting protection.[1]

Objectives

- This study aims at estimating clinical and economic benefits of providing an alternative meningococcal meningitis conjugate vaccine (MenACWY-DT) in high-risk toddlers in Egypt.
- A budget impact analysis is constructed to adopt the MenACWY-DT vaccine in the Egyptian national immunization program to high-risk toddlers.

Methods and Materials

- A decision tree model was developed to simulate the costs and health outcomes through 5-year time horizon from healthcare system perspective.
- The analysis is performed to compare between two scenarios; two doses MenACWY-DT vaccine compared with no vaccination.

References

1. Who.int. 2021. Meningitis. [online] Available at: <https://www.who.int/news-room/fact-sheets/detail/meningitis> [Accessed 28 September 2021].
2. Nakhla, I., Frenck, R., Teleb, N., Oun, S., Sultan, Y., Mansour, H. and Mahoney, F., 2005. The changing epidemiology of meningococcal meningitis after introduction of bivalent A/C polysaccharide vaccine into school-based vaccination programs in Egypt. *Vaccine*, (23), pp.3288-3293.
3. Cdc.gov. 2019. Meningococcal Disease: Technical and Clinical Information | CDC. [online] Available at: <https://www.cdc.gov/meningococcal/clinical-info.html> [Accessed 10 November 2021].

Materials and Methods (cont.)

- The reported health outcomes were estimated per annual IMM cases collected by literature rapid review.
- The total annual costs include the vaccination cost and IMM treatment cost derived from semi-structured interviews through a Delphi panel.
- The reported budget impact was the difference in the total annual costs of the two scenarios. Uncertainty was handled through one-way sensitivity analysis.

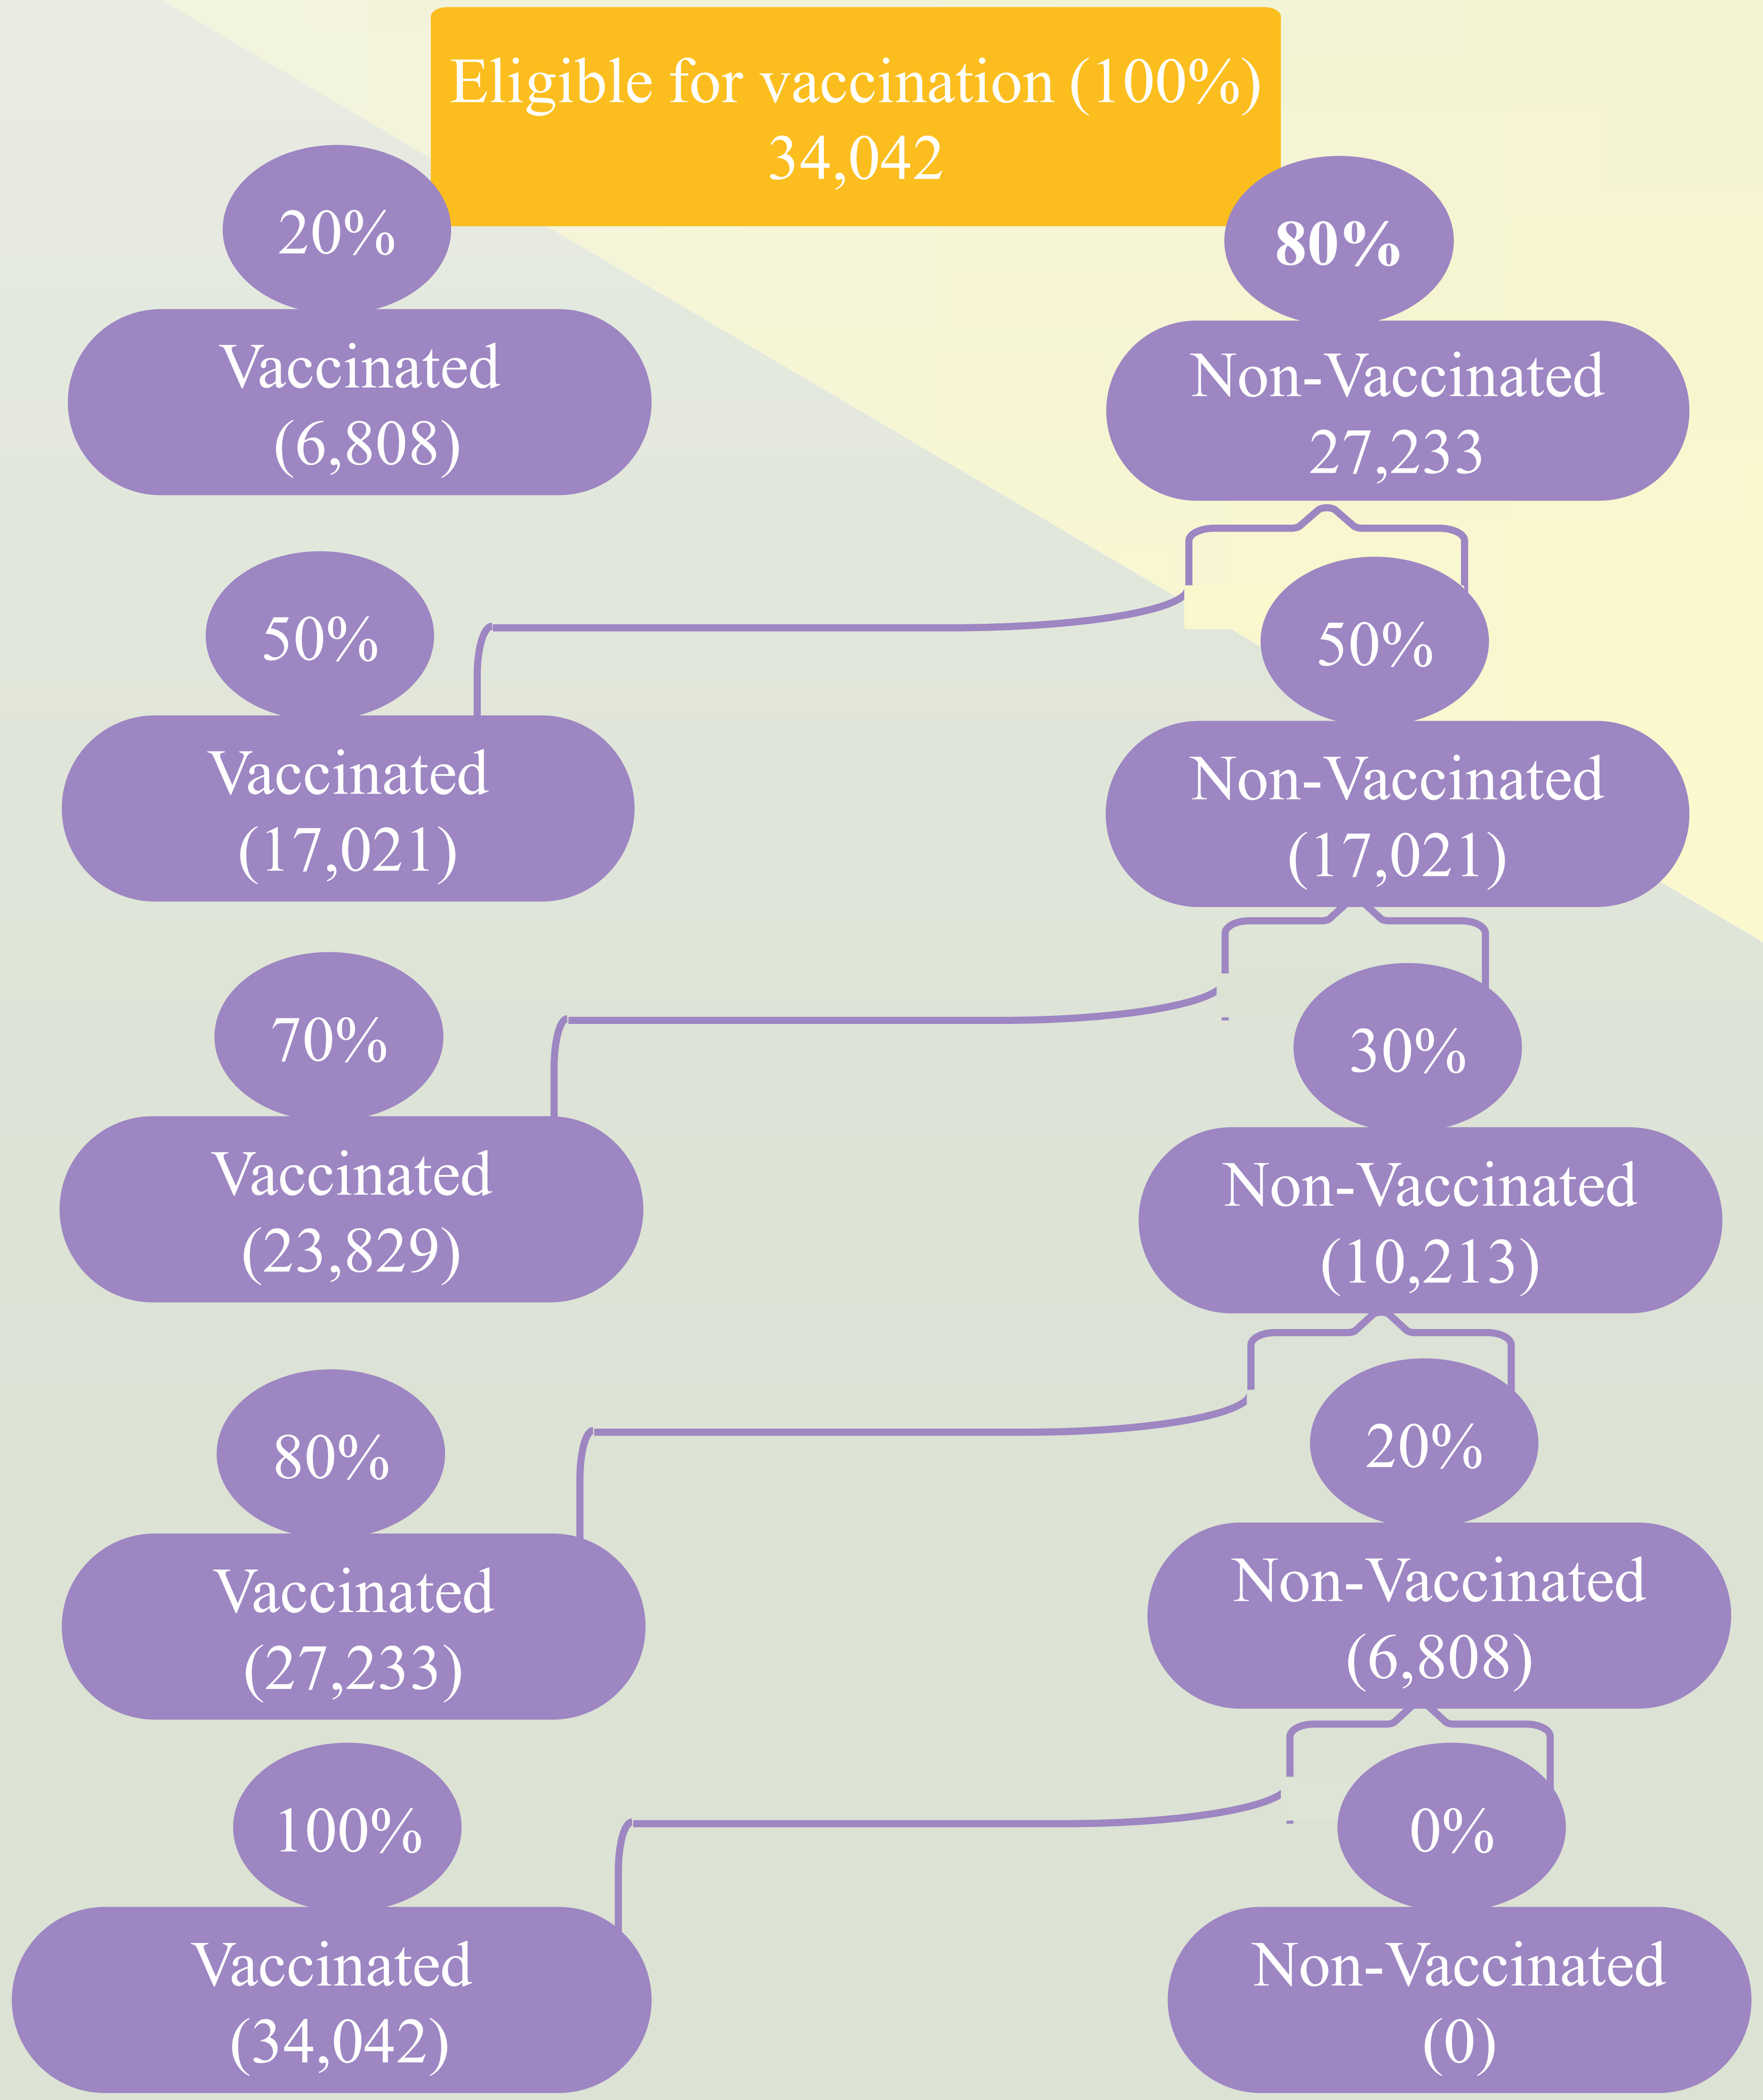
Eligible for vaccination (100%) 34,042
20%
Vaccinated (6,808)
80%
Non-Vaccinated 27,233
50%
Vaccinated (17,021)
50%
Non-Vaccinated (17,021)
70%
Vaccinated (23,829)
30%
Non-Vaccinated (10,213)
80%
Vaccinated (27,233)
20%
Non-Vaccinated (6,808)
100%
Vaccinated (34,042)
0%
Non-Vaccinated (0)

Materials and Methods (cont.)

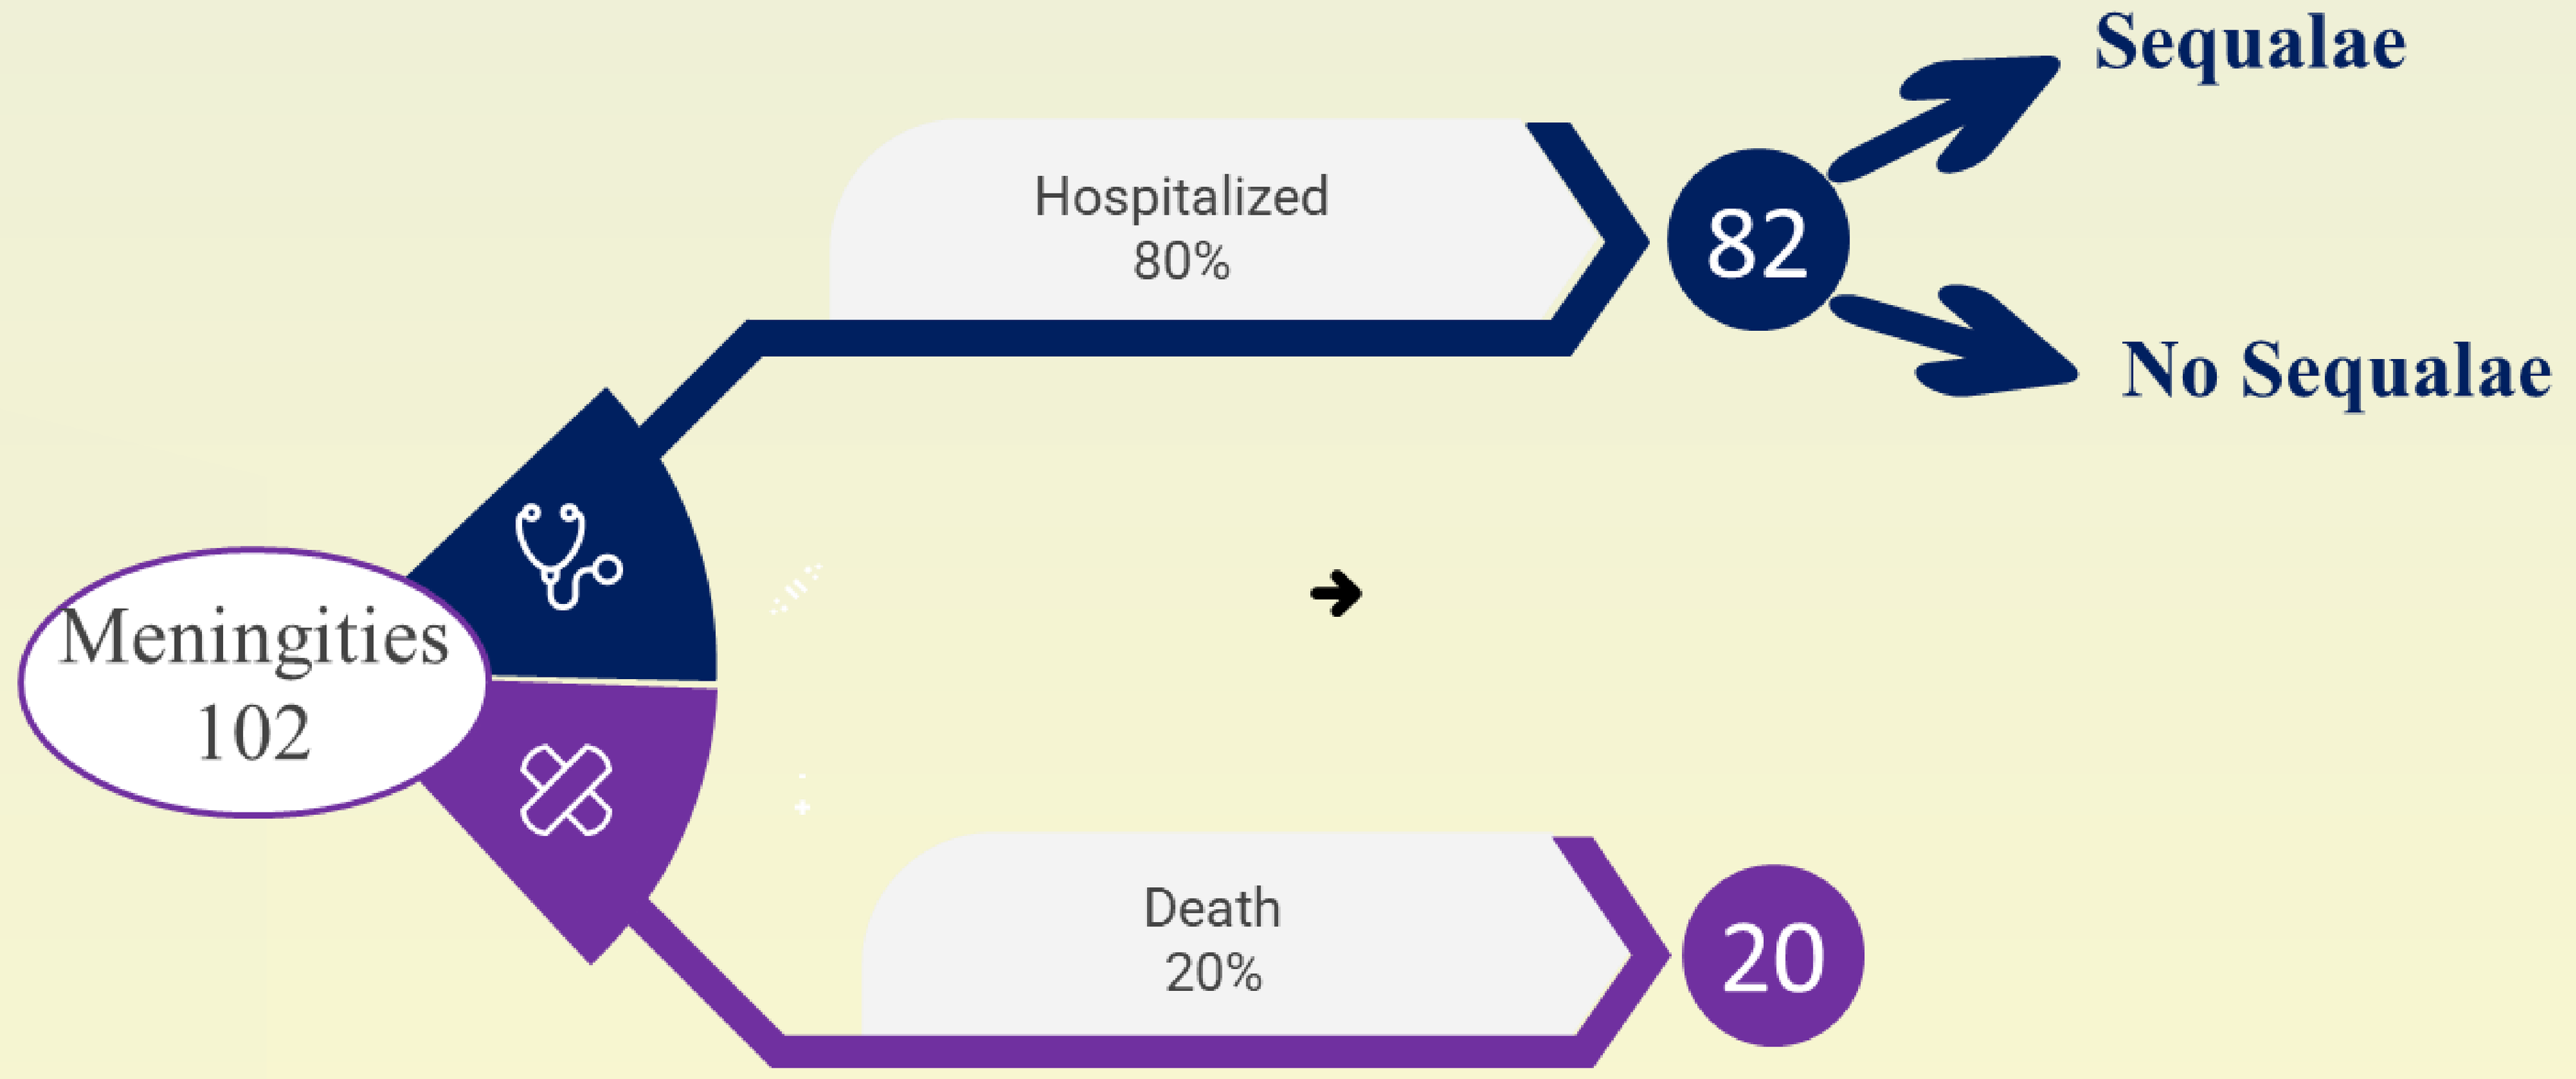
Meningities 102
Hospitalized 80%
82
Sequalae
No Sequalae
Death 20%
20

Results

The inclusion of MenACWY-DT vaccine for high-risk toddlers would contribute in:

- Treatment cost saving by 154,912,699 EGP over 5 years due to reduction in IMM cases.
- An incremental cost on the healthcare budget expenditure of 2,699,122,096 EGP.

Conclusion

- The implementation of the conjugate meningococcal meningitis vaccine for high-risk toddlers in the Egyptian national immunization program has a substantial impact on public health and subsequent health outcomes.
- MenACWY- DT vaccine adoption will endure significant incremental cost yet will reflect on the decrease in incidence rate, reduce number of deaths due to IMM and prevent its sequalae, preventable complications and disabilities.
- Public health planning may consider an alternative strategy for high risk groups rather than meningitis polysaccharide vaccine at pre-school age.