COST OF ILLNESS BY DISEASE STAGE OF PATIENTS WITH MYCOSIS FUNGOIDES AND SÉZARY SYNDROME: A REAL-WORLD ANALYSIS IN SPANISH PATIENTS (MICADOS STUDY)

STUDY MICADOS

POSB48

Navarro Matilla B,[1] Ortiz Romero PL,[2] Pujol Vallverdú RM,[3] Combalia Escudero A,[4] Zapata Paz I,[1] González Barca E,[5] Muniesa Montserrat C,[6] Morillo Andújar M,[7] Pérez Ferriols A,[8] Román Curto C,[9] Fernández de Misa Cabrera R,[10] Hospital Gil M,[1] Marín Niebla A,[11] Rios Rull PJ,[10] de la Cruz F,[7] Izu Belloso RM,[12] Martín García-Sancho A,[13] Parera Amer ME,[14] Córdoba R,[15] Ramón Quiles MD,[16] Saus Carreres A,[16] Del Campo García R,[14] Machan S,[15] Viguera Ester P,[17] Blanco Garnelo J,[17] Silva Romero I,[17] Soler López B[18]

1. Hospital Universitario Puerta de Hierro, Madrid, Spain; 2. Hospital Universitario 12 de Octubre, Madrid, Spain; 3. Hospital del Mar, Barcelona, Spain; 4. Hospital Universitario Clínic i Provincial, Barcelona, Spain; 5. Institut Catalá D'Oncología - Hospitalet, Barcelona, Spain; 6. Hospital Universitario de Bellvitge, Barcelona, Spain; 7. Hospital Universitario Virgen del Rocío, Sevilla, Spain; 8. Hospital General Universitario de Valencia, Valencia, Spain; 9. Hospital General Universitario de Salamanca, Salamanca, Spain; 10. Hospital Universitario Nuestra Señora de Candelaria, Santa Cruz de Tenerife, Spain; 11. Hospital Universitario Vall d'Hebrón/VHIO, Barcelona, Spain; 12. Hospital de Basurto, Bilbao, Spain; 13. Hospital General Universitario de Salamanca, Salamanca, Spain; 14. Hospital Son Llàtzer, Palma de Mallorca, Spain; 15. Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; 16. Hospital Clínico Universitario, Valencia, Spain; 17. Kyowa Kirin Farmacéutica, S.L., Madrid, Spain; 18. E-C-BIO,S.L., Las Rozas, M, Spain

BACKGROUND

Cutaneous T-cell lymphoma (CTCL) has been associated with a higher annual incremental cost than any other skin disease and is the costliest from 24 skin diseases meaning about **$3,182.60** annual cost by patient.[1-3]

No data about cost of illness has been quantified in Spain up till now. The economic burden of CTCL for patients and society remains to be quantified.

OBJECTIVES

The study objective was to determine the total and by patient cost of illness of cutaneous T-cell lymphoma(CTCL) in Spain by clinical stage.

METHODS

A one-year retrospective pharmacoeconomic cost of illness study was designed.

A total of 23 dermatologists and hematologists from 15 Spanish public hospitals completed the study.

Adult patients with mycosis fungoides (MF) and Sézary syndrome (SS) were selected with diagnosis established at least one year before the study.

The economic study was done under the National Health Service perspective, with a time horizon of one year. The direct medical costs comprised: visits and admissions, laboratory tests, imaging tests, pharmacy, and therapeutic intervention costs.

Table 1. Mycosis fungoide and Sézary síndrome total annual direct costs in Spain (€2020). Data diaggregated by disease stage, and direct resource

Stage	I	II	III	IV	All stages
MF/SS prevalence (n)	4,872	247	11	61	5,295
Medical visits and hospital Admissions (€)	8,387,245	711,059	514,037	438,542	10,050,883
Laboratory tests (€)	3,878,599	356,465	227,837	156,624	4,619,525
Imaging tests (€)	1,244,796	163,378	93,388	71,869	1,573,431
Pharmacological treatments (€)	20,008,281	3,454,707	2,313,135	2,996,871	28,772,994
Non pharmacological treatments (€)	30,163,770	1,120,424	1,310,361	689,782	33,284,337
Total annual direct costs (€)	**63,682,692**	**5,806,034**	**4,458,758**	**4,353,687**	**78,301,171**

RESULTS

A total of 141 patients were included, 81 male (57.4%), with a mean age of 63.6 years old (95% CI 61.4-65.7).

MF was diagnosed in 109 patients (77.3%) and SS in 32 patients (22.7%). ECOG-0 was the functional status of 99 patients (70.2%), ECOG-1 in 29 patients (20.6%), ECOG-2 in 12 patients (8.5%).

The mean direct annual cost by patient at any stage was **€34,214**, being €11,953 at stage I, €23,506 at stage II, €38,772 at stage III and €72,749 at stage IV.

With an estimated annual incidence of 260 patients with MF/SS in Spain, the expected prevalence is about 5,295 patients.

The estimated total direct annual cost of MF/SS resulted in **€78,301,171** in Spain, where 81% of costs were attributable to patients in clinical stage I, 7% to stage II, 6% to stage III and 6% to stage IV. See disaggregated costs in Table 1 and percentual costs distribution in Figure 1.

Figure 1. Disaggregated total anual direct costs distribution by resource and Clinical stage in Spain (year 2020)

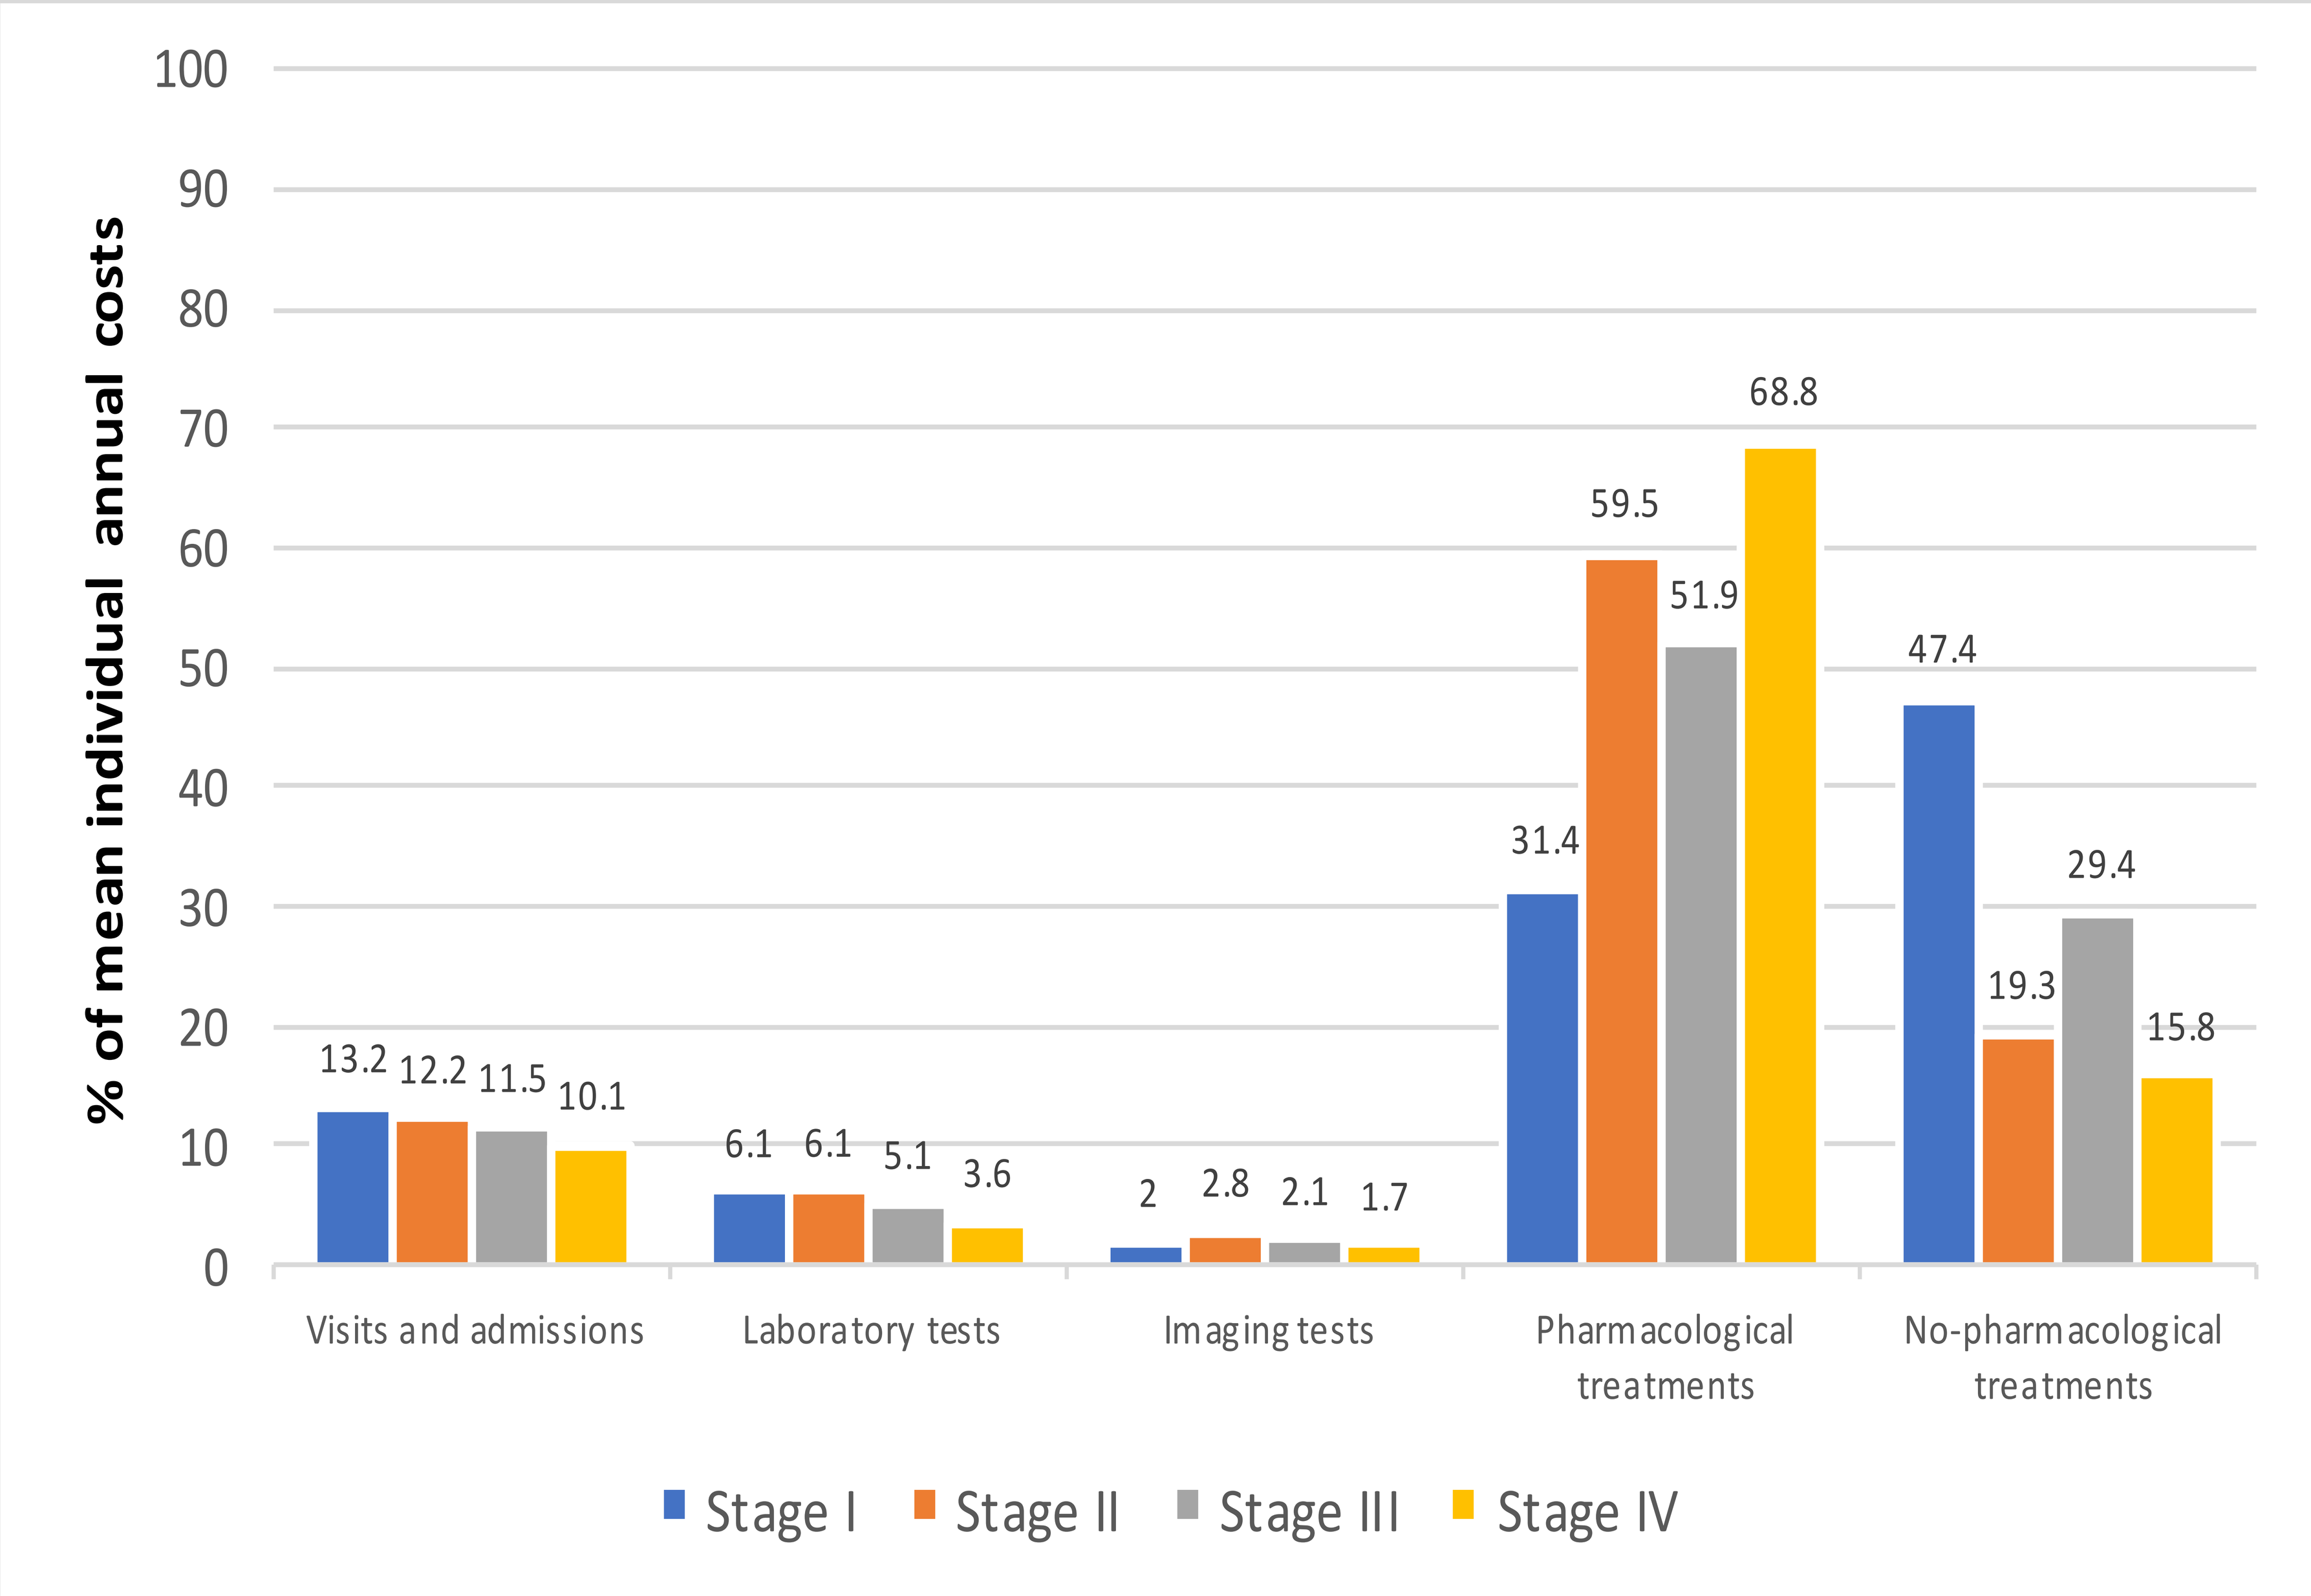
% of mean individual annual costs
100
90
80
70
60
50
40
30
20
10
0
13.2 12.2 11.5 10.1
6.1 6.1 5.1 3.6
2 2.8 2.1 1.7
31.4 59.5 51.9 68.8
47.4 19.3 29.4 15.8
Visits and admissions
Laboratory tests
Imaging tests
Pharmacological treatments
No-pharmacological treatments
Stage I
Stage II
Stage III
Stage IV

CONCLUSIONS

- CTCL has been described as the costliest of 24 skin diseases in the United States meaning **$3,183.60** mean annual cost by patient.[1-3]
- Ten-fold higher costs in CTCL Spanish patients that the costs reported for similar population in US was observed, considering only direct costs, being **€34,214** .
- The estimated total direct annual cost of MF/SS in Spain resulted in **€78,301,171**.
- Patient's costs and indirect costs due to loss of productivity need to be considered to describe the real burden of the disease and will further increase this difference.

TOPIC – Economic Evaluation DISEASE – Oncology, Rare and Orpham Diseases

REFERENCES

1. Lim HW, Collins SAB, Resneck JS, et al. Contribution of health care factors to the burden of skin disease in the United States. J Am Acad Dermatol 2017;76:1151-60. Doi: https://dx.doi.org/10.1016/j.jaad.2017.03.006
2. Lim HW, Collins SAB, Resneck JS, et al. The burden of skin disease in the United States. J Am Acad Dermatol 2017;76:958-972. DOI: https://dx.doi.org/10.1016/j.jaad.2016.12.043
3. Lim HW, Collins SAB, Resneck JS, et al. A risk adjustment approach to estimating the burden of skin disease in the United States. J Am Acad Dermatol 2017;78:129-140. Doi: https://doi.or/10.106/j.jaad.2017.08.06

Virtual ISPOR Europe 2021 Presenter

The study was sponsored by: KYOWA KIRIN