Capturing The Value Of Potentially Curative Oncology Therapies: Lessons From The Use And Acceptance Of Cure Modelling Assumptions In NICE Technology Appraisals

POSA266

Micallef J[1], New E[1], Satija A[1], Subramaniyan SV[1], Porteous A[1]

[1]Costello Medical, London, UK

OBJECTIVE

To explore the use and acceptance of cure assumptions in cost-effectiveness analyses as part of National Institute for Health and Care Excellence (NICE) health technology appraisals of oncology therapies.

BACKGROUND

- Novel oncology treatments, such as chimeric antigen receptor T-cell (CAR-T) therapies, immuno-oncology therapies and highly effective targeted treatments have emerged in the last few years as potential treatments for a variety of cancers. Due to their novel mechanism of action, these treatments may offer long-term survival and even cure.[1]
- Owing to this potential, these treatments often exhibit complex survival functions which standard methods for survival extrapolation may not adequately reflect. Accordingly, cure assumptions (where a proportion of the modelled population are implicitly or explicitly assumed to achieve long-term survival in line with the general population) are becoming increasingly common in cost-effectiveness analyses submitted as part of NICE appraisals.

METHODS

- The NICE website was searched on 20th May 2021 until the ten most recent completed technology appraisals for oncology therapies where the manufacturer's cost-effectiveness analysis included a cure assumption were identified.
- Information relating to the choice of modelling approach (including the Evidence Review Group comments and Appraisal Committee response) was extracted from these ten appraisals.

RESULTS

- Ten appraisals incorporating a cure assumption into the manufacturer's cost-effectiveness analyses were identified after searching the forty-four most recent oncology appraisals.
- Three appraisals utilised mixture cure models (MCMs) and six modelled cure (three explicitly, three implicitly) where survival was informed by general population mortality (GPM) after a certain timepoint for a proportion of patients **(Figure 1)**. One appraisal used a combination of MCM and GPM-based assumptions.
- A summary of the key features of these 10 appraisals is presented in **Table 1**.
- Manufacturers justified the use of cure-based modelling approaches by claiming that standard methods for survival extrapolation may not accurately reflect the survival profile of patients who had received novel oncology treatments.

1 Summary of appraisals that included a cure assumption in the cost-effectiveness analysis

Ref. number	Intervention	Indication	Modelling approach used	Cure assumption	Was the modelling approach accepted by the Committee?	Criticisms of the model: Limited data follow up	Lack of an observed plateau	Use of surrogate outcomes to justify cure	External choice of the proportion of patients cured
TA693	Olaparib plus bevacizumab	Advanced ovarian, fallopian tube or primary peritoneal cancer	MCM and GPM (explicit cure assumption)	PFS was extrapolated using an MCM. Patients who were progression-free at 5 years were considered 'cured' and considered to have GPM, with OS set equal to PFS	No	✓	✓	✓	
TA689	Acalabrutinib	Untreated and treated chronic lymphocytic leukaemia	GPM (implicit cure assumption)	Modelling of post-progression survival using OS for subsequent treatments resulted in an implicit assumption that a large proportion of patients were functionally cured after treatment with acalabrutinib and follow GPM	No	✓			
TA677	Autologous anti-CD19-transduced CD3+ cells	Relapsed or refractory mantle cell lymphoma in adults who have previously had a BTK inhibitor	MCM	OS and PFS were extrapolated using MCMs, where 'cured' patients had an increased relative risk of mortality (SMR of 1.09) versus GPM	Yes	✓			
TA673	Niraparib	Advanced ovarian, fallopian tube and peritoneal cancer after response to first line platinum-based chemotherapy	GPM (explicit cure assumption)	Patients who were progression-free after 7 years were assumed to never relapse and incur GPM	No	✓			
TA650	Pembrolizumab with axitinib	Untreated advanced renal cell carcinoma	GPM (implicit cure assumption)	The company capped overall survival by GPM. For pembrolizumab/axitinib, this happened at 20 years, suggesting 17% of people were effectively 'cured'	No		✓	✓	✓
TA649	Polatuzumab vedotin with bendamustine and rituximab	Relapsed/refractory DLBCL who are not candidates for haematopoietic stem cell transplant	MCM	OS and PFS were extrapolated using MCMs, where 'cured' patients followed GPM	No	✓	✓	✓	
TA642	Gilteritinib	Adults with relapsed or refractory FLT3 mutation positive acute myeloid leukaemia	GPM (explicit cure assumption)	All patients alive after Year 3 were assumed to be cured and follow GPM with an SMR of 2.0	Yes	✓			
TA638	Atezolizumab with carboplatin and etoposide	Adults with untreated extensive-stage, small-cell lung cancer	MCM	Extrapolations of OS and PFS using MCMs were explored. However, both the Company and the ERG agreed that that the use of MCMs was not appropriate for extrapolation	NA	✓	✓	✓	
TA632	Trastuzumab emtansine	HER2-positive early breast cancer in adults who have residual invasive disease after neoadjuvant therapy	GPM (explicit cure assumption)	The proportion of patients being 'cured' (no longer at risk of recurrence and only subject to GPM) linearly increased with time from 0% at 36 months to 95% at 120 months	Yes	✓			
TA600	Pembrolizumab with carboplatin and paclitaxel	Untreated metastatic squamous non-small-cell lung cancer	GPM (implicit cure assumption)	The Company capped overall survival by GPM. This happened at approximately 18 years, suggesting 10% of people were "cured"	No				✓

Abbreviations: BTK: bruton tyrosine kinase; DLBCL: diffuse large B-cell lymphoma; ERG: Evidence Review Group; GPM: general population mortality; MCM: mixture cure model; NA: not applicable; NICE: National Institute for Health and Care Excellence; OS: overall survival; PFS: progression-free survival; SMR: standardised mortality ratio.

1 Cure assumptions in recent NICE health technology appraisals of oncology therapies

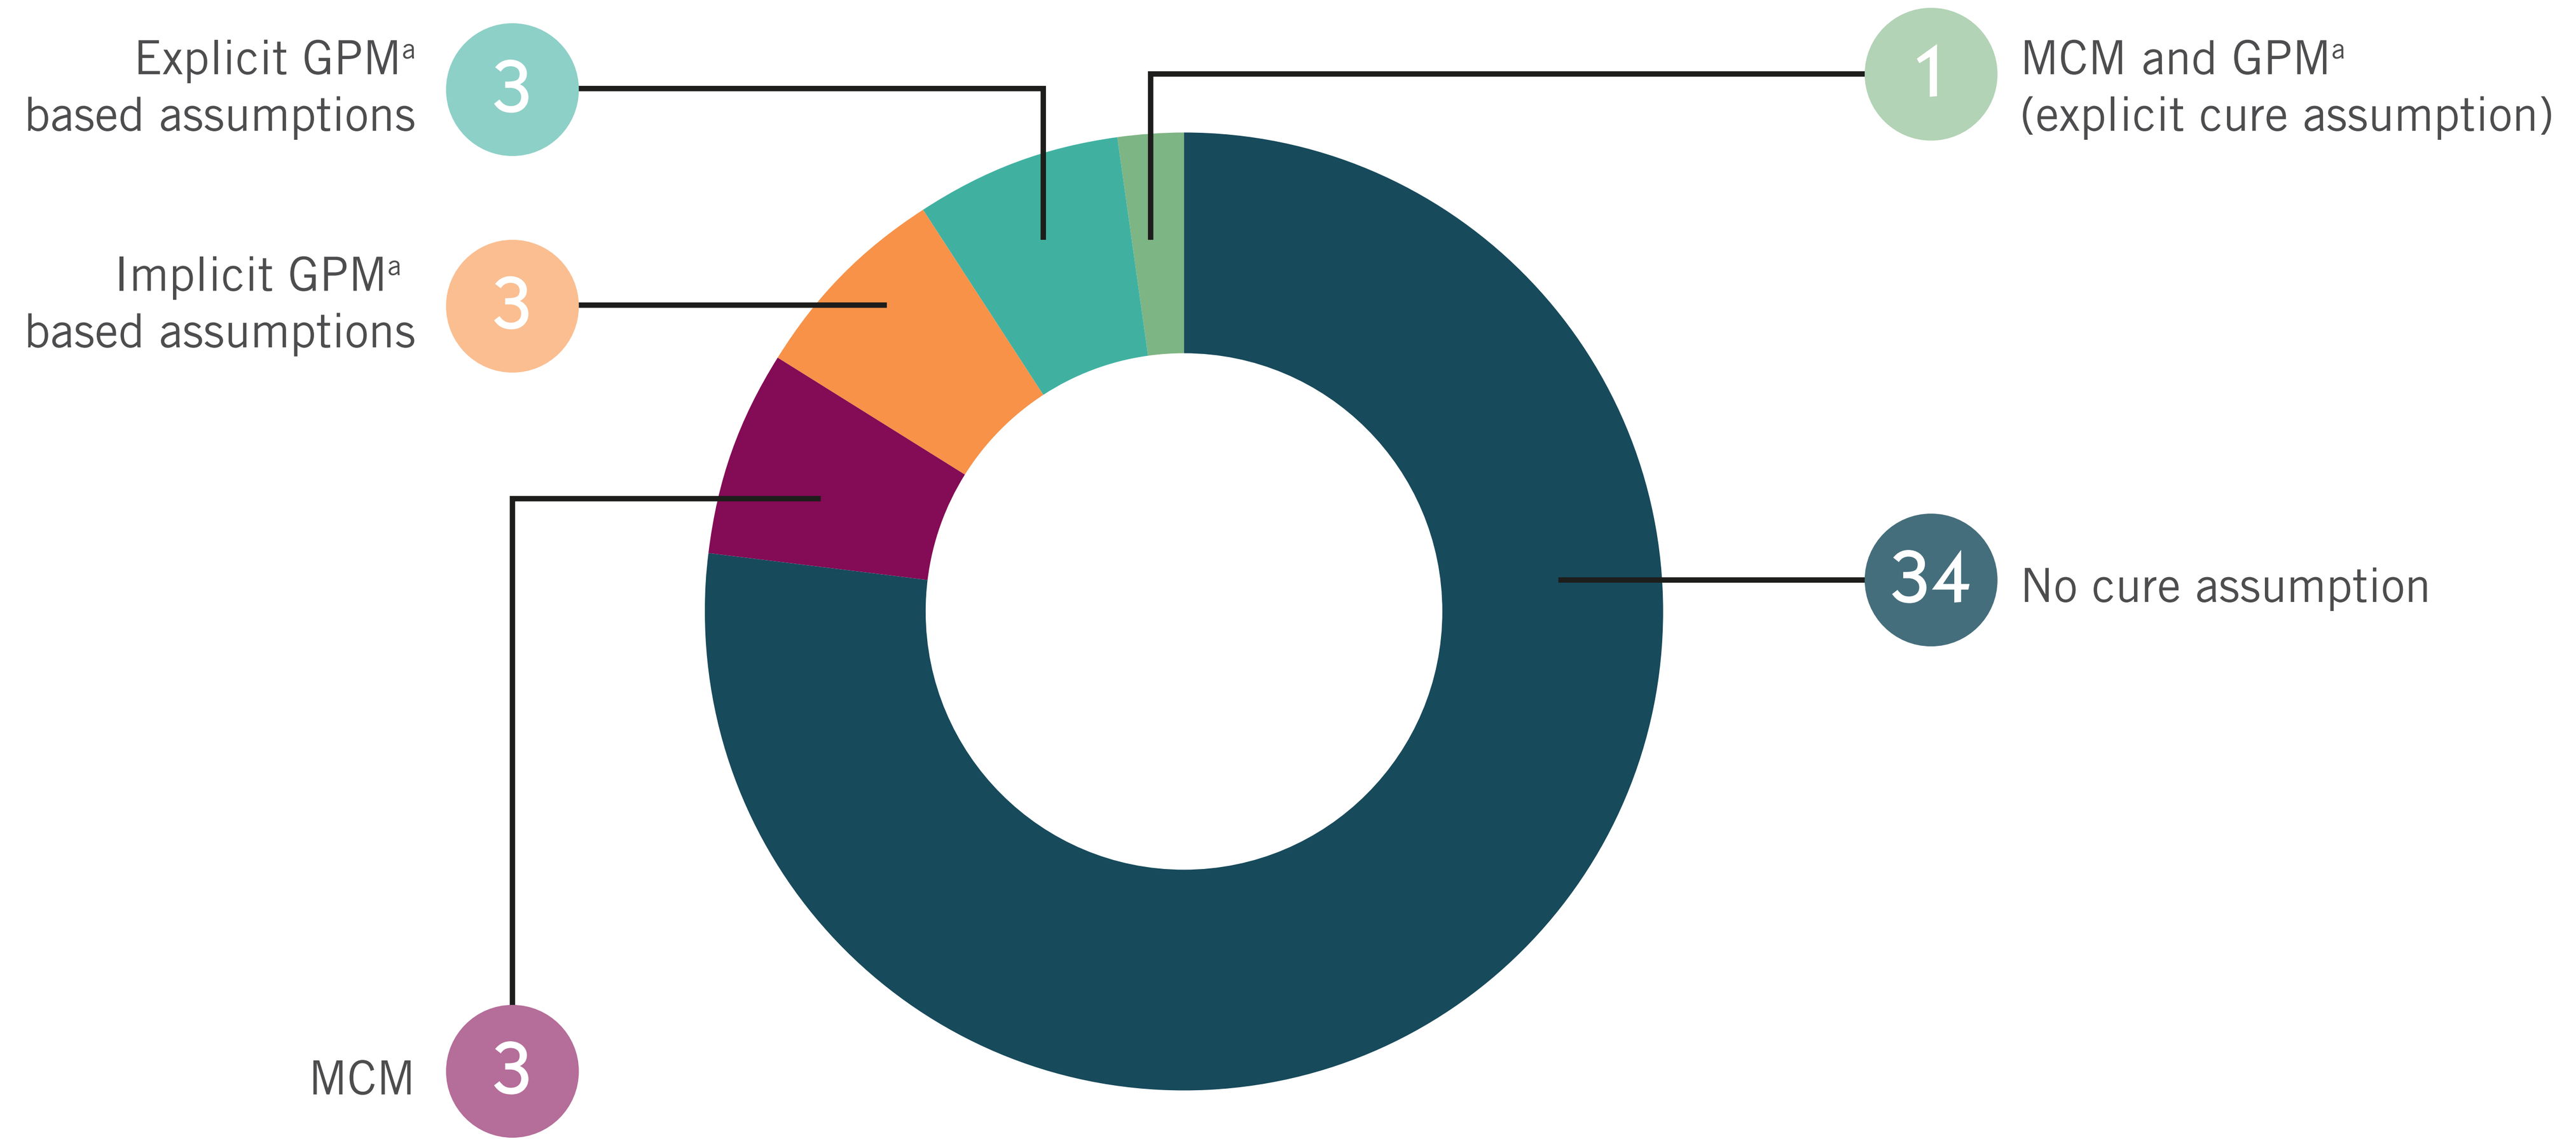
Explicit GPM[a] based assumptions
3
Implicit GPM[a] based assumptions
3
MCM
3
1
MCM and GPM[a] (explicit cure assumption)
34
No cure assumption

[a]Survival was informed by GPM after a certain timepoint for a proportion of patients.

- The modelled cure assumption was rejected in the majority (N=7) of appraisals **(Table 1)**. **Table 1** summarises the criticisms frequently associated with cure assumptions, which included limited data follow-up, the lack of an observed plateau, and the reliance on surrogate outcomes, such as progression-free survival. For MCMs, limited data follow-up can result in a wide range of possible cure fractions, introducing additional uncertainty.
- Appraisal Committees considered generalisable external data with long-term follow up, precedence from previous appraisals in the same indication and clinical expert opinion more robust justifications for cure versus short-term observed survival profiles.
- For non-MCM approaches, Appraisal Committees cited methodological concerns with the external choice of the proportion of patients who were considered cured and the timepoint beyond which patients are considered cured (N=2).
- In appraisals where a GPM-based assumption was solely used to model cure (N=6), evidence review groups recommended the consideration of an MCM to model cure in most cases (4/6).

CONCLUSIONS

Cure assumptions were included in a quarter of the most recent oncology appraisals to NICE but were rarely considered appropriate for decision-making.

The absence of long-term trial overall survival data, or the lack of an observed plateau, were frequent limitations associated with the plausibility of cure assumptions. Generalisable and robust external data or clinical expert opinion supporting the cure assumption have mitigated these limitations, such as in TA642.

In general, MCMs appeared to be considered more robust methods for modelling cure versus GPM-based models. This may be due to the calculation of a cure fraction based on available trial data in an MCM, rather than relying on the determination of proportion of patients cured using an external assumption.

References

1. Michielin O, Atkins MB, Koon HB, *et al.* Evolving impact of long-term survival results on metastatic melanoma treatment. Journal for ImmunoTherapy of Cancer 2020;8:e000948.

Acknowledgements

The authors thank Fay Angel, Costello Medical, for graphic design assistance. We also thank Matt Griffiths, Costello Medical, for his review and editorial assistance in the preparation of this poster, and for his guidance and support of this research.

Presented at Virtual ISPOR Europe 2021 | 30 November–3 December 2021

Copyright © Costello Medical Consulting Ltd