Treatment patterns, survival, and healthcare resource use of patients with advanced Merkel cell carcinoma in England from 2013 to 2018: results of a longitudinal observational cohort study

R. Palencia,[1] C. Knott,[2,3] S. H. Mahmoudpour,[1] F. Kolitsopoulos,[4] A. Amin,[5] P. Verpillat[1]

[1]the healthcare business of Merck KGaA, Darmstadt, Germany; [2]Health Data Insight CIC, Fulbourn, UK; [3]National Disease Registration Service, NHS Digital, Leeds, UK; [4]Pfizer, New York, NY, USA; [5]Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany

SCOPE

- **The objective of this study was to report patient characteristics, treatment patterns, survival rates, and healthcare resource use (HRU) among patients with advanced Merkel cell carcinoma (aMCC) in England**

CONCLUSIONS

- **This study provides important insights into historic treatment patterns, outcomes, and HRU among patients with newly diagnosed aMCC in England, identified from January 2013 to December 2018, with follow-up until December 2019**
- **Given the low percentage of patients who receive systemic treatment attributable to a dearth of approved and effective treatment options, a need exists to treat patients with aMCC with the most effective and tolerable frontline agents that can be administered by a broad range of treating physicians**
- **Data from this cohort suggest that the treatment landscape for patients with aMCC is being shifted to immune checkpoint inhibitor (ICI) therapy, incorporated into clinical guidelines by the National Institute for Care and Health Excellence in April 2018, which was the last year of inclusion of this study**
- **This study can serve as a useful benchmark for future real-world studies in aMCC, thereby enabling clinicians to better understand the utilization of and outcomes associated with systemic agents**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR Europe and the author of this poster

Correspondence: Roberto Palencia, **roberto.palencia@merckgroup.com**

BACKGROUND

- Merkel cell carcinoma (MCC) is a rare neuroendocrine, cutaneous malignancy characterized by firm, painless, rapidly enlarging, dome-shaped nodules on sun-exposed areas[1-3]
- MCC accounts for 29.3% of rare skin cancers and is known to have a poor prognosis[4]
- The incidence of MCC is low (0.13/100,000 population in Europe), and it occurs more commonly in men than women. The proportion of patients diagnosed with distant metastatic disease has been reported to be between 4% and 12% of MCC cases[5-9]
- The incidence of MCC is rising; rates in England increased from 0.1 per 100,000 in 1998 to 0.2 per 100,000 in 2008[4]
- Survival is poor for patients with MCC, with a disease-associated 5-year mortality rate of up to 46%; 5-year survival rates for metastatic MCC (mMCC) range from 0% to 16%[8-10]
- There are few approved treatment options for patients with mMCC;[11,12] chemotherapy has short-lived efficacy and survival in both treatment-naive and previously treated patients with mMCC[13-15]

METHODS

Design

- A descriptive, noninterventional study of a real-world cohort of aMCC patients based on patient-level data collected by the National Disease Registration Service in England

Inclusion criteria (at date of diagnosis)

- Resident of England aged ≥18 years
- ≥1 incident primary diagnosis (index) of aMCC (stage III/IV) between 1 January 2013 and 31 December 2018

Exclusion criteria

- No recorded date or stage of diagnosis; no recorded age at diagnosis
- Cases identified via death certificate only
- Receipt of a nonsponsor systemic therapy funded through the Cancer Drug Fund
- Participation in a clinical trial

Statistical analyses

- Patients were followed-up until the earliest of death, loss to follow-up, or end of study period (December 2019)
- Lines of therapy were derived via algorithms
- Descriptive statistics were used for baseline characteristics and treatment patterns
- Overall survival (OS) data were analysed from diagnosis and from the start of first-line (1L) systemic therapy using Kaplan-Meier survival curves

RESULTS

- There were 505 patients diagnosed with stage III/IV MCC in England between January 2013 and December 2018 inclusive (**Figure 1**)

Figure 1. Flow of patients

Patients diagnosed with MCC in England between January 2013 and December 2018 inclusive (N=1,911; 100.0%)

Patients excluded due to poor vital status data quality (n=3; 0.2%)

Patients excluded due to missing stage at diagnosis (n=602; 31.5%)

Patients excluded due to stage I or II at diagnosis (n=801; 41.9%)

Patients diagnosed with stage III/IV MCC in England between January 2013 and December 2018 inclusive (n=505; 26.4%)

MCC, Merkel cell carcinoma.

Comorbidities

- The most frequently occurring comorbidities documented during the baseline period (between 3 and 27 months prior to index date) were hypertension (n=174; 34.5%), type 2 diabetes without complications (n=56; 11.1%), and atrial fibrillation (n=48; 9.5%) (**Table 1**)

Table 1. Patient characteristics

	N=505
Age at diagnosis, years	
Mean (SD)	77.8 (10.3)
Median (Q1-Q3)	80.0 (71.0-86.0)
Sex, n (%)	
Male	298 (59.0)
Female	207 (41.0)
Stage at diagnosis, n (%)	
III	340 (67.3)
IV	165 (32.7)
Duration of follow-up from diagnosis, months	
Mean (SD)	22.8 (20.8)
Median (Q1-Q3)	15.9 (6.6-33.5)
Modified Deyo-Charlson comorbidity index score at baseline*†	
Mean (SD)	4.1 (1.8)
Median (Q1-Q3)	4.0 (3.0-5.0)

* The modified Deyo-Charlson score quantifies the burden of patient comorbidity from 17 prespecified medical conditions, including but not limited to myocardial infarction, dementia, liver disease, and metastatic disease other than the primary disease of interest; it also factors in age.

† Baseline was 3 and 27 months prior to index date (ie, date of diagnosis).

Q, quartile.

Treatment patterns

- In total, 345 (68.3%) patients with aMCC received 1 resection during the primary course of treatment, 311 (61.6%) received radiation therapy, and 123 (24.4%) had ≥1 derived lines of systemic therapy
- Of the 123 patients with derived lines of therapy, 15 (12.2%) moved to second-line therapy, with no patients reaching a third line of therapy during the study period (**Table 2**)
- The classes of drugs most commonly received by patients were platinum compounds (n=95; 77.2%) and other cytotoxic compounds (n=83; 67.5%) (**Table 2**)
- ICIs across all lines of therapy were received by 24.4% (n=30) of patients

Table 2. Patients with aMCC treated with systemic treatment classes by line of therapy

	First Line (n=123)		Second Line (n=15)	
	n	%	n	%
Platinum compounds	92	74.8	6	40.0
Other cytotoxic agents	81	65.9	9	60.0
Immune checkpoint inhibitors*	24	19.5	6	40.0
Antimetabolites	5	4.1	0	0.0
Monoclonal antibodies	3	2.4	0	0.0
Alkylating agents	2	1.6	5	33.3
Targeted therapies	2	1.6	0	0.0
Anthracyclines	1	0.8	5	33.3

Patients could have received multiple classes of drug within a single line; National Institute for Health and Care Excellence (NICE) recommendation for second-line immune checkpoint inhibitors were published in 2018; systemic therapies funded through the Cancer Drug Fund are excluded.

* Twenty-four patients (100%) with aMCC received first-line avelumab; 5 patients (83.3%) received second-line avelumab, and 1 (16.7%) received second-line treatment with pembrolizumab.

aMCC, advanced Merkel cell carcinoma.

OS

- In the overall population (N=505), there were 353 (69.9%) deaths following diagnosis, resulting in a median OS of 16.9 months (95% CI, 14.0-19.2 months) from diagnosis; the 12-month OS rate was 58.6% (95% CI, 54.2%-62.8%), and the 24-month OS rate was 41.1% (95% CI, 36.8%-45.5%) (**Figure 2**)

Figure 2. Kaplan-Meier curve of OS from diagnosis (N=505)

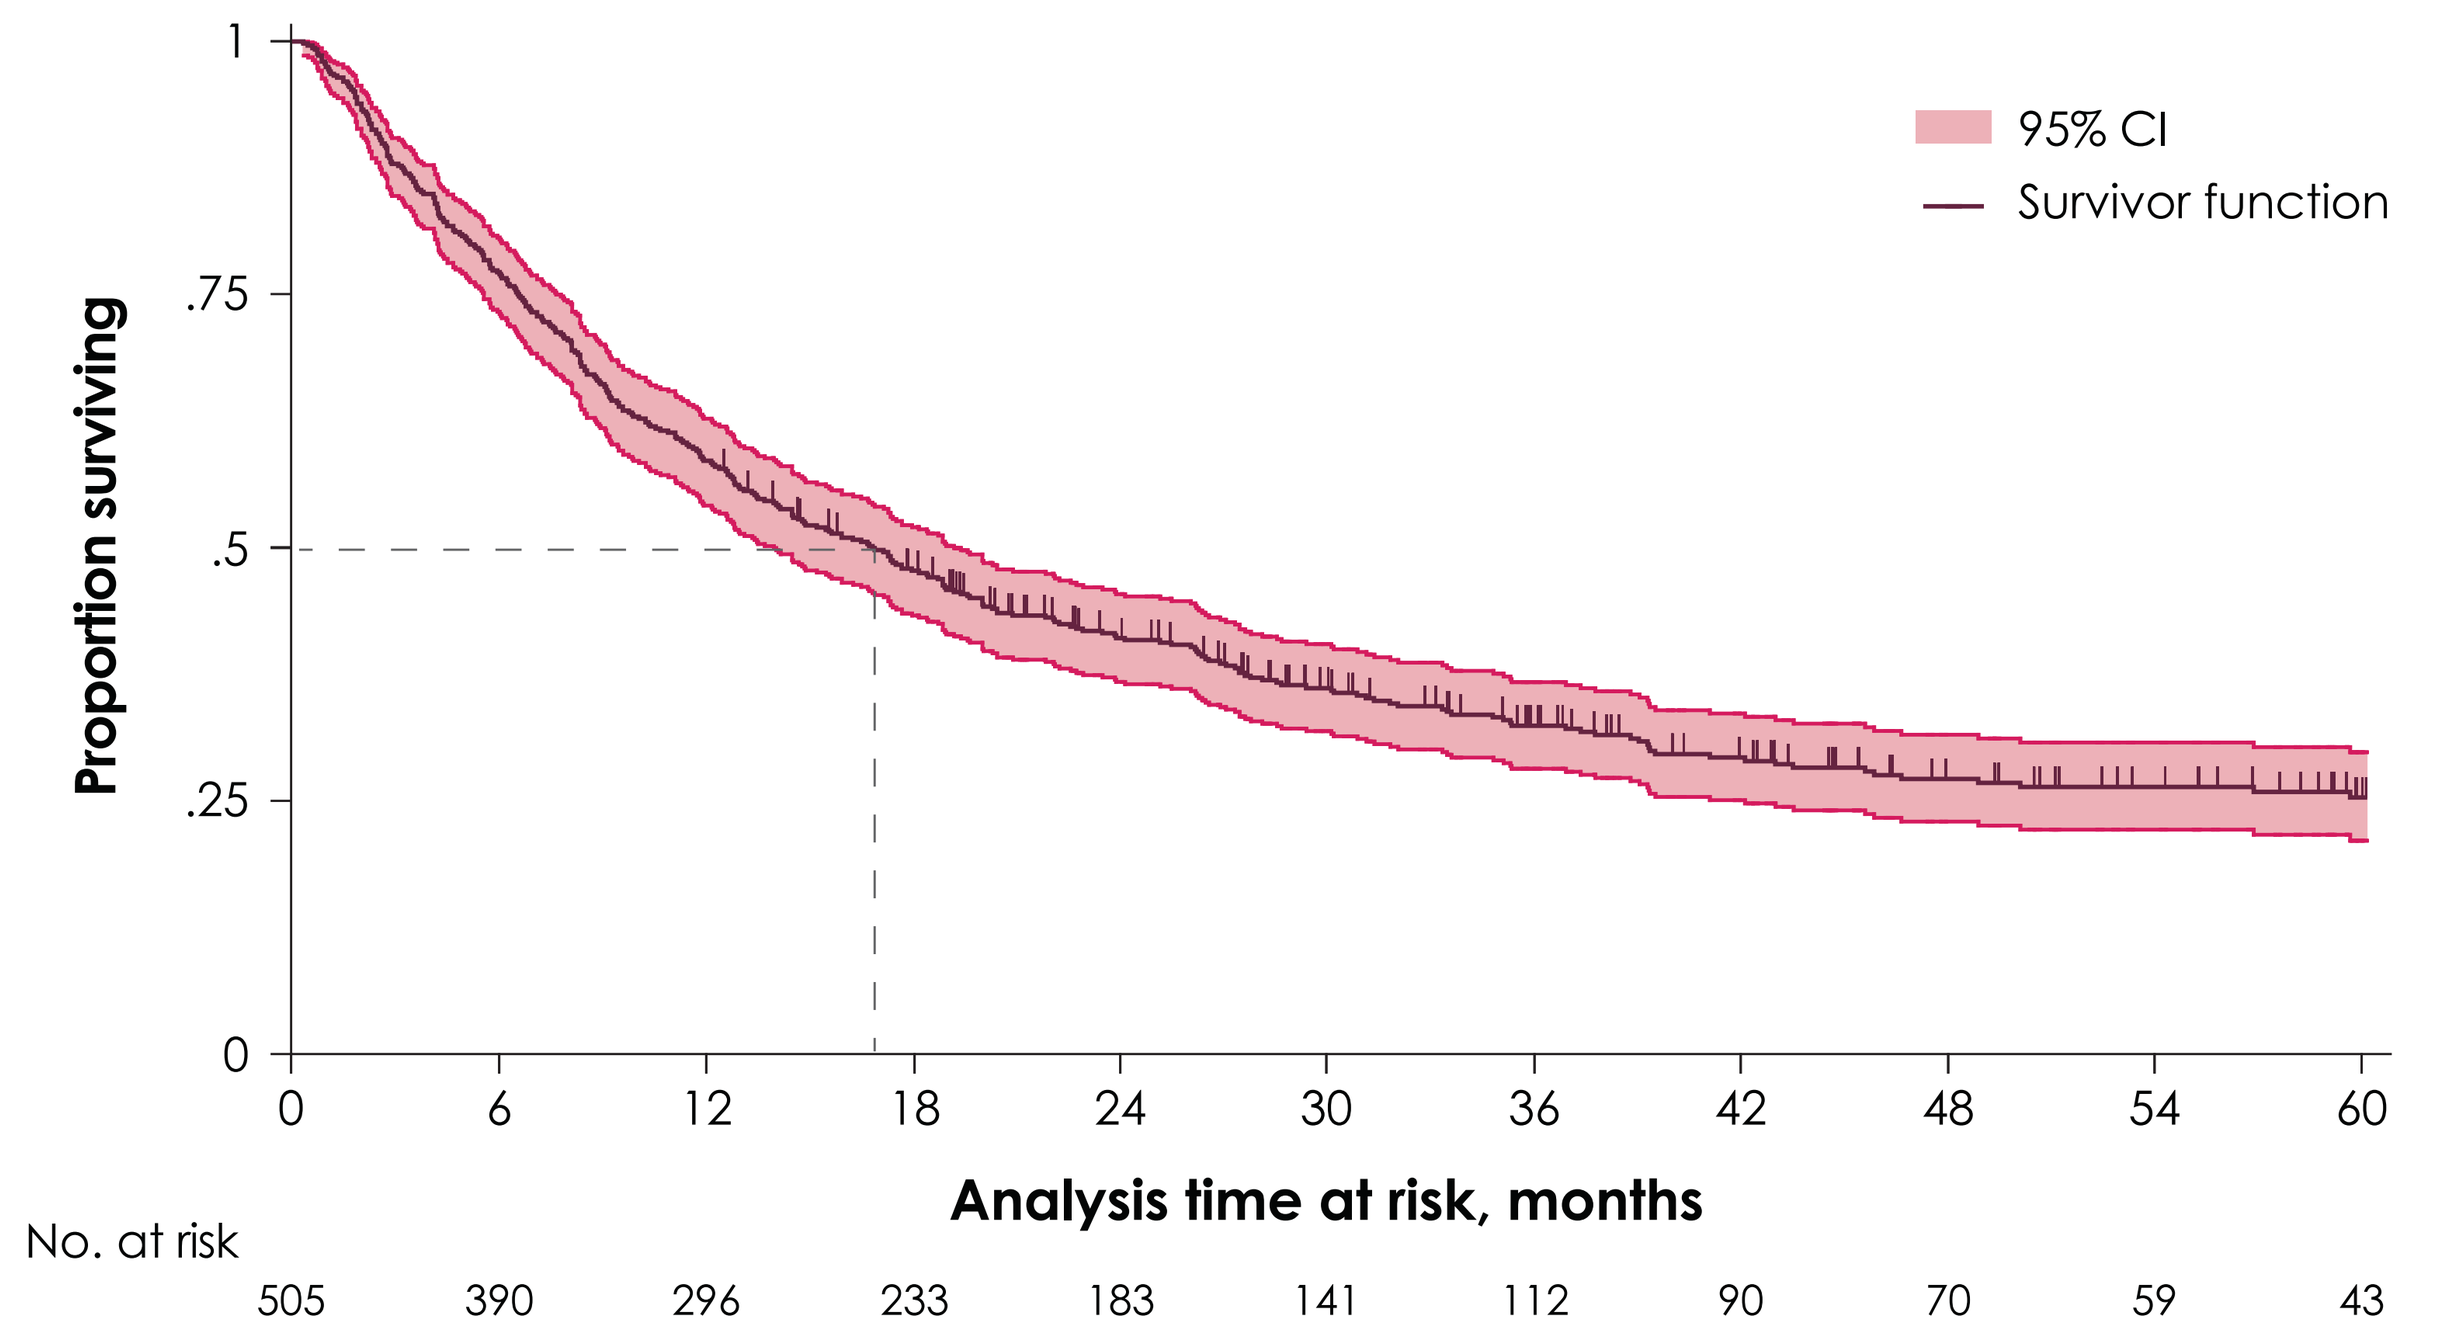
95% CI
Survivor function
Proportion surviving
1
.75
.5
.25
0
0 6 12 18 24 30 36 42 48 54 60
Analysis time at risk, months
No. at risk
505 390 296 233 183 141 112 90 70 59 43

OS, overall survival.

- From the initiation of 1L systemic therapy (N=123), there were 84 (68.3) deaths during follow-up, resulting in a median OS of 9.1 months (95% CI, 7.2-12.0 months); the 12-month OS rate was 41.6% (95% CI, 32.5%-50.4%), and the 24-month OS rate was 28.3% (95% CI, 20.1%-37.1%) (**Figure 3**)

Figure 3. Kaplan-Meier curve of OS from initiation of 1L systemic therapy (N=123)

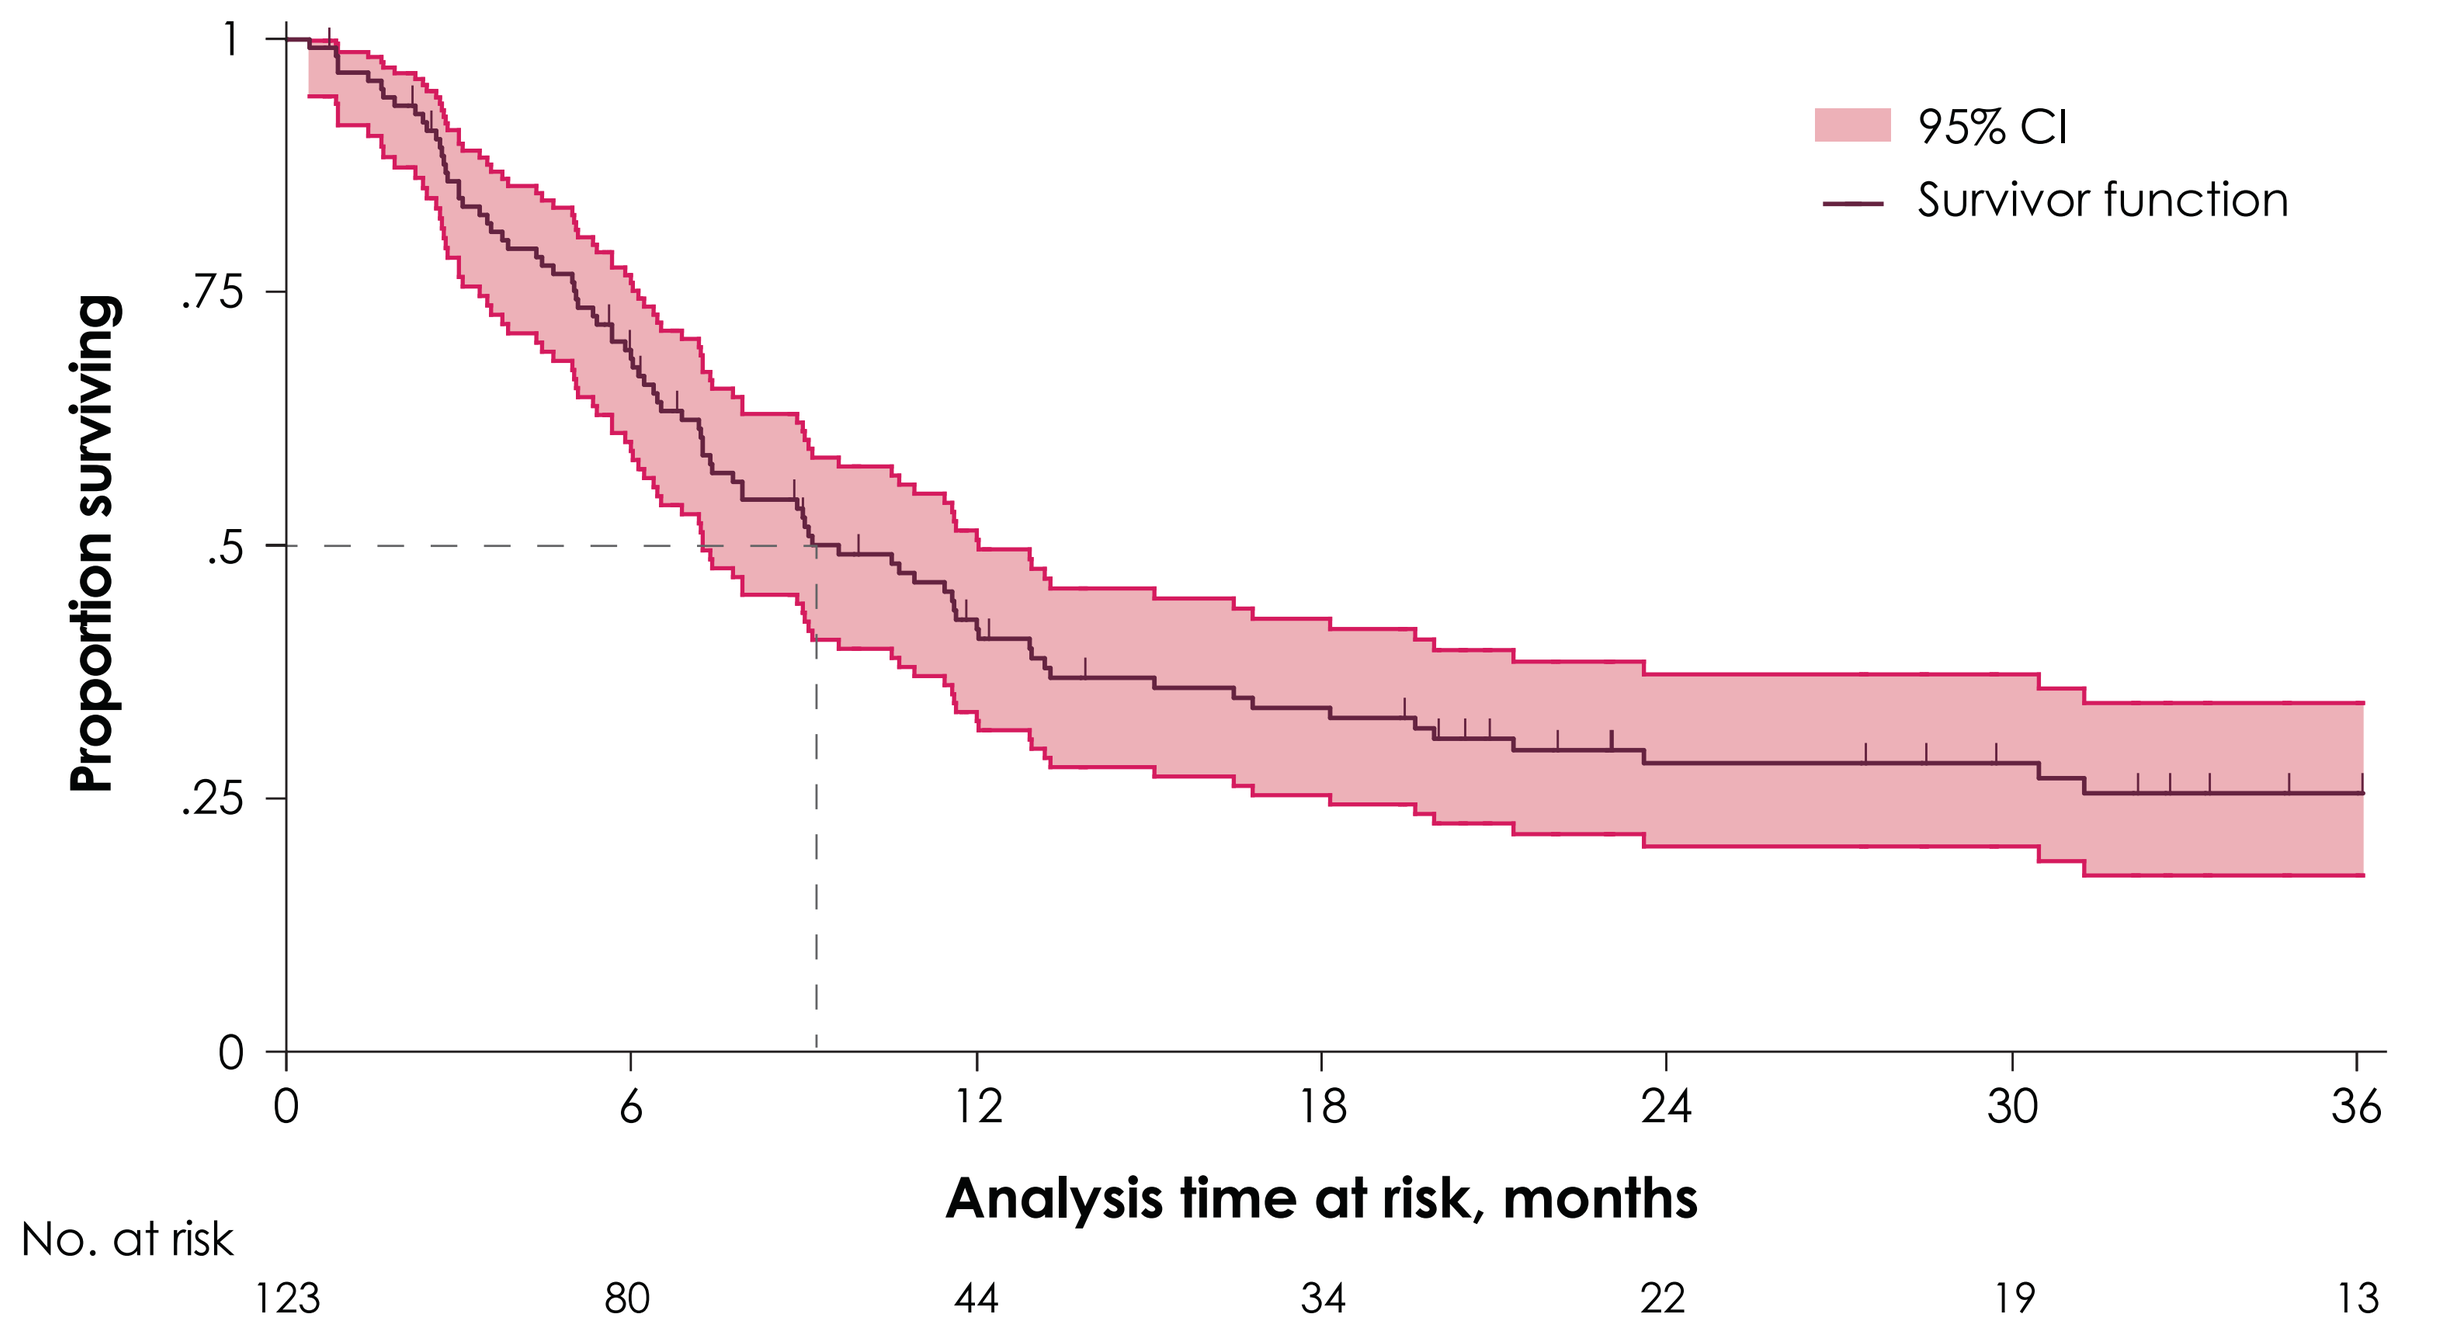
95% CI
Survivor function
Proportion surviving
1
.75
.5
.25
0
0 6 12 18 24 30 36
Analysis time at risk, months
No. at risk
123 80 44 34 22 19 13

1L, first-line; **OS**, overall survival.

Strengths

- The multiple linked datasets held by the National Cancer Registration and Analysis Service (NCRAS) provide national coverage of all people diagnosed with cancer in England
- The cancer registry benefits from mandatory data submission and robust validation procedures, maximizing completeness
- Standardized recording reduces the risk of misclassification
- Possibility of long-term follow-up and rich clinical detail

Limitations

- Lack of information on disease-specific outcomes, such as cancer recurrence and disease progression
- Systemic anticancer therapy dataset contains data for systemic therapy prescribed in secondary but not primary or private care settings
- Lines of systemic therapy were derived using an algorithm
- Incompleteness of data items such as *International Classification of Diseases, Tenth Revision* diagnosis codes and OPCS Classification of Interventions and Procedures version 4 procedure codes in hospital episode statistics
- Procedures undertaken exclusively in outpatient settings are likely to be missed

REFERENCES 1. Bichakjian CK, et al. J Natl Compr Canc Netw. 2014;12(3):410-24. **2.** Agelli M, Clegg LX. J Am Acad Dermatol. 2003;49(5):832-41. **3.** Smith VA, et al. Laryngoscope. 2012;122(6):1283-90. **4.** National Cancer Intelligence Network. Rare skin cancer in England. Accessed 16 September 2021. http://www.ncin.org.uk/publications/data_briefings/rareskincancer **5.** Schadendorf D, et al. Eur J Cancer. 2017;71:53-69. **6.** Allen PJ, et al. J Clin Oncol. 2005;23(10):2300-9. **7.** Albores-Saavedra J, et al. J Cutan Pathol. 2010;37(1):20-7. **8.** Lemos BD, et al. J Am Acad Dermatol. 2010;63(5):751-61. **9.** Kukko H, et al. Eur J Cancer. 2012;48(5):737-42. **10.** Kaae J, et al. J Natl Cancer Inst. 2010;102(11):793-801. **11.** Lebbe C, et al. Eur J Cancer. 2015;51(16):2396-403. **12.** NCCN Clinical Practice Guidelines. Merkel Cell Carcinoma. V2.2019 https://merkelcell.org/wp-content/uploads/2017/10/MCC_v.2.2019-2.pdf **13.** Becker J, et al. Ann Oncol. 2016;27(suppl 6):379-400. **14.** Becker JC, et al. Oncotarget. 2017;8(45):79731-41. **15** .Cowey CL, et al. Future Oncol. 2017;13(19):1699-710. **DISCLOSURES R. Palencia, S. H. Mahmoudpour, and P. Verpillat** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **C. Knott** is an employee of Health Data Insight CIC, which received funding from Merck KGaA and Pfizer. **F. Kolitsopoulos** is an employee of Pfizer, New York, NY, USA. **A. Amin** is an employee of Merck Serono Ltd., Feltham, UK; an affiliate of Merck KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS** The authors would like to thank Health Data Insight CIC for its analytical support and Public Health England (PHE) for providing access to the data. This work uses data that has been provided by patients and collected by the NHS as part of their care and support. The data are collated, maintained and quality assured by the National Disease Registration Service, which is part of NHS Digital. The data are collated and maintained, and quality is assured by the NCRAS, which is part of PHE. This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and is part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer.

Poster No. POSC139. Presented at Virtual ISPOR Europe 2021, 30 November - 3 December 2021.