Oncologists' preferences for first-line (1L) treatment of locally advanced/unresectable or metastatic urothelial carcinoma (aUC)

P. Grivas,[1] P. Veeranki,[2]* K. Chiu,[2] V. Pawar,[3] J. Chang,[4] M. Bharmal[3]

[1]University of Washington and Fred Hutchinson Cancer Research Center, Seattle Cancer Care Alliance, Seattle, WA, USA; [2]PRECISIONheor, Los Angeles, CA, USA; [3]EMD Serono, Billerica, MA, USA; [4]Pfizer, New York, NY, USA
*Affiliation at the time the analysis was conducted.

SCOPE

- **This is a cross-sectional survey study assessing oncologists' preferences for treating aUC using a discrete choice experiment (DCE)**

CONCLUSIONS

- **Overall survival (OS) was the most important attribute driving decision-making among US oncologists for selecting 1L treatment for patients with aUC**
- **Relative to other attributes, oncologists were indifferent between choices of treatment regimens administered once every 3 weeks and those administered with other frequencies, including those administered once per week or once every 2 weeks**
- **Oncologists preferred 1L immune checkpoint inhibitor (ICI) maintenance therapy over other treatment regimens for patients diagnosed with aUC**
 - **This is consistent with the category 1 recommendation for the JAVELIN Bladder regimen from both the National Comprehensive Cancer Network and European Society for Medical Oncology clinical practice guidelines for bladder cancer[1,2]**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR Europe and the author of this poster

Correspondence: Murtuza Bharmal, **murtuza.bharmal@emdserono.com**

BACKGROUND

- The treatment landscape for patients with aUC has rapidly evolved in recent years[3]
- Platinum-based chemotherapy regimens are still the standard of care in the 1L setting, but ICIs have expanded treatment options for these patients
- Avelumab, a PD-L1 inhibitor, was approved as 1L maintenance therapy for patients with aUC whose disease has not progressed with platinum-based chemotherapy[3]
- Evidence is limited on oncologists' preferences for different 1L pharmacological treatments for patients with aUC

METHODS

- A DCE was used to estimate oncologists' preferences; ISPOR best practices for a conjoint analysis were used to calibrate the attributes and levels of the DCE module[4-6]
 - **Figure 1** illustrates an example DCE choice set
- The survey was administered in March 2021
- Along with 12 discrete choice sets, 18 additional questions were asked to ascertain the oncologists' practice type, years in practice, size of hospital, health insurance, and type of patients
- The survey was completed by a convenience sample of 151 board-certified US oncologists who manage ≥1 patient with advanced bladder cancer per month; oncologists were identified from market research databases
- Random parameters logit was used to estimate weights of oncologists' preferences
- Preference weight was used to derive attribute importance and preference shares

METHODS

Figure 1. Example DCE choice set

	Treatment A	Treatment B
Treatment course		
Number and duration of medicines given	A B 4 → B Two different medicines (A and B) given for 4 months followed by 1 medicine (B) given until disease progression or unacceptable toxicity	A 4 → B One medicine (A) given for 4 months followed by a different medicine (B) given until disease progression or unacceptable toxicity
Likelihood of experiencing a grade 3 or 4 adverse event	90% chance of side effects → 15% chance of side effects	75% chance of side effects → 15% chance of side effects
Frequency of administration	Once every 3 weeks 3 weeks	Once every 2 weeks 2 weeks
Survival	16 months	24 months
Which treatment would you recommend?	○	○

RESULTS

- Most of the oncologists were men (72%), White (56%), in practice >10 years (81%), and managed ≥5 aUC patients per month (92%) (**Table 1**; **Table 2**)
- OS was the most important attribute, contributing to 74.3% of the decision for choice of treatment regimen (**Figure 2**)
 - 19.7% of the decision was determined by regimen type or grade 3 or 4 treatment-related adverse events
 - Frequency of administration was the least important attribute, contributing to 6% of the decision for choice of treatment regimen
- Among 1L treatments for aUC, 1L ICI maintenance therapy had the highest preference (73%), followed by 1L ICI monotherapy (19%), 1L ICI combination therapy (7%), and 1L chemotherapy alone (1%) (**Figure 3**)

Figure 2. Oncologists' preference weights for attributes

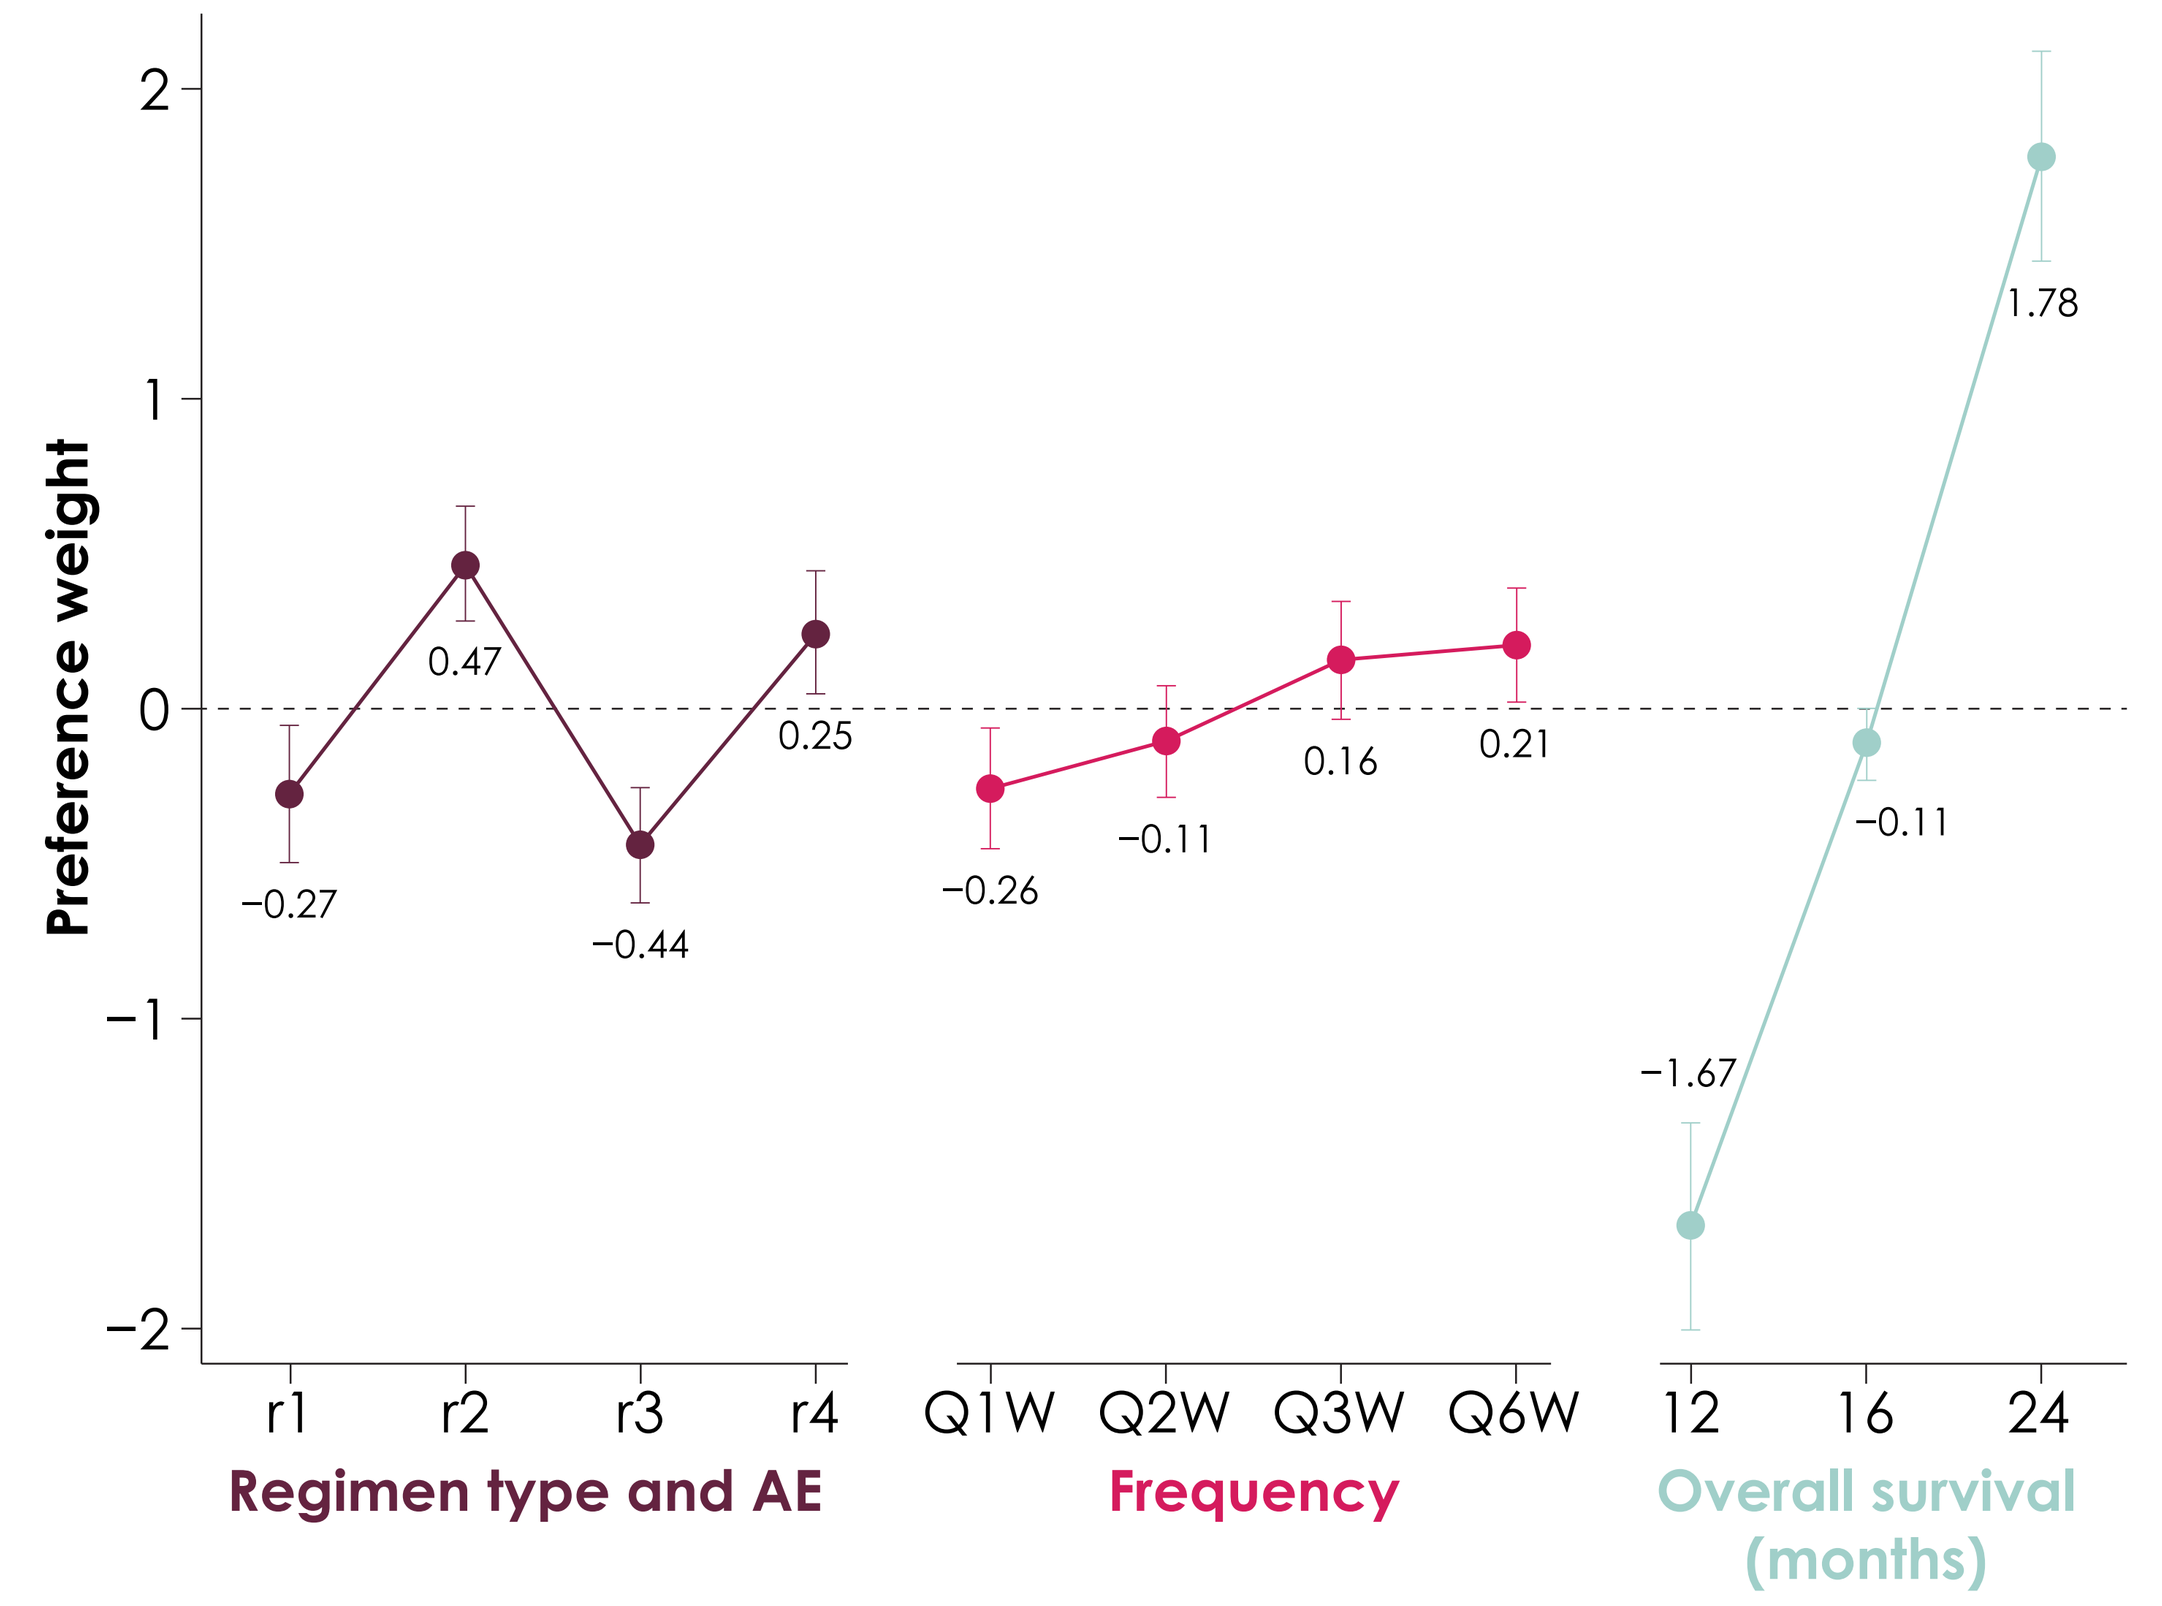

Preference weight
2
1
0
−1
−2
0.47
0.25
−0.27
−0.44
−0.26
−0.11
0.16
0.21
1.78
−0.11
−1.67
r1 r2 r3 r4
Regimen type and AE
Q1W Q2W Q3W Q6W
Frequency
12 16 24
Overall survival (months)

AE, adverse event; **Q1W**, every week; **Q2W**, every 2 weeks; **Q3W**, every 3 weeks; **Q4W**, every 4 weeks; **r1**, 1 medication with moderate AE; **r2**, 1 medication with low AE; **r3**, 2 medications with high AE followed by 1 medication with moderate AE; **r4**, 1 medication with moderate AE for 4 months followed by 1 medication with low AE.

Table 1. Oncologist demographics

Demographic characteristics	N=151
Age, mean ± SD	49.9±10.2
Sex, n (%)	
Male	109 (72.2)
Female	31 (20.5)
Not reported	11 (7.3)
Race, n (%)	
White	85 (56.3)
African American	4 (2.6)
Asian	37 (24.5)
Other	6 (4.0)
Not reported	19 (12.6)

Figure 3. Oncologists' preference for different 1L treatments

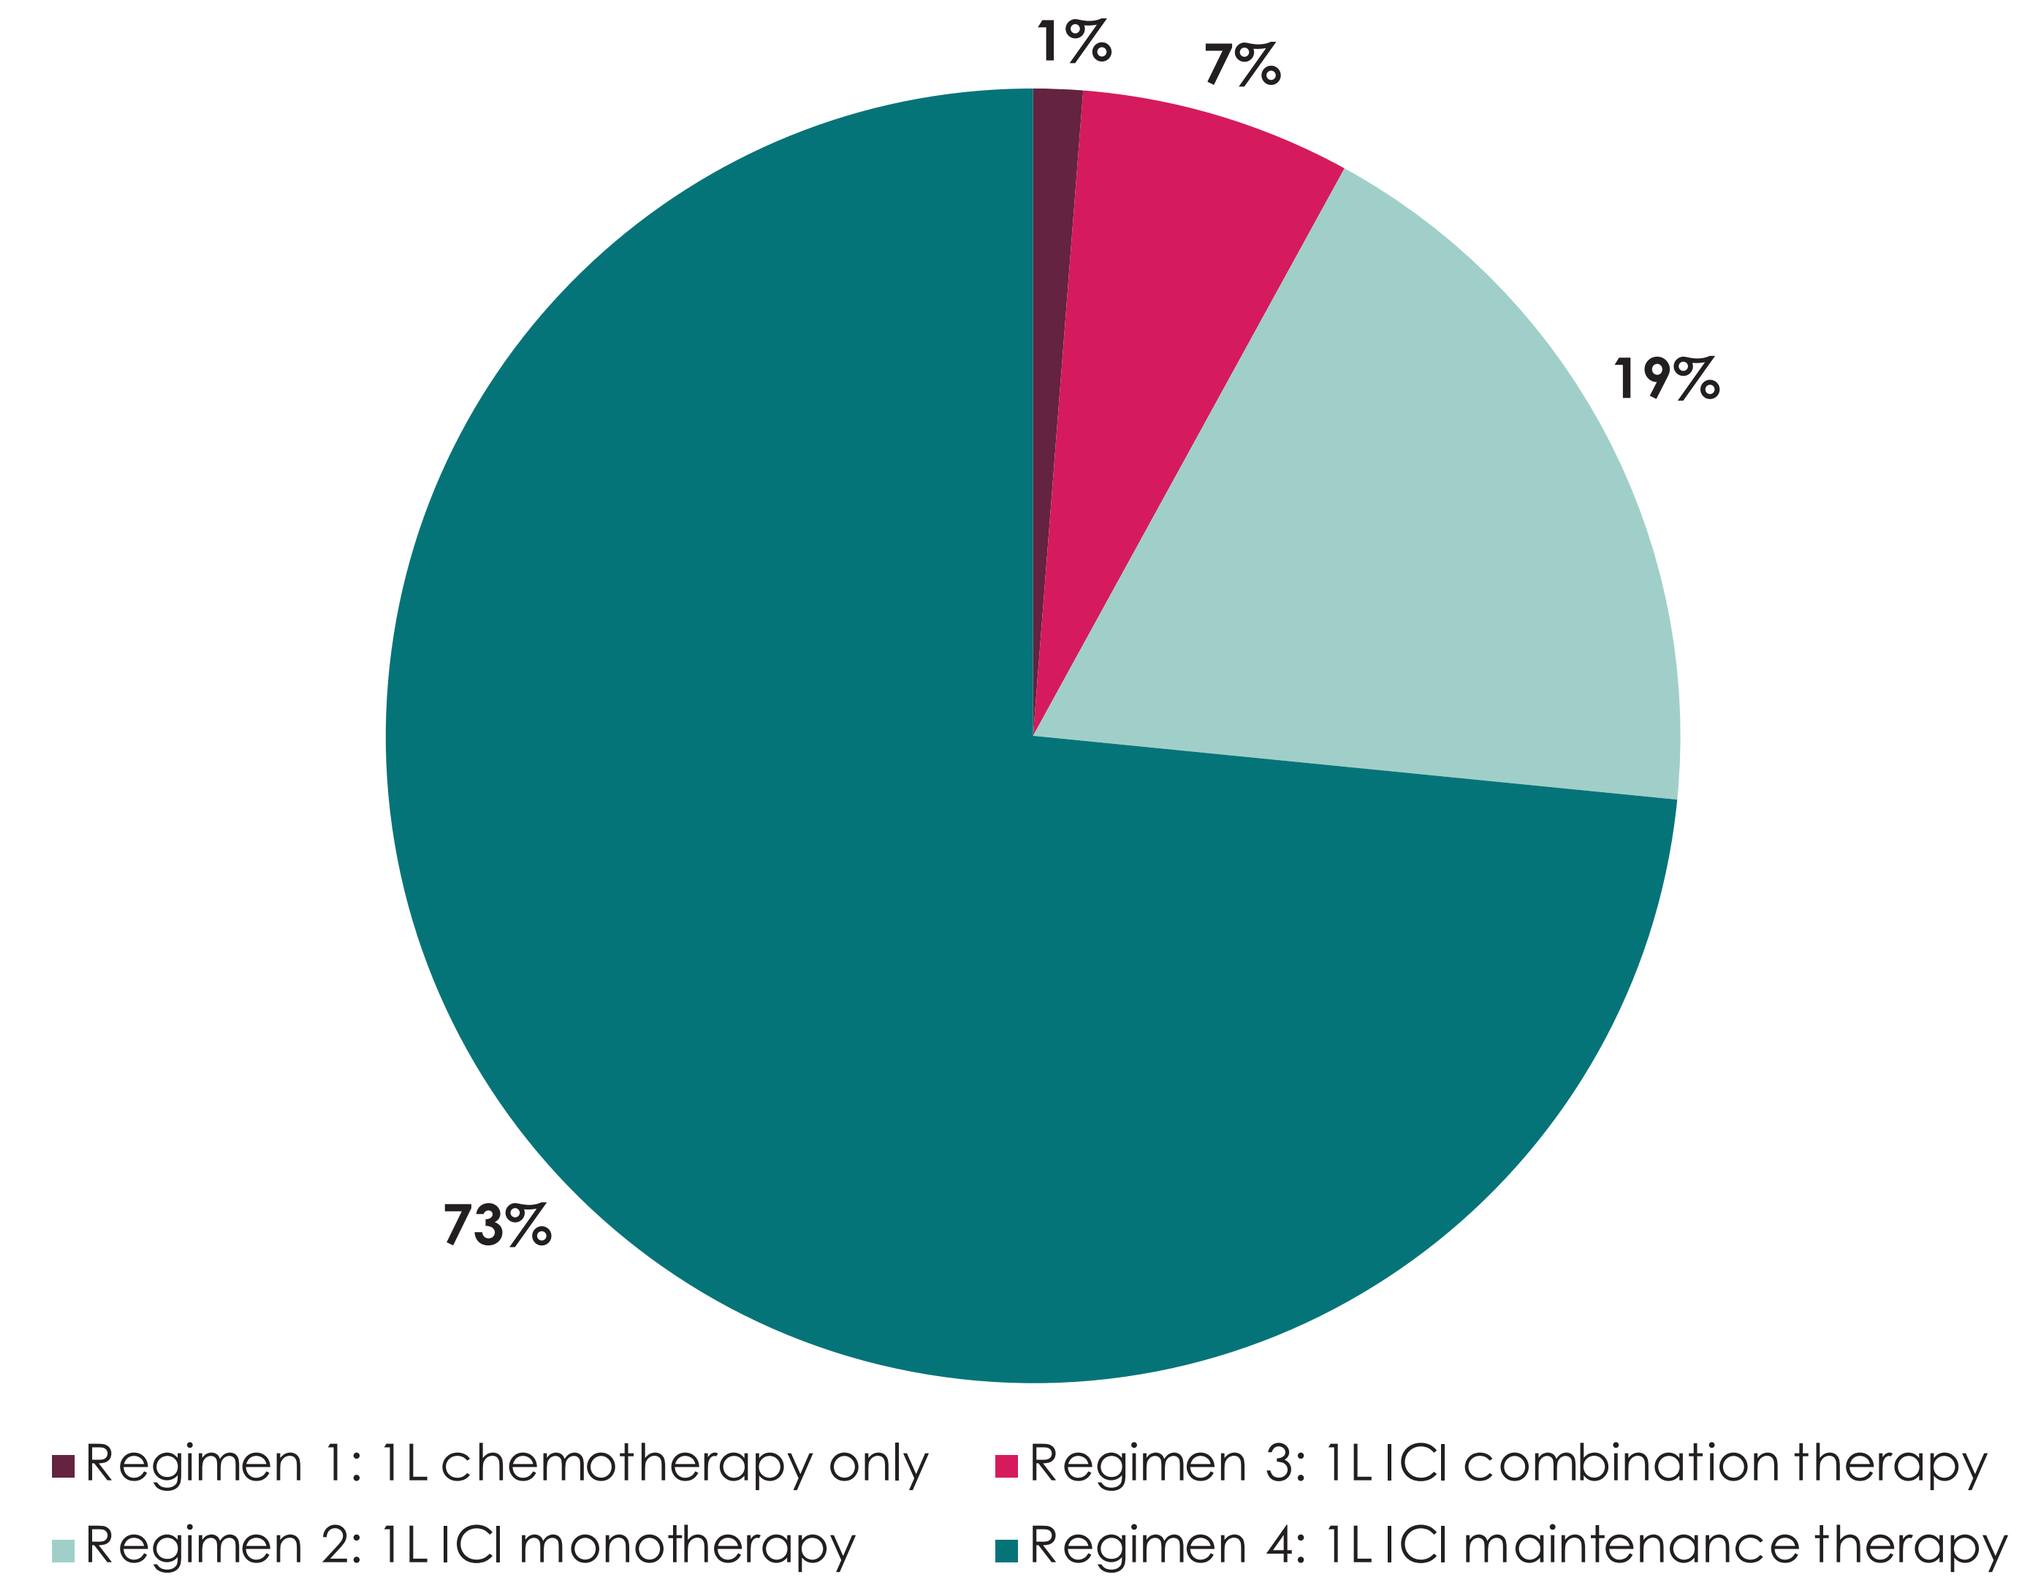

1%
7%
19%
73%
Regimen 1: 1L chemotherapy only
Regimen 3: 1L ICI combination therapy
Regimen 2: 1L ICI monotherapy
Regimen 4: 1L ICI maintenance therapy

1L, first-line; **ICI**, immune checkpoint inhibitor.

Table 2. Practice-related characteristics

Practice-related characteristic	N=151
Primary specialty, n (%)	
Internal medicine	3 (2.0)
Oncology	147 (97.4)
Radiology	1 (0.7)
Practice location, n (%)	
Solo practice	14 (9.3)
Partnership/group practice	35 (23.2)
Government, university, or comprehensive cancer center	41 (27.2)
Specialty group practice	43 (28.5)
Hospital-based practice	16 (10.6)
100-499 beds	12 (7.9)
≥500 beds	4 (2.6)
Not reported	2 (1.3)
Years in practice, n (%)	
0-10	28 (18.5)
11-20	80 (53.0)
>20	41 (27.2)
Not reported	2 (1.3)

Limitations

- This study used a convenience sample; therefore, study findings cannot be generalized to all US oncologists
- This study tested a limited number of treatment attributes
 - All DCE studies are affected by this limitation, as survey tractability is important, and a balance needs to be achieved in testing the most important attributes and pragmatic length of the survey

REFERENCES 1. NCCN Clinical Practice Guidelines. Bladder Cancer. V5.2021. Accessed 25 October 2021. https://www.nccn.org/professionals/physician_gls/pdf/bladder.pdf. **2.** European Society for Medical Oncology. e-Update – bladder cancer treatment recommendations. Accessed 25 October 2021. https://www.esmo.org/guidelines/genitourinary-cancers/bladder-cancer/eupdate-bladder-cancer-treatment-recommendations4. **3.** Powles T, et al. N Engl J Med. 2020;383(13):1218-1230. **4.** Bridges JFP, et al. Value Health. 2011;14(4):403-413. **5.** Reed Johnson F, et al. Value Health. 2013;16(1):3-13. **6.** Hauber AB, et al. Value Health. 2016;19(4):300-315.

DISCLOSURES P Grivas has received honoraria from AstraZeneca, BMS, Exelixis, Incyte, Ipsen, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Seattle Genetics, the healthcare business of Merck KGaA, Darmstadt, Germany, Astellas Pharma, Johnson & Johnson, Eisai, and Roche; has served in a consulting or advisory role at AstraZeneca, BMS, Exelixis, Incyte, Ipsen, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Seattle Genetics, the healthcare business of Merck KGaA, Darmstadt, Germany, Astellas Pharma, Johnson & Johnson, Eisai, and Roche; has received research funding from AstraZeneca, BMS, Exelixis, Ipsen, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Seattle Genetics, the healthcare business of Merck KGaA, Darmstadt, Germany, Astellas Pharma, Johnson & Johnson, Eisai, and Roche; and has received travel and accommodations expenses from Roche, Pfizer, Merck & Co., Kenilworth, NJ, AstraZeneca, and Ipsen. **P Veeranki** and **K Chiu** are employees of PRECISIONheor, which received funding from the healthcare business of Merck KGaA, Darmstadt, Germany to conduct the study. **V Pawar** and **M Bharmal** are employees of EMD Serono, Billerica, MA, USA. **J Chang** is an employee of Pfizer. **ACKNOWLEDGMENTS** This study was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) as part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer.

Poster No. POSB243. Presented at Virtual ISPOR Europe 2021, 30 November - 3 December 2021.