Approaches to immune checkpoint inhibitor first-line maintenance therapy in unresectable locally advanced or metastatic urothelial cancer in France, Germany, Italy, Spain, and the UK (the Eu5)

N. Milloy,[1] M. Berry,[1] M. Kostikas,[1] S. Li,[2] M. Kearney,[3] M. Bharmal,[4] N. Costa,[5] J. Chang[2]

[1]Adelphi Real World, Bollington, UK; [2]Pfizer, New York, NY, USA; [3]the healthcare business of Merck KGaA, Darmstadt, Germany; [4]EMD Serono, Billerica, MA, USA; [5]Pfizer, Porto Salvo, Portugal

SCOPE

- **This point-in-time survey was conducted in the Eu5 from November 2020 through April 2021**
- **In January 2021, avelumab was approved in Europe as first-line (1L) maintenance therapy (MT) for patients with unresectable locally advanced (LA) or metastatic urothelial carcinoma (mUC) that has not progressed with 1L platinum-containing chemotherapy**
- **The aim of this study was to assess how European physicians might incorporate this novel treatment approach into clinical practice**

CONCLUSIONS

- **Despite being approved only shortly before the completion of study data collection, most Eu5 physicians surveyed were familiar with avelumab as 1L-MT, and the majority indicated that they would use it in eligible patients after 4-6 cycles of chemotherapy and a period of 4-6 weeks between end of chemotherapy and start of 1L-MT**
- **These are important data for European health technology agencies and payers to consider as they assess reimbursement for avelumab**
- **This research suggests that a substantial number of clinicians in the Eu5 would consider this treatment for eligible 1L patients with LA/mUC**
- **Further real-world research is needed to examine how physicians' perceptions and treatment patterns will evolve with newer guideline recommendations and to examine the growing use and experience with avelumab as 1L-MT for patients with LA/mUC**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR Europe and the author of this poster

Correspondence: Neil Milloy, **neil.milloy@adelphigroup.com**

BACKGROUND

- UC is the fourth most common tumour type in developed countries[1]
- The prognosis of locally advanced (T3/T4 or N1) and metastatic disease is poor, with few approved therapeutic options[2]
- Systemic chemotherapy (cisplatin- or carboplatin-based combination) is the standard approach for initial treatment of patients with inoperable LA/mUC[3,4]
- Avelumab was approved by the European Medicines Agency in January 2021 as 1L-MT for patients with LA/mUC that has not progressed with 1L platinum-containing chemotherapy[5]

METHODS

- Real-world data were drawn from the Adelphi mUC Disease-Specific Programme™, a point-in-time physician survey of oncologists/urologists conducted in November 2020-April 2021 across the Eu5
- Physicians completed questionnaires capturing data points that included their demographics, workload, clinical practice, and perspectives on the use of avelumab for 1L-MT
- Descriptive analyses were conducted
- Physician inclusion criteria:
 - Personally responsible for treatment and management of patients with mUC*
 - Spends ≥50% of time in management of patients
 - Sees ≥8 patients with mUC per month

*The inclusion criteria for physicians to participate in the survey included the management of patients with mUC only. Nonetheless, some questions referred to the management of both patients with LA and patients with mUC.

RESULTS

- 232 physicians participated in the study, with oncologists making up the majority of responders, ranging from 66% (Germany) to 100% (Italy) (**Table 1**)
- Physicians saw the greatest proportion of their patients in public hospitals in Spain, France, and Italy; comprehensive cancer centers in the UK; and public office-based practices in Germany (**Table 1**)
- On average, physicians estimated that they would consider chemotherapy-containing regimens at 1L for 73% of their patients, followed by immune checkpoint inhibitor (ICI) monotherapy (17%) and ICI in combination (7%) (**Figure 1**)
- Among the 232 participating physicians, knowledge of switch maintenance was generally high across the Eu5 (79%), with the highest familiarity being in France (86%) and the UK (82%), followed by Spain (78%), Germany (74%), and Italy (73%)
 - Of the physicians who were familiar with 1L-MT (Eu5 n=183), 78% would consider treating eligible patients with 1L-MT when the treatment is available (**Figure 2**)
- Of the physicians who were familiar with 1L-MT, 22% would not consider any of their patients for treatment with ICIs as 1L-MT whereas 9% would consider >70% of patients (**Figure 3**)

Figure 1. Percentage of patients who physicians typically consider for each regimen at 1L*

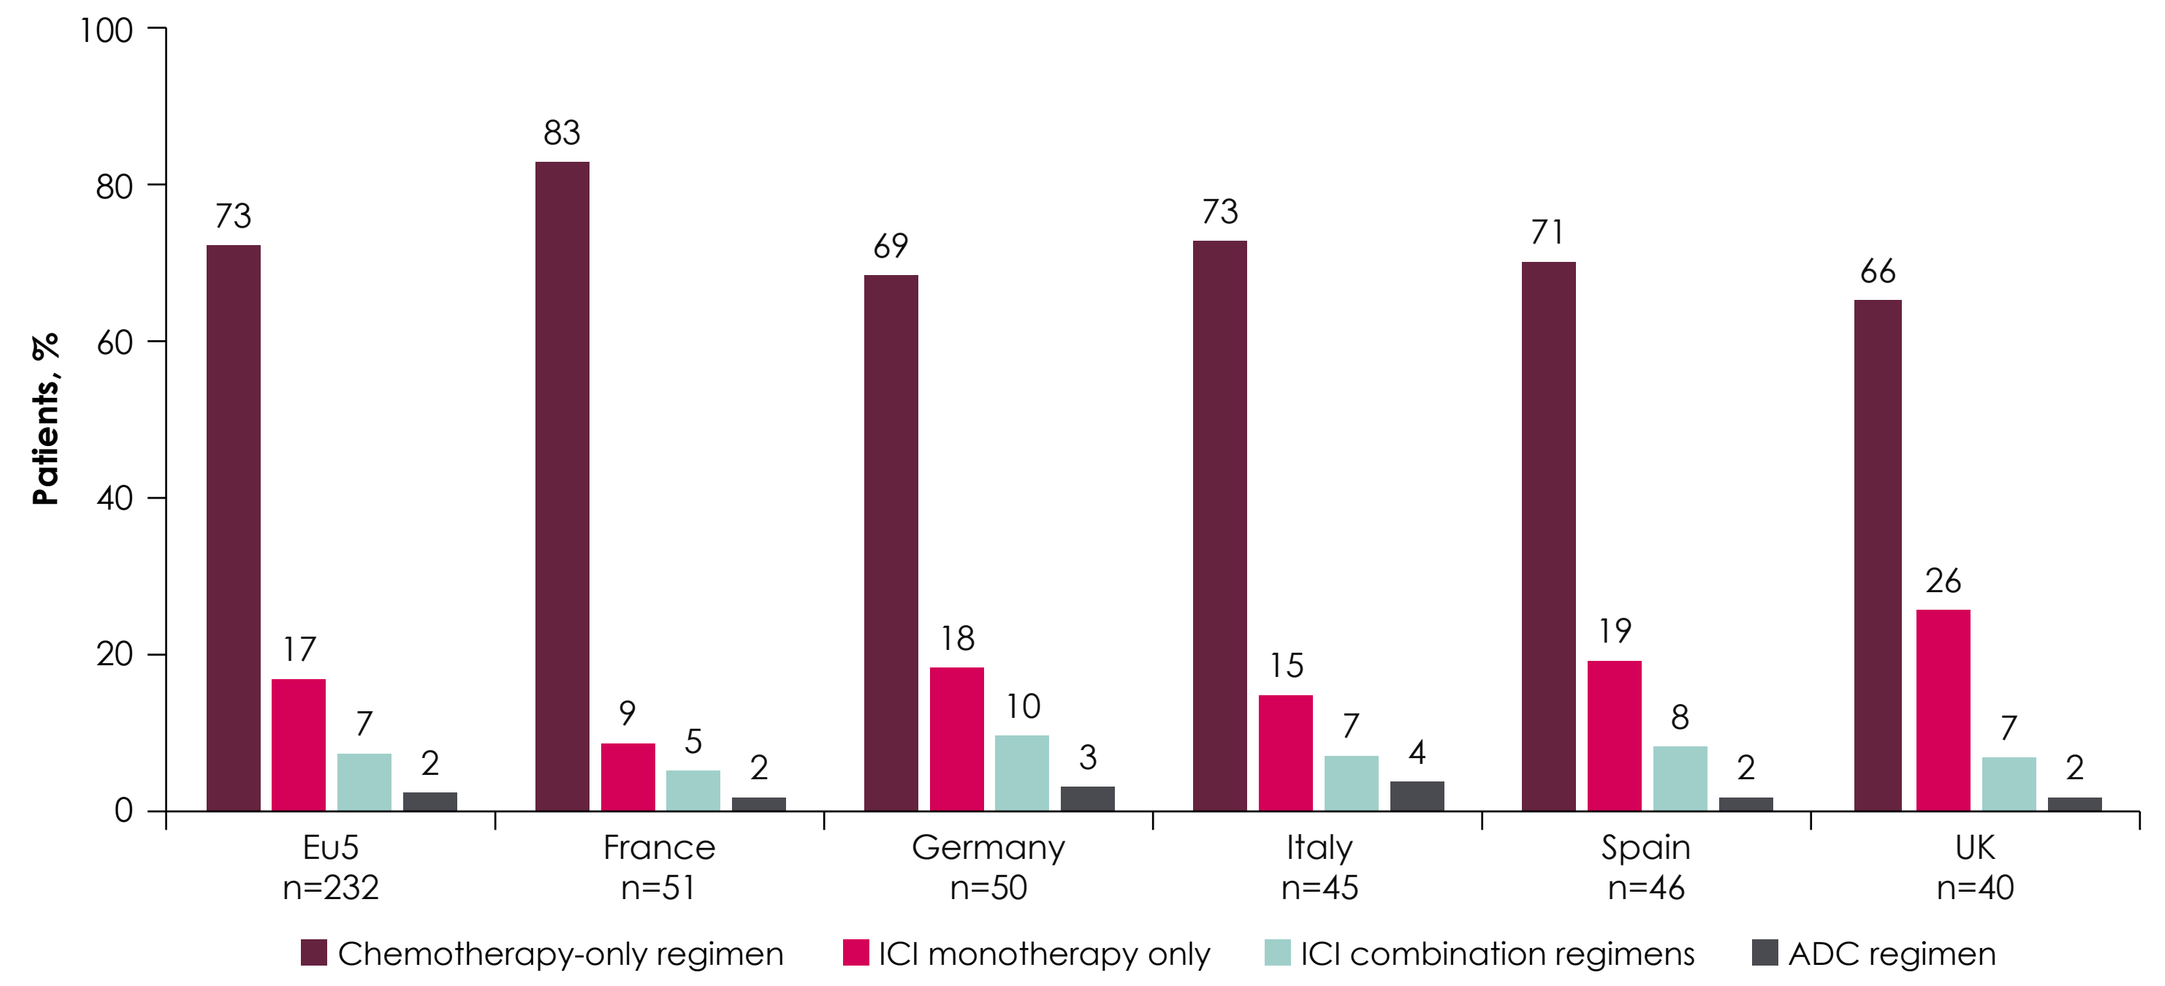
Patients, %
100
80
60
40
20
0
Eu5 n=232
France n=51
Germany n=50
Italy n=45
Spain n=46
UK n=40
73 17 7 2
83 9 5 2
69 18 10 3
73 15 7 4
71 19 8 2
66 26 7 2
Chemotherapy-only regimen
ICI monotherapy only
ICI combination regimens
ADC regimen

1L, first-line; **ADC**, antibody-drug conjugate; **Eu5**, France, Germany, Italy, Spain, and UK together; **ICI**, immune checkpoint inhibitor.
*Chemotherapy regimen includes cisplatin, carboplatin, gemcitabine, methotrexate, vinblastine, doxorubicin, etc. ICI monotherapy includes atezolizumab, avelumab, durvalumab, nivolumab, and pembrolizumab. ICI combination includes ICI + ICI or ICI + chemotherapy. ADC includes enfortumab vedotin. These drug choices reflect the treatments physicians would consider prescribing, not what they actually prescribed. Nonapproved agents at the time of data collection, such as enfortumab vedotin and ICI-chemotherapy combination regimens, have been included.

Figure 2. Familiarity with and intended use of 1L-MT

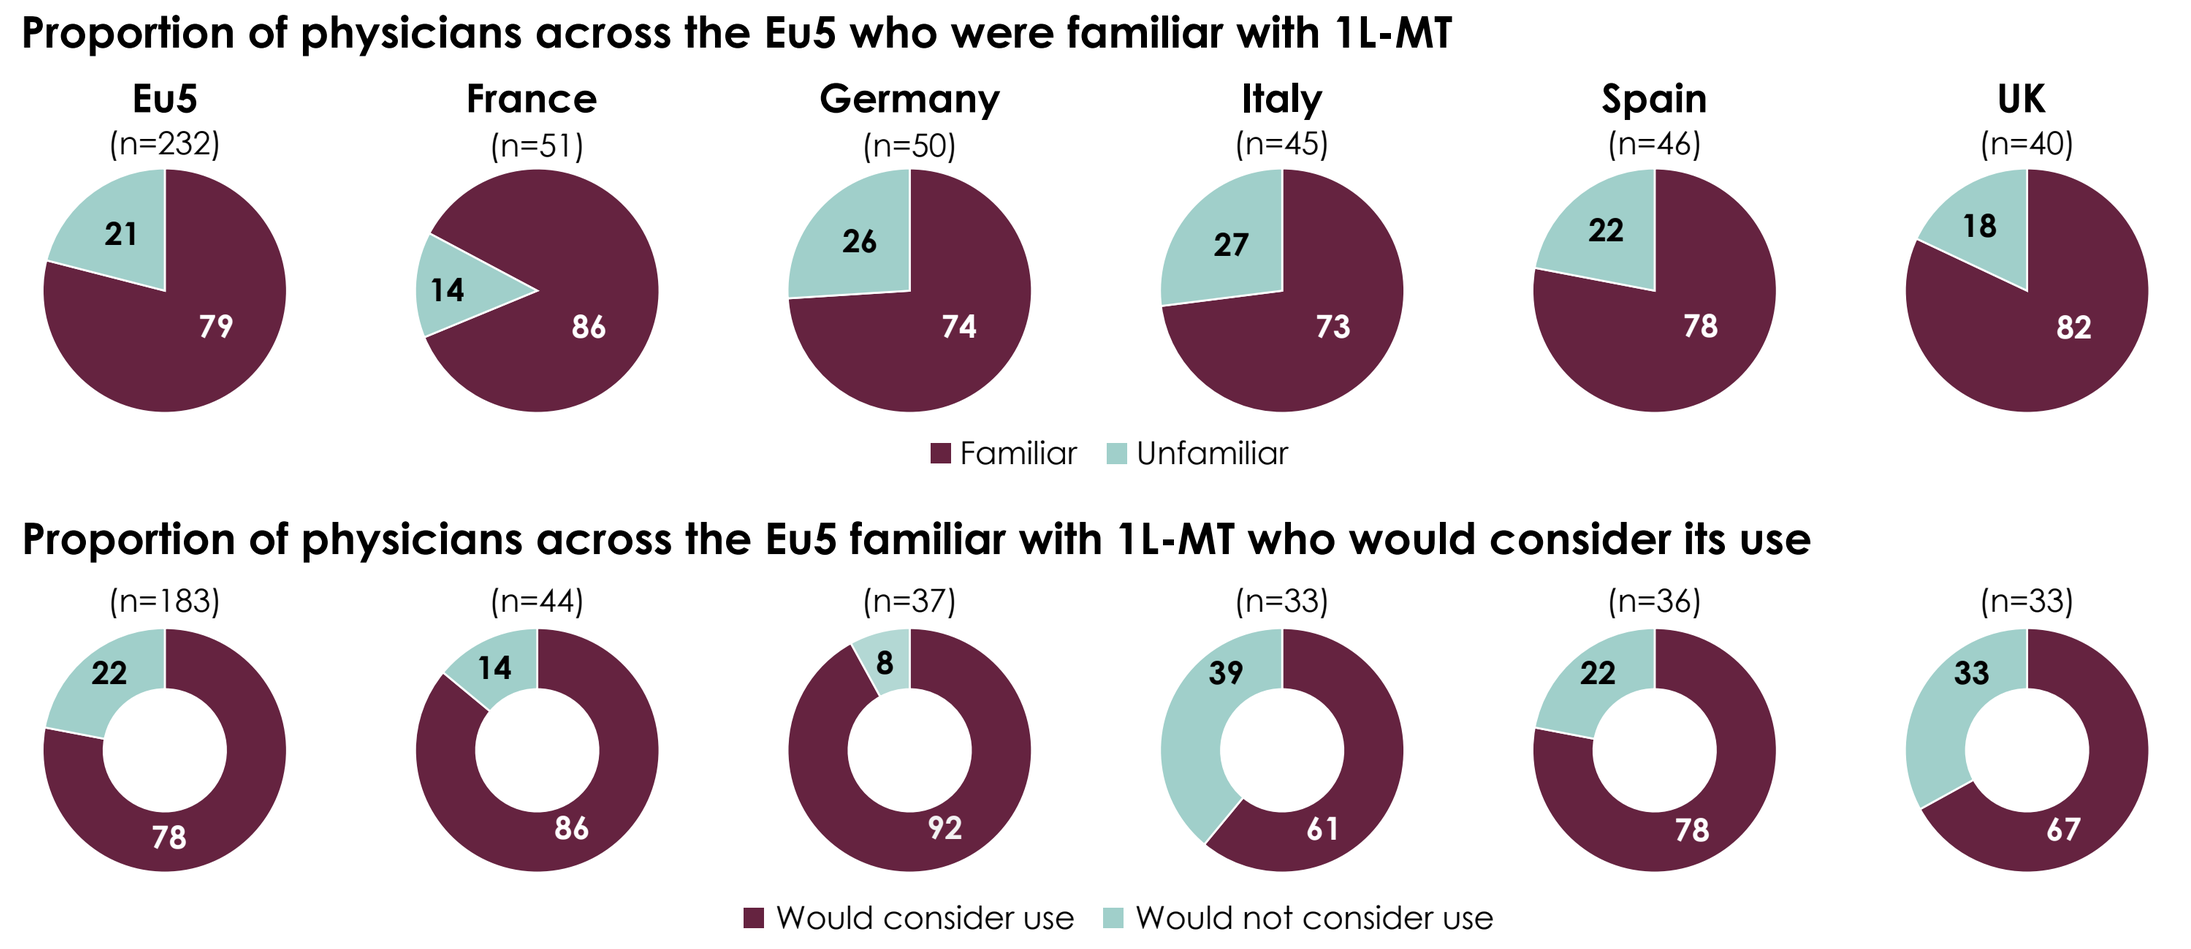
Proportion of physicians across the Eu5 who were familiar with 1L-MT
Eu5 (n=232) 21 79
France (n=51) 14 86
Germany (n=50) 26 74
Italy (n=45) 27 73
Spain (n=46) 22 78
UK (n=40) 18 82
Familiar
Unfamiliar
Proportion of physicians across the Eu5 familiar with 1L-MT who would consider its use
(n=183) 22 78
(n=44) 14 86
(n=37) 8 92
(n=33) 39 61
(n=36) 22 78
(n=33) 33 67
Would consider use
Would not consider use

1L-MT, first-line maintenance therapy; **Eu5**, France, Germany, Italy, Spain, and UK together.

Figure 3. Proportion of patients who physicians would consider treating with ICIs as 1L-MT

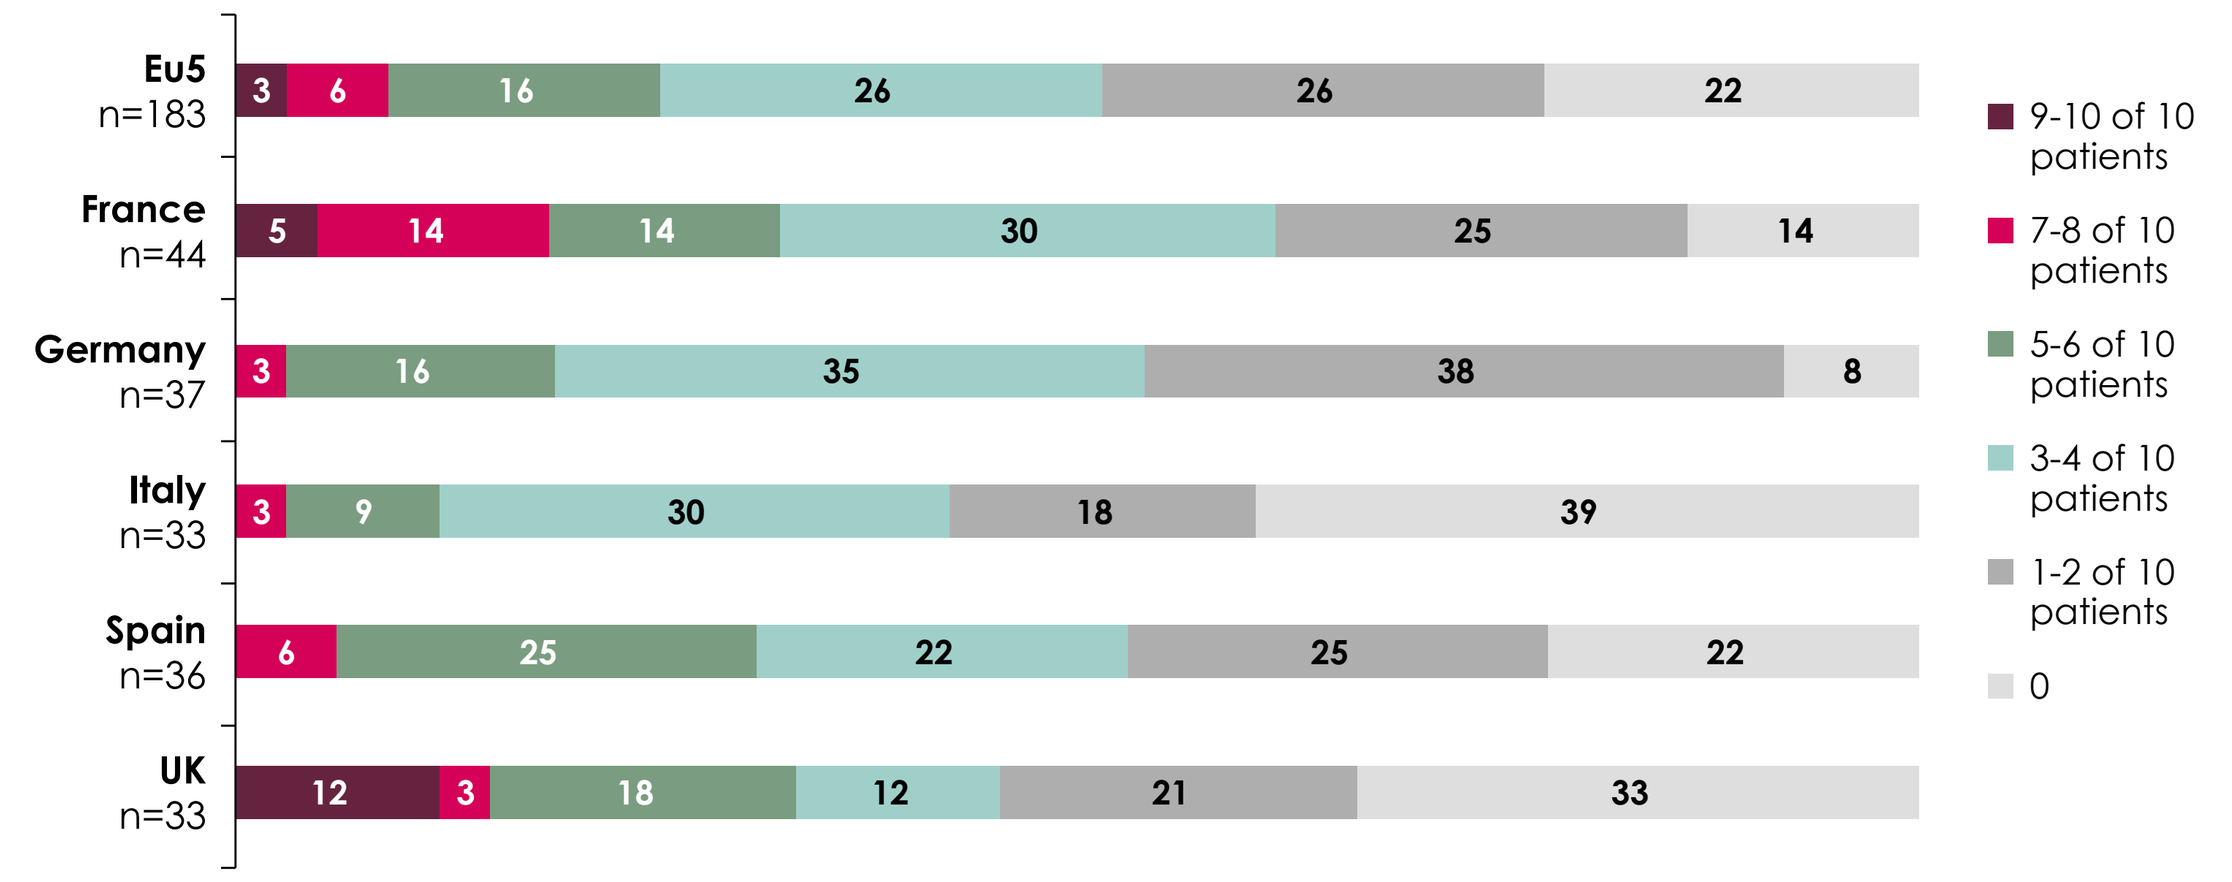
Eu5 n=183 3 6 16 26 26 22
France n=44 5 14 14 30 25 14
Germany n=37 3 16 35 38 8
Italy n=33 3 9 30 18 39
Spain n=36 6 25 22 25 22
UK n=33 12 3 18 12 21 33
9-10 of 10 patients
7-8 of 10 patients
5-6 of 10 patients
3-4 of 10 patients
1-2 of 10 patients
0

1L-MT, first-line maintenance therapy; **Eu5**, France, Germany, Italy, Spain, and UK together.
Base: All Eu5 physicians who are aware of 1L-MT.

Figure 4. Timing for initiating 1L-MT in patients with LA/mUC

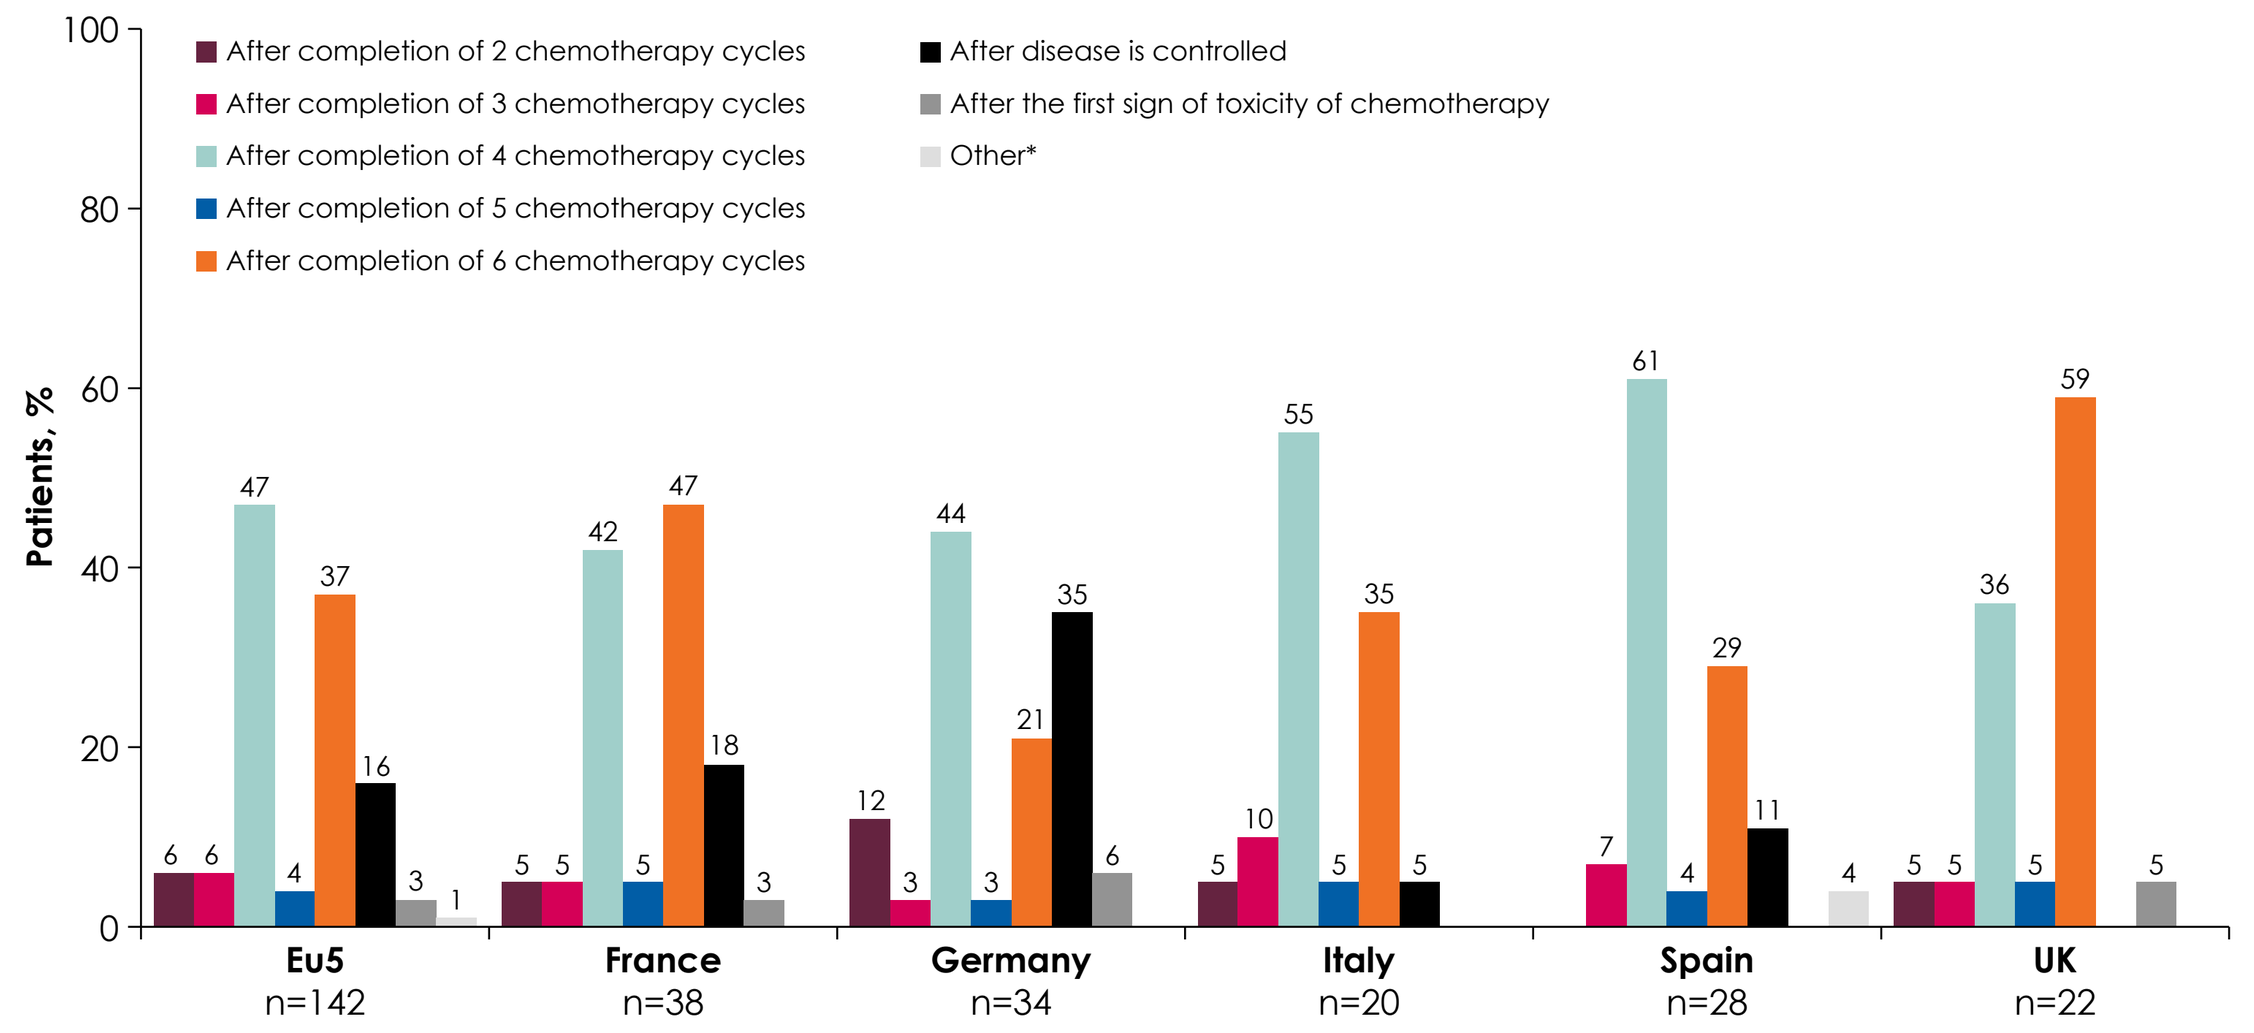
Patients, %
100
80
60
40
20
0
After completion of 2 chemotherapy cycles
After completion of 3 chemotherapy cycles
After completion of 4 chemotherapy cycles
After completion of 5 chemotherapy cycles
After completion of 6 chemotherapy cycles
After disease is controlled
After the first sign of toxicity of chemotherapy
Other*
Eu5 n=142
France n=38
Germany n=34
Italy n=20
Spain n=28
UK n=22
6 6 47 4 37 16 3 1
5 5 42 5 47 18 3
12 3 44 3 21 35 6
5 10 55 5 35 5
7 61 4 29 11 4
5 5 36 5 59 5

1L-MT, first-line maintenance therapy; **Eu5**, France, Germany, Italy, Spain, and UK together; **LA/mUC**, locally advanced or metastatic urothelial cancer.
Base: All Eu5 physicians who would consider treating their patients with 1L-MT at data abstraction.
*1 physician reported other: "With at least stable disease after undergoing 4 cycles of chemotherapy treatment."

Table 1. Physicians' demographics and workload

	Eu5	France	Germany	Italy	Spain	UK
Base (all physicians)	232	51	50	45	46	40
Physician gender, n (%)						
Female	73 (31)	14 (27)	9 (18)	26 (58)	20 (43)	4 (10)
Male	159 (69)	37 (73)	41 (82)	19 (42)	26 (57)	36 (90)
Year of qualification, n (%)						
2006 or before	138 (59)	44 (86)	34 (68)	15 (33)	25 (54)	20 (50)
2007 onward	94 (41)	7 (14)	16 (32)	30 (67)	21 (46)	20 (50)
Primary specialty, n (%)*						
Oncologist	203 (88)	44 (86)	33 (66)	45 (100)	44 (96)	37 (93)
Urologist	29 (12)	7 (14)	17 (34)	0	2 (4)	3 (8)
Ambulatory care/outpatients setting, mean (SD) %*						
Comprehensive cancer center†	22.6 (39.6)	13.5 (33.8)	2.2 (14.2)	25.4 (41.9)	14.8 (34.0)	65.6 (40.5)
Public hospital	57.7 (46.4)	74.9 (42.1)	41.4 (48.3)	61.2 (46.6)	78.5 (36.2)	28.4 (38.4)
Private hospital	5.9 (20.7)	9.6 (27.8)	2.2 (14.2)	11.3 (31.7)	2.6 (6.4)	3.4 (5.8)
Public office	13.0 (32.1)	0	54.0 (48.7)	1.9 (8.4)	3.5 (14.3)	1.6 (7.0)
Private office	0.8 (7.0)	2.0 (14.0)	0.2 (1.4)	0.1 (0.8)	0.7 (4.4)	0.9 (2.7)

Eu5, France, Germany, Italy, Spain, and UK together.
*Not all categories add up to 100% due to rounding.
†A comprehensive cancer center is a cancer-specific center, not a general hospital.

- Of the physicians across the Eu5 who would consider treating with 1L-MT, respondents would consider this therapy*:
 - After 2-3 cycles of 1L chemotherapy in 11%; this ranged from 7% in Spain to 15% in Italy and Germany
 - After 4-6 cycles of 1L chemotherapy in 89%; this ranged from 59% in Germany to 100% in the UK
 - After disease control in 16%; this ranged from 0% in the UK to 35% in Germany (**Figure 4**)
- Of the physicians who would consider treating with 1L-MT†:
 - 56% would start 1L-MT 4-6 weeks post chemotherapy
 - 25% would start 1L-MT 2-3 weeks post chemotherapy
 - 17% would start 1L-MT 6-9 weeks post chemotherapy
 - 3% would start 1L-MT at another point post chemotherapy

*The sum of values exceeds 100% because physicians were allowed to select more than 1 option.
†The sum of values exceeds 100% due to rounding.

REFERENCES 1. Siegel RL, et al. CA Cancer J Clin. 2017;66:7. **2.** Di Lorenzo G, et al. Medicine. 2015;94(51):e2297. **3.** Bellmunt J, et al. Ann Oncol. 2014;25(suppl 3):iii40-8. **4.** Witjes JA, et al. Eur Urol. 2021;79(1):82-104. **5.** Powles T, et al. N Engl J Med. 2020;383(13):1218-30. **DISCLOSURES M. Neil**, **B. Mia**, and **K. Michael** are employees of Adelphi Real World. **S. Li** is an employee of St. John University and a Pfizer Fellow. **M. Kearney** is an employee of the healthcare business of Merck KGaA and has stock/shares of Merck KGaA, Darmstadt, Germany, Novartis Pharma, and UCB Biopharma SPRL. **M. Bharmal** is an employee of EMD Serono, Billerica, MA, USA. **N. Costa** is an employee of Pfizer. **J. Chang** is an employee of Pfizer and has stock/shares of Pfizer, BMS, and Bayer. **ACKNOWLEDGMENTS** This study was funded by Pfizer as part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and Pfizer.

Poster No. POSA243. Presented at Virtual ISPOR Europe 2021, 30 November - 3 December 2021.