Health utility values in the STEP 1–4 trials of semaglutide 2.4 mg in obesity

Tobias Sydendal Grand[1]; Jakob Bue Bjorner[2,3]; Thomas Holst-Hansen[1]

[1]Novo Nordisk A/S, Søborg, Denmark; [2]QualityMetric Inc., LLC, Johnston, RI, USA; [3]Department of Public Health, University of Copenhagen, Copenhagen, Denmark

Introduction

- Obesity has a detrimental effect on health-related quality of life (HRQoL). Clinicians and regulatory authorities are putting increasing focus on assessing HRQoL when evaluating the efficacy of new therapeutic agents.
- In health economic assessments, HRQoL is often summarized as a single value (health utility) anchored by the values 1 (perfect health) and 0 (equivalent to death). Health utilities are the basis for estimation of quality-adjusted life years (QALYs), when combined with time in a particular health state and are often used in cost-effectiveness analysis.
- Here, we report health utility values in the Semaglutide Treatment Effect in People with obesity (STEP) trials 1–4.[1–4]

Methods

- The STEP trials 1–4 were phase 3a, 68-week, double-blind randomized controlled trials that assessed the efficacy and safety of semaglutide 2.4 mg versus placebo in individuals with obesity (body mass index [BMI] ≥ 30 kg/m^2) or who were overweight (BMI ≥ 27 kg/m^2) with at least one comorbidity (STEP 1, 3 and 4) or had type 2 diabetes (STEP 2).
- Patients in STEP 1 (NCT03548935),[1] STEP 2 (NCT03552757)[2] and STEP 4 (NCT03548987)[4] received lifestyle intervention (500-kcal/day deficit and a recommended 150 min/week of physical activity), and patients in STEP 3 (NCT03611582)[3] received intensive behavioural therapy.
 - In STEP trials 1, 2, 3 and 4, the estimated mean weight changes from randomization to week 68 with semaglutide 2.4 mg (placebo) were –14.9% (–2.4%), –9.6% (–3.4%), –16.0% (–5.7%) and –7.9% (+6.9%), respectively.[1–4]
- HRQoL was assessed in STEP trials 1–4 using the 36-item Short-Form Health Survey version 2 (SF-36v2), a generic health survey composed of 36 questions. Participants completed the SF-36v2 at baseline and week 68.
 - 'Baseline' corresponded to week 0 in all trials except STEP 4, in which randomization (baseline) followed a 20-week run-in period during which all patients received semaglutide 2.4 mg.
- SF-36v2 scores were mapped to health utility indices using validated methods, either by directly converting into Short-Form Six-Dimension version 2 (SF-6Dv2) utility index (UI)[5] or by mapping onto the European Quality of Life Five-Dimension Three-Level (EQ-5D-3L) UI (Figure 1).[6] Both methods were based on UK health utility weights.
- Utility values were analysed per trial using a mixed model for repeated measurements, without adjustment for multiplicity. All responses before first discontinuation of treatment were included in a mixed model with randomized treatment as a factor and baseline utility score as covariate, all nested within visit.
 - For STEP 2, the stratification groups (oral antidiabetic treatment status and haemoglobin A1c category at screening) and the interaction between stratification groups were included in the statistical model as additional factors.

Figure 1. Mapping SF-36v2 data onto the (A) SF-6Dv2 and (B) EQ-5D-3L UIs

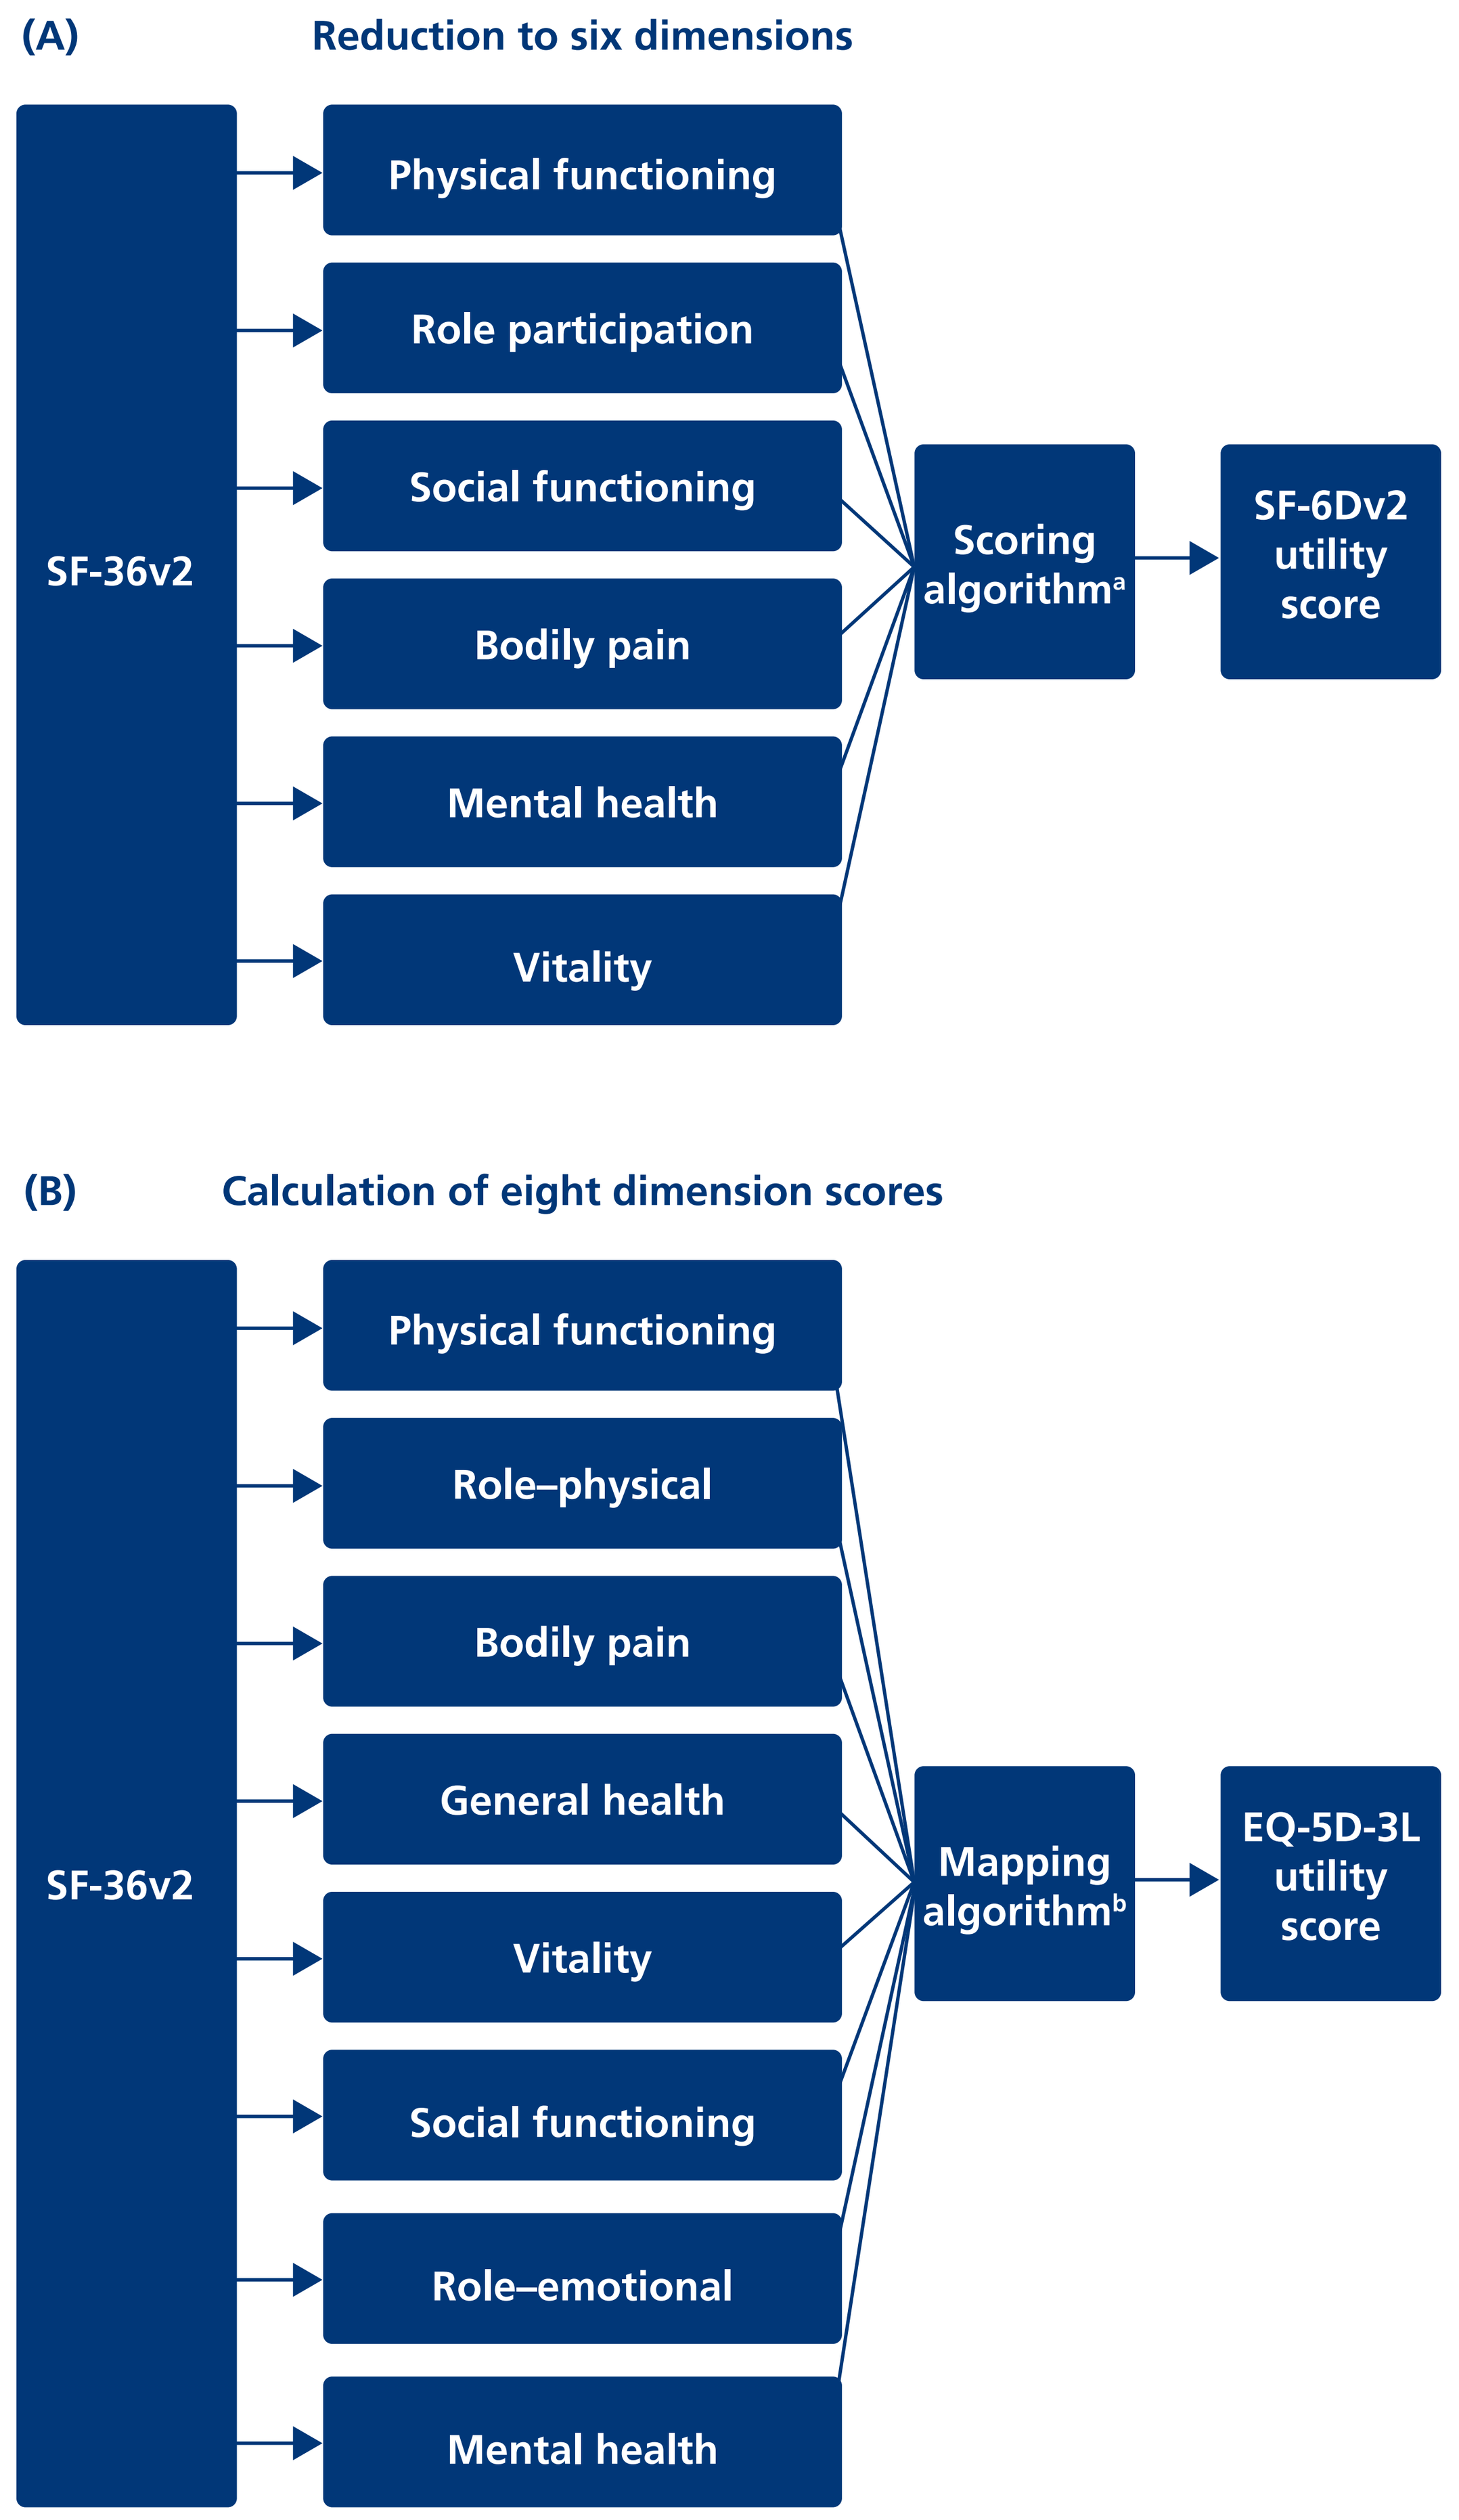
(A)
Reduction to six dimensions
SF-36v2
Physical functioning
Role participation
Social functioning
Bodily pain
Mental health
Vitality
Scoring algorithm[a]
SF-6Dv2 utility score
(B)
Calculation of eight dimension scores
SF-36v2
Physical functioning
Role–physical
Bodily pain
General health
Vitality
Social functioning
Role–emotional
Mental health
Mapping algorithm[b]
EQ-5D-3L utility score

[a]Based on the methods of Mulhern et al. (2020).[5]
[b]Based on the methods of Rowen et al. (2009).[6]
EQ-5D-3L, European Quality of Life Five-Dimension Three-Level; SF-36v2, Short-Form-36 version 2; SF-6Dv2, Short-Form Six-Dimension version 2; UI, utility index.

Results

Table 1. Frequency of comorbidities[a] at baseline

Number of comorbidities:		n	None	1	2	3	4	5 or more
			Percentage of patients having specified number of comorbidities					
STEP 1	Semaglutide 2.4 mg	1306	25.1	25.8	22.8	14.0	7.4	4.9
	Placebo	655	24.9	28.5	20.6	14.7	6.6	4.7
STEP 2	Semaglutide 2.4 mg	404	–[b]	11.6	19.1	24.3	22.0	23.0
	Placebo	403	–[b]	8.2	17.9	27.8	26.1	20.1
STEP 3	Semaglutide 2.4 mg	407	24.3	22.9	23.6	15.2	7.6	6.4
	Placebo	204	24.0	26.0	21.1	18.6	6.9	3.4
STEP 4	Semaglutide 2.4 mg	535	26.9	29.9	19.3	14.4	7.1	2.4
	Placebo	268	26.1	29.1	25.4	12.7	5.6	1.1

[a]Comorbidities evaluated include: dyslipidaemia, hypertension, coronary artery disease, cerebrovascular disease, obstructive sleep apnoea, impaired glucose metabolism, reproductive system disease, liver disease, kidney disease, osteoarthritis, gout and asthma.
[b]All patients had at least one comorbidity (type 2 diabetes) as an inclusion criterion.
STEP, Semaglutide Treatment Effect in People with obesity.

Table 2. Indexation of estimated mean utility scores in STEP trials 1–4[a]

		SF-6Dv2 UI		EQ-5D-3L UI	
		Semaglutide 2.4 mg	Placebo	Semaglutide 2.4 mg	Placebo
STEP 1	Estimate at week 68	0.843	0.814	0.908	0.879
	Change from baseline (week 0) to week 68	0.008	–0.021	0.013	–0.015
STEP 2	Estimate at week 68	0.847	0.827	0.900	0.877
	Change from baseline (week 0) to week 68	0.013	–0.008	0.018	–0.005
STEP 3	Estimate at week 68	0.864	0.851	0.923	0.907
	Change from baseline (week 0) to week 68	0.012	–0.001	0.018	0.002
STEP 4	Estimate at week 68	0.878	0.833	0.933	0.899
	Change from baseline (week 20) to week 68	0.009	–0.036	0.007	–0.027

[a]Data are mean scores at the end of treatment (week 68) and estimated mean change from baseline, analysed using a mixed model for repeated measurements, including all responses before first discontinuation of treatment. *n* semaglutide/placebo: STEP 1, 942/439; STEP 2, 318/311; STEP 3, 289/138; STEP 4, 467/219. Change from baseline may not correspond exactly to the estimated mean treatment differences in Figure 2 owing to rounding errors.
EQ-5D-3L, European Quality of Life Five-Dimension Three-Level; SF-6Dv2, Short-Form Six-Dimension version 2; STEP, Semaglutide Treatment Effect in People with obesity; UI, utility index.

Key result

Figure 2. Estimated treatment differences in SF-6Dv2 UI and EQ-5D-3L UI scores[a]

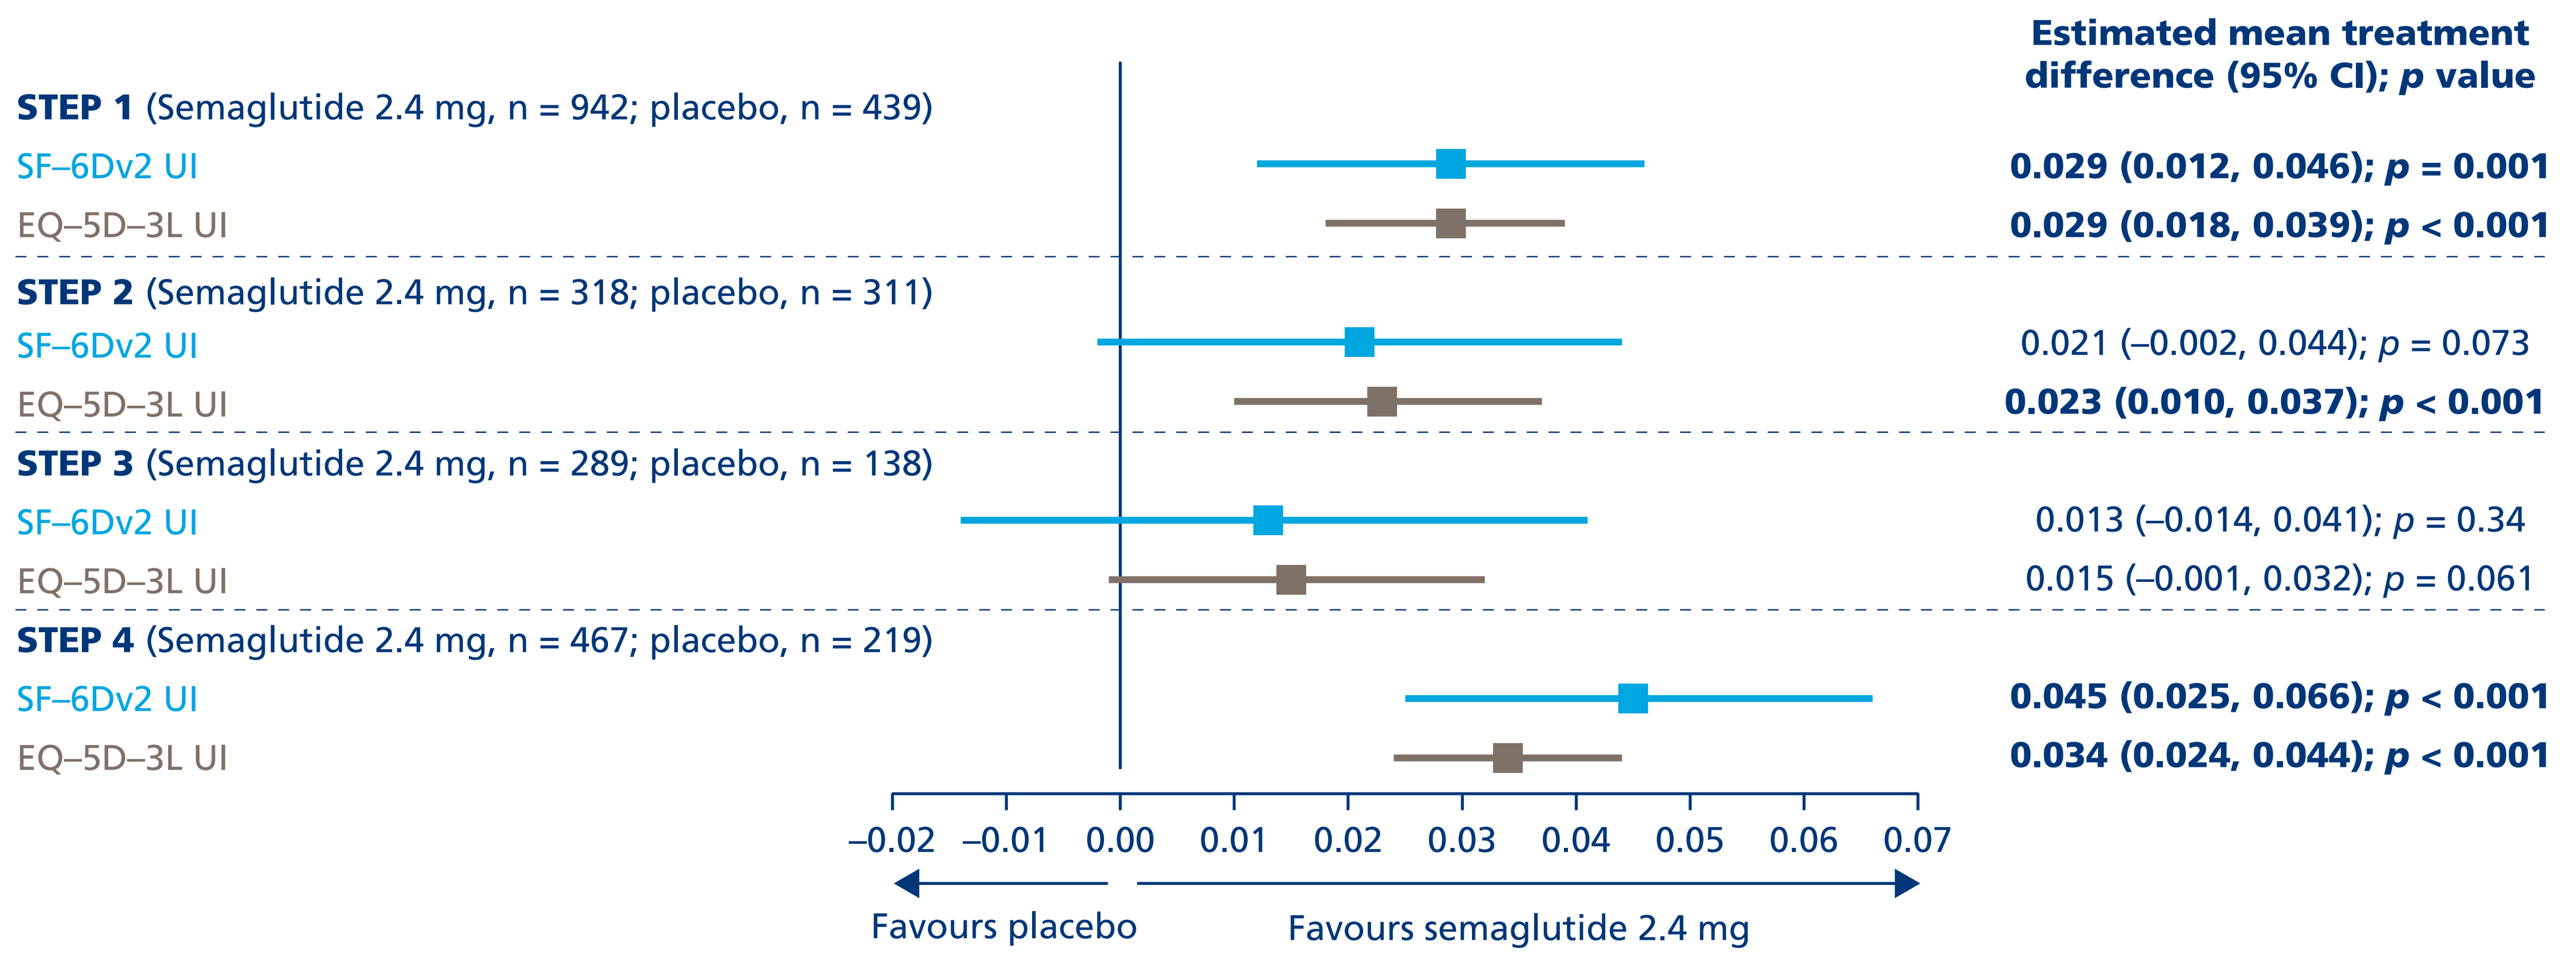
Estimated mean treatment difference (95% CI); p value
STEP 1 (Semaglutide 2.4 mg, n = 942; placebo, n = 439)
SF–6Dv2 UI
0.029 (0.012, 0.046); p = 0.001
EQ–5D–3L UI
0.029 (0.018, 0.039); p < 0.001
STEP 2 (Semaglutide 2.4 mg, n = 318; placebo, n = 311)
SF–6Dv2 UI
0.021 (–0.002, 0.044); p = 0.073
EQ–5D–3L UI
0.023 (0.010, 0.037); p < 0.001
STEP 3 (Semaglutide 2.4 mg, n = 289; placebo, n = 138)
SF–6Dv2 UI
0.013 (–0.014, 0.041); p = 0.34
EQ–5D–3L UI
0.015 (–0.001, 0.032); p = 0.061
STEP 4 (Semaglutide 2.4 mg, n = 467; placebo, n = 219)
SF–6Dv2 UI
0.045 (0.025, 0.066); p < 0.001
EQ–5D–3L UI
0.034 (0.024, 0.044); p < 0.001
–0.02 –0.01 0.00 0.01 0.02 0.03 0.04 0.05 0.06 0.07
Favours placebo
Favours semaglutide 2.4 mg

[a]Statistically significant improvements versus placebo are indicated in bold.
CI, confidence interval; EQ-5D-3L, European Quality of Life Five-Dimension Three-Level; SF-6Dv2, Short-Form Six-Dimension version 2; STEP, Semaglutide Treatment Effect in People with obesity; UI, utility index.

- The frequency of comorbidities at baseline in STEP 1–3 (week 0) and STEP 4 (week 20) is shown in Table 1.
 - Approximately half of the patients in each group across STEP trials 1, 3 and 4 had either no comorbidities or one comorbidity.
 - In STEP 2, in which all participants had type 2 diabetes as an inclusion criterion, most participants had multiple additional comorbidities.
- In STEP 1–3, mean baseline (week 0) SF-6Dv2 UI scores ranged from 0.835 to 0.851, and EQ-5D-3L UI scores from 0.881 to 0.905. In STEP 4, baseline (week 20; after run-in semaglutide treatment for all participants) SF-6Dv2 UI and EQ-5D-3L UI scores were 0.869 and 0.926, respectively.
 - Therefore, baseline health status in STEP 1–4 was generally good despite the presence of comorbidities.
- At week 68, semaglutide 2.4 mg was associated with small utility score improvements from baseline in all trials, whereas utility scores for placebo typically decreased (Table 2).
- The treatment differences in SF-6Dv2 by week 68 in the semaglutide group versus placebo were significant ($p \leq 0.001$) in STEP 1 and STEP 4 but not in STEP 2 ($p = 0.073$) or STEP 3 ($p = 0.34$) (Figure 2).
- EQ-5D-3L treatment differences in the semaglutide group versus placebo were significant in STEP 1, 2 and 4 ($p < 0.001$ for all) but not in STEP 3 ($p = 0.061$; Figure 2).
 - The lack of significant treatment differences for either utility score in STEP 3 may be due to the effects of intensive behavioural therapy offered to both the semaglutide and the placebo groups,[3] such that there was no (or negligible) decline in scores in the placebo group (Table 2).
 - The larger treatment differences in mean utility score observed in STEP 4 may be related to trial design and weight gain in the placebo group post randomization.[4]

Conclusions

- **Semaglutide 2.4 mg was associated with treatment differences in health utility scores compared with placebo, reaching statistical significance in STEP 1, 2 (EQ-5D-3L) and 4.**
 - Baseline utility scores were generally high, and treatment differences were typically driven by an increase in mean utility score in the semaglutide 2.4 mg group and a decrease in mean utility score in the placebo group.
 - The modest treatment effect may be because participants in the STEP trials 1–4 had an overall good baseline health status, which limited the effect of semaglutide 2.4 mg on utility scores.

TSG and TH-H are employees of Novo Nordisk. JBB is an employee of QualityMetric Inc., LLC. The authors would like to acknowledge the contribution of Henrik Hjorth Meinke to the analysis of the data and the development of the abstract.
This study was funded by Novo Nordisk A/S. Medical writing support was provided by Oxford PharmaGenesis, with funding from Novo Nordisk A/S.
Presented at Virtual ISPOR Europe, 30 November–3 December 2021.

References: (**1**) Wilding et al. N Engl J Med. 2021;384:989; (**2**) Davies et al. Lancet. 2021;397:971–84; (**3**) Wadden et al. JAMA. 2021;325:1403–13; (**4**) Rubino et al. JAMA. 2021;325:1414–25; (**5**) Mulhern et al. Med Care. 2020;58:566–73; (**6**) Rowen et al. Health Qual Life Outcomes. 2009;7:27; (**7**) Luo et al. Med Care. 2010; 1:365–71; (**8**) Warkentin et al. BMC Med. 2014;12:1–9.