Leveraging Real-World Data for Time-to-Event Endpoints in Clinical Trials

Deepak Parashar (University of Warwick), **Peter Almgren** (LEO Pharma), **Anders Berglund** (Daiichi Sankyo), **Alessandro Guasconi** (Chiesi Farmaceutici), **Elizabeth Merrall** (Janssen), **Claire Smith** (AstraZeneca), **Barbara Torlinska** (University of Birmingham), **Jixian Wang** (Bristol-Myers Squibb), **Qing Wang** (Roche)
PSI Real-World Data Special Interest Group Methodology Team

The Issue: Baseline of therapy initiation may not match for subjects in clinical trial and those in RWD
Uncertainty in definition of "time-zero"
Differences in (structured) missing events between Time-to-Event endpoints between trial data and RWD

Proposal: Adjust baseline and missing events to 'softer' events for multiple diseases

Examples from therapeutic areas

- **Respiratory (COPD)**
 - *Time to treatment escalation* (Real-world study comparing effectiveness of LABA/LAMA (*intervention*) vs. LAMA(*control*))
 - *Time to first COPD exacerbation* (Real-world study comparing effectiveness of ICS/LABA/LAMA (*intervention*) vs. LABA/LAMA (*control*))
- **Dermatitis**
 - *Time to drug discontinuation or treatment change* (comparing 'drug survival' (*e.g. due to non-efficaciousness, adverse events etc*.) between treatment and external control)
 - *Estimation of disease trajectories over time*: time-zero important for comparison.
- **Multiple Sclerosis** (Safety)
 - *Time to first occurrence of malignancy* (Registry case-control study; Exposure: patients exposed to at least 1 dose of therapy; comparing rate of malignancies between Exposed/Unexposed; Long latency)
- **Endocrinology**
 - *Time from final treatment to major cardiovascular events (MACE)* (Real-world study comparing long-term risks of the three main treatments for hyperthyroidism)
 - *Estimation of weight over time: time-zero needed for aligning trajectories.*
- **Oncology**
 - *Multiple starting times* for each patient (mitigating time-zero)
 - *Sufficient follow-up*
 - *Match each trial patient to at least one RWD patient* (to balance cancer stage/history/treatment)
 - *Match interim-decision* in cancer trials to treatment change or subpopulation change (targeted anti-cancer therapies)
- **Vaccines**
 - *Time to Covid-19 related hospitalisation* (Primary outcome of Observed Covid-19)
 - Claims data (Cohort study)
 - Each participant in Vaccination cohort *matched* (location, age, gender, comorbidity) *to 10 participants* with no evidence of Covid-19 vaccine.
 - Follow-up (from 14 days from cohort entry to Receiving Covid-19 vaccine/ Death/ Survival time)

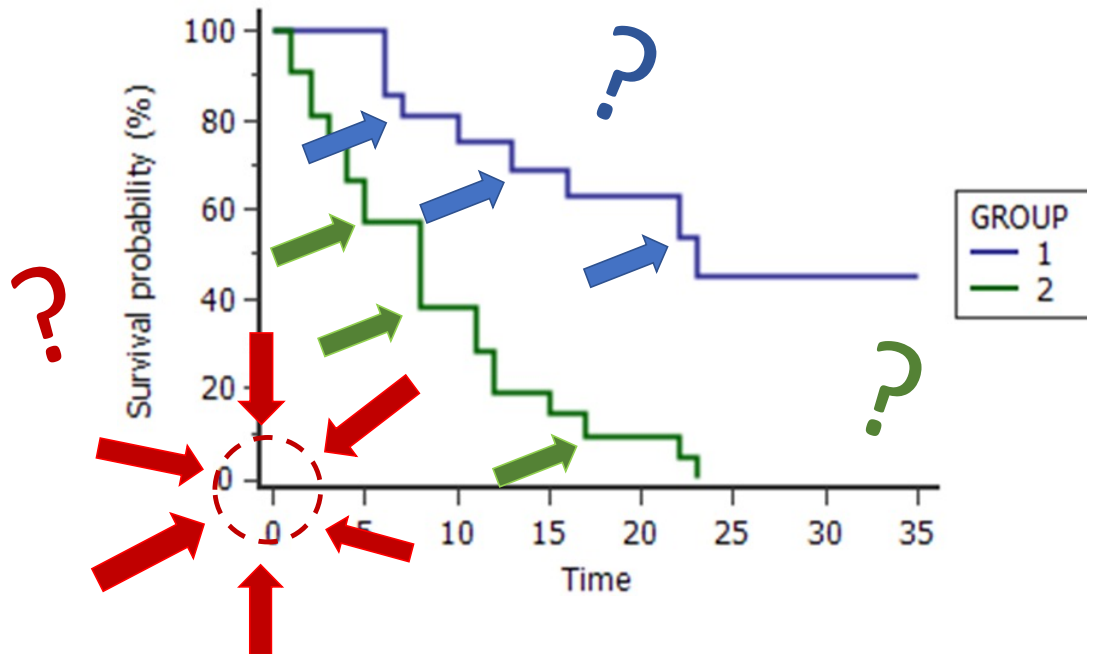
Survival probability (%)
100
80
60
40
20
0
0
5
10
15
20
25
30
35
Time
GROUP
1
2
?
?
?

References: DOI: 10.1183/23120541.00106-2019; DOI: 10.1186/s12931-021-01776-y; DOI: 10.1136/bmjopen-2021-055219; DOI: 10.1002/pds.4107; DOI: 10.1093/aje/kwv254; https://www.medrxiv.org/content/10.1101/2021.09.10.21263385v2.full.pdf