An Economic Analysis of Empagliflozin versus Sacubitril/Valsartan in Patients with Heart Failure with Reduced Ejection Fraction (HFrEF) in the United Kingdom (UK)

Reifsnider O[1], Tafazzoli A[1], Bellanca L[2], Litkiewicz M[3], Stargardter M[1], Linden S[4]

[1]Evidera, Bethesda, MD, USA; [2]Boehringer Ingelheim Ltd., Berkshire, UK; [3]Evidera, London, UK; [4]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany

INTRODUCTION

- The sodium-glucose cotransporter 2 inhibitor (SGLT2i) empagliflozin and angiotensin receptor neprilysin inhibitor (ARNi) sacubitril/valsartan have both shown to reduce cardiovascular (CV) death or hospitalization for heart failure (HHF) in patients with heart failure with reduced ejection fraction (HFrEF).
- Empagliflozin 10 mg once daily was compared with placebo as an add-on to standard of care (SoC) for HFrEF in the EMPEROR-Reduced trial, demonstrating a 25% (HR: 0.75; 95% confidence interval [CI]: 065–0.86) reduction in the combined relative risk of CV death or HHF, leading to recent authorization by the European Medicines Agency (EMA) in this indication.[1]
 - Effect of empagliflozin + SoC on this outcome was observed irrespective of the use of sacubitril/valsartan at baseline.[1]
 - Risk reduction for CV death or HHF among patients not using sacubitril/valsartan: HR: 0.77; 95% CI: 0.66–0.90.[1]
- Sacubitril/valsartan is authorized for use by EMA for management of HFrEF based on results from the PARADIGM-HF trial.[2]
- Comparative value for money for empagliflozin vs. sacubitril/valsartan in treatment of patients with HFrEF is unknown.

OBJECTIVE

- To conduct a cost-minimization analysis of empagliflozin vs. sacubitril/valsartan in HFrEF patients who were already treated with SoC including angiotensin-converting enzyme inhibitors (ACEi) or angiotensin receptor blockers (ARB), beta blockers, mineralocorticoid receptor antagonists (MRAs), and/or ivabradine.

METHODS

Model Overview

- Cost-minimization analysis was conducted from the United Kingdom (UK) National Health Service (NHS) perspective over a lifetime horizon.
- A Markov cohort state-transition model was used to simulate the clinical course of HFrEF (Figure 1).
- Health states based on Kansas City Cardiomyopathy Questionnaire—Clinical Summary Score (KCCQ-CSS) quartiles tracked patients' disease severity over time.
- At the start of the model, the patient cohort entered following the baseline distribution of KCCQ-CSS quartiles per the EMPEROR-Reduced trial data.
- During each model cycle, patients could transition between KCCQ-CSS health states and/or death (CV or non-CV) and were subject to risk of HHF, treatment-related adverse events (AEs), and treatment discontinuation.
 - First and subsequent HHF and treatment-related AEs were captured as transient events and patients who discontinued treatment with empagliflozin or sacubitril/valsartan were treated with SoC alone.

Figure 1. Model Diagram

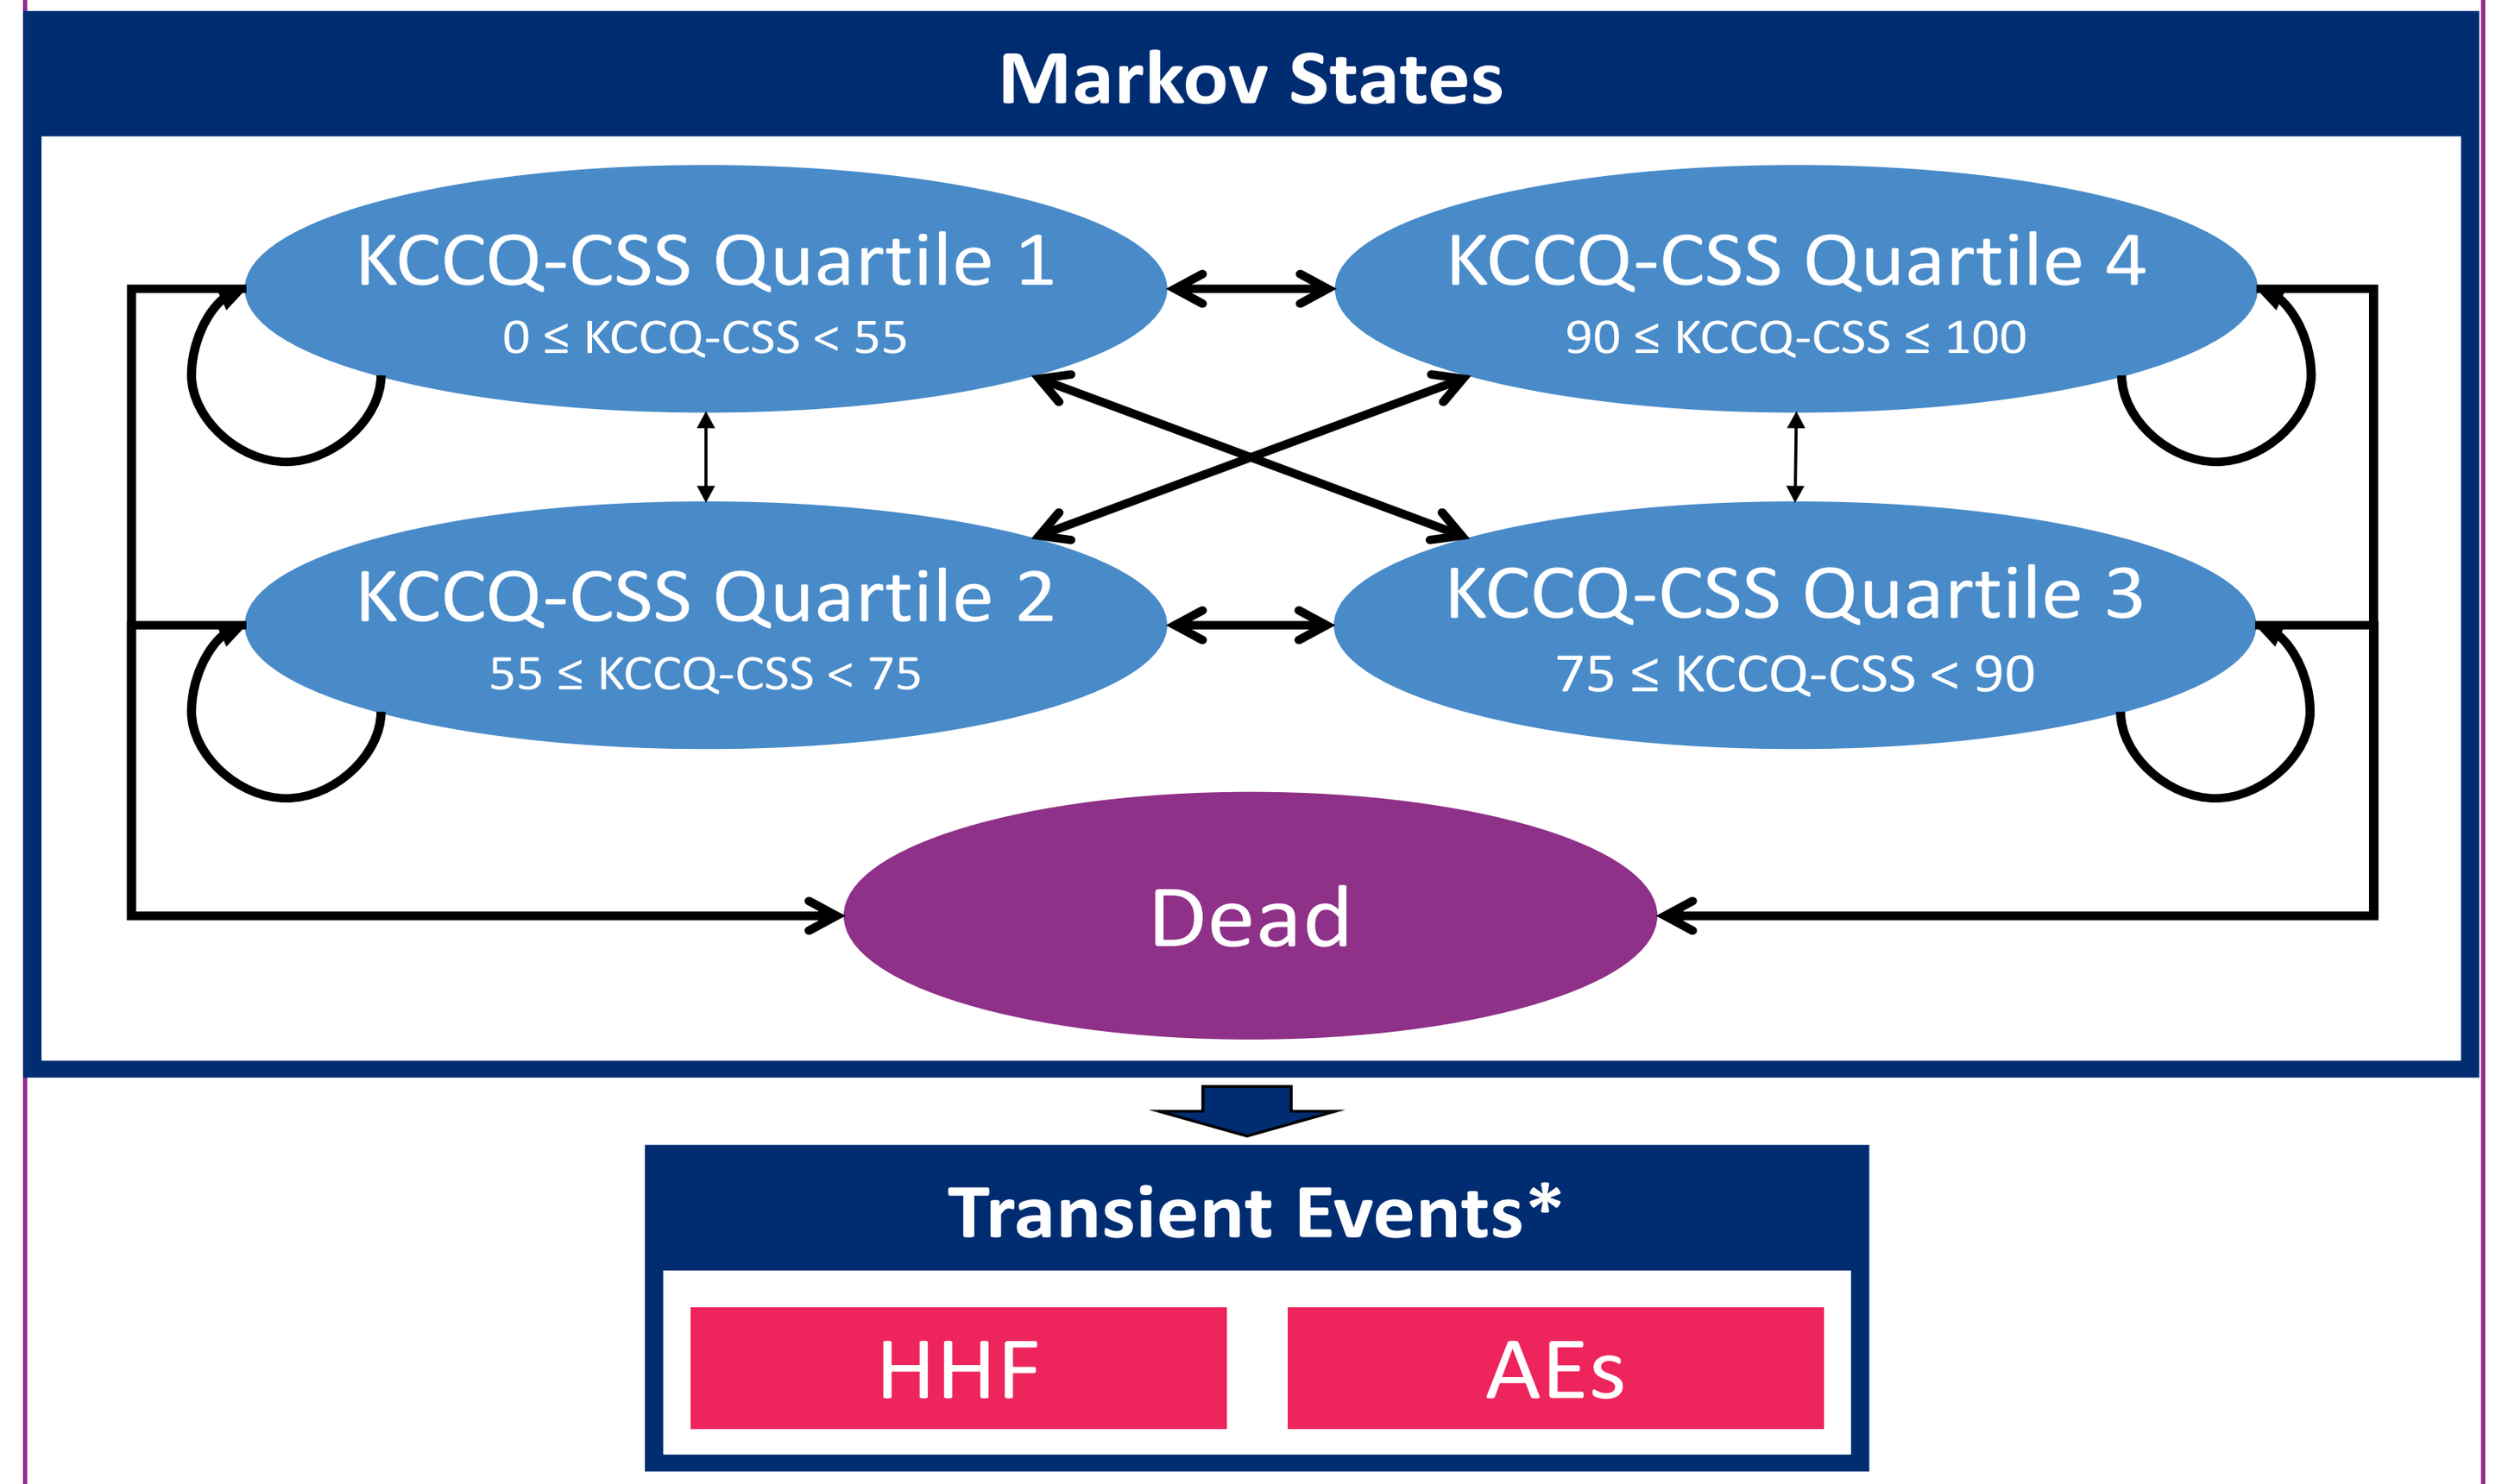
Markov States
KCCQ-CSS Quartile 1
0 ≤ KCCQ-CSS < 55
KCCQ-CSS Quartile 4
90 ≤ KCCQ-CSS ≤ 100
KCCQ-CSS Quartile 2
55 ≤ KCCQ-CSS < 75
KCCQ-CSS Quartile 3
75 ≤ KCCQ-CSS < 90
Dead
Transient Events*
HHF
AEs

Abbreviations: AE = adverse event; KCCQ-CSS = Kansas City Cardiomyopathy Questionnaire Clinical Symptom Score; HHF = hospitalization for heart failure
* Transient events may occur in any non-death health state.

METHODS (CONT.)

Population

- Modeled population was the same as participants with HFrEF who were not treated with ARNi at baseline in the EMPEROR-Reduced trial (constituting approximately 80.5% of the trial population)
 - Adults with New York Heart Association (NYHA) class II-IV and left ventricular ejection fraction (LVEF) ≤40%. Mean age was 67 years, and 76% of participants were male.
 - Initial proportion of patients distributed in KCCQ-CSS Quartile 1 (0 ≤KCCQ-CSS <55), Quartile 2 (55 ≤KCCQ-CSS <75), Quartile 3 (75 ≤KCCQ-CSS <90), and Quartile 4 (90 ≤KCCQ-CSS ≤100) was 24.6%, 25.7%, 27.0%, and 22.7%, respectively.

Clinical Parameters

- Monthly transitions between KCCQ-CSS health states for three time periods (baseline–week 12, week 12–32, and week 32–52) were derived from patient-level EMPEROR-Reduced trial data. Transition probabilities from week 32–52 were used for the remaining model time horizon.
- Risk equations for HHF and death were based on a sub-analysis of patients with HFrEF not treated with ARNi at baseline in the EMPEROR-Reduced trial and included KCCQ-CSS quartiles as time-varying predictors.
 - Rate of first and recurrent HHF was captured using a Poisson model fitted to patient-level data with generalised estimating equations to account for repeated measures.
 - Parametric survival analysis was used to project event-free survival curves for CV death and all-cause death.
 - Proportion of non-CV deaths calculated from the risk equations was compared with UK age- and sex-specific life tables (adjusted to exclude CV deaths), and the least optimistic data was applied.
- Parametric survival analysis of EMPEROR-Reduced trial data informed time to active treatment discontinuation, considering KCCQ-CSS quartiles as time-varying predictors.

Treatment-related Parameters

- Equal effectiveness was assumed for empagliflozin + SoC and sacubitril/valsartan + SoC based on Bucher indirect treatment comparison (ITC).
 - Comparison of the EMPEROR-Reduced trial subpopulation without ARNi use vs. all patients in PARADIGM-HF was performed.
 - Empagliflozin vs. sacubitril/valsartan on time to CV death or HHF was not statistically significant (HR: 0.96; 0.81–1.15); similarly, no difference in treatment effect was shown on individual outcomes.
- AEs for empagliflozin + SoC reflected EMPEROR-Reduced trial data, and AEs for sacubitril/valsartan were informed by published PARADIGM-HF trial data; AEs were assumed to be constant over time.

Cost Inputs

- Drug acquisition, clinical event management, and disease management costs in £2021 were derived from public sources[3-6] and discounted at 3.5% per year.
- A weighted average drug acquisition cost was applied for each treatment regimen, adjusted for the utilization of HF medication classes in the EMPEROR-Reduced trial subpopulation without ARNi use at baseline (Table 1).
 - Stable maintenance dosage for each SoC treatment was applied.
- HHF, CV death, and AEs incurred a one-time cost per event in the model (Table 2).
 - AE expenditures were a weighted average based on whether patients were expected to receive outpatient or inpatient care.
- Frequency of use and unit costs for GP visits, cardiologist visits, and accident and emergency referrals were used to estimate the total HF-related disease management cost per patient (Table 3).

Table 1. Drug Costs by Treatment Regimen (£2021)

	Cost[3] (Utilization)*	
Treatment	Empagliflozin + SoC	Sacubitril/Valsartan + SoC
SGLT2i: empagliflozin	£39.78 (100%)	--
ARNi: sacubitril/valsartan	--	£99.53 (100%)
ACEi: captopril, enalapril, lisinopril, ramipril, trandolapril**	£6.03 (41%)	£6.03 (41%)
ARB: candesartan, valsartan, losartan**	£11.05 (29%)	£11.05 (29%)
MRA: eplerenone	£7.63 (71%)	£7.63 (71%)
BB: bisoprolol, carvedilol, metoprolol, nebivolol**	£9.50 (95%)	£9.50 (95%)
HCN channel blocker: ivabradine	£4.83 (6%)	£4.83 (6%)
Estimated weighted drug cost per month	**£60.13**	**£119.89**

Abbreviations: ACEi = angiotensin-converting enzyme inhibitor; ARB = angiotensin receptor blocker; ARNi = angiotensin receptor neprilysin inhibitor; BB = beta blocker; HCN = hyperpolarization-activated cyclic nucleotide-gated; MRA = mineralocorticoid receptor antagonist; SGLT2i = sodium-glucose cotransporter 2 inhibitor; SoC= standard of care
Note: Cost of SoC alone was estimated to be £20.35.
*Utilization among the subgroup of patients in EMPEROR-Reduced who were not treated with ARNi at baseline.
**Treatments within each class were assumed to be uniformly distributed.

METHODS (CONT.)

Table 2. Acute Clinical Event Management Cost (£2021)

Event	Cost per Event
HHF[4]	£3,072
CV Death[5]	£4,146
Urinary Tract Infection[a6]	£40
Genital Mycotic Infection[a6]	£40
Acute renal failure[b4]	£1,906
Hepatic injury[c4,6]	£1,274
Volume depletion[a6]	£40
Hypotension[a6]	£40
Hypoglycemic event[c4,6]	£627
Bone fracture[b4]	£2,710
Hyperkalemia[a6]	£40
Cough[a6]	£40
Dizziness[a6]	£40

Abbreviations: CV = cardiovascular; HHF = hospitalization for worsening heart failure
Note: Cost of non-CV death was assumed to be zero.
[a]Patients were assumed to receive outpatient care (per patient contact lasting 9.22 minutes).
[b]Patients were assumed to receive inpatient care (weighted mean by national average unit costs and number of finished consultant episodes).
[c]Patients were assumed to receive outpatient (50%) or inpatient (50%) care.

Table 3. Disease Management Costs (£2021)

	KCCQ-CSS Quartile 1–4	
Resource	Monthly Frequency	Monthly Unit Cost
GP visit[a6]	1.9283	£40
Cardiologist visit[b4]	0.0042	£140
A&E referral[c4]	0.0008	£153
Estimated total monthly cost per patient	**£77**	

Abbreviations: A&E = accident and emergency; GP = general practitioner; KCCQ-CSS = Kansas City Cardiomyopathy Questionnaire Clinical Summary Score
[a]Outpatient visit with per patient contact lasting 9.22 minutes.
[b]Cardiology, non-admitted, face to face, follow-up visit.
[c]Weighted mean by national average unit costs and number of finished consultant episodes for emergency medicine.

RESULTS

- In validation, event rates for empagliflozin + SoC vs. SoC estimated by the model over 18 months (available upon request) were consistent with the EMPEROR-Reduced trial results.
- Total discounted costs per patient were £15,347 with empagliflozin and £17,220 with sacubitril/valsartan, resulting in total lifetime cost savings of £1,873/patient which is equivalent to a 12.2% cost reduction (Figure 2).
 - Incremental cost was driven primarily by the lower drug acquisition cost for empagliflozin vs. sacubitril/valsartan
- Empagliflozin remained cost-saving by 10.1% to 22.1% vs. sacubitril/valsartan under a range of sensitivity analyses (Table 4).

Figure 2. Estimated Total Per Patient Lifetime Cost of Treatment

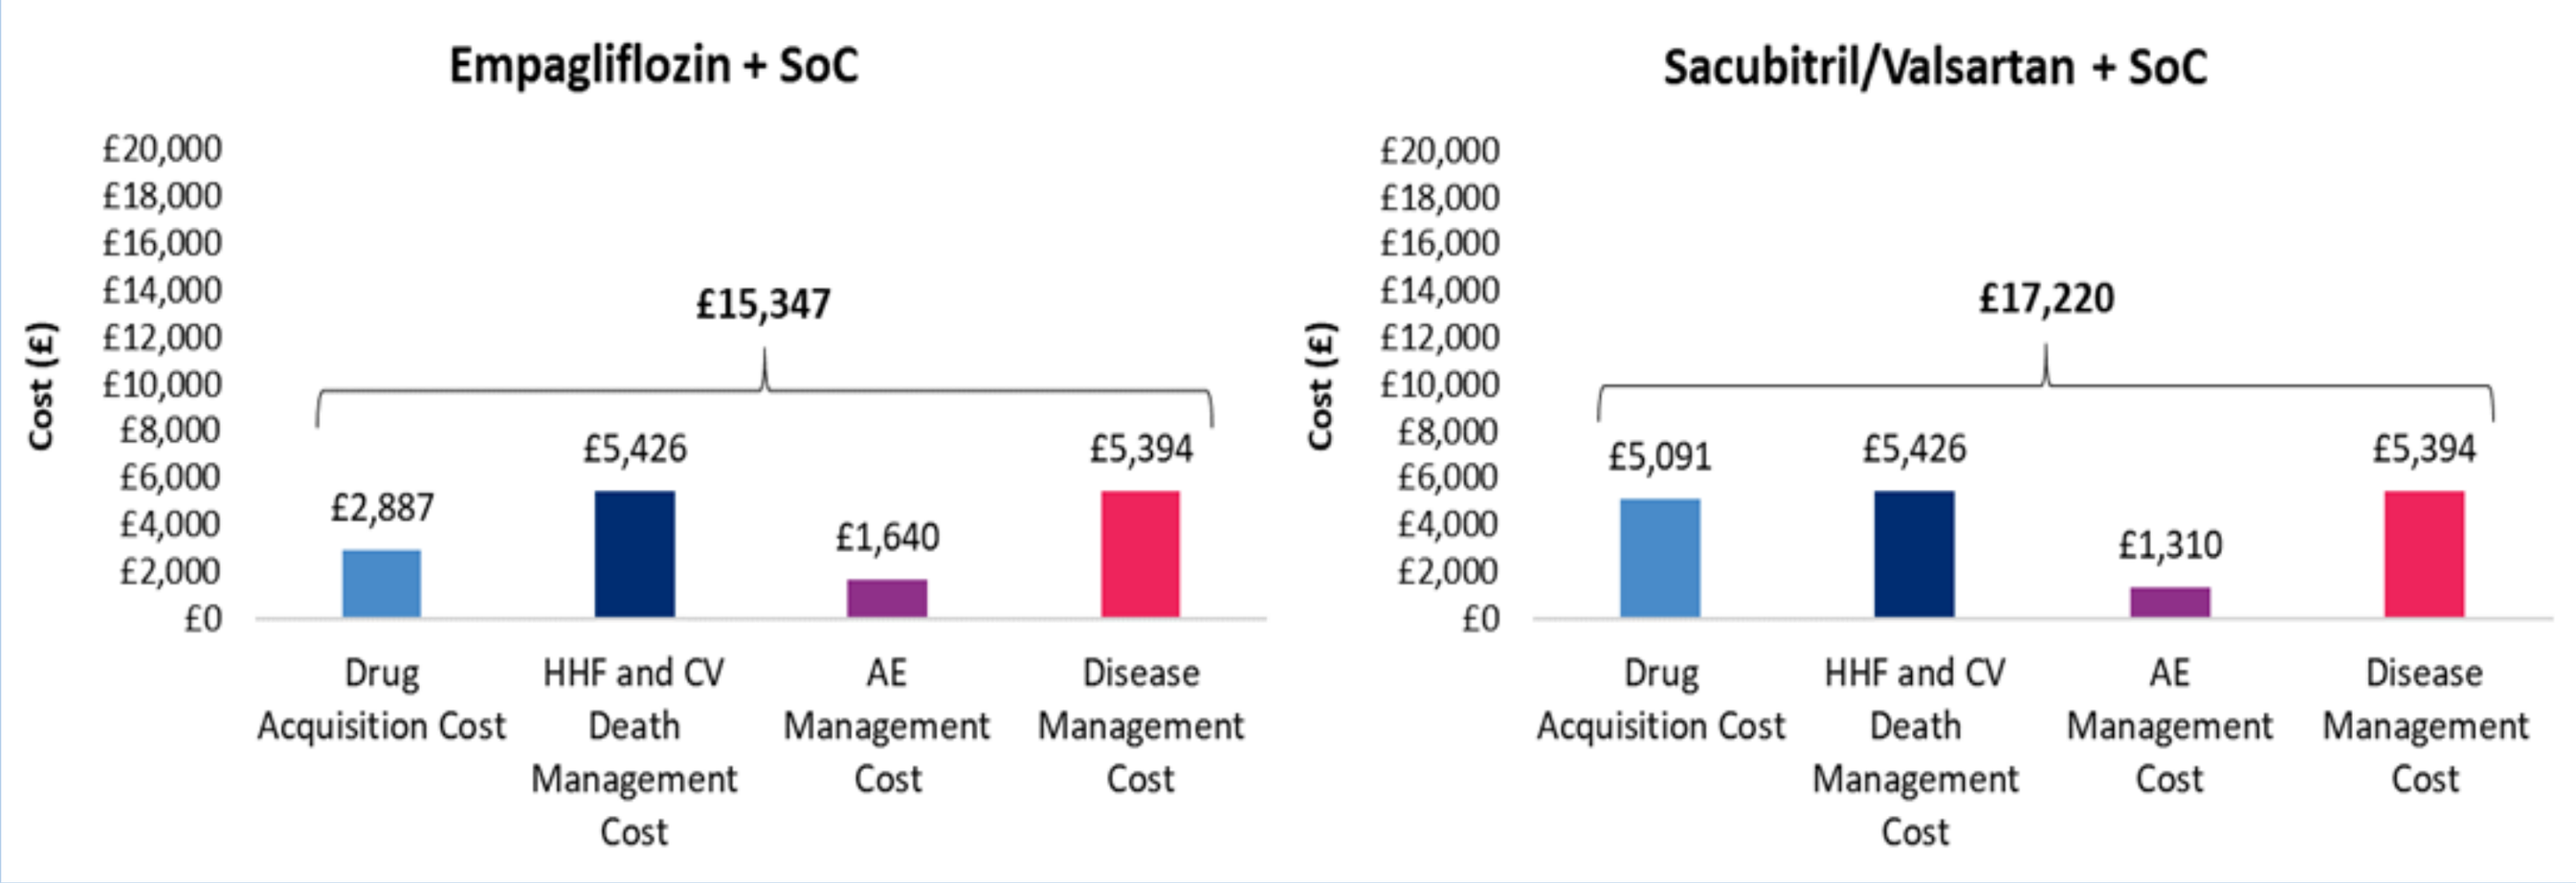
Empagliflozin + SoC
Sacubitril/Valsartan + SoC
Cost (£)
£20,000
£18,000
£16,000
£14,000
£12,000
£10,000
£8,000
£6,000
£4,000
£2,000
£0
£15,347
£2,887
£5,426
£1,640
£5,394
£17,220
£5,091
£5,426
£1,310
£5,394
Drug Acquisition Cost
HHF and CV Death Management Cost
AE Management Cost
Disease Management Cost

Abbreviations: AE = adverse event; CV = cardiovascular; HHF = hospitalization for worsening heart failure; SoC= standard of care

RESULTS (CONT.)

Table 4. Sensitivity Analysis Results

Scenario	Total Cost: Empagliflozin + SoC	Total Cost: Sacubitril/Valsartan + SoC	Cost Difference	Saving in Annual Cost (%)
Base case	£15,347	£17,220	-£1,873	-12.2%
Sensitivity Analysis				
Exclude treatment discontinuation	£16,228	£19,820	-£3,592	-22.1%
Empagliflozin acquisition cost, -20%	£15,054	£17,220	-£2,167	-14.4%
Time horizon, 10 years	£13,253	£15,067	-£1,814	-13.7%
Treatment discontinuation, alternative distribution*	£15,403	£17,386	-£1,983	-12.9%
AE management cost, -20%	£15,019	£16,959	-£1,939	-12.9%
Discount rate on cost, 5%	£14,302	£16,099	-£1,797	-12.6%
Time horizon, 20 years	£15,160	£17,032	-£1,872	-12.3%
AE management cost, +20%	£15,675	£17,482	-£1,807	-11.5%
Discount rate on cost, 0%	£18,511	£20,596	-£2,085	-11.3%
Empagliflozin acquisition cost, +20%	£15,641	£17,220	-£1,580	-10.1%

Abbreviations: AE = adverse event; SoC= standard of care
*Weibull instead of exponential in the base case.

LIMITATIONS

- Analyses were based on the subpopulation of EMPEROR-Reduced not treated with ARNi at baseline, and the trial was not powered to assess treatment benefits in a subgroup.

CONCLUSIONS

- Treatment with empagliflozin added to SoC resulted in significant cost savings compared to sacubitril/valsartan added to SoC for the management of HFrEF in the UK.
- For decision makers, treatment with empagliflozin may reduce the cost burden of HFrEF.

REFERENCES

1. Packer M. N Engl J Med. 2020; 383:1413-1424.
2. McMurray J. N Engl J Med. 2014; 371:993-1004.
3. MIMS Drug Database 2021.
4. NHS Reference Costs 2018-2019.
5. Alva ML. Diabet Med. 2015 Apr; 32(4):459-466.
6. Personal Social Services Research Unit (PSSRU) 2019.

Poster Presented at: International Society for Pharmacoeconomics and Outcomes Research Europe | 30 November–3 December 2021 | Virtual

Brought to you by:
EMPOWER
Empowering patients and healthcare professionals to do more