POSB24

Long-Term Survival Associated With Next-Generation Sequencing Versus Standard Diagnostic Tests to Detect Epidermal Growth Factor Receptor Exon 20 Insertion Variants in Non–Small Cell Lung Cancer: A Decision-Analytic Model

Luis G Hernandez[1], Eric N Churchill[1], Laura J Walton [2]

[1]Millennium Pharmaceuticals, Inc., a wholly owned subsidiary of Takeda Pharmaceutical Company Limited, Cambridge, MA, USA, [2]Takeda Pharmaceuticals, Zurich, Switzerland

Background

- Non–small cell lung cancer (NSCLC) accounts for 80% to 85% of all lung cancers.[1] Adenocarcinoma is the most common histologic NSCLC subtype and is divided further into molecular subtypes with distinct and often actionable oncogenic driver mutations in the epidermal growth factor receptor (*EGFR*) and anaplastic lymphoma kinase genes[1,2]
- *EGFR* exon 20 insertions (ex20ins) are a group of *EGFR*-activating mutations.[3] The frequency of the ex20ins mutations is estimated at approximately 2% among all patients with NSCLC and 4% to 12% among those with *EGFR* mutations[4-7]
- Unlike *EGFR* exon 19 deletions and exon 21 point mutations, ex20ins are not associated with sensitivity to classic EGFR tyrosine kinase inhibitors (TKIs)[3]
- With the rapid increase in drug development for patients with this disease and the approval in the United States of amivantamab and mobocertinib for patients with locally advanced or metastatic NSCLC harboring *EGFR*ex20ins mutations whose disease has progressed on or after platinum-based chemotherapy,[8,9] it is becoming increasingly important to identify these ex20ins variants to optimize treatment outcomes
- Although commonly used, single-gene polymerase chain reaction (sgPCR) tests are limited for detecting all *EGFR* variants[10]
- Next-generation sequencing (NGS) testing can detect all ex20ins variants,[11] allowing for more patients to be accurately identified and appropriately treated

Objective

- To develop a decision-analytic model to quantify and understand how testing with NGS versus sgPCR impacts long-term outcomes for patients with *EGFR*ex20ins-positive NSCLC

Methods

- A Markov model was developed in Microsoft Excel. The model structure is presented in **Figure 1**
- The model considered 2 identical cohorts with *EGFR*ex20ins-positive NSCLC, followed from the time of diagnosis and biomarker testing until death. One cohort underwent biomarker testing for *EGFR* mutations with NGS and the other with sgPCR
- NGS was assumed to detect 100% of ex20ins variants in the base-case scenario (varied between 90% and 100%)
- Ex20ins variant detectability with sgPCR was determined from a retrospective analysis in NSCLC patients whose *EGFR*ex20ins were identified by NGS from a global trial of mobocertinib (NCT02716116; n=145) and 3 real-world data sets: US COTA Electronic Health Record database (n=30), chart review study across 12 academic sites in Germany (n=80), and Japanese genome screening project (LC-SCRUM-Japan; n=95)[10]
 - In the retrospective study, the proportions of patients with *EGFR*ex20ins variants identifiable by 6 commercially available PCR kits were projected based on manufacturer-provided coverage information. These PCR tests were projected to detect 12.4% to 51.0% of patients in the trial, 13.3% to 46.7% in US COTA, 17.5% to 50.0% in the German data set, and 21.1% to 58.9% in Japan LC-SCRUM[10]
 - sgPCR was assumed to conservatively detect 59% of ex20ins variants in the base-case scenario (varied between 12% and 59%; the lowest and highest detectability of ex20ins variants for PCR tests)
- Patients correctly identified as having ex20ins received platinum-based chemotherapy (PBC) as first-line treatment, while patients incorrectly identified as not having ex20ins were assumed to receive first-line PBC, immuno-oncology (IO) agents or classic EGFR TKIs (**Table 1**)

Figure 1. Model Structure

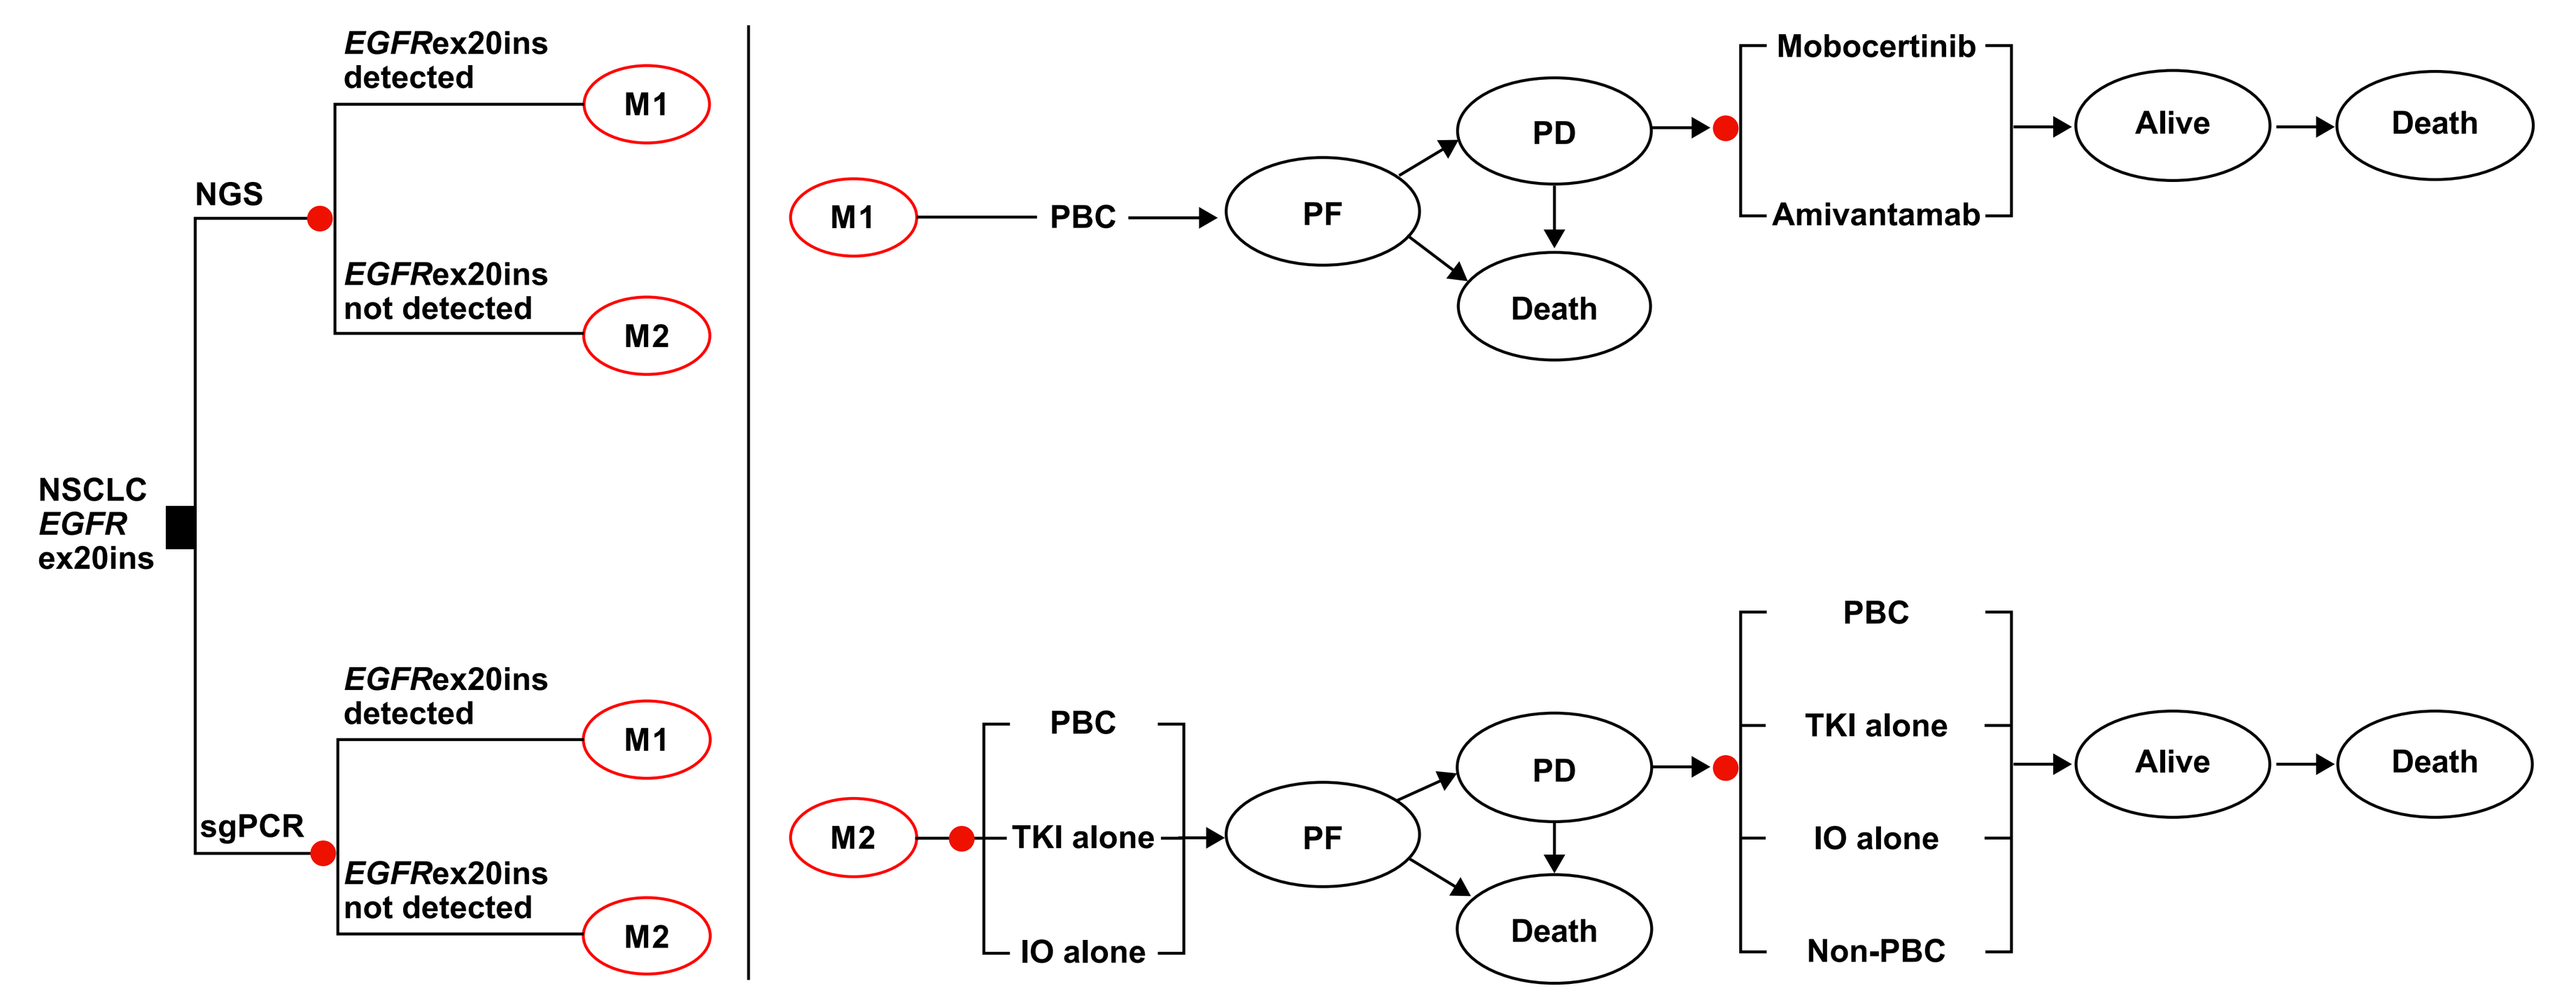

NSCLC
EGFR
ex20ins
NGS
sgPCR
EGFRex20ins detected
EGFRex20ins not detected
M1
M2
PBC
TKI alone
IO alone
PF
PD
Death
Mobocertinib
Amivantamab
Non-PBC
Alive

EGFR, epidermal growth factor receptor gene; ex20ins, exon 20 insertions; IO, immune-oncology therapy; NGS, next-generation sequencing; NSCLC, non–small cell lung cancer; PBC, platinum-based chemotherapy; PD, progressive disease; PF, progression free; sgPCR, single-gene polymerase chain reaction; TKI, tyrosine kinase inhibitor

- For each first-line treatment, the model calculated the proportion of patients who remained progression-free, had progressed, or had died every month (model cycle length), using a partitioned survival structure (**Figure 1**)
- Patients who experienced disease progression switched to a second-line treatment (**Table 1**) and were followed until death (**Figure 1**)
 - The model assumes that the impact of any subsequent treatment post-second line was implicitly captured in the overall survival (OS) of each second-line treatment
- The progression-free survival (PFS) and OS for each treatment were extrapolated based on an exponential model, ie, assuming a constant hazard of progression and death calculated based on the median PFS and OS (**Table 1**)
- The model calculated the average number of life years for both cohorts (undergoing NGS vs sgPCR) from the time of diagnosis and biomarker testing until death

Table 1. Distribution and Median PFS and OS of First- and Second-Line Treatments

	Line 1				Line 2		
	Treatment Distribution		Median (95% CI), Months[12]		Treatment Distribution		Median (95% CI), Months[12-14]
Treatment	*EGFR*ex20ins detected	*EGFR*ex20ins not detected	PFS	OS	*EGFR*ex20ins detected	*EGFR*ex20ins not detected	OS
PBC	100%	33.3%	6.60	17.40	0%	25%	14.20
TKI alone	0%	33.3%	2.90	9.50	0%	25%	14.00
IO alone	0%	33.3%	3.10	12.10	0%	25%	9.10
Non-PBC	0%	0%	—	—	0%	25%	8.70
Amivantamab	0%	0%	—	—	50%	0%	23.95
Mobocertinib	0%	0%	—	—	50%	0%	22.80

*EGFR*ex20ins, epidermal growth factor receptor gene exon 20 insertions; IO, immune-oncology therapy; OS, overall survival; PBC, platinum-based chemotherapy; PFS, progression-free survival

Results

- In the base-case scenario, the mean number of life years associated with NGS and sgPCR were 3.21 and 2.62, respectively, for an additional 0.59 years (7.08 months) in favor of NGS
- Varying the accuracy of NGS and sgPCR simultaneously, the difference in number of incremental life years between NGS and sgPCR ranged from 0.44 to 1.24 years, with an average of 0.84 years (**Figure 2**)

Figure 2. Incremental Life Years for NGS vs sgPCR Varying Ex20ins Variant Detectability

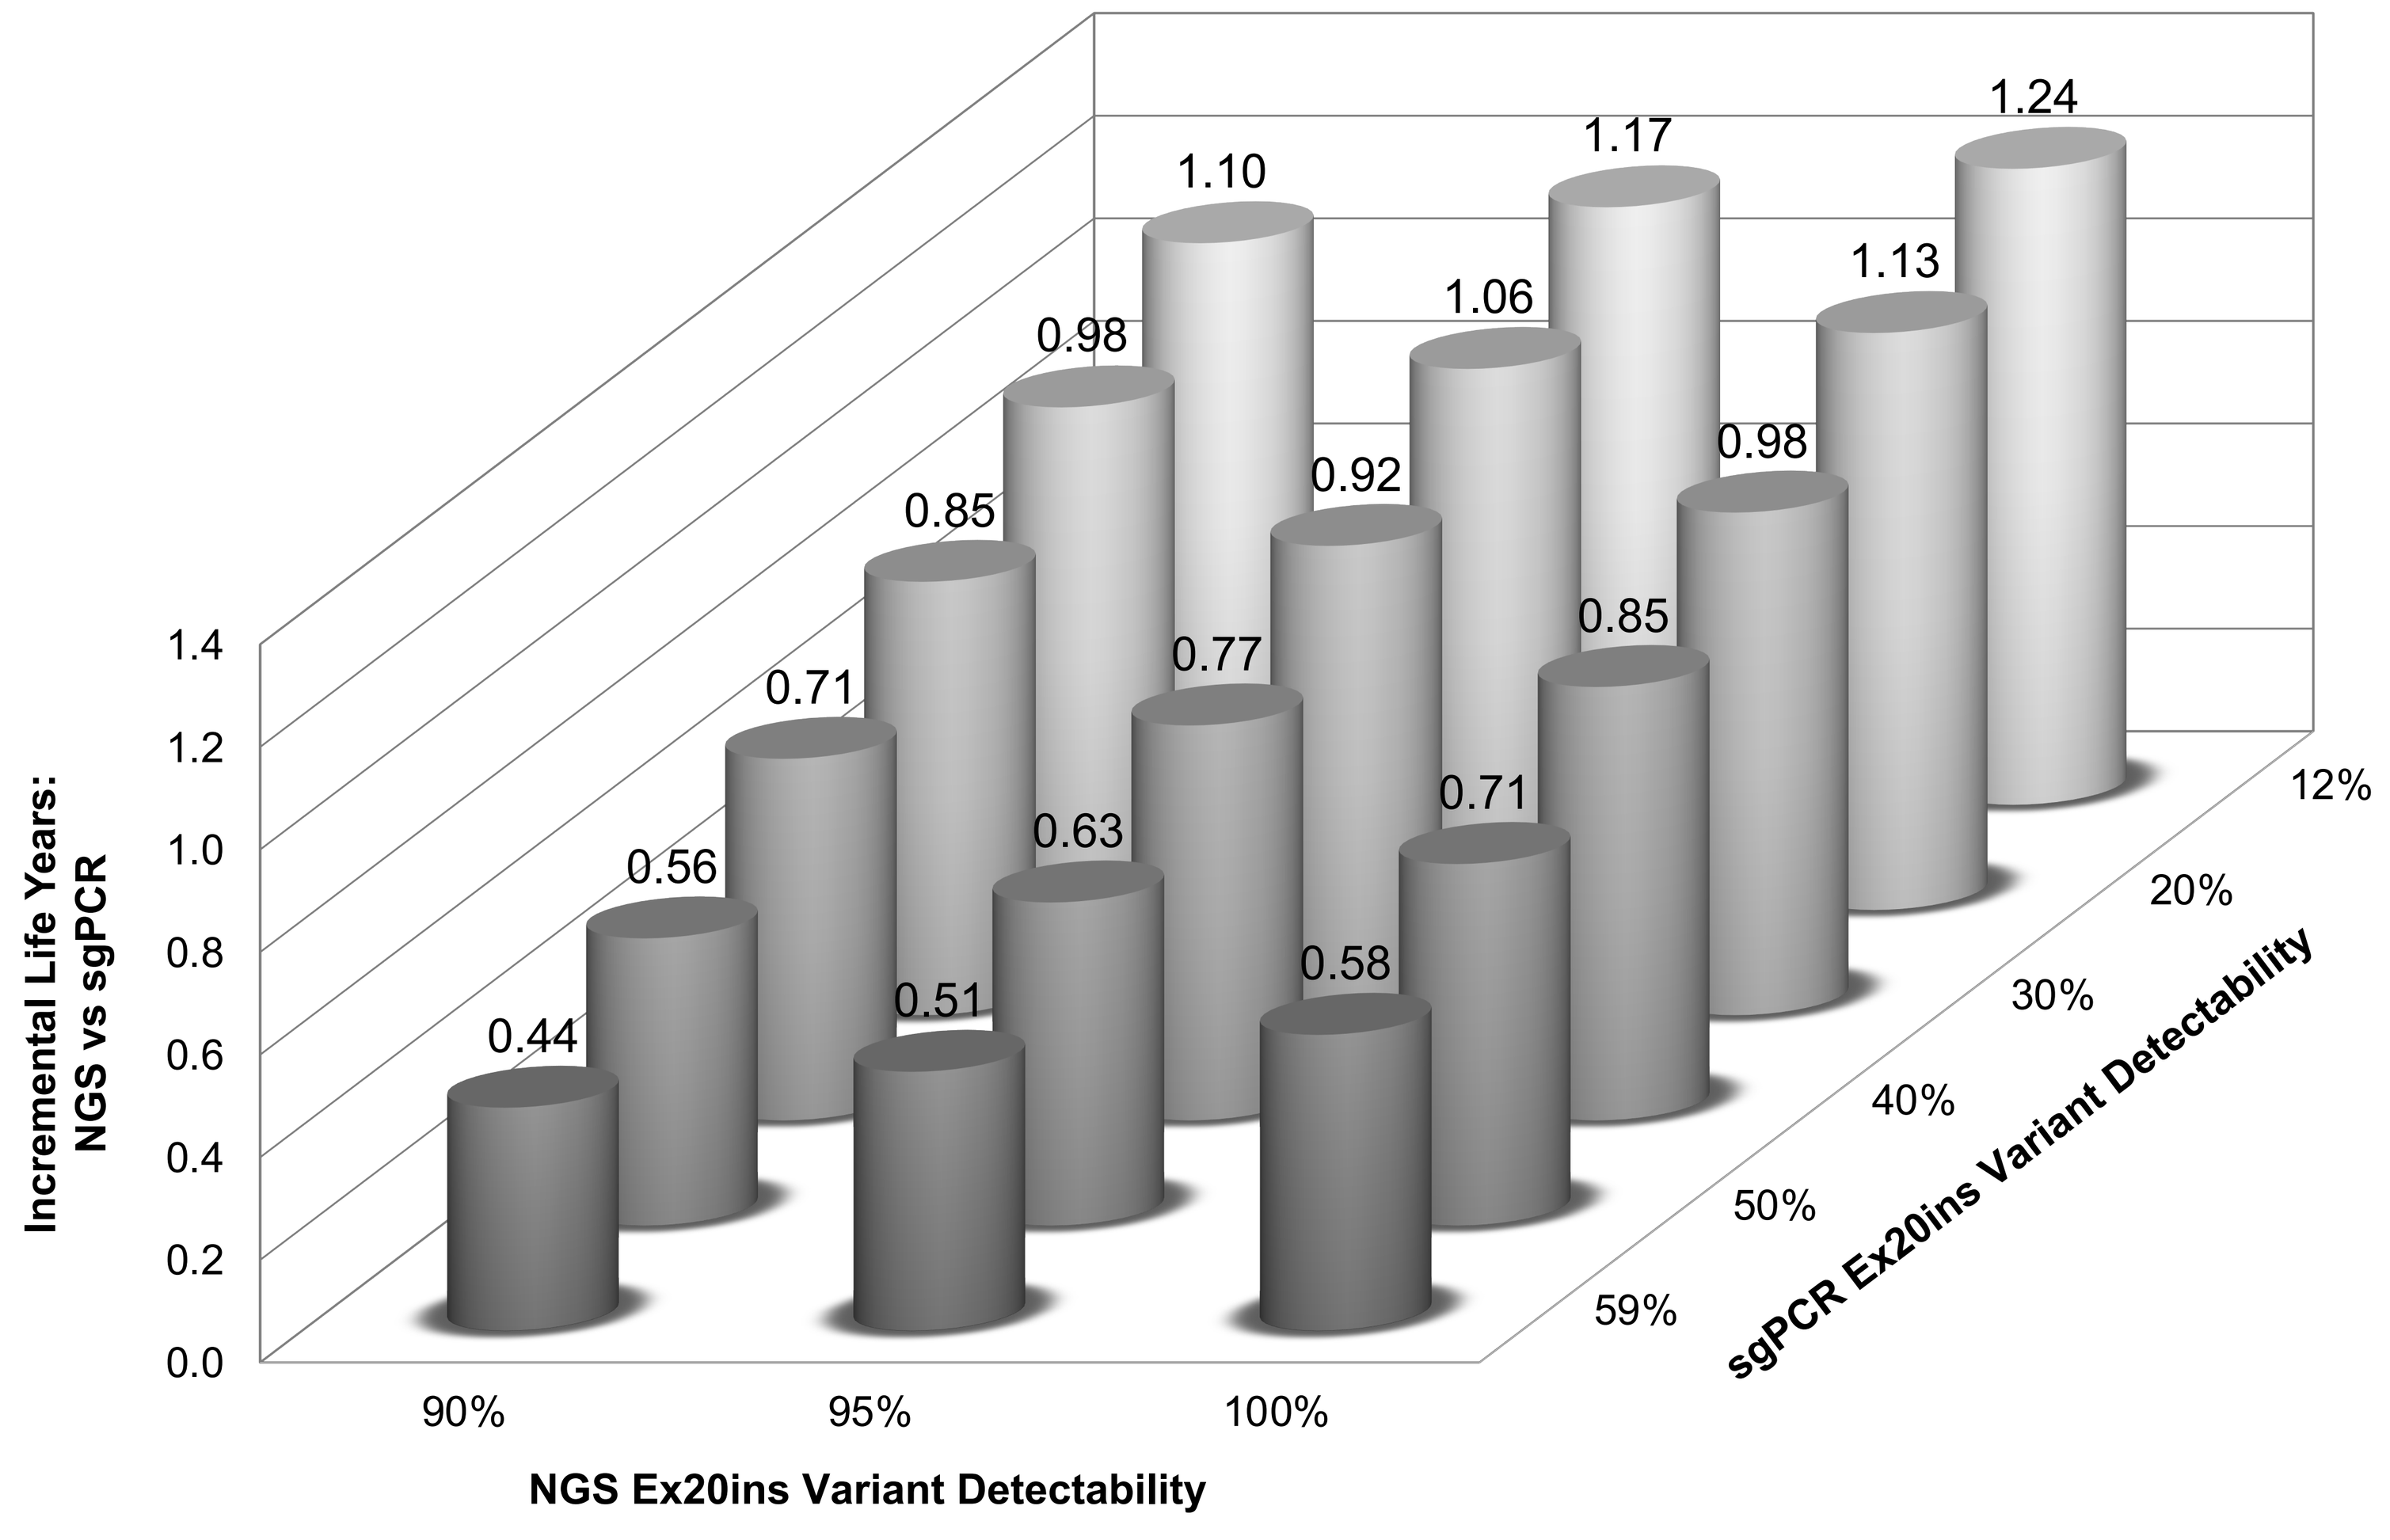

Incremental Life Years: NGS vs sgPCR
0.0
0.2
0.4
0.6
0.8
1.0
1.2
1.4
NGS Ex20ins Variant Detectability
90%
95%
100%
sgPCR Ex20ins Variant Detectability
59%
50%
40%
30%
20%
12%
0.44
0.56
0.71
0.85
0.98
1.10
0.51
0.63
0.77
0.92
1.06
1.17
0.58
0.71
0.85
0.98
1.13
1.24

ex20ins, exon 20 insertions; NGS, next-generation sequencing; sgPCR, single-gene polymerase chain reaction

- Additional scenarios were evaluated, varying the ex20ins variant detectability with NGS and sgPCR and the distribution of treatments in first line and second line. In all scenarios, testing with NGS was associated with additional life years vs testing with sgPCR ranging from 0.40 to 1.36 years (**Table 2**)
 - The highest number of life years associated with NGS and sgPCR were 3.26 and 2.85, respectively (highlighted in black in **Table 2**). The lowest number of life years associated with NGS and sgPCR were 2.97 and 1.62, respectively (highlighted in red in **Table 2**)

Table 2. Scenarios Varying Distribution of Treatments in First and Second Line

Scenario								
Ex20ins Detectability		Line 1		Line 2		Mean Life Years		Incremental Life Years
NGS (%)	sgPCR (%)	*EGFR*ex20ins detected	*EGFR*ex20ins not detected	*EGFR*ex20ins detected*	*EGFR*ex20ins not detected	NGS	sgPCR	NGS vs sgPCR
100	59	PBC	PBC	Mobocertinib/ Amivantamab	TKI	3.21	2.81	0.40
100	59	PBC	PBC	Mobocertinib/ Amivantamab	IO	3.21	2.61	0.60
100	59	PBC	PBC	Mobocertinib/ Amivantamab	Non-PBC	3.21	2.59	0.62
100	59	PBC	TKI	Mobocertinib/ Amivantamab	PBC	3.21	2.70	0.51
100	59	PBC	TKI	Mobocertinib/ Amivantamab	IO	3.21	2.46	0.75
100	59	PBC	TKI	Mobocertinib/ Amivantamab	Non-PBC	3.21	2.45	0.76
100	59	PBC	IO	Mobocertinib/ Amivantamab	PBC	3.21	2.73	0.48
100	59	PBC	IO	Mobocertinib/ Amivantamab	TKI	3.21	2.72	0.49
100	59	PBC	IO	Mobocertinib/ Amivantamab	Non-PBC	3.21	2.47	0.74
100	59	PBC	PBC	Mobocertinib	TKI	3.26	2.85	0.41
90	12	PBC	TKI	Amivantamab	Non-PBC	2.97	1.62	1.36

* If more than one treatment, they were equally distributed.
*EGFR*ex20ins, epidermal growth factor receptor gene exon 20 insertions; IO, immune-oncology therapy; NGS, next-generation sequencing; PBC, platinum-based chemotherapy; sgPCR, single-gene polymerase chain reaction; TKI, tyrosine kinase inhibitor

Limitations

- In the absence of patient-level data for all comparators, PFS and OS were extrapolated based on an exponential model
- The impact of any subsequent treatment post-second line was assumed to be implicitly captured in the OS of the second-line treatments and there could be imbalances in the distribution of subsequent treatment post-second line between the data sources used to inform the median PFS and OS

Conclusions and Recommendations

- With the development and recent approval of targeted therapies for patients with *EGFR*ex20ins-positive NSCLC, understanding the impact of sgPCR detection limitations on outcomes is becoming more important
- NGS can improve the detection of ex20ins variants and is associated with additional life years vs sgPCR. In markets where NGS may not be routinely used, other technologies with similar detection rates, such as multiplex PCR,[15] would also be expected to be associated with additional life years vs sgPCR
- Future models should quantify the impact of improved detection of ex20ins variants with NGS vs sgPCR in quality-adjusted life years and costs

References

1. Duma N, Santana-Davila R, Molina JR. Non-small cell lung cancer: epidemiology, screening, diagnosis, and treatment. *Mayo Clin Proc*. 2019;94(8):1623-1640. **2.** Gregg JP, Li T, Yoneda KY. Molecular testing strategies in non-small cell lung cancer: optimizing the diagnostic journey. *Transl Lung Cancer Res*. 2019;8(3):286-301. **3.** Oxnard GR, Lo PC, Nishino M, et al. Natural history and molecular characteristics of lung cancers harboring *EGFR* exon 20 insertions. *J Thorac Oncol*. 2013;8(2):179-184. **4.** Burnett H, Emich H, Carroll C, Stapleton N, Mahadevia P, Li T. Epidemiological and clinical burden of *EGFR* exon 20 insertion in advanced non-small cell lung cancer: a systematic literature review. *PLoS One*. 2021;16(3):e0247620. **5.** Riess JW, Gandara DR, Frampton GM, et al. Diverse *EGFR* exon 20 insertions and co-occurring molecular alterations identified by comprehensive genomic profiling of NSCLC. *J Thorac Oncol*. 2018;13(10):1560-1568. **6.** Fang W, Huang Y, Hong S, et al. EGFR exon 20 insertion mutations and response to osimertinib in non-small-cell lung cancer. *BMC Cancer*. 2019;19(1):595. **7.** Yasuda H, Kobayashi S, Costa DB. *EGFR* exon 20 insertion mutations in non-small-cell lung cancer: preclinical data and clinical implications. *Lancet Oncol*. 2012;13(1):e23-31. **8.** Rybrevant (amivantamab-vmjw) injection, for intravenous use - Prescribing Information. 2021. https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/761210s000lbl.pdf. Accessed: June 25, 2021. **9.** Exkivity (mobocertinib) capsules, for oral use - Prescribing Information. 2021. https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/215310s000lbl.pdf. Accessed: October 7, 2021. **10.** Ou S, Hong J, Lin H, et al. Distribution and detectability of EGFR exon 20 insertion variants in non–small cell lung cancer [abstract MA14.01]. 2021. https://library.iaslc.org/conference-program?product_id=24. Accessed: October 7, 2021. **11.** Behjati S, Tarpey PS. What is next generation sequencing? *Arch Dis Child Educ Pract Ed*. 2013;98(6):236-238. **12.** Girard N, Bazhenova L, Minchom A, et al. Comparative clinical outcomes for patients with NSCLC harboring EGFR exon 20 insertion mutations and common EGFR mutations [abstract MA04.07]. *J Thorac Oncol*. 2021;16(3):S145-S146. **13.** Park K, Haura EB, Leighl NB, et al. Amivantamab in EGFR exon 20 insertion-mutated non-small-cell lung cancer progressing on platinum chemotherapy: initial results from the CHRYSALIS Phase I Study. *J Clin Oncol*. 2021:JCO2100662. **14.** Ramalingam SS, Zhou C, Kim TM, et al. Mobocertinib (TAK-788) in *EGFR* exon 20 insertion (ex20ins)+ metastatic NSCLC (mNSCLC): additional results from platinum-pretreated patients (pts) and EXCLAIM cohort of phase 1/2 study [abstract 9014]. *J Clin Oncol*. 2021;39(15_suppl):9014. **15.** Cernomaz AT, Macovei II, Pavel I, et al. Comparison of next generation sequencing, SNaPshot assay and real-time polymerase chain reaction for lung adenocarcinoma EGFR mutation assessment. *BMC Pulm Med*. 2016;16(1):88.

Abbreviations

EGFR, epidermal growth factor receptor gene; ex20ins, exon 20 insertions; IO, immune-oncology therapy; NGS, next-generation sequencing; NSCLC, non–small cell lung cancer; OS, overall survival; PBC, platinum-based chemotherapy; PCR, polymerase chain reaction; PD, progressive disease; PF, progression free; PFS, progression-free survival; sgPCR, single-gene polymerase chain reaction; TKI, tyrosine kinase inhibitor

Acknowledgments

This study was sponsored by Millennium Pharmaceuticals, Inc., Cambridge, MA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

Disclosures

Luis G Hernandez: Employment (Takeda)
Eric N Churchill: Employment (Takeda)
Laura J Walton: Employment (Takeda)

Email for questions or comments: luis.hernandez3@takeda.com

For an e-Print, scan this QR code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at ISPOR-EU (European Congress of the International Society for Pharmacoeconomics and Outcomes Research), 30 November – 3 December 2021 (Virtual)