POSC28

Systematic Literature Review of Second-line Treatments for Small-cell Lung Cancer

Virtual International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe

30 November-3 December 2021

Raj Hanvesakul,[1,*] Badri Rengarajan,[2] Catharine Stack,[3,†] Florence R. Wilson,[4] Adekemi Adeyemi[4]

[1]Jazz Pharmaceuticals, Oxford, England, UK; [2]Jazz Pharmaceuticals, Palo Alto, CA, USA; [3]Former employee of Jazz Pharmaceuticals, Philadelphia, PA, USA; [4]PRECISIONheor, Vancouver, BC, Canada.

*Presenting author.

†Catharine Stack was an employee of Jazz Pharmaceuticals, Inc., at the time this research was conducted.

Background

- Lurbinectedin (3.2 mg/m² every 3 weeks; intravenous [IV], 1-hour infusion) received accelerated approval from the US Food and Drug Administration (June 2020) for the treatment of adult patients with metastatic small-cell lung cancer (SCLC) with disease progression on or after platinum-based chemotherapy[1] based on results of the SCLC cohort from a phase 2 basket trial[2]
 - A single-arm, phase 2 basket trial of lurbinectedin (ClinicalTrials.gov Identifier: NCT02454972) demonstrated an overall response rate (ORR) of 35.2%, a median duration of response (DoR) of 5.3 months, and a median overall survival (OS) of 9.3 months, as measured by investigator assessment (data cutoff: January 15, 2019)[2]
- Head-to-head randomised controlled trials (RCTs) comparing the efficacy of lurbinectedin monotherapy with other second-line SCLC therapies are not available

Objective

- We conducted a systematic literature review (SLR) to identify clinical trials of second-line SCLC therapies recommended by the National Comprehensive Cancer Network (NCCN) guidelines and assessed the feasibility of an indirect treatment comparison (ITC) to estimate the relative efficacy and safety of lurbinectedin versus relevant comparators

Methods

SLR

- A comprehensive database search using predefined criteria (summarised in **Table 1**) included:
 - Published data from EMBASE, MEDLINE, and the Cochrane Central Register of Controlled Trials
 - Grey literature search results from the US National Institutes of Health Clinical Trial Registry, the American Society of Clinical Oncology (ASCO), the American Association for Cancer Research (AACR), and the International Association for the Study of Lung Cancer Word Conference on Lung Cancer (IASLC WCLC)
 - The bibliographies of recently published relevant systematic reviews and meta-analyses were also searched
- Searches were executed on January 6, 2021; only studies published in English from 1990 to January 2021 were included
- Two independent reviewers conducted abstract/full-text selection and data extraction

Unanchored Matching-adjusted Indirect Comparison (MAIC) and Network Meta-analysis (NMA)

- An unanchored MAIC was performed to connect the single-arm lurbinectedin study (Trigo 2020) to the connected network of RCTs identified through the SLR
- A fixed-effect NMA using a Bayesian framework was subsequently performed to evaluate the comparative efficacy of lurbinectedin versus relevant competing interventions
 - Although both fixed- and random-effects models were considered for all analyses, fixed-effect models were preferred given that all evidence networks consisted of relatively few trials, which caused the random-effects models to lead to unstable estimates
- Relative effectiveness was assessed with hazard ratios (HRs) and odds ratio (ORs) with 95% credible intervals (CrIs)

Table 1. Eligibility Criteria for Study Screening

Criteria	Inclusion
Population	Adults ≥18 years of age with 2L SCLC and ≥1 prior platinum-based chemotherapy regimen **Subgroups of interest:** • Age: <65 years; >65 years • Platinum-sensitivity status: sensitive (CTFI ≥90 days); resistant (CTFI <90 days) • Performance status: ECOG = 0; ECOG = 1; ECOG = 2 • Stage at diagnosis: LS SCLC; ES SCLC
Interventions[a]	Treatments recommended as subsequent systemic therapy in the NCCN guidelines (v1.2021)[b]: • Lurbinectedin • Topotecan (oral or IV) • Platinum rechallenge with carboplatin or cisplatin ± etoposide or irinotecan • Paclitaxel, docetaxel, irinotecan, nivolumab, pembrolizumab
Comparators	• Placebo or BSC • Any intervention of interest • Any treatment that facilitates an indirect comparison
Outcomes	At least 1 of the following efficacy outcomes: • OS, PFS/TTP, tumor response (ORR, CR, PR, SD, PD, CBR, DCR), and DoR
Study design[c]	• Phase 1/2, 2, or 3 RCTs • In a stepwise manner, where no RCTs were available, nonrandomised prospective clinical trials or single-arm clinical trials (phase 1/2, 2, or 3) were included[d]

2L, second-line; SCLC, small-cell lung cancer; CTFI, chemotherapy-free interval; ECOG, Eastern Cooperative Oncology Group; LS, limited-stage; ES, extensive-stage; NCCN, National Comprehensive Cancer Network; IV, intravenous; BSC, best supportive care; OS, overall survival; PFS, progression-free survival; TTP, time to progression; ORR, overall response rate; CR, complete response; PR, partial response; SD, stable disease; PD, progressive disease; CBR, clinical benefit rate; DCR, disease control rate; DoR, duration of response; RCT, randomised controlled trial; CAV, cyclophosphamide + doxorubicin + vincristine.
[a]Only monotherapy regimens were included unless otherwise specified. [b]Excluded treatments: nonpharmacologic interventions (eg, surgery, radiotherapy), lurbinectedin combination therapy, nivolumab + ipilimumab, gemcitabine, bendamustine, etoposide monotherapy, temozolomide, vinorelbine, and CAV. [c]Long-term follow-up and *post hoc* analyses of relevant studies were included. [d]Included for an intervention of interest only when comparative studies were unavailable.

Results

- Of 4,769 citations identified, 10 unique relevant trials were included (5 RCTs, 2 single-arm trials, 2 RCTs with 1 arm of interest, and 1 trial with randomised and nonrandomised cohorts with 1 arm of interest; **Figure 1**)

Figure 1. Flow Diagram for the SLR

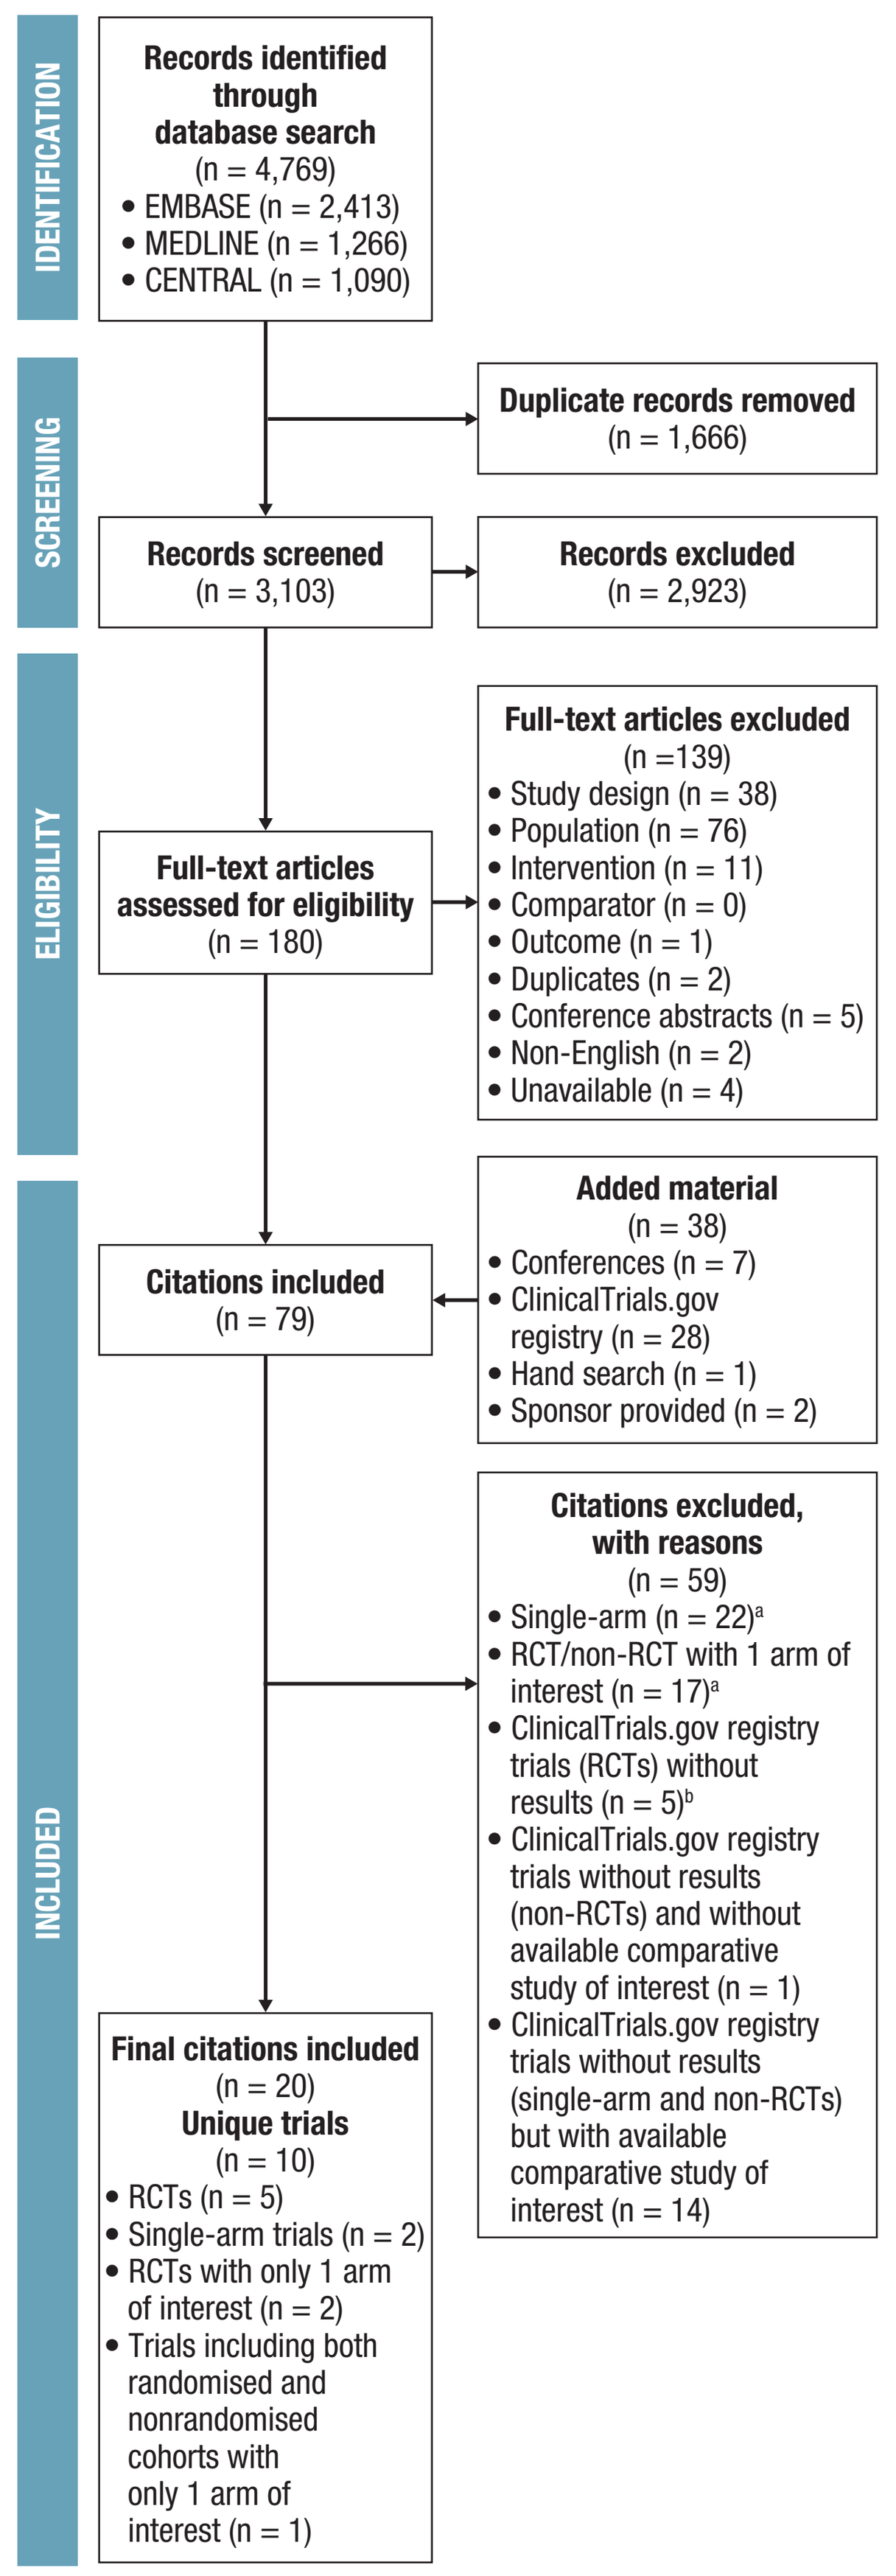

IDENTIFICATION
Records identified through database search (n = 4,769)
• EMBASE (n = 2,413)
• MEDLINE (n = 1,266)
• CENTRAL (n = 1,090)
SCREENING
Duplicate records removed (n = 1,666)
Records screened (n = 3,103)
Records excluded (n = 2,923)
ELIGIBILITY
Full-text articles assessed for eligibility (n = 180)
Full-text articles excluded (n =139)
• Study design (n = 38)
• Population (n = 76)
• Intervention (n = 11)
• Comparator (n = 0)
• Outcome (n = 1)
• Duplicates (n = 2)
• Conference abstracts (n = 5)
• Non-English (n = 2)
• Unavailable (n = 4)
INCLUDED
Added material (n = 38)
• Conferences (n = 7)
• ClinicalTrials.gov registry (n = 28)
• Hand search (n = 1)
• Sponsor provided (n = 2)
Citations included (n = 79)
Citations excluded, with reasons (n = 59)
• Single-arm (n = 22)[a]
• RCT/non-RCT with 1 arm of interest (n = 17)[a]
• ClinicalTrials.gov registry trials (RCTs) without results (n = 5)[b]
• ClinicalTrials.gov registry trials without results (non-RCTs) and without available comparative study of interest (n = 1)
• ClinicalTrials.gov registry trials without results (single-arm and non-RCTs) but with available comparative study of interest (n = 14)
Final citations included (n = 20)
Unique trials (n = 10)
• RCTs (n = 5)
• Single-arm trials (n = 2)
• RCTs with only 1 arm of interest (n = 2)
• Trials including both randomised and nonrandomised cohorts with only 1 arm of interest (n = 1)

SLR, systematic literature review; CENTRAL, Cochrane Central Register of Controlled Trials; RCT, randomised controlled trial.
[a]Single-arm studies were excluded when comparative RCTs were available for the intervention of interest. RCTs and non-RCTs with only 1 arm of interest were treated in a similar fashion to single-arm studies and excluded when comparative studies were available.
[b]Including 2 studies already captured by database search.

- Baseline patient characteristics were generally consistent across trials (**Table 2**), although there were differences in the proportion of patients with:
 - Limited-stage and extensive-stage SCLC
 - Eastern Cooperative Oncology Group (ECOG) performance status
 - Platinum-sensitive and platinum-resistant disease

Table 2. Summary of Baseline Patient Characteristics of Trials Identified in the SLR

Trial ID	Treatment	Analysis population	Line of therapy	N	ECOG PS, n (%) 0	ECOG PS, n (%) 1	ECOG PS, n (%) 2	Disease stage, n (%) LS	Disease stage, n (%) ES	Response to 1L, n (%) Resistant	Response to 1L, n (%) Sensitive
RCTs											
Baize 2020[3] (N = 164)	Topotecan (oral)	ITT, CTFI ≥90 d	2L	81	26 (32.1)	48 (59.3)	7 (8.6)	30 (37.0)	51 (63.0)	0 (0)	81 (100)
	Carboplatin + etoposide	ITT, CTFI ≥90 d	2L	81	28 (34.6)	48 (59.3)	5 (6.2)	29 (35.8)	52 (64.2)	0 (0)	81 (100)
O'Brien 2006[4] (N = 141)	BSC	ITT	2L	70	6 (8.6)	41 (58.6)	23 (32.9)	27 (38.6)	43 (61.4)	35 (50.0)	35 (50.0)
	Topotecan (oral) + BSC	ITT	2L	71	8 (11.3)	44 (62.0)	19 (26.8)	23 (32.4)	48 (67.6)	41 (57.7)	30 (42.3)
Eckardt 2007[5] (N = 309)	Topotecan (oral)	ITT, CTFI ≥90 d[a]	2L	153	48 (31.4)	85 (55.6)	20 (13.1)	51 (33.3)	102 (67.7)	15[b] (9.8)	134[b] (87.6)
	Topotecan (IV)	ITT, CTFI ≥90 d[a]	2L	151	35 (23.2)	98 (64.9)	18 (11.9)	45 (29.8)	106 (70.2)	13[b] (8.6)	137[b] (90.7)
von Pawel 2001[6] (N = 106)	Topotecan (oral)	ITT, CTFI ≥90 d[c]	2L	52	10[d] (19.2)	34[d] (65.4)	8[d] (15.4)	14[a] (26.9)	37[b] (71.2)	1 (1.9)	51 (98.1)
	Topotecan (IV)	ITT, CTFI ≥90 d[c]	2L	54	18[d] (33.3)	21[d] (38.9)	15[d] (27.8)	14[a] (25.9)	39[a] (72.2)	1 (1.9)	53 (98.1)
United Therapeutics 2017[7] (N = 483)	Irinotecan	ITT	2L	190	40 (21.1)	150 (78.9)	0 (0)	–	–	–	–
	Topotecan (IV)	ITT	2L	94	19 (20.2)	75 (79.8)	0 (0)	–	–	–	–
RCTs with only 1 arm of interest											
Owonikoko 2020[8] (N = 178)	Paclitaxel + placebo	ITT	2L	89	18 (20.2)	71 (79.8)	0 (0)	8[a] (9.0)	62[b] (69.7)	33[b,e] (37.1)	35[b,e] (39.3)
CheckMate 331[9] (N = 569)	Nivolumab	ITT	2L	284	75 (26.4)	209 (73.6)	0 (0)	74 (26.1)	210 (73.9)	121 (42.6)	163 (57.4)
Non-RCTs/RCTs with only 1 arm of interest											
CheckMate 032 – **Antonia 2016[10] (non-RCT; N = 216)**	Nivolumab	Overall	2L+	98[f]	–	–	–	–	–	30[b,g] (30.6)	55[b,g] (56.1)
CheckMate 032 – **Ready 2020[11] (RCT; N = 243)**	Nivolumab	ITT	2L+	147[f]	49 (33.3)	98 (66.7)	–	–	–	73[b] (49.7)	73[b] (49.7)
Single-arm trials											
Trigo 2020[2] (N = 105)[h]	Lurbinectedin	Treated population	2L	105	38 (36.2)	59 (56.2)	8 (7.6)	32 (30.5)	73 (69.5)	45 (42.9)	60 (57.1)
Smyth 1994[12] (N = 34)	Docetaxel	Overall	1L+	34	8[d] (23.5)	22[d] (64.7)	4[d] (11.8)	0 (0)	34 (100)	–	–

ECOG PS, Eastern Cooperative Oncology Group performance status; 1L, first-line; LS, limited-stage; ES, extensive-stage; RCT; randomised controlled trial; ITT, intent-to-treat; CTFI, chemotherapy-free interval; d, days; 2L, second-line; BSC, best supportive care; IV, intravenous; WHO, World Health Organization.
Note: Highlighting indicates similarity (green), some differences (yellow), and major differences (red) in characteristics based on visual assessment.
[a]Approximately 10% of patients in both treatment groups had a CTFI <90 days at study entry. [b]Missing patient data was reported. [c]Approximately 2% of patients in both treatment groups had a CTFI <90 days at study entry. [d]WHO performance status. [e]Platinum refractory, 16/89 (18%). [f]Mixed population, baseline characteristics for the 2L population not provided. [g]Three patients in the nivolumab 3 mg/kg group did not receive 1L platinum therapy and did not meet eligibility criteria, although they were treated and included in the analysis. [h]The study enrolled 110 patients; however, 5 patients did not receive treatment.

- Data extraction of patient characteristics and outcomes from the SLR does not reflect statistical comparison (**Table 3**)
- For ORR, tumour assessment was based on investigator assessment in 4 trials, independent review committee in 3 trials, both in 1 trial, and unknown assessors in 2 trials
- Among patients with platinum-sensitive disease (CTFI ≥90 days):
 - ORR ranged from 3.0% (O'Brien 2006) to 25.3% (Baize 2020) for oral or IV topotecan, 45.0% (Trigo 2020) for lurbinectedin, and 49.4% (Baize 2020) for platinum rechallenge with carboplatin plus etoposide
 - Median OS ranged from 5.8 months (von Pawel 2001) to 8.1 months (Eckardt 2007) for oral or IV topotecan, 7.5 months (Baize 2020) for platinum rechallenge with carboplatin plus etoposide, 7.6 months (CheckMate 331) for nivolumab, and 11.9 months (Trigo 2020) for lurbinectedin

Table 3. Select Efficacy Outcomes by Treatment in Patients With SCLC Receiving 2L Therapy in Trials Identified in the SLR

Trial ID	Treatment	Analysis population	N	CTFI ≥90 d, n (%)	ORR, % Overall	ORR, % CTFI ≥90 d	Median OS, mos (95% CI) Overall	Median OS, mos (95% CI) CTFI ≥90 d
RCTs								
Baize 2020[3] (N = 164)	Topotecan (oral)	ITT, CTFI ≥90 d	81	81 (100.0)	25.3[a,b]		7.4 (6.0, 8.7)	
	Carboplatin + etoposide	ITT, CTFI ≥90 d	81	81 (100.0)	49.4[a,b]		7.5 (5.4, 9.5)	
O'Brien 2006[4] (N = 141)	BSC	ITT	70	35 (50.0)	–	–	3.2[c] (2.6, 4.3)	–
	Topotecan (oral) + BSC	ITT	71	30 (42.3)	7.0[a]	3.0[a]	6.0[c] (4.2, 7.3)	–
Eckardt 2007[5] (N = 309)	Topotecan (oral)	ITT, CTFI ≥90 d[d]	153	134 (87.6)[e]	18.3[f]		7.6[c] (6.7, 9.8)	
	Topotecan (IV)	ITT, CTFI ≥90 d[d]	151	137 (90.7)[e]	21.9[f]		8.1[c] (7.1, 8.6)	
von Pawel 2001[6] (N = 106)	Topotecan (oral)	ITT, CTFI ≥90 d[g]	52	51 (98.1)	23.1[f]		7.4[c] (range: 0.1, 15.9)	
	Topotecan (IV)	ITT, CTFI ≥90 d[g]	54	53 (98.1)	14.8[f]		5.8[c] (range: 0.1, 15.0)	
United Therapeutics 2017[7] (N = 483)	Irinotecan	ITT	190	–	18.9	–	7.0 (5.6, 8.9)	–
	Topotecan (IV)	ITT	94	–	20.2	–	7.4 (6.1, 9.3)	–
RCTs with only 1 arm of interest								
Owonikoko 2020[8] (N = 178)	Paclitaxel + placebo	ITT, CTFI <180 d[h]	89	35 (39.3)[e]	18.0[f]	–	5.42	–
CheckMate 331[9] (N = 569)	Nivolumab	ITT	284[i]	163 (57.4)	13.7[a]	–	7.5 (5.6, 9.2)	7.6 (5.6, 11.2)
Non-RCTs/RCTs with only 1 arm of interest								
CheckMate 032 – **Antonia 2016[10] (non-RCT; N = 216)**	Nivolumab	Subgroup, 2L	40[j]	–	10.0[a]	–	–	–
CheckMate 032 – **Ready 2020[11] (RCT; N = 243)**	Nivolumab	Subgroup, 2L	–	–	–	–	6.0 (3.8, 9.4)	–
Single-arm trials								
Trigo 2020[2] (N = 105)[k]	Lurbinectedin	Treated population	105	60 (57.1)	35.2[a]	45.0[a]	9.3 (6.3, 11.8)	11.9 (9.7, 16.2)
Smyth 1994[12] (N = 34)	Docetaxel	Subgroup, 2L	22[l]	–	18.0	–	–	–

SCLC, small-cell lung cancer; 2L, second-line; SLR, systematic literature review; CTFI, chemotherapy-free interval; d, days; ORR, overall response rate; OS, overall survival; mos, months; CI, confidence interval; RCT, randomised controlled trial; ITT, intent-to-treat; BSC, best supportive care; IV, intravenous; IA, investigator assessment; IRC, independent review committee; 1L, first-line.
[a]IA. [b]Nonevaluable patients were excluded from analysis (n = 6 in topotecan arm; n = 2 in carboplatin + etoposide arm). [c]Median OS was reported in weeks. [d]Approximately 10% of patients in both treatment groups had a CTFI <90 days at study entry. [e]Missing patient data was reported. [f]IRC assessed. [g]Approximately 2% of patients in both treatment groups had a CTFI <90 days at study entry. [h]Protocol Amendment 2 (January 2015) corrected the stratification definition for time to relapse to follow standard guidance by counting from the date of the last dose of frontline chemotherapy as opposed to counting from the initial response to 1L platinum-based chemotherapy, as originally set out in the protocol. Data shown in the table are for analysis done after protocol amendment. [i]13% and 19.3% of patients had missing data in the nivolumab and investigator's choice arms, respectively. [j]3% of patients had missing data. [k]The study enrolled 110 patients; however, 5 patients did not receive treatment. [l]A total of 28 patients were evaluable; of these, 22 were 2L patients.

- Of the 10 trials included in the feasibility assessment, 5 RCTs formed a connected network (Baize 2020, O'Brien 2006, von Pawel 2001, Eckardt 2007, United Therapeutics 2017) while 5 trials were disconnected (Trigo 2020, Smyth 1994, CheckMate 032, CheckMate 331, Owonikoko 2020; **Figure 2**)

Figure 2. Full Evidence Network of Trials Identified From the SLR and Included in the Feasibility Assessment

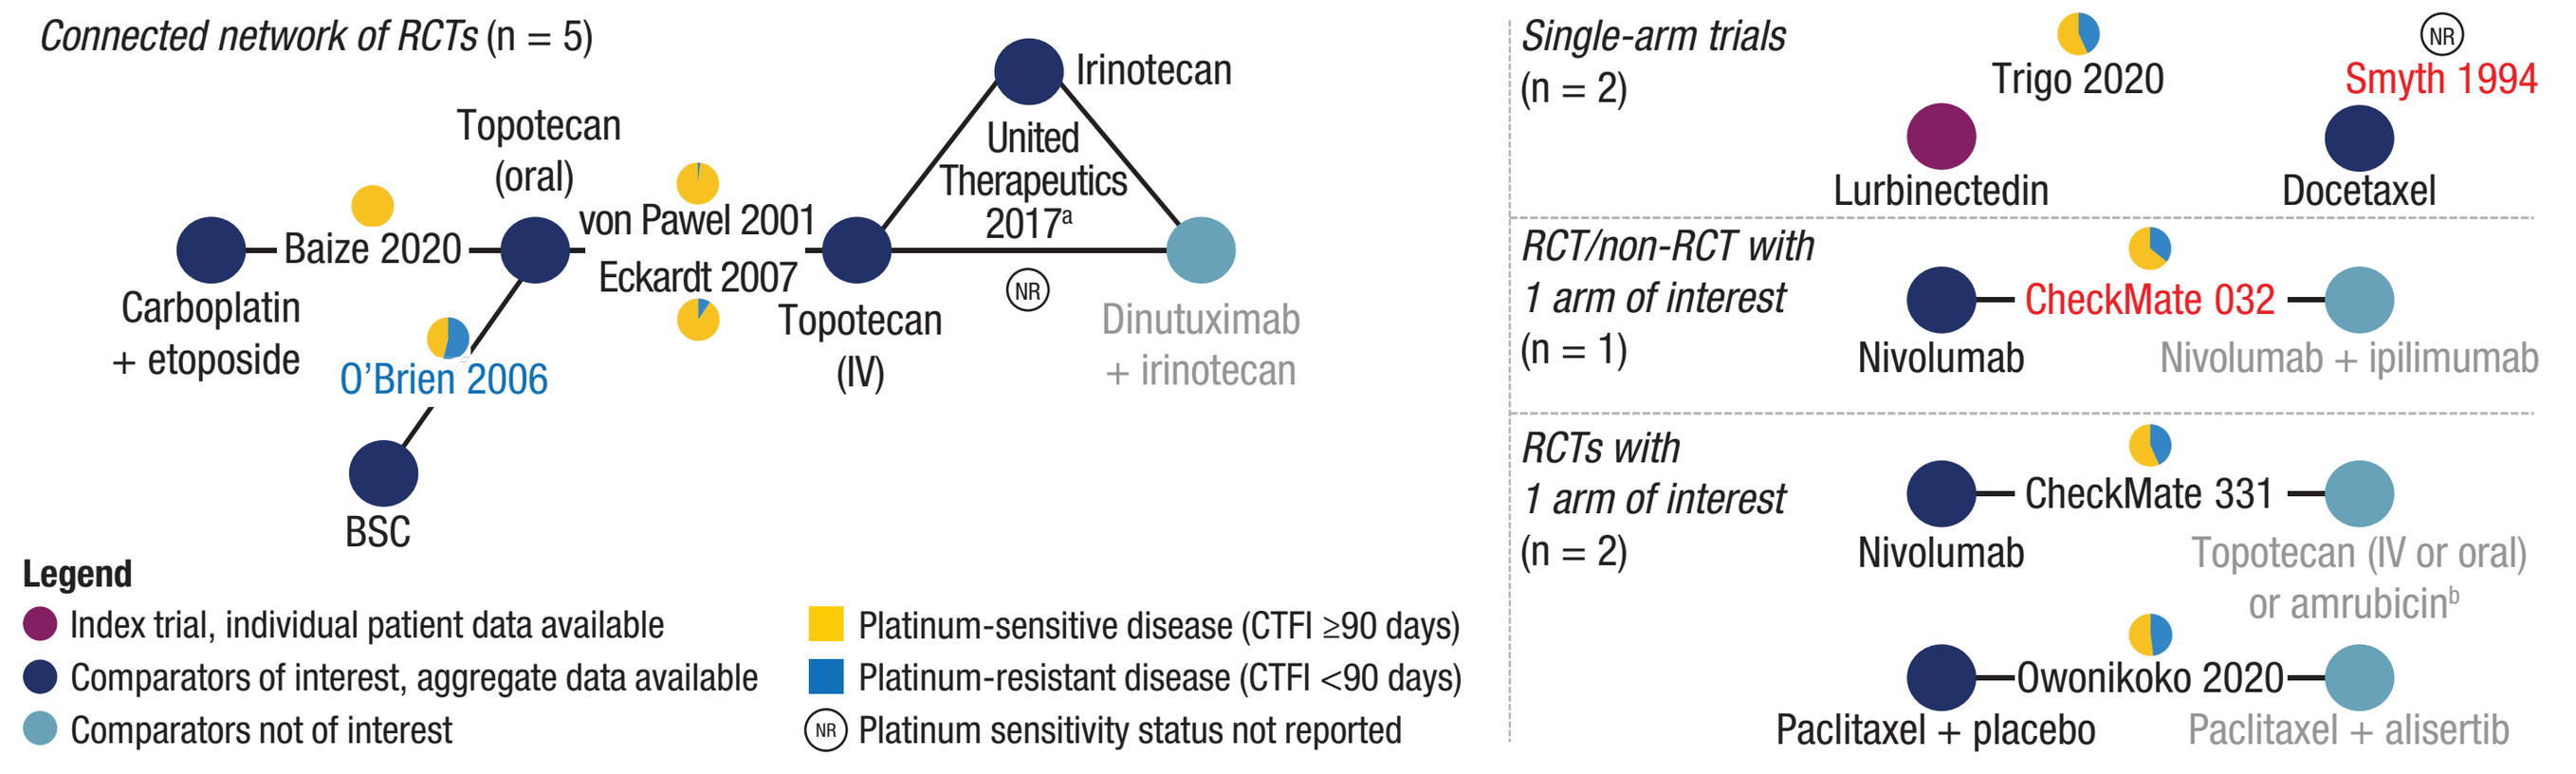

Connected network of RCTs (n = 5)
Carboplatin + etoposide
Baize 2020
Topotecan (oral)
O'Brien 2006
BSC
von Pawel 2001
Eckardt 2007
Topotecan (IV)
Irinotecan
United Therapeutics 2017[a]
NR
Dinutuximab + irinotecan
Single-arm trials (n = 2)
Trigo 2020
Lurbinectedin
NR
Smyth 1994
Docetaxel
RCT/non-RCT with 1 arm of interest (n = 1)
Nivolumab
CheckMate 032
Nivolumab + ipilimumab
RCTs with 1 arm of interest (n = 2)
Nivolumab
CheckMate 331
Topotecan (IV or oral) or amrubicin[b]
Paclitaxel + placebo
Owonikoko 2020
Paclitaxel + alisertib
Legend
Index trial, individual patient data available
Comparators of interest, aggregate data available
Comparators not of interest
Platinum-sensitive disease (CTFI ≥90 days)
Platinum-resistant disease (CTFI <90 days)
NR Platinum sensitivity status not reported

SLR, systematic literature review; RCT, randomised controlled trial; NR, not reported; IV, intravenous; BSC, best supportive care; CTFI, chemotherapy-free interval.
Note: Red text: trials excluded from analyses. Blue text: trial excluded from the base case. Grey text: interventions not relevant based on predefined eligibility criteria for the SLR.
[a]United Therapeutics 2017 is a 2-part study; only Part 2 is the relevant RCT (Part 1 = dose escalation). [b]Investigator's choice: intervention considered not relevant given that results are not stratified by treatment.

- Six of the 10 trials included in the feasibility assessment were excluded from the ITC due to various reasons (eg, lack of data availability, differences in patient characteristics, and being disconnected from the network of RCTs)
- Given the differences in the disease sensitivity status of patients across trials, the base case ITC analyses were restricted to the platinum-sensitive population
- Three RCTs (Baize 2020, von Pawel 2001, Eckardt 2007) had sufficiently similar platinum-sensitive populations and were assessed to identify which should be used to connect Trigo 2020 to the network for the base case ITC analyses (**Figure 3**)

Figure 3. Network of Trials Included in the Base Case ITC for Patients With Platinum-sensitive Disease

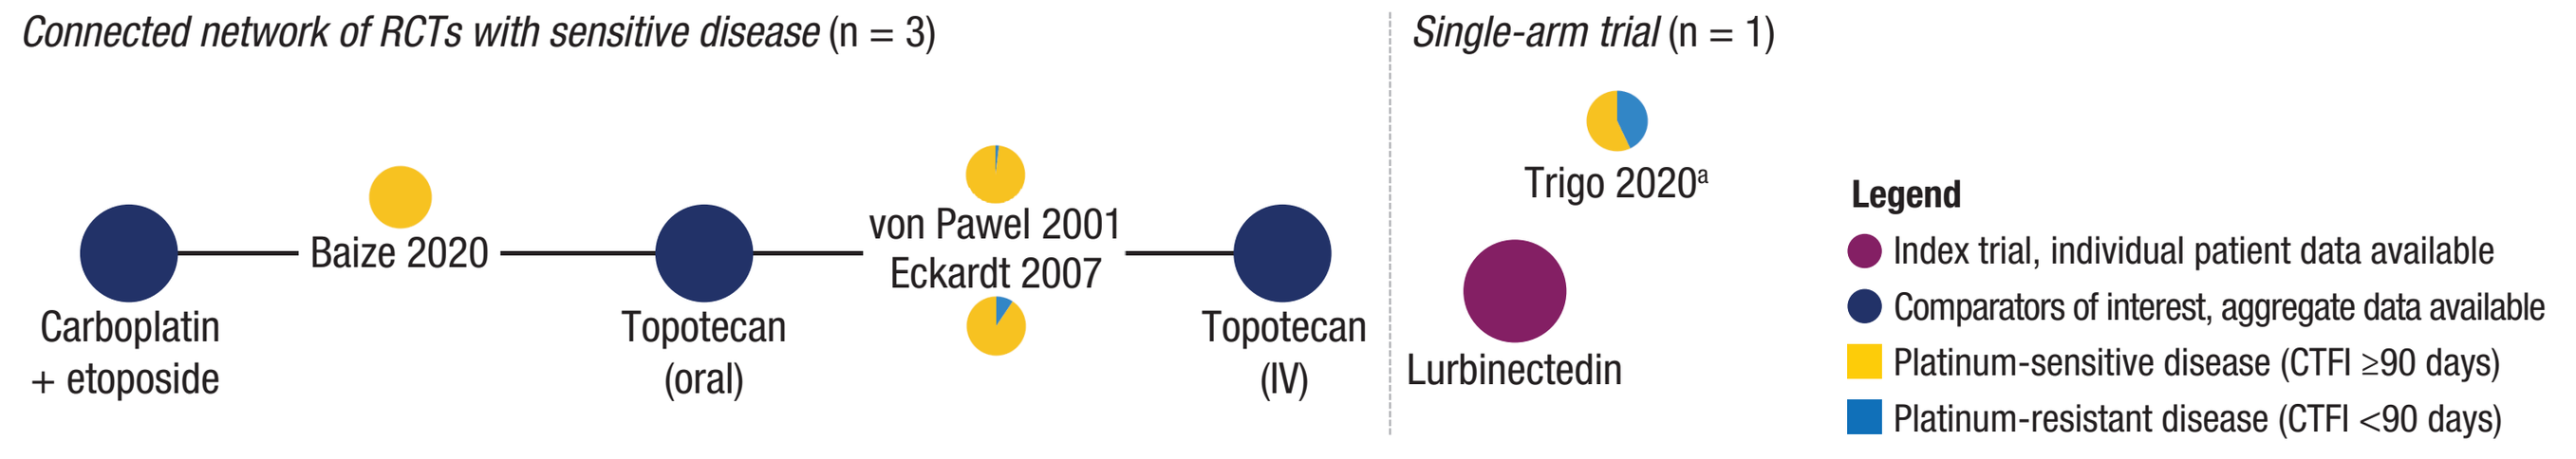

Connected network of RCTs with sensitive disease (n = 3)
Carboplatin + etoposide
Baize 2020
Topotecan (oral)
von Pawel 2001
Eckardt 2007
Topotecan (IV)
Single-arm trial (n = 1)
Trigo 2020[a]
Lurbinectedin
Legend
Index trial, individual patient data available
Comparators of interest, aggregate data available
Platinum-sensitive disease (CTFI ≥90 days)
Platinum-resistant disease (CTFI <90 days)

ITC, indirect treatment comparison; RCT, randomised controlled trial; IV, intravenous; CTFI, chemotherapy-free interval.
[a]Based on the CTFI ≥90 days subgroup.

- Based on the similarity of patient characteristics, Baize 2020 was selected as the external RCT to connect Trigo 2020 to the base case network in patients with platinum-sensitive disease (**Figure 4**)
 - In the base case fixed-effect NMA, lurbinectedin demonstrated a significantly greater OS compared to oral topotecan (HR: 0.43; 95% CrI: 0.27, 0.67), IV topotecan (HR: 0.43; 95% CrI: 0.26, 0.70), and carboplatin plus etoposide (HR: 0.42; 95% CrI: 0.30, 0.58)
 - Odds ratio estimates from the base case fixed-effect NMA also found lurbinectedin to be associated with a significantly greater ORR than oral topotecan (OR: 2.58; 95% CrI: 1.12, 5.94); however, statistically significant differences were not observed for lurbinectedin versus IV topotecan (OR: 2.36; CrI: 0.89, 6.23) and versus carboplatin plus etoposide (OR: 0.85; 95% CrI: 0.40, 1.83)

Figure 4. Evidence Network for Base Case ITC Analysis in Patients With Platinum-sensitive Disease for ORR and OS

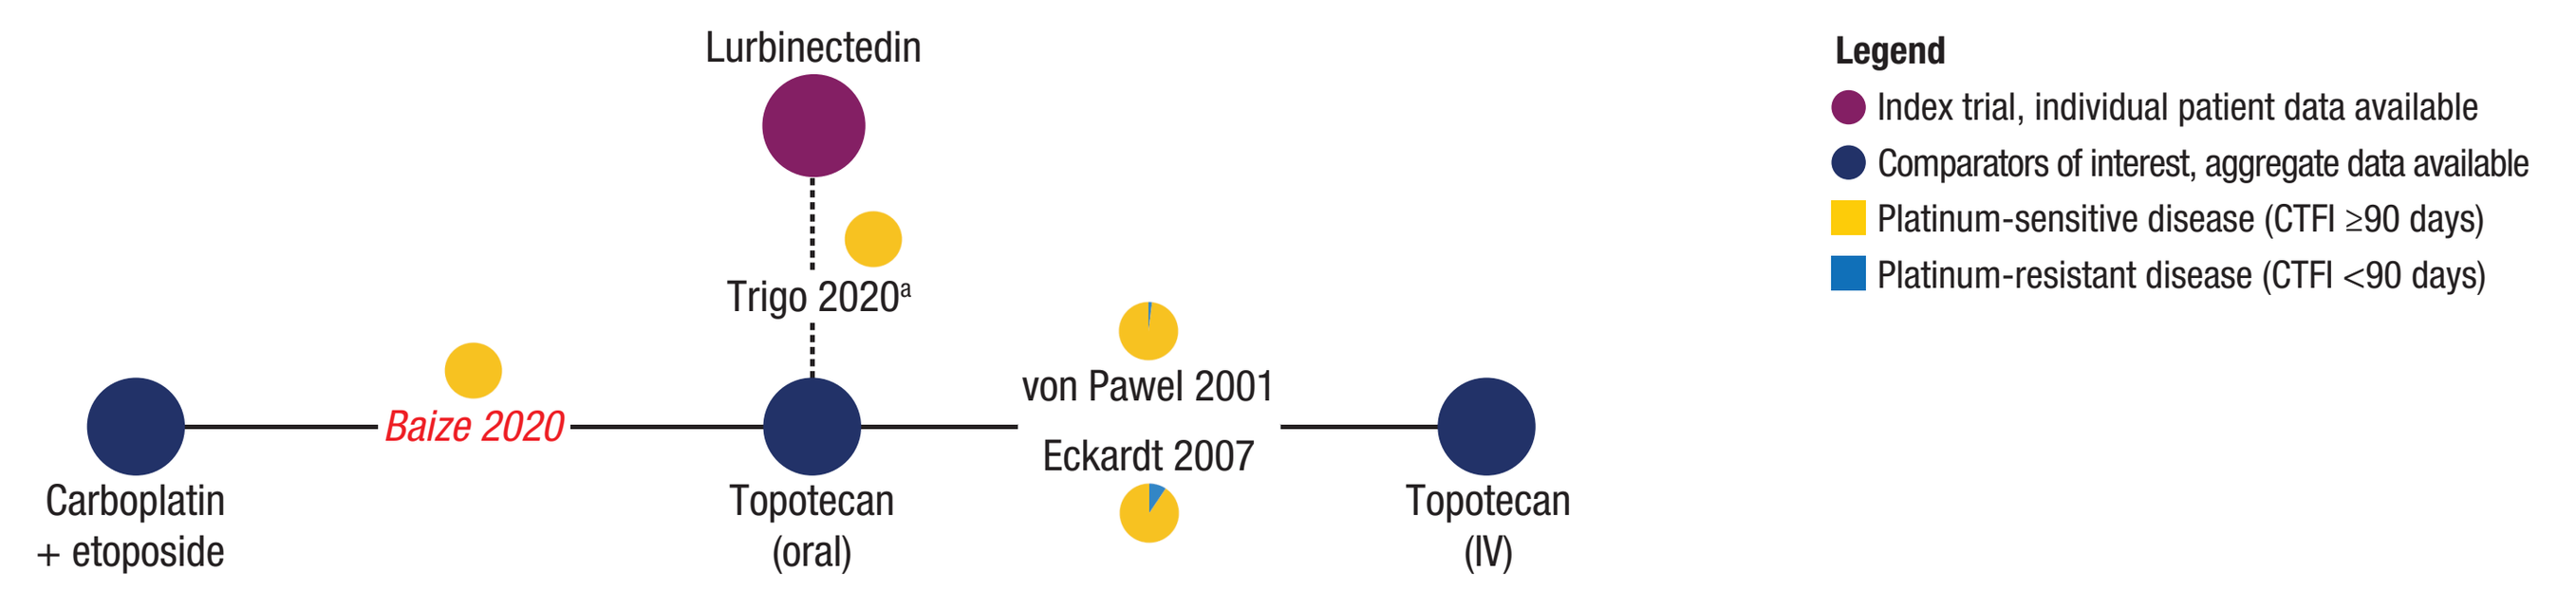

Lurbinectedin
Trigo 2020[a]
Carboplatin + etoposide
Baize 2020
Topotecan (oral)
von Pawel 2001
Eckardt 2007
Topotecan (IV)
Legend
Index trial, individual patient data available
Comparators of interest, aggregate data available
Platinum-sensitive disease (CTFI ≥90 days)
Platinum-resistant disease (CTFI <90 days)

ITC, indirect treatment comparison; ORR, overall response rate; OS, overall survival; IV, intravenous; CTFI, chemotherapy-free interval; MAIC, matching-adjusted indirect comparison.
Note: Trial name in *red italic* represents the trial used to connect Trigo 2020 to the network via a MAIC.
[a]Based on the CTFI ≥90 days subgroup (n = 60).

Conclusions

- Results from the SLR confirmed that it was feasible to evaluate the comparative efficacy of lurbinectedin versus key comparators of interest, as required assumptions were not prohibitive
- Given the inclusion of a disconnected single-arm trial, an unanchored MAIC was used to compare lurbinectedin with other treatments
- Restricting the ITC to patients with platinum-sensitive disease receiving second-line therapy for SCLC reduced heterogeneity
- In the base case fixed-effect NMA, lurbinectedin showed a robust survival benefit for the second-line treatment of patients with platinum-sensitive SCLC compared with topotecan and platinum rechallenge
 - Lurbinectedin had a significantly greater OS versus oral topotecan, IV topotecan, and carboplatin plus etoposide, and a significantly greater ORR versus oral topotecan

References: 1. ZEPZELCA® (lurbinectedin) for injection, for intravenous use [prescribing information]. Palo Alto, CA: Jazz Pharmaceuticals, Inc. **2.** Trigo J, et al. *Lancet Oncol.* 2020;21(5):645-654. **3.** Baize N, et al. *Lancet Oncol.* 2020;21(9):1224-1233. **4.** O'Brien ME, et al. *J Clin Oncol.* 2006;24(34):5441-5447. **5.** Eckardt JR, et al. *J Clin Oncol.* 2007;25(15):2086-2092. **6.** von Pawel J, et al. *J Clin Oncol.* 2001;19(6):1743-1749. **7.** Costa R, et al. *Oncotarget.* 2017;8(5):8910-8920. **8.** Owonikoko TK, et al. *J Thorac Oncol.* 2020;15(2):274-287. **9.** Spigel DR, et al. *Ann Oncol.* 2021;32(5):631-641. **10.** Antonia SJ, et al. *Lancet Oncol.* 2016;17(7):883-895. **11.** Ready NE, et al. *J Thorac Oncol.* 2020;15(3):426-435. **12.** Smyth JF, et al. *Eur J Cancer.* 1994;30(8):1058-1060.

Support: This study was supported by Jazz Pharmaceuticals.

Acknowledgements: Medical writing and editorial assistance were provided by Tamara K. Stevenson, PhD, of Cello Health Communications/SciFluent Communications, Inc., and were financially supported by Jazz Pharmaceuticals.

Disclosures: RH and **BR** are employees of and hold stock in Jazz Pharmaceuticals. **CS** is a former employee of Jazz Pharmaceuticals. **AA** and **FRW** are employees of PRECISIONheor.