POSB197

Impact of immune checkpoint inhibitors on the management of advanced non-small cell lung cancer in real-world practice at Karolinska University Hospital, Stockholm between 2012 and 2018

Simon Ekman,[1] Odd Terje Brustugun,[2] Jens Benn Sørensen,[3] Anne Mette Tranberg Kejs,[4] Quratul Ann,[5] Margherita Bortolini,[6] Alan Calleja,[7] Linda Rosengren,[8] Pernilla Huetson,[8] Melinda J. Daumont,[9] John R. Penrod,[10] Hazel Jacobs,[11] Laure Lacoin,[12] Hirsh Koyi[13]

[1]Thoracic Oncology Center, Karolinska University Hospital and Department of Oncology-Pathology, Karolinska Institute, Stockholm, Sweden; [2]Section of Oncology, Drammen Hospital, Vestre Viken Hospital Trust, Drammen, Norway; [3]Department of Oncology, Rigshospitalet, Copenhagen, Denmark; [4]Real World & Analytics Solutions, IQVIA, Copenhagen, Denmark; [5]EMEA Real World Methods & Evidence Generation, IQVIA, London, UK; [6]EMEA Real World Methods & Evidence Generation, IQVIA, London, UK; [7]Real World Solutions, IQVIA, London, UK; [8]Bristol Myers Squibb, Solna, Sweden; [9]Worldwide Health Economics & Outcomes Research, Bristol Myers Squibb, Braine-L'Alleud, Belgium; [10]Worldwide Health Economics & Outcomes Research, Bristol Myers Squibb, Princeton, NJ, USA; [11]Syneos Health, Farnborough, UK; [12]Epi-Fit, Bordeaux, Nouvelle-Aquitaine, France; [13]Department of Oncology-Pathology, Karolinska Institute, Stockholm, Sweden and Department of Pulmonary Medicine, Gävle Hospital, Sweden

Background

- Immune checkpoint inhibitors (ICIs) have improved survival outcomes in patients with advanced/metastatic non-small cell lung cancer (NSCLC) as shown in patients receiving ICI monotherapy in 2L versus conventional therapy[1-4] and more recently in patients receiving ICI in 1L (monotherapy in PD-L1 ≥ 50% or in combination with chemotherapy)[5-9]
- In Sweden, ICIs have been reimbursed since 2015 for locally advanced/metastatic NSCLC
- ICI monotherapy in Sweden has been reimbursed for:
 - Second-line (2L) treatment for squamous carcinoma (SQ) since December 2015
 - 2L treatment for non-squamous carcinoma (NSQ) since June 2016
 - 1L treatment for high programmed death ligand 1 (PD-L1) expressors (≥ 50%) since March 2017
- This study from the SCAN LEAF project, part of the I-O Optimise initiative,[10] assessed the early adoption of ICIs for the real-world management of advanced/metastatic NSCLC in Sweden

Methods

Settings and participants

- This retrospective cohort study included adult patients (≥ 18 years) without *ALK* rearrangements/*EGFR* mutations diagnosed with advanced/metastatic NSCLC at Karolinska University Hospital between January 2012 and December 2017 who started a 1L treatment between January 2012 and December 2018
- Patients with advanced/metastatic NSCLC were defined as patients with stage IIIB-IV NSCLC at initial diagnosis or patients initially diagnosed at stage I-IIIA who then progressed and started a 1L treatment
- Patients were followed until December 2018
- Patients were categorized into 3 subgroups based on the year of 1L treatment start:
 - Pre-ICI (2012-2014): more than 1 year prior to first 2L ICI approval in advanced NSCLC
 - Washout (2015-2016): in the year prior and after the first 2L ICI approval in advanced NSCLC
 - Early post-ICI (2017-2018): at least 1 year after the first 2L ICI approval in advanced NSCLC
- Electronic medical records data were linked to national registries data

Analyses

- Index date was defined as the 1L treatment initiation date
- Patient characteristics were described by histology and time period using summary characteristics
- Distinct lines of therapy (LoTs) were identified using an algorithm based on the type and date of systemic anti-cancer therapy (SACT) administered and the length of the time period between 2 treatments
- Treatment sequences across LoTs were displayed using Sankey diagrams and included the first 4 LoTs

Results

Patient characteristics

- In 2012-2018, 1485 patients with advanced/metastatic NSCLC without *ALK* rearrangements or *EGFR* mutations received a 1L SACT during the study period (**Table 1**)
 - Baseline characteristics at the start of 1L treatment were broadly similar across the time periods

Table 1. Baseline characteristics at time of 1L treatment start in patients with advanced/metastatic NSCLC without *ALK* rearrangements/*EGFR* mutations

	All years 2012-2018			Pre-ICI 2012-2014		Washout 2015-2016		Early post-ICI 2017-2018	
	All (N = 1485)	NSQ/Other (n = 1217)	SQ (n = 268)	NSQ/Other (n = 549)	SQ (n = 116)	NSQ/Other (n = 437)	SQ (n = 100)	NSQ/Other (n = 231)	SQ (n = 52)
Age									
Median (IQR), years	69.7 (63.6-74.5)	69.5 (63.6-74.2)	70.8 (64.0-75.4)	68.9 (63.5-74.1)	70.3 (63.7-76.5)	69.7 (63.0-74.2)	69.6 (62.9-74.2)	70.5 (64.1-74.8)	72.7 (67.0-76.1)
Range, years	34.0-91.7	34.0-91.7	34.5-86.5	34.0-91.7	35.7-86.5	39.7-85.9	34.5-84.6	39.7-91.1	51.3-84.7
Aged ≥ 75 years	401 (27.0)	316 (26.0)	85 (31.7)	138 (25.1)	40 (34.5)	112 (25.6)	26 (26.0)	66 (28.6)	19 (36.5)
Sex									
Male	710 (47.8)	543 (44.6)	167 (62.3)	251 (45.7)	75 (64.7)	190 (43.5)	64 (64.0)	102 (44.2)	28 (53.8)
Female	775 (52.2)	674 (55.4)	101 (37.7)	298 (54.3)	41 (35.3)	247 (56.5)	36 (36.0)	129 (55.8)	24 (46.2)
Smoking status									
Never smoker	63 (4.2)	61 (5.0)	< 6	25 (4.6)	< 6	19 (4.3)	< 6	17 (7.4)	0
Former smoker	834 (56.2)	689 (56.6)	145 (54.1)	308 (56.1)	60 (51.7)	247 (56.5)	50 (50.0)	134 (58.0)	35 (67.3)
Current smoker	356 (24.0)	280 (23.0)	76 (28.4)	129 (23.5)	37 (31.9)	101 (23.1)	28 (28.0)	50 (21.6)	11 (21.2)
Missing/unknown	232 (15.6)	187 (15.4)	< 50 (< 20.0)	87 (15.8)	< 20 (< 20.0)	70 (16.0)	< 25 (< 25.0)	30 (13.0)	6 (11.5)
ECOG PS									
0-1	969 (65.3)	800 (65.7)	169 (63.1)	367 (66.8)	73 (62.9)	263 (60.2)	60 (60.0)	170 (73.6)	36 (69.2)
≥ 2	127 (8.6)	91 (7.5)	36 (13.4)	43 (7.8)	13 (11.2)	34 (7.8)	15 (15.0)	14 (6.1)	8 (15.4)
Missing/unknown	389 (26.2)	326 (26.8)	63 (23.5)	139 (25.3)	30 (25.9)	140 (32.0)	25 (25.0)	47 (20.3)	8 (15.4)
TNM stage at initial diagnosis[a]									
I-IIIA	404 (27.2)	299 (24.6)	105 (39.2)	123 (22.4)	44 (37.9)	124 (28.4)	36 (36.0)	52 (22.5)	25 (48.1)
IIIB	157 (10.6)	99 (8.1)	58 (21.6)	37 (6.7)	25 (21.6)	39 (8.9)	23 (23.0)	23 (10.0)	10 (19.2)
IV	924 (62.2)	819 (67.3)	105 (39.2)	389 (70.9)	47 (40.5)	274 (62.7)	41 (41.0)	156 (67.5)	17 (32.7)
Histopathology									
NSQ	998 (67.2)	998 (82.0)	0	437 (79.6)	0	353 (80.8)	0	208 (90.0)	0
SQ	268 (18.0)	0	268 (100)	0	116 (100)	0	100 (100)	0	52 (100)
NOS/Other specified	195 (13.1)	195 (16.0)	0	100 (18.2)	0	74 (16.9)	0	< 25 (< 10.8)	0
Clinically diagnosed[b]	24 (1.6)	24 (2.0)	0	12 (2.2)	0	10 (2.3)	0	< 6	0
PD-L1 testing	106 (7.1)	79 (6.5)	27 (10.1)	< 6	0	< 6	< 6	74 (32.0)	26 (50.0)
PD-L1 status									
PD-L1+	51 (3.4)	37 (3.0)	14 (5.2)	0	0	< 6	0	36 (15.6)	14 (26.9)
PD-L1+ ≥ 50%	13 (0.9)	10 (0.8)	< 6	0	0	0	0	10 (4.3)	< 6
PD-L1−	28 (1.9)	21 (1.7)	7 (2.6)	0	0	< 6	0	19 (8.2)	7 (13.5)
Missing/non-valid	1406 (94.7)	1159 (95.2)	247 (92.2)	549 (100)	116 (100)	434 (99.3)	100 (100)	176 (76.2)	31 (59.6)

Data are n (%) unless otherwise stated.
[a]Stage at initial diagnosis was based on the 7th edition of the TNM classification system through the full study period; [b]No histology or pathology available.
ECOG PS, Eastern Cooperative Oncology Group performance status; IQR, interquartile range; NOS, not otherwise specified.

Figure 1. Treatment sequencing of patients with advanced/metastatic NSCLC[a]

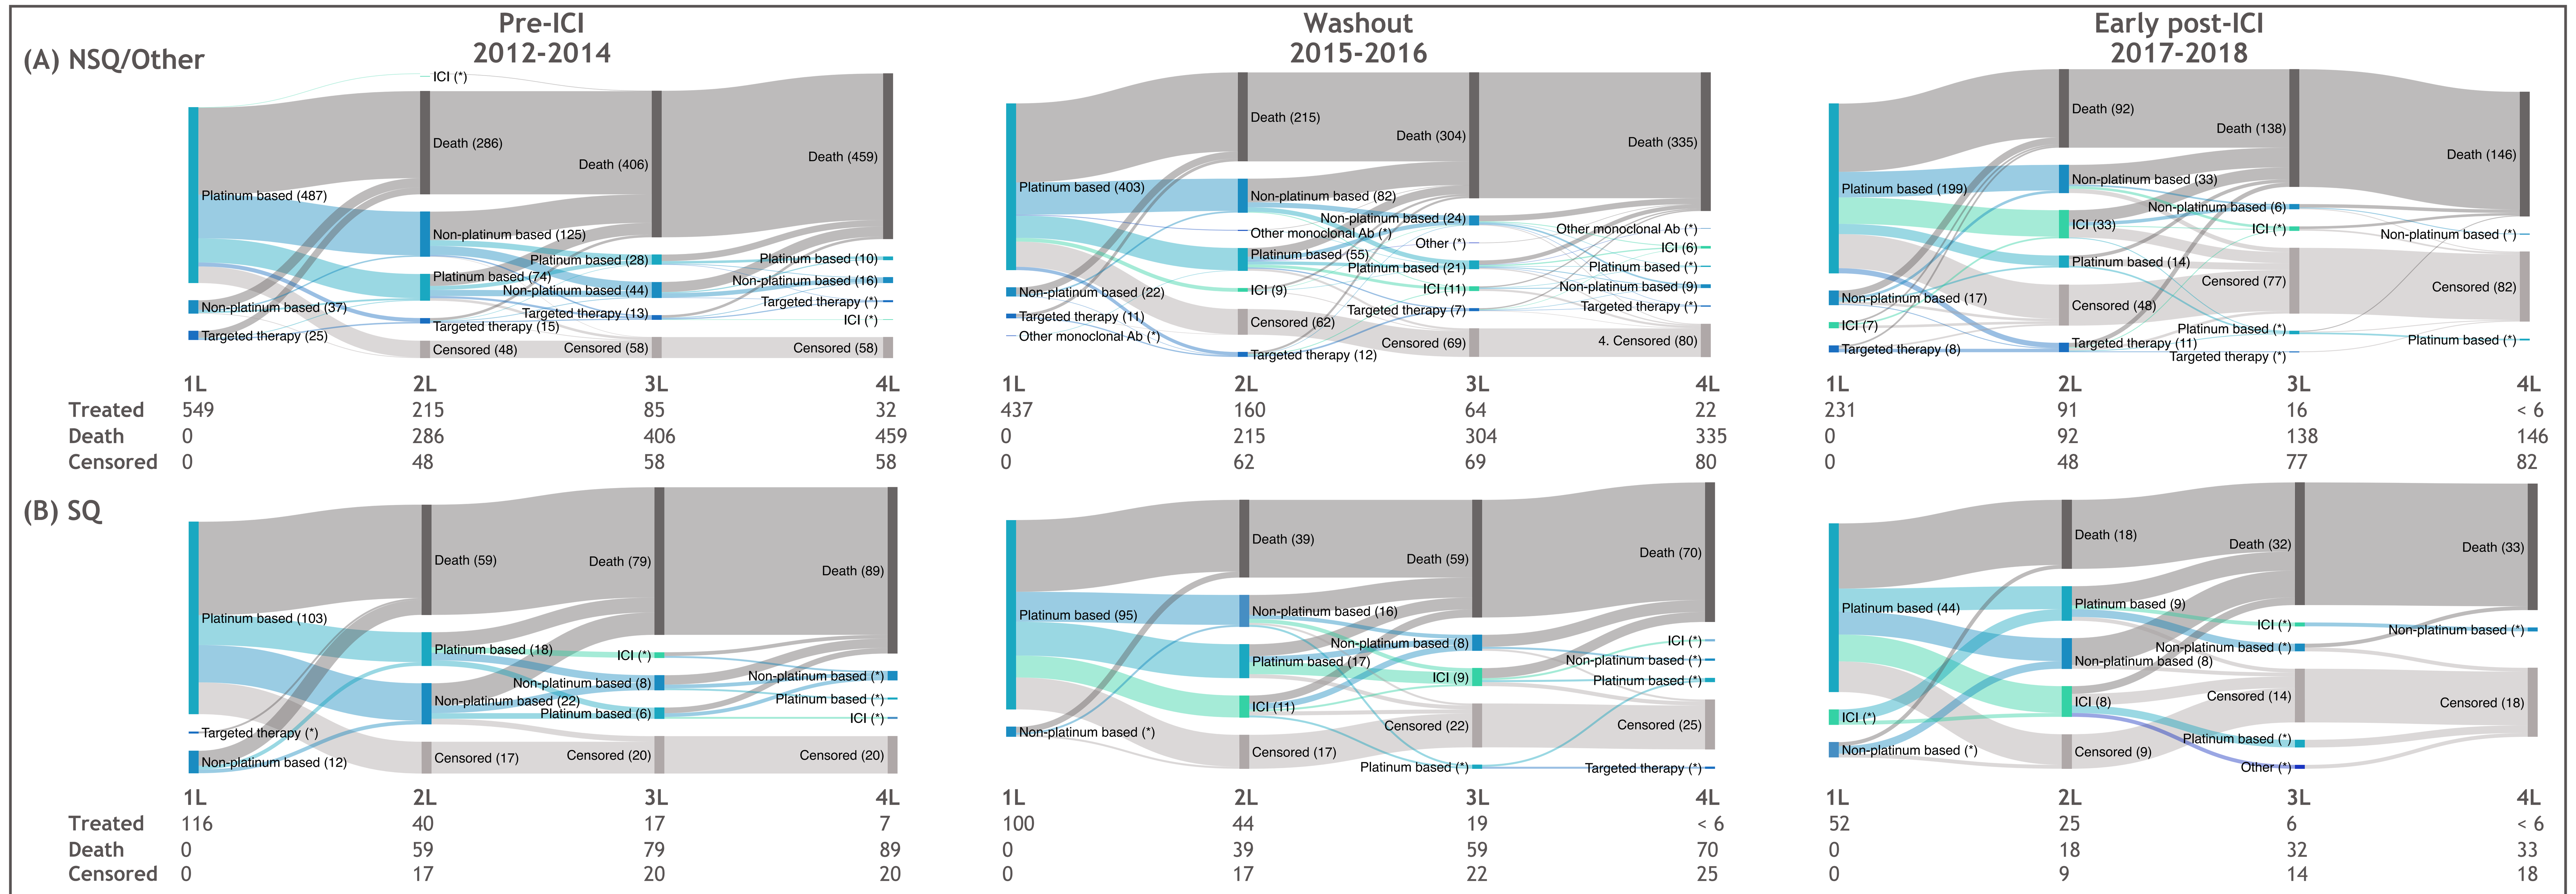

Pre-ICI 2012-2014
Washout 2015-2016
Early post-ICI 2017-2018
(A) NSQ/Other
Platinum based (487)
Non-platinum based (37)
Targeted therapy (25)
ICI (*)
Death (286)
Non-platinum based (125)
Platinum based (74)
Targeted therapy (15)
Censored (48)
Death (406)
Platinum based (28)
Non-platinum based (44)
Targeted therapy (13)
Censored (58)
Death (459)
Platinum based (10)
Non-platinum based (16)
Targeted therapy (*)
ICI (*)
Censored (58)
Platinum based (403)
Non-platinum based (22)
Targeted therapy (11)
Other monoclonal Ab (*)
Death (215)
Non-platinum based (82)
Other monoclonal Ab (*)
Platinum based (55)
ICI (9)
Censored (62)
Targeted therapy (12)
Death (304)
Non-platinum based (24)
Other (*)
Platinum based (21)
ICI (11)
Targeted therapy (7)
Censored (69)
Death (335)
Other monoclonal Ab (*)
ICI (6)
Platinum based (*)
Non-platinum based (9)
Targeted therapy (*)
4. Censored (80)
Platinum based (199)
Non-platinum based (17)
ICI (7)
Targeted therapy (8)
Death (92)
Non-platinum based (33)
ICI (33)
Platinum based (14)
Censored (48)
Targeted therapy (11)
Death (138)
Non-platinum based (6)
ICI (*)
Censored (77)
Platinum based (*)
Targeted therapy (*)
Death (146)
Non-platinum based (*)
Censored (82)
Platinum based (*)
1L 2L 3L 4L
Treated 549 215 85 32
Death 0 286 406 459
Censored 0 48 58 58
1L 2L 3L 4L
Treated 437 160 64 22
Death 0 215 304 335
Censored 0 62 69 80
1L 2L 3L 4L
Treated 231 91 16 < 6
Death 0 92 138 146
Censored 0 48 77 82
(B) SQ
Platinum based (103)
Targeted therapy (*)
Non-platinum based (12)
Death (59)
Platinum based (18)
Non-platinum based (22)
Censored (17)
Death (79)
ICI (*)
Non-platinum based (8)
Platinum based (6)
Censored (20)
Death (89)
Non-platinum based (*)
Platinum based (*)
ICI (*)
Censored (20)
Platinum based (95)
Non-platinum based (*)
Death (39)
Non-platinum based (16)
Platinum based (17)
ICI (11)
Censored (17)
Death (59)
Non-platinum based (8)
ICI (9)
Censored (22)
Platinum based (*)
Death (70)
ICI (*)
Non-platinum based (*)
Platinum based (*)
Censored (25)
Targeted therapy (*)
Platinum based (44)
ICI (*)
Non-platinum based (*)
Death (18)
Platinum based (9)
Non-platinum based (8)
ICI (8)
Censored (9)
Death (32)
ICI (*)
Non-platinum based (*)
Censored (14)
Platinum based (*)
Other (*)
Death (33)
Non-platinum based (*)
Censored (18)
1L 2L 3L 4L
Treated 116 40 17 7
Death 0 59 79 89
Censored 0 17 20 20
1L 2L 3L 4L
Treated 100 44 19 < 6
Death 0 39 59 70
Censored 0 17 22 25
1L 2L 3L 4L
Treated 52 25 6 < 6
Death 0 18 32 33
Censored 0 9 14 18

*Data are not shown when n < 6.
[a]Without *ALK* rearrangements/*EGFR* mutations.
3L, third line; 4L, fourth line.

- Between the pre-ICI period and the early post-ICI period, PD-L1 testing prior to 1L treatment start increased from < 1% to 32% for NSQ/other and from 0% to 50.0% for SQ

Treatment patterns

- Treatment sequencing for patients with NSQ/other and SQ histology by treatment period is shown in **Figure 1**
- There were no major changes in 1L treatment received between the pre-ICI period and the early post-ICI period
 - Overall, 89.5% of patients with NSQ/other and 90.3% of patients with SQ histology received 1L platinum-based chemotherapy
 - In the early post-ICI period, 3% of patients with NSQ/other histology and less than 6 patients with SQ histology received an ICI monotherapy as 1L treatment
- There was no change in the proportion of patients receiving a 2L treatment over the study period
 - Overall, 38.7% of patients received a 2L therapy
 - Further follow-up is needed in the early post-ICI period to appropriately estimate this proportion, as 17.3% and 20.8% of the patients with NSQ/other and SQ who were alive were censored while still on 1L treatment during this period
- Receipt of ICI by treatment period is shown in **Figure 2**
 - For patients with NSQ/other histology, an ICI was received by 1.1% of patients (mostly 3L or later) in the pre-ICI period and 18.2% (mostly 2L) in the early post-ICI period
 - For patients with SQ histology, an ICI was received by 3.4% of patients (3L or later) in the pre-ICI period and 23.1% in the early post-ICI period (majority as 2L)

Figure 2. Receipt of ICI in patients with advanced/metastatic NSCLC[a]

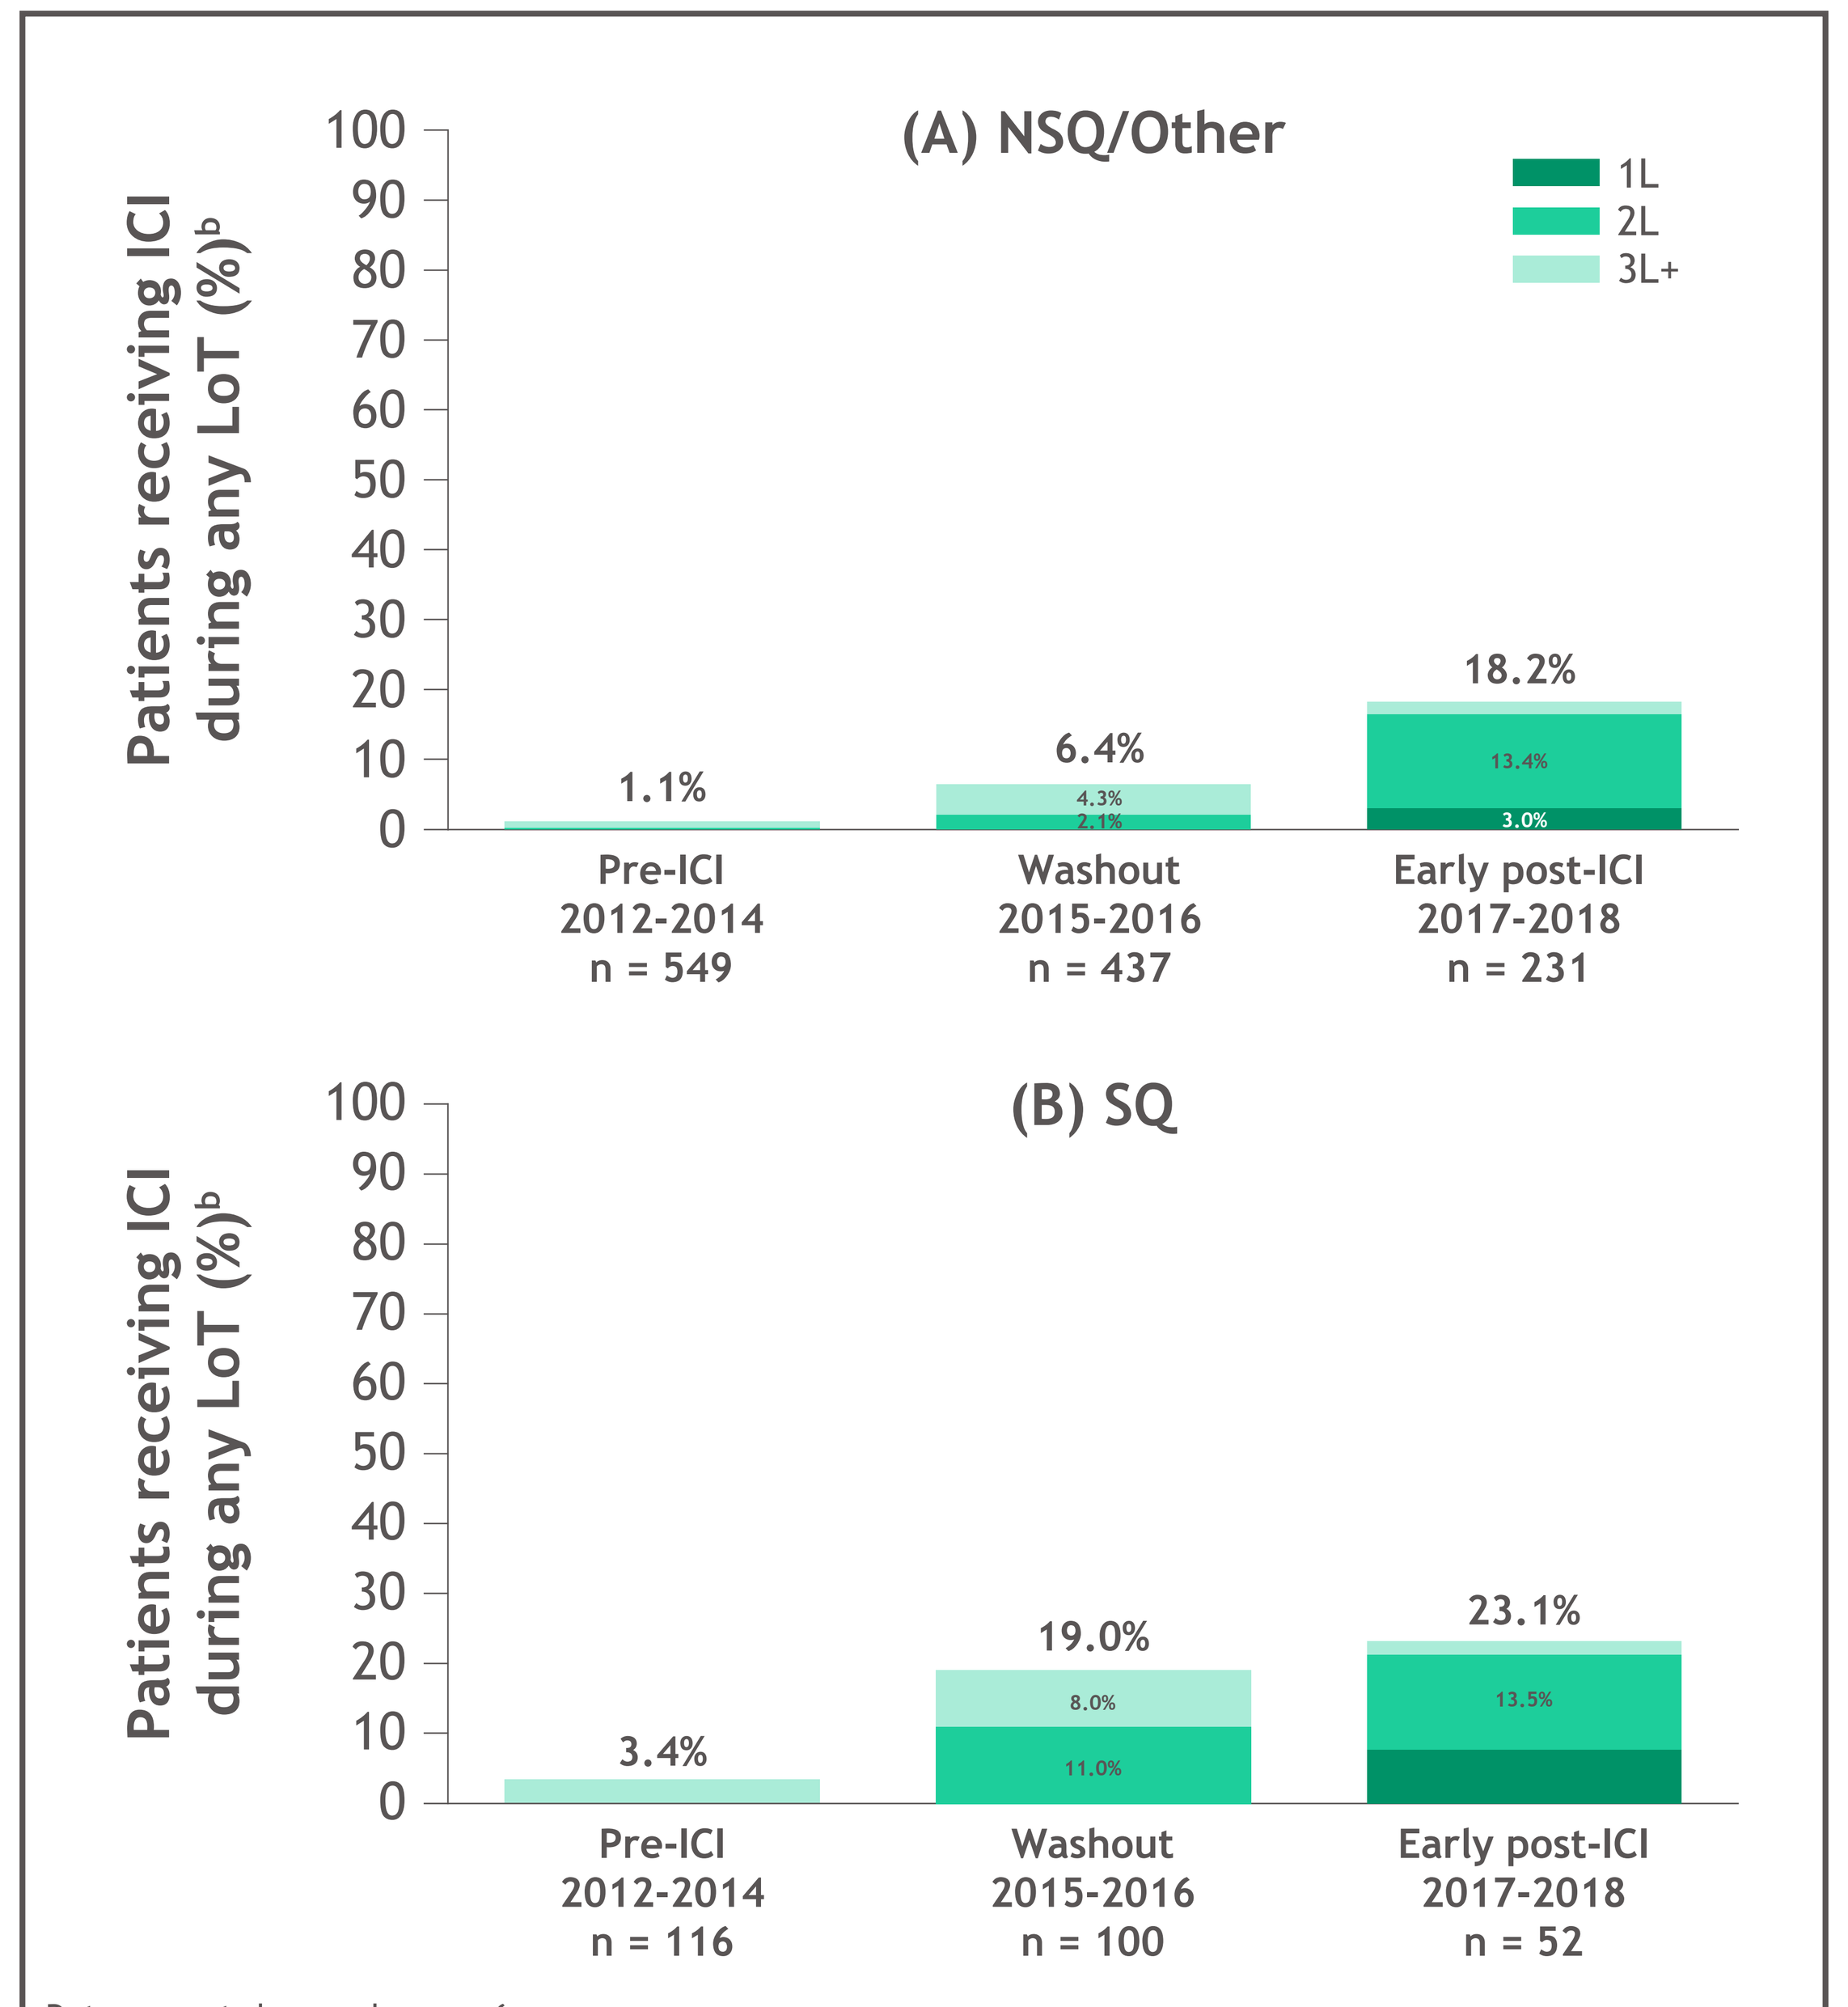

(A) NSQ/Other
1L
2L
3L+
Patients receiving ICI during any LoT (%)b
100 90 80 70 60 50 40 30 20 10 0
1.1%
6.4%
4.3%
2.1%
18.2%
13.4%
3.0%
Pre-ICI 2012-2014 n = 549
Washout 2015-2016 n = 437
Early post-ICI 2017-2018 n = 231
(B) SQ
3.4%
19.0%
8.0%
11.0%
23.1%
13.5%
Pre-ICI 2012-2014 n = 116
Washout 2015-2016 n = 100
Early post-ICI 2017-2018 n = 52

Data are not shown when n < 6.
[a]Without *ALK* rearrangements/*EGFR* mutations; [b]ICIs up to the 11th LoT were included.

Conclusions

- In the early post-ICI period, approximately one-fifth of patients with advanced/metastatic NSCLC treated at Karolinska University Hospital received an ICI, mainly in the 2L setting
 - The proportion of patients in the post-ICI period receiving an ICI as 2L+ was similar between NSQ/other and SQ histology
- Future studies will evaluate the longer-term adoption and continued use of ICIs, as well as the impact of this use on survival outcomes

References

1. Borghaei H, et al. *N Engl J Med* 2015;373:1627-1639.
2. Brahmer J, et al. *N Engl J Med* 2015;373:123-135.
3. Herbst R, et al. *Lancet* 2016;387:1540-1550.
4. Rittmeyer A, et al. *Lancet* 2017;389:255-265.
5. Reck M, et al. *N Engl J Med* 2016;375:1823-1833.
6. Gandhi L, et al. *N Engl J Med* 2018;378:2078-2092.
7. Paz-Ares L, et al. *N Engl J Med* 2018;379:2040-2051.
8. Socinski MA, et al. *N Engl J Med* 2018;378:2288-2301.
9. West H, et al. *Lancet Oncol* 2019;20:924-937.
10. Ekman S, et al. *Future Oncol* 2019;15:1551-1563.

Acknowledgments

- This work is part of the I-O Optimise programme, an initiative of Bristol Myers Squibb
- This study was supported by Bristol Myers Squibb
- All authors contributed to and approved the presentation; professional writing and editorial services were provided by Parexel, funded by Bristol Myers Squibb

Disclosures

- Simon Ekman is employed by an institution that was remunerated by Bristol Myers Squibb for this study
- Odd Terje Brustugun has received honoraria from AstraZeneca, Bristol Myers Squibb, Boehringer Ingelheim, Eli Lilly, MSD, Novartis, Pfizer, Pierre Fabre, Roche, and Takeda; and received research funding from Pfizer, AstraZeneca, Roche, GlaxoSmithKline, and Boehringer Ingelheim
- Jens Benn Sørensen has received speakers fee and honoraria for advisory boards from Bristol Myers Squibb
- Linda Rosengren, Pernilla Huetson, Melinda J. Daumont, and John R. Penrod are employees of Bristol Myers Squibb
- Anne Mette Tranberg Kejs, Quratul Ann, Margherita Bortolini, and Alan Calleja are employees of IQVIA
- Laure Lacoin is an employee of Epi-Fit and was contracted as a consultant by Bristol Myers Squibb to support the I-O Optimise initiative
- Hazel Jacobs is an employee of Syneos Health and was contracted as a consultant by Bristol Myers Squibb to support the I-O Optimise initiative
- Hirsh Koyi reports no disclosures

Presented at Virtual ISPOR Europe 2021, November 30-December 3, 2021

Email: simon.ekman@ki.se

Copies of this poster are for personal use only and may not be reproduced without written permission from the author of this poster.