Poster POSA357

Jyoti Chauhan (jyoti.chauhan@novartis.com)

Social Media Listening for Melanoma Care Across European Countries

Jyoti Chauhan,[1] Sathyaraj Aasaithambi,[1] Alexandros Sagkriotis,[2] Mike Lau,[2] Iván Márquez Rodas,[3] Luigi Formisano,[4] Sophie Papa,[5] Nicolas Meyer,[6] Andrea Forschner,[7] Guy Faust[8]

[1]Novartis Healthcare Pvt. Ltd, Hyderabad, India; [2]Novartis Pharma AG, Basel, Switzerland; [3]Department of Medical Oncology, Hospital General Universitario Gregorio Marañón, Instituto de Investigación Sanitaria Gregorio Marañón (IISGM), CIBERONC, Universidad Complutense, Madrid, Spain; [4]Department of Clinical Medicine and Surgery, University of Naples "Federico II", Naples, Italy; [5]School of Cancer and Pharmaceutical Studies, King's College London, London, UK; [6]Department of Onco-Dermatology, Toulouse Cancer Institute (IUC) and University Hospital (CHU), Toulouse, France; [7]Department of Dermatology, University Hospital Tübingen, Tübingen, Germany; [8]Department of Oncology, University Hospitals of Leicester, Leicester, UK

Scan to obtain:
- Poster

https://www.medicalcongress.novartisoncology.com/ISPORQRG/ispor/qrgispor.html#Chauhan345

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- SML studies allow for enhanced understanding of the quality of care and treatment approach to improve the wellbeing of patients with melanoma
- The study identifies emotional burden as the main QoL impact, emphasizing a need for the emotional support of patients with melanoma
- This understanding may aid HCP-patient communication and inform the adequate measurement of QoL concepts in clinical trials and other research studies
- Surgery was the most frequently discussed treatment for melanoma (particularly in 1L), and had the highest number of positive mentions compared with other treatments, while chemotherapy was often associated with negative sentiments, which was largely attributed to treatment side effects, but may also result from the palliative, non-curative approach of this treatment
- Despite the limitations mentioned, SML can provide person-focused, real-world insights into the lived experience of melanoma that are not typically available in the published literature or in databases

This study is sponsored by Novartis Pharma AG

Poster presented at ISPOR Europe 2021, held virtually on 30 November-3 December 2021

INTRODUCTION

- Melanoma is an aggressive malignancy arising from pigmented cells
 - Advanced disease occurs in about 30–40% of cases, and has an average 5-year survival rate of 5–19%[1]
- Incidence of melanoma has been increasing worldwide for the last 50 years[2]
- Melanoma is a major public health burden in Europe due to increasing incidence, high mortality, complexity of care for advanced stage disease, and impact on quality of life (QoL), and is estimated to cost >20,000 lives every year[3]
- The ubiquitous use of social media (SM) allows disease-specific insights to be obtained from publically available sources
- There is currently limited qualitative research on melanoma in the SM population
- This study used a SM listening (SML) approach to generate insights into the needs and perceptions of patients with melanoma from across European countries

OBJECTIVE

- To investigate the needs and perceptions of melanoma stakeholders using SML analysis to generate patient insights from across European countries

METHODS

- SML data was collected retrospectively for 2 years (Nov 2018–Nov 2020) for melanoma-specific terms across a number of European countries (Austria, Belgium, Denmark, Finland, France, Germany, Italy, Netherlands, Norway, Portugal, Spain, Sweden, Switzerland, and UK), in 11 languages (Danish, Dutch, English, Finnish, French, German, Italian, Norwegian, Portuguese, Spanish, and Swedish), from blogs, forums, and SM platforms
- Melanoma-relevant search strings were developed for each language, and were used to identify all viable data sources and extract data for in-scope geographies (**Figure 1**)
- Contextualized data obtained from relevant sources was further analyzed by excluding irrelevant and non-insightful data, to provide melanoma-related insights
- An iterative random sampling procedure was used as per an agreed sample size for SM use/country to filter the dataset and obtain a manageable number of posts for filtering
- Posts were filtered using an automated relevancy approach (containing keyword-based relevancy algorithms) and manual review to ensure conversations provided relevant insights to the patient journey stage and other patient-centric topics for melanoma

METHODS (CONTINUED)

Figure 1. Study Methodology

Step 1 — **Develop Search String**
Example Keywords (in English): Melanoma*; #Melanoma*; #MetastaticMelanoma; metastaticmelanoma; #mMelanoma; mMelanoma; #amelanoticMelanoma; amelanoticMelanoma; #invasiveMelanoma; superficialspreadingmelanoma; nodularmelanoma; lentigomalignamelanoma; malignamelanoma; malignantmelanoma; #malignantmelanoma; Acrallentiginousmelanoma; amelanoticmelanoma; nevoidmelanoma; spitzoidmelanoma; desmoplasticmelanoma; cutaneousmelanoma

Step 2 — **Search and Extract Data**
Identify all viable data sources (including forums, blogs, Twitter, Facebook, and YouTube), and extract data for in-scope geographies

Step 3 — **Data Analysis**
Filter dataset by excluding irrelevant information using both automated and manual relevancy approaches to remove data that lacked meaningful insights

Step 4 — **Generate Insights**
Deep-dive on relevant discussions using an iterative random sampling procedure to draw insights from key stakeholders who provided information on specific research domains

RESULTS

SM Overview and Population Demographics

- A total of 182.4K posts were extracted, of which a total of 864 relevant conversations were included for analysis
- Twitter was the primary source of information (82% of analyzed posts; n = 864) in most countries, except for Germany and Austria, where forums were more commonly used among melanoma stakeholders (**Table 1**)
- Most analyzed posts were from the UK (n = 200) when compared with other European countries included in the study (**Table 1**)
- For all countries, patients were the largest contributors to conversations (62%), followed by either caregivers, or patients' friends and family (**Table 1**)
- Conversations were predominantly female-led (55%), which was consistent for most countries except Switzerland and the Nordic countries, where male conversations were more common, and Spain, where the gender split was 50% (**Table 1**)
- 31–50 years was the most common age group contributing to discussions, although France and Belgium mostly had stakeholders in younger age groups (**Table 1**); and age was rarely mentioned in Austria, Portugal, and Switzerland
- Malignant and metastatic disease accounted for 77% of the melanoma types mentioned in the analyzed dataset (n = 864), and advanced stage melanoma (which also included the terms 'stage IV,' 'late stage,' and 'metastatic stage' disease), was the most frequently discussed disease stage (63%)

Table 1. SM Population Demographics by Country

Key Channels and Key Stakeholders:

Countries	Twitter	Forums	Blogs	n	Patients	Caregivers	Friends & Family	HCPs	n
All (Overall Data)	82%	18%	0.1%	n = 864	62%	22%	12%	2%	n = 864
UK	90%	10%	0%	n = 200	69%	24%	5%	2%	n = 200
Spain	98%	2%	0%	n = 150	69%	14%	13%	2%	n = 150
France	82%	18%	0%	n = 100	45%	28%	17%	7%	n = 100
Italy	89%	11%	0%	n = 140	42%	36%	18%	1%	n = 140
Germany	24%	76%	0%	n = 100	77%	5%	8%	0%	n = 100
Netherlands	96%	4%	0%	n = 50	72%	18%	8%	4%	n = 50
The Nordics	94%	6%	0%	n = 50	56%	30%	10%	4%	n = 50
Switzerland	100%	0%	0%	n = 20	70%	5%	25%	0%	n = 20
Austria	25%	75%	0%	n = 20	70%	10%	10%	0%	n = 20
Belgium	88%	8%	4%	n = 25	68%	20%	12%	0%	n = 25
Portugal	100%	0%	0%	n = 9	44%	22%	33%	0%	n = 9

Gender and Key Age Groups:

Countries	Gender		n	20 yrs and below	21 - 30 yrs	31 - 40 yrs	41 - 50 yrs	51 - 60 yrs	61 - 70 yrs	71 - 80 yrs	Above 80 yrs	n
All (Overall Data)	45%	55%	n = 768	5%	14%	29%	24%	14%	7%	4%	3%	n = 307
UK	38%	62%	n = 181		7%	33%	33%	18%	6%	4%		n = 107
Spain	50%	50%	n = 145	15%			38%	15%	15%		15%	n = 13
France	46%	54%	n = 83	38%	14%	19%	5%	5%	10%	5%	5%	n = 21
Italy	48%	52%	n = 132	10%	10%	24%	14%	10%	7%	17%	10%	n = 42
Germany	30%	70%	n = 66		6%	31%	19%	25%	6%		13%	n = 42
Netherlands	45%	55%	n = 49		20%	34%	24%	17%	5%			n = 41
The Nordics	69%	31%	n = 49	2%	26%	30%	26%	9%	6%	2%		n = 47
Switzerland	88%	12%	n = 17				100%					n = 1
Austria	25%	75%	n = 12									NA
Belgium	48%	52%	n = 25		28%	44%	11%	11%	6%			n = 18
Portugal	33%	77%	n = 9		100%							n = 1

RESULTS (CONTINUED)

Patient Journey for Melanoma

- The study provided key insights into the patient journey of those living with melanoma. Treatment (55%), and diagnosis and tests (33%) were the two most frequently discussed melanoma-related topics across countries (**Figure 2**)

Figure 2. Patient Journey by Country

	Causes	Pre-diagnosis Symptoms	Diagnosis & Tests	Treatment	Management	Remission	Recurrence	Death	n
Overall data (n = 792)	14%	10%	33%	55%	21%	24%	3%	13%	
UK	25%	8%	12%	58%	47%	47%	2%	9%	n = 195
Spain	9%	3%	33%	47%	7%	30%	1%	9%	n = 135
France	3%	3%	10%	52%	16%	14%	3%	22%	n = 77
Italy	7%	0%	30%	58%	18%	1%	2%	21%	n = 121
Germany	9%	24%	72%	48%	6%	7%	4%	4%	n = 97
Netherlands	21%	30%	55%	55%	28%	34%	11%	6%	n = 47
The Nordics	16%	20%	41%	76%	10%	22%	6%	29%	n = 49
Switzerland	5%	0%	25%	60%	0%	15%	0%	30%	n = 20
Austria	5%	25%	75%	65%	15%	0%	5%	0%	n = 20
Belgium	26%	26%	43%	39%	30%	22%	9%	13%	n = 23
Portugal	38%	0%	13%	75%	25%	25%	13%	25%	n = 8

High to low Prevalence

- In the countries where diagnostics were the most commonly discussed topic of the patient journey (Austria, Germany, and Belgium; **Figure 2**), confirmed diagnosis (60%), diagnostic tests/biopsy (40%), and other diagnosis/test topics (66%), were the most frequently discussed diagnosis and tests topics, respectively (**Table 2**)
 - Biopsy was frequently communicated as the confirmatory diagnostic test across most countries (with the exception of Austria and Portugal, where it was not mentioned)

Table 2. Patient Journey in Melanoma: Diagnosis and Tests

Diagnosis & tests	Confirmed diagnosis	Screening & diagnostics tests in general	Diagnosis in general	Biopsy	Others	Total N/n
All	36%	20%	15%	18%	40%	255
UK	22%	22%	35%	26%	4%	23
Spain	73%	5%	2%	2%	20%	44
France	0%	25%	25%	13%	50%	8
Italy	0%	61%	12%	12%	33%	33
Germany	40%	9%	4%	23%	66%	70
Netherlands	42%	27%	12%	23%	58%	26
The Nordics	20%	0%	55%	35%	25%	20
Switzerland	0%	20%	40%	20%	40%	5
Austria	60%	27%	0%	0%	40%	15
Belgium	20%	40%	30%	40%	20%	10
Portugal	0%	0%	100%	0%	0%	1

High to low prevalence

- With the exception of Switzerland, where immunotherapy was most commonly mentioned (50% of treatment discussions), surgery was the most frequently mentioned treatment type across all other countries (**Table 3**)

Table 3. Patient Journey in Melanoma: Treatment

	Surgery	Immunotherapy/ Biological	Radiation Therapy	Targeted Therapy	Chemotherapy	Palliative Care	Others	Total (N/n)
All	67%	12%	5%	4%	3%	1%	20%	437
UK	61%	16%	4%	5%	3%	0%	23%	114
Spain	83%	6%	3%	0%	5%	2%	10%	64
France	64%	10%	3%	3%	3%	0%	26%	39
Italy	70%	7%	4%	1%	3%	0%	22%	70
Germany	64%	13%	21%	4%	2%	0%	19%	47
Netherlands	81%	19%	0%	8%	0%	0%	4%	26
The Nordics	59%	0%	0%	5%	0%	8%	30%	37
Switzerland	33%	50%	0%	0%	0%	8%	16%	12
Austria	69%	23%	0%	15%	0%	0%	38%	13
Belgium	67%	33%	22%	22%	0%	0%	0%	9
Portugal	50%	0%	0%	0%	50%	0%	0%	6

High to low prevalence

- Surgery was the primary first-line (1L) treatment mentioned across all countries, particularly by stakeholders in Spain (78%; n = 32), and the UK (73%; n = 60)
- Surgery was also discussed as a frequently used second-line treatment in the UK (36%; n = 11), Spain (67%; n = 6), and the Netherlands (75%; n = 8)

Impact of Melanoma on Patients' QoL and Unmet Needs

- Discussions around impact on QoL were particularly prominent in the UK (n = 74), Italy (n = 47), Spain (n = 37), and Germany (n = 32); emotional impact was a frequently discussed topic across most countries (**Table 4**)

Table 4. QoL Impact on Patients with Melanoma

QoL Impact	Emotional	Physical	Social	Financial	Total N/n
All	70%	24%	17%	4%	255
UK	85%	19%	7%	1%	74
Spain	41%	19%	49%	5%	37
France	44%	33%	0%	22%	9
Italy	77%	28%	15%	0%	47
Germany	84%	19%	13%	0%	32
Netherlands	58%	37%	11%	11%	19
The Nordics	39%	33%	17%	17%	18
Switzerland	0%	100%	0%	0%	3
Austria	83%	0%	0%	17%	6
Belgium	100%	25%	50%	0%	4
Portugal	100%	17%	33%	0%	6

High to low prevalence

- The two most frequently mentioned unmet needs were availability of effective treatments and access to good HCPs/treatment, both constituting a quarter of conversations on unmet needs; these were most prominently discussed in the UK and Italy (**Figure 3**)
- Lack of empathy, limited communication, and lack of trust in HCPs was particularly prominent in conversations from Germany (**Figure 3**)
- Safe access to care during the COVID-19 pandemic was mentioned in 21% of conversations about unmet needs and was particularly prominent in the UK, Spain, France, and Belgium (**Figure 3**)

Figure 3: Unmet Needs of Patients with Melanoma

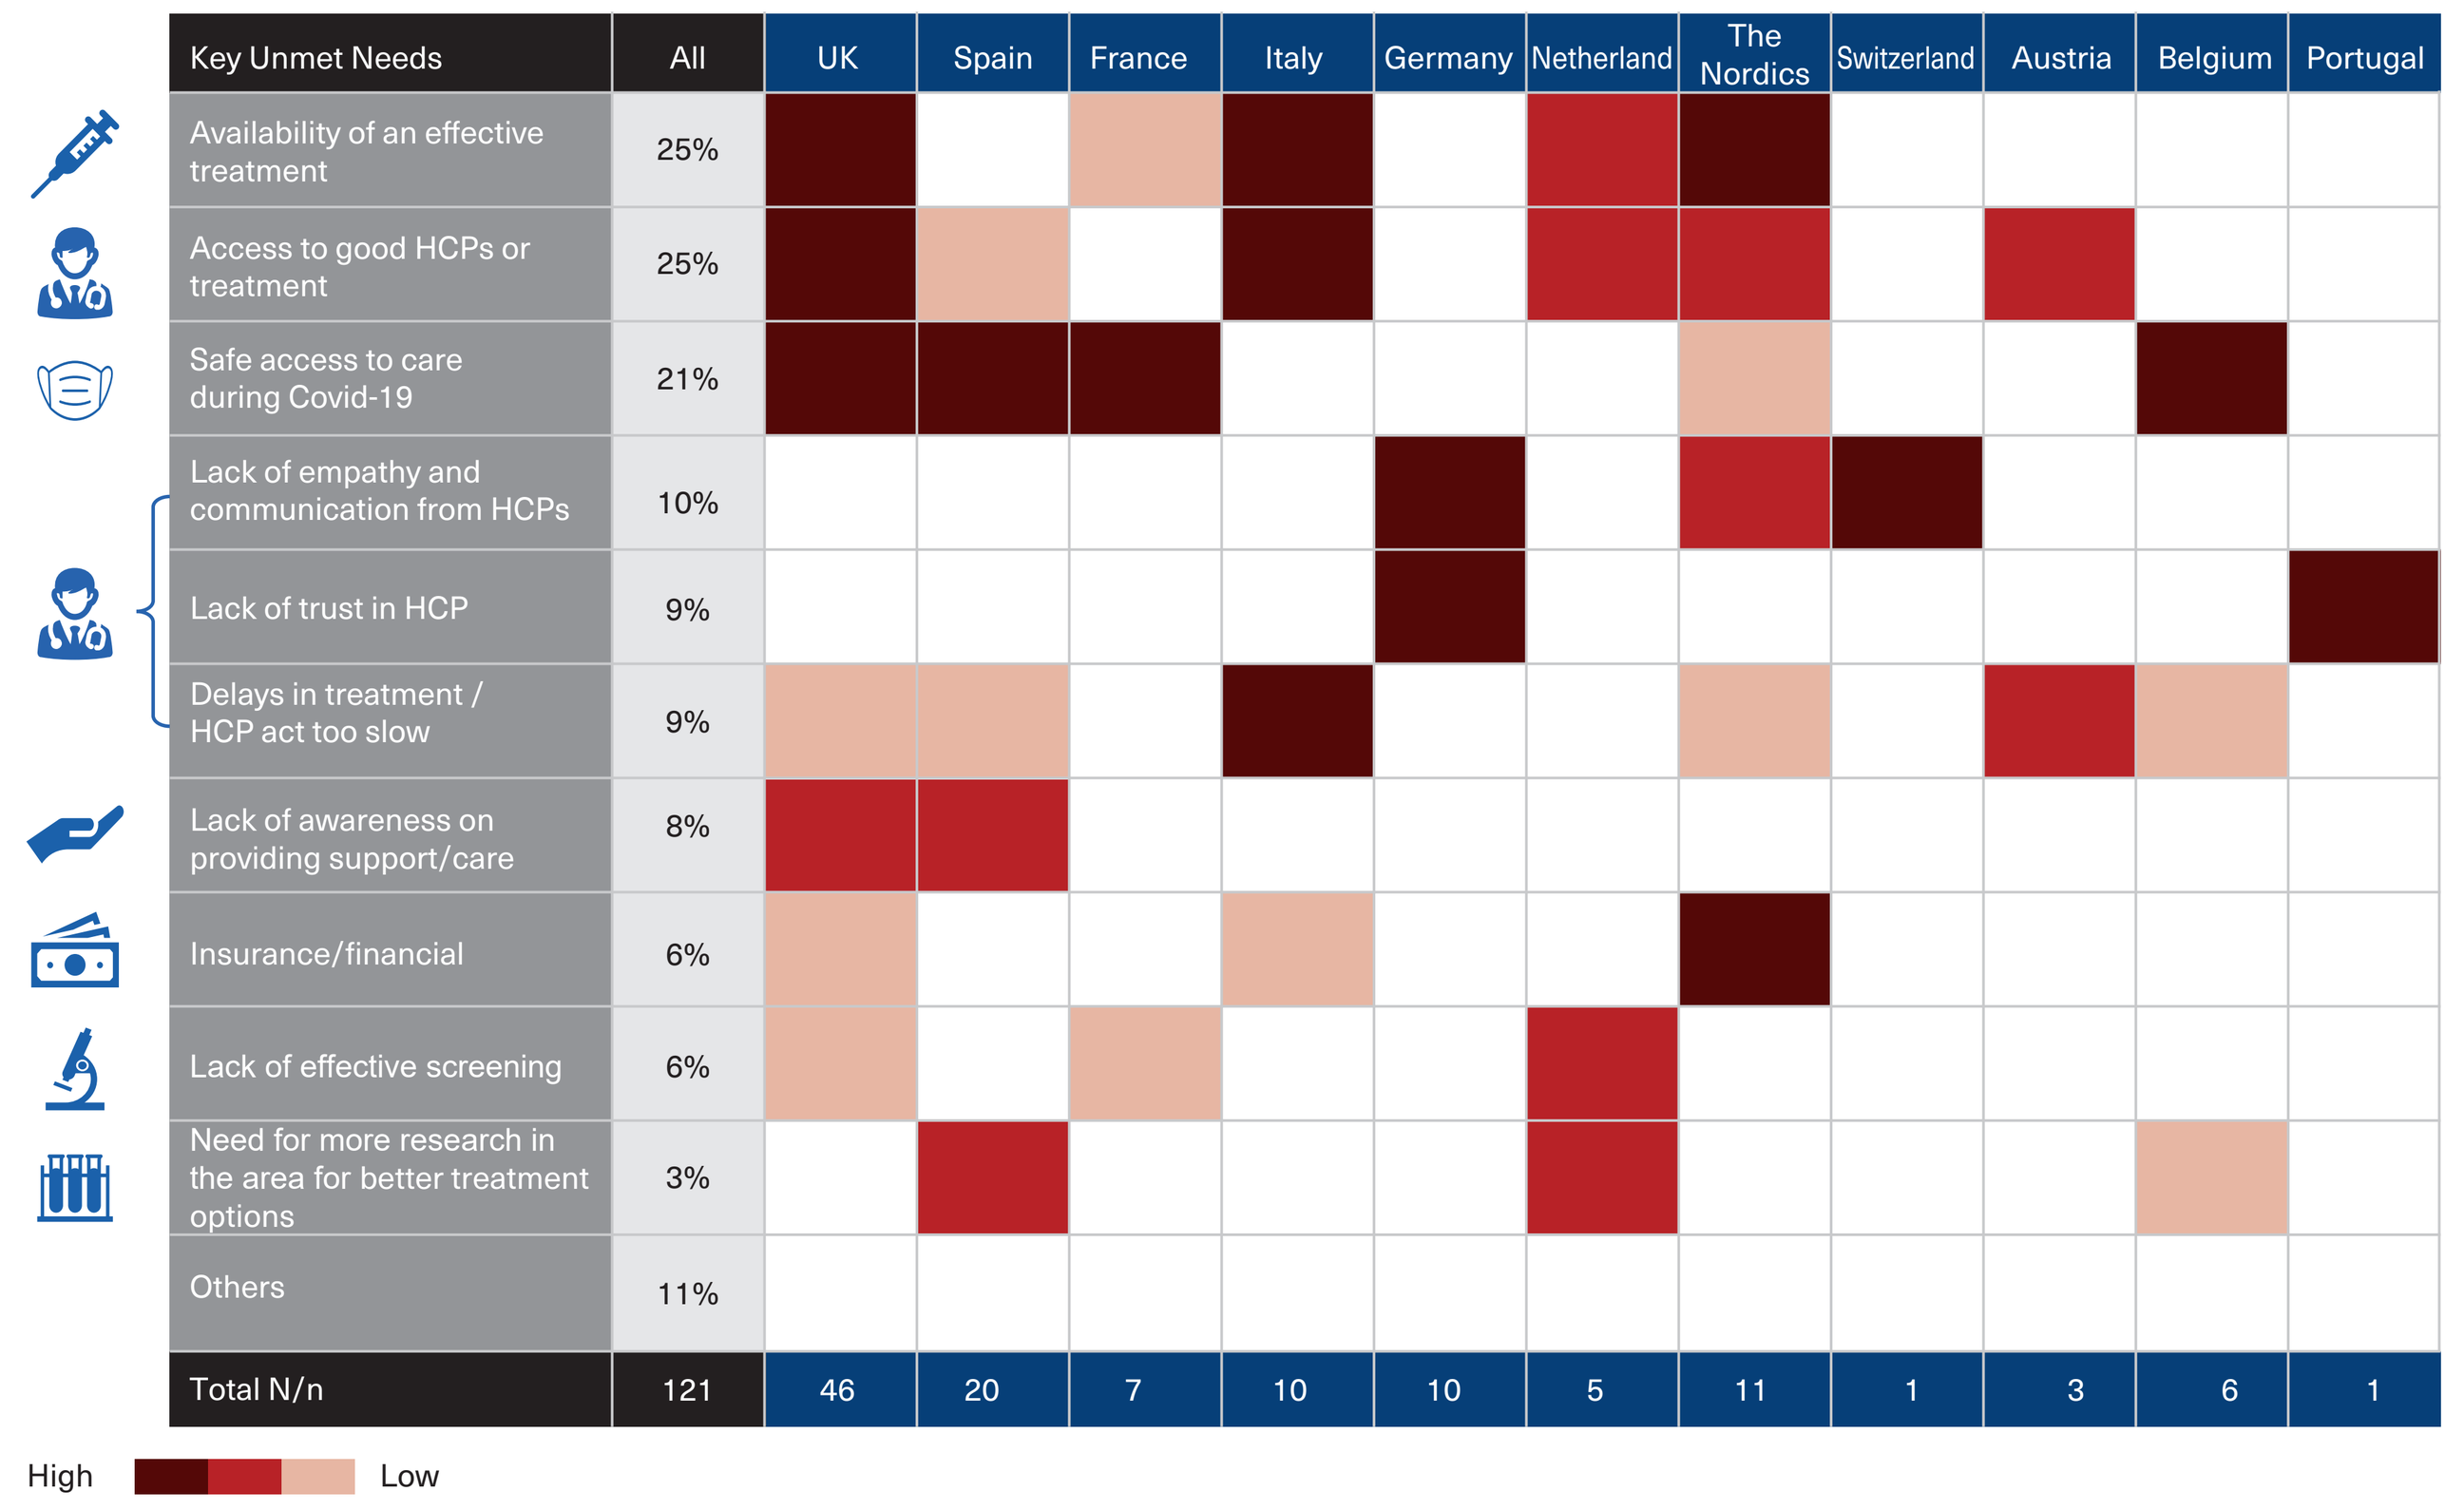
Key Unmet Needs
All
UK
Spain
France
Italy
Germany
Netherland
The Nordics
Switzerland
Austria
Belgium
Portugal
Availability of an effective treatment 25%
Access to good HCPs or treatment 25%
Safe access to care during Covid-19 21%
Lack of empathy and communication from HCPs 10%
Lack of trust in HCP 9%
Delays in treatment / HCP act too slow 9%
Lack of awareness on providing support/care 8%
Insurance/financial 6%
Lack of effective screening 6%
Need for more research in the area for better treatment options 3%
Others 11%
Total N/n 121 46 20 7 10 10 5 11 1 3 6 1
High
Low

STUDY LIMITATIONS

- Data gathered through SML cannot be confirmed as correct, therefore it is acknowledged that some information may be categorized incorrectly
- The quality of SML insights depends on the richness of conversations, and all data needs to be objectively determined to assess if it supports specific research questions
- Although in the public domain, SM users cannot fully consent to the use of their data in the study
- The SM population may not fully represent the whole community affected by melanoma

Acknowledgements

We acknowledge Victoria Sherwood, PhD (Novartis Ireland Ltd, Dublin, Ireland), for medical editorial assistance with this poster.
This study was sponsored by Novartis Pharmaceuticals.

Disclosures

JC is an employee of Novartis

References

1. Sandru, A., Voinea, S., Panaitescu, E. & Blidaru, A. Survival rates of patients with metastatic malignant melanoma. *J Med Life* 7, 572-576 (2014).
2. Erdmann, F. et al. International trends in the incidence of malignant melanoma 1953-2008--are recent generations at higher or lower risk? *Int J Cancer* 132, 385-400 (2013).
3. Forsea, A. M. Melanoma Epidemiology and Early Detection in Europe: Diversity and Disparities. *Dermatol Pract Concept* 10, e2020033 (2020).