Clinical and Economic Burden Attributable to Serotypes Included in Current and Future Pneumococcal Conjugate Vaccines in Argentinean Children Under Five Years of Age

Mercedes Mac Mullen,[1] M. Gabriela Abalos,[1] Carolina Carballo,[1] Emily Horn,[2] Johnna Perdrizet[2]

[1]*Pfizer Argentina*

[2]*Pfizer Inc, New York, NY, USA*

BACKGROUND

- Argentina introduced the 13-valent pneumococcal conjugate vaccine (PCV13) into its pediatric national immunization program (NIP) in 2012, upon which a significant reduction of pneumococcal disease was observed.[1,2]
- Two investigational higher-valent PCVs, a 15-valent (PCV15) and a 20-valent vaccine (PCV20), are currently in clinical development to include an additional 2 and 7 serotypes, respectively, in their formulations **(Figure 1)**.
- Despite the substantial impact of infant PCV13 vaccination in Argentina, disease burden attributable to non-PCV13 serotypes remains. Investigational PCVs may further reduce pneumococcal disease due to their broader serotype coverage.

Figure 1. Licensed and Investigational PCVs and Accompanying Serotypes Contained in Each Formulation

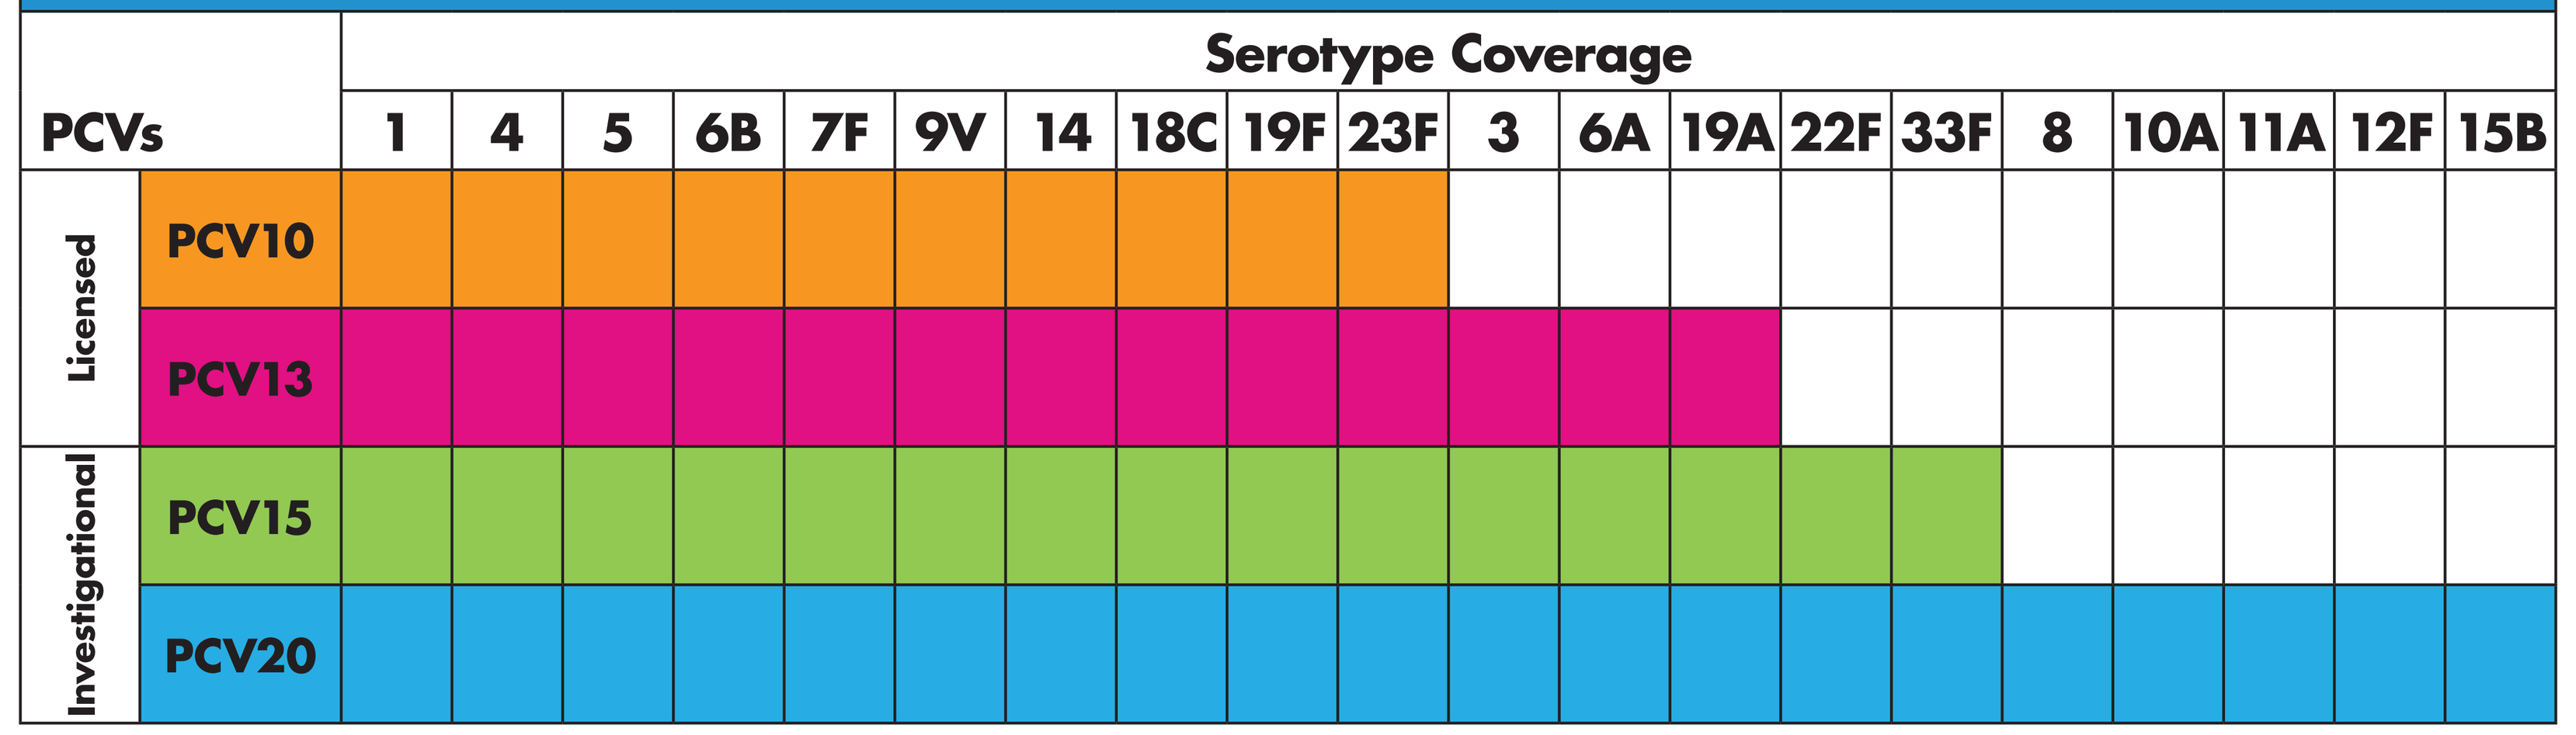
Serotype Coverage
PCVs
1 4 5 6B 7F 9V 14 18C 19F 23F 3 6A 19A 22F 33F 8 10A 11A 12F 15B
Licensed
PCV10
PCV13
Investigational
PCV15
PCV20

OBJECTIVE

- To estimate and compare the annual clinical and economic burden caused by serotypes contained in licensed PCVs (PCV10 and PCV13) and investigational PCVs (PCV15 and PCV20) among Argentinean children <5 years of age.

METHODS

- A decision-analytic model was used to estimate the annual number of disease cases, deaths, and associated costs resulting from invasive pneumococcal disease (IPD), pneumonia, and acute otitis media (AOM) caused by PCV serotypes among children <5 years old in Argentina.

Data inputs

- Population data from 2021 were sourced from Instituto Nacional de Estadistica y Censos (INDEC) for children <5 years of age.[3]
- The latest PCV serotype coverage estimates were calculated using the reported serotypes responsible for IPD cases in the 2018 SIREVA II *Streptococcus pneumoniae* surveillance report.[4]
- Due to a lack of nationally representative current data, the incidence of IPD and all-cause AOM was estimated by averaging the reported incidences in Italy,[5] France,[6-8] and Spain.[9,10] The incidence of all-cause pneumonia was sourced from published literature.[11]
- Among IPD cases, an estimated 23% were meningitis and 77% were bacteremia.[4]
- It was assumed that 74% and 40% of all-cause pneumonia and AOM cases, respectively, were attributable to *Streptococcus pneumoniae*.[12,13]
- Bacteremia, meningitis, and inpatient pneumonia carry a risk of death, and their respective case fatality rates (CFRs) were taken from published reports and literature.[14-16]
- Direct costs associated with each disease state and payer subsector were derived from Urueña et al. (2011) and adjusted to 2020 US dollars (USD).[17] An estimated 13.5% of the population uses the private healthcare subsector, 31.5% uses the public healthcare subsector, and 55.0% uses the social health insurance (social security) subsector.[18]

Analysis

- The serotype distribution was extrapolated to calculate the number of cases of bacteremia, meningitis, inpatient and outpatient pneumonia, and AOM.
- CFRs and associated costs were applied to relevant outcomes to calculate the number of deaths and total direct costs.

RESULTS

- PCV20 is estimated to cover 55% of current pneumococcal disease, with PCV10, PCV13, and PCV15 covering 19%, 36%, and 40%, respectively **(Figure 2)**.
- PCV20 serotypes are responsible for 175 IPD cases, 137,114 inpatient and outpatient pneumonia cases, 215,684 AOM cases, and 147 deaths annually in children <5 years of age, which is substantially more than the pneumococcal disease burden attributable to serotypes contained in lower-valent PCVs **(Table 1)**.
- Pneumococcal disease in Argentinean children <5 years old represents a substantial economic burden, with serotypes contained in PCV20 costing $3.93 million in the private healthcare system, $7.02 million in the public healthcare system, and $14.82 million in the social security system, amounting to more than $25.77 million annually **(Table 2)**.

Figure 2. Serotype Coverage for Licensed and Investigational PCVs in Argentinean Children <5 Years of Age

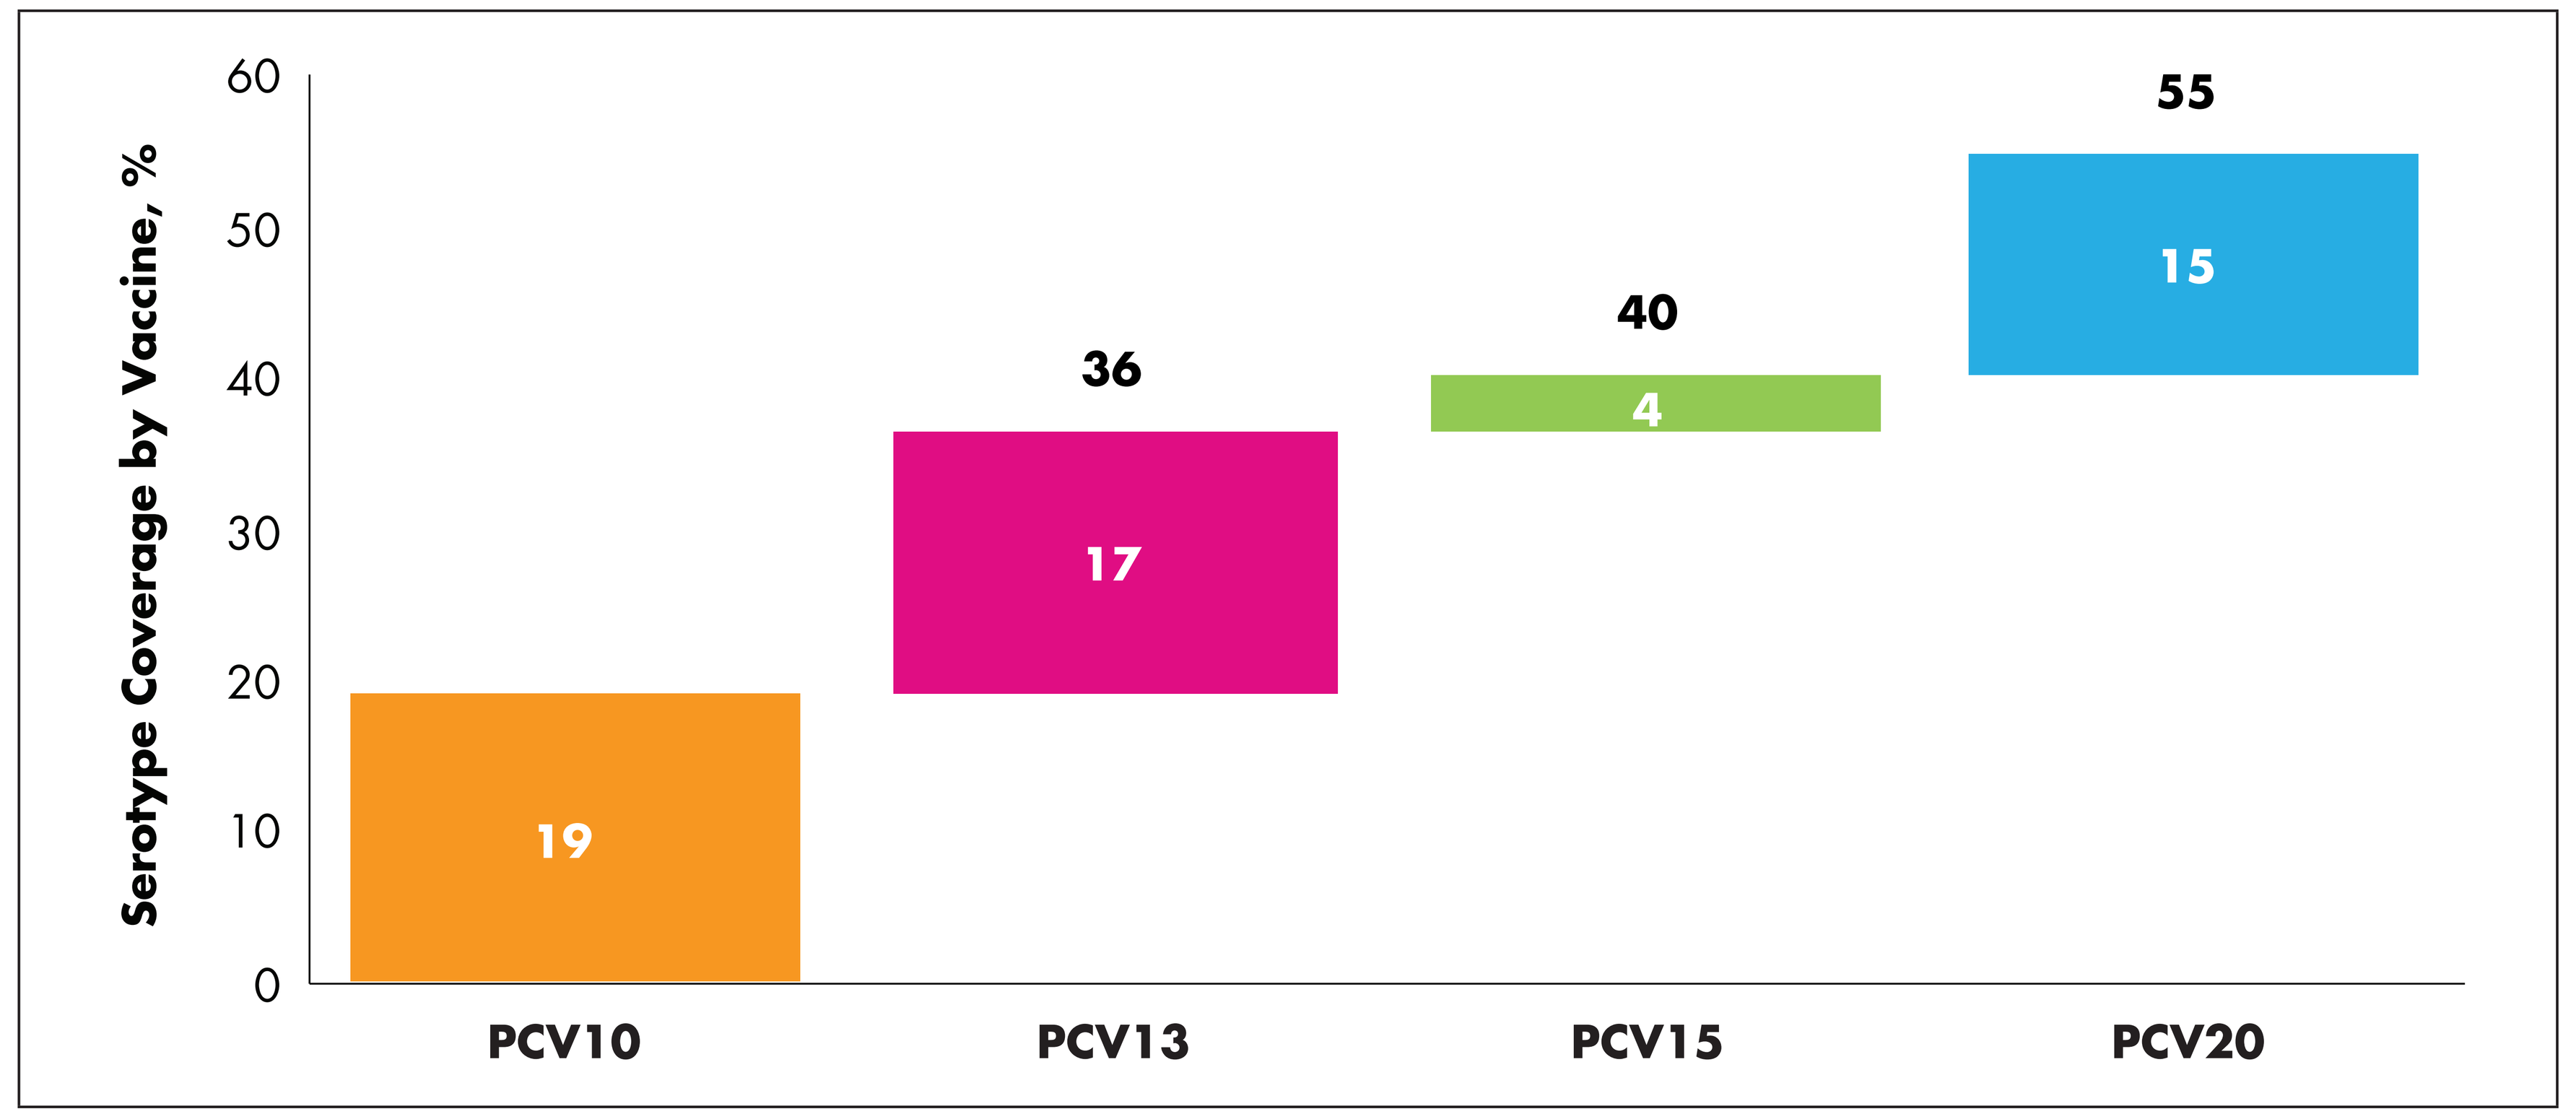
Serotype Coverage by Vaccine, %
0
10
20
30
40
50
60
19
17
36
4
40
15
55
PCV10
PCV13
PCV15
PCV20

Table 1. Estimated Annual Clinical and Economic Burden Caused by Serotypes Contained in Licensed and Investigational PCVs

Outcome	PCV10	PCV13	PCV15	PCV20
Total cases	123,770	233,787	258,999	352,973
IPD cases	61	116	128	175
Inpatient PNE cases	5,495	10,379	11,498	15,670
Outpatient PNE cases	42,584	80,437	89,111	121,444
AOM cases	75,630	142,856	158,262	215,684
Total deaths	52	97	108	147
Total cost ($USD)	$9,037,133	$17,070,140	$18,911,037	$25,772,564
IPD cost	$37,736	$71,278	$78,965	$107,616
Inpatient PNE cost	$5,366,184	$10,136,126	$11,229,237	$15,303,562
Outpatient PNE cost	$1,663,406	$3,141,989	$3,480,831	$4,743,787
AOM cost	$1,969,807	$3,720,747	$4,122,004	$5,617,598

AOM=acute otitis media; IPD=invasive pneumococcal disease; PCV=pneumococcal conjugate vaccine; PNE=pneumonia; USD=US Dollars.

Table 2. Estimated Annual Economic Burden Caused by Serotypes Contained in Licensed and Investigational PCVs by Healthcare System Subsector ($USD)

Cost	PCV10	PCV13	PCV15	PCV20
Private subsector	$1,379,040	$2,604,854	$2,885,769	$3,932,818
IPD costs	$5,250	$9,917	$10,986	$14,973
Inpatient PNE costs	$900,008	$1,700,016	$1,883,351	$2,566,691
Outpatient PNE costs	$224,150	$423,394	$469,054	$639,242
AOM costs	$249,632	$471,527	$522,378	$711,913
Public subsector	$2,462,927	$4,652,195	$5,153,902	$7,023,902
IPD costs	$10,958	$20,698	$22,931	$31,251
Inpatient PNE costs	$1,420,909	$2,683,939	$2,973,383	$4,052,222
Outpatient PNE costs	$459,234	$867,441	$960,989	$1,309,666
AOM costs	$571,826	$1,080,116	$1,196,599	$1,630,764
Social security subsector	$5,195,166	$9,813,091	$10,871,366	$14,815,843
IPD costs	$21,528	$40,663	$45,048	$61,393
Inpatient PNE costs	$3,045,267	$5,752,171	$6,372,503	$8,684,650
Outpatient PNE costs	$980,022	$1,851,154	$2,050,788	$2,794,879
AOM costs	$1,148,349	$2,169,104	$2,403,027	$3,274,921

AOM=acute otitis media; IPD=invasive pneumococcal disease; PCV=pneumococcal conjugate vaccine; PNE=pneumonia; USD=US Dollars.

CONCLUSIONS

- **Pneumococcal disease due to PCV13 serotypes in Argentina has decreased because of continued PCV13 use in the pediatric NIP.**
- **Substantial clinical and economic burden remains in Argentina even after more than a decade of PCV use in infants, which is largely driven by serotypes not contained in currently licensed PCVs.**
- **The investigational PCV20 covers an additional 36% and 19% of serotypes compared with the currently available PCV10 and PCV13, respectively, and has the potential to provide substantial public health and economic impact in Argentina.**
- **It will be important to continue monitoring local epidemiology in the future to better understand the potential clinical and economic burden that investigational vaccines may be able to address.**

REFERENCES

1. López EL, et al. *J Pediatric Infect Dis Soc.* 2018;7(1):30-5.
2. Chapman R, et al. *Vaccine.* 2020;38(45):7138-45.
3. Instituto Nacional de Estadistica y Censos. Proyecciones Provinciales de Población por Pexo y Prupo de Pdad 2010-2040. 1a ed. 2013. Available at: https://www.indec.gob.ar/ftp/cuadros/publicaciones/proyecciones_prov_2010_2040.pdf. Accessed October 19, 2021.
4. Efron A, et al. Información Sobre la Vigilancia de las Neumonías y Meningitis Bacterianas. SIREVA II. OPS. 2018 *Streptococcus pneumoniae.* 2018. Available at http://antimicrobianos.com.ar/ATB/wp-content/uploads/2019/10/Tablas-vigilancia-SIREVA-II-Spn-2018-1.pdf. Accessed October 19, 2021.
5. Ansaldi F, et al. *Pathogens.* 2020;9(2):76.
6. Santé Publique France, Institut de Veille Sanitaire. Bulletin de Reseau de Surveillance des Infections Invasives Bactériennes. 2016. Available at: https://www.santepubliquefrance.fr/maladies-et-traumatismes/maladies-et-infections-respiratoires/infections-a-pneumocoque/documents/bulletin-national/bulletin-epibac-n-6-du-reseau-de-surveillance-des-infections-invasives-bacteriennes. Accessed October 19, 2021.
7. Marom T, et al. *JAMA Pediatr.* 2014;168(1):68-75.
8. Caeymaex L, et al. *Pediatr Infect Dis J.* 2014;33(5):533-6.
9. Ministerio de Sanidad, Servicios Sociales e Igualdad. Instituto de Información Sanitaria. Registro de Altas – CMBD. 2017. Available at: https://pestadistico.inteligenciadegestion.mscbs.es/publicoSNS/S. Accessed October 20, 2021.
10. Liese JG, et al. *Epidemiol Infect.* 2014;142(8):1778-88.
11. Rudan I, et al. *J Glob Health.* 2013;3(1):010401.
12. Gentile A, et al. Etiology of pneumonia and invasive bacterial disease (IBD) among hospitalized children in Argentine highlights importance of S. *Pneumoniae* and its prevention. Poster presented at: 4th Vaccine and ISV Annual Global Congress; October 3-5, 2010; Vienna, Austria.
13. Paganini, HR. Las Infecciones en Pediatria. Diagnostico y Tratamiento. Imprecolor SA. Chapter 2. 2009.
14. Berberian G. *Arch Argent Pediatr.* 2014;112(4):332-6.
15. Minesterio de Salud Argentina. Introducción de la Vacuna Conjugada Contra el Neumococo al Calendario Nacional de Inmunizaciones de la República Argentina. 2011. Available at: https://bancos.salud.gob.ar/sites/default/files/2018-10/0000000438cnt-2011-10_lineamientos-neumococo-2011.pdf. Accessed October 19, 2021.
16. Gentile A, et al. *Arch Argent Pediatr* 2015;113(6):502-9.
17. Urueña A, et al. *Vaccine.* 2011;29(31):4963-72.
18. Caeme Innovación Para la Salud. El Sistema de Salud Argentino. 2020. Available at: https://www.caeme.org.ar/archivo-weber/el-sistema-de-salud-argentino/. Accessed October 19, 2021.

DISCLOSURES

This study was funded by Pfizer Inc.

For more information please contact:
Emily Horn
Pfizer Inc
Email: Emily.Horn@pfizer.com

International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe 2021 Virtual Conference; November 30–December 3, 2021