Poster XXXX

TBC

Patient Reported Experience Measure for Adult Patients with Sickle Cell Disease (SCD) in Greece

Markouri A[1], Tripsa T[2], Vareli K[2], Papastefanou V[1]

[1]Novartis Hellas, Athens, A1, Greece; [2]IQVIA, Athens, Greece

Scan to obtain:
- Poster

http://novartis.medicalcongressposters.com/Default.aspx?doc=fe99a
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- There is a significantly high cost burden among SCD patients, mainly driven by diagnostic tests, medication costs and travel expenses.
- Patients express their need for involvement in treatment choice, while SCD impacts their everyday activities and work.
- A better healthcare system, working opportunities and financial support is required for SCD patients.
- SCD patients believe that there is a need for getting more information on new treatments, while also improvement on pain crisis care and training of HCPs is also needed.
- This study provided a national patient's perspective on SCD healthcare that may help shape future SCD service delivery, design, and funding in Greece

This study is sponsored by Novartis Pharma AG

Poster presented at ISPOR Europe 2021, held virtually on 30 November-3 December 2021

BACKGROUND AND OBJECTIVE

- Sickle cell disease (SCD) is an inherited blood disorder that causes episodic pain, persistent organ damage, and a reduction of life expectancy. (1)
- Vaso-occlusive crises (VOC) and pain are hallmark symptoms of SCD. These painful episodes have been reported as intense stabbing or throbbing, resulting in frequent ED (Emergency Department) and hospital visits. (2)
- Understanding patient experiences, quality of life, and treatment needs is essential to efficient healthcare delivery. A better patient experience leads to higher treatment adherence, preventive services use, and healthcare utilization. (1, 3)
- Therefore, understanding and measuring SCD patients' care experiences is essential in order to inform healthcare improvement. (4)
- The objective of the current study is to develop and implement a patient-reported experience measure (PREM) for patients with SCD, to gather insights on patient's experiences related to healthcare services they receive, and to assess SCD impact on the patient's daily and social life.

METHODS

- The study was conducted in two phases:
 - The first phase included a qualitative assessment using a focus group discussion with adult patients and representatives of patient association groups (PAGs) across Greece via IQVIA digital platform. Patients/PAGs members provided information about living with SCD, their personal journey, the burden of disease, difficulties in everyday life, information they seek for SCD, the role of family and PAGs. Apart from the group discussion, the SCD participants validated the questionnaire that would be used in the second phase. They were asked to evaluate each question when regards to its relevance with SCD and its clarity. They were also asked if there was any other issue/topic that the questionnaire did not cover that should be added. Patients' comments and suggestions were incorporated into the final version of the quantitative questionnaire.
 - The second phase was a quantitative study, wherein the SCD patients participated in an online survey. The survey was implemented with the usage of a Picker methodology based structured questionnaire, tested by participants of phase 1. Patients participated into the quantitative part were recruited by physicians working in centers throughout Greece. (**Figure 1**)

Figure 1. Methodology used for PREM tool assessment

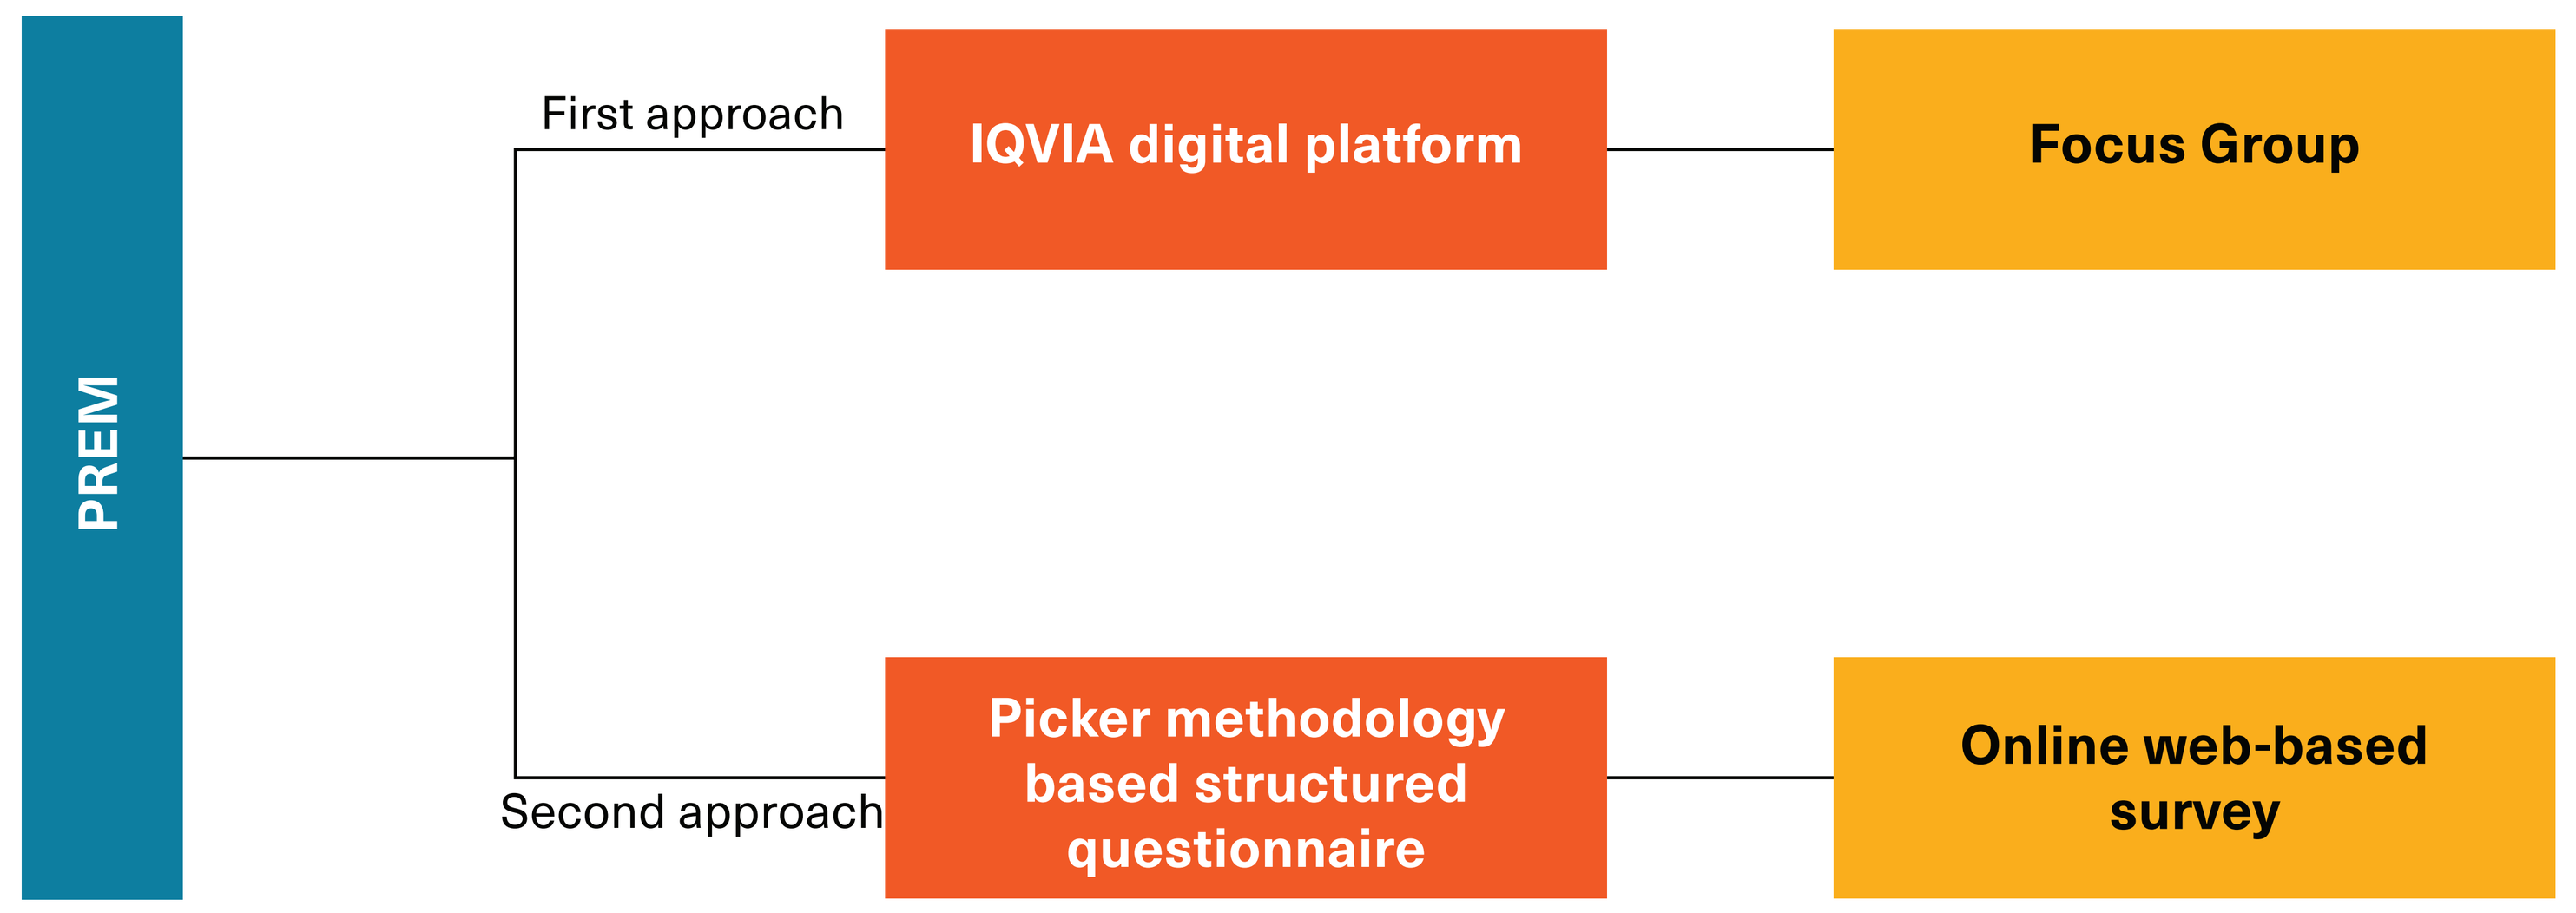

PREM
First approach
IQVIA digital platform
Focus Group
Second approach
Picker methodology based structured questionnaire
Online web-based survey

PREM: Patient Reported Experience Measure

RESULTS

SCD Patients in qualitative phase:

- Six SCD patients who were PAG members participated and shared their experiences via group discussion.
 - Patients described pain episodes or crisis as the worse part of the disease, that burdens them, both psychologically and practically.
 - Patients also referred to the limitations and barriers that disease has placed upon them in the physical realm of their lives.
 - Patients separated or distinguish their experience of living with SCD as 'before and after hydroxyurea and referred to hydroxyurea as a milestone on treating SCD.
 - In rural locations, health care professionals (HCP's) were less aware on SCD crisis management while patient in crisis were not adequately managed due to lack of hospital facilities in the emergency rooms.
 - Patient's family, HCPs and the patient's associations were mentioned as the three main pillars of support for SCD patients.
 - Financial burden from the disease was a key issue among SCD patients, driven mainly from expenses for supportive medications, medical tests and out of pocket expenses.
- A web-based survey included 40 SCD patients, the majority of them were female (57%) and (40%) of them were under 40.

RESULTS (CONTINUED)

Patient journey through the Health Care System:

- A total of (93%) SCD patients visited hematologists on a regular basis, and (43%) had consultation by their physician more frequently than 3 months.
- Patients were very satisfied with their physicians (93%) in terms of discussion upon issues related to treatment/medication, whereas evaluated the nurses lower in SCD knowledge (65%).
- Nearly (53%) of the patients preferred to discuss with HCPs and be informed on new advancements.
- Visits to emergency room (ER) were declared by (68%) of the patients, and (73%) of these found the ER staff to be friendly: however, the ratings for SCD knowledge were lower (43%).

Sources of information for SCD:

- All patients claimed to be informed on SCD, with (58%) expressed a need to receive more information.
- (80%) of total patients searched for new developments, (63%) for new treatments, and (48%) exchanged views with other patients, especially the younger ones (63%).

Living with SCD:

- Patients felt adequately informed on how to take their medication (80%) but desired more involvement in treatment alternatives (78%).
- Nearly (65%) of the patients found enough support dealing with SCD, while (35%) would like to have more help.
- Only (13%) of SCD patients felt their disease was adequately treated by the healthcare system.
- A total of (43%) of patients stated negative impact in finding a job due to their disease whereas (45%) indicated that pain crises caused difficulties in their work.
- Regarding disease burden, (42%) of patients reported that they had out of pocket expenses for treating their disease, with diagnostic tests being the top mentioned type of care they had to pay followed by medication costs and doctor visits. Moreover, (58%) of additional disease burden costs were travel costs. (**Figure 2**)

Figure 2. Out of Pocket cost for SCD

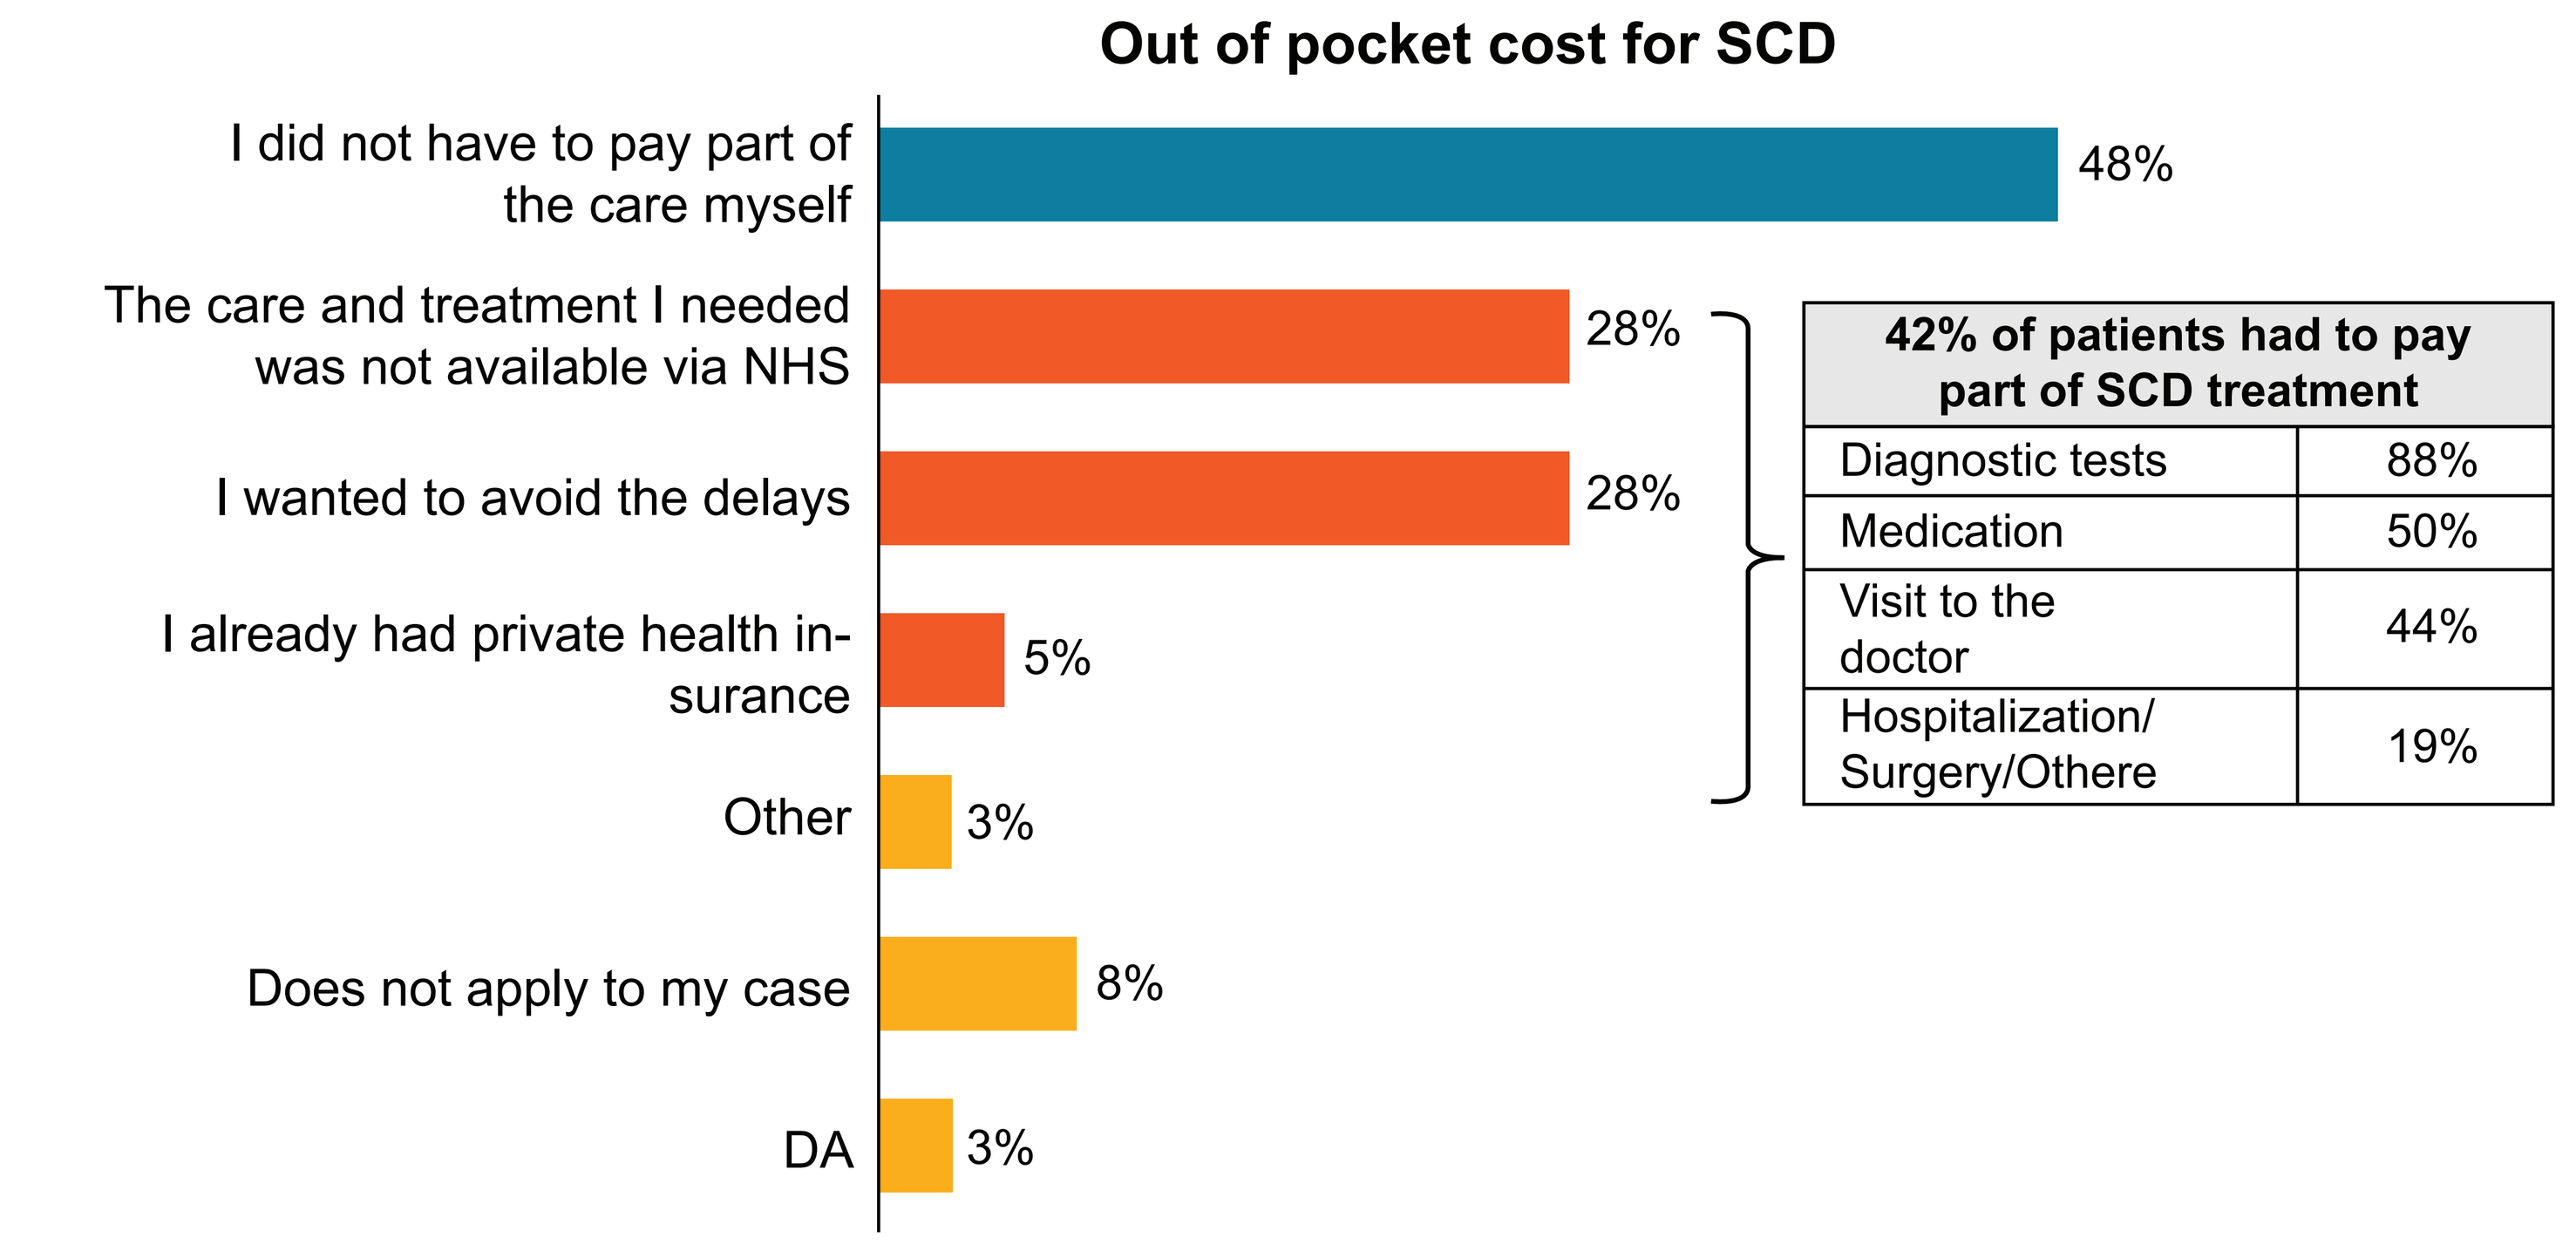

Out of pocket cost for SCD
I did not have to pay part of the care myself
48%
The care and treatment I needed was not available via NHS
28%
I wanted to avoid the delays
28%
I already had private health in-surance
5%
Other
3%
Does not apply to my case
8%
DA
3%

42% of patients had to pay part of SCD treatment	
Diagnostic tests	88%
Medication	50%
Visit to the doctor	44%
Hospitalization/ Surgery/Othere	19%

DA: Do not answer
Source: IQVIA Health Economics • April 2021
IQVIA • Novartis • Prem Tool SCD • May 2021

Quality of Life (QoL)-attitudes and behaviours:

- More than half of the patients (55%) were happy with their health status, and (45%) with their SCD management.
- Patients stated that SCD impacted mostly their social life (hobbies, social activities, contacts) and less everyday life. (**Figure 3**)
- Nearly (50%) of the patients felt uncomfortable, sad (48%) and felt different from others (43%) at least once per month.
- Patients expressed the need for a better healthcare system followed by jobs with less physical requirements and better financial assistance. (**Figure 4**)

Figure 3. Constraints to social & everyday life due to SCD

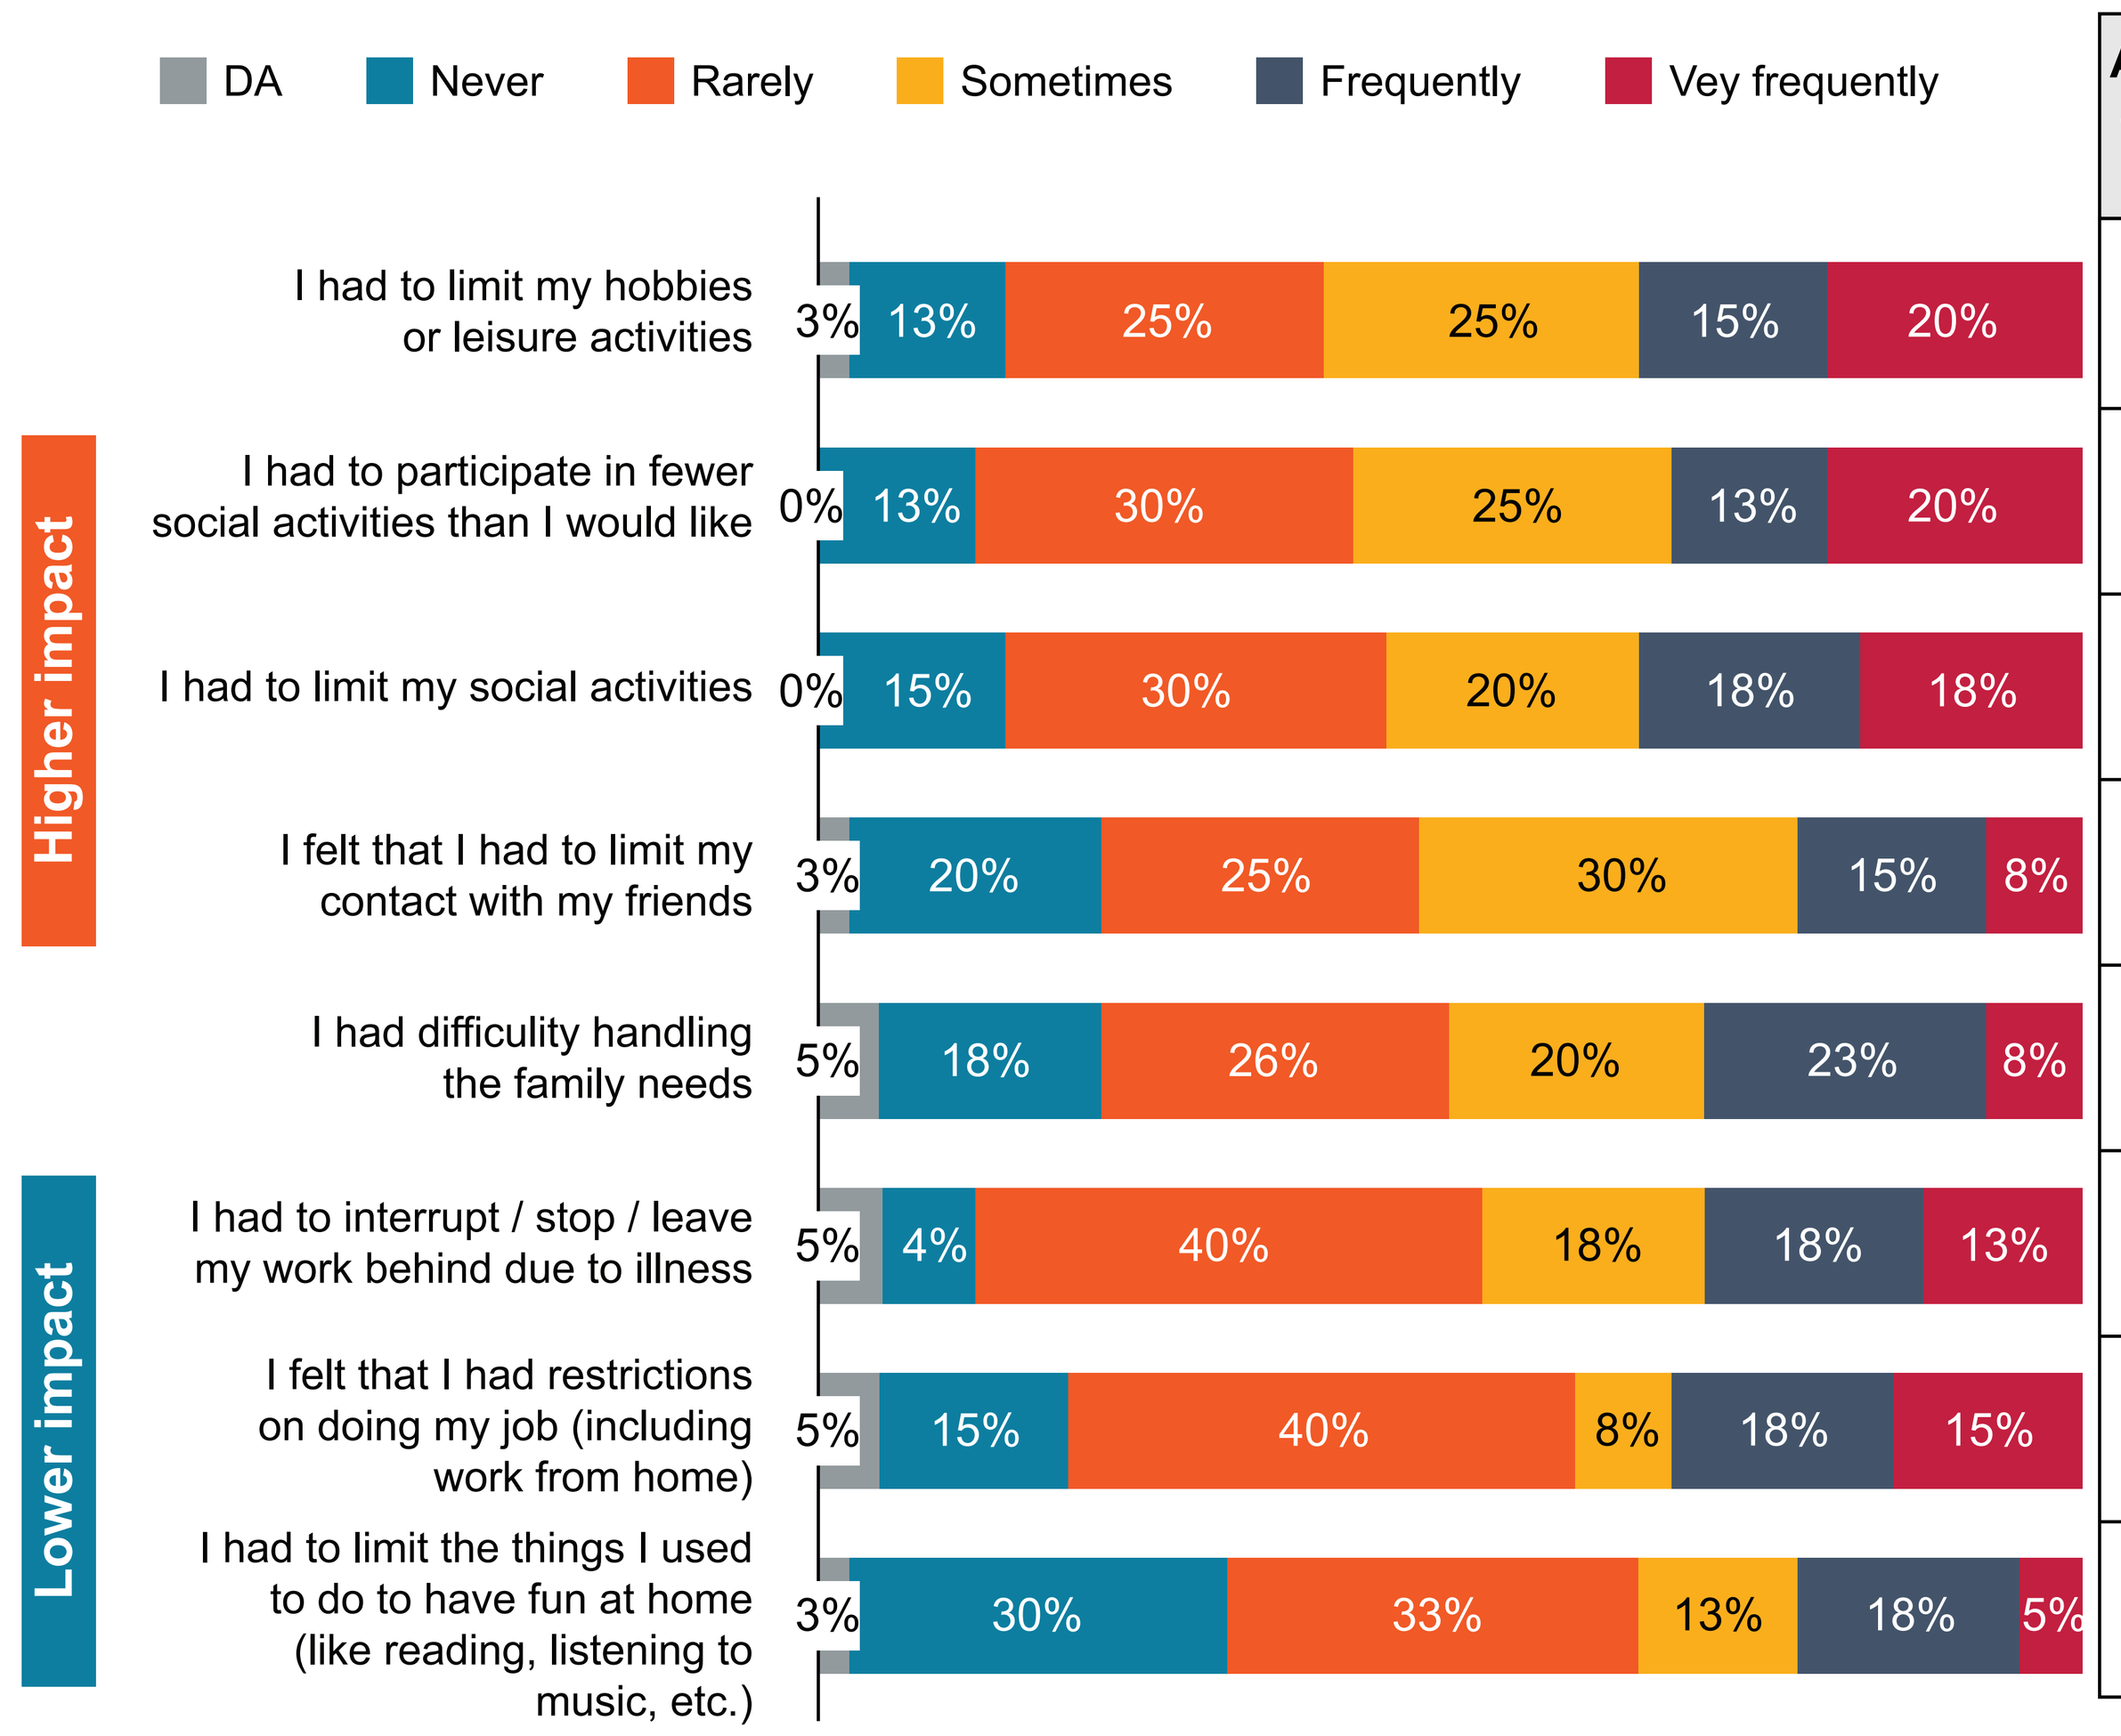

DA
Never
Rarely
Sometimes
Frequently
Vey frequently
Higher impact
Lower impact
I had to limit my hobbies or leisure activities
3% 13% 25% 25% 15% 20%
I had to participate in fewer social activities than I would like
0% 13% 30% 25% 13% 20%
I had to limit my social activities
0% 15% 30% 20% 18% 18%
I felt that I had to limit my contact with my friends
3% 20% 25% 30% 15% 8%
I had difficulty handling the family needs
5% 18% 26% 20% 23% 8%
I had to interrupt / stop / leave my work behind due to illness
5% 4% 40% 18% 18% 13%
I felt that I had restrictions on doing my job (including work from home)
5% 15% 40% 8% 18% 15%
I had to limit the things I used to do to have fun at home (like reading, listening to music, etc.)
3% 30% 33% 13% 18% 5%

	At least once a month	Less than once per month	Never
I had to limit my hobbies or leisure activities	60%	25%	13%
I had to participate in fewer social activities than I would like	58%	30%	13%
I had to limit my social activities	55%	30%	15%
I felt that I had to limit my contact with my friends	53%	25%	20%
I had difficulty handling the family needs	50%	28%	18%
I had to interrupt / stop / leave my work behind due to illness	48%	40%	8%
I felt that I had restrictions on doing my job (including work from home)	40%	40%	15%
I had to limit the things I used to do to have fun at home (like reading, listening to music, etc.)	35%	33%	30%

DA: Do not answer
Source: IQVIA Health Economics • April 2021
IQVIA • Novartis • Prem Tool SCD • May 2021

Figure 4. SCD patient's needs that state should offer

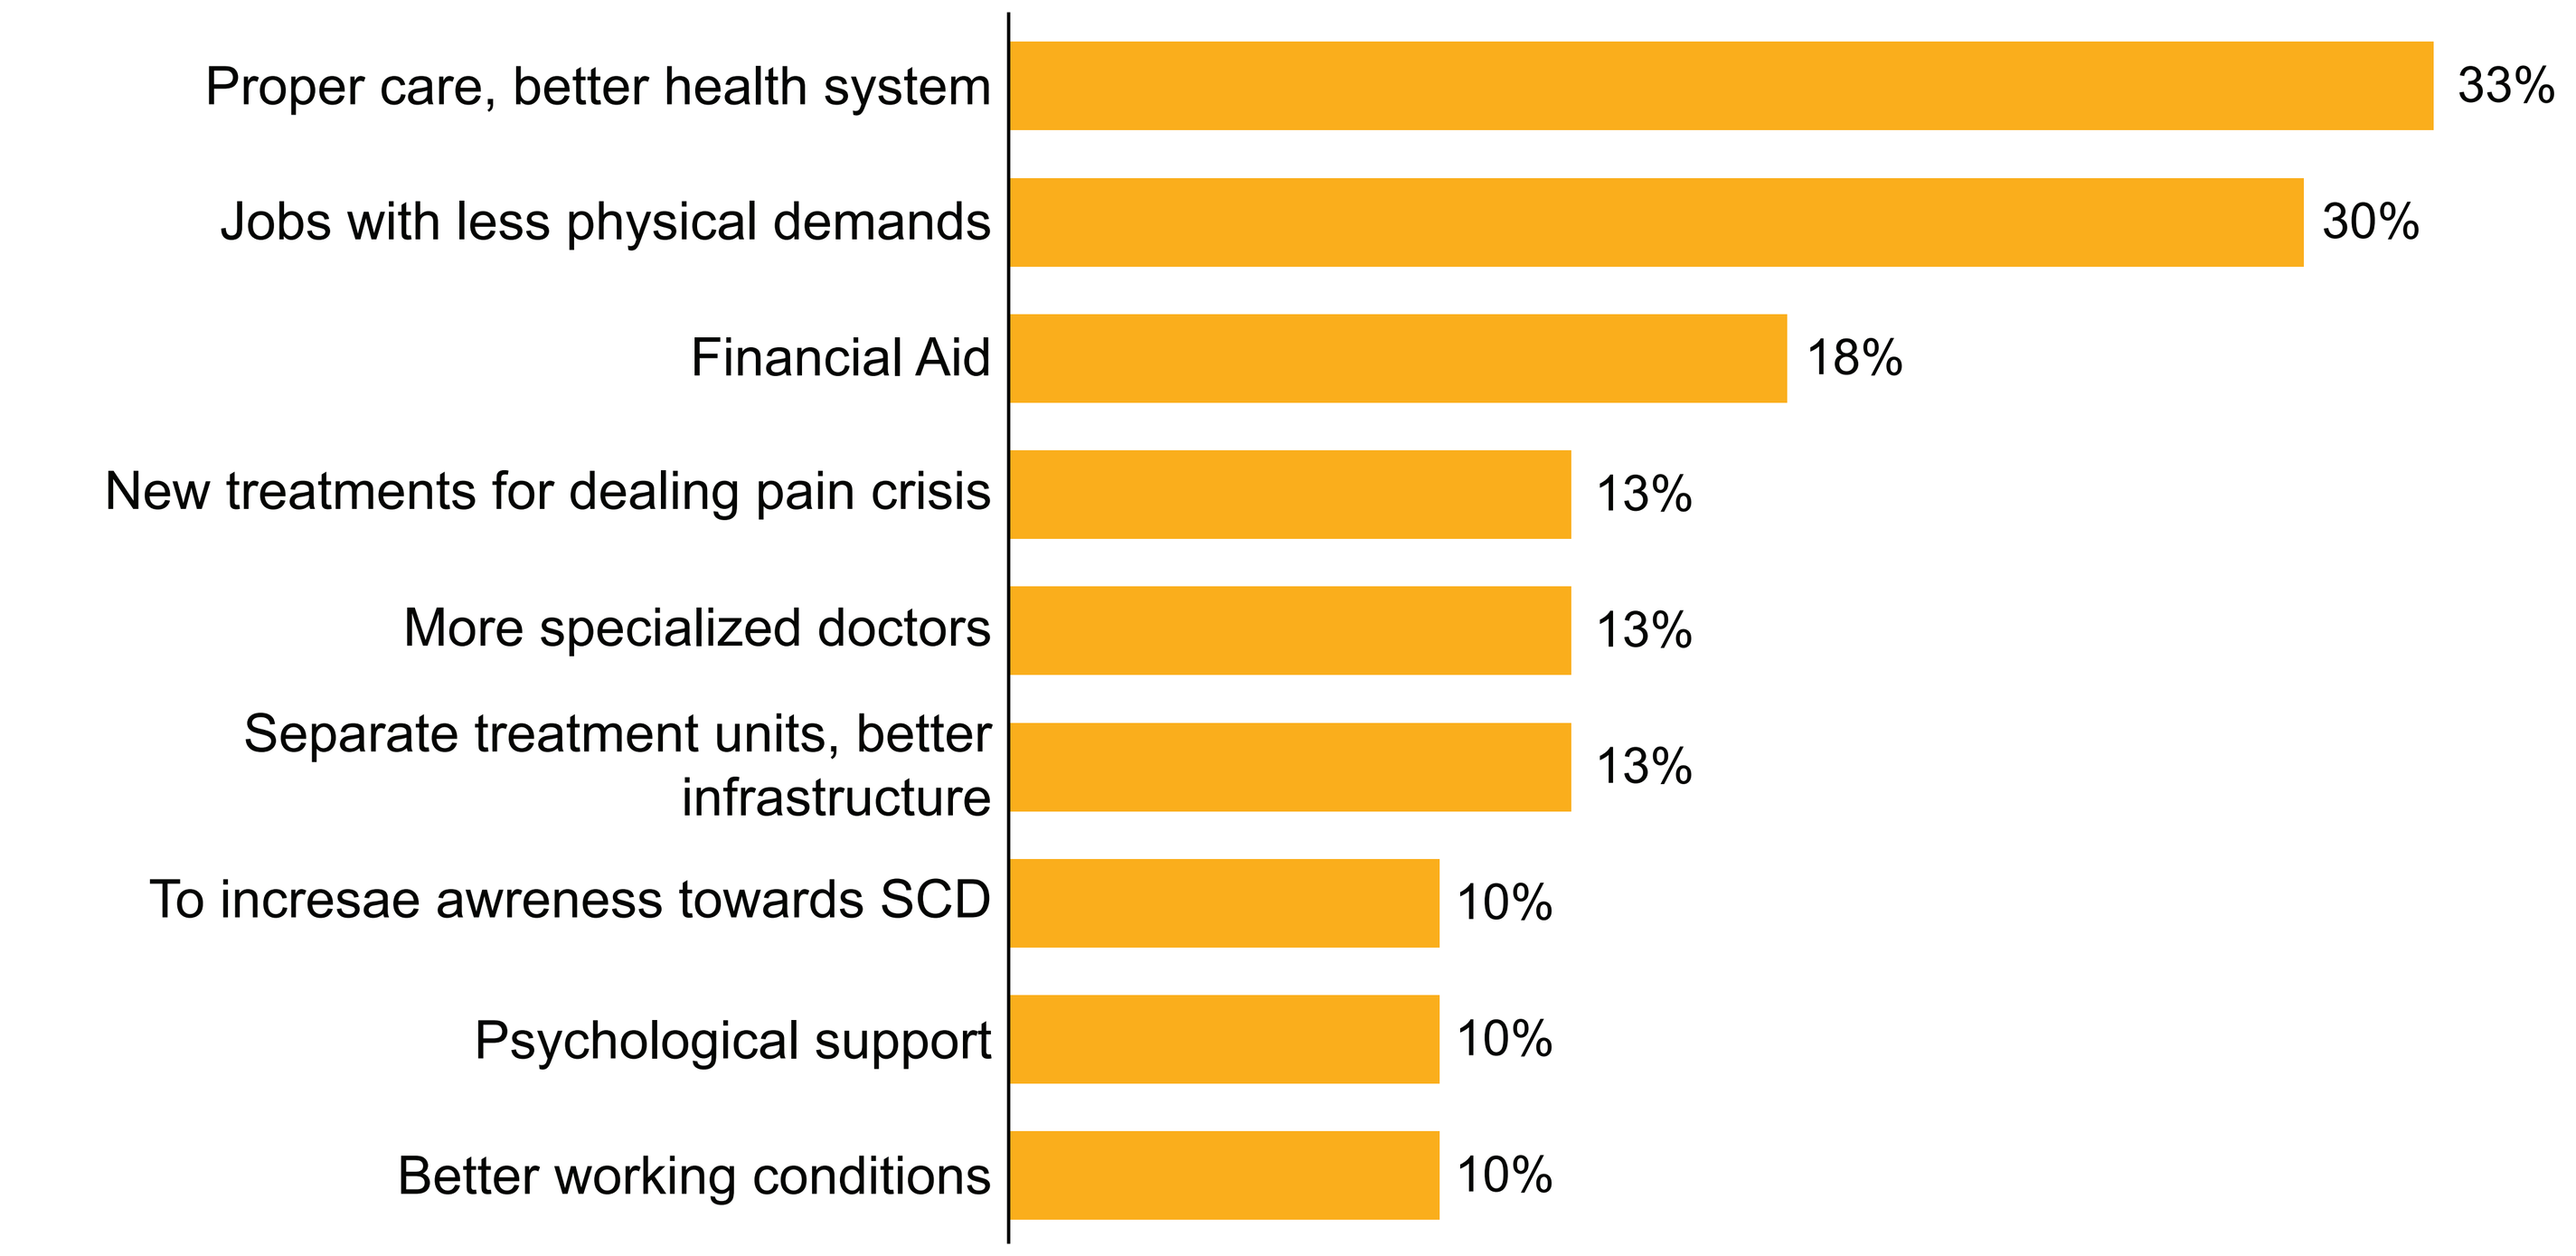

Proper care, better health system
33%
Jobs with less physical demands
30%
Financial Aid
18%
New treatments for dealing pain crisis
13%
More specialized doctors
13%
Separate treatment units, better infrastructure
13%
To incresae awreness towards SCD
10%
Psychological support
10%
Better working conditions
10%

Source: IQVIA Health Economics • April 2021
IQVIA • Novartis • Prem Tool SCD • May 2021

Acknowledgements

The authors acknowledge Rahul Kumar (Novartis, Hyderabad) for providing medical writing assistance with this poster.

Conflict of Interest

Argyro Markouri is a former employee of Novartis that worked for Novartis during study conduction
Tina Tripsa, Katerina Vareli are employees of IQVIA Athens Greece
Vasilis Papastefanou is employee of Novartis Hellas Athens Greece

References

1. Chakravorty S TA, Witwicki C, Hay H, Mkandawire C, Ogundipe A, Ojeer P, Whitaker A, Thompson J, Siz-mur S, Sathyamoorthy G. Patient-reported experience measure in sickle cell disease. *Archives of disease in childhood.* 2018;103(12):1104-9.
2. Sarri G, Bhor M, Abogunrin S, Farmer C, Nandal S, Halloway R, Revicki DA. Systematic literature review and assessment of patient-reported outcome instruments in sickle cell disease. *Health and quality of life outcomes.* 2018 Dec;16(1):1-3.
3. Knisely MR, Pugh N, Kroner B, Masese R, Gordeuk V, King AA, Smith SM, Gurney JG, Adams R, Wun T, Snyder A. Patient-reported outcomes in sickle cell disease and association with clinical and psychoso-cial factors: Report from the sickle cell disease implementation consortium. *American journal of hema-tology.* 2020 Sep;95(9):1066-74.
4. Witwicki C, Hay H, Tallett A. Piloting a new Patient Reported Experience Measure for Sickle Cell Dis-ease: A report of the findings. *Picker Institute Europe.* 2015 Dec:1-55.