Patient Experience Information (PEI) and Patient Involvement in Health Technology Assessment (HTA) Processes in 7 European Countries Using Immuno-Oncology Examples: How Can the Patient Voice Make an Impact?

Poster No. PCN318

A. P. Brogan[1], C. K. Patel[1], N. Schoenherr[2], S. Bobiak[3]

[1]AESARA, Chapel Hill, NC, USA [2]Merck KGaA, Darmstadt, Germany, [3]EMD Serono Research & Development Institute, Inc., Billerica, MA, USA; an affiliate of Merck KGaA, Darmstadt, Germany

BACKGROUND

- Patients can provide unique insights into diseases they have and the treatments they are offered (**Figure 1**). To advance the inclusion of the patient perspective in medicine development, the European Patients' Academy of Therapeutic Innovation (EUPATI) was formed and released guidance in 2018 for activities to support patient involvement in HTA decision-making.[1] The guidance suggests:
 - Involving patients in the identification and prioritization of health technologies for assessment
 - Inviting patient organizations to comment on draft scoping documents
 - Including the patient perspective as part of the HTA evaluation
 - Describing how the patient perspective was included in the assessment and impacted the decision in final published assessments
- Recent HTA appraisals in immuno-oncology (IO) and rare cancers provide an opportunity to analyze the inclusion of the patient perspective in HTA decisions and identify areas of improvement to best meet the guidance outlined by EUPATI

Ways in which patients can be included in HTA processes

Patient involvement

- Defined as patients, patient organizations, or caregivers engaging with HTA bodies in the evaluation of a new medicine. This could include:
 - Participating as part of an HTA review panel
 - Fulfilling requests for information from an HTA review committee

Patient experience information (PEI)

- Defined as evidence about the patient experience regarding experiences with the disease or treatment of interest provided. This could include:
 - Patient-reported outcomes (PROs)
 - Patient interviews
 - Patient journaling
 - Patient activity tracking
 - Patient preferences

Figure 1. Patients provide unique insights into diseases they have and the impact on their daily lives

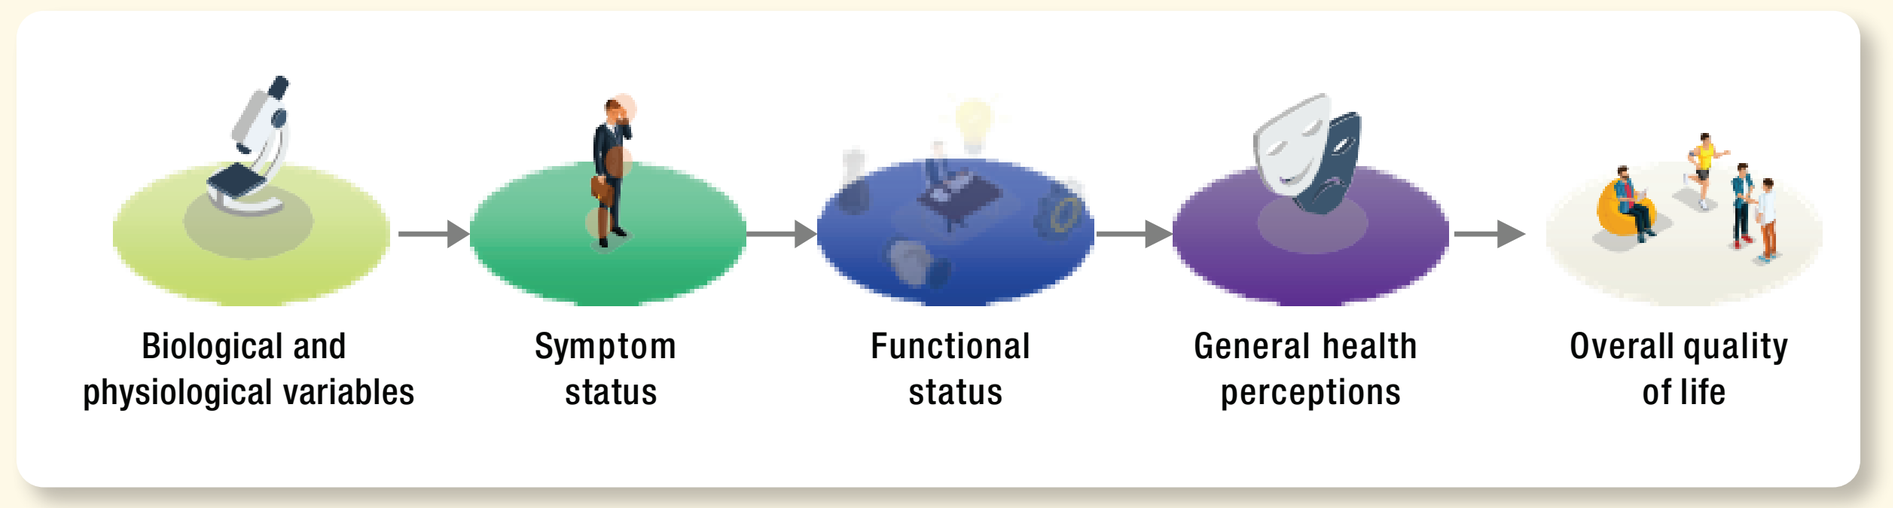
Biological and physiological variables
Symptom status
Functional status
General health perceptions
Overall quality of life

OBJECTIVE

- To describe how the new EUPATI guidance on patient involvement is being implemented practically at the country level and how the patient voice affects HTA recommendations using recent IO and rare cancer examples in 7 European countries

METHODS

- Targeted literature review of recently published HTA guidance by Institut für Qualität und Wirtschaftlichkeit im Gesundheitswesen (IQWiG), Haute Autorité de Santé (HAS), Tandvårds–och Läkemedelsförmånsverket (TLV), Zorginstituut Nederland (ZIN), and National Institute for Health and Care Excellence (NICE) on IO treatments across different cancer types to describe current implementation of PEI and patient involvement in HTA
- In-depth, qualitative interviews with seasoned HTA advisors in 7 European countries (**Table 1**) to identify gaps and potential impact of PEI/patient involvement on future HTA processes and decisions

Table 1. HTA advisor profiles and details about HTA processes in countries examined

Country	HTA advisor profile	HTA archetype country	Formal patient involvement in HTA	Patient voting rights for HTA decisions
France	Health economics professor and HAS advisor	Comparative effectiveness	Yes	Yes (potentially 3 of 21 voting seats at Transparency Commission)
Germany	Health economics professor and member of the arbitration board for drug pricing	Comparative effectiveness	Yes	No
Italy	Health economics professor and advisor to regional and national health authorities	Budget impact	No (currently AIFA has to make formal request; expected to change in 3-5 years)	No
The Netherlands	Health economics professor and ZIN advisor	Cost-effectiveness	Yes	No
Spain	Health economics professor and advisor to regional and national health authorities	Budget impact	Yes	No
Sweden	Professor and TLV advisor	Cost-effectiveness	Yes	Yes (1 board member for pricing and reimbursement)
United Kingdom	Health economics professor and advisor to NICE and SMC	Cost-effectiveness	Yes	No

AIFA, Agenzia Italiana del Farmaco; **SMC**, Scottish Medicines Consortium.

RESULTS

Targeted Literature Review

- Patient organizations are made aware of medicine appraisals by 2 basic mechanisms across all countries examined:
 - Public announcement, including social media
 - HTA agencies have running lists of accredited patient organizations that are notified when appraisals in their purview are underway
- At the country level, there is room for more participation from patient associations:
 - For example, 19 patient/care groups were invited to comment on the NICE appraisal of atezolizumab for breast cancer, but only 2 submitted written responses to the NICE appraisal committee[2]
 - A recent study by HAS in France found that patient association input was included in only 20% of medicines reviewed (2017-2018)[3]
- Among the published HTA documents reviewed (pembrolizumab, nivolumab, cemiplimab, avelumab, atezolizumab, durvalumab), there was a general lack of specificity about how the patient perspective was included in the assessment and its impact on the final HTA decision
 - Published HTA appraisal documents from Germany provided the most detail about how the patient perspective was included and to what extent it impacted the assessment
 - Pembrolizumab (first-line metastatic non-small cell lung cancer) and nivolumab (advanced renal cell carcinoma) were awarded "hints of added benefit" (hinweis auf zusatznutzen) ratings in the outcome category "health-related quality of life" (QOL) for providing statistically significant evidence regarding patients' health status and PRO data
 - Other IO appraisals resulted in "no added benefit" in the outcome category "health-related quality of life" when IQWiG was not able to find a statistically significant difference in PRO data between intervention and comparator
- In "cost-effectiveness countries" (the Netherlands, Sweden, the United Kingdom), PROs are frequently used as part of cost/quality-adjusted life years (QALYs) analyses. This analysis only captures a small fraction of the actual patient experience via a generic PRO tool that is often not sensitive to specific diseases. Few additional details are provided in the published HTA about the patient experience and to what extent it is captured in the cost-effectiveness analysis

HTA Advisor Interviews

Relative importance of PEI for HTA decisions

- PEI has a varied impact across European countries and may become more relevant in the future as the EUPATI guidance and the importance of the patient perspective continues to gain traction (**Figure 2**)
- The importance of PEI (as perceived by interviewees) did not differ between oncology in general or more specifically in IO indications, except for France. The HTA advisor from France noted that PEI can be more important for end-stage or metastatic cancer
- Lower ratings for PEI for IO medicines in France and Spain were attributed to the lack of familiarity with IO
- PEI can have a significant impact in Germany, particularly if there are safety/tolerability issues associated with the medicine under assessment or the comparator
- In order to maximize impact, PEI must be translated into country-specific outcomes that resonate with the respective payers and HTA archetypes (e.g. cost-effectiveness, comparative effectiveness or budget impact)

> "Spain is not a 'QALY country,' but a 'budget impact country.' Side effects have high costs to the healthcare system. If manufacturers can translate how adverse events lower costs to the system that would be helpful. Showing QOL/PRO data has impact to budget impact—[and] that would be powerful!"
>
> ~Spanish HTA Advisor

Figure 2. Importance of PEI for HTA decision-making*

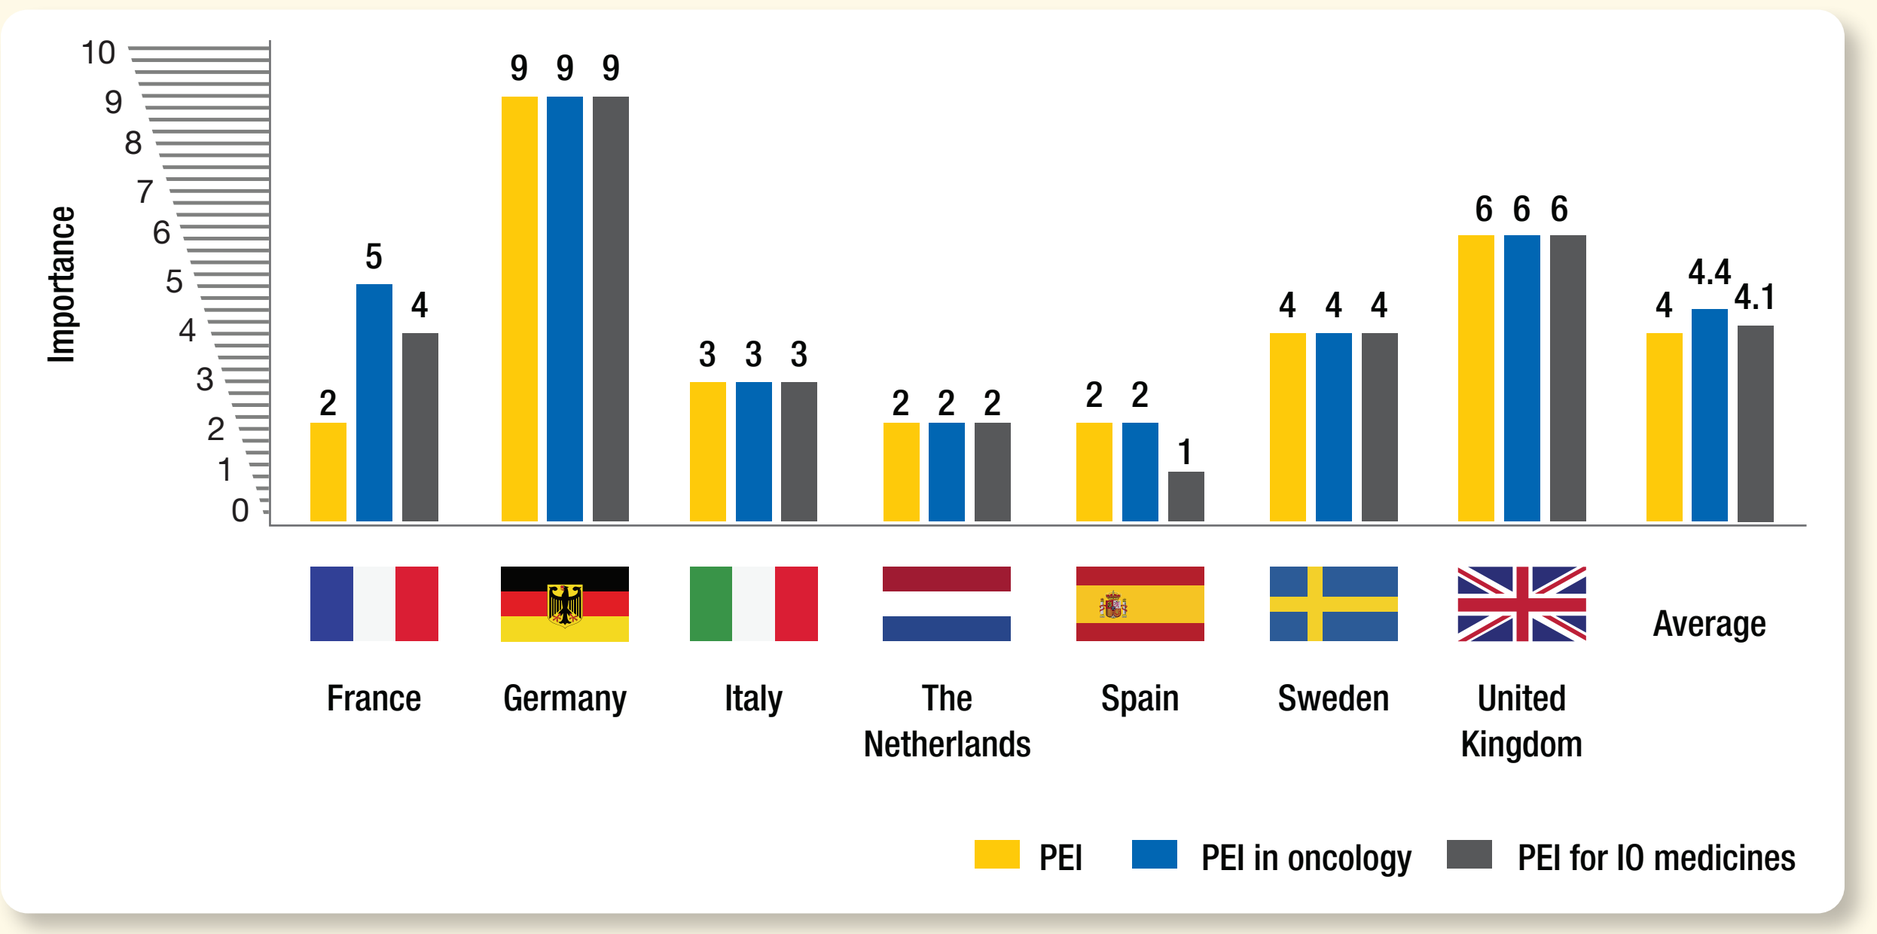
Importance
10
9
8
7
6
5
4
3
2
1
0
2 5 4
9 9 9
3 3 3
2 2 2
2 2 1
4 4 4
6 6 6
4 4.4 4.1
France
Germany
Italy
The Netherlands
Spain
Sweden
United Kingdom
Average
PEI
PEI in oncology
PEI for IO medicines

* Rated on a scale of 0 to 10, for which 0 is not important at all and 10 is extremely important.

> "PEI is critically important if there are safety/tolerability issues associated with the drug or comparator."
>
> ~Germany HTA Advisor

Importance of PEI for rare cancers

- Including patient information and input in HTA decisions for rare cancers is particularly relevant (**Figure 3**)
- Lack of information for rare cancers make PEI in rare cancers more important than in general oncology
- There is a higher need for information on rare cancers, for which the number of affected patients is lower; therefore, the patient voice can play a more important role in HTA decision-making

> "PEI is more important in rare or less common cancers. [There is a] need [to capture more] information about the disease and the patient's voice can play [an] important role for this!"
>
> ~Italian HTA Advisor

Figure 3. Importance of PEI for rare cancer types for HTA decision-making*

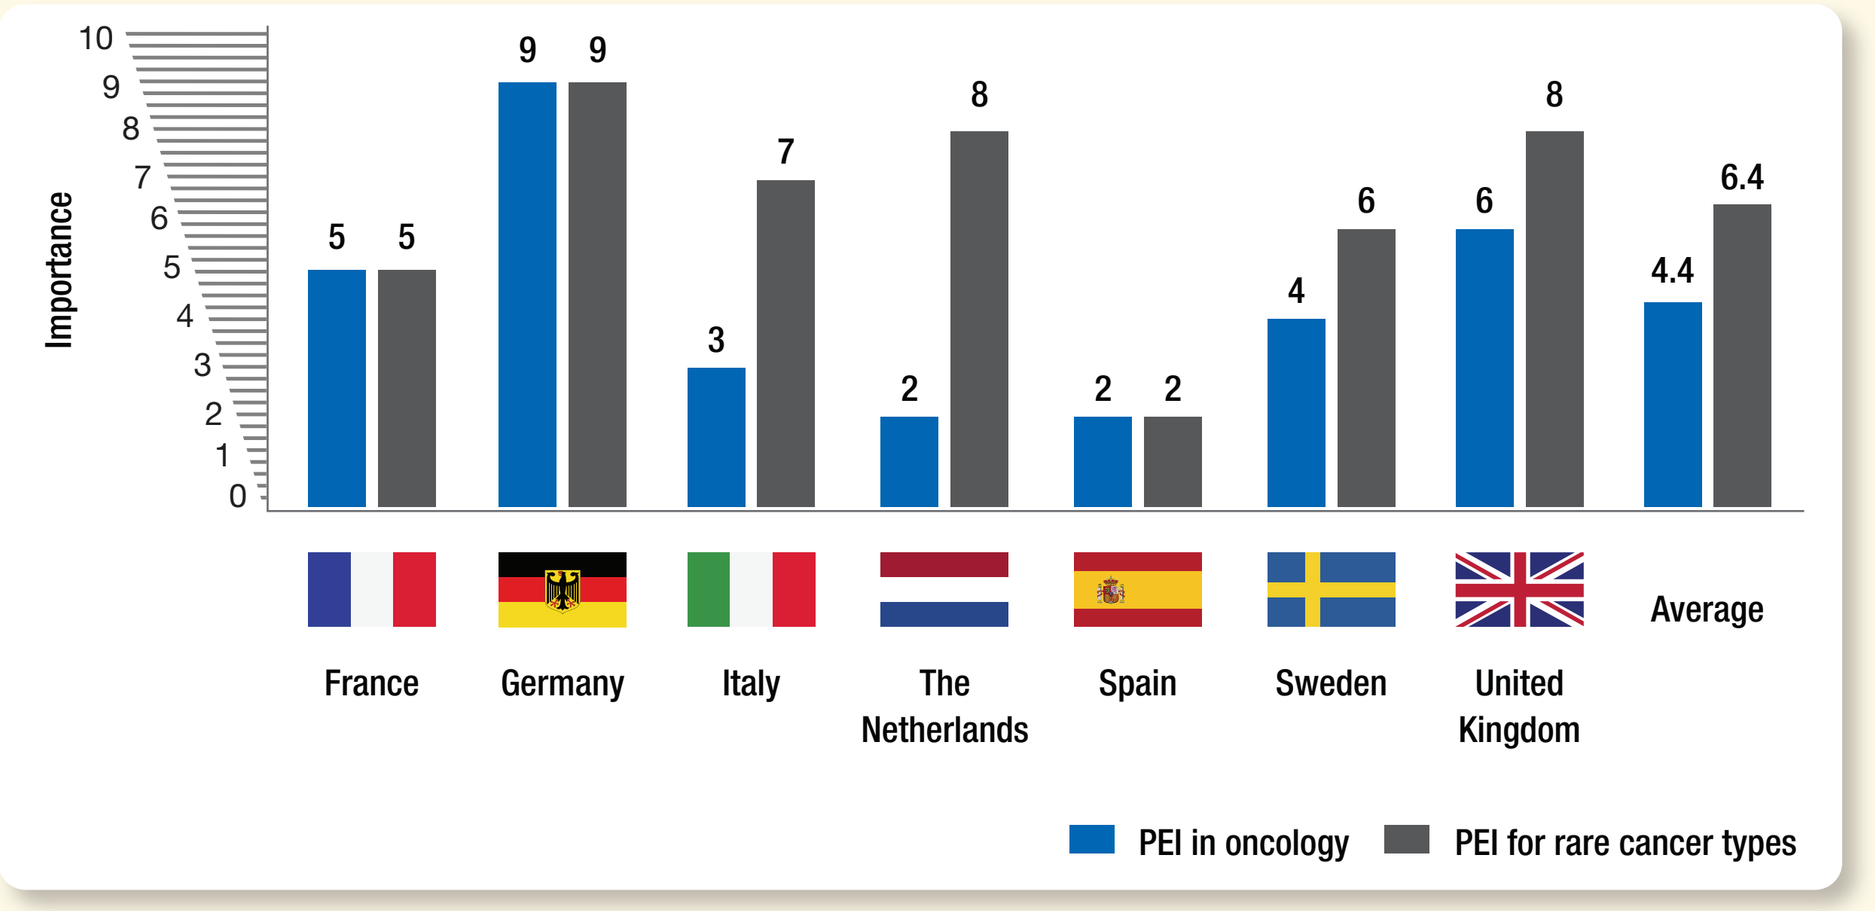
Importance
10
9
8
7
6
5
4
3
2
1
0
5 5
9 9
3 7
2 8
2 2
4 6
6 8
4.4 6.4
France
Germany
Italy
The Netherlands
Spain
Sweden
United Kingdom
Average
PEI in oncology
PEI for rare cancer types

* Rated on a scale of 0 to 10, for which 0 is not important at all and 10 is extremely important.

Importance of postprogression PEI for oncology HTA decision-making in IO indications

- Postprogression PEI provides additional insight to the patient perspective and can impact HTA decisions, not only in "cost-effectiveness countries" but also, for example, in Germany, where the Gemeinsamer Bundesausschuss requests postprogression PRO data (**Figure 4**)
- HTA advisors further indicated that postprogression PEI is most important for rare cancer types that are the least understood
- Postprogression PEI has been perceived as a missed opportunity in cost-effectiveness countries, particularly cost/QALY evaluations with extended time horizons
- Historically, postprogression PRO data has not been captured due to trial designs and definitions. HTA evaluators in "cost-effectiveness countries" are keen to understand the level of QALY decrease from pre- to postprogression as this will impact the cost/QALY analysis

> "Patient is in 3 health states—preprogression, postprogression, and dead… [Postprogression PRO] is very important, but poorly done because when patients progress, the [manufacturers] stop collecting data. But we want to know in cost/QALY model, what is the drop in progression based on QOL or symptoms—continue PRO follow-up during progression phase."
>
> ~UK HTA Advisor

Figure 4. Importance of postprogression PEI for HTA decision-making*

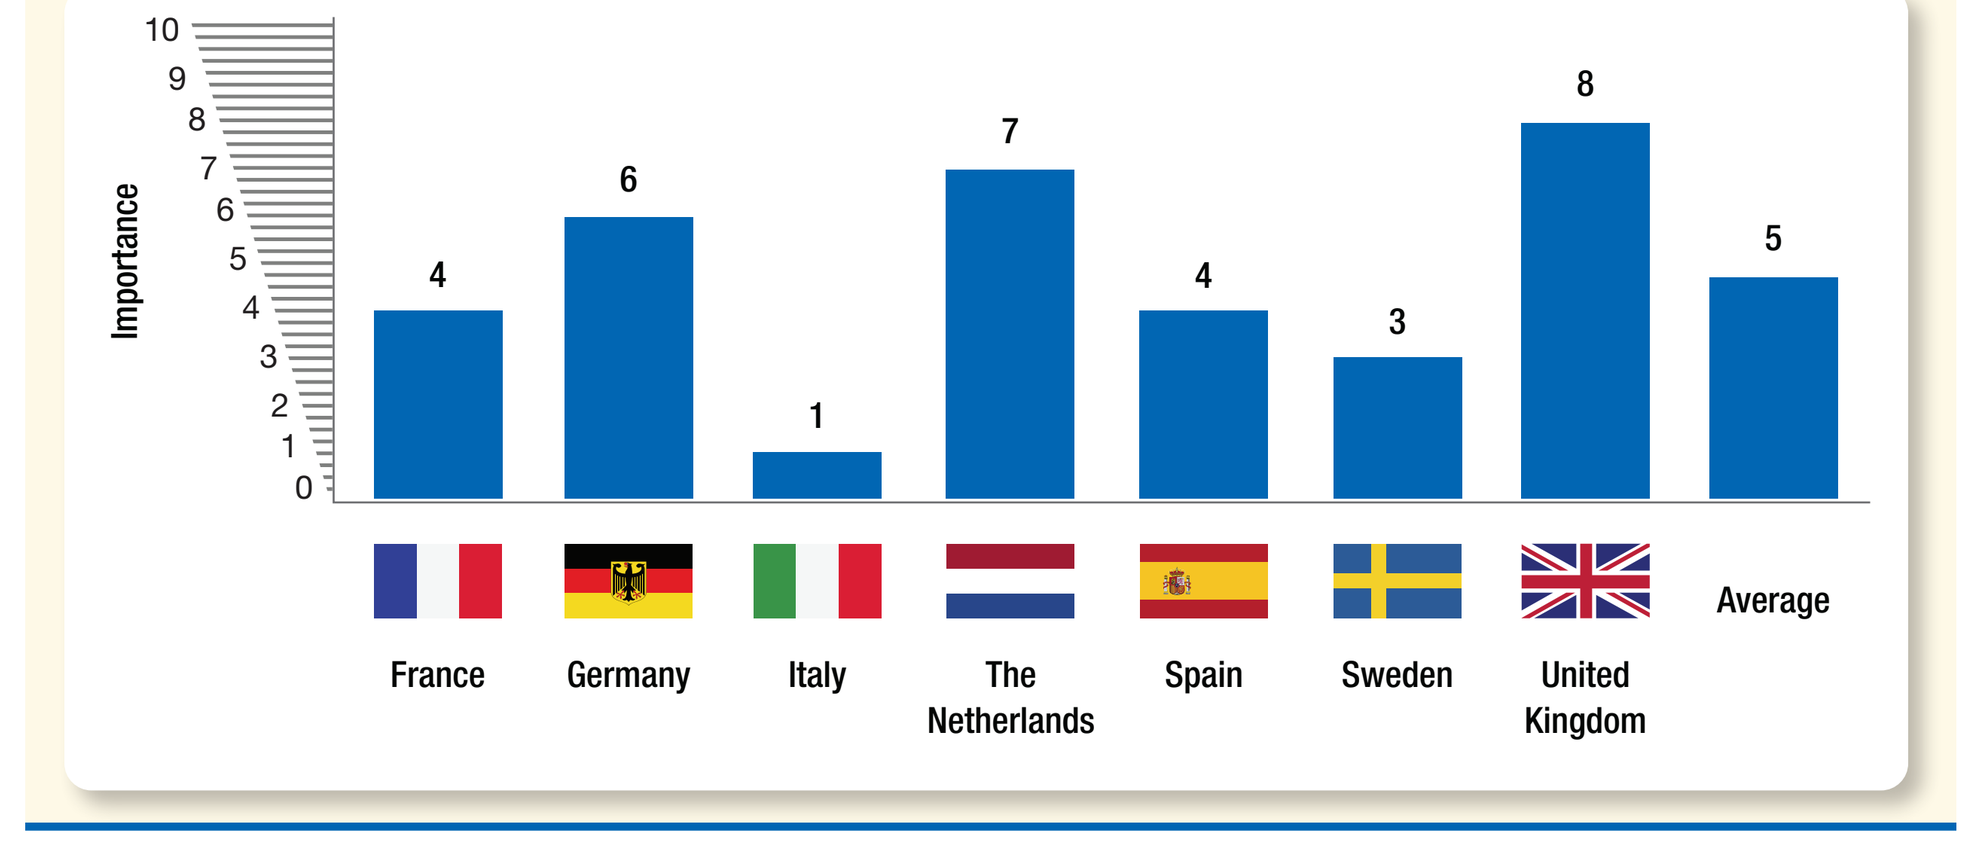
Importance
10
9
8
7
6
5
4
3
2
1
0
4
6
1
7
4
3
8
5
France
Germany
Italy
The Netherlands
Spain
Sweden
United Kingdom
Average

* Rated on a scale of 0 to 10, for which 0 is not important at all and 10 is extremely important.

> "[It would be good to show] if additional efficacy is still being seen. Postprogression PEI is important if patient continues to benefit in some way."
>
> ~German HTA Advisor

Examples of patient involvement significantly impacting HTA decisions

- Patient involvement and PEI can impact and, in rare instances, even reverse HTA decisions
- Using the patient perspective to evolve inputs into HTA submissions beyond overall survival (OS) gain is one example (**Figure 5**)

Figure 5. Oncology example of patient-relevant endpoints impacting HTA decisions in Sweden

Identify and include endpoints that are meaningful to patients

Decision criteria in Sweden have evolved beyond OS gain for innovative medicines to increase patient relevance

Bringing in the patient voice enabled a new focus on endpoints, such as "deepening of response" and "minimum residual disease" as additional critical outcome measures

→

Including the patient perspective as part of the HTA evaluation

Driven by patient input on endpoints, with robust economic modeling, TLV has changed their HTA decisions in 5 cases (2018-2020)

- Acute lymphocytic leukemia
- Breast cancer
- Non-small cell lung cancer
- Lymphoma
- CAR-T

CAR, chimeric antigen receptor.

Manufacturers can play a critical role in bringing the patient perspective into the HTA process

- Manufacturers can bring in the patient voice during the HTA appraisal process (eg, by showing the patient journey and burden)
- The holistic view on patients' experiences should start even before the clinical trial design and continue throughout the drug life cycle (**Figure 6**)

Figure 6. Patient input should start early in clinical development and continue throughout the HTA process

Incorporate PEI and PRO as part of clinical trials → Understand how patient experience can complement clinical data (eg, deep response, minimum residual disease). Identify data gaps and ways to address those → Prioritize funding for validating economic models and defining the patient journey → Translate into cost/QALY, budget impact, or comparative effectiveness, depending on country

> "Target patient experience as part of the trial. Has to be tied to the impact of the drug."
>
> ~French HTA Advisor

> "With IO therapies, there is little patient experience information. A competitive opportunity exists in certain therapy areas because clinical data is similar and then the patient perspective and PEI can differentiate among treatment options."
>
> ~Italian HTA Advisor

CONCLUSIONS

- Most HTA processes in European countries do not meet EUPATI guidelines regarding the level of patient involvement
- There are opportunities for PEI to provide differentiation among treatment options and affect HTA decisions, particularly for the treatment of rare cancers
- PEI collected post progression can be useful, especially (but not exclusively) in "cost-effectiveness countries"
- Manufacturers should collect PEI in clinical trials and real-world evidence studies, analyze these data with scientific rigor, and translate this evidence into country-specific outcomes (eg, cost/QALYs or budget impact) to maximize the contribution of the patient voice to HTA decisions
- The results of this study might be limited by the small sample size of experts participating
- Increased patient involvement and use of PEI will drive better informed HTA decisions

REFERENCES

1. Hunter A, et al. EUPATI guidance for patient involvement in medicines research and development: health technology assessment. *Front Med (Lausanne)*. 2018;5:231.
2. National Institute for Health and Care Excellence. Accessed October 8, 2020. https://www.nice.org.uk/guidance/ta639/
3. Haite Autorité de Santé. Accessed October 8, 2020. https://www.has-sante.fr/upload/docs/application/pdf/2020-04/analyse_descriptive_contributions.pdf

ACKNOWLEDGMENTS

Kenneth W. K. Wu and Angelo Ramessar developed the graphics for this poster.

Correspondence: **Andy P Brogan, andy@aesara.com**

DISCLOSURES

Study sponsored by Merck KGaA, Darmstadt, Germany, as part of an alliance between Merck KGaA, Darmstadt, Germany, and GlaxoSmithKline.

APB and **CP** report employment at AESARA. **NS** reports employment at Merck KGaA, Darmstadt, Germany. **SB** reports employment at EMD Serono Research and Development Institute, Inc., Billerica, MA, USA; an affiliate of Merck KGaA, Darmstadt, Germany

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ISPOR EU and the author of this poster.

For questions, please contact **andy@aesara.com**.

GET POSTER PDF

Abstract No. 105741. Presented at the Virtual ISPOR Europe 2020 Meeting, 16-19 November 2020