CONTRASTING AVAILABLE THERAPY FOR PATIENTS WITH LIMITED OPTIONS DUE TO EXTREMELY DRUG-RESISTANCE BACTERIA WITH A NOVEL OPTION FOR COMPLICATED NOSOCOMIAL INFECTIONS

Serrano CE[1], Valdez-Vázquez RR[2], Sánchez KX[3]

[1]HEOR Coordinator, Pfizer Mexico, Mexico City, Mexico; [2]Antibiotic Medical Lead, Pfizer Mexico, Mexico City, Mexico; [3]Former full-time employee of Pfizer when the model presented in this work was first developed.

For more information please contact:
Cintia Serrano
Pfizer Mexico
Mexico City, Mexico
Phone: 55-1137-0518
cintiaestefania.serrano@pfizer.com
www.pfizer.com

BACKGROUND

- Antimicrobial resistance (AMR) is a worldwide imminent risk that is taking 200,000 lives every year because of multidrug resistance (MDR) and extensively drug resistance (XDR) infections[1].
- That risk will be the cause of 10 millions of deaths attributable to AMR by 2050, causing more deaths than all types of cancer by that year[1].
- World Health Organization has defined a list of the most dangerous pathogens for human health and called for innovation and development as priority. The three most dangerous pathogens in this list are: carbapenem-resistant *Enterobacteriaceae (CRE)*, *Pseudomonas aeruginosa* and *Acinetobacter baumannii*[2].
- In Mexico, there is an increasing resistance rate to currently available antimicrobials among isolated pathogens, same pathogens that cause numerous nosocomial infections and expose an unmet need for patients[3].
- The INFORM[4] surveillance program, monitoring the in-vitro activity of numerous antimicrobial agents against *Enterobacteriaceae* and *Pseudomonas aeruginosa* isolates worldwide, reports:
 - Antimicrobial susceptibility of the most common types of infection stratified by pathogen
 - Resistance and susceptibility rates of each pathogen by its MDR, XDR and CRE severity groups based on Clinical Laboratory Standards Institute (CLSI) and European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoints.
- A retrospective study (INVIFAR 2019[5]) driven on 47 Mexican centers collected 22,943 isolations from Gram-negatives and Gram-positives pathogens aiming to settle the state of AMR in Mexican hospitals and finding MDR, XDR and possible pandrug-resistance (PDR) in *Acinetobacter sp.*, *Escherichia coli*, *Klebsiella sp.*, *Enterobacter sp.*, and *Pseudomonas aeruginosa*.
- RHOVE[3], a data base collecting data regarding antimicrobial susceptibility in nosocomial infections in Mexican public health institutions, shows an incremental rate of AMR to currently available therapies.

OBJECTIVE

Appraise the economic value of introducing a novel antimicrobial in the Mexican Public Healthcare System for hospitalized patients with limited treatment options due to infections caused by aerobic Gram-negative organisms resistant to available therapies.

METHODS

- A cost-effectiveness model developed for hospitalized adults with complicated nosocomial infections entering a last line of treatment due to failure to all previous antimicrobial therapies.
- Infection is controlled and intended to cure through:
 - (a) Ceftazidime-Avibactam
 - (b) Colistin + Carbapenem
- Ceftazidime-Avibactam, an extended-spectrum cephalosporin along with a novel non-β-lactam β-lactamase inhibitor, is compared with a combined therapy to assess effectiveness measured as[6]:
 - (a) Proportion of cured patients
 - (b) Proportion of alive patients
- Deterministic and probabilistic sensitivity analyses were conducted to evaluate the robustness of clinical and economic outcomes and to evaluate the impact of uncertainty within the model[6].
- Time horizon considers full treatment of antimicrobial, since it considers a period shorter than a year, no discount rate were used[6].
- The study was conducted considering a Mexican public healthcare system perspective, thus only direct costs were incorporated.

Evidence of Efficacy and Safety

- Observational studies have been conducted on this population evaluating clinical cure and mortality of new and current therapies. Reporting higher proportion of patients cured, less adverse events and lower mortality rates with novel alternatives, novel therapies has proven to be feasible alternatives in the treatment of complicated infections[6].
- Shields (2017)[6] observational study compares Ceftazidime-Avibactam with combined therapies to evaluate clinical success (defined as patients with no infection) and overall survival in patients with carbapenem-resistant *Klebsiella pneumoniae* bacteremia.
- Carbapenem-resistant *Enterobacteriaceae* is a subgroup of resistance severity that sometimes belongs to MDR and sometimes to XDR.

Costs

- The analysis was performed with a public institutional perspective as payer, considering sector specific resource use and costs.
- Costs for comparators were obtained from Mexican public health system public sources considering an average length of treatment[7].
- Proportion of patients wit acute kidney injury (AKI) at end of treatment was considered the side effect of antimicrobial treatments for complicated infections and the cost was estimated through a Diagnosis Related Groups methodology based on a publication in 2014 by the main public healthcare institution in Mexico (IMSS)[8] and updated through the official accumulated inflation rate.
- The model structure considers different heath states based on published evidence by Shields (2017)[6], Figure 1.

Figure 1. Cost-Effectiveness Model

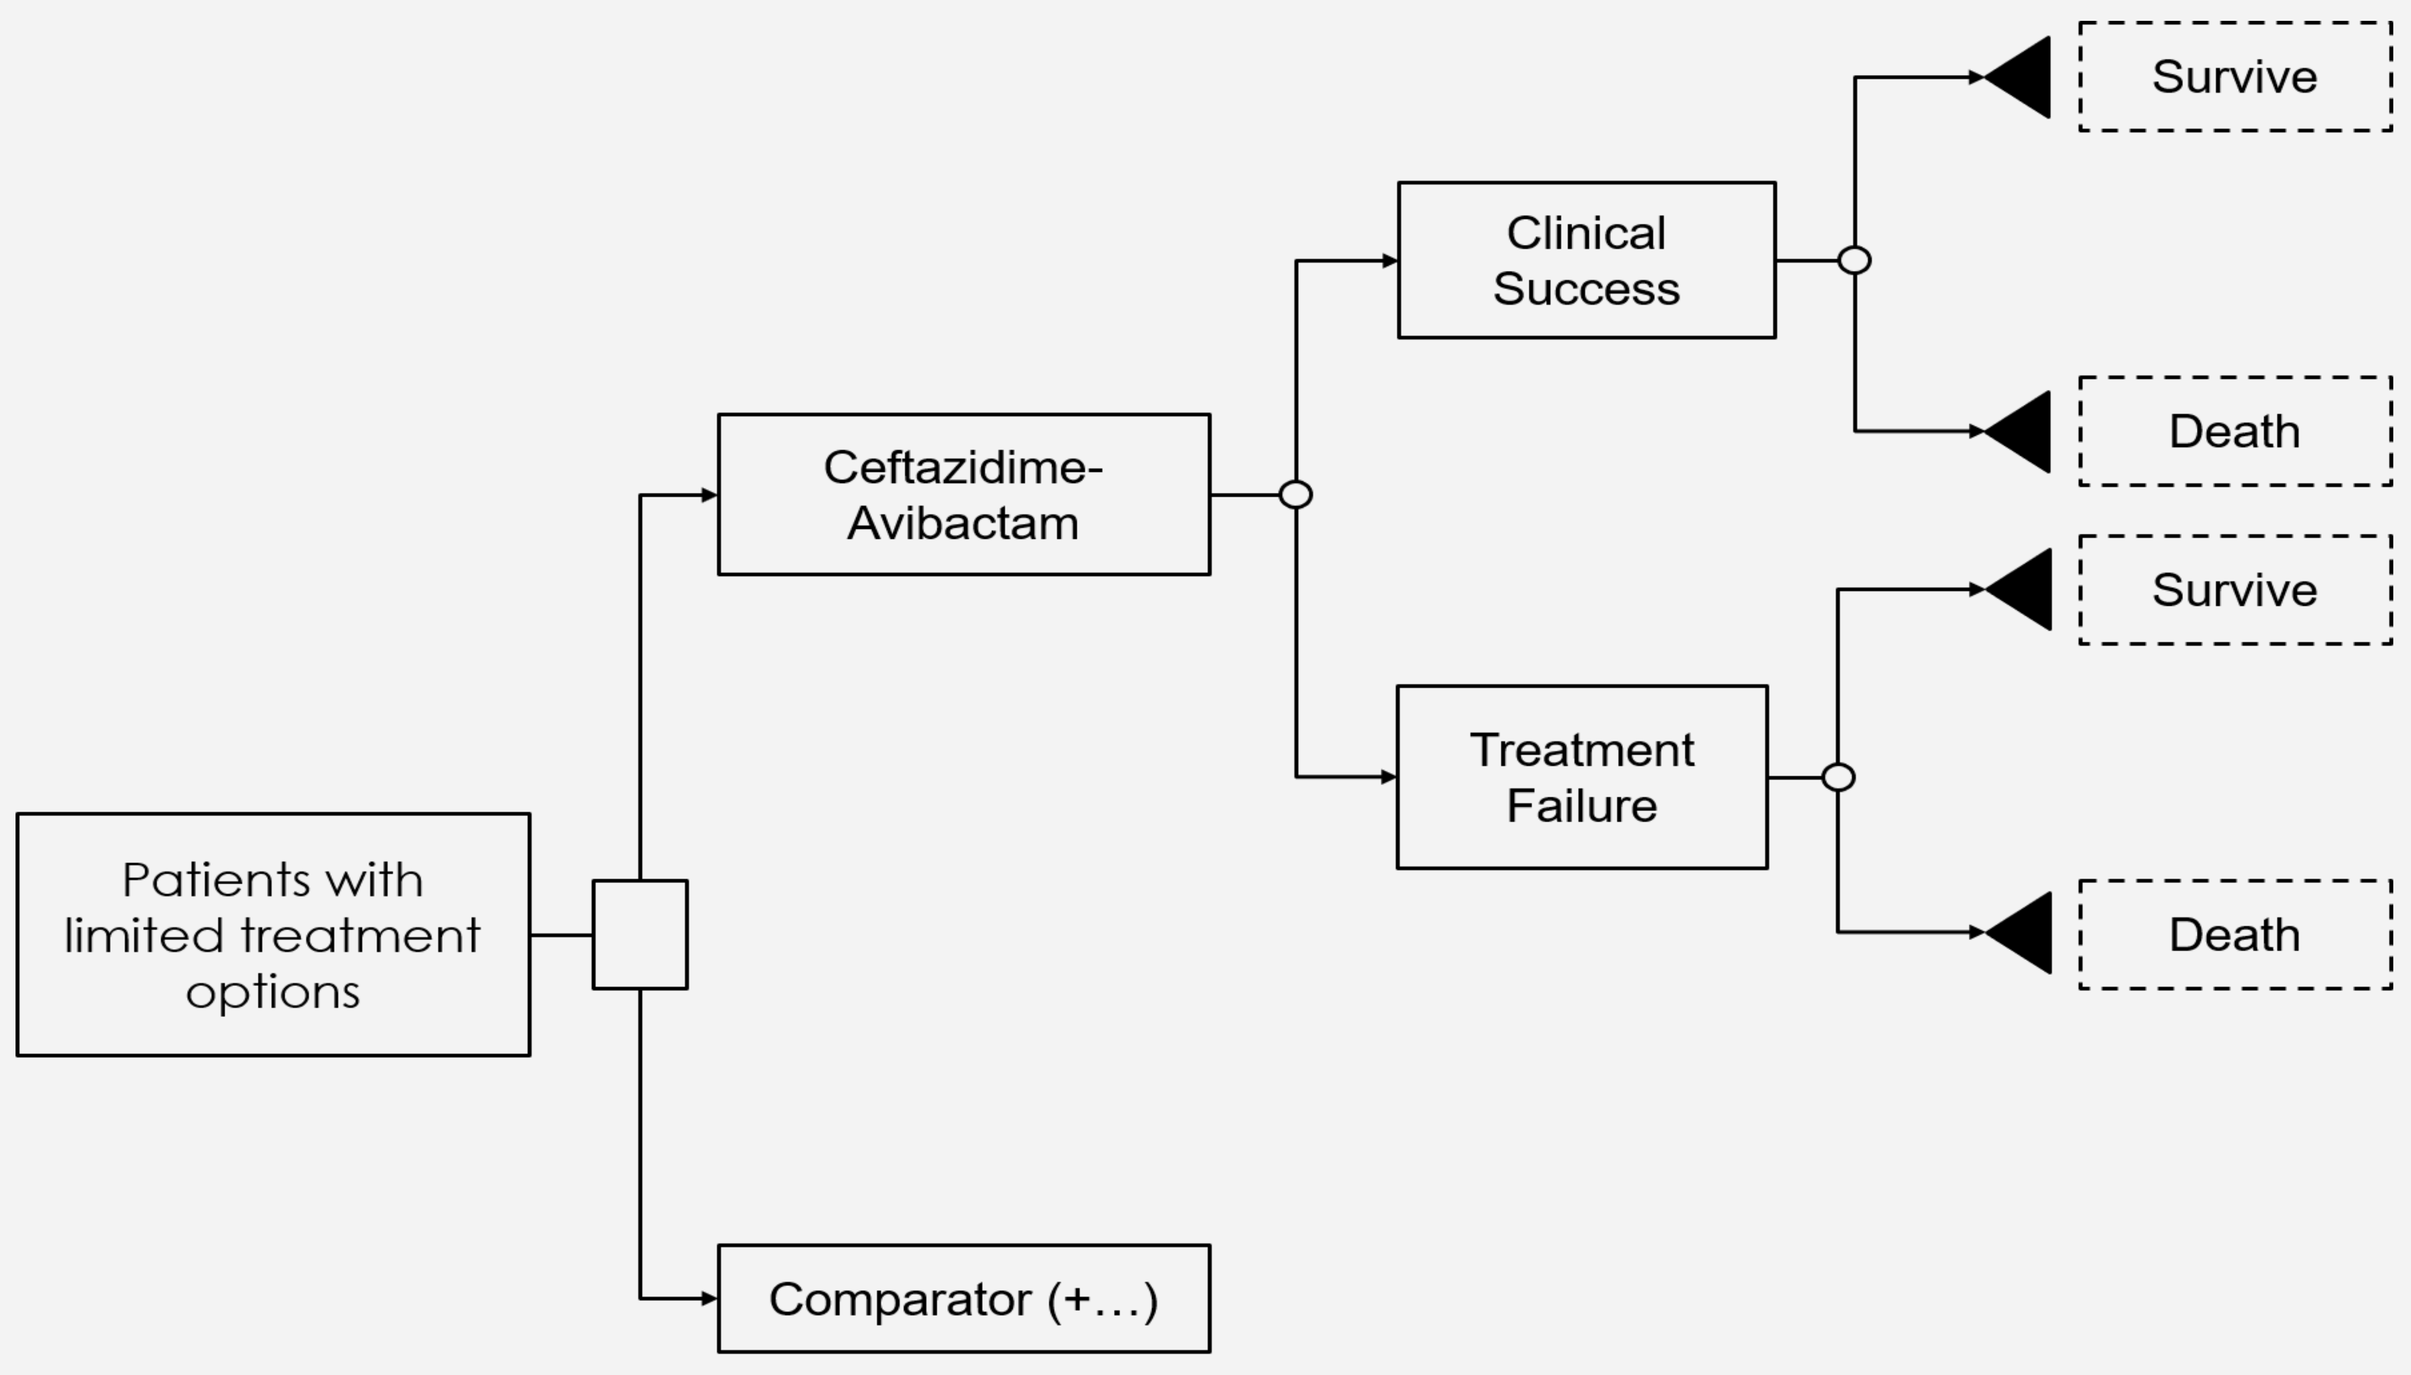

Patients with limited treatment options
Ceftazidime-Avibactam
Comparator (+…)
Clinical Success
Treatment Failure
Survive
Death
Survive
Death

This work was funded by Pfizer Mexico.

RESULTS

Effectiveness Result: Clinical Success

- Ceftazidime-Avibactam offers a higher proportion of patients achieving clinical success[6] of 85% versus 40% of Colistin + Carbapenem, Table 1.
- Ceftazidime-Avibactam offers a lower cost per patient with no infection in comparison to Colistin + Carbapenem: USD 4,542 versus USD 13,596, respectively (Table 1).
- Ceftazidime-Avibactam is a dominant alternative in comparison to Colistin + Carbapenem in the treatment of complicated infections in patients with limited options due to the raising AMR rates present in Mexico.

Table 1. Effectiveness Results measured in Clinical Success

Intervention	Cost	Incremental Cost	Effectiveness	Incremental Effectiveness	ICER	C/E
Colistin +Carbapenem	USD 5,439		40%			USD 13,596
Ceftazidime-Avibactam	USD 3,861	-USD 1,578	85%	45%	Dominant	USD 4,542

Effectiveness Result: 90-Day Survival

- Ceftazidime-Avibactam is a dominant alternative in 90-day survival offering a higher proportion of patients alive[6] at end of treatment of 92% versus *55%* with Colistin + Carbapenem, Table 2.
- Furthermore, Ceftazidime-Avibactam results in a lower cost per patient alive compared with Colistin + Carbapenem: USD 4,196 versus USD 9,888 (Table 2).

Table 2. Effectiveness Result measured in 90-day survival

Intervention	Cost	Incremental Cost	Effectiveness	Incremental Effectiveness	ICER	C/E
Colistin +Carbapenem	USD 5,439		55%			USD 9,888
Ceftazidime-Avibactam	USD 3,861	-USD 1,578	92%	37%	Dominant	USD 4,196

SENSITIVITY ANLYSIS

- A probabilistic sensitivity analysis was driven to assess the impact of uncertainty on model results around cost-effectiveness of Ceftazidime-Avibactam versus Colistin + Carbapenem regarding clinical success.
- Ceftazidime-Avibactam is dominant with a probability of 70%, cost-effective and no cost-effective with a probability of 18% and 12%, respectively.

Figure 2. Cost-Effectiveness Chart on Clinical Success

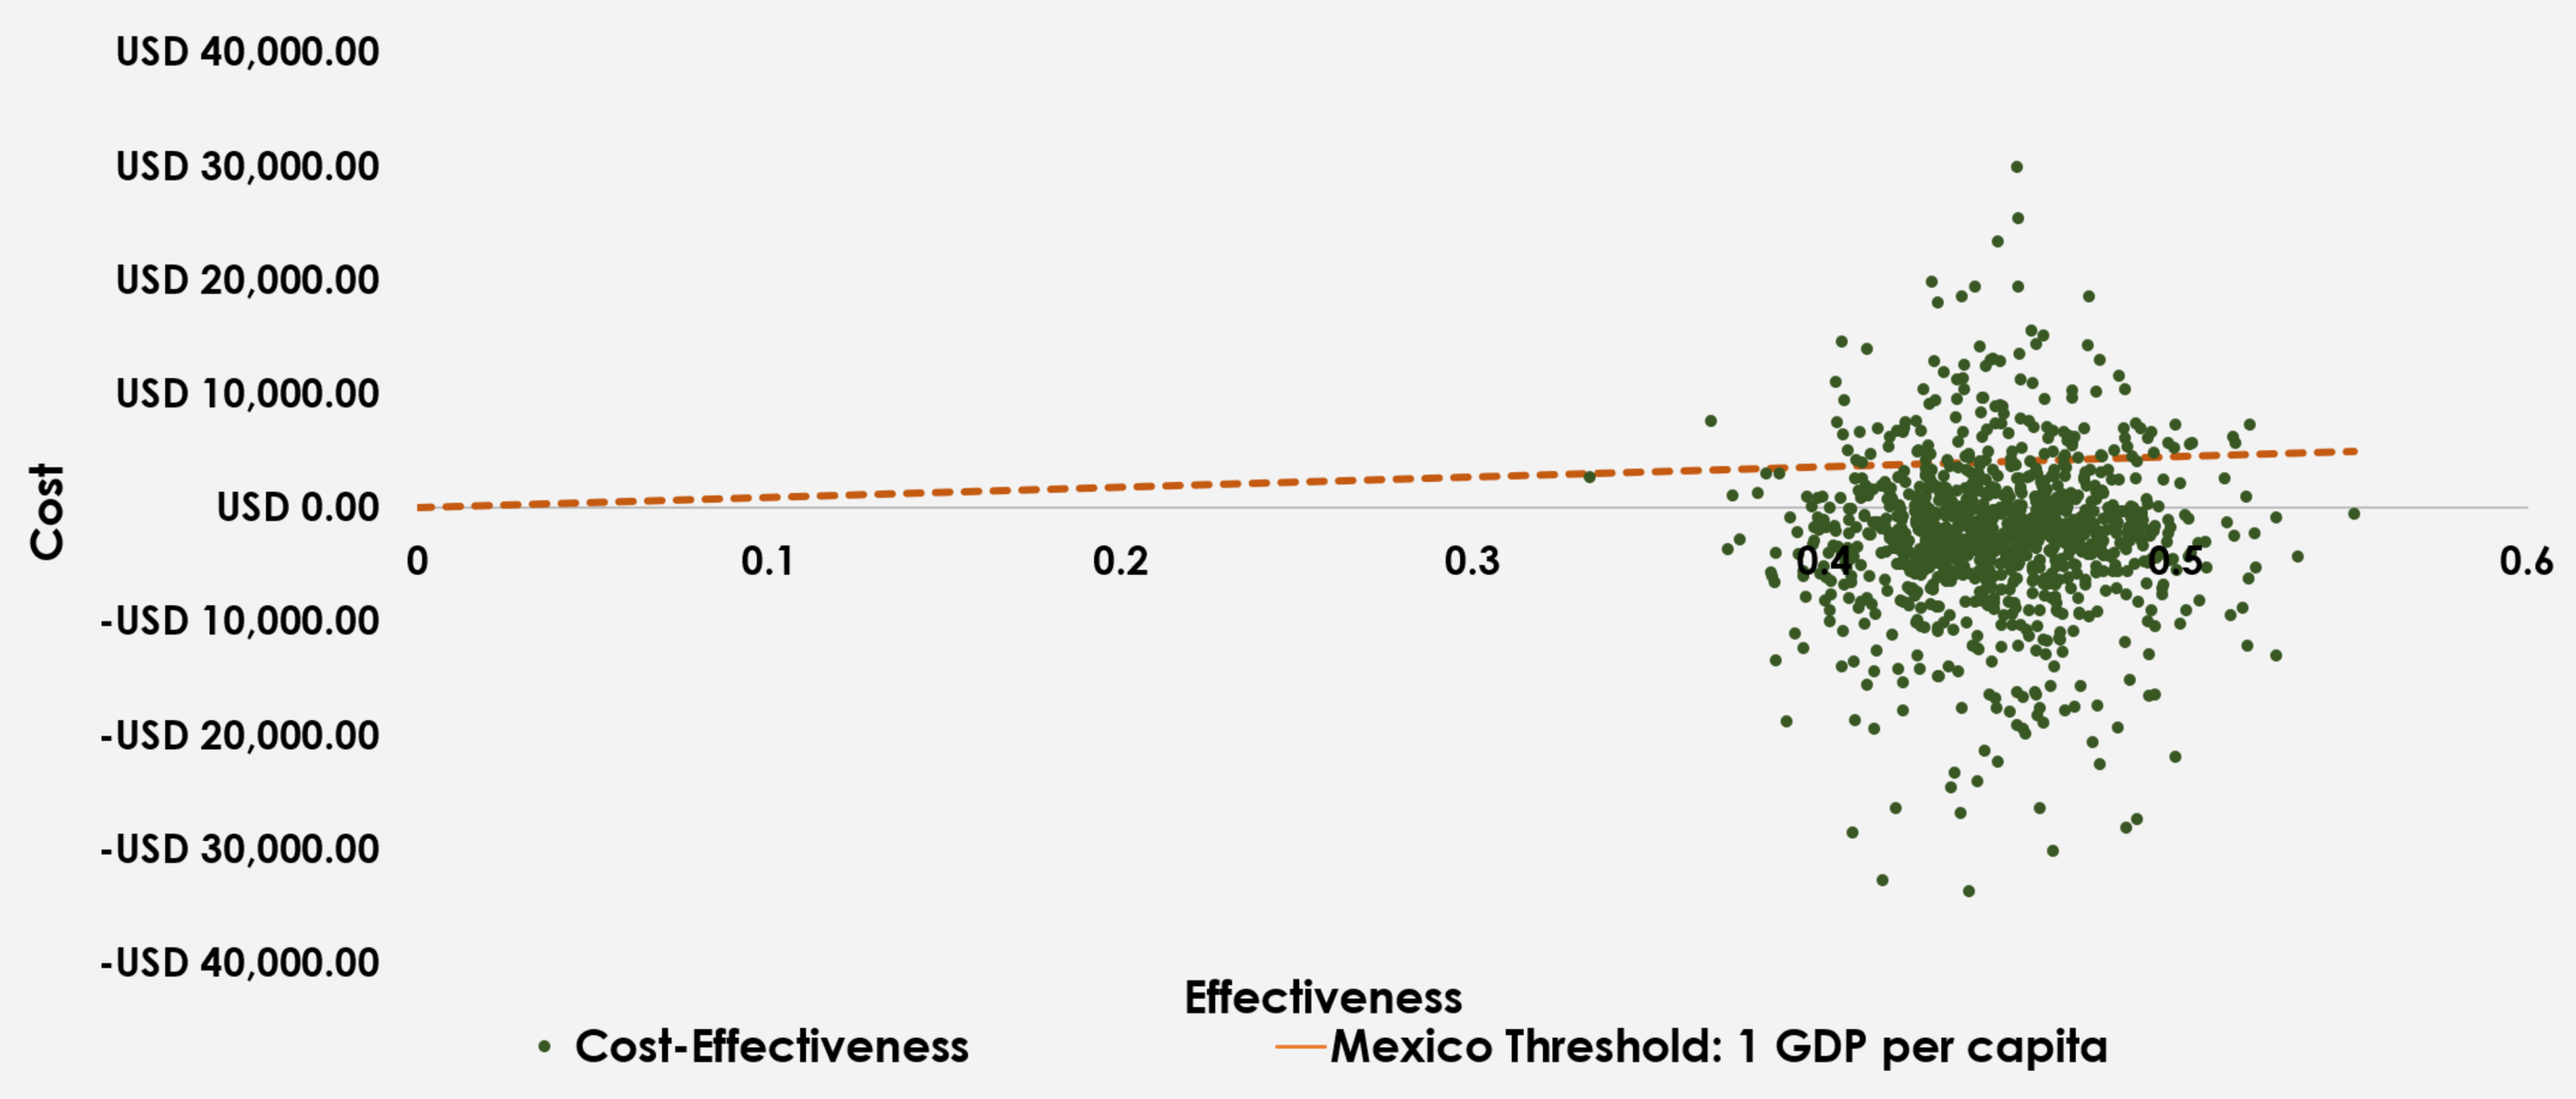

USD 40,000.00
USD 30,000.00
USD 20,000.00
USD 10,000.00
USD 0.00
-USD 10,000.00
-USD 20,000.00
-USD 30,000.00
-USD 40,000.00
Cost
0
0.1
0.2
0.3
0.4
0.5
0.6
Effectiveness
Cost-Effectiveness
Mexico Threshold: 1 GDP per capita

CONCLUSION

- **Ceftazidime-Avibactam is a dominant alternative in the treatment of complicated infections caused by multidrug-resistant pathogens in patients with limited treatment options due to estimated higher clinical success and survival rates.**
- **Given the 700k deaths occurring today per year due to AMR, the 200k lives in risk because MDR and XDR and the estimate of 10 million lives at risk per year by 2050 due to the increasing drug-resistant infections, WHO[9] and Mexican[10] government have released an action plan against AMR through strategic objectives that needs to be taken into practice.**
- **Ceftazidime-Avibactam represents a feasible alternative for Mexican public healthcare system in the fight against AMR and an additional option to the current available therapies aiming to complement the range of antimicrobial options.**
- **The present model has proven robustness due to probabilistic sensitivity analysis results.**
- **All efforts aiming the adequate use of antimicrobials in Mexico and over the world are needed and claimed since the increasing rates of AMR reduces the opportunity to adequately treat patients with nosocomial infections.**

REFERENCES

1 O'Neill (2016). Tackling Drug-Resistant Infections Globally: Final Report and Recommendations. The Review on Antimicrobial Resistance, 1-84.
2 WHO (2017). List of bacteria for which new antibiotics are urgently needed. Press Center, Ginebra. Web page: https://www.who.int/es/news-room/detail/27-02-2017-who-publishes-list-of-bacteria-for-which-new-antibiotics-are-urgently-needed
3 General Directorate of Epidemiology (2015). Epidemiological Surveillance Hospital Network. RHOVE. Health Secretary, First Edition.
4 Sader HS, Castanheira M, Duncan LR, Flamm RK (2018). Antimicrobial Susceptibility of Enterobacteriaceae and Pseudomonas aeruginosa Isolates from United States Medical Centers Stratified by Infection Type: Results from the International Network for Optimal Resistance Monitoring (INFORM) Surveillance Program, 2015–2016. Diagnostic Microbiology and Infectious Disease 92 (2018) 69–74.
5 Garza-González E, Morfín-Otero R, Mendoza-Olazarán S, Bocanegra-Ibarias P, Flores-Treviño S, Rodríguez-Noriega E, et al. (2019) A snapshot of antimicrobial resistance in Mexico. Results from 47 centers from 20 states during a six-month period. INVIFAR. PLoS ONE 14(3): e0209865.
Magiorakos et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. Clin Microbiol Infect 2012; 18: 268–281.
6 Shields, R., Nguyen, M., Chen, L., Press, E., Potoski, B., Marini, R., y otros. (2017). Ceftazidime-Avibactam Is Superior to Other Treatment Regimens againsy Carbapenem-Resistant *Klebsiella pneumoniae* Bacteremia. Antimicrobial Agents and Chemotherapy, 61(8), 1-7.
7 IMSS (2019). National tender cost data base. Mexico. Web page: http://compras.imss.gob.mx/?P=search_alt
8 IMSS, Dirección de Prestaciones Médicas(2014). Diagnosis Related Groups, 1-443.
9 WHO (2016). World Action Plan on Antimicrobial Resistance. WHO Document Production Services, 1-30.
10 General Health Council (2018). Agreement declaring the obligatory nature of the National Action Strategy Against Antimicrobial Resistance. Federal Official Journal 1-20.

Presented at ISPOR Europe 2019; Nov 4-6; Copenhagen, Denmark