COST-UTILITY ANALYSIS OF ABIRATERONE ACETATE PLUS PREDNISONE VERSUS ENZALUTAMIDE IN THE TREATMENT OF METASTATIC CASTRATION-RESISTANT PROSTATE CANCER AFTER FAILURE OF ANDROGEN DEPRIVATION THERAPY

Miguel LS[1], Paquete AT[1], Borges M[1,2]

[1] Centro de Estudos de Medicina Baseada na Evidência, Faculdade de Medicina, Universidade de Lisboa. Lisboa, Portugal. [2] Laboratório de Farmacologia Clínica e Terapêutica, Faculdade de Medicina, Universidade de Lisboa. Lisboa, Portugal.

ID: PDG88

BACKGROUND

- Prostate cancer is the most prevalent oncology disease among men.[1] In metastatic disease, androgen deprivation therapy is the first therapeutic approach. In recent years, some innovative options have been introduced for patients who develop resistance to hormonal treatment, asymptomatic or mildly asymptomatic, non-eligible for chemotherapy.
- This study aims to assess the cost-utility of abiraterone acetate plus prednisone versus enzalutamide in the treatment of metastatic castration-resistant prostate cancer (mCRPC) in asymptomatic or mildly symptomatic patients, after failure of androgen deprivation therapy (ADT) in Portugal, according to the payers' perspective.

METHODS

ECONOMIC MODEL

- The model, developed by Evidera Inc, is based on survival functions that simulate individual progression, by estimating the time each patient remains in each health state. Health states are defined by sequential therapeutic regimens, intercalated by periods free from active treatment or in best supportive care (BSC) – Figure 1. In every health state, patients may progress to death or progress to the following health state.
- Life years (LY), quality-adjusted life years (QALY) and costs were estimated assuming a lifetime horizon and 5% discount rate.

Figure 1. Model structure.

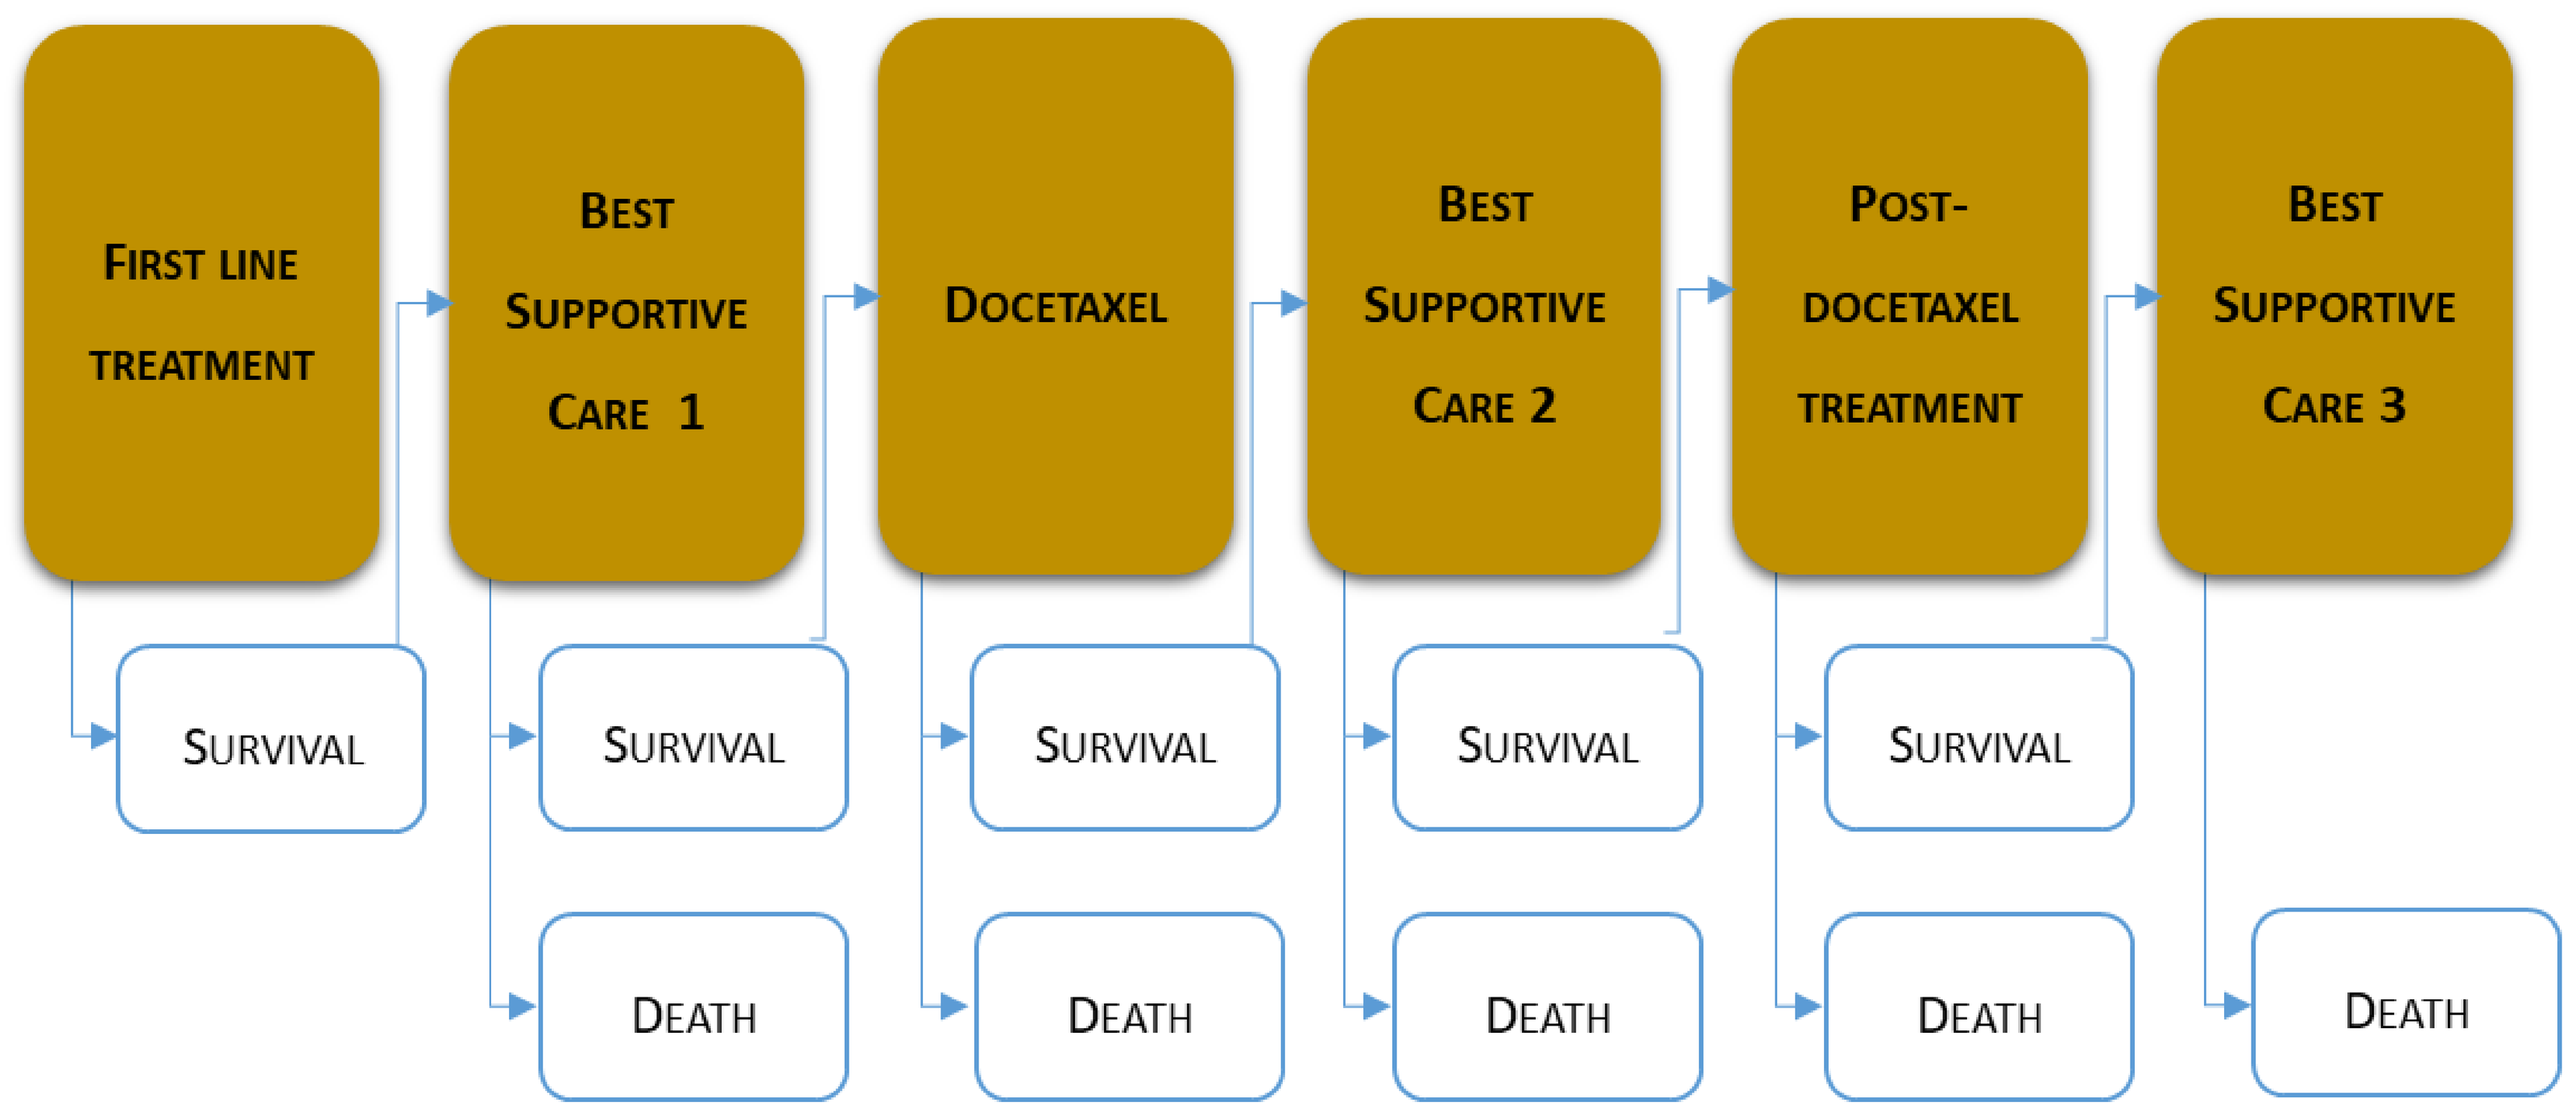
First line treatment
Best Supportive Care 1
Docetaxel
Best Supportive Care 2
Post-docetaxel treatment
Best Supportive Care 3
Survival
Survival
Survival
Survival
Survival
Death
Death
Death
Death
Death

CLINICAL DATA

- Survival functions were based on the reference clinical trial of abiraterone acetate plus prednisone in the treatment of mCRPC after failure of ADT [1,2], depending on patients profile. For patients treated with enzalutamide, time on treatment was adjusted by the treatment coefficient of the survival function, based in the differences of median time on treatment from the reference clinical trials (13.8 months in abiraterone[2] and 16.6 months in enzalutamide[3]).

UTILITIES

- Quality of life weights were obtained from a study developed by Oxford Outcomes[4]. Added utility from using abiraterone, estimated in the reference clinical trial[1,2], was applied to both alternatives in the model. Utility of terminal phase of the disease was based on Sandblom *et al*. study[5].

COSTS

- Resource consumption was based on an expert panel with six specialists in mCRPC in several institutions and geographical areas, highly representative of the Portuguese National Health Service (NHS).
- Unit costs were based on official sources: Hospital Morbidity database from NHS hospitals for the year 2016; NHS prices per Diagnosis Related Group (Portaria 262/2015, Portaria 254/2018); NHS prices per palliative care (Portaria 354/2017); drug cost database (Infomed) [6]; hospital drug catalog database (SPMS) [7].

- Monthly costs and utilities per health state are presented on Table 1.

Table 1. Monthly costs and utilities, per health state.

Health State	Monthly cost (€)	Utilities
First line treatment	450	0.851
Best Supportive Care 1	450	0.625
Docetaxel	835	0.692
Best Supportive Care 2	1,025	0.700
Post-docetaxel treatment	1,067	0.700
Best Supportive Care 3	1,025	0.500

RESULTS

Base case scenario

- Abiraterone acetate plus prednisone delays disease progression, increasing quality of life and delaying subsequent treatment with docetaxel.
- Globally, abiraterone acetate plus prednisone allows an increase of 0.102 life years and 0.003 QALYs.
- Abiraterone is a less expensive option than enzalutamide, due to less time on treatment (1.78 years *vs*. 2.12 years).

Figure 2. Base case results.

Abiraterone acetate plus prednisone increases LY and QALY

↑ **0.102 LY** (3.303 *vs*. 3.201)

↑ 0.117 LY pre-docetaxel
↓ 0.015 LY during and after docetaxel

↑ **0.003 QALY** (2.355 *vs*. 2.352)

↑ 0.012 QALY pre-docetaxel
↓ 0.010 QALY during and after docetaxel

Abiraterone acetate plus prednisone is associated with decreasing costs

↓ **12,564 Total costs** (88.260 *vs*. 100.824)

Pre-docetaxel

↓ 13,020 € in drug acquisition and administration (56,637€ *vs*. 69,658€)

↑ 181 € in adverse events

↑ 544 € in follow-up care

During and after docetaxel

↓ 16 € in drug acquisition and administration (4,214€ *vs*. 4,230€)

↓ 17€ in adverse events

↓ 176 € in follow-up care

Abiraterone acetate plus prednisone is a dominant option (more effective and less expensive)

Sensitivity analyses

- Probabilistic sensitivity analyses was undertaken in order to evaluate the impact of uncertainty on clinical inputs, costs and utilities. Figure 3 illustrates 10,000 simulations, showing the dispersion of incremental costs and QALYs.
- In 91% of simulations, abiraterone acetate plus prednisone is a less expensive option. In 66% and 50% of simulations, abiraterone increases LY and QALY, respectively.

Figure 3. Cost-effectiveness plane: cost per QALY.

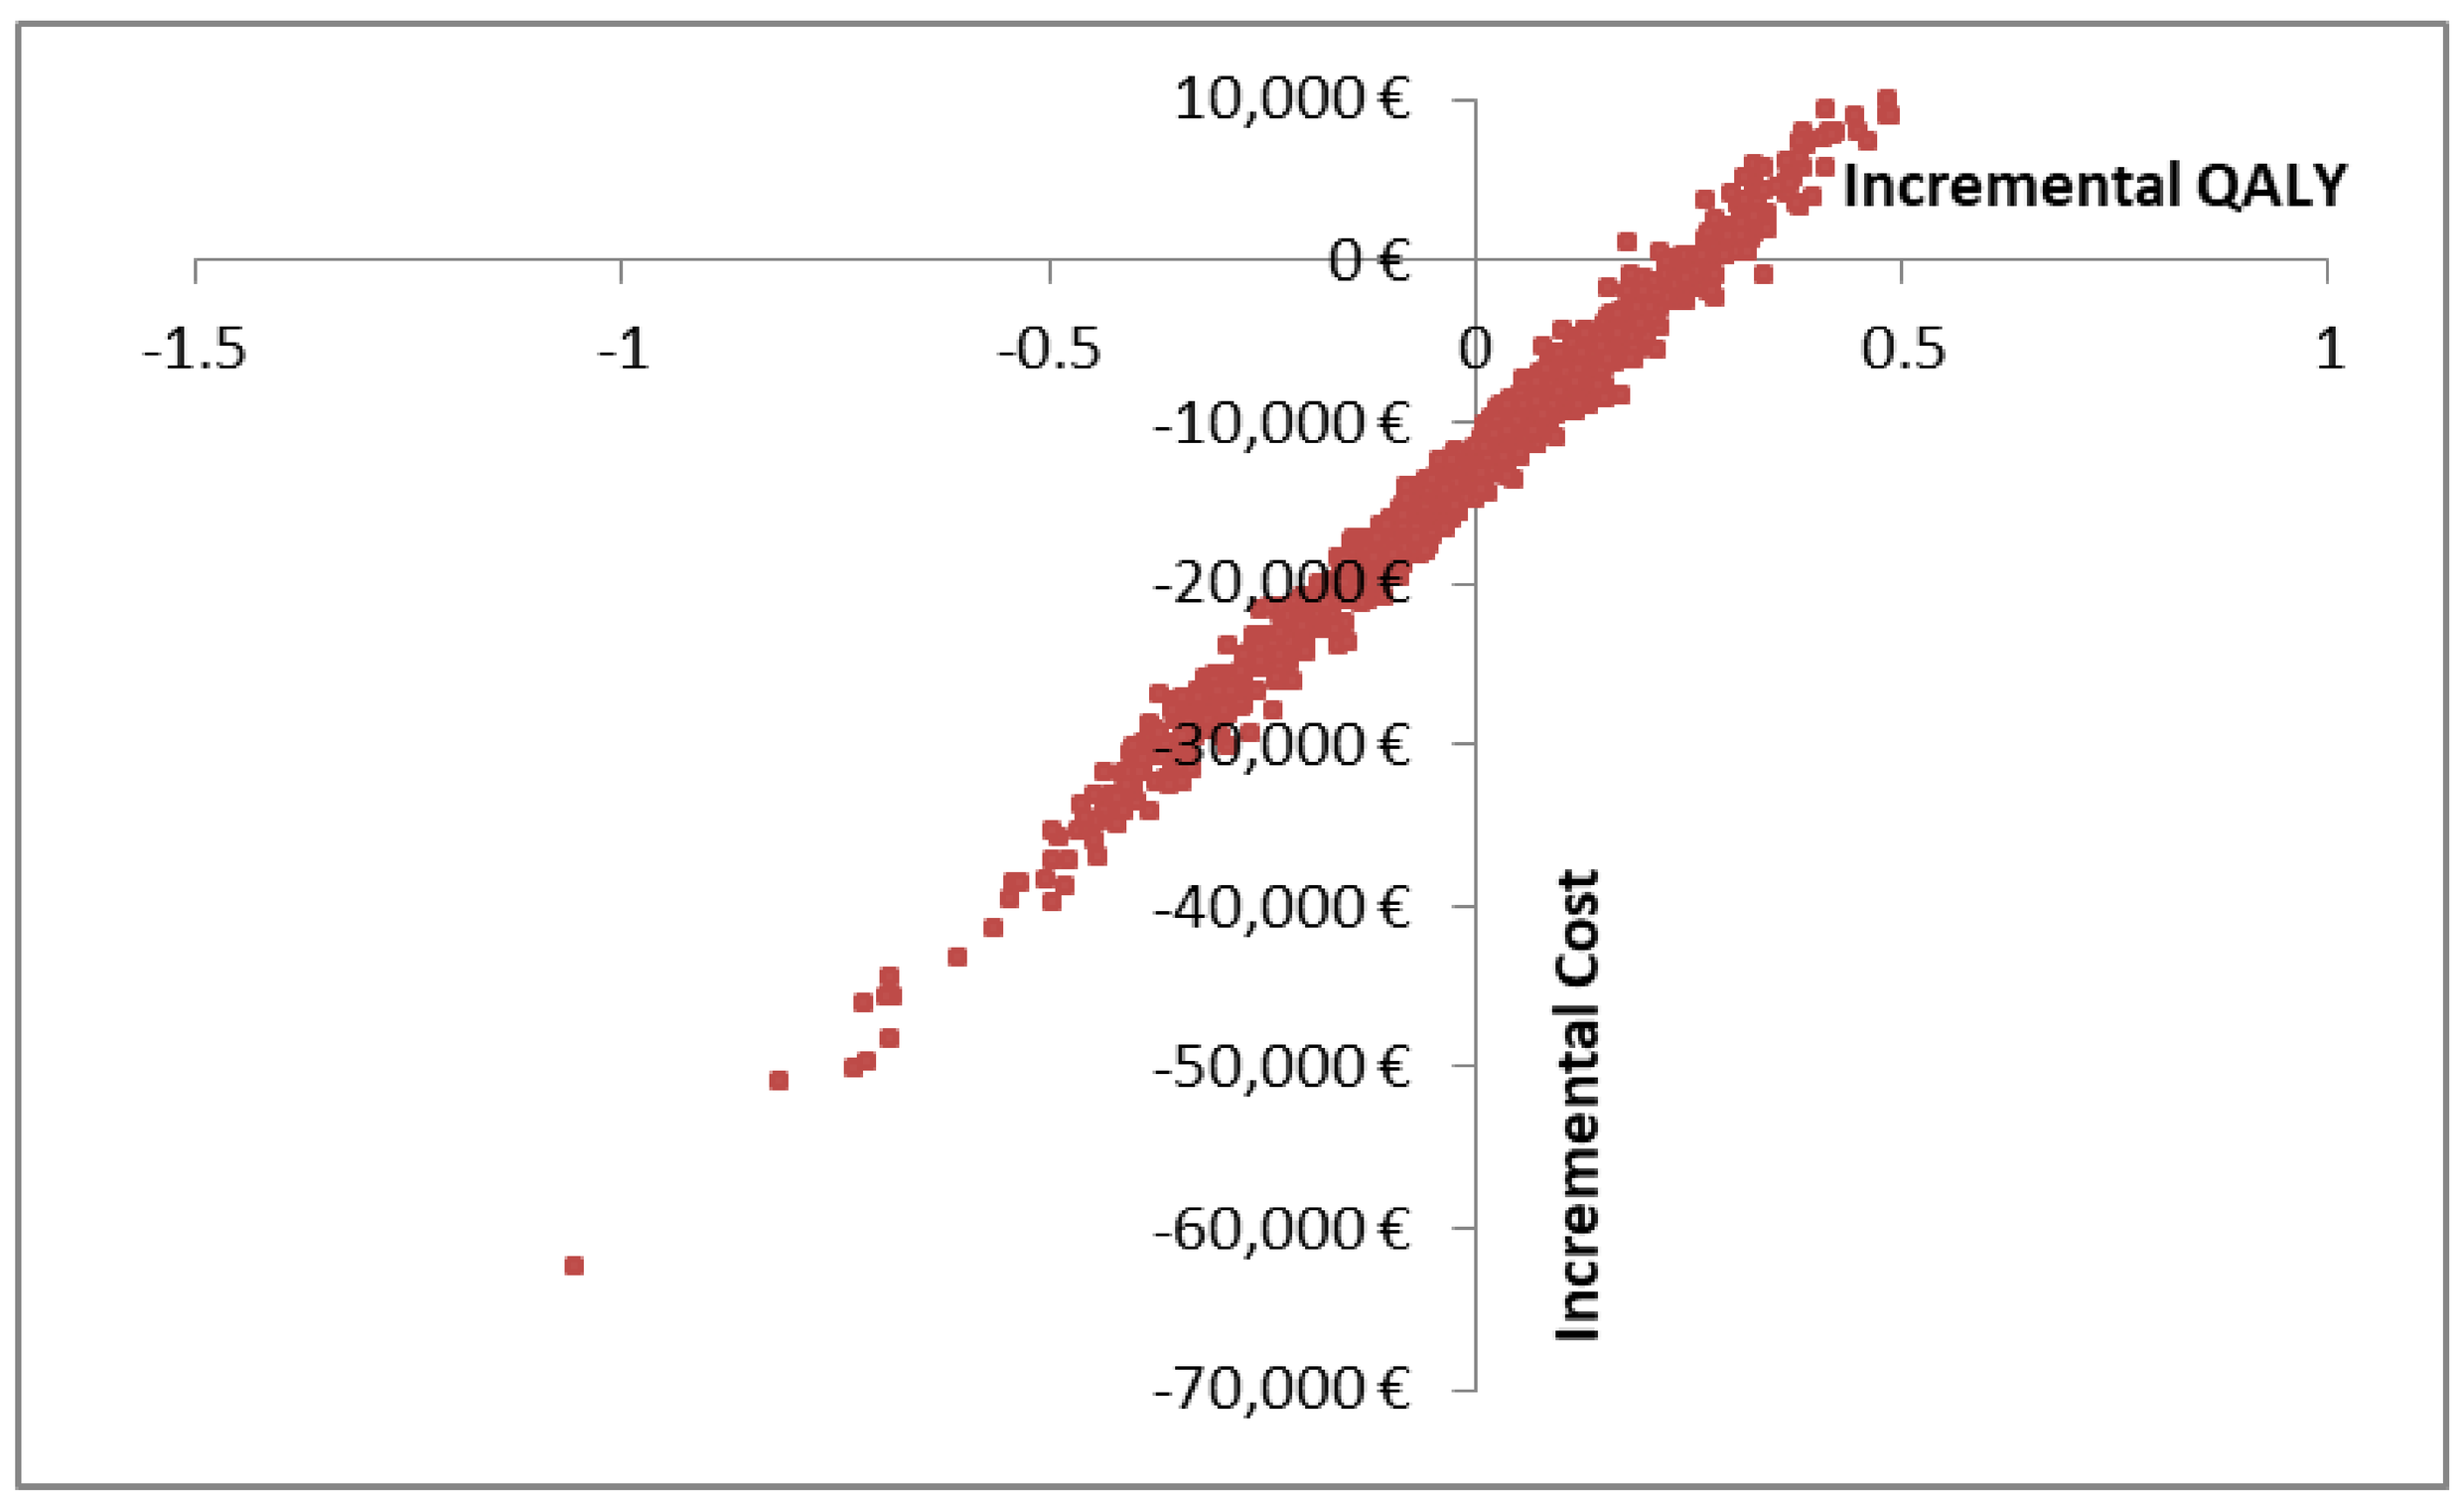
10,000 €
0 €
-10,000 €
-20,000 €
-30,000 €
-40,000 €
-50,000 €
-60,000 €
-70,000 €
-1.5
-1
-0.5
0
0.5
1
Incremental QALY
Incremental Cost

CONCLUSIONS

- Abiraterone acetate plus prednisone in the treatment of metastatic castration-resistant prostate cancer in asymptomatic or mildly symptomatic patients is a less expensive and more effective option than enzalutamide, being considered a dominant alternative in the Portuguese setting.

ACKNOWLEDGEMENTS

- This study was sponsored by Janssen-Cilag Farmacêutica, Lda. Funding was independent of the study outcomes.

REFERENCES

[1] Ryan CJ, Smith MR, de Bono JS e col. (2013) Abiraterone in Metastatic Prostate Cancer without Previous Chemotherapy. N Engl J Med 368:138-48. [2] Ryan CJ, Smith MR, Fizazi K e col. (2015) Abiraterone acetate plus prednisone versus placebo plus prednisone in chemotherapy naive men with metastatic castration-resistant prostate cancer(COU-AA-302): final overall survival analysis of a randomised, double-blind, placebo-controlled phase 3 study. Lancet Oncol 2015; 16: 152–60. [3] Beer, T. M., Armstrong, A. J., Rathkopf, D. E., Loriot, Y., Sternberg, C. N., Higano, C. S., ... & Davis, I. D. (2014). Enzalutamide in metastatic prostate cancer before chemotherapy. New England Journal of Medicine, 371(5), 424-433.[4] Oxford Outcomes (2013) Utility measurement study in metastatic castrate resistant prostate cancer. *Data on file*. [5] Sandblom G, Carlsson P, Sennfalt K, e col. (2004) A population-based study of pain and quality of life during the year before death in men with prostate cancer. *British Journal of Cancer* 90(6):1163-1168. [6] Infomed, base de dados Infarmed. [7] SPMS, Catálogo de Aprovisionamento Público de Saúde (www.catalogo.min-saude.pt).

Corresponding author: luis.silvamiguel@medicina.ulisboa.pt

ISPOR Europe 2019. 2-6 November 2019. Copenhagen, Denmark.