COST-MINIMIZATION OF VENETOCLAX WITH RITUXIMAB VERSUS IBRUTINIB IN R/R CLL IN THE BRAZILIAN PRIVATE HEALTHCARE SYSTEM

PCN243

Clóvis de Aguiar Brito Filho[1], Fernando Meton Vieria[2], Vinícius Vitale[1]

[1]AbbVie Brazil, São Paulo/SP, Brazil; [2]Instituto COI de Educação e Pesquisa, Rio de Janeiro/RJ, Brazil

Presented at the ISPOR Europe, 11/04/2019, Denmark.

INTRODUCTION

- CLL is a lymphproliferative disease caracterized by the persistency of a minimum of 5 × 10^9/L type B monoclonal lymphocytes. These lymphocytes can accumulate in periferal blood, lymph nodes, blood marrow and spleen. More frequent in elders and considered a rare disease, CLL has a global incidence between <1 and 5.5 cases per 100,000 inhabitants. (1,2)
- Althought a rare disease, the CLL is the most common type of of leukemia in adults, being reponsible for about 32% of total leukemias. (3)
- It is an indolent disease, in which about 60% of patients at the time of diagnosis may be assymtomatic, while other 40% may have several inespecific clinical and laboratory changes, such as fatigue, weakness, or more specific symptoms like lymphadenomegaly, hepatomegaly, splenomegaly, cytopenias for autoimmune mechanisms (autoimmune anemia and thrombocytopenia) and constitutional symptoms such as fever, weight loss and night sweats. (4,5)
- The presence of 17p deletion (!7p Del), TP53 mutation and unmutated IGHV indicate worse clinical results with fast disease evolution. The 17p Del varies between 30% to 50% in R/R CLL patients. (6)
- A commonly used regimen for chronic lymphocitic leukemia has been bendamustine in combination with rituximab. Recently, novel targeted oral agents, such as venetoclax and ibrutinib, have provided greater survival with modest toxicity. (7)
- Venetoclax is an orally administered potent and selective BH3 mimetic that targets the BCL-2 (B-Cell Leukemia/Lymphoma 2 Gene) inhibitor, exhibiting significant apoptotic activity. While Ibrutinib is a once-daily, oral covalent inhibitor of Bruton's tyrosine kinase, which inhibits B-cell receptor signaling (7)
- NICE's committee believe venetoclax plus rituximabe to have similar or better efficacy to ibrutinib, and that venetoclax is unlikely to be inferior to ibrutinib. (8)
- According to ANS' rol (mandatory list of drugs and procedures), with regards to R/R CLL, only patients with 17p Del are contemplated with ibrutinib.

OBJECTIVE

Develop a cost-minimization analysis of venetoclax plus rituximab (V+R) *versus* ibrutinib In adult patients with Chronic Lymphocytic Leukemia (R/R CLL) who have received at least one previous therapy, according to the perspective of the Brazilian private healthcare system.

METHODS

- In a systematic review developed by authors, it was not found any direct-comparison between venetoclax and ibrutinib. Once indirect-comparisons have limitations, it was developed a cost-minimization analysis assuming equivalency in therapeutic efficacy between V+R and ibrutinib.
- The cost-minimization analysis has compared in terms of incremental costs the total drugs-related treatment costs in a pre-defined time horizon (24 months).

Figure 1. Treatment scheme: venetoclax + rituximab.

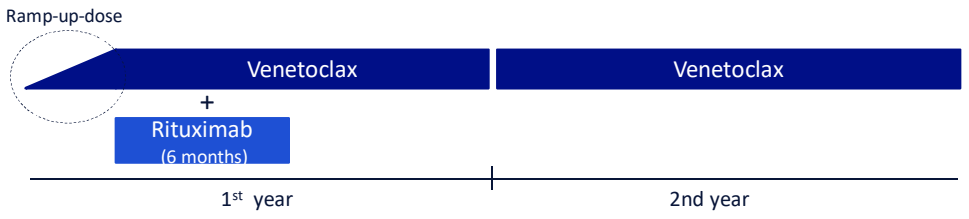
Ramp-up-dose
Venetoclax
Venetoclax
+
Rituximab
(6 months)
1st year
2nd year

Figure 2. Treatment scheme: ibrutinib.

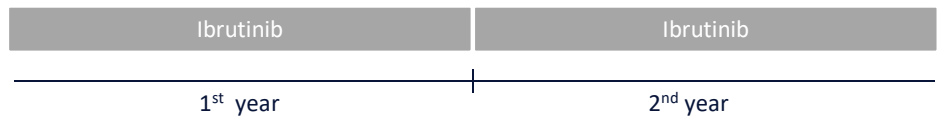
Ibrutinib
Ibrutinib
1st year
2nd year

- The innitial dose of venetoclax is 20mg (02 tablets of 10mg), weekly ramping-up untill reaching 400mg (04 tablets of 100mg) at the begining of week 5. From that point on patient will follow the maintenance 400mg dosage per day until the end of the 24th month.
- Drugs acquisition costs were obtained from the price-reference list, published by the Brazilian Regulatory Agency (ANVISA) considering list prices (PF), with 18% tax (PF 18%) over circulation of merchandise and services (ICMS). The prices considered in this analysis were based in the April 2019 list edition. (9)
- Table 1 shows treatment monthly cost of venetoclax, rituximab and ibrutinib. For the calculation of monthly costs it was considered that one months had 30 days.
- All costs are presented in american dollars (USD). Currency rate used was R$3.92 to USD 1.00. It represents the avarage rate of last 60 days (07/08/2019 to 09/06/2019). (10)

Table 1. Price of drugs used in the analysis.

Drugs	Dosage Forms	List Price (PF18%)
Venetoclax (start-kit)*	(10 mg x 14 tablets – week 1) (50 mg x 7 tablets – week 2) (100 mg x 7 tablets – week 3) 100 mg x 14 tablets – week 4)	$ 1,899.27
Venetoclax (maintenance dose)	100 mg x 120 tablets	$ 8,799.67
Rituximab	10 mg/mL x 10 mL bottle	$ 349.06**
Ibrutinib	140 mg x 90 tablets	$ 9,852.72

** Related to the 1st month of treatment.*
*** PF 0%: product exempt from ICMS.*

- Rituximab dose calculation was based on patients body surface area, defined according to Mosteller formula, based on Brazilian antropometric data. (11)
 - Weight: 67.2 kg
 - Hight: 164.5 cm;
 - Body surface area: 1.75 m².

METHODS (CONTINUED)

Table 2. Monthly treatment cost.

Drugs	Dosage Forms	List Price (PF18%)
Venetoclax	1st month (start-kit)	$ 1,899.27
Venetoclax	Following months (maitenance dosage)	$ 8,799.67
Rituximab	1st Cicle: 375 mg/m² (month 1) 2-6 Cicles: 500 mg/m² (months 2-6)	$ 2,293.46 $ 3,057.94
Ibrutinib	30 days (bottle with 90 tablets)	$ 9,852.72

Table 3. Annual treatment cost.

Drugs	1st year	2nd year
Venetoclax + rituximab	$ 116,278.83	$ 105,596.06
Ibrutinib	$ 118,232.04	$ 118,232.04

- Cost-minimization is exclusivelly based in two parameters, drugs price and dosage, thus the only parameter subject to uncertainty is purchasing price of the drugs analyzed. These prices are uncertain and depend on negotiations between pharmaceutical companies and HMOs, which makes it unfair any comparison based on prices different than those defined by Anvisa's list of price-reference.
- Considering these uncertainties it was developed an univariate sensitivity analysis of the results presented in the monthly treatment costs (Table 4), applying a variation of -10% over 18% PF prices of venetoclax, rituximab and ibrutinib in order to analyze the impact of treatment costs. Superior limit was not applied once PF represents the maximum for drugs price. Results are presented in Table 6.

Table 4. Treatment monthly cost.

Purchasing cost of...	Base Cost	Inferior limit	Superior limit
Ibrutinib	$ 9,852.71	$ 8,867.44	-
Venetoclax (start-kit)	$ 1,899.26	$ 1,709.33	-
Venetoclax (maintenance)	$ 8,799.67	$ 7,919.70	-
Rituximab	$ 349.06	$ 314.15	-

- An alternative scenario of 56 months time-horizon was also considered, according to NICE's appraisal of V+R (8), which assumed that the average treatment duration with ibrutinib by R/R CLL patients was 56 months, and 22 months for V+R. However, it was adopted a more conservative approach in this analysis, considering 25 months of treatment duration for venetoclax + rituximab, according to product label.

RESULTS

Table 5. Treatment yearly costs.

Drugs	1st year of treatment	Incremental	% of savings	2nd year of treatment	Incremental	% of savings
Venetoclax + rituximab	$ 116,278.83	-	0%	$ 105,596.06	-	0%
Ibrutinib	$ 118,232.04	-$ 1,953.81	-2%	$ 118,232.04	-$ 12,636.59	-11%

Figure 3. Yearly treatment costs comparison.

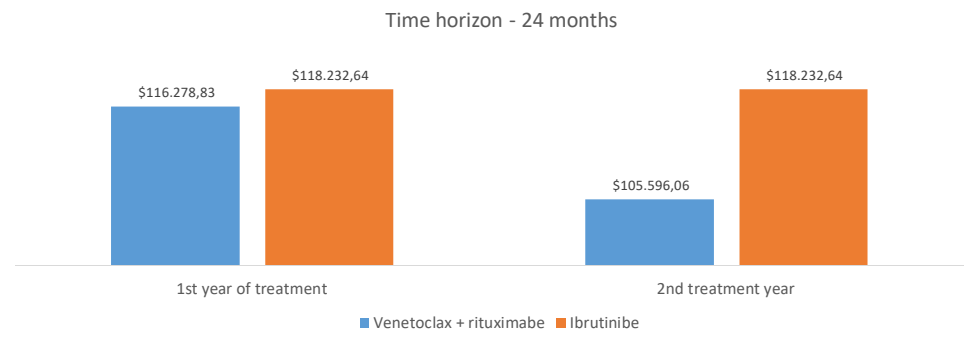
Time horizon - 24 months
$116.278,83
$118.232,64
$105.596,06
$118.232,64
1st year of treatment
2nd treatment year
Venetoclax + rituximabe
Ibrutinibe

- According to treatment costs analyzed V+R demonstrates being the most economic alternative when compared to ibrutinib in R/R CLL patients. The adoption of V+R offers cost savings of about USD 2,000 (2%) per patient in the 1st year, even with the addition of rituximab. In the 2nd year, this adoption allows cost savings of approximately USD 12,500 (11%) per patient.

Table 6. Univariate analysis results (incremental cost of V+R *versus* ibrutinib considering inferior limit of prices).

Default Parameters	1st year	2nd year
Cost of venetoclax (start-kit)	$ -2,143.73	$ -12,636.59
Cost of venetoclax (maintenance)	$ -11,633.45	$ -23,196.19
Cost of rituximab	$ -3,712.13	$ -12,636.59
Cost of ibrutinib	$ 9,869.46	$ -813,32

Figure 4. Univariate sensitivity analysis – Tornado Diagram.

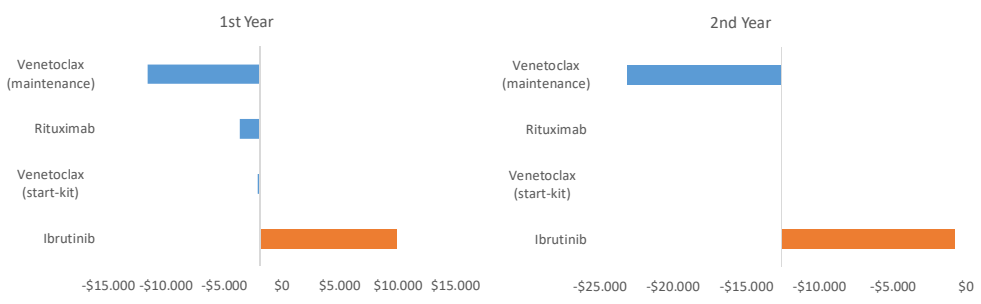
1st Year
Venetoclax (maintenance)
Rituximab
Venetoclax (start-kit)
Ibrutinib
-$15.000 -$10.000 -$5.000 $0 $5.000 $10.000 $15.000
2nd Year
Venetoclax (maintenance)
Rituximab
Venetoclax (start-kit)
Ibrutinib
-$25.000 -$20.000 -$15.000 -$10.000 -$5.000 $0

RESULTS (CONTINUED)

- Results from the univariate sensitivity analysis indicate that if applied a 10% discount in ibrutinib over its PF18% price, the 1st year with V+R show an incremental cost of about $9,950. This results is directly related to the addition of rituximab. However, even applying this discount, the following years show savings of around $800, in favor of V+R. Discounts around 10% to the other parameters do not have significantly influentiated the analysis results. Therefore, independently of the discounts applied to the other drugs (venetoclax and rituximab), the treatment with V+R remains with the lowest cost in comparison to ibrutinib.

Table 7. Results of cost-minimization analysis (per patient) – 56 months.

Medicamento	Venetoclax + rituximab	Ibrutinib	Incremental	% of savings
Cost of treatment	$ 230,674	$ 551,752	-$ 321,077	-58%
Time in treatment	25 months (complete duration of treatment)	56 months average duration of treatment)	-	-
Drug	Venetoclax + rituximab	Ibrutinib		

- With duration of treatment of 56 months for ibrutinib and 25 months for V+R, V+R demonstrates resource savings of about $321,077 per patient (58% cost savings). Which means that the cost of treating 10 R/R CLL patients with ibrutinib is the same cost of treating about 24 patients with venetoclax + rituximab.

Figure 5. Results of cost-minimization analysis (per patient) – 56 months.

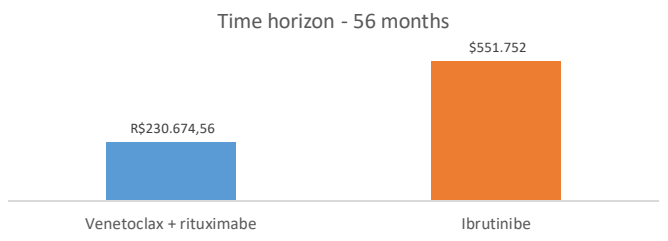
Time horizon - 56 months
R$230.674,56
$551.752
Venetoclax + rituximabe
Ibrutinibe

Figure 6. Accumulated costs (USD) comparison: 1st year.

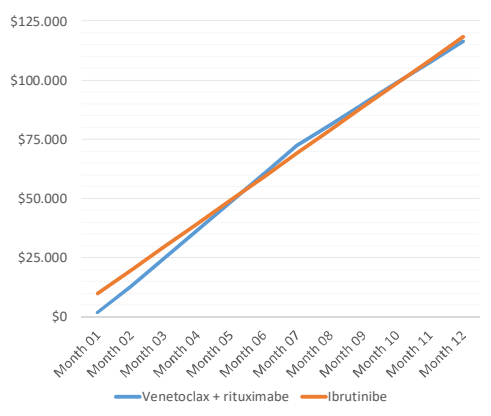
$125.000
$100.000
$75.000
$50.000
$25.000
$0
Month 01
Month 02
Month 03
Month 04
Month 05
Month 06
Month 07
Month 08
Month 09
Month 10
Month 11
Month 12
Venetoclax + rituximabe
Ibrutinibe

Figure 7. Accumulated costs (USD) comparison: from 1st to 5th year.

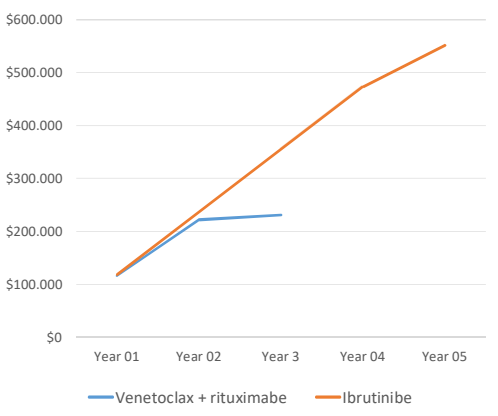
$600.000
$500.000
$400.000
$300.000
$200.000
$100.000
$0
Year 01
Year 02
Year 3
Year 04
Year 05
Venetoclax + rituximabe
Ibrutinibe

CONCLUSIONS

- Economic analysis suggest that combined treatment with V+R brings resource savings to the Brazilian private healthcare system. This results are specially related to its fixed treatment duration, with treatment interruption in 25 months.
- According to the cost-minimization analysis of V+R *versus* ibrutinib, V+R offers $2,000 (2%) per patient in cost savings in the 1st year, and $12,500 (11%) per patient in the 2nd year.
- In 56 months time-horizon cost-minimization analysis, based on NICE's average treatment duration, the adoption of V+R shows $321,077 in resource savings per patient. Therefore, the cost of treating 10 R/R CLL patients with ibrutinib is the same cost of treating about 24 patients with venetoclax + rituximab.

REFERENCES

1. Hallek M. Chronic lymphocytic leukemia: 2017 update on diagnosis, risk stratification, and treatment. Am J Hematol. 2017;92(9):946–65.
2. Scarfò L, Ferreri AJM, Ghia P. Chronic lymphocytic leukaemia. Crit Rev Oncol Hematol. Elsevier Ireland Ltd; 2016 Aug;104:169–82.
3. Siegel R, Naishadham D, Jemal A. Cancer statistics. CA Cancer J Clin 2013; 63: 11–30.
4. Rodrigues CA, Gonçalves MV, Ikoma MRV, Lorand-Metze I, Pereira AD, Farias DLC de, et al. Diagnosis and treatment of chronic lymphocytic leukemia: recommendations from the Brazilian Group of Chronic Lymphocytic Leukemia. Rev Bras Hematol Hemoter. 2016;38(4):346–57.
5. Hallek M, Cheson BD, Catovsky D, Caligaris-cappio F, Dighiero G, Dohner H, et al. Guidelines for the diagnosis and treatment of chronic lymphocytic leukemia: a report from the International Workshop on Chronic Lymphocytic Leukemia updating the National Cancer Institute-Working Group 1996 guidelines. Blood. 2008 Jun 15;111(12):5446–56.
6. Döhner H, Fischer K, Bentz M, Hansen K, Benner A, Cabot G, et al. p53 gene deletion predicts for poor survival and non-response to therapy with purine analogs in chronic B-cell leukemias. Blood. 1995 Mar 15;85(6):1580–9.
7. Po-Huang Chen 1 , Ching-Liang Ho, et al. Treatment Outcomes of Novel Targeted Agents in Relapse/Refractory Chronic Lymphocytic Leukemia: A Systematic Review and Network Meta-Analysis, J. Clin. Med. 2019 May, 8:737.
8. National Institute for Health and Clinical Excellence (NICE). Venetoclax with rituximab for pre previously viously treated chronic lymphocytic leukaemia. Technology appraisal guidance TA561. Geneva: NICE; 2019.
9. Ministério da Saúde (Brasil). Agência Nacional de Vigilância Sanitária (ANVISA). Listas de preços de medicamentos [Internet]. 2018. Available from: http://portal.anvisa.gov.br/listas-de-precos.
10. Ministério da Economia (Brasil). Cotações e boletins [Internet]. 2019. Available from: https://www.bcb.gov.br/
11. Instituto Brasileiro de Geografia e Estatítica (IBGE). Pesquisa de Orçamentos Familiares: 2008-2009. Análise do Consumo Alimentar Pessoal no Brasil. Rio de Janeiro: IBGE; 2011. 150 p.

ACKNOWLEDGEMENTS

- Financial support was provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the poster. All authors contributed to the development of the publication and maintained control over the final content

DISCLOSURES

Vieira, Fernando Meton: is researcher at Instituto COI de Educação e Pesquisa.

Brito Filho, Clóvis de A; Vitale, Vinícius: are employees and stockholders of AbbVie.

Vinicius Vitale coauthored the abstract/poster as a member of the Market Access organization and transferred to the Commercial organization after the content was finalized.

Placeholder for a QR code. Use if applicable.

To view an electronic version of this poster and other AbbVie <Congress> scientific presentations, scan QR code or visit http://abbviescience.com/pdf/<fill in website here>. QR code expiration: <date>.