VIEW ONLINE
http://loremipsum.com/
index.html?l=orem

PCV74

Cost-effectiveness of Rivaroxaban 2.5mg twice daily in combination with Acid acetylsalicylic in treatment of patients with Coronary Artery Disease (CAD) or Symptomatic Peripheral Artery Disease (PAD) in Greece

Kalogeropoulou M[1], Gkouvitsos Y[2], Barlogiannis T[3], Bowrin K[4], Briere J-B[5], Millier A[6], Karabis A[7]

[1] IQVIA, Athens, Greece
[2] IQVIA, Athens, Greece (at the time that the study was conducted)
[3] Bayer AG, Athens, Greece
[4] Bayer Plc, London, United Kingdom
[5] Bayer AG, Berlin, Germany
[6] Creativ-Ceutical, Paris, France
[7] IQVIA, Amsterdam, Netherlands

CONCLUSION

- **The study indicates that, from a Greek payer perspective, rivaroxaban in combination with acid acetylsalicylic is a cost-effective treatment of patients with CAD or symptomatic PAD. Specifically, it found that a lifetime treatment with rivaroxaban 2.5 mg bid in combination with ASA 100 mg once daily (od) compared to ASA 100 mg od alone is associated with an increase in QALYs (0.33) and LYs (0.38) and with an increase in costs (€3,815) in the population of patients with CAD or PAD. Clinical benefits and associated costs of RIV+ASA vs ASA result in an ICER of €11,534 per QALY gained and of €10,146 per LY saved.**
- **Overall, the results of the cost-effectiveness model support the use of rivaroxaban 2.5 mg bid in combination with ASA as a cost-effective treatment option, at cost-effectiveness threshold of €30,000/QALY, in patients with CAD or PAD, compared to ASA alone, in the Greek setting.**

BACKGROUND & OBJECTIVE

- **Both coronary artery disease (CAD) and peripheral artery disease (PAD) are clinical presentations of atherosclerosis which is a progressive condition affecting the large and medium-sized arteries [1, 2]. CAD is caused by atherosclerosis of the coronary arteries that leads to a restriction of blood flow to the heart [3]. Clinical presentations of CAD include acute coronary syndrome (ACS) in the form of unstable angina (UA) or myocardial infection (MI), as well as stable angina [4, 5]. PAD is caused by atherosclerosis of the arteries of the lower extremities and sometimes including carotid artery stenosis [6]. Clinical presentations of PAD include asymptomatic and symptomatic disease, the latter including intermittent claudication (IC), critical (or chronic) limb ischemia [7] and acute limb ischemia (ALI) [8].**
- **Rivaroxaban is a selective direct factor Xa inhibitor that is used to prevent and treat venous thromboembolism and to prevent stroke or systemic embolism in atrial fibrillation (AF). The Cardiovascular Outcomes for People Using Anticoagulation Strategies (COMPASS) trial (NCT01776424) [9] demonstrated that rivaroxaban 2.5 mg bid in combination with acid acetylsalicylic (ASA) is more effective than acid acetylsalicylic alone in preventing cardiovascular events, with acceptable safety, in patients with coronary artery disease (CAD) and symptomatic peripheral artery disease (PAD).**
- **The objective of this study was to evaluate the cost-effectiveness of rivaroxaban 2.5 mg bid in combination with acid acetylsalicylic 100 mg once daily (RIV+ASA) compared to acid acetylsalicylic 100 mg od alone (ASA), in patients with CAD or symptomatic PAD over a lifetime horizon, to the Greek setting.**

METHODS

Model structure

- A schematic of the Markov model is presented in Figure 1.
- Event-free health state is the starting point, patients can remain there, or experience a first main event, a second acute main event or death.
- As "Main events" we define Myocardial infraction (MI), Ischaemic Stroke (IS) or Intracranial Haemorrhage (ICH) that are opposed to other events that a patient may experience, such as bleeds or limb events (health events), during the model simulation.
- As "Health Events" we define extracranial bleed event or limb events:
 - Extracranial bleed event: Major extracranial non-fatal bleeds (as modified by the International Society on Thrombosis and Haemostasis-ISTH)
 - Limb events: Acute Limb Ischemia (ALI), Minor amputation, Major amputation, Venous Thrombo-Embolism (VTE)
- When experiencing the first main event, patients transition to an acute health state for one cycle and to a post-acute health state for subsequent cycles, if not fatal
- Again, patients can remain in the post-acute health state or experience a second event. When patients experience a second main event, patients transition to another acute health state for one cycle and to a post-acute health state for subsequent cycles, if not fatal
- At any point of the model, patients can experience death

Figure 1. Model diagram

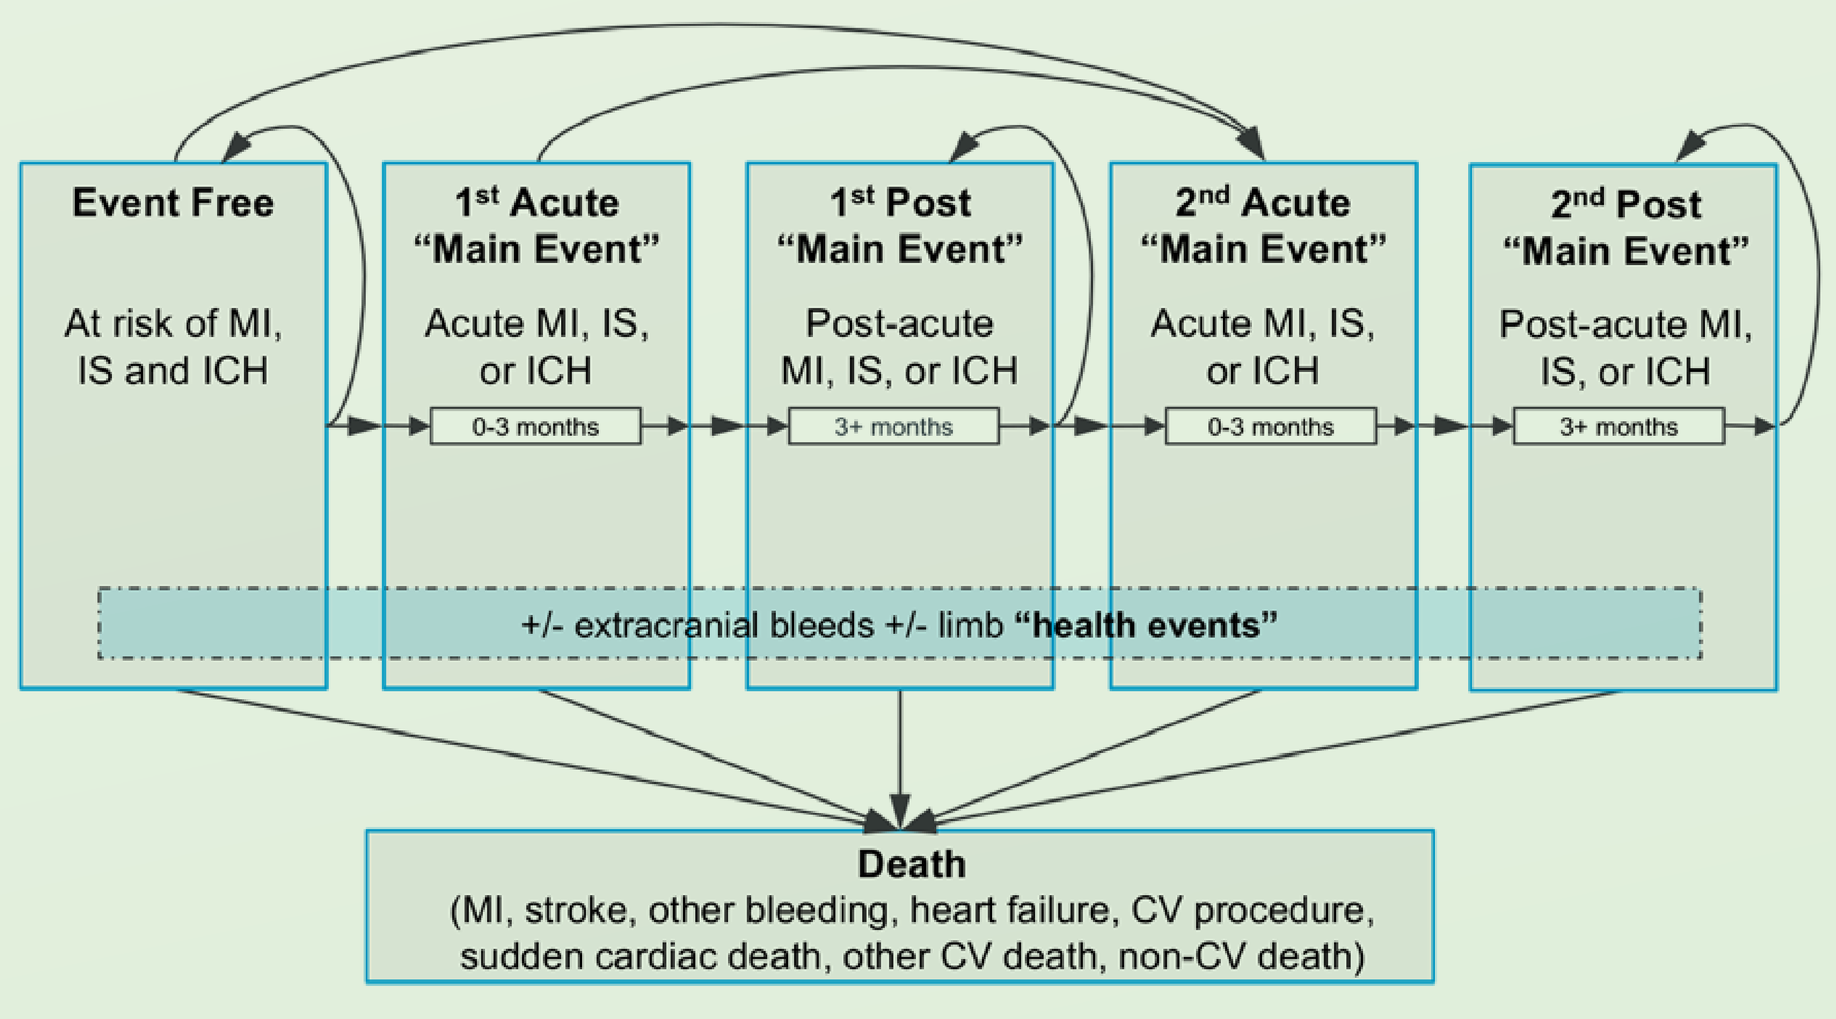

Event Free
At risk of MI, IS and ICH
1st Acute "Main Event"
Acute MI, IS, or ICH
0-3 months
1st Post "Main Event"
Post-acute MI, IS, or ICH
3+ months
2nd Acute "Main Event"
Acute MI, IS, or ICH
0-3 months
2nd Post "Main Event"
Post-acute MI, IS, or ICH
3+ months
+/- extracranial bleeds +/- limb "health events"
Death
(MI, stroke, other bleeding, heart failure, CV procedure, sudden cardiac death, other CV death, non-CV death)

CV: cardiovascular; ICH: Intracranial Haemorrhage; IS: Ischaemic Stroke; MI: Myocardial Infraction

Model features

Population
- The populations modelled reflect the COMPASS trial population [9], and, as such, include the overall COMPASS population (CAD or PAD)

Time horizon: lifetime
- Rationale
 - CV risks and outcomes are relevant for the duration of a patient's life
 - Treatment is continued over the patient's lifetime
- Simulation of patients trajectories up to 100 years old
- Mean age in COMPASS: 68y ➝time horizon is 33 years

Cycle length: 3 month
- Rationale
 - Relevant granularity to capture multiple events and associated impact on costs and health outcomes
 - In line with previous models, and also validated with experts
- Conservative assumption: 1 single event by cycle, supported by
- COMPASS results:
 - Patients with ≥2 within 3 months: 5 in RIV+ASA arm (0.055%), 8 in ASA arm (0.087%)
- Half cycle correction applied

Discount rate: 3.5%

Comparators
The main comparison of the model is between the two regimens studied in COMPASS [9]:
- rivaroxaban 2.5 mg bid in combination with acid acetylsalicylic 100 mg once daily (od)
- acid acetylsalicylic 100 mg od alone [9-19]

MODEL INPUTS

Clinical efficacy and utility input

- The main clinical data inputs were based on the COMPASS study [9] and data on file.
- Utility values for the base case are sourced from COMPASS EQ-5D multivariate analyses [9]. Utility values for health states and utility decrements for health events are assumed to be the same in all treatment arms, as there is no evidence to suggest that treatment choice has any impact on quality of life. In addition, utility for death is considered to be 0.

Cost inputs

- The adapted cost-effectiveness model considers the Greek National Healthcare system perspective and thus it does include only direct costs. The following types of direct costs are considered in the model: treatment costs and cost of management of potential associated main events, such as Myocardial Infarction (MI), Ischaemic Stroke (IS), Intracranial Haemorrhage (ICH), CV death and bleeding death and the management of potential health events, such as Acute Limb Ischemia (ALI), minor and major amputation, Venous Thrombo-Embolism (VTE).
- The model assigns medication costs to each patient and a cost to each health state. These costs are then combined with the number of patients in that health state over the time horizon. Costs of non-fatal and fatal health events are then applied to the proportion of patients experiencing those events in order to calculate the total average costs over the time horizon of the model.
- Direct cost was based on national reimbursed prices and Diagnosis Related Groups tariffs per main and health event (e.g. Official Price Bulletin, Government Gazettes) [20-23] (Table 2-4). Where appropriate, costs are inflated to 2018 Euros (€), using the EURO price deflator data from the Hellenic Statistical Authority [24].

Medication costs

Table 2: Daily medication costs

Drug	Daily dose	Pack size	Pack price*
ASA	100mg od	20	€1.17
RIV	2.5mg bid	56	€77.44
CLO	75mg od	28	€3.50

ASA: acid acetylsalicylic; CLO: clopidogrel; RIV: rivaroxaban
Source: Greek Ministry of Health. Official Pharmaceutical Price Bulletin [in Greek]. Available at http://www.moh.gov.gr/articles/times-farmakwn/deltia-timwn/5723-laquo-tropopoihsh-ths-d3-a-44071-11-6-2018-apofashs-laquo-deltio-timwn-farmakwn-anthrwpinhs-xrhshs-maioy-2018-raquo-raquo-18-7-2018. Accessed 19 April 2019.
* Pack price refers to the public list price. Cost-Effectiveness results are based on confidential net prices

Health state costs

Table 3: Health state costs

Health state	DRG per event	Reference
Acute MI	€2,823.31	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: K10M, K10X, K31M, K31X, K40M, K40X, updated to 2018 cost using price deflator)
Post-acute MI	€171.38	Government Gazette 3054/B/18-11-2012, Government Gazette 2816/B/22-12-2015 Cost includes consultation and diagnostic tests, based on KOLs input, such as a) Fatigue TEST includes: Electrocardiogram (ECG) at pre- and post-fatigue (with or without Laser), Fatigue Test on Rolled Carpet, Radioelectrohybrid; b) Myocardial SPECT imaging; c) Ultrasound Complete Study: 2-Dimensional Real-Time Bifurcation, M-Mode Record. Doppler Spectrum Ultrasound and Color Imaging Flow; d) stress echocardiography updated to 2018 cost using price deflator; adjusted for cycle length of three months
Death related to MI	€1,295.44	Proportional estimation to Acute MI cost, assuming that death related to MI cost accounts for 46% (based on the estimated costs from Rivaroxaban 2.5mg NICE submission) of Acute MI cost.
Acute IS	€1,008.79	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: N29M, N29X, N30A, N30Ma, N30Mb, N30X, updated to 2018 cost using price deflator)
Post-IS	€2,708.42	Government Gazette 3054/B/18-11-2012, Government Gazette 2816/B/22-12-2015 Cost includes consultation, imaging (CT and MRI) and rehabilitation (physiotherapy or rehabilitation center), based on KOLs input, updated to 2018 cost using price deflator; adjusted for cycle length of three months.
Acute ICH	€1,156.12	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: N30A, N30Ma, N30Mb, N30X, updated to 2018 cost using price deflator)
Post-ICH	€4,121.57	Government Gazette 3054/B/18-11-2012, Government Gazette 2816/B/22-12-2015 Cost includes consultation, imaging (CT, MRI and MRA) and rehabilitation (physiotherapy or rehabilitation center), based on KOLs input, updated to 2018 cost using price deflator; adjusted for cycle length of three months.
Death related to stroke	€3,887.34	Proportional estimation to Acute MI cost, assuming that death related stroke cost accounts for 138% (based on the estimated costs from Rivaroxaban 2.5mg NICE submission) of Acute MI cost.
Bleeding death and heart failure death	€259.15	Proportional estimation to Acute MI cost, assuming that bleeding death and heart failure death account for 9% (based on the estimated costs from Rivaroxaban 2.5mg NICE submission) of Acute MI cost.
CV procedure death and other CV death	€2,591.56	Proportional estimation to Acute MI cost, assuming that CV procedure and other CV death account for 92% (based on the estimated costs from Rivaroxaban 2.5mg NICE submission) of Acute MI cost.
Other mortality	€1,297.31	In absence of other estimates, a weighted average of CV death costs was applied to 'all CV death'. Zero costs to sudden cardiac death and non-CV death were applied

DRGs: Diagnosis Related Groups; ALI: Acute limb ischemia; ICH: Intracranial Haemorrhage; IS: Ischaemic Stroke; MI: Myocardial infraction; VTE: Venous Thrombo-Embolism

Health Events costs

Table 4: Health events costs

Health event	DRG per event	Reference
ALI	€2,208.82	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: K14Mβ, K14X, K35M, K35X, updated to 2018 cost using price deflator)
Minor amputation	€2,381.99	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: K13M, K13X, updated to 2018 cost using price deflator)
Major amputation	€8,089.18	Government Gazette 946/B/27-3-2012, FEK 3054/B/18-11-2012 • Inpatient cost: Weighted average of DRGs: K11M, K11X, M07A, updated to 2018 cost using price deflator • Equipment cost: Wheelchair and Prosthesis cost • Rehabilitation cost: Physiotherapy cost
VTE	€1,215.08	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: A21M, A21X, K33M, K33X, updated to 2018 cost using price deflator)
Major non-fatal extracranial bleeds (ISTH)	€650.92	Government Gazette 946/B/27-3-2012 (Weighted average of DRGs: Π41M, Π41X, Y25M, Y25X, updated to 2018 cost using price deflator)

DRGs: Diagnosis Related Groups; ALI: Acute limb ischemia; ICH: Intracranial Haemorrhage; IS: Ischaemic Stroke; ISTH: International Society on Thrombosis and Haemostasis; MI: Myocardial infraction; VTE: Venous Thrombo-Embolism

RESULTS

Cost-effectiveness ratio

- Over a lifetime horizon, costs are higher for RIV+ASA compared to ASA (€13,030 vs. €9,215), with incremental costs reaching €3,815 (Table 6 & Figure 2). This is largely due to the higher drug costs (€6,081), although savings are associated with the reduced rates for events.
- The benefits result to 0.33 additional Quality Adjusted Life Years (QALYs) and 0.38 additional Life Years (LYs) for RIV+ASA, compared to ASA. As shown in Table 6 and Figure 2, clinical benefits and associated costs vs ASA result in an Incremental Cost Effectiveness Ratio (ICER) of €11,534 per QALY gained and of €10,146 per LY saved.
- Based on the analysis, RIV+ASA is cost-effective vs. ASA in patients with CAD or PAD as ICER falls below the acceptable threshold considered to be €30,000/ QALY.

Table 6: Costs effectiveness results

	RIV+ASA	ASA	Incr. vs ASA
Drug costs	6,221€	140€	6,081€
Ongoing medical care	5,769€	7,811€	-2,042€
Non-fatal acute CV events	535€	647€	-111€
Mortality	227€	285€	-58€
Health events (direct costs)	278€	332€	-54€
	RIV+ASA vs ASA		
Outcome	Incr. costs (€)	Incr. outcomes	ICER (€/outcome)
Per QALY gained	3,815	0.33	**11,534**
per LY gained	3,815	0.38	**10,146**

CV: cardiovascular; QALY: Quality Adjusted Life Year; LY: Life Year

Figure 2: Cost-effectiveness plane

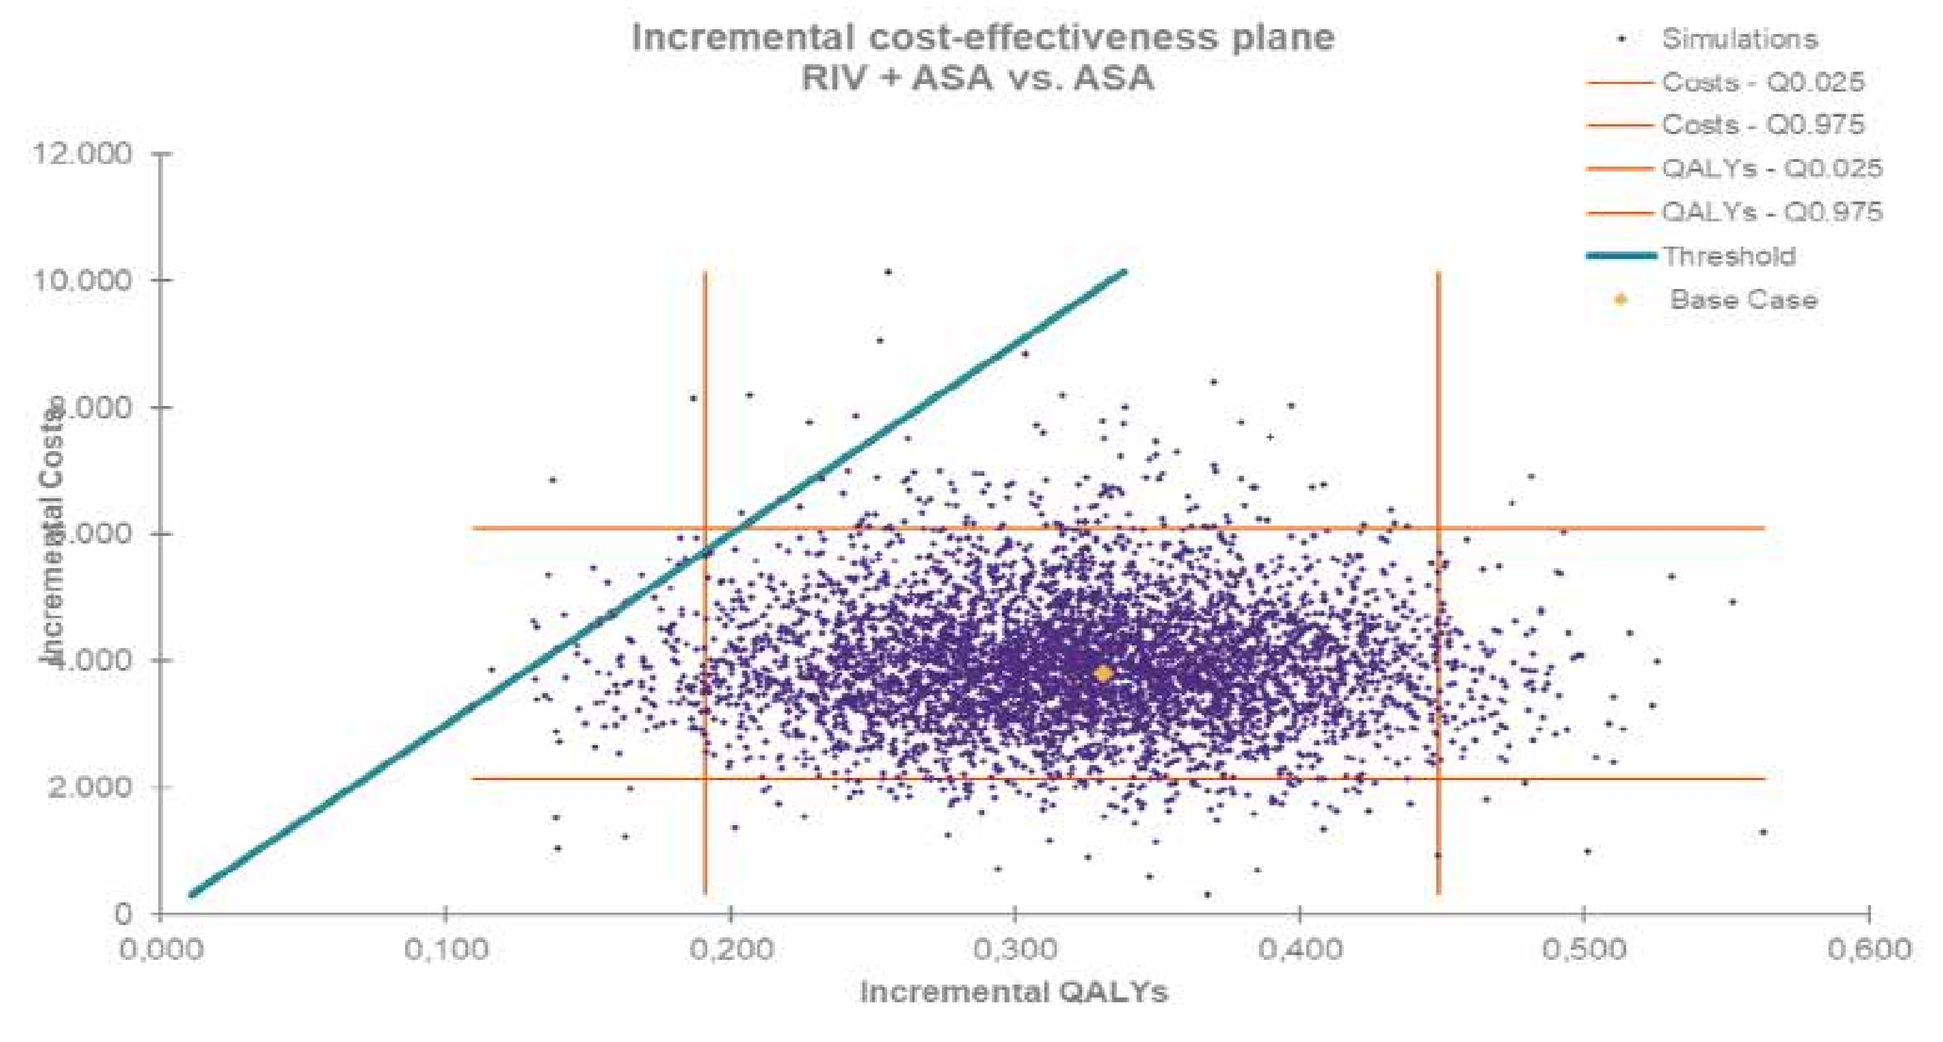

Incremental cost-effectiveness plane
RIV + ASA vs. ASA
Simulations
Costs - Q0.025
Costs - Q0.975
QALYs - Q0.025
QALYs - Q0.975
Threshold
Base Case
12.000
10.000
8.000
6.000
4.000
2.000
0
Incremental Costs
0,000
0,100
0,200
0,300
0,400
0,500
0,600
Incremental QALYs

Sensitivity analyses

- Overall, sensitivity analyses confirm that the model is robust to changes in the majority of input parameters.
- The One-way sensitivity analysis (OWSA) shows that the main ICER drivers in CAD or PAD population includes efficacy data related to Ischaemic Stroke (IS) and Intracranial Hemorrhage (ICH) (Figure 3).
- Using 5,000 simulations, the Probabilistic Sensitivity Analysis (PSA) shows that the mean ICER is €12,313/QALY with the probability that RIV+ASA is cost-effective against ASA being 93.1% (at cost-effectiveness threshold of €20,000/QALY) and 99.4% (at cost-effectiveness threshold of €30,000/QALY) (Table 7).

Figure 3: OWSA results

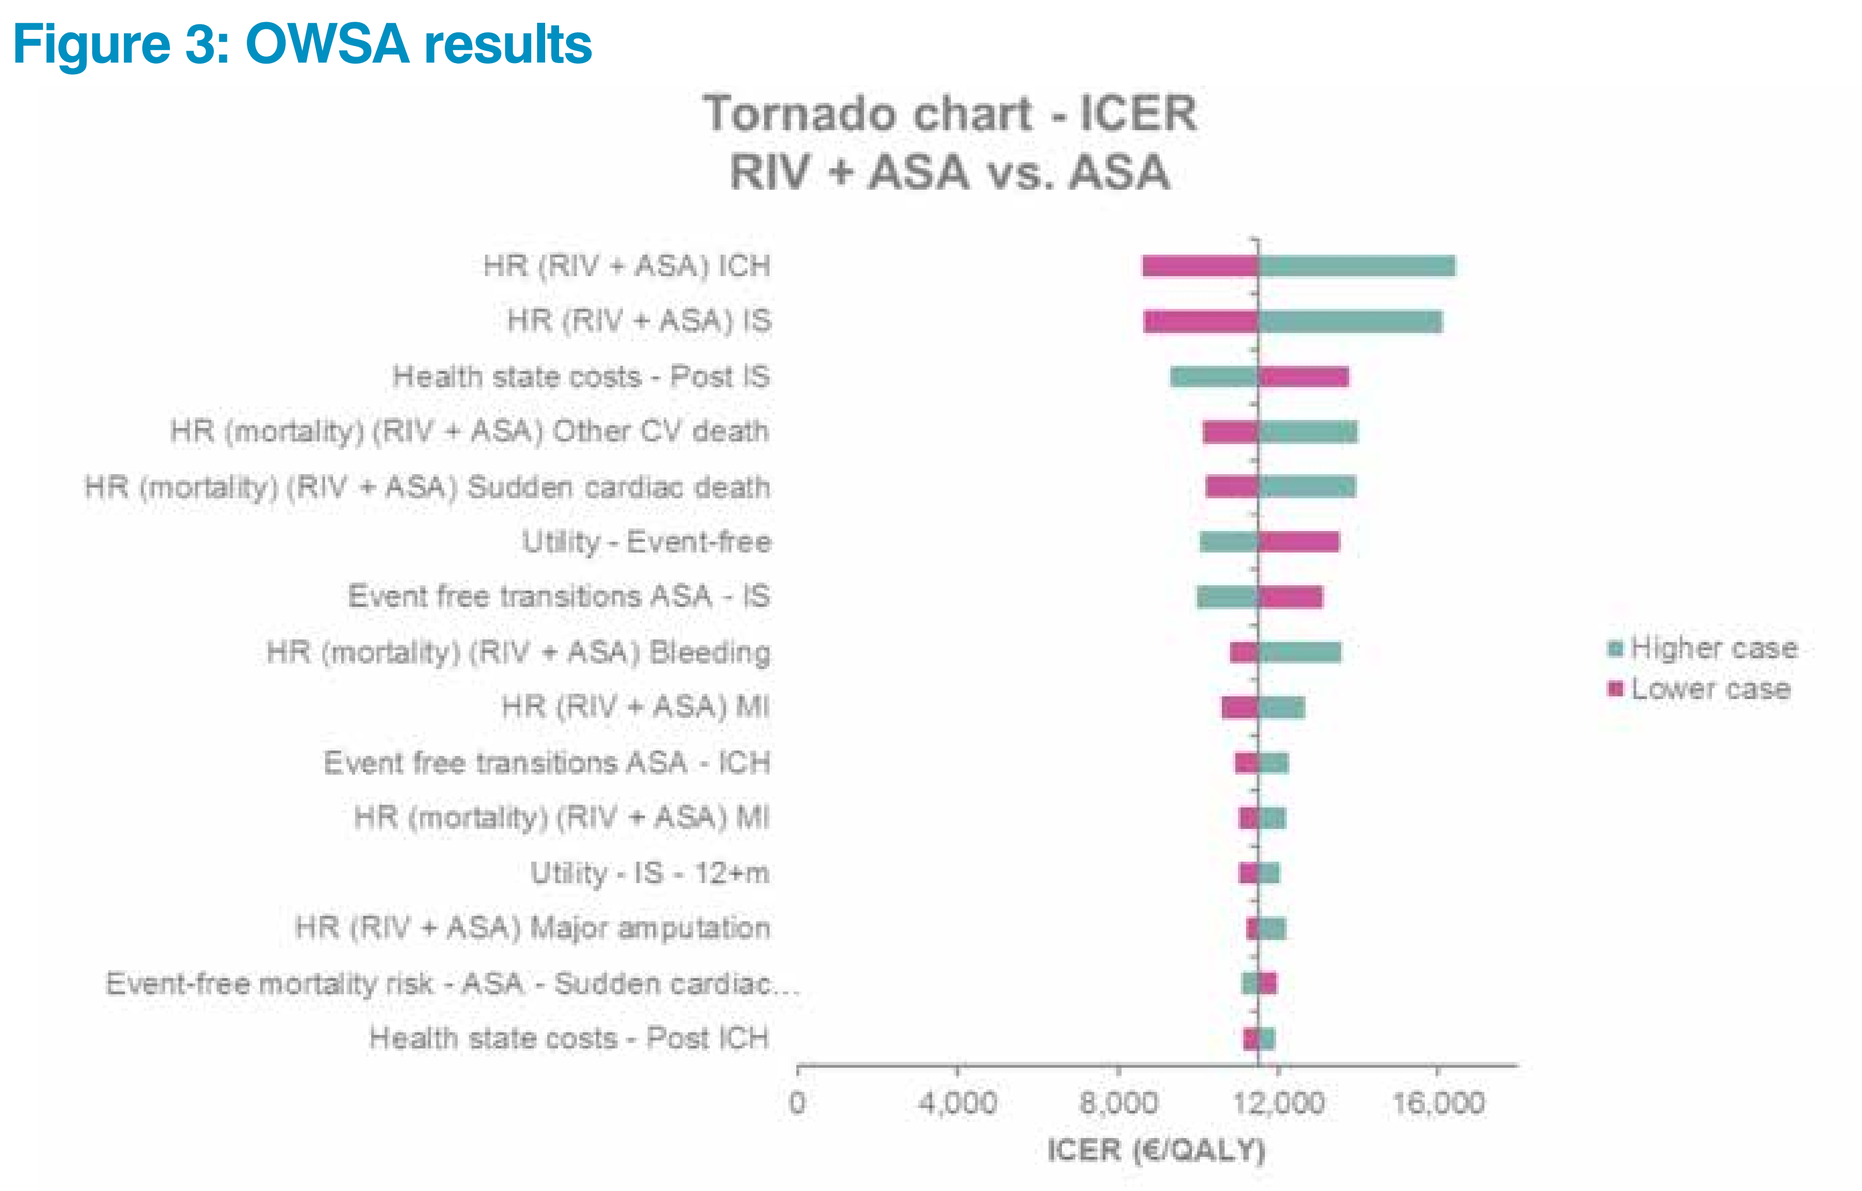

Tornado chart - ICER
RIV + ASA vs. ASA
HR (RIV + ASA) ICH
HR (RIV + ASA) IS
Health state costs - Post IS
HR (mortality) (RIV + ASA) Other CV death
HR (mortality) (RIV + ASA) Sudden cardiac death
Utility - Event-free
Event free transitions ASA - IS
HR (mortality) (RIV + ASA) Bleeding
HR (RIV + ASA) MI
Event free transitions ASA - ICH
HR (mortality) (RIV + ASA) MI
Utility - IS - 12+m
HR (RIV + ASA) Major amputation
Event-free mortality risk - ASA - Sudden cardiac...
Health state costs - Post ICH
Higher case
Lower case
0
4,000
8,000
12,000
16,000
ICER (€/QALY)

Table 7: PSA results

	Incr. costs (€)	Incr. QALYs	ICER (€/QALYs)
Base Case	**3,815**	**0.33**	**11,534**
Mean	3,953	0.32	12,313
Std Deviation	981	0.07	4,508
Median	3,895	0.32	12,189
Min	325	0.11	885
Q1	2,144	0.19	6,239
Q3	6,106	0.45	23,871
Max	10,138	0.56	49,719
Probability (<€20,000 threshold)			93.08%
Probability (<€30,000 threshold)			99.44%

References
1. Mozaffarian D, Wilson PW, Kannel WB. Beyond established and novel risk factors: lifestyle risk factors for cardiovascular disease. Circulation 2008;117(23):3031-8.
2. O'Flaherty M, Sans-Menendez S, Capewell S, Jørgensen T. Epidemiology ofatherosclerotic cardiovascular disease: scope of the problem and itsdeterminants. ESC Textbook Prev Cardiol. 2015;1.
3. Sanchis-Gomar F, Perez-Quilis C, Leischik R, Lucia A. Epidemiology of coronary heart disease and acute coronary syndrome. Ann Transl Med 2016;4(13):256-267.
4. Montalescot G, Sechtem U, Achenbach S, Andreotti F, Arden C, Budaj A, et al. 2013 ESC guidelines on the management of stable coronary artery disease: the Task Force on the management of stable coronary artery disease of the European Society of Cardiology. Eur Heart J 2013;34(38):2949-3003.
5. Fihn SD, Gardin JM, Abrams J, et al. 2012 ACCF/AHA/ACP/AATS/PCNA/SCAI/STS Guideline for the diagnosis and management of patients with stable ischemic heart disease: a report of the American College of Cardiology Foundation/American Heart Association Task Force on Practice Guidelines, and the American College of Physicians, American Association for Thoracic Surgery, Preventive Cardiovascular Nurses Association, Society for Cardiovascular Angiography and Interventions, and Society of Thoracic Surgeons. J Am Coll Cardiol 2012;60(24):e44-e164.
6. Gerhard-Herman MD, Gornik HL, Barrett C, et al. 2016 AHA/ACC Guideline on the Management of Patients with Lower Extremity Peripheral Artery Disease: Executive Summary. Vasc Med 2017;22(3):NP1-NP43
7. Howard DPJ, Banerjee A, Fairhead JF, et al. Population-Based Study of Incidence, Risk Factors, Outcome, and Prognosis of Ischemic Peripheral Arterial Events: Implications for Prevention. Circulation 2015;132(19): 1805–1815.
8. Tendera M, Bartelink V, Baumgartner ML, et al. ESC Guidelines on the diagnosis and treatment of peripheral artery diseases: Document covering atherosclerotic disease of extracranial carotid and vertebral, mesenteric, renal, upper and lower extremity arteries: the Task Force on the Diagnosis and Treatment of Peripheral Artery Diseases of the European Society of Cardiology (ESC). Eur Heart J 2011;32(22):2851-906.
9. Eikelboom JW, Connolly SJ, Bosch J, et al. Rivaroxaban with or without aspirin in stable cardiovascular disease. N Engl J Med 2017;377(14):1319-30.
10. Montalescot G, Sechtem U, Achenbach S, et al. 2013 ESC guidelines on the management of stable coronary artery disease: the Task Force on the management of stable coronary artery disease of the European Society of Cardiology. Eur Heart J 2013;34(38):2949-3003.
11. NICE. Stable angina: management [Clinical guideline]. 2011 (updated 2016). Available from: https://www.nice.org.uk/guidance/cg126/resources/stable-angina-management-pdf-35109453262021.
12. Mancini GB, Gosselin G, Chow B, et al. Canadian Cardiovascular Society guidelines for the diagnosis and management of stable ischemic heart disease. Can J Cardiol 2014;30(8):837-49.
13. Gerhard-Herman MD, Gornik HL, Barrett C, et al. 2016 AHA/ACC Guideline on the Management of Patients With Lower Extremity Peripheral Artery Disease: A Report of the American College of Cardiology/American Heart Association Task Force on Clinical Practice Guidelines. Circulation 2017;135(12):e726-e79.
14. Aboyans V, Ricco JB, Bartelink MEL, et al. 2017 ESC Guidelines on the Diagnosis and Treatment of Peripheral Arterial Diseases, in collaboration with the European Society for Vascular Surgery (ESVS): Document covering atherosclerotic disease of extracranial carotid and vertebral, mesenteric, renal, upper and lower extremity arteries, Endorsed by: the European Stroke Organization (ESO)The Task Force for the Diagnosis and Treatment of Peripheral Arterial Diseases of the European Society of Cardiology (ESC) and of the European Society for Vascular Surgery (ESVS). Eur Heart J 2017;39(9):763-816 .
15. Levine G.N, Bates E R, Bittl JA, et al. 2016 ACC/AHA guideline focused update on duration of dual antiplatelet therapy in patients with coronary artery disease: A report of the American College of Cardiology/American Heart Association Task Force on Clinical Practice Guidelines. J Thorac Cardiovasc Surg 2016;152(5):1243-1275.
16. Neumann F-J, Sousa-Uva M, Ahlsson A, et al. 2018 ESC/EACTS Guidelines on myocardial revascularization: the Task Force on Myocardial Revascularization of the European Society of Cardiology (ESC) and the European Association for Cardio-Thoracic Surgery (EACTS). Developed with the special contribution of the European Association of Percutaneous Cardiovascular Interventions (EAPCI). Eur Heart J 2019;40:87-165.
17. Valgimigli M, Bueno H, Byrne RA, et al. 2017 ESC focused update on dual antiplatelet therapy in coronary artery disease developed in collaboration with EACTS: The Task Force for dual antiplatelet therapy in coronary artery disease of the European Society of Cardiology (ESC) and of the European Association for Cardio-Thoracic Surgery (EACTS). Eur Heart J 2018;39(3):213-260.
18. Roffi M, Patrono C, Collet JP, et al. 2015 ESC Guidelines for the management of acute coronary syndromes in patients presenting without persistent ST-segment elevation: Task Force for the Management of Acute Coronary Syndromes in Patients Presenting without Persistent ST-Segment Elevation of the European Society of Cardiology (ESC). Eur Heart J 2016;37(3):267-315.
19. Brott TG, Halperin JL, Abbara S, et al. 2011 ASA/ACCF/AHA/AANN/AANS/ACR/ASNR/CNS/SAIP/SCAI/SIR/SNIS/SVM/SVS guideline on the management of patients with extracranial carotid and vertebral artery disease: executive summary. A report of the American College of Cardiology Foundation/American Heart Association Task Force on Practice Guidelines, and the American Stroke Association, American Association of Neuroscience Nurses, American Association of Neurological Surgeons, American College of Radiology, American Society of Neuroradiology, Congress of Neurological Surgeons, Society of Atherosclerosis Imaging and Prevention, Society for Cardiovascular Angiography and Interventions, Society of Interventional Radiology, Society of NeuroInterventional Surgery, Society for Vascular Medicine, and Society for Vascular Surgery. Circulation 2011;124(4): 489-532.
20. Greek Ministry of Health and Greek Ministry of Finance. Ministerial Decision Government Gazette 946/B/27-3-2012 [In Greek]. Available at https://www.e-nomothesia.gr/kat-ygeia/perithalpse/kya-4a-oik-18051-2012.html. Accessed 19 April 2019.
21. Greek Ministry of Health and Greek Ministry of Finance. Government Gazette 3054/B/18-11-2012 [in Greek]. Available at https://www.e-nomothesia.gr/kat-ygeia/perithalpse/kya-emp-5-2012.html. Accessed 19 April 2019.
22. Greek Ministry of Health. Government Gazette 2816/B/22-12-2015 [in Greek]. Available at https://www.e-nomothesia.gr/kat-ygeia/perithalpse/upourgike-apophase-g3g-oik-98494-2015.html. Accessed 19 April 2019
23. Greek Ministry of Health. Official Pharmaceutical Price Bulletin [in Greek]. Available at http://www.moh.gov.gr/articles/times-farmakwn/deltia-timwn/5723-laquo-tropopoihsh-ths-d3-a-44071-11-6-2018-apofashs-laquo-deltio-timwn-farmakwn-anthrwpinhs-xrhshs-maioy-2018-raquo-raquo-18-7-2018. Accessed 19 April 2019.
24. Hellenic Statistical Authority. Evolution of the annual rates of change of the Overall Consumer Price Index. Available at https://www.statistics.gr/en/statistics/-/publication/DKT87/-

Funding & Acknowledgments
- The study was funded by Bayer Hellas
- The authors would like to thank Prof. Dimitrios Alexopoulos, Dr. Vassilis Voudris and Ass. Prof. John Kakisis for their contribution in providing inputs for the Cost-Effectiveness model adaptation to the Greek setting