Anti-VEGF injections in Portugal: the first decade 2007-2017

Ana Patrícia Marques[1], Antonio Filipe Macedo[2], João Victor Rocha[1], Marta Afonso-Silva[3], Pedro A. Laires[3], Rui Santana[1]

1 NOVA National School of Public Health, Public Health Research Centre, Universidade NOVA de Lisboa, Portugal
2 Department of Medicine and Optometry, Linnaeus University, Kalmar, Sweden
3 Novartis Farma, Produtos Farmacêuticos SA, Porto Salvo, Portugal

Introduction

- Antivascular endothelial growth factor (anti-VEGF) intravitreal injections are sight saving treatments for patients with neovascular age-related macular degeneration (nAMD), diabetic macular edema (DME), retinal vein occlusion (RVO) and other indications [1,2,3].
- Anti-VEGFs were licensed for the treatment of nAMD by the European Medicines Agency (EMA) in 2007.
- Since 2008 that they have been covered by the Portuguese National Health Service (NHS) [4].

Objectives

- The aim of this study was to examine the dissemination of anti-VEGFs injections in the Portuguese NHS.

Methods

We used an administrative database of inpatient and day cases stays from all Portuguese NHS hospitals during the period of 2007 - 2017.

Setting
- Portuguese NHS Hospitals

Period of analysis
- From 2007 to 2017 (data for the year 2017 is preliminary, therefore the values for this year may be underestimated)

Database
- Database for inpatient and day cases episodes

Eligibility criteria
- Surgical codes for eye procedures coded with ICD9CM and ICD10

Outcome measures
- Absolute values of episodes and treated patients
- Yearly episodes and treated patients
- Yearly rates of episodes per 100,000 habitants
- Yearly rates of treated patients per 100,000 habitants

Results

The sample included 319,320 episodes and 131,816 patients from 2007 to 2017. Patients had a mean age of 68.0 years (standard deviation = 13.9) and 50.0% were men.

Figure 1 - *Annual number of hospital episodes of anti-VEGFs treatments and annual number of treated patients from 2007 to 2017*

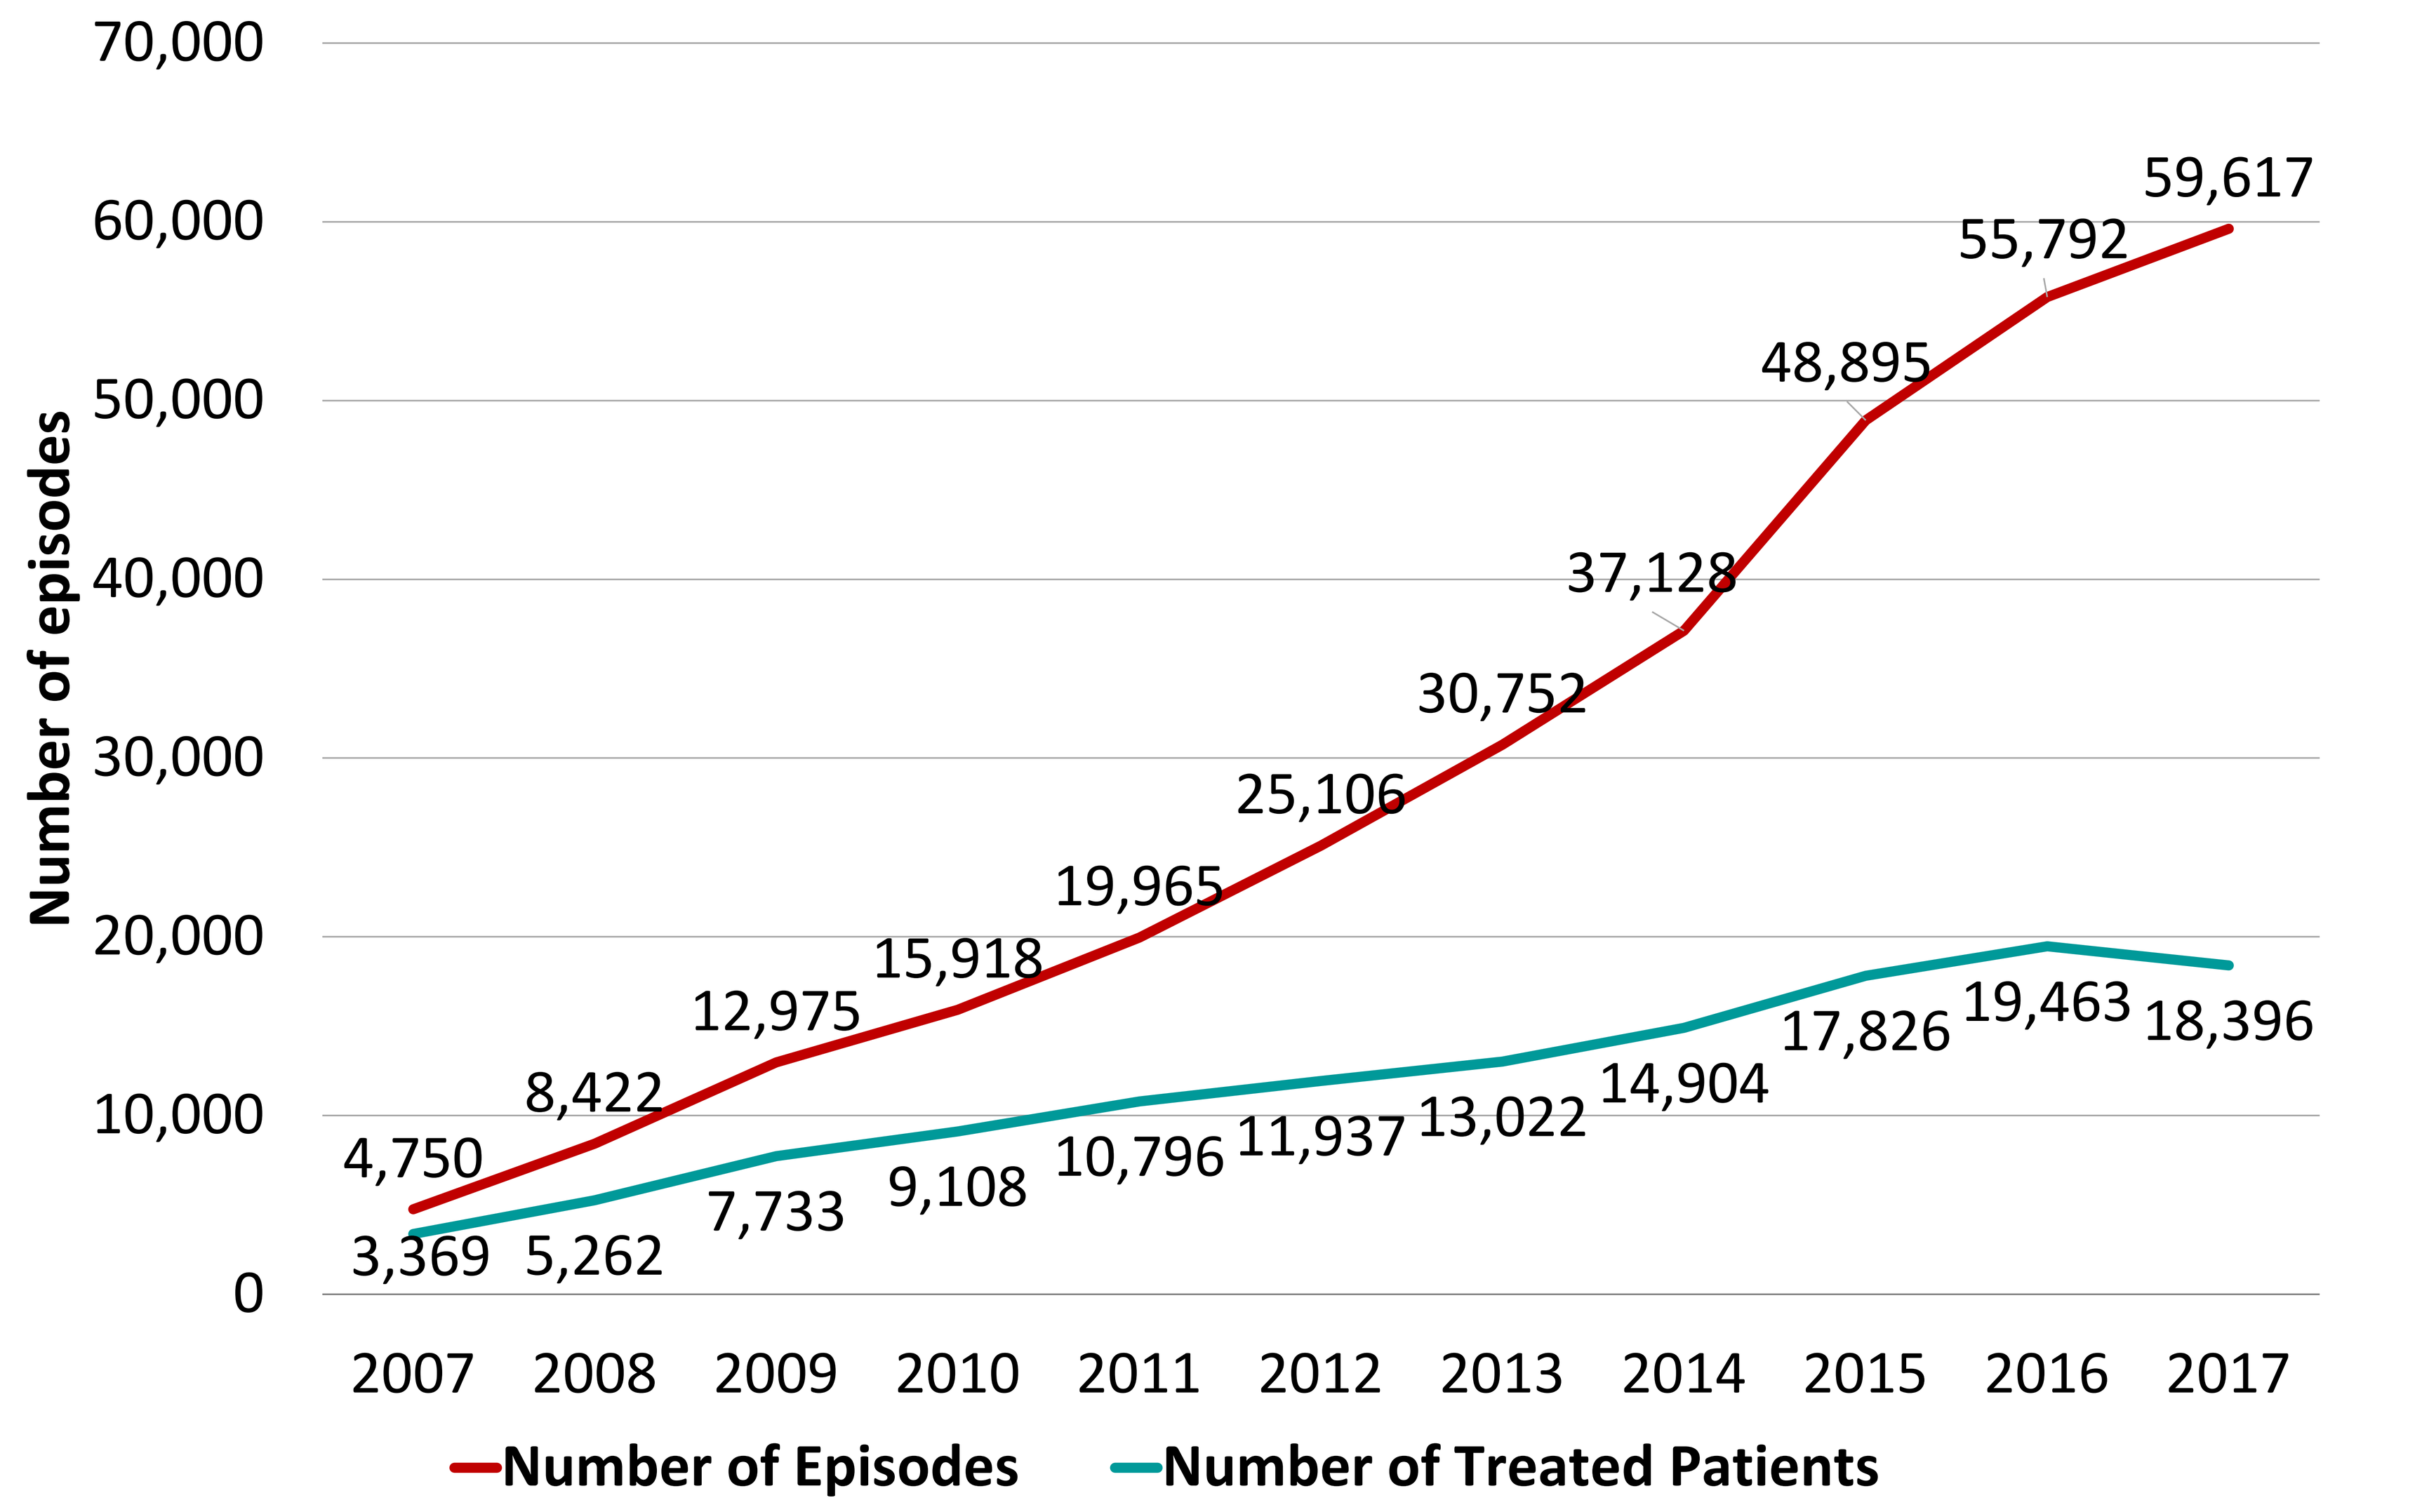
Number of episodes
70,000
60,000
50,000
40,000
30,000
20,000
10,000
0
2007
2008
2009
2010
2011
2012
2013
2014
2015
2016
2017
4,750
8,422
12,975
15,918
19,965
25,106
30,752
37,128
48,895
55,792
59,617
3,369
5,262
7,733
9,108
10,796
11,937
13,022
14,904
17,826
19,463
18,396
Number of Episodes
Number of Treated Patients

Figure 2 - *Annual number of hospital episodes and treated patients by diagnoses from 2007 to 2017*

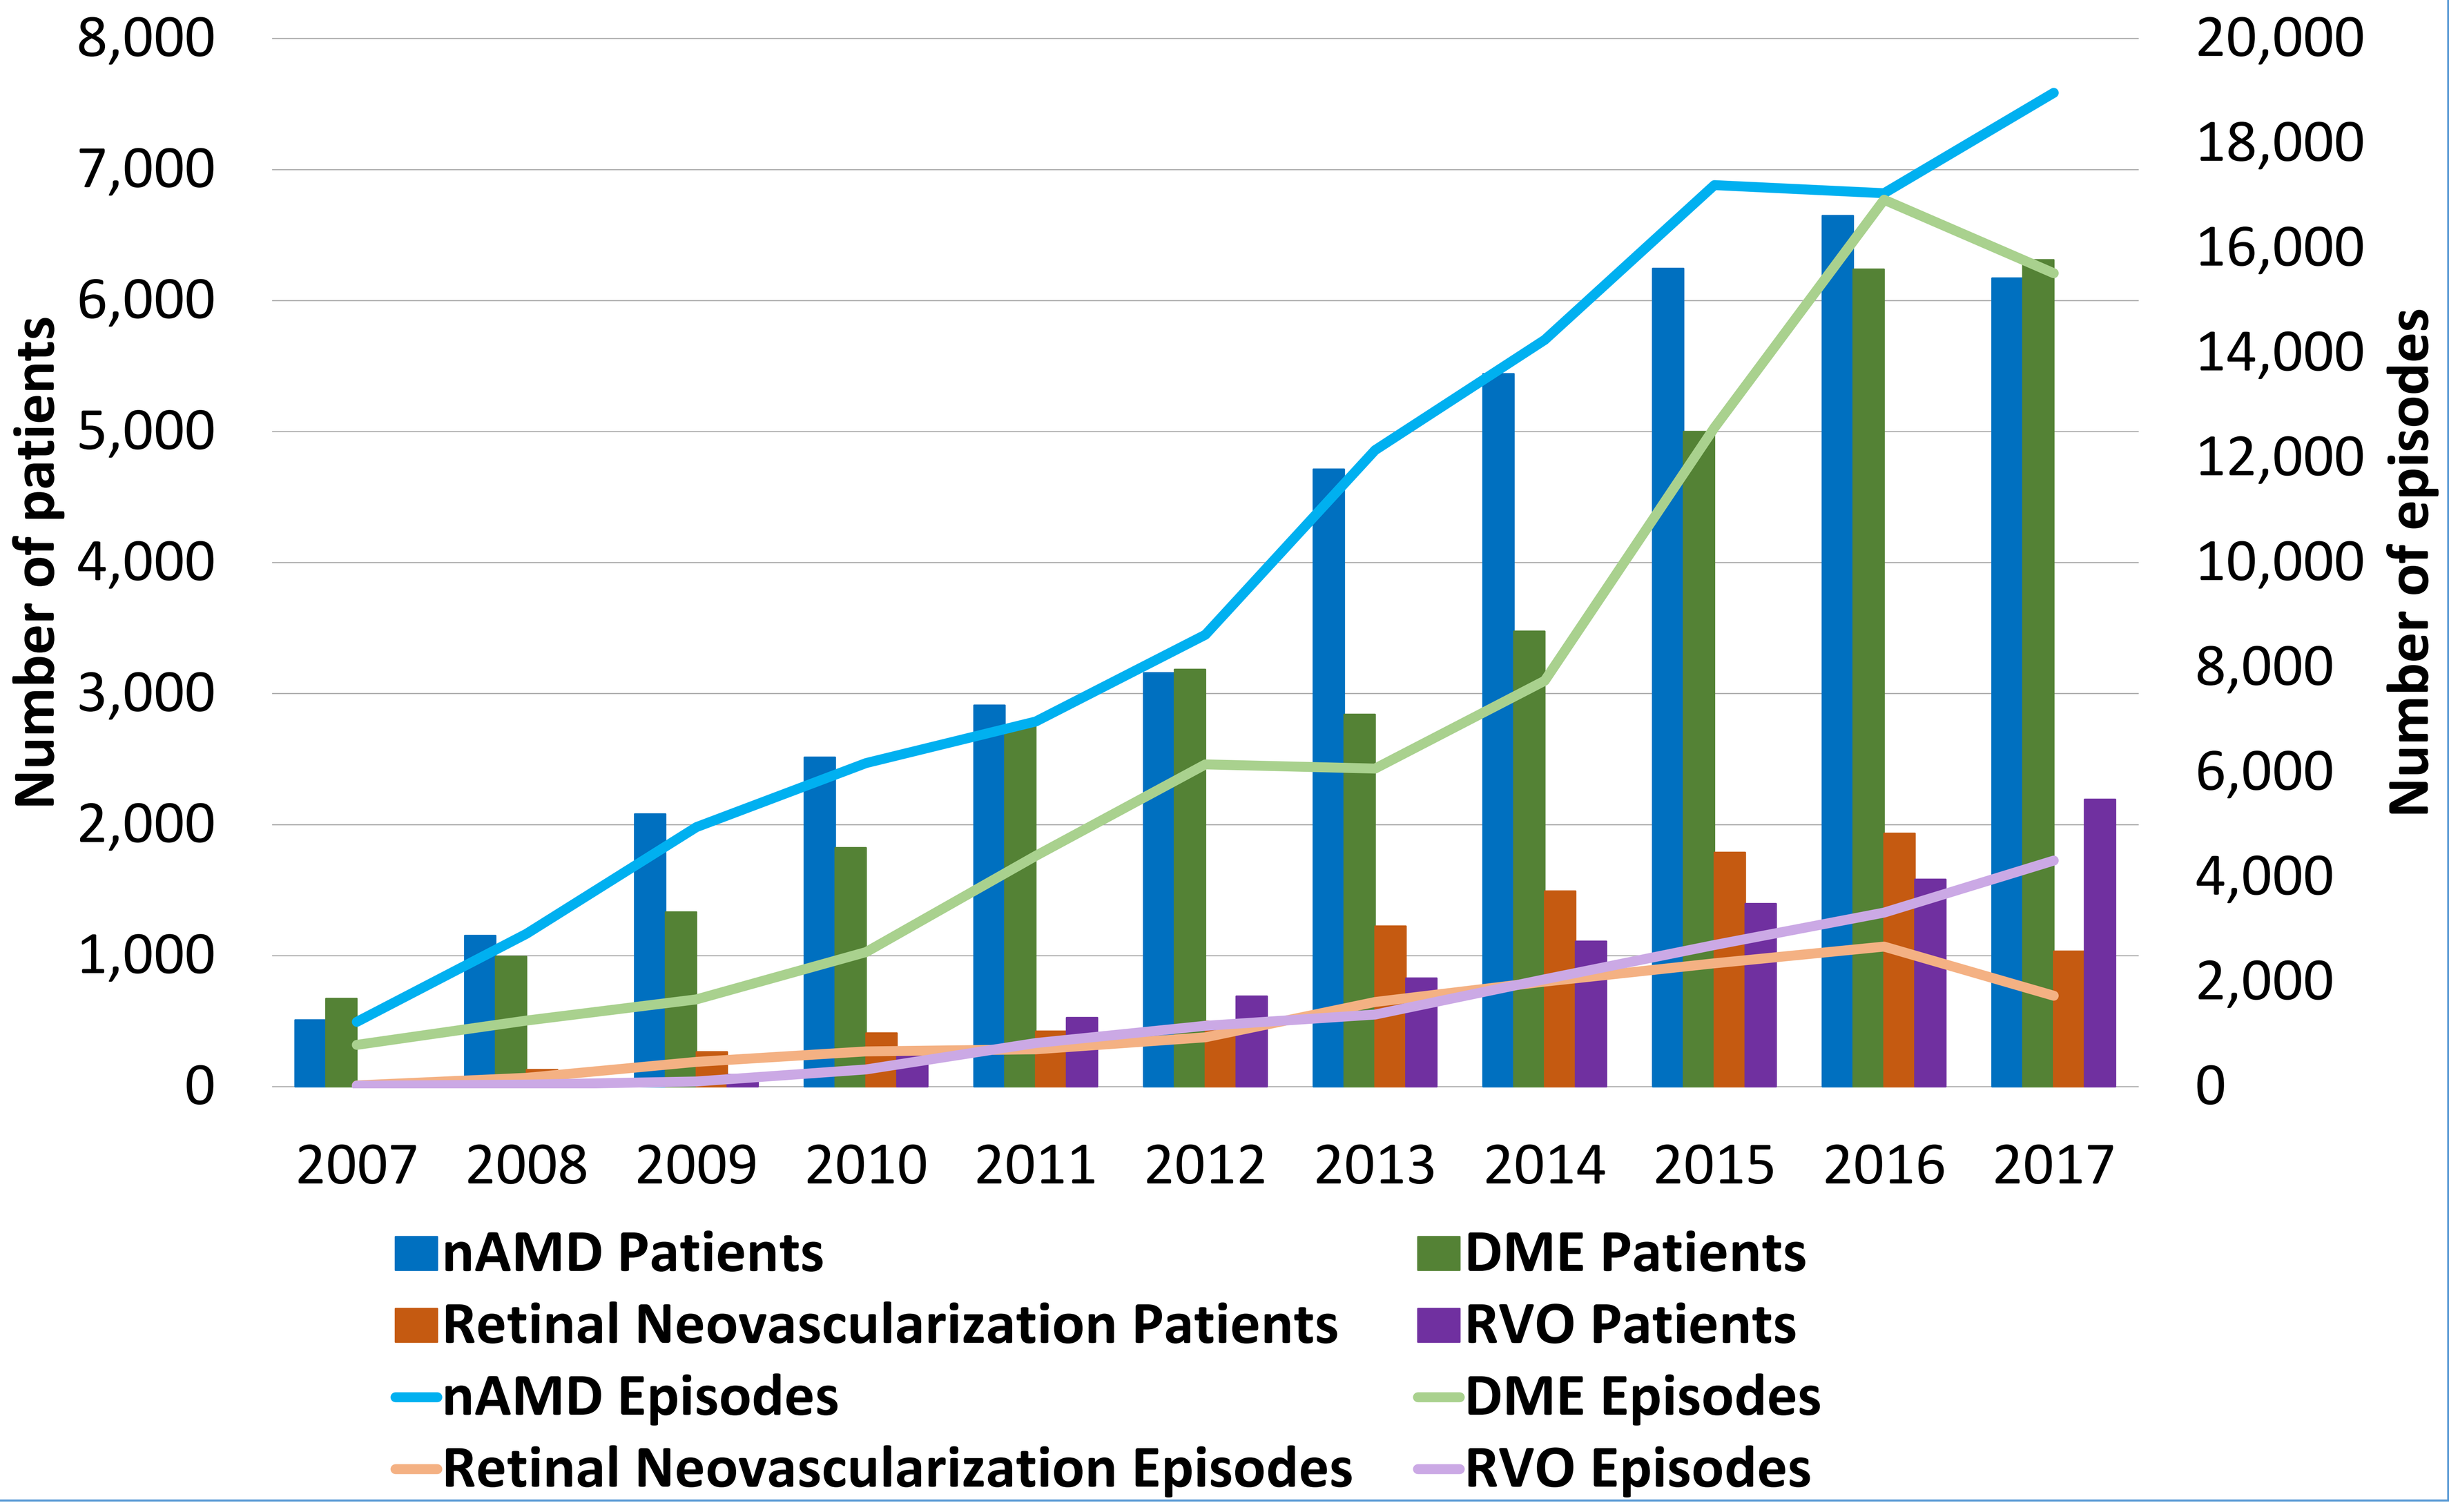
Number of patients
8,000
7,000
6,000
5,000
4,000
3,000
2,000
1,000
0
Number of episodes
20,000
18,000
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
2007
2008
2009
2010
2011
2012
2013
2014
2015
2016
2017
nAMD Patients
DME Patients
Retinal Neovascularization Patients
RVO Patients
nAMD Episodes
DME Episodes
Retinal Neovascularization Episodes
RVO Episodes

Figure 1 shows that the total number of episodes increased from 4,750 in 2007 to 59,617 in 2017. These values corresponded to a mean annual increase of episodes of 30%, to an increase in rates of episodes related to anti-VEGFs from 47.3 to 568.8 per 100,000 habitants and to an increase of rates of treated patients from 33.6 to 187.9 per 100,000 habitants.

Figure 2 shows that nAMD and DME are the two most frequent diagnoses associated with anti-VEGF treatments. The four diagnoses analyzed have shown increase in trends for number of episodes and patients for most of the analyzed period.

Conclusion

The number of procedures related with anti-VEGF treatments grew exponentially since its approval. These events can impose challenges to the NHS in terms of budget and access. Planning the dissemination of innovative treatments is important to ensure these interventions are available to all people that can benefit from it.

[1] Cheung N, Lam DS, Wong TY. Anti-vascular endothelial growth factor treatment for eye diseases. BMJ 2012;344:e2970
[2] Saeed MU, Gkaragkani E, Ali K. Emerging roles for antiangiogenesis factors in management of ocular disease. Clin Ophthalmol 2013;6:533–43. 21.
[3] Kovach JL, Schwartz SG, Flynn HW Jr, et al. Anti-VEGF treatment strategies for wet AMD. J Ophthalmol 2012;2012:786870
[4] Marques AP, Macedo AF, Perelman J, Aguiar P, Rocha-Sousa A, Santana R. Diffusion of anti-VEGF injections in the Portuguese National Health System. BMJ Open. 2015 Nov 23;5(11):e009006

IOPH/518/102019