PATIENT PREFERENCES FOR MAINTENANCE PARP IN OVARIAN CANCER TREATMENT

Laura J. Havrilesky[1,2,5], Stephanie Lim[2], Jessie A. Ehrisman[1,2], Amelia Lorenzo[1,2], Angeles Alvarez Secord[1,2,5], Jui-Chen Yang [6], F. Reed Johnson[3,4,5], Shelby D. Reed[3,4,5]

[1] Division of Gynecologic Oncology, [2] Department of Obstetrics and Gynecology, [3]Department of Medicine, Duke University Medical Center, Durham, NC, USA; [4]Duke Clinical Research Institute, Durham, NC, USA; [5]Duke Cancer Institute, Durham, NC, USA, [6]Pacific Economic Research, Bellevue, WA, USA

INTRODUCTION

- Prior to the introduction of poly ADP ribose polymerase (PARP) inhibitors, there were few maintenance therapy options for high grade serous ovarian cancer.
- Women with either germline or somatic BRCA mutations, or a loss of heterozygosity genetic defect, have substantially higher PFS gains, while those without these aberrations achieve an average PFS benefit of only a few months.
- This differential effect based on tumor characteristics leads to the question of risk-benefit balance, especially among those without these genetic defects.
- It is likely that women have personal preferences that drive the decision to take a PARP inhibitor or a treatment break after completing an effective course of chemotherapy. No prior studies have queried women on the risks and benefits of PARP maintenance or its attributes.

OBJECTIVE

- To examine the preferences of women with ovarian cancer regarding the risks, side effects and benefits afforded by maintenance therapies

METHODS

- **Survey design:** We performed a prospective discrete choice experiment survey of women with a personal history of ovarian cancer, regarding their preferences for the potential harms and benefits associated with PARP inhibitor maintenance therapy. (Table 4).
- Participants were then asked to choose between 3 clinical scenarios: Two scenarios representing attributes of maintenance therapy and a constant scenario representing a treatment break, with multiple iterations. In each maintenance therapy scenario presented, the levels of each attribute was varied experimentally.
- Participants viewed 8 to 10 individual iterations of this 3 way choice
- **Participants:** Women with a diagnosis of ovarian cancer were approached for participation, regardless of current treatment status or history of PARP inhibitor use.
- **Analysis:** Random-parameters logit regression was applied to model participants' choices as a function of attribute levels.

RESULTS

- Of 150 women with ovarian cancer recruited, 95 were eligible and completed the survey. The mean age was 62, 48% had recurrent ovarian cancer, and 13% were currently taking a PARP inhibitor.
- Participants always significantly preferred better clinical outcomes to worse (Figure 4).
- Participants valued overall survival most of all the attributes (Figure 2).
- On average, participants would tolerate a 2% additional risk of MDS/AML in exchange for 2 additional months of PFS and 7% additional risk of MDS/AML in exchange for 6 additional months of PFS.

Attribute	Attribute Levels
Monthly out of pocket cost	$0 $50 $500 $1000
Nausea	None Mild Moderate
Fatigue	None Mild Moderate
Probability of dying from myelodysplastic syndrome (MDS)	0% 1% 5% 10%
Progression-free survival	15 months 17 months 21 months
Overall survival	36 months 38 months 42 months

Table 1. Attributes, levels and descriptions. The attributes and corresponding levels were described by a clinician in a video. Video is available online at http://vimeo.com/61893779

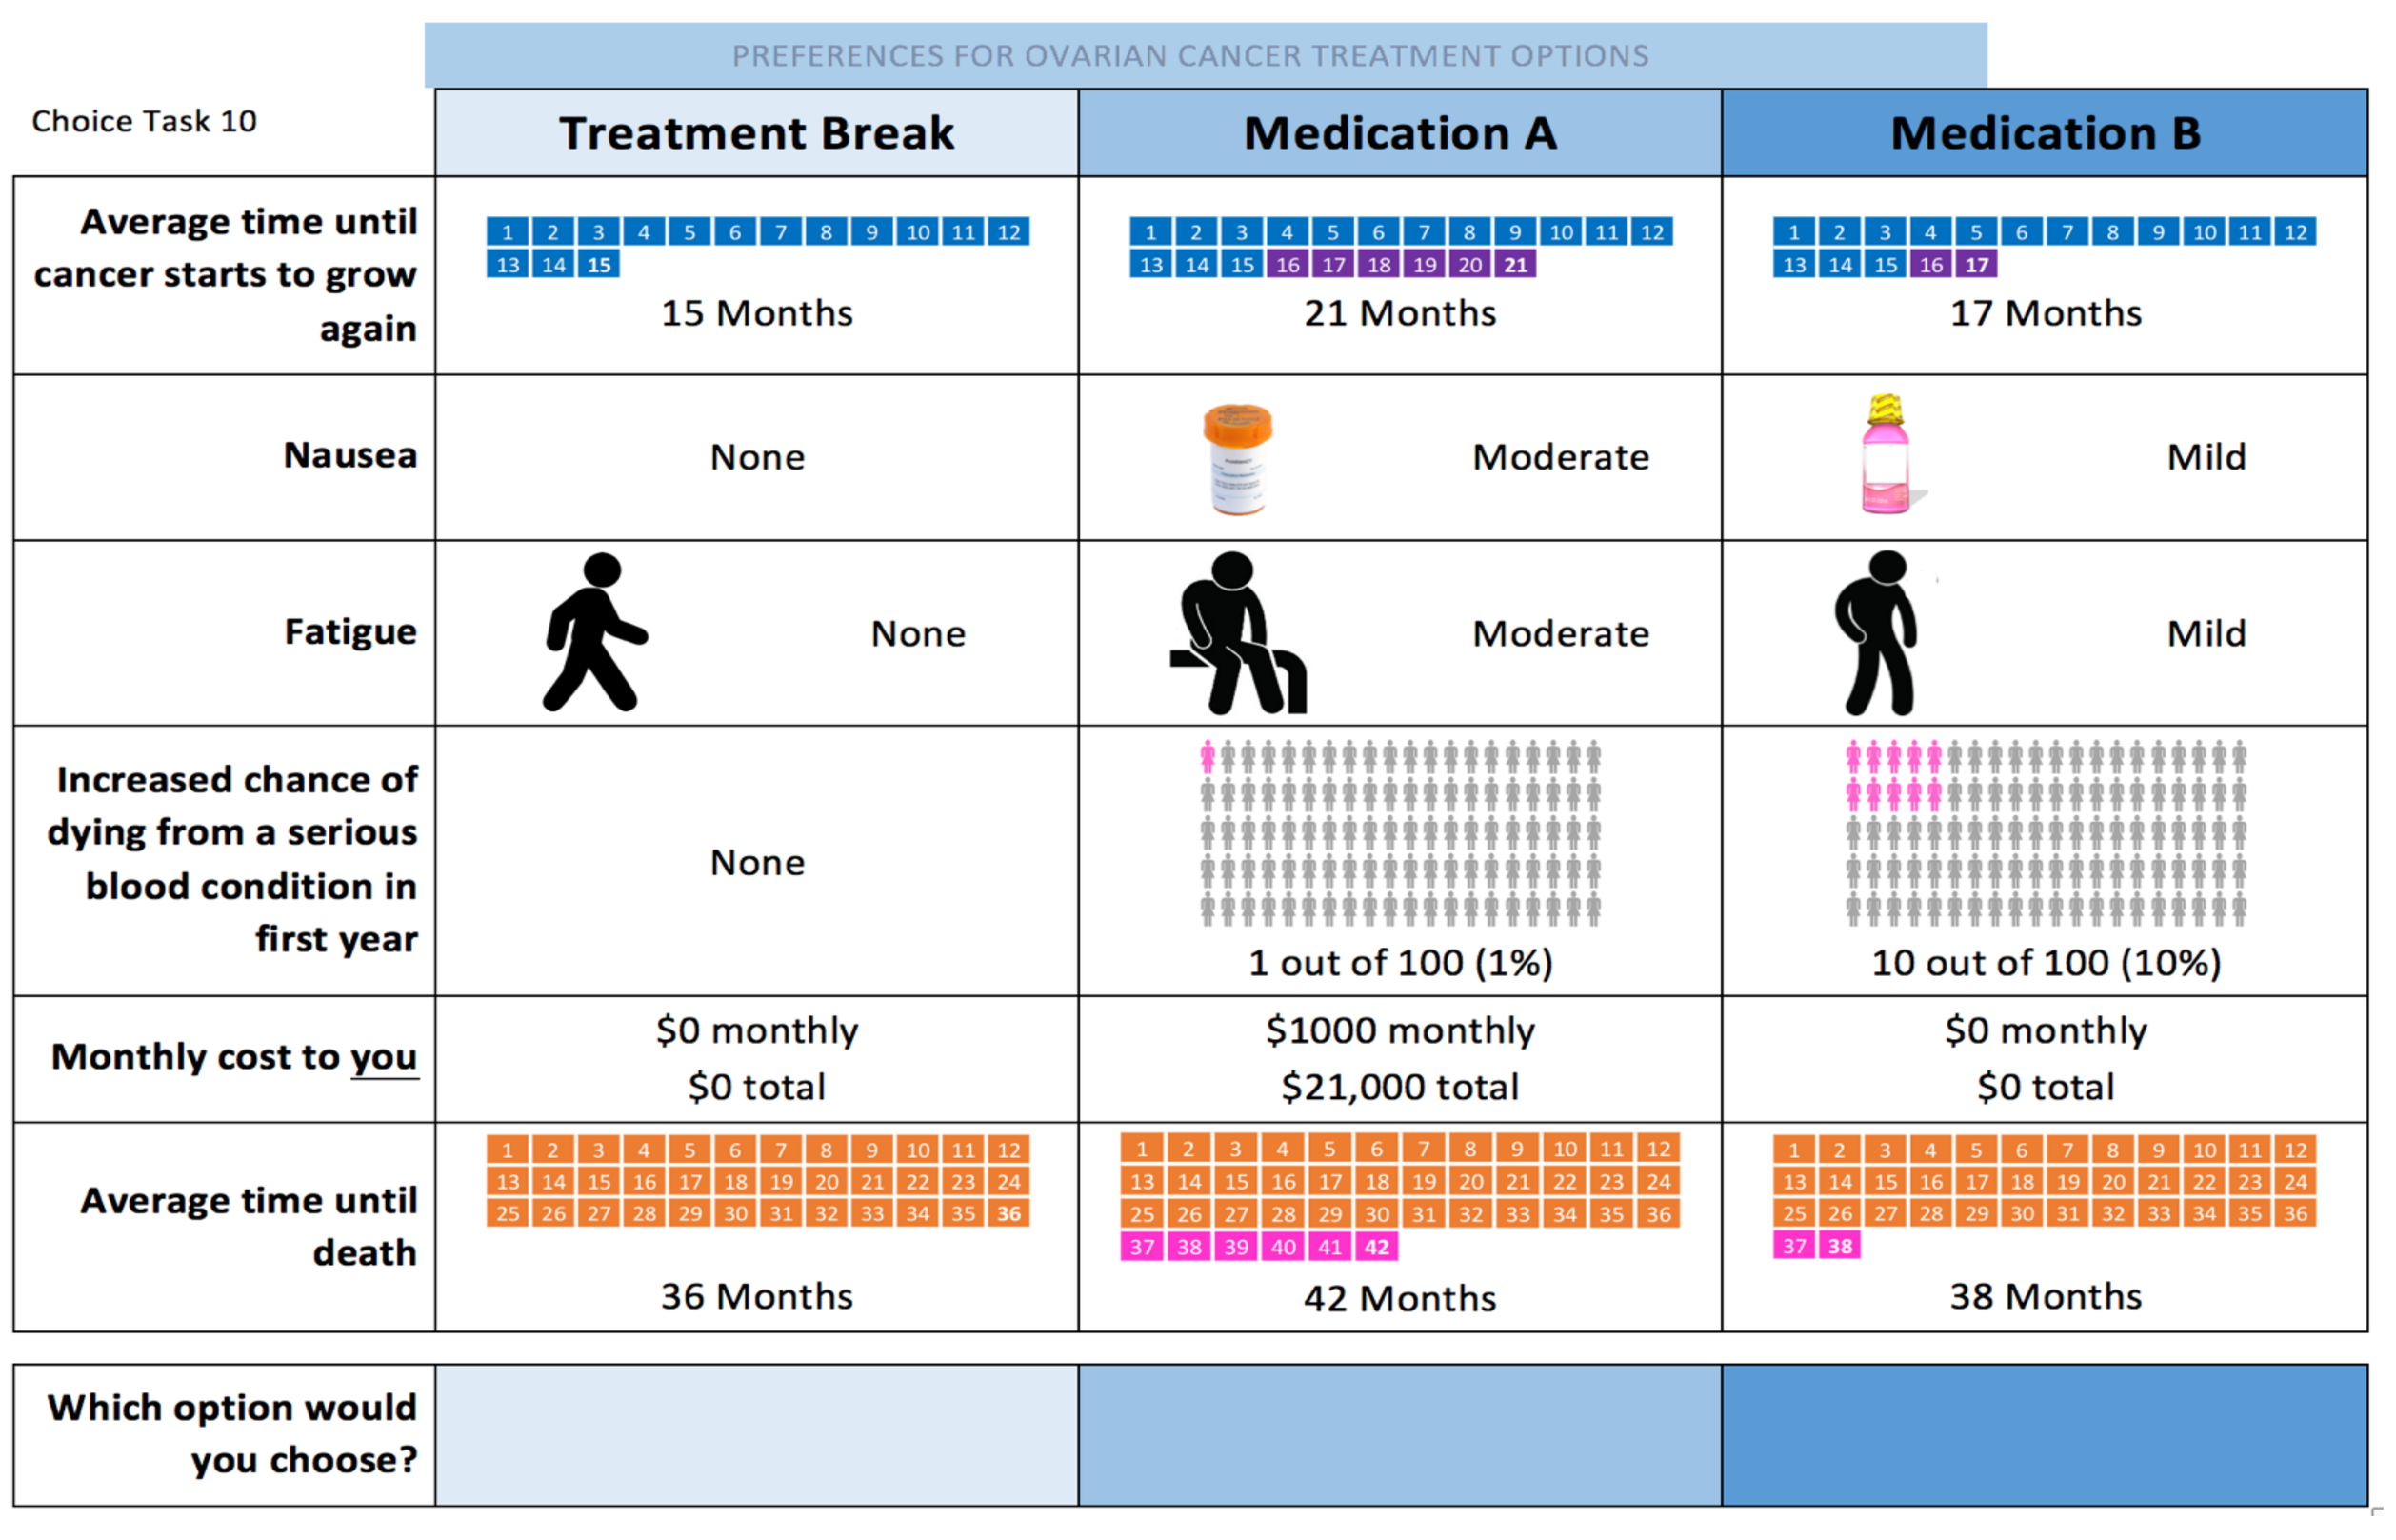
PREFERENCES FOR OVARIAN CANCER TREATMENT OPTIONS
Choice Task 10
Treatment Break
Medication A
Medication B
Average time until cancer starts to grow again
15 Months
21 Months
17 Months
Nausea
None
Moderate
Mild
Fatigue
None
Moderate
Mild
Increased chance of dying from a serious blood condition in first year
None
1 out of 100 (1%)
10 out of 100 (10%)
Monthly cost to you
$0 monthly
$0 total
$1000 monthly
$21,000 total
$0 monthly
$0 total
Average time until death
36 Months
42 Months
38 Months
Which option would you choose?

Figure 1. Example Choice Task.

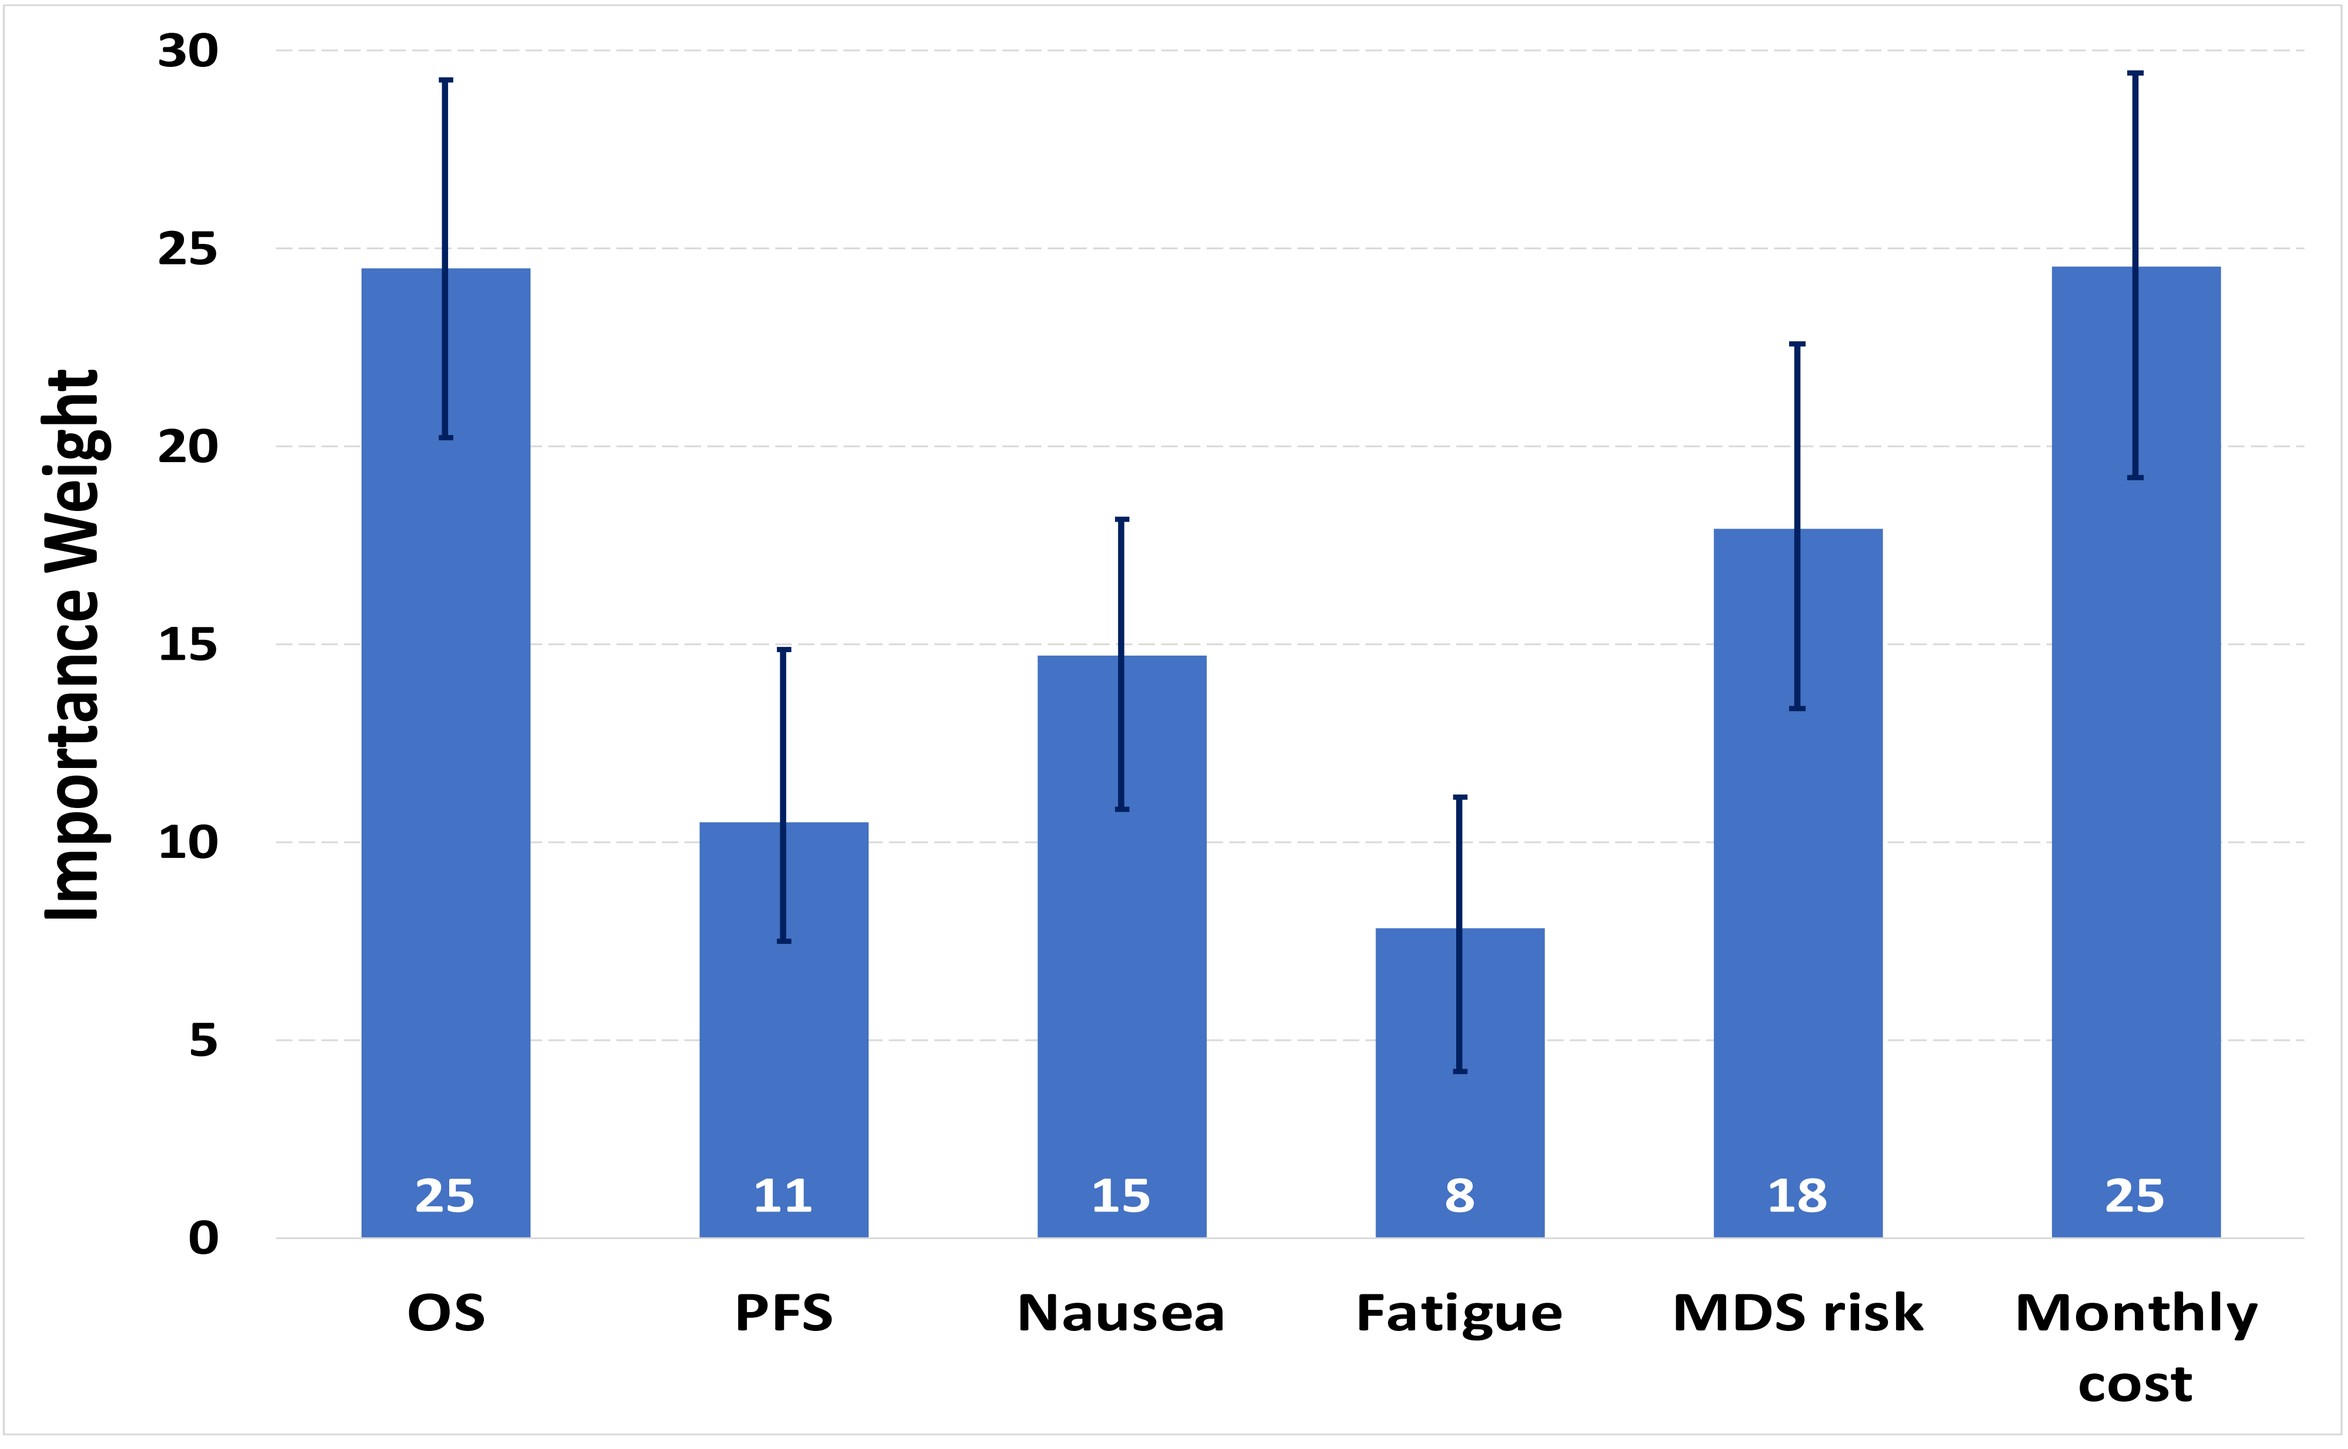
Importance Weight
30
25
20
15
10
5
0
25
11
15
8
18
25
OS
PFS
Nausea
Fatigue
MDS risk
Monthly cost

Figure 2. Preference Weights (sum=100)

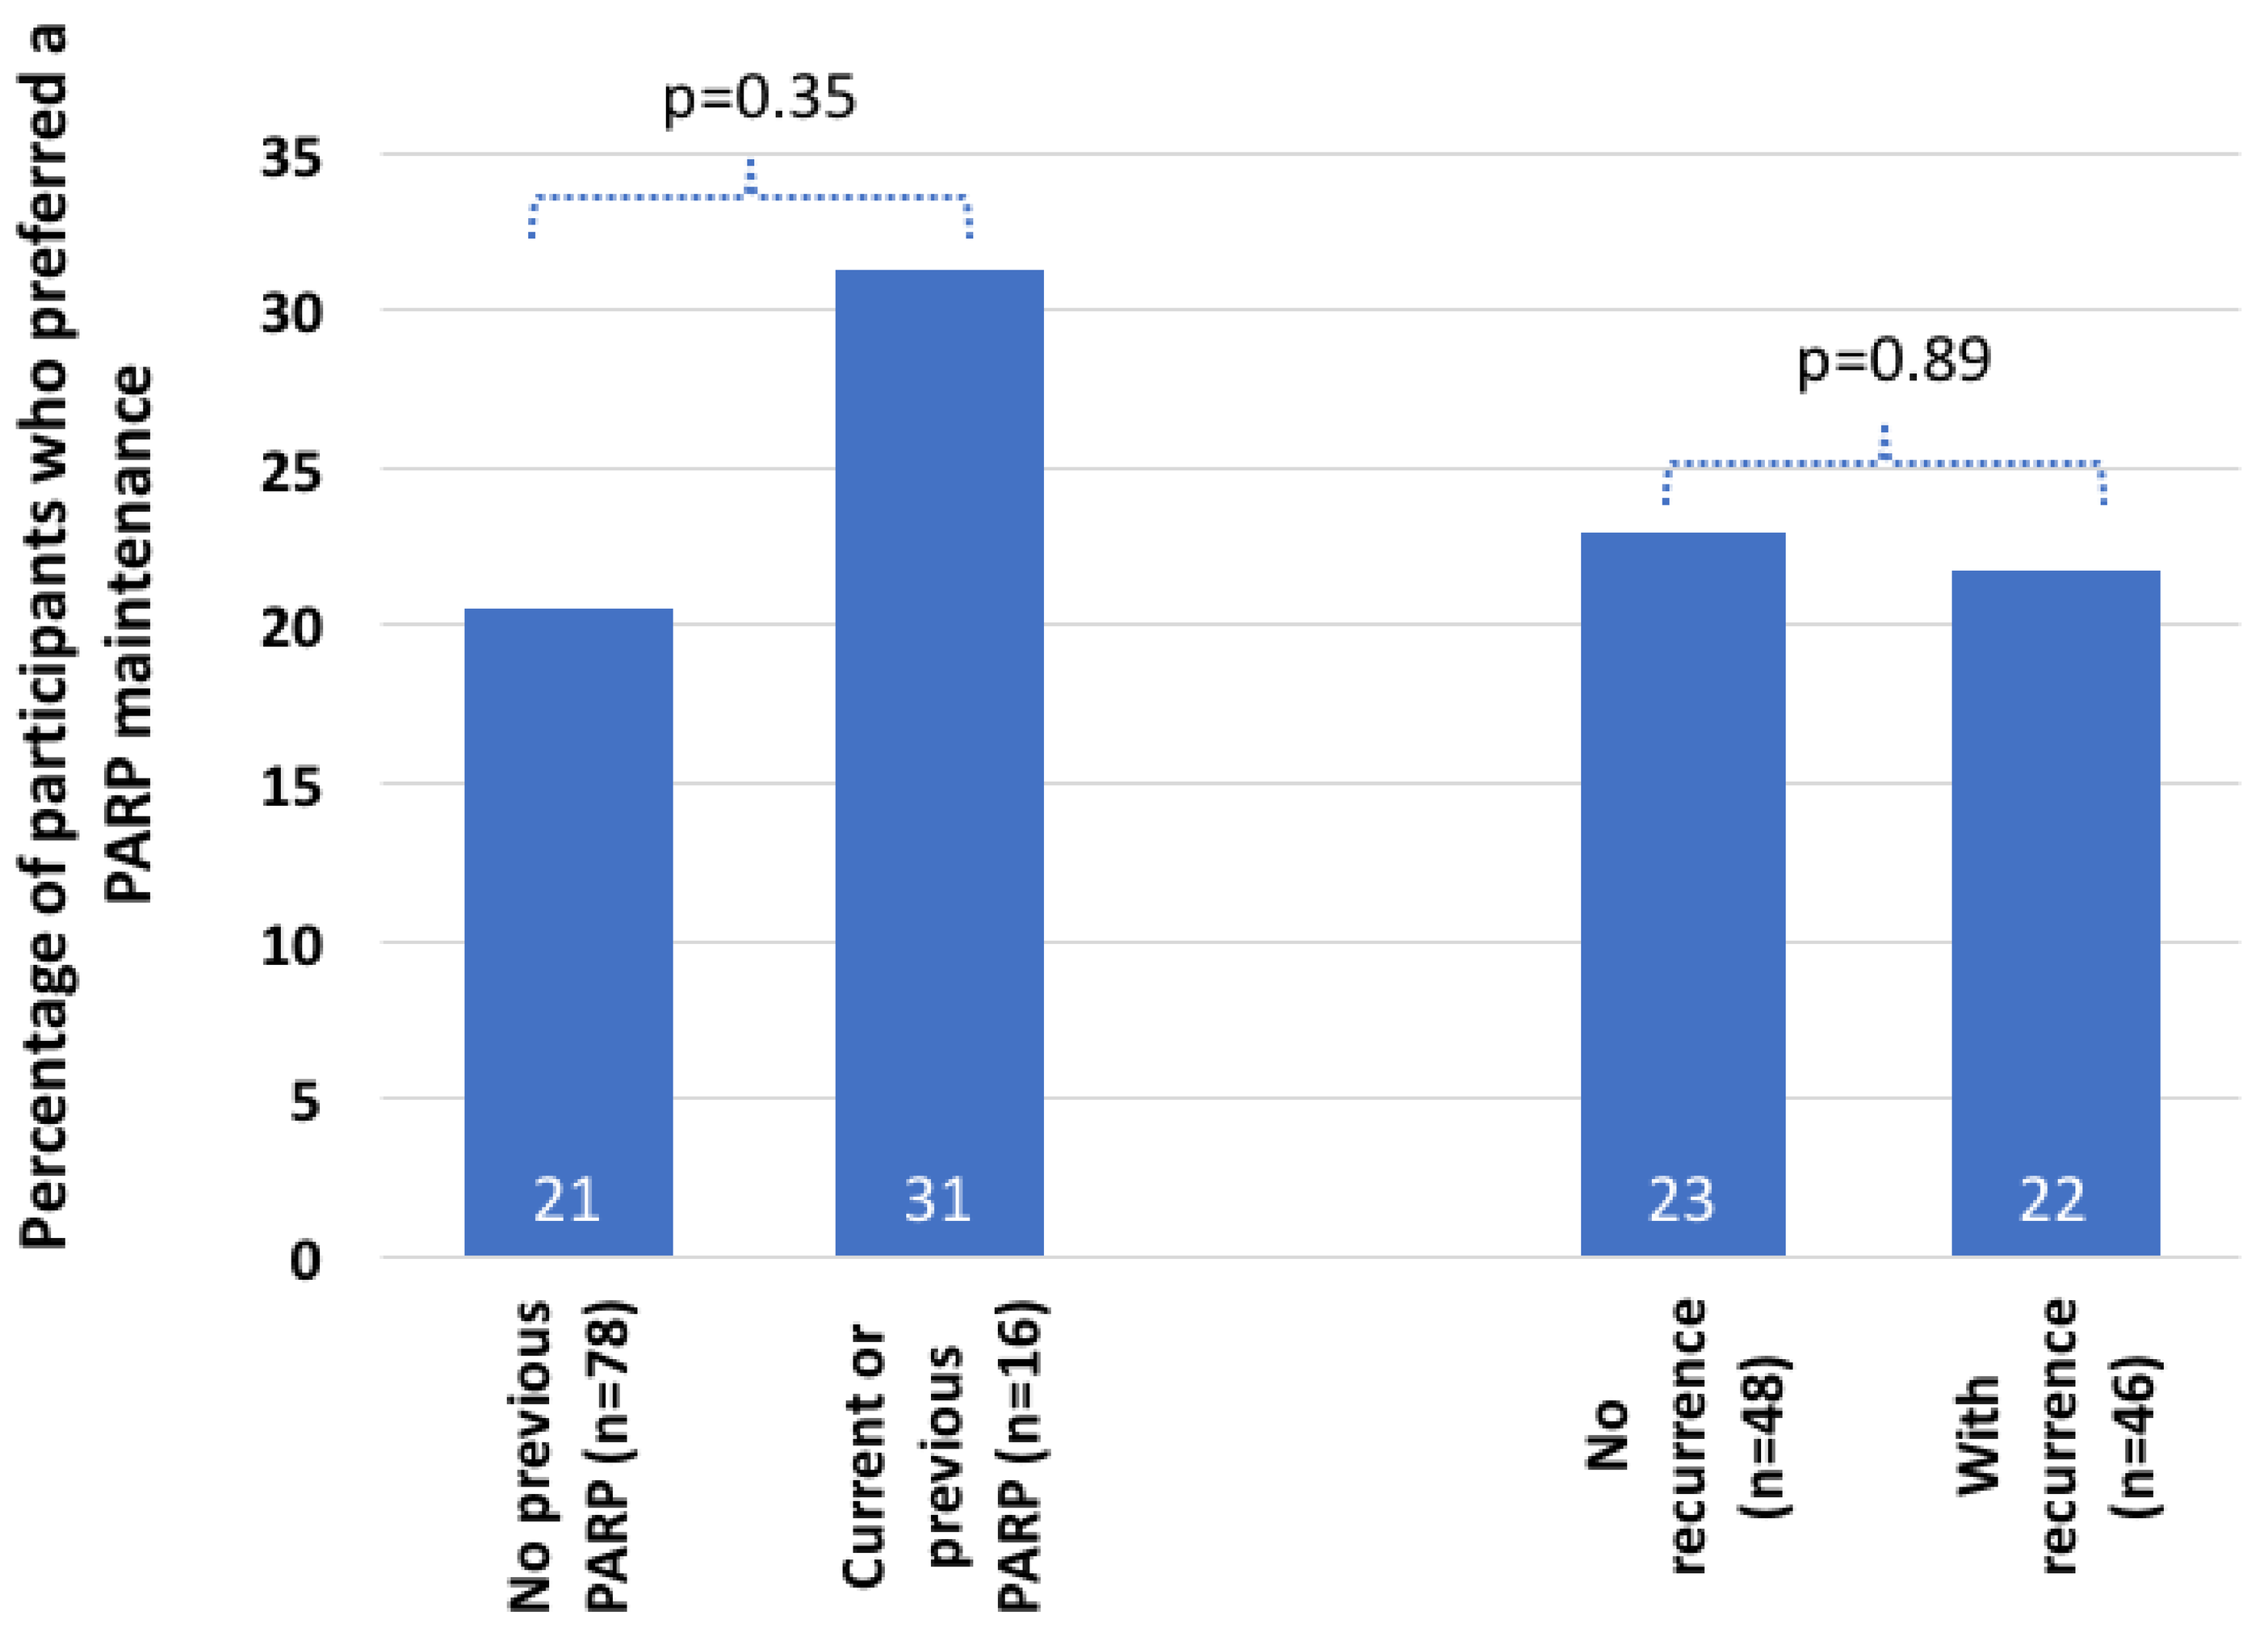
Percentage of participants who preferred a PARP maintenance
p=0.35
p=0.89
35
30
25
20
15
10
5
0
21
31
23
22
No previous PARP (n=78)
Current or previous PARP (n=16)
No recurrence (n=48)
With recurrence (n=46)

Figure 3. Percentage of participants who preferred 'PARP maintenance therapy' over 'Treatment break' in fixed choice scenario, by subgroup.

Treatment Outcome	Treatment benefit	From	To	MDS/AML Risk considered acceptable (95% CI)	Monthly cost considered acceptable (95% CI)
PFS (months)	2-month gain	15	17	1% (-4%, 6%)	$6 (-$40, $181)
	6-month gain	15	21	6% (4%, 10%)	$424 ($50, $702)
OS (months)	2-month gain	36	38	4% (2%, 7%)	$159 ($24, $516)
	6-month gain	36	42	13% (10%, 23%)	$998 ($771, $1,455)

Table 2. Risk and Cost Equivalents.

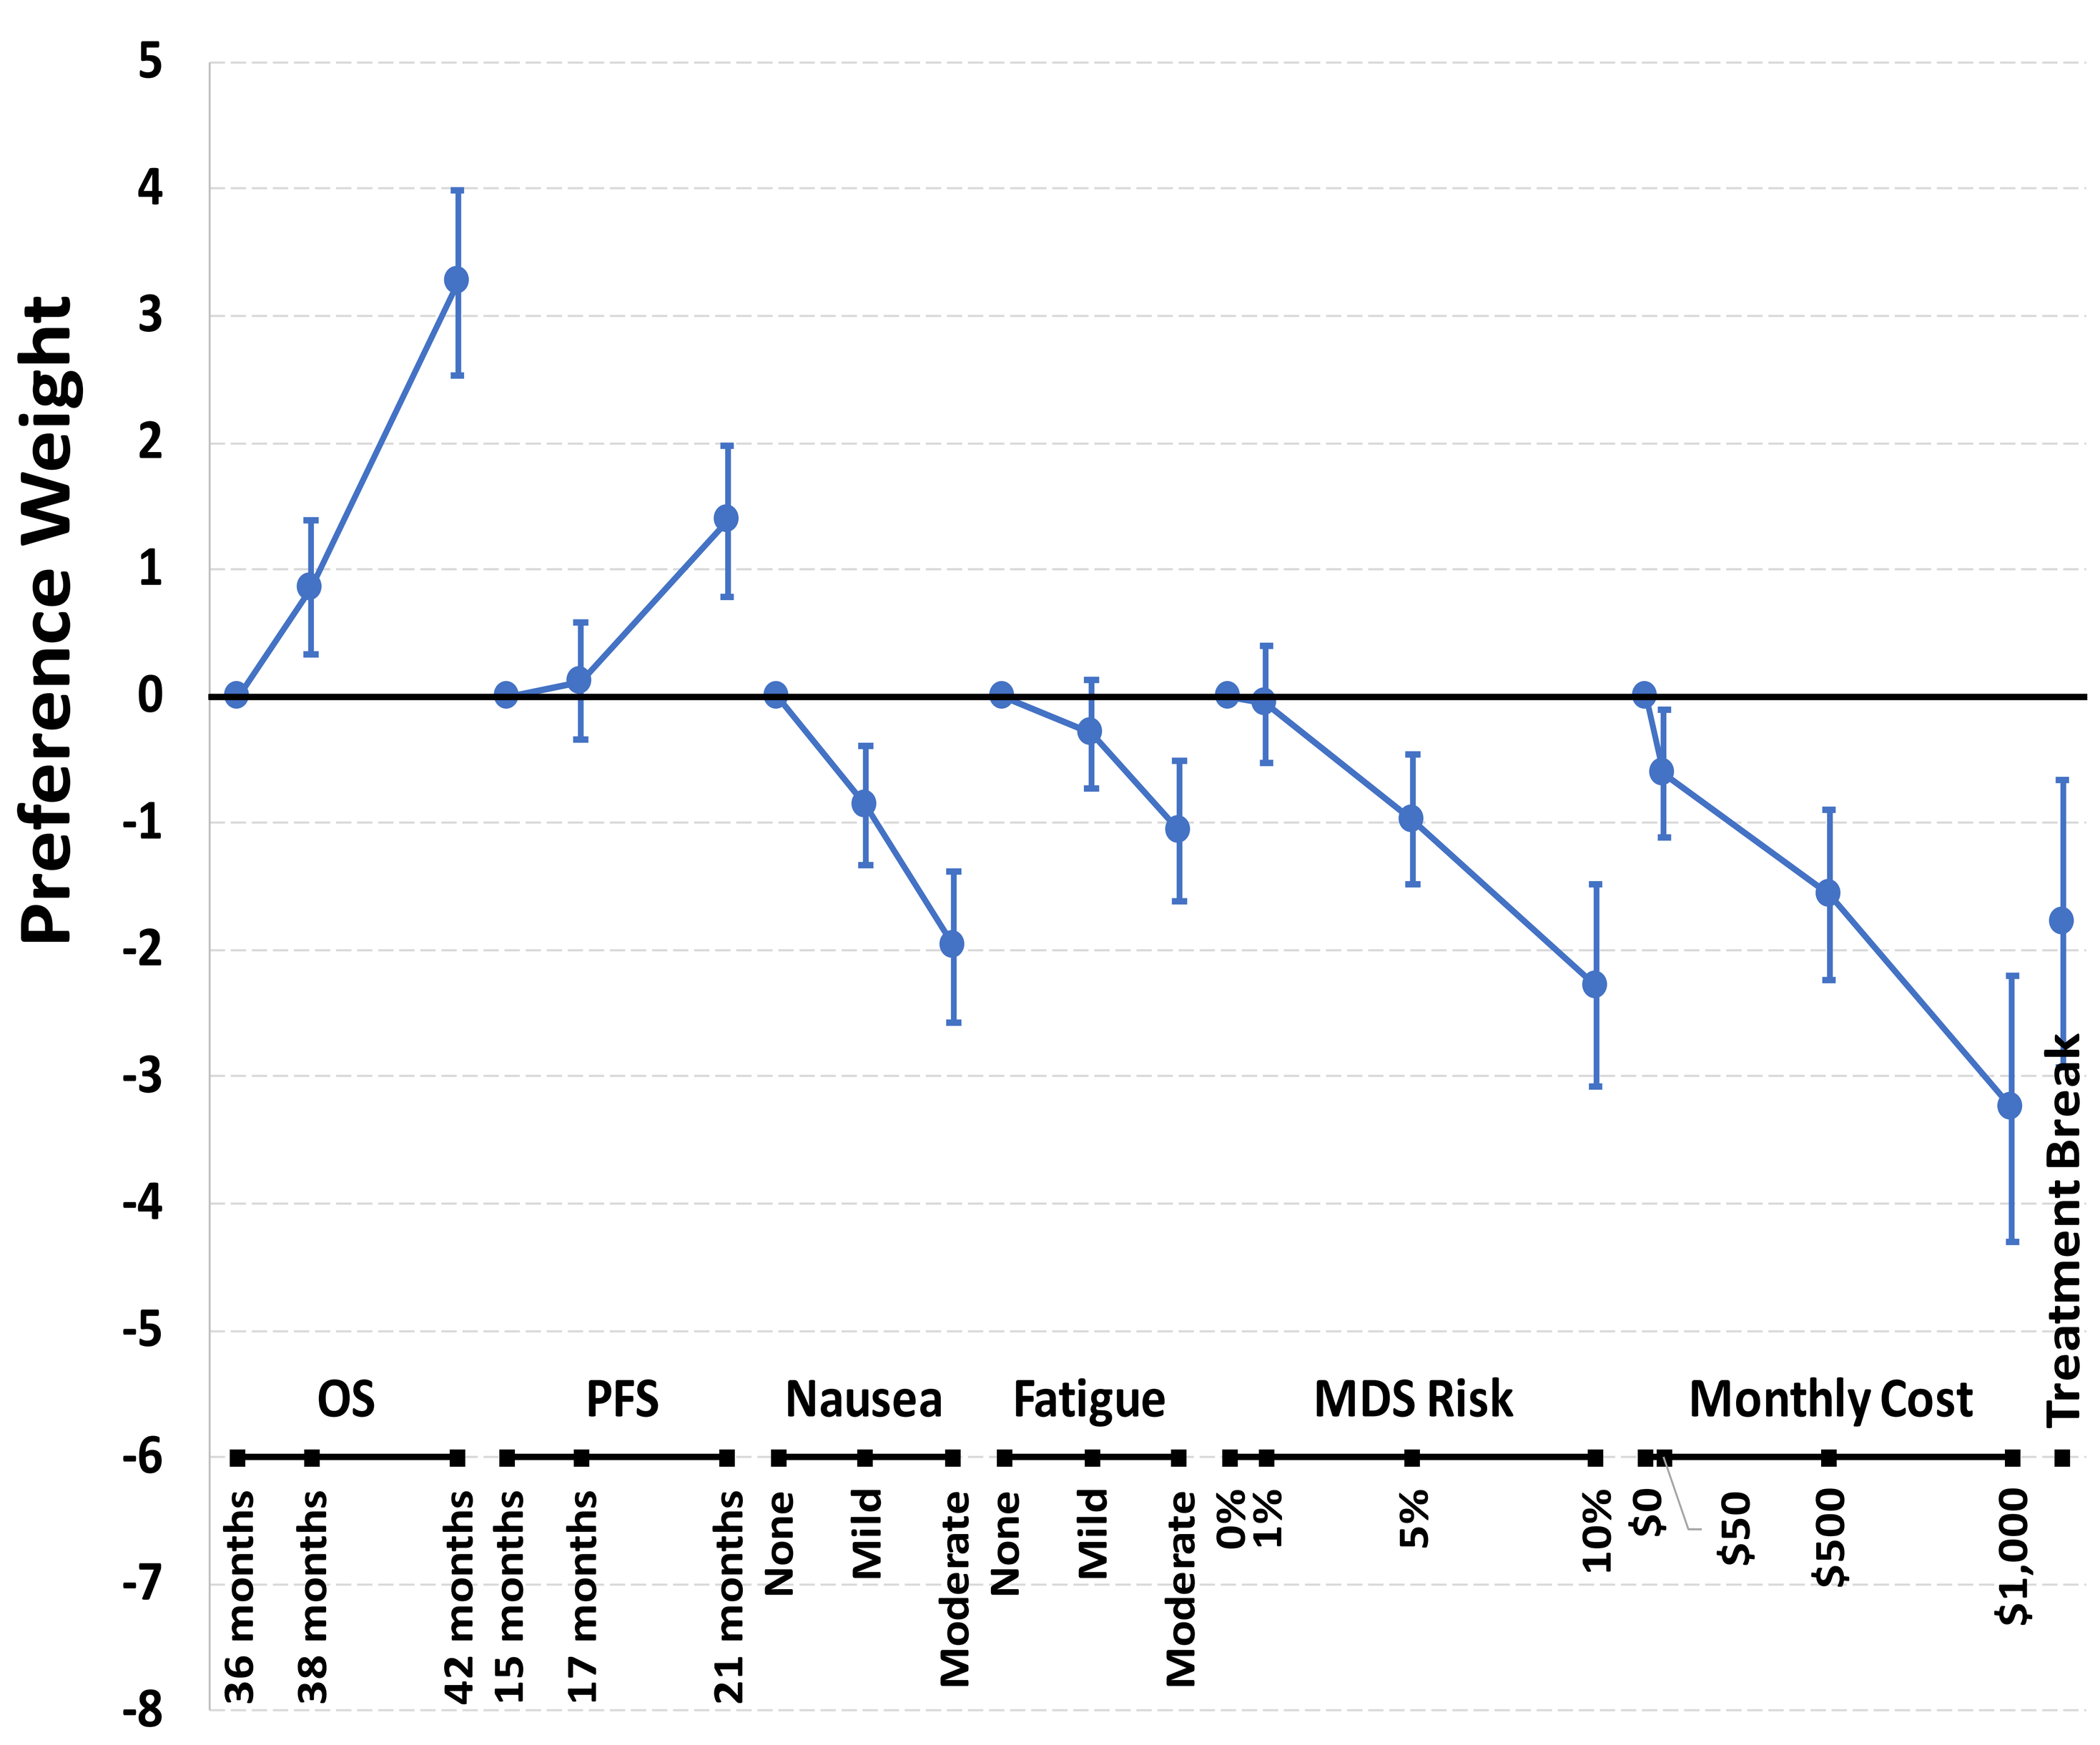
Preference Weight
5
4
3
2
1
0
-1
-2
-3
-4
-5
-6
-7
-8
OS
PFS
Nausea
Fatigue
MDS Risk
Monthly Cost
Treatment Break
36 months
38 months
42 months
15 months
17 months
21 months
None
Mild
Moderate
None
Mild
Moderate
0%
1%
5%
10%
$0
$50
$500
$1,000

Figure 4. Relative impact of attribute levels on treatment choice.

CONCLUSIONS

- Women with ovarian cancer are willing to accept side effects of maintenance PARP inhibitors and to pay some money out of pocket in exchange for modest OS gains or more substantial PFS gains.

REFERENCES

1. Burger, R.A., et al., *Incorporation of bevacizumab in the primary treatment of ovarian cancer.* N Engl J Med, 2011. 365(26): p. 2473-83.
2. Ledermann, J., et al., *Olaparib maintenance therapy in platinum-sensitive relapsed ovarian cancer.* N Engl J Med, 2012. 366(15): p. 1382-92.
3. Perren, T.J., et al., *A phase 3 trial of bevacizumab in ovarian cancer.* N Engl J Med, 2011. 365(26): p. 2484-96.
4. Ryan M, Farrar S. *Using conjoint analysis to elicit preferences for health care*. BMJ. Jun 3 2000;320(7248):1530-1533.
5. Bridges JF, Mohamed AF, Finnern HW, Woehl A, Hauber AB. *Patients' preferences for treatment outcomes for advanced non-small cell lung cancer: a conjoint analysis.* Lung Cancer. Jul 2012;77(1):224-231.

ACKNOWLEDGEMENTS

- This study was funded by an investigator-initiated research grant from Astra Zeneca (PI: Laura Havrilesky and Shelby Reed).
- The authors acknowledge Dr. Sue Friedman and Facing Our Risk of Cancer Empowered for assisting the study team with accrual.