PCN55

Network Meta-Analysis Comparing Palbociclib Plus Fulvestrant With Everolimus Plus Exemestane For HR+/HER2- Advanced Breast Cancer That Has Become Resistant To Previous Endocrine Therapy

LeReun C[1], Gentilini A[2], Large S[3], Chowdhury M[3], Smith TA[4], Rinciog C[2]

[1]Independent Biostatistician, Sainte-Anne, Guadeloupe, France; [2]Symmetron Limited, London, UK; [3]Pfizer Limited, Walton Oaks, UK; [4]Pfizer Inc, New York, USA

Objective

To indirectly estimate comparative OS between PAL+FUL and EVE+EXE.

Conclusion

The NMAs found that PAL+FUL was associated with increased OS compared to EVE+EXE.

Context

PAL+FUL and EVE+EXE are two treatments licensed for the treatment of breast cancer resistant to prior endocrine therapy.

Introduction

- Fulvestrant (FUL) and everolimus plus exemestane (EVE+EXE) are licensed therapies for advanced breast cancer resistant to prior endocrine therapy.[1,2] EVE+EXE was found to be superior in terms of overall survival (OS) compared to exemestane alone in the phase 3 BOLERO-2 trial (hazard ratio [HR]: 0.89; 95% confidence interval [CI]: 0.73, 1.10).[3] Palbociclib is a first-in-class selective small-molecule inhibitor of the cyclin dependent kinases (CDKs) types 4 and 6, which play a role in proliferation of breast cancer cells. A phase 3 trial, PALOMA-3, found that palbociclib plus fulvestrant (PAL+FUL) was associated with clinically meaningful improvements in OS versus FUL (HR: 0.81; 95% CI: 0.64, 1.03).[4] PAL+FUL is now also licensed in breast cancer resistant to prior endocrine therapy,[5] hence it is of interest to determine the comparative effectiveness of PAL+FUL versus EVE+EXE.

Methods

- An existing systematic literature review (SLR) was updated to identify all relevant published clinical data of pre/peri/post-menopausal women with HR+, HER2- locally advanced or metastatic breast cancer receiving first- or second-line therapy and who have been exposed to prior endocrine therapy, either in the (neo)adjuvant or advanced/metastatic setting.[6]
- Searches were performed in EMBASE, MEDLINE, Medline In-Process, the Cochrane Library and relevant conference proceedings, published up to February 2019. The SLR was performed in accordance with standard guidelines.[7,8]
- The OS proportional hazards assumption was assessed using log cumulative hazard and Schoenfeld residual plots for all studies in the network.
- Two NMA methodologies were utilised to conduct analyses using a network of studies linking OS evidence from PAL+FUL to EVE+EXE:
 1. **Proportional hazards (PH) NMA** which assumes constant treatment effects over time.
 2. **Fractional polynomial (FP) NMA** was used to capture a multi dimensional treatment effect and does not require the PH assumption to be valid.
- The PH NMA was conducted using Bayesian methods outlined in NICE Decision Support Unit guidelines.[9] Fixed-effects (FE) and random effects (RE) models were considered; the analysis utilised hazard ratios (HRs) as inputs.
- The FP NMA was conducted using methodology reported in Jansen (2011).[10] First- and second-order FE models were fitted to the data representing all possible combinations of powers from the following set: –2.0, –1.0, –0.5, 0.0, 0.5, 1.0, 2.0, 3.0. The model with the lowest DIC was selected as the best-fitting model. Other models with a DIC within 5 points of the best model were also considered as possible candidates. The OS projections obtained from suitable candidates were also checked visually for clinical validity and were compared against Kaplan Meier OS observations.
- Both NMA methodologies were implemented in WinBUGS (version 1.4.3).

Results

- The SLR was conducted in February 2019; 142 publications reporting on 94 unique studies were ultimately found to be relevant. Four of these studies (BOLERO-2,[3] CONFIRM,[11] PALOMA-3[4] and SoFEA[12]) were found feasible for inclusion in the OS NMAs (**Figure 1**). A further study, EFECT,[13] was included after completion of the SLR using 2007 conference data. These data were not identified in the SLR as only conference proceedings up to three years prior to the SLR were hand-searched. EFECT was identified once the updated SLR was cross-checked against another SLR from NICE appraisal TA593[14] in a similar population.

Figure 1. Network plot

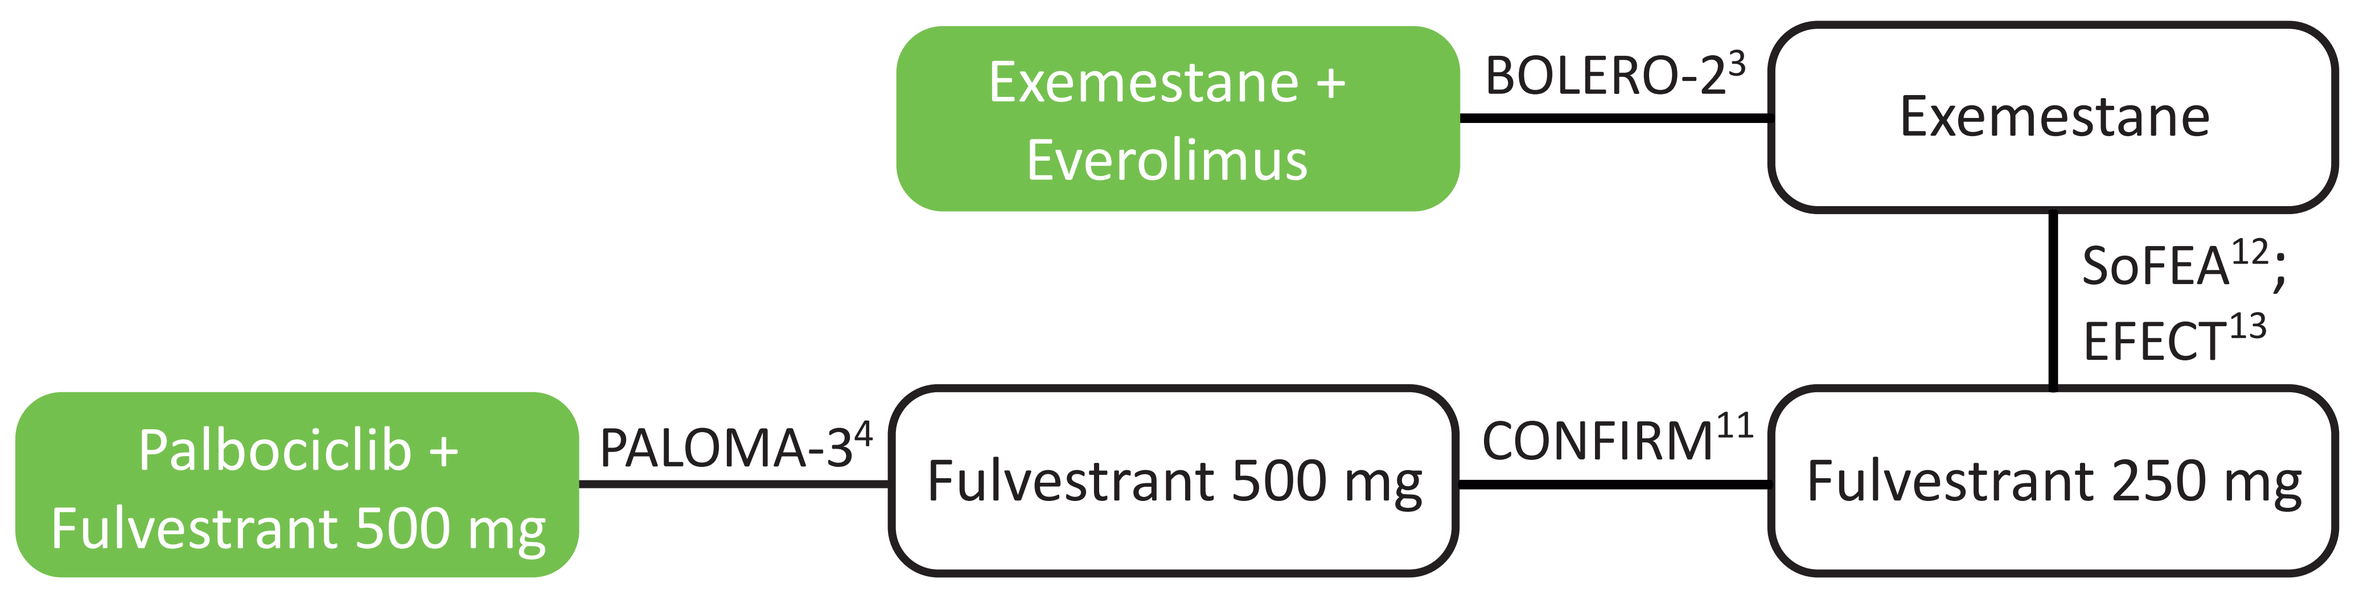
Exemestane + Everolimus
BOLERO-2[3]
Exemestane
SoFEA[12]; EFECT[13]
Palbociclib + Fulvestrant 500 mg
PALOMA-3[4]
Fulvestrant 500 mg
CONFIRM[11]
Fulvestrant 250 mg

Comparison of interest highlighted in green

- Key patient characteristics were found to be similar across trials in the feasibility assessment.
- The PH assumption was met for three of the five studies, including PALOMA-3. The PH assumption was violated for two studies (BOLERO-2 and EFECT); log cumulative hazard plots of the treatment arms were not parallel and produced significant p-values (p<0.05) when considering the Schoenfeld residuals (**Figure 2**).

Figure 2. Diagnostic plots showing proportional hazards assumption violations for two studies

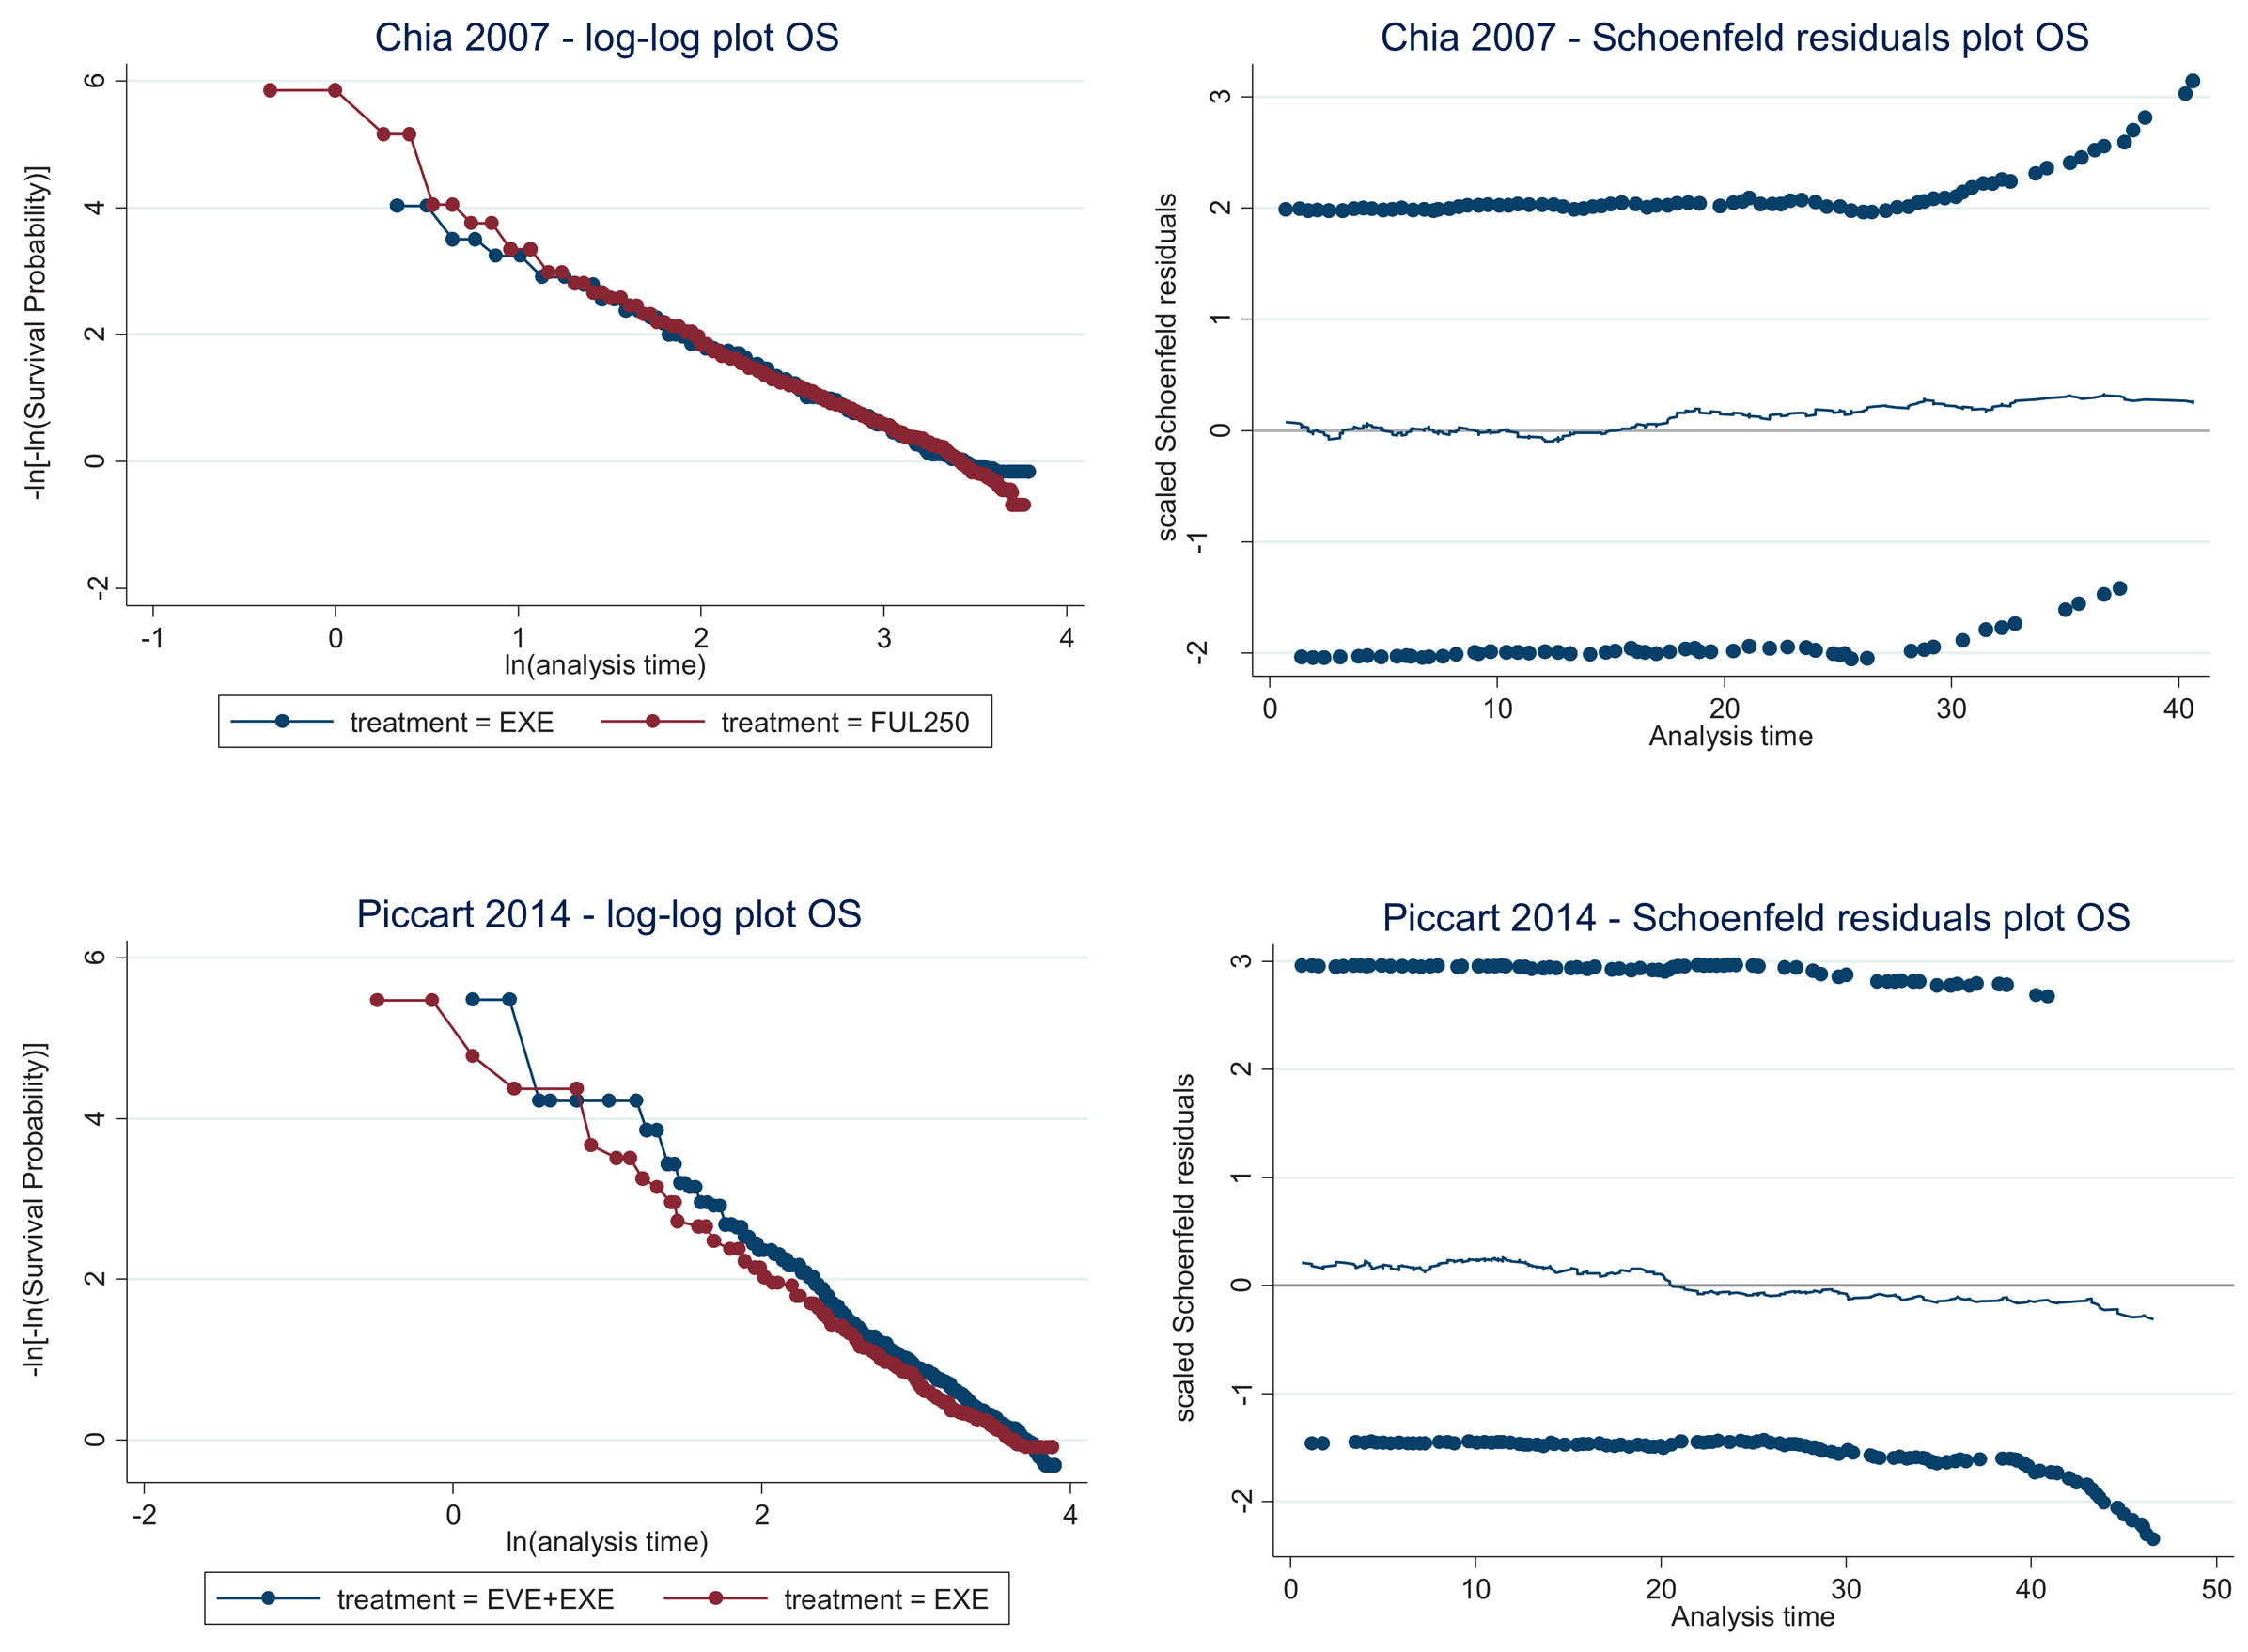
Chia 2007 - log-log plot OS
-ln[-ln(Survival Probability)]
ln(analysis time)
treatment = EXE
treatment = FUL250
Chia 2007 - Schoenfeld residuals plot OS
scaled Schoenfeld residuals
Analysis time
Piccart 2014 - log-log plot OS
treatment = EVE+EXE
treatment = EXE
Piccart 2014 - Schoenfeld residuals plot OS

Schoenfeld residuals p-value: p=0.007 (Chia 2007 [EFECT]) and p=0.001 (Piccart 2014 [BOLERO-2])

- RE models were not run in the PH-based or FP-based NMAs as the majority of the connections between interventions were informed by single studies in the OS network, and the ability to reliably estimate between-study variance was very limited. The RE NMAs would therefore carry a lot of uncertainty and give results that are not clinically plausible due to extremely wide credible intervals.

PH-based model

- The results of the PH-based NMA showed an OS benefit for PAL+FUL over EVE+EXE with a median HR of 0.74 (95% credible interval [CrI]: 0.51, 1.09).

FP-based model

- Two models met the selection criteria. The best-fitting model was the second-order FE model with powers 0 and 0.5 (DIC: 2333.47), while the second best-fitting model was the second-order FE model with powers 0.5 and –0.5 (DIC: 2337.39).
- In the best-fitting model, the median HR decreased from 0.87 at 6 months (95% CrI: 0.44, 1.74) to 0.43 at 60 months (95% CrI: 0.11, 1.59), as shown in **Figure 3**. The second best-fitting model produced very similar results (**Table 1**); the median HR decreased from 0.88 at 6 months (95% CrI: 0.45, 1.70) to 0.42 at 60 months (95% CrI: 0.13, 1.39).

Figure 3. Median HR (PAL+FUL versus EVE+EXE) and 95% credible interval over time for best FP-based model

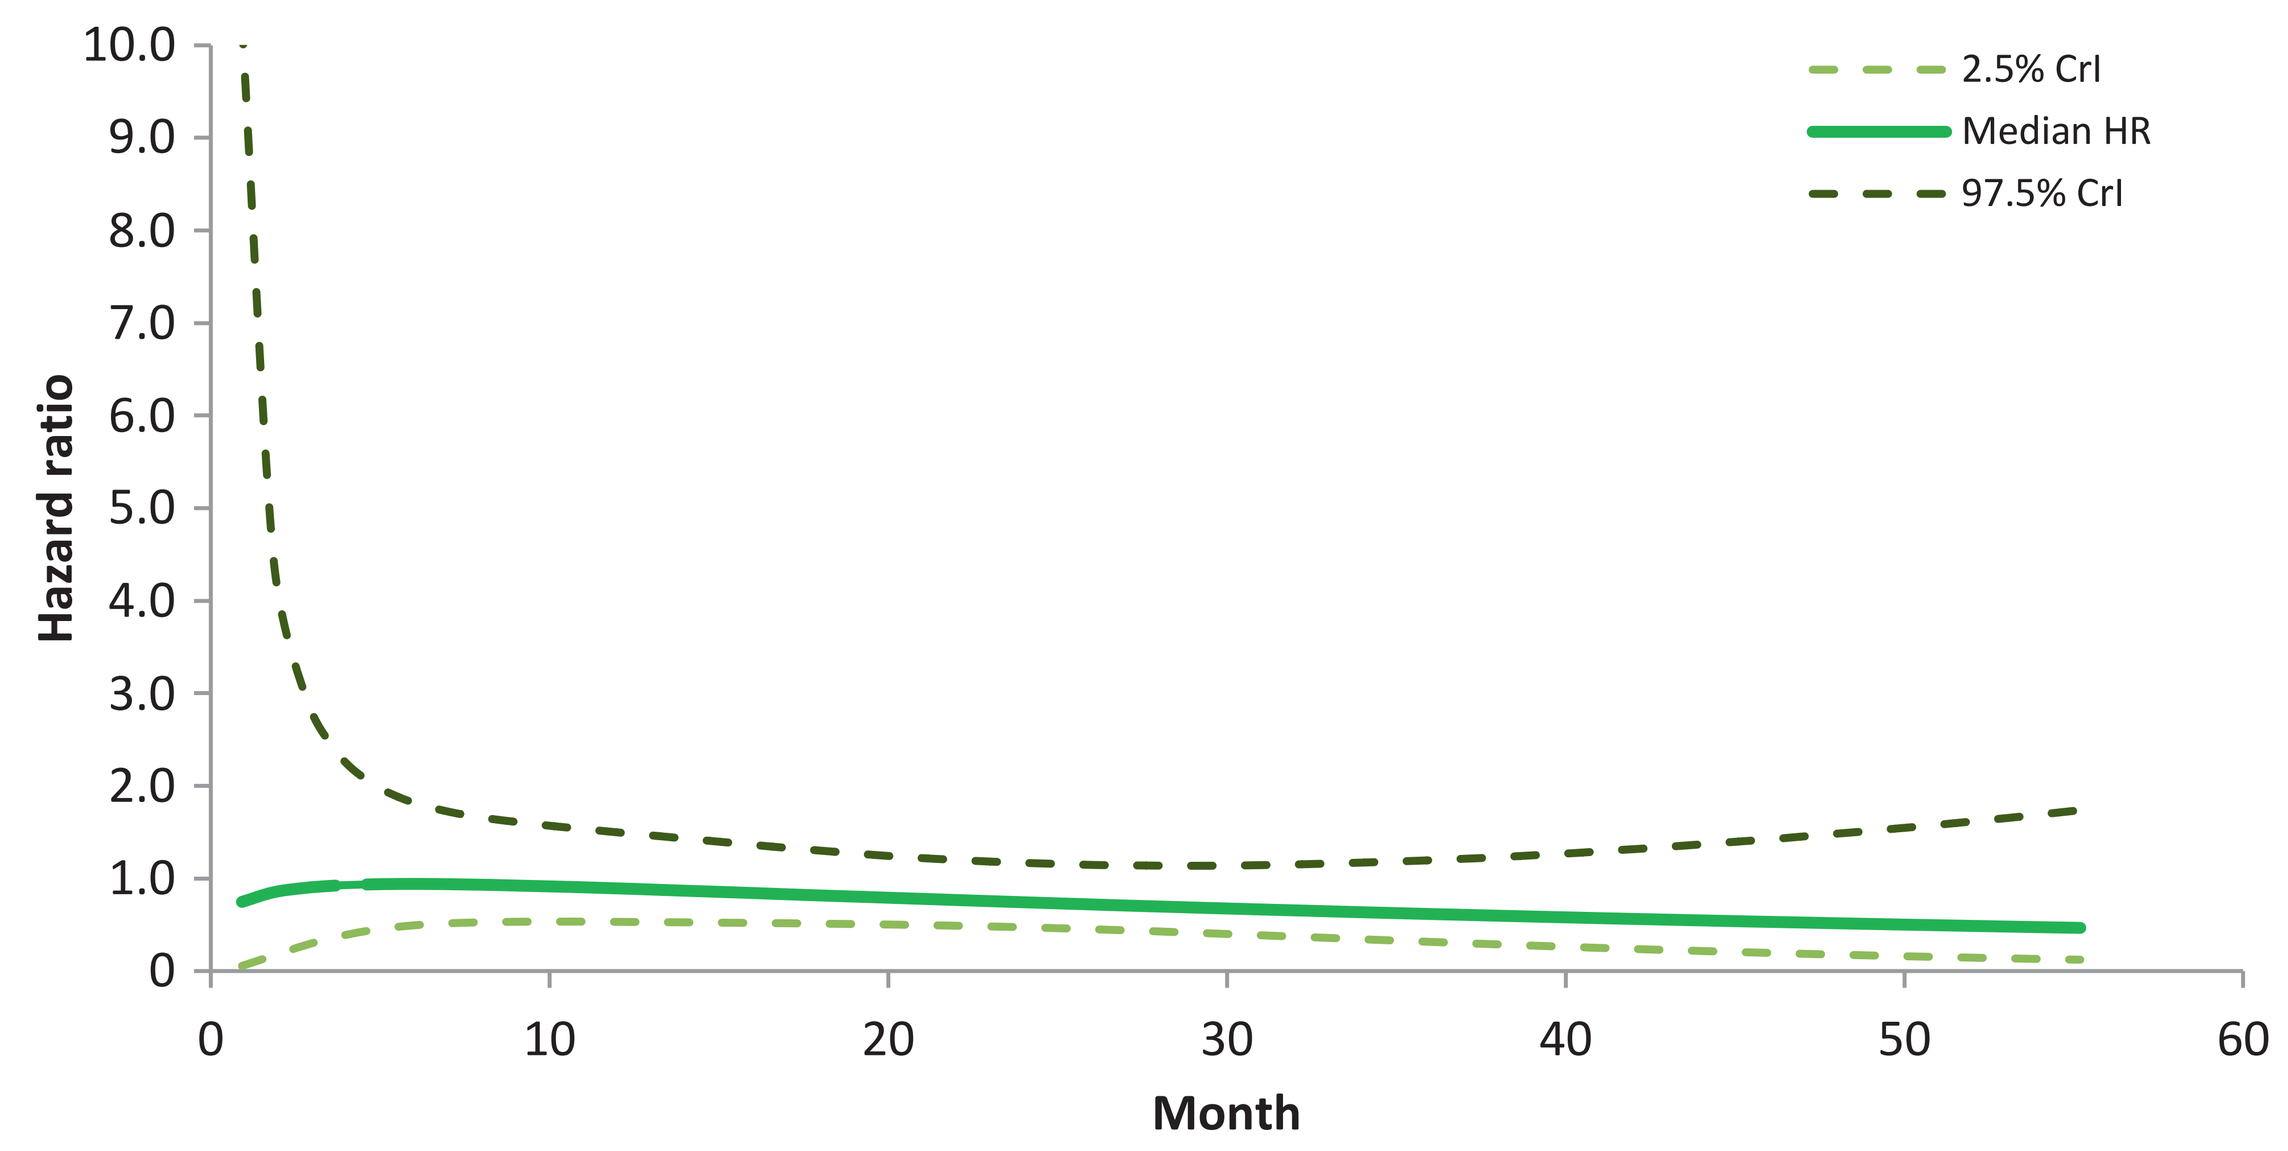
2.5% CrI
Median HR
97.5% CrI
Hazard ratio
Month

Abbreviations: CrI, credible interval; HR, hazard ratio

Table 1. FP-based NMA results (PAL+FUL vs. EVE+EXE)

Time (months)	Median HR (95% CrI)	
	Model 1 (powers 0 and 0.5)	Model 2 (powers -0.5 and 0.5)
6	0.87 (0.44, 1.74)	0.88 (0.45, 1.70)
12	0.83 (0.49, 1.42)	0.84 (0.49, 1.46)
24	0.70 (0.45, 1.11)	0.70 (0.46, 1.08)
36	0.59 (0.33, 1.07)	0.58 (0.32, 1.05)
48	0.50 (0.20, 1.26)	0.49 (0.20, 1.18)
60	0.43 (0.11, 1.59)	0.42 (0.13, 1.39)

Abbreviations: CrI, credible interval; EVE+EXE, everolimus plus exemestane; HR, hazard ratio; PAL+FUL, palbociclib plus fulvestrant

- The FP-based survival curves produced by the best-fitting model closely matched the corresponding Kaplan-Meier curves for PAL+FUL and EVE+EXE, as shown in **Figure 4**.

Figure 4. Plot of FP-based survival curves along with Kaplan-Meier curves for PAL+FUL and EVE+EXE

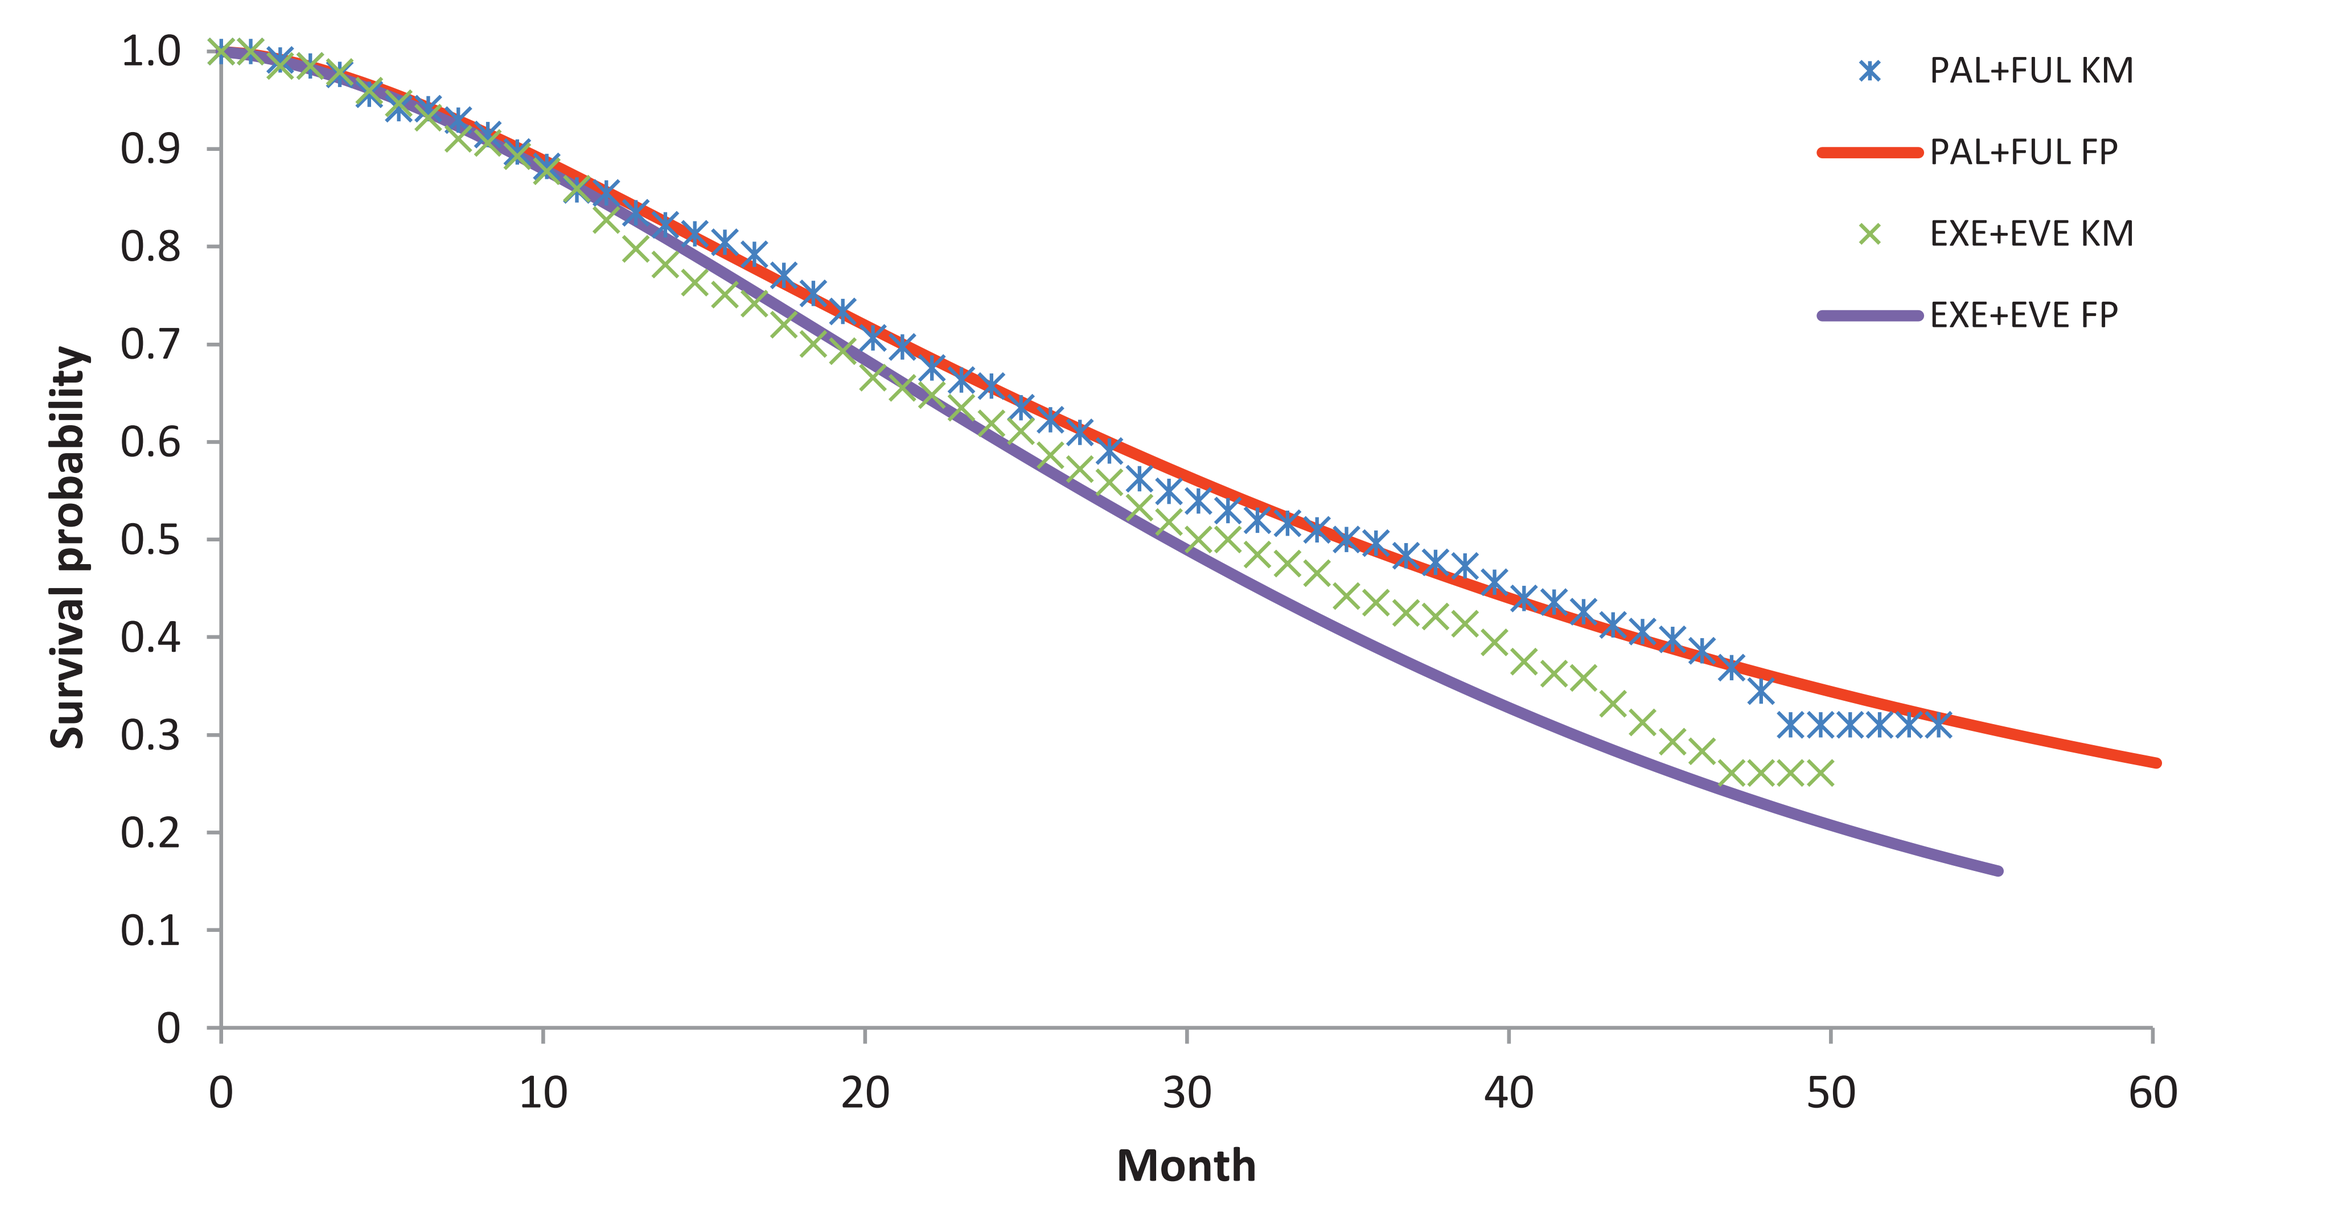
PAL+FUL KM
PAL+FUL FP
EXE+EVE KM
EXE+EVE FP
Survival probability
Month

Abbreviations: EVE+EXE, everolimus plus exemestane; FP, fractional polynomial; KM, Kaplan-Meier; PAL+FUL, palbociclib plus fulvestrant

- The results of the PH-based NMA are similar to the FP-based NMA at 24 months (difference in median HR: 0.04). However, the 6-month and 12-month median HR estimates from the FP-based NMA are higher with larger credible intervals.

Discussion and Conclusion

- Both the FP-based NMA and PH-based NMA found PAL+FUL was associated with increased OS compared to EVE+EXE.
- It was possible to compare PAL+FUL to EVE+EXE indirectly via three additional treatments in a small network using results from studies identified in the SLR.
- The PH assumption did not hold for two of the five studies in the network, suggesting that the FP-based NMA results may be more reliable and valid than those produced by the PH-based NMA approach.
- The results of the NMAs are consistent with the Kaplan-Meier curves for both PAL+FUL and EVE+EXE, and PAL+FUL is observed to have a greater survival probability over time compared to EVE+EXE.
- Lack of statistical significance may be expected due to studies not being powered to detect differences in OS, between-trial variance and the small number of studies in the NMA network.

Electronic Poster

An electronic version of this poster may be obtained by scanning this QR code with your smartphone app. Copies of this poster obtained through the QR Code are for personal use only and may not be reproduced without permission from the author of this poster.

References:

1. Faslodex - Summary of Product Characteristics (SPC), accessed October 2019.
2. Afinitor - Summary of Product Characteristics (SPC), accessed October 2019
3. Piccart M, Hortobagyi GN, Campone M, et al. Everolimus plus exemestane for hormone-receptor-positive, human epidermal growth factor receptor-2-negative advanced breast cancer: overall survival results from BOLERO-2. Annals of oncology 2014;25:2357-2362.
4. Turner NC, Slamon DJ, Ro J, et al. Overall survival with palbociclib and fulvestrant in advanced breast cancer. New England Journal of Medicine 2018;379:1926-1936.
5. IBRANCE - Summary of Product Characteristics (SPC), accessed October 2019
6. Wilson FR VA, Mitra D, Cameron C, Iyer S. Systematic review and network meta-analysis comparing palbociclib with chemotherapy agents for the treatment of postmenopausal women with HR-positive and HER2-negative advanced/metastatic breast cancer. Breast cancer research and treatment 2017;166:167-77.
7. Moher D, Liberati A, Tetzlaff J, et al. Preferred reporting items for systematic reviews and meta-analyses: the PRISMA statement. BMJ 2009;339:b2535.
8. Centre for Reviews and Dissemination. Systematic reviews: CRD's guidance for undertaking reviews in health care. In: York Uo, ed. York, 2009.
9. Dias S, Welton NJ, Sutton AJ, et al. NICE DSU technical support document 2: a generalised linear modelling framework for pairwise and network meta-analysis of randomised controlled trials. 2011.
10. Jansen JP. Network meta-analysis of survival data with fractional polynomials. BMC medical research methodology 2011;11:61.
11. Di Leo A, Jerusalem G, Petruzelka L, et al. Final overall survival: fulvestrant 500 mg vs 250 mg in the randomized CONFIRM trial. Journal of the National Cancer Institute 2014;106:djt337-djt337.
12. Johnston SR, Kilburn LS, Ellis P, et al. Fulvestrant plus anastrozole or placebo versus exemestane alone after progression on non-steroidal aromatase inhibitors in postmenopausal patients with hormone-receptor-positive locally advanced or metastatic breast cancer (SoFEA): a composite, multicentre, phase 3 randomised trial. The lancet oncology 2013;14:989-998.
13. Chia S, Gradishar W, Mauriac L, et al. Double-blind, randomized placebo controlled trial of fulvestrant compared with exemestane after prior nonsteroidal aromatase inhibitor therapy in postmenopausal women with hormone receptor-positive, advanced breast cancer: results from EFECT. J Clin Oncol 2008;26:1664-70.
14. National Institute for Health and Care Excellence. Ribociclib with fulvestrant for treating hormone receptor-positive, HER2-negative, advanced breast cancer. Technology appraisal guidance [TA593]. 2019.

Acknowledgments: The clinical SLR update was conducted by Ingress Health.

Copyright ©2019. All rights reserved.

Presented at ISPOR Europe 2019 | 2-6 November 2019 | Copenhagen, Denmark