Leveraging Innovative Digital Technology: A Novel Approach to Service Optimisation

Kailey RS[1], Lee JM[1], Hart J[2], Clarke SEC[3], Jaffe JN[3], McGeagh JD[3]

[1]Janssen, High Wycombe, UK, [2]The Oxford Academic Health Science Network, Oxford, UK, [3]Accession Healthcare Consulting Ltd., Henley-on-Thames, UK

BACKGROUND

Treatment resistant depression (TRD) is a debilitating subtype of major depressive disorder (MDD), defined as a patient who has failed to respond to at least two different oral anti-depressants (OADs) during their current depressive episode.[1,2]

There is a significant unmet need for this patient cohort. Despite this, pathways are generally not optimised and the majority of TRD patients (90-95%) are managed in primary care with patients cycling through different treatment options leading to further treatment resistance and chronicity of the disease over time.

Service optimisation is critical so that that patients are managed in an appropriate care setting and to ensure that resources are used appropriately.

However, implementing service optimisation can have unforeseen downstream impacts meaning estimated cost savings or patient outcomes may not be realised. A novel digital technology, pathwaypro®, aims to provide local health economies (LHEs) with tools to explore the effects before implementing an intervention in a real-world setting.

This poster explores service optimisation initiatives within the MDD pathway and the integration into pathwaypro® to facilitate wider dissemination and exploration of potential solutions in other LHEs.

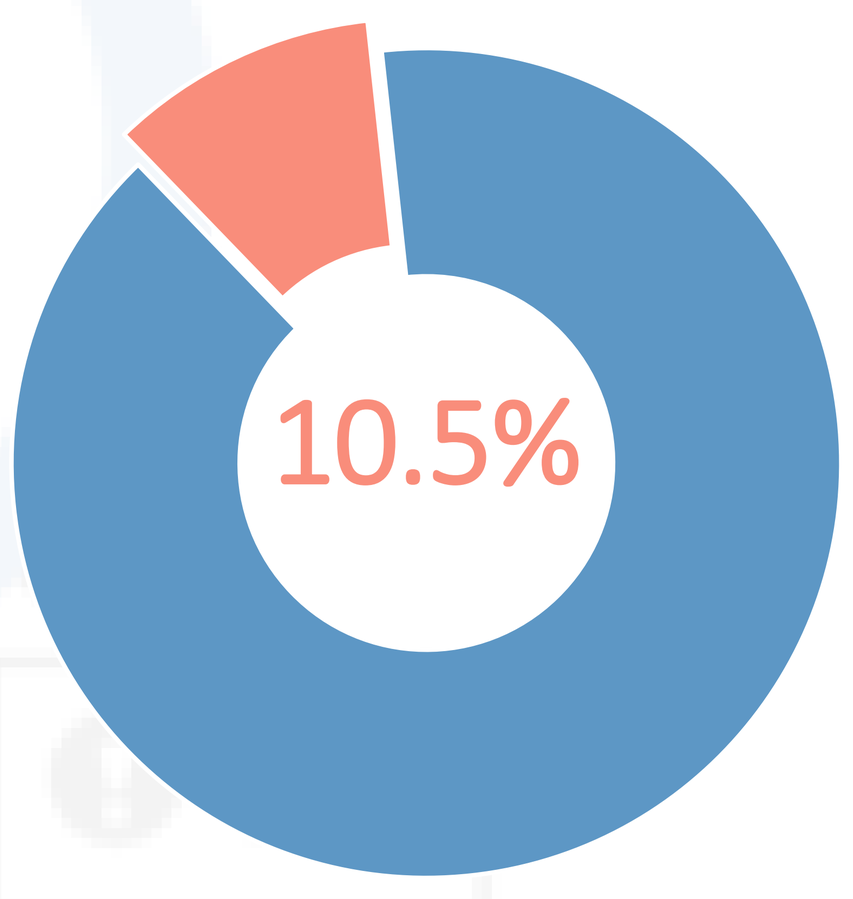
10.5%

UK data suggests about 10.5% of current MDD patients suffer from TRD, equating to over 130,000 patients.[3]

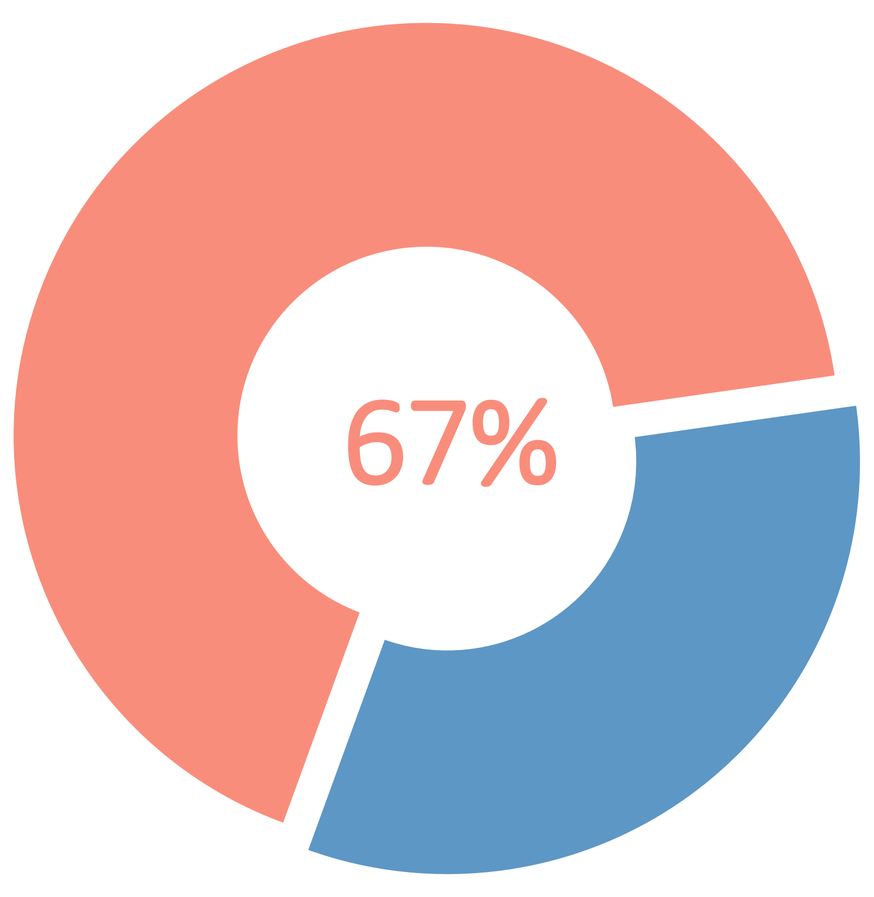
67%

TRD patients account for a disproportionate 67% (£3.9 billion) of the total cost of depression in the UK.[4,5]

pathwaypro®

Create the case for change

Setup the pathway to reflect your local health economy

Analyse the pathway for system pressure points

Implement an intervention, which may be based on outcomes from a case study from another locality

Review the whole system impact of the intervention

1. pathwaypro® is a communication tool that facilitates the communication of complex pathways and economic models within a local health economy
2. pathwaypro® enables the identification and analysis of pressure points within the pathway and provides the facility to overlay best practice pathway improvements to alleviate issues

DISCUSSION

- New models of care for the MDD pathway are in development across multiple academic health science networks (AHSN) in the UK.
- These studies aim to understand the impact that a new technology will have on an existing LHE and investigate new models of care that optimise and streamline primary to secondary care interactions.
- These studies are in the early stages of development; data will be collected over the next 6-24 months to demonstrate outcomes and impact of the interventions.

One of these case studies, conducted within the Oxford Academic Health Science Network (Oxford AHSN), is highlighted below.

CASE STUDY

Oxford AHSN are carrying out a lean assessment process, which aims to provide promising technologies coming to market an all-around assessment of the value proposition within a four month timeframe. By carrying out a stakeholder analysis and economic modelling, evidence will be produced to support key decision-making for the use of novel technologies in the NHS in England.

STAGE 1: STUDY INITIATION	STAGE 2: STUDY IMPLEMENTATION	STAGE 3: STUDY WRITE-UP	STAGE 4: BEST PRACTICE DISSEMINATION
The study will be conducted by Oxford AHSN in collaboration with Janssen. The initial phase will comprise of a workshop for knowledge sharing, identifying local variation and the mapping of the care pathway.	Interviews with key decision makers across the LHE will be conducted in combination with preliminary local health economic assessment to assess the impact on healthcare providers.	The evidence generated through this study will be developed into a report with guidance that will be applicable to the NHS in England.	The outcomes of this study will be integrated into pathwaypro®, which will facilitate dissemination across the UK by putting the findings in the context of each LHE and providing a localised cost impact of interventions.

ADDITIONAL PILOTS

A number of additional pilots are being explored across the UK, focused on three key areas that aim to improve services, capture data, identify patients sooner, and normalise mental health treatment nationally.

1. PATHWAY AND FUNDING
2. SERVICE DELIVERY
3. OUTCOMES AND MEASUREMENT

CONCLUSION

Dissemination of successful interventions across the NHS is essential for implementing best practice and reducing variation. Conducting pilots within the AHSN footprints before leveraging innovative digital technologies to assess how such interventions may translate into a new locality is a novel approach that will demonstrate a localised 'invest to save' business case to facilitate widespread adoption.

REFERENCES

1. Johnston KM, Powell LC, et al. The burden of treatment-resistant depression: A systematic review of the economic and quality of life literature. J Affect Disord. 2019;242:195-210.
2. Adult Psychiatric Morbidity Survey: Survey of Mental Health and Wellbeing, England, 2014. Available at: https://digital.nhs.uk/data-and-information/publications/statistical/adult-psychiatric-morbidity-survey/adult-psychiatric-morbidity-survey-survey-of-mental-health-and-wellbeing-england-2014#key-facts
3. Janssen. Data on File. Esketamine_DoF_12June2019_HEMAR_TM_002.
4. McCrone P, Dhanasiri S, et al. Paying the price. The cost of mental health care in England to 2026. 2008.
5. Ivanova JI, Birnbaum HG, et al. Direct and indirect costs of employees with treatment-resistant and non-treatment-resistant major depressive disorder. Curr Med Res Opin. 2010;26(10):2475-84.

Accession

Janssen

Oxford Academic Health Science Network

pathwaypro® and device is a registered trademark of Accession Healthcare Consulting Limited. All content, software and algorithms are proprietary to and copyright © 2019 Accession Healthcare Consulting Limited. For use only by Accession Healthcare Consulting licensees under written licence terms. All rights reserved