Incidence and Burden of Disease from Shingles and Post-Herpetic Neuralgia in the over 80 year olds in the UK

PIH31

AUTHORS

Ian Matthews*[1], Boriana Guimicheva[1], Mai Duong[2], Victoria Parsons[2], Michelle Johnson[2], Yash Patel[1], Bayad Nozad[3]

1. Merck, Sharp & Dohme Ltd, Hoddesdon, UK. 2. Oxon Epidemiology, London, UK 3. Imperial College London, UK

BACKGROUND

The UK herpes zoster (HZ) national immunisation programme currently covers individuals aged 70-79 years-old, although the exact eligibility criteria are complicated and have changed since the start of the programme. Due to the programme complexity and other factors, evidence[1] suggests a significant proportion of ≥80 year-olds, including those previously eligible, may have missed out on vaccination, thereby remaining at increased risk of disease. Furthermore, literature on the burden of disease in these older individuals is limited[2].

OBJECTIVES

The study describes the disease burden of HZ and post-herpetic neuralgia (PHN) in the immunocompetent unvaccinated UK population aged ≥80 years. The number of individuals who lost eligibility due to the 80-year cut-off in the UK national vaccination programme has also been estimated.

METHODS

Immunocompetent individuals unvaccinated against HZ and aged ≥80 years were identified using Clinical Practice Research Datalink (CPRD) / Hospital Episode Statistics (HES) data* during the study period (1st September 2013 until 30th June 2018). From this the HZ and PHN incidence rates were calculated per 1,000 person-years and stratified by age groups (80-84, 85-90 and ≥90 years). Additionally, the number of individuals who have lost eligibility for vaccination due to the age cut-off was calculated and extrapolated to the overall UK population, using data from the Office of National Statistics.

RESULTS

In the study (Figure 1) 255,613 individuals contributed 580,326 person-years of follow-up. Overall HZ incidence in those ≥80 was 8.43/1,000 person-years and decreased with age from 8.85 in 80-84 year-olds to 7.37 in the ≥90 year-olds (Table 1). Further investigations showed HZ rate was lowest in those aged ≥96 years (5.27/1,000 person-years). Overall PHN incidence was 0.80/1,000 person-years, and also decreased with age from 0.85 in the 80-84 cohort to 0.56 in those aged ≥90 (Table 2). Results show that 16.8% of the study population had lost eligibility to the vaccination programme whilst 83.6% had never been eligible for the vaccine. When extrapolated to the UK population this suggests that 2.2 million people over 80 remain unprotected from shingles (Figure 2).

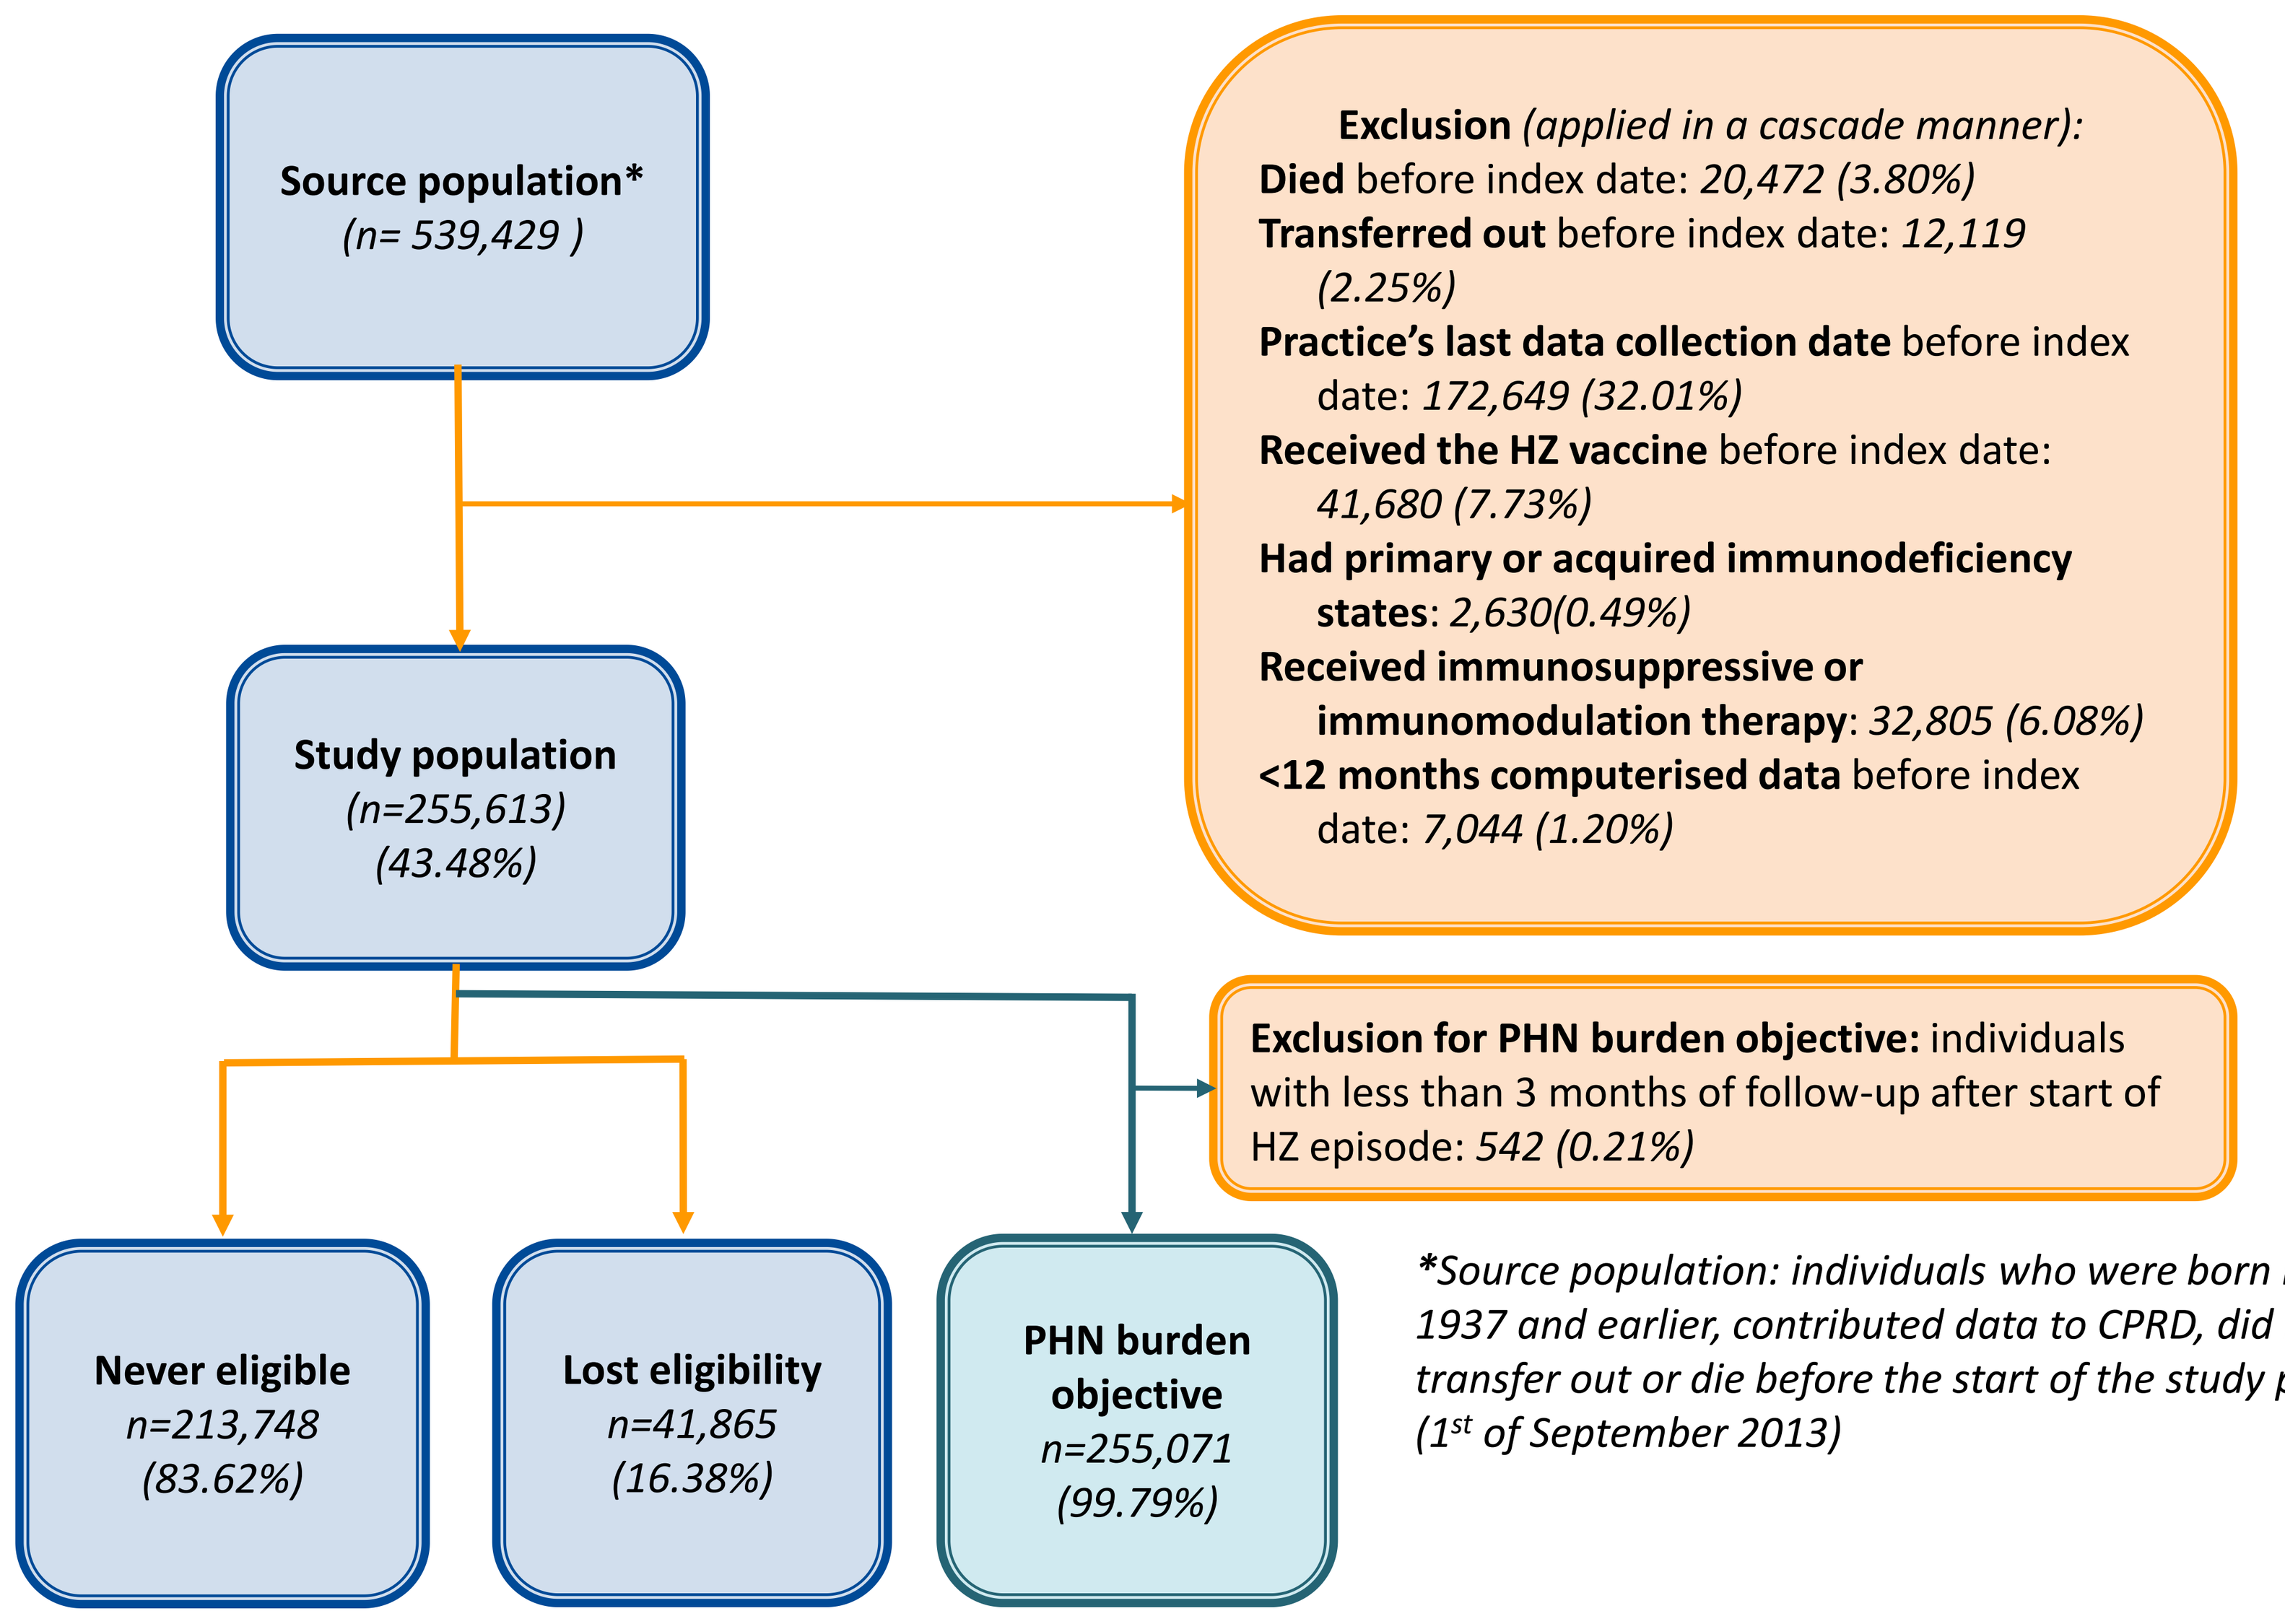

Source population*
(n= 539,429)
Exclusion (applied in a cascade manner):
Died before index date: 20,472 (3.80%)
Transferred out before index date: 12,119 (2.25%)
Practice's last data collection date before index date: 172,649 (32.01%)
Received the HZ vaccine before index date: 41,680 (7.73%)
Had primary or acquired immunodeficiency states: 2,630(0.49%)
Received immunosuppressive or immunomodulation therapy: 32,805 (6.08%)
<12 months computerised data before index date: 7,044 (1.20%)
Study population
(n=255,613)
(43.48%)
Exclusion for PHN burden objective: individuals with less than 3 months of follow-up after start of HZ episode: 542 (0.21%)
Never eligible
n=213,748
(83.62%)
Lost eligibility
n=41,865
(16.38%)
PHN burden objective
n=255,071
(99.79%)

*Source population: individuals who were born in 1937 and earlier, contributed data to CPRD, did not transfer out or die before the start of the study period (1st of September 2013)

Figure 1: Study population

	Number of patients	Number of episodes	Number of PY at-risk	Rate per 1,000 PY	[95%CI]
Overall	4,832	4,858	576,421	8.43	[8.19 - 8.66]
HES linkage status					
With HES linkage	2,287	2,293	253,317	9.05	[8.68 - 9.42]
Without HES linkage	2,545	2,565	323,104	7.94	[7.63 - 8.24]
HZ vaccine eligibility					
Never eligible*	4,354	4,380	513,700	8.53	[8.27 - 8.78]
Lost eligibility**	478	478	62,721	7.62	[6.94 - 8.30]
Age stratification¤					
80-84 years	2,915	2,051	231,852	8.85	[8.46 - 9.23]
85-89 years	3,126	1,815	209,973	8.64	[8.25 - 9.04]
≥90 years	1,428	992	134,595	7.37	[6.91 - 7.83]
Sex					
Male	1,722	1,730	218,762	7.91	[7.54 - 8.28]
Female	3,110	3,128	357,659	8.75	[8.44 - 9.05]
Ethnicity (n, %)					
White	2,193	2,199	238,943	9.20	[8.82 - 9.59]
Others	44	44	7,578	5.81	[4.10 - 7.52]
Social deprivation (Townsend score)					
1 (most affluent)	620	622	70,301	8.85	[8.16 - 9.54]
2	649	652	68,052	9.58	[8.85 - 10.31]
3	518	518	60,914	8.50	[7.77 - 9.23]
4	410	410	47,871	8.56	[7.74 - 9.39]
5 (least affluent)	190	191	25,625	7.45	[6.40 - 8.51]
Care home residency					
Living in care home	123	124	15,085	8.22	[6.78 - 9.66]
Living arrangements					
Living alone	140	141	16,614	8.49	[7.09 - 9.88]
Cohabiting	382	383	44,560	8.60	[7.74 - 9.45]
Urban versus rural location					
Urban	2010	2,015	245,760	8.20	[7.84 - 8.56]
Rural	393	394	42,969	9.17	[8.27 - 10.07]

PY: person-years
*Those who aged ≥80 years old at the start of the vaccination programme (including everyone who were born in 1933 as only year of birth is available in CPRD)
**Those turned 80 years old after the start of the vaccination programme (i.e. born between 1934 and 1937)
¤ Since age increases during follow-up, some individuals can contribute to more than one age stratification

Table 1: HZ rates in overall study population and for each stratification

	Number PHN cases	Number of PY at-risk	Rate per 1,000 PY	[95%CI]
Overall	464	578,800	0.80	[0.73 - 0.87]
HES linkage status				
With HES linkage	245	254,313	0.96	[0.84 - 1.08]
Without HES linkage	219	324,488	0.67	[0.59 - 0.76]
HZ vaccine eligibility				
Never eligible*	423	515,867	0.82	[0.74 - 0.90]
Lost eligibility**	41	62,934	0.65	[0.45 - 0.85]
Age stratification¤				
80-84 years	198	232,804	0.85	[0.73 - 0.97]
85-89 years	172	210,693	0.82	[0.69 - 0.94]
≥90 years	76	134,999	0.56	[0.44 - 0.69]
Sex				
Male	160	219608	0.73	[0.62 - 0.84]
Female	304	359193	0.85	[0.75 - 0.94]
Ethnicity (n, %)				
White	238	239,890	0.99	[0.87 - 1.12]
Others	<5			
Social deprivation (Townsend score)				
1 (most affluent)	60	70,588	0.85	[0.64 - 1.06]
2	74	68,347	1.08	[0.84 - 1.33]
3	57	61,143	0.93	[0.69 - 1.17]
4	39	48,045	0.81	[0.56 - 1.07]
5 (least affluent)	19	25,698	0.74	[0.41 - 1.07]
Urban versus rural location				
Urban	205	246,712	0.83	[0.72 - 0.94]
Rural	46	43,079	1.07	[0.76 - 1.38]

PY, *, ** and ¤ defined as per Table 1
Care home residency and living arrangements data are not reported due to small patient numbers

Table 2: PHN rates in the overall population and for each stratification

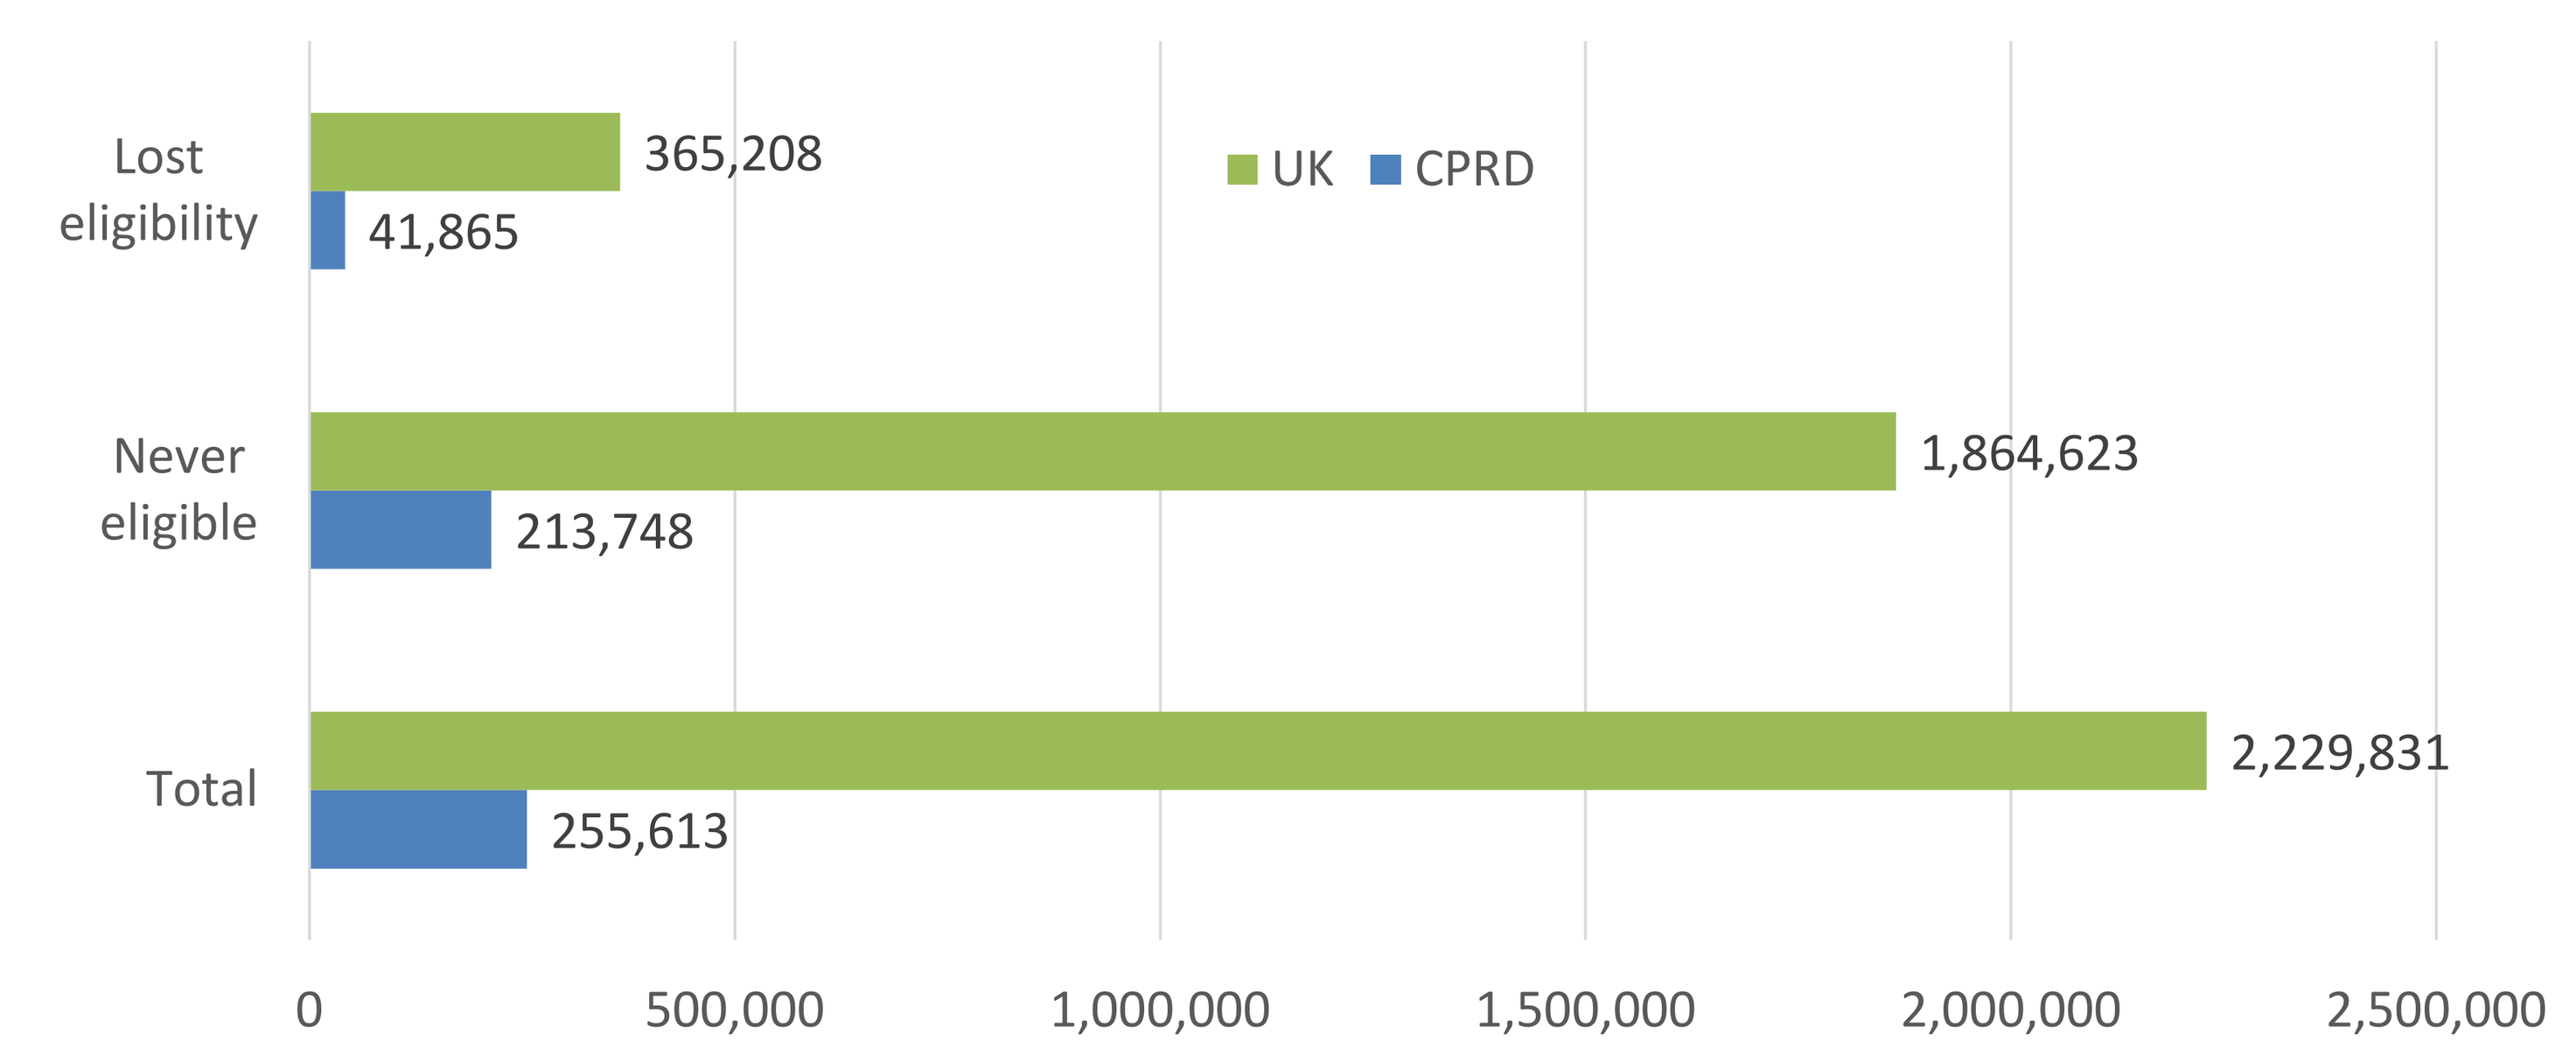

Lost eligibility
365,208
41,865
Never eligible
1,864,623
213,748
Total
2,229,831
255,613
UK
CPRD
0
500,000
1,000,000
1,500,000
2,000,000
2,500,000

Figure 2: Extrapolated number of individuals who have lost eligibility for HZ vaccination due to the 80 year old age limit in the UK from September 2013 to December 2017

CONCLUSIONS

This is the first study to look in detail at the burden of shingles disease in very old individuals across the UK. It is apparent that the incidence and burden of HZ and PHN remains significant in the those ≥80 years old. However, interestingly the disease burden seems to decline in those ≥90 years. Nonetheless the importance of vaccination to the oldest individuals in the UK remains high and a significant number of people are missing out on the available protection. From this evidence it seems timely for the UK to revisit the current shingles vaccination age cap and consider removing it so that fewer people remain at risk from shingles.

* The protocol for this research was approved by the Independent Scientific Advisory Committee (ISAC) of the Medicines and Healthcare Products Regulatory Agency (protocol number 18_320).

References:

1. Public Health England. Herpes zoster (shingles) immunisation programme 2017 to 2018: evaluation report. Health Protection Report. 2018;12(42).
2. A. Gautier et al, Epidemiol. Infect. (2009), 137, 38–47.

This study was organised and funded by Merck Sharp & Dohme Ltd, UK.

MSD INVENTING FOR LIFE