PUK28

EMULATING EQ-5D DATA FROM AVAILABLE PROS TO OPTIMIZE CLINICAL TRIAL PLANNING IN CHRONIC KIDNEY DISEASE-ASSOCIATED PRURITUS

Schaufler T [1], Ramirez de Arellano Serna A [1], Munera C [2], Menzaghi F [2]

[1] Vifor Pharma, Glattbrugg, ZH, Switzerland; [2] Cara Therapeutics, Inc., Stamford, CT, USA

Background and Objectives:

Chronic kidney disease-associated pruritus (CKD-aP) is a debilitating itching condition, significantly impacting health-related quality of life (HRQoL) of patients with progressive kidney disease. It is associated with depression, poor sleep quality and increased mortality. Epidemiologic data from several studies demonstrate that at least 30-40% of patients with end-stage renal disease suffer from moderate-to-severe pruritus. At the same time, data shows that CKD-aP is underdiagnosed by nephrologists and underreported by patients. There is currently no approved pharmaceutical treatment option in Europe or the US for the treatment of CKD-aP. [i]

In the absence of published reference data for this multi-morbid hemodialysis (HD) patient population suffering from moderate or severe CKD-aP, we aimed to assess the expected sensitivity of EQ-5D-5L to inform design and sample size calculation for potential studies intended to collect payer-relevant HRQoL and utility data.

Methods:

Difelikefalin (DFK) is a first-in-class, highly selective kappa opioid receptor (KOR) agonist that targets KOR located on peripheral sensory neurons and immune cells , which is currently in clinical development to treat CKD-aP in patients on hemodialysis (HD). It has demonstrated significant improvements of itch intensity and HRQoL as measured by the Skindex-10 (Sk-10) and 5-D Itch scale in HD patients with moderate-to-severe CKD in a recently completed double blind placebo-controlled phase-3-study (CR845-CLIN3102, KALM-1). [ii]

For the purpose of emulating EQ-5D-5L health states, we used available patient-level data from a phase-2-study (CR845-CLIN2101) in 174 CKD-aP patients treated with difelikefalin (DFK) 0.5, 1.0, and 1.5 mcg/kg or placebo that collected daily worst itch intensity (numerical rating scale from 0 = no itch to 10 = worst itch imaginable), Sk-10 and 5D-Itch Scale. We emulated EQ-5D-5L health states by matching related questions from available patient-reported outcomes (PRO) instruments to the relevant EQ-5D dimension and applied the United Kingdom (UK) and United States (US) valuation sets. [iii] To account for differences in the questions and rating scales used in each instrument, different mapping scenarios were assessed (cf. Figure 1).

Figure 1: Mapping models M1 to M4 of Skindex-10 and 5D Itch questions to EQ-5D domains

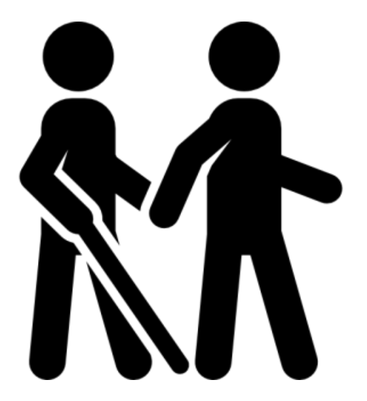

Mobility

- M1: 5D Itch Disability questions on (c) housework/errands, (d) work/school
- M2: 5D Itch Disability questions on (c) housework/errands, (d) work/school (constant baseline value)
- M3: same as M2
- M4: same as M2

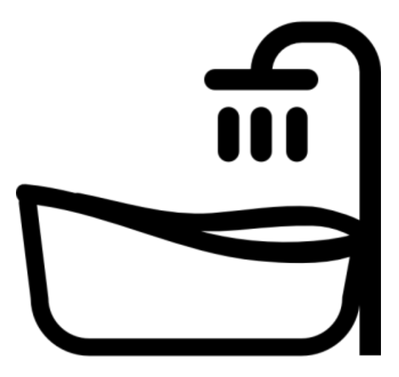

Self-Care

- M1: None
- M2: Skindex Q4: Frustration about your itching
- M3: same as M2
- M4: same as M2

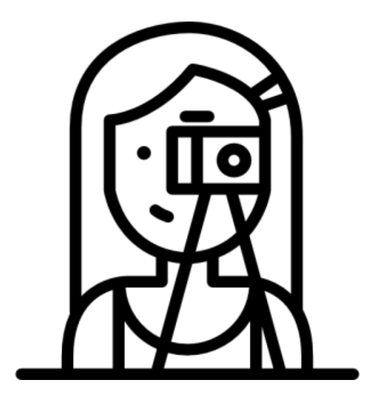

Usual Activities

- M1: 5D Itch Disability questions on (a) sleep, (b) leisure/social, (c) housework/errands, (d) work/school
- M2: Skindex Q10: Effect of itching making it hard to work or do what you enjoy
- M3: same as M2
- M4: Skindex Q8: Effects of itching on interactions with others

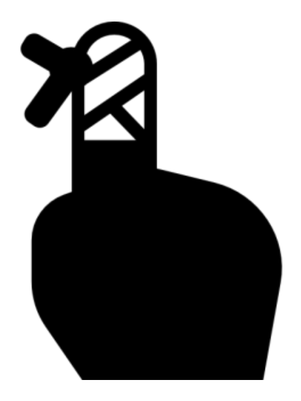

Pain / Discomfort

- M1: 5D Itch question: Duration of itch (hours per day)
- M2: 5D Itch question: Degree (intensity) of itch
- M3: Skindex Q1: Degree of being bothered by itching
- M4: same as M3

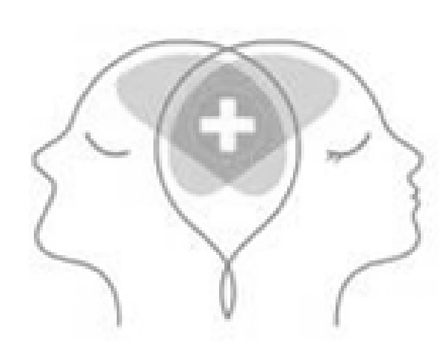

Anxiety / Depression

- M1: Skindex Q6: Feeling depressed about your itching
- M2: same as M1
- M3: same as M1
- M4: same as M1

For the *Mobility* domain of the EQ-5D, we referred to the reply to the third and fourth sub-question of the disability item of the 5-D Itch scale, asking how much the itch impacted housework / errands and work / school activities in the past two weeks. The higher value (reflecting the more severe impairment) of the two questions was mapped. However, as it is not clear whether improving the itch would actually result in an improvement of self-reported mobility in the EQ-5D, we kept the value at baseline constant for Models M2 to M4.

Based on the same logic and because neither Sk-10 nor 5-D Itch include any question directly related to the *Self-care* domain of the EQ-5D, we assigned a value of 3 to all patients at baseline and end of study in model M1. For models M2 to M4 we mapped question 4 of Sk-10 ("frustration about your itching") as the closest match.

The domains *Usual Activities* and *Pain / Discomfort* can be linked to several individual questions of the two PRO scales used in the study (cf. Figure 1) and we therefore tested several models to check for major discrepancies in the resulting health states and resulting utility values respectively.

Finally, for the *Anxiety / Depression* domain, the Sk-10 was an obvious match with Sk-10 question 6 ("feeling depressed about your itching").

The 5 point scale of 5D Itch was directly mapped to the respective values of the EQ-5D. To map the 7 point Sk-10 scale the following assumptions were used: 0 – no problems; 1, 2 – slight problems; 3, 4 some problems; 5 – severe problems; 6 – extreme problems.

Results:

The frequency distribution of emulated health states per domain implies that the **main driver for reduced HRQoL is the *Pain / Discomfort* domain** with two thirds of CKD-aP patients indicating severe or extreme problems, followed by *Self-care*, for which almost half of patients indicate severe or extreme problems (cf. Figure 2).

Aggregating these results into health states and calculating the respective utility values confirm that patients with moderate-to-severe CKD-aP experienced a substantial utility impairment (baseline score between 0.15 and 0.35 with the UK tariff, depending on mapping used). The US tariff resulted in a somewhat lower, but still very relevant impairment (baseline of 0.39 – 0.53), reflecting different valuations in diverse populations.

Treatment with DFK resulted in a statistically significant 8.2 point higher improvement on the Skindex-10 and of 2.5 points on the 5D-Itch scale (both p<0.001 versus placebo). Depending on the mapping applied to emulate EQ-5D-5L health states, **this translated into 0.1 to 0.21 higher HRQoL scores vs. Placebo at week 8** based on the UK tariff and 0.07 to 0.14 with the US tariff. Emulated QoL scores of DFK-treated patients at week 8 are similar to those directly measured with EQ-5D-5L in HD patients in other studies, supporting the validity of the results.

Main driver of the higher HRQoL were lower severity of reported problems in the EQ-5D *Self-Care* and *Pain / Discomfort* domains.

Figure 2: Distribution of emulated EQ-5D health states at baseline (based on model M4)

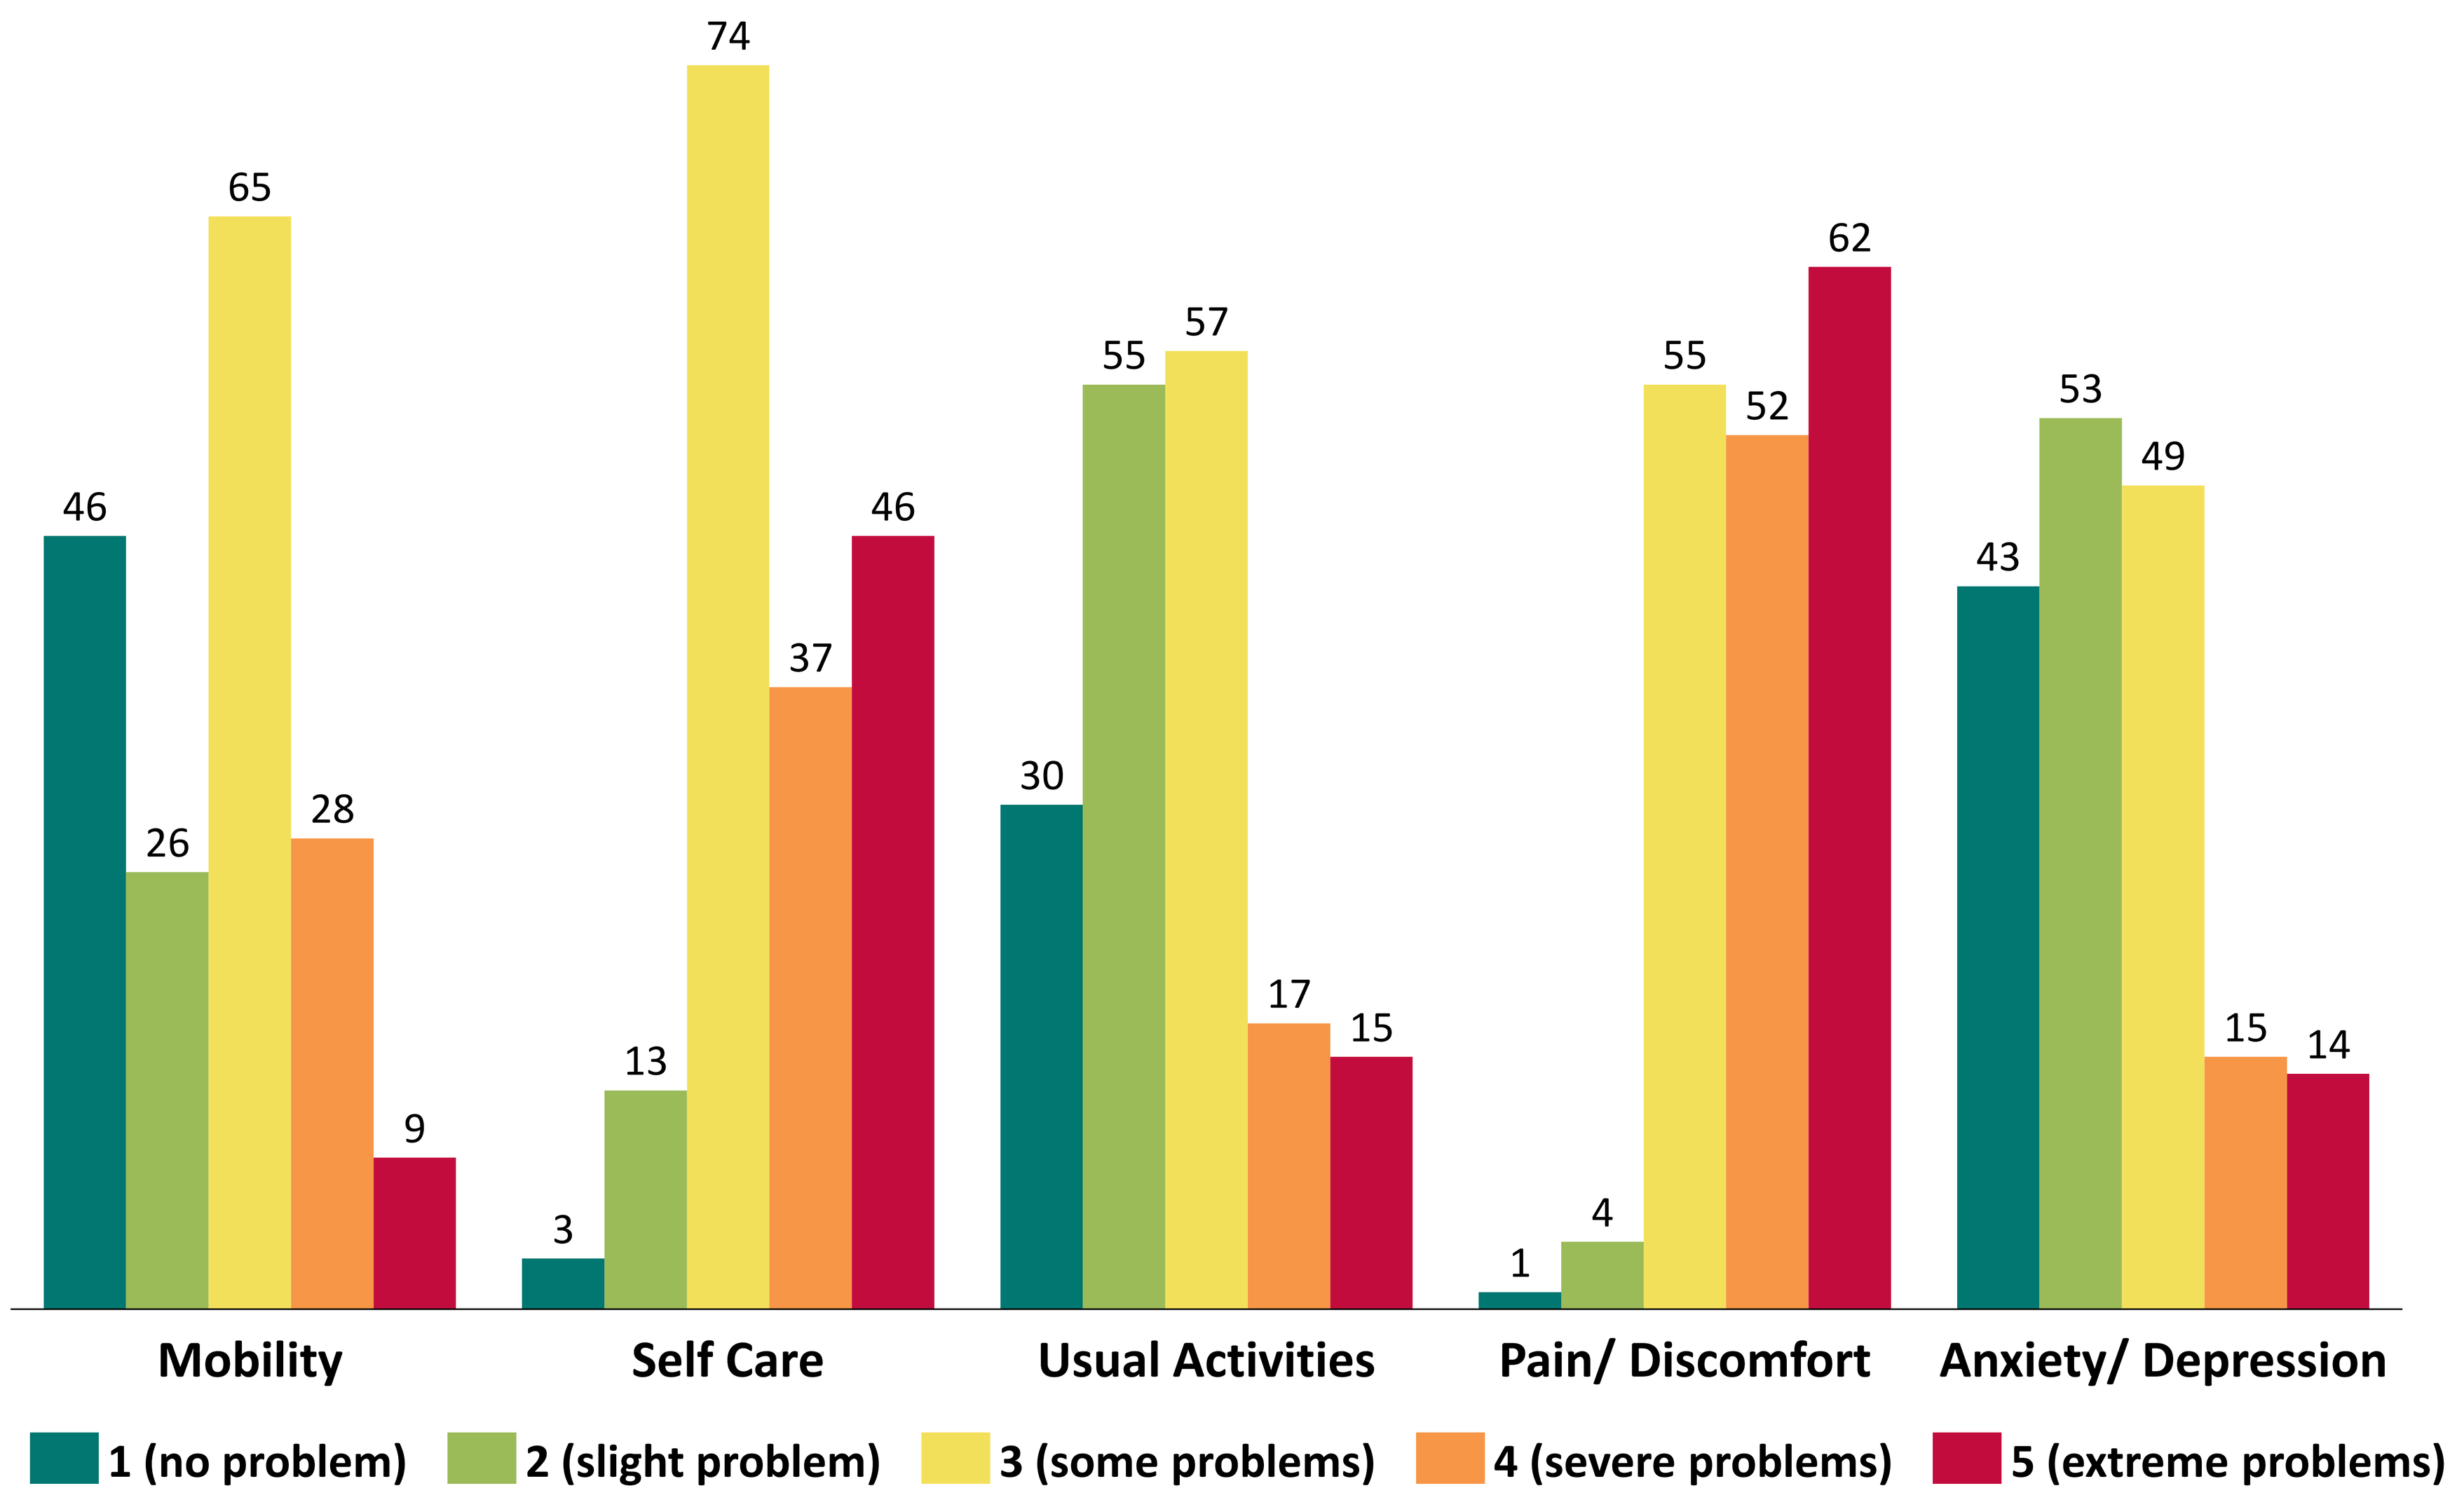
46
26
65
28
9
3
13
74
37
46
30
55
57
17
15
1
4
55
52
62
43
53
49
15
14
Mobility
Self Care
Usual Activities
Pain/ Discomfort
Anxiety/ Depression
1 (no problem)
2 (slight problem)
3 (some problems)
4 (severe problems)
5 (extreme problems)

N = 174, except 1 missing value for *Self-care* domain

Conclusion:

This emulation increased the confidence in the ability of EQ-5D-5L to detect relevant differences in patients' utilities when treating CKD-aP, despite potential confounding by the underlying morbidity. While these results do not allow to claim any actual effect on patients' utility, the quantification of the magnitude of the expected effect can be helpful to appropriately power studies focused on collecting HRQoL data in CKD-aP patients in the absence of published results or analogue cases.

i) Rayner et al., *Clinical Journal of the American Society of Nephrology*, 2017; Pisoni et al., *Nephrol Dial Transplant Journal,* 2006; Fishbane et al., *NDT Journal*, 2001; ii) Cara Therapeutics, press release, May 2019; iii) Devlin et al., *Health Economics*, 2017; Pickard et al., *Value Health,* 2019;