CHARACTERISTICS OF PATIENTS WITH TYPE 2 DIABETES MELLITUS NEWLY TREATED WITH BASAL INSULIN THERAPY: A POPULATION-BASED UK STUDY

Muna Adan[1], Seidu S[1,2], Zaccardi F[1,2], Khunti K[1,2], Webb Dr[1,2], Tyrer F[1], Gillies C[1], Saremi A[3], Lubwama R[3], Boss A[3], Dex T[3]

[1] Department of Health Sciences, University of Leicester, Leicester, UK, [2] Diabetes Research Centre, Leicester General Hospital NHS Trust, Leicester, UK, [3] Sanofi, New Jersey, USA

Background

In 2016, the World Health Organization reported that type 2 diabetes (T2DM) was the seventh leading cause of mortality with approximately 1.6 million attributed deaths. Similarly, T2DM is a major public health concern in the UK and currently there are 3.9 million people living with T2DM.The condition is progressive and the resulting hyperglycaemia can lead to both micro and macro-vascular complications, thus timely clinical interventions are vital. As T2DM progresses, pharmacological management are frequently needed including intensive treatment with insulin-therapy. However, little is known about patients with T2DM newly initiated with basal insulin therapy in UK primary care.

Aim

The aims of this study were to:

- Determine the prevalence of patients with T2DM newly initiating basal insulin between January 2004-December 2016
- Describe the demographic and clinical traits of T2DM patients newly initiating basal insulin therapy between January 2004-December 2016

Methods

Data Source and Study Population

The Clinical Practice Research Datalink (CPRD) was used to identify adult patients with type 2 diabetes and with a first eligible prescription of basal insulin (index-date) between 1st January 2004 and 31st December 2016.

Exclusion Criteria

- Type 1 Diabetes Mellitus (identified by Read codes)
- Less than 12-monts practice registration prior to index date
- ≤18 year of age at index-date
- No HbA1c-recordings 6-12months within the index-date

Exposure

- Exposure to first prescription of a long-acting or intermediate acting insulin were measured between 1st January-31st December 2016:
- Number of days' of drug supply from index-date determined medication continuity
 - Long term continuous course was defined as <60 days supply/gap/year

Measures

Patient characteristics were assessed prior to insulin treatment initiation (index-date) and from first T2DM-diagnosis, the measures included;

- Socio-demographic factors
- Comorbidities (Charlson comorbidity index/score)
- Lifestyle/clinical measures (smoking, BMI, HBA1c-levels)
- Co-prescriptions (oral antidiabetic medications, cardiovascular medications, psychotropic medications)

Data Analysis

Descriptive statistics were used to describe patients' demographics, clinical traits. Characteristic differences between gender were compared using Student T-test and Pearson chi-squared and *p*-value of 0.05 was considered statistically significant. Results were expressed as percentage, mean±sd unless indicated otherwise.

Results

- There were a total of 13,811 T2DM patients newly prescribed insulin between 2004-2016.
- Majority (55.43%) of patients were male, and the average age of the cohort at index-date was 64.90±12.88 years (Figure 1).

Figure 1. Age distribution of patients initiated basal insulin by gender

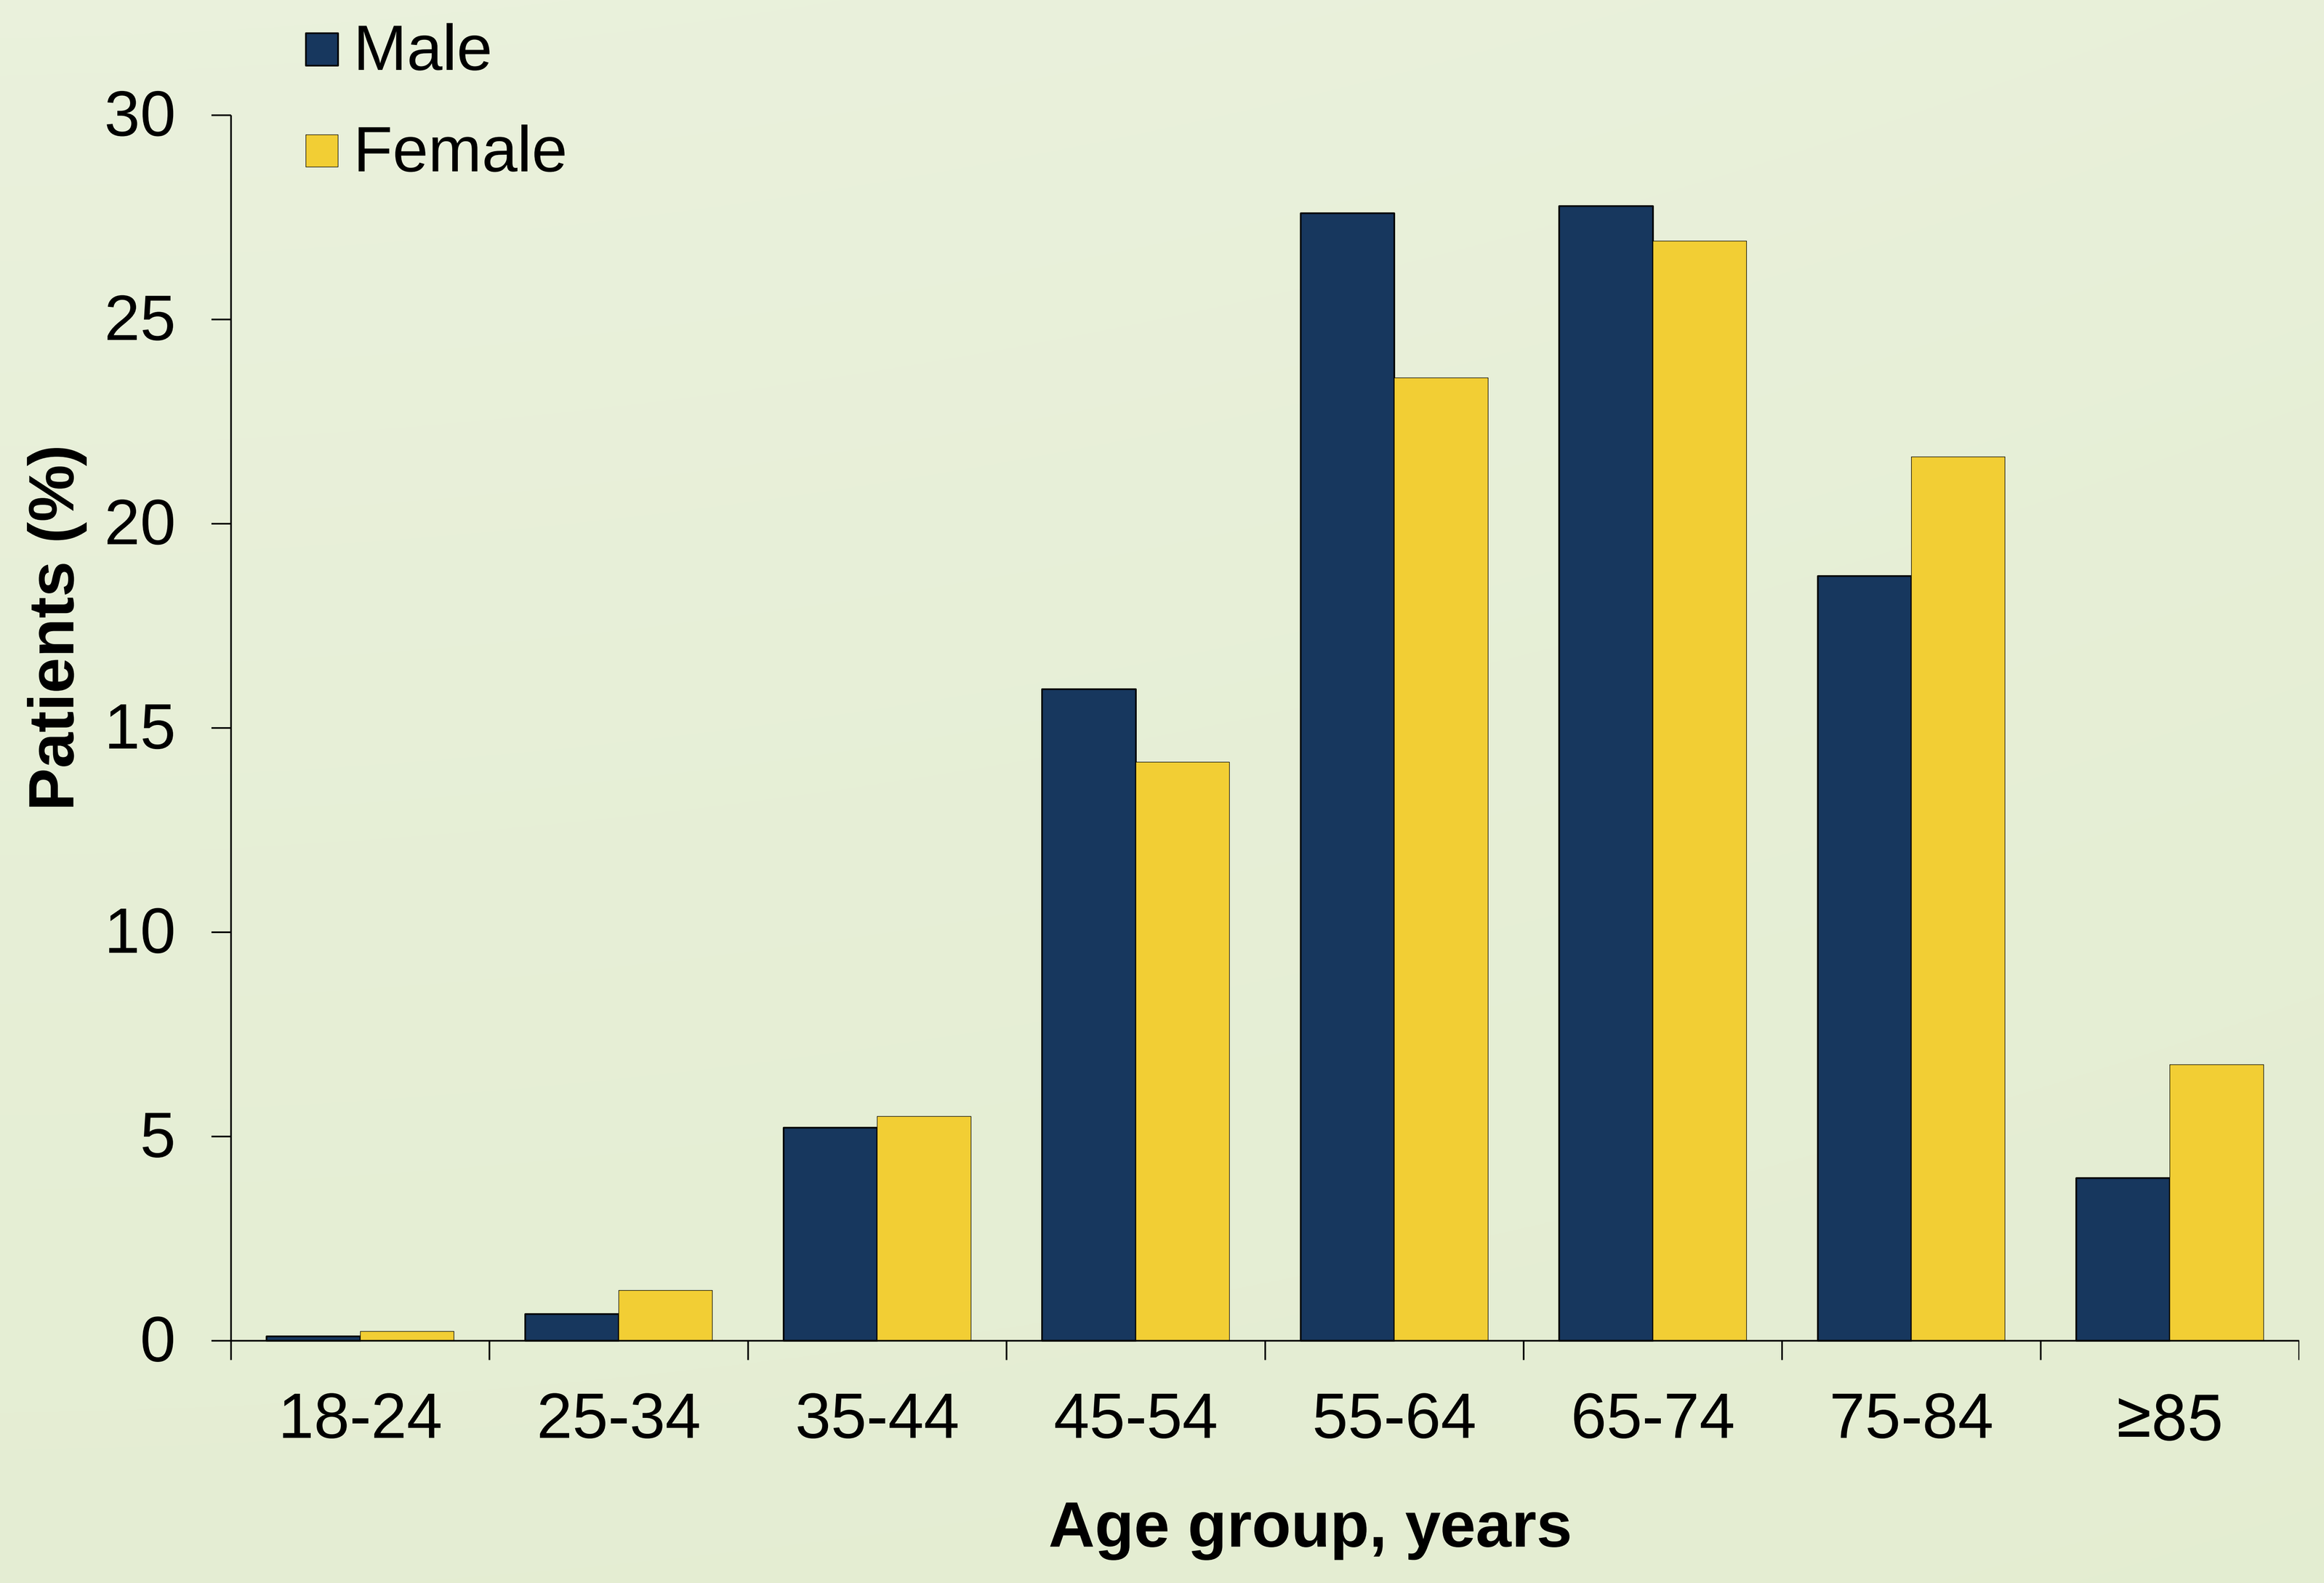
Male
Female
30
25
20
15
10
5
0
Patients (%)
18-24
25-34
35-44
45-54
55-64
65-74
75-84
≥85
Age group, years

Results

More than a third of the study cohort (38.01%) had ≥2 chronic comorbidities prior to basal insulin therapy and hypertension, renal disease, ischaemic heart disease, COPD, depression were most common (Table 1). Eighty-seven percent (n=12,029) of patients had a record of prior use of OAD, with 61.74% having had two or more OAD prior to insulin therapy. A large proportion of the study population were also on antiplatelet, antihypertensive and lipid-lowering drugs prior to the initial use of insulin.

Table 1. Clinical characteristics of study cohort

Characteristics	Study sample		
	Male	Female	*P*-value
	n=7,655	**n=6,156**	
Duration of T2DM (years, Median, IQR)	8 (4-12)	7 (4-12)	<0.001
Socioeconomic status			
1-least deprived	1,453 (18.98)	1,018 (16.53)	
2	1,632 (21.23)	1,249 (20.29)	
3	1,619 (21.14)	1,349 (21.91)	
4	1,666 (21.76)	1,343 (21.81)	
5-most deprived	1,285 (16.79)	1,198 (19.46)	0.01
Smoking status			
Smoker	1,469 (20.06)	1,030 (17.84)	
Ex-smoker	3,264 (44.57)	1,688 (29.24)	
Non-smoker	2,591 (35.38)	3,055 (52.92)	<0.001
BMI-category			
Normal	627 (8.34	493 (8.24)	
Overweight	1,843 (24.51)	1,116 (18.64)	
Obese	4,308 (57.30)	3,857 (64.43)	<0.001
Comorbidities			
Hypertension	2,683 (35.05)	1,904 (30.93)	<0.05
Renal disease	1,159 (15.14)	1,104 (17.93)	<0.001
Ischaemic heart disease	867 (11.33)	485 (7.88)	<0.001
COPD	379 (4.95)	449 (7.29)	<0.01
Depression	1,122 (14.66)	1,361 (22.11)	<0.01
Co-prescribed drugs			
Lipid-lowering	3,042 (39.74)	2,495 (40.53)	<0.001
Antihypertensive	2,649 (34.60)	1,864 (30.28)	<0.001
Antiplatelet	1,248 (16.30)	1,379 (22.40)	<0.001
Antidepressants	691 (9.03)	897 (14.57)	<0.001
Antipsychotics	803 (10.49)	893 (14.51)	<0.001
Benzodiazepines	2,649 (34.60)	1,864 (30.28)	<0.001

**p-value measure of statistical significance*

Figure 2. Changes in glycaemic control

The median glycated haemoglobin measured up to 12-months prior to basal insulin therapy was 9.60% (IQR, 8.30,11.10) in males and 9.40% (IQR, 8.10, 11.20) in females (Figure 2). At 12-months after insulin initiation HbA1c-levels was; 8.40% (IQR, 7.30, 9.80) in males and 8.30% (7.40,9.70) in females. A reduction of 1.2% in males and 1.1% in females.

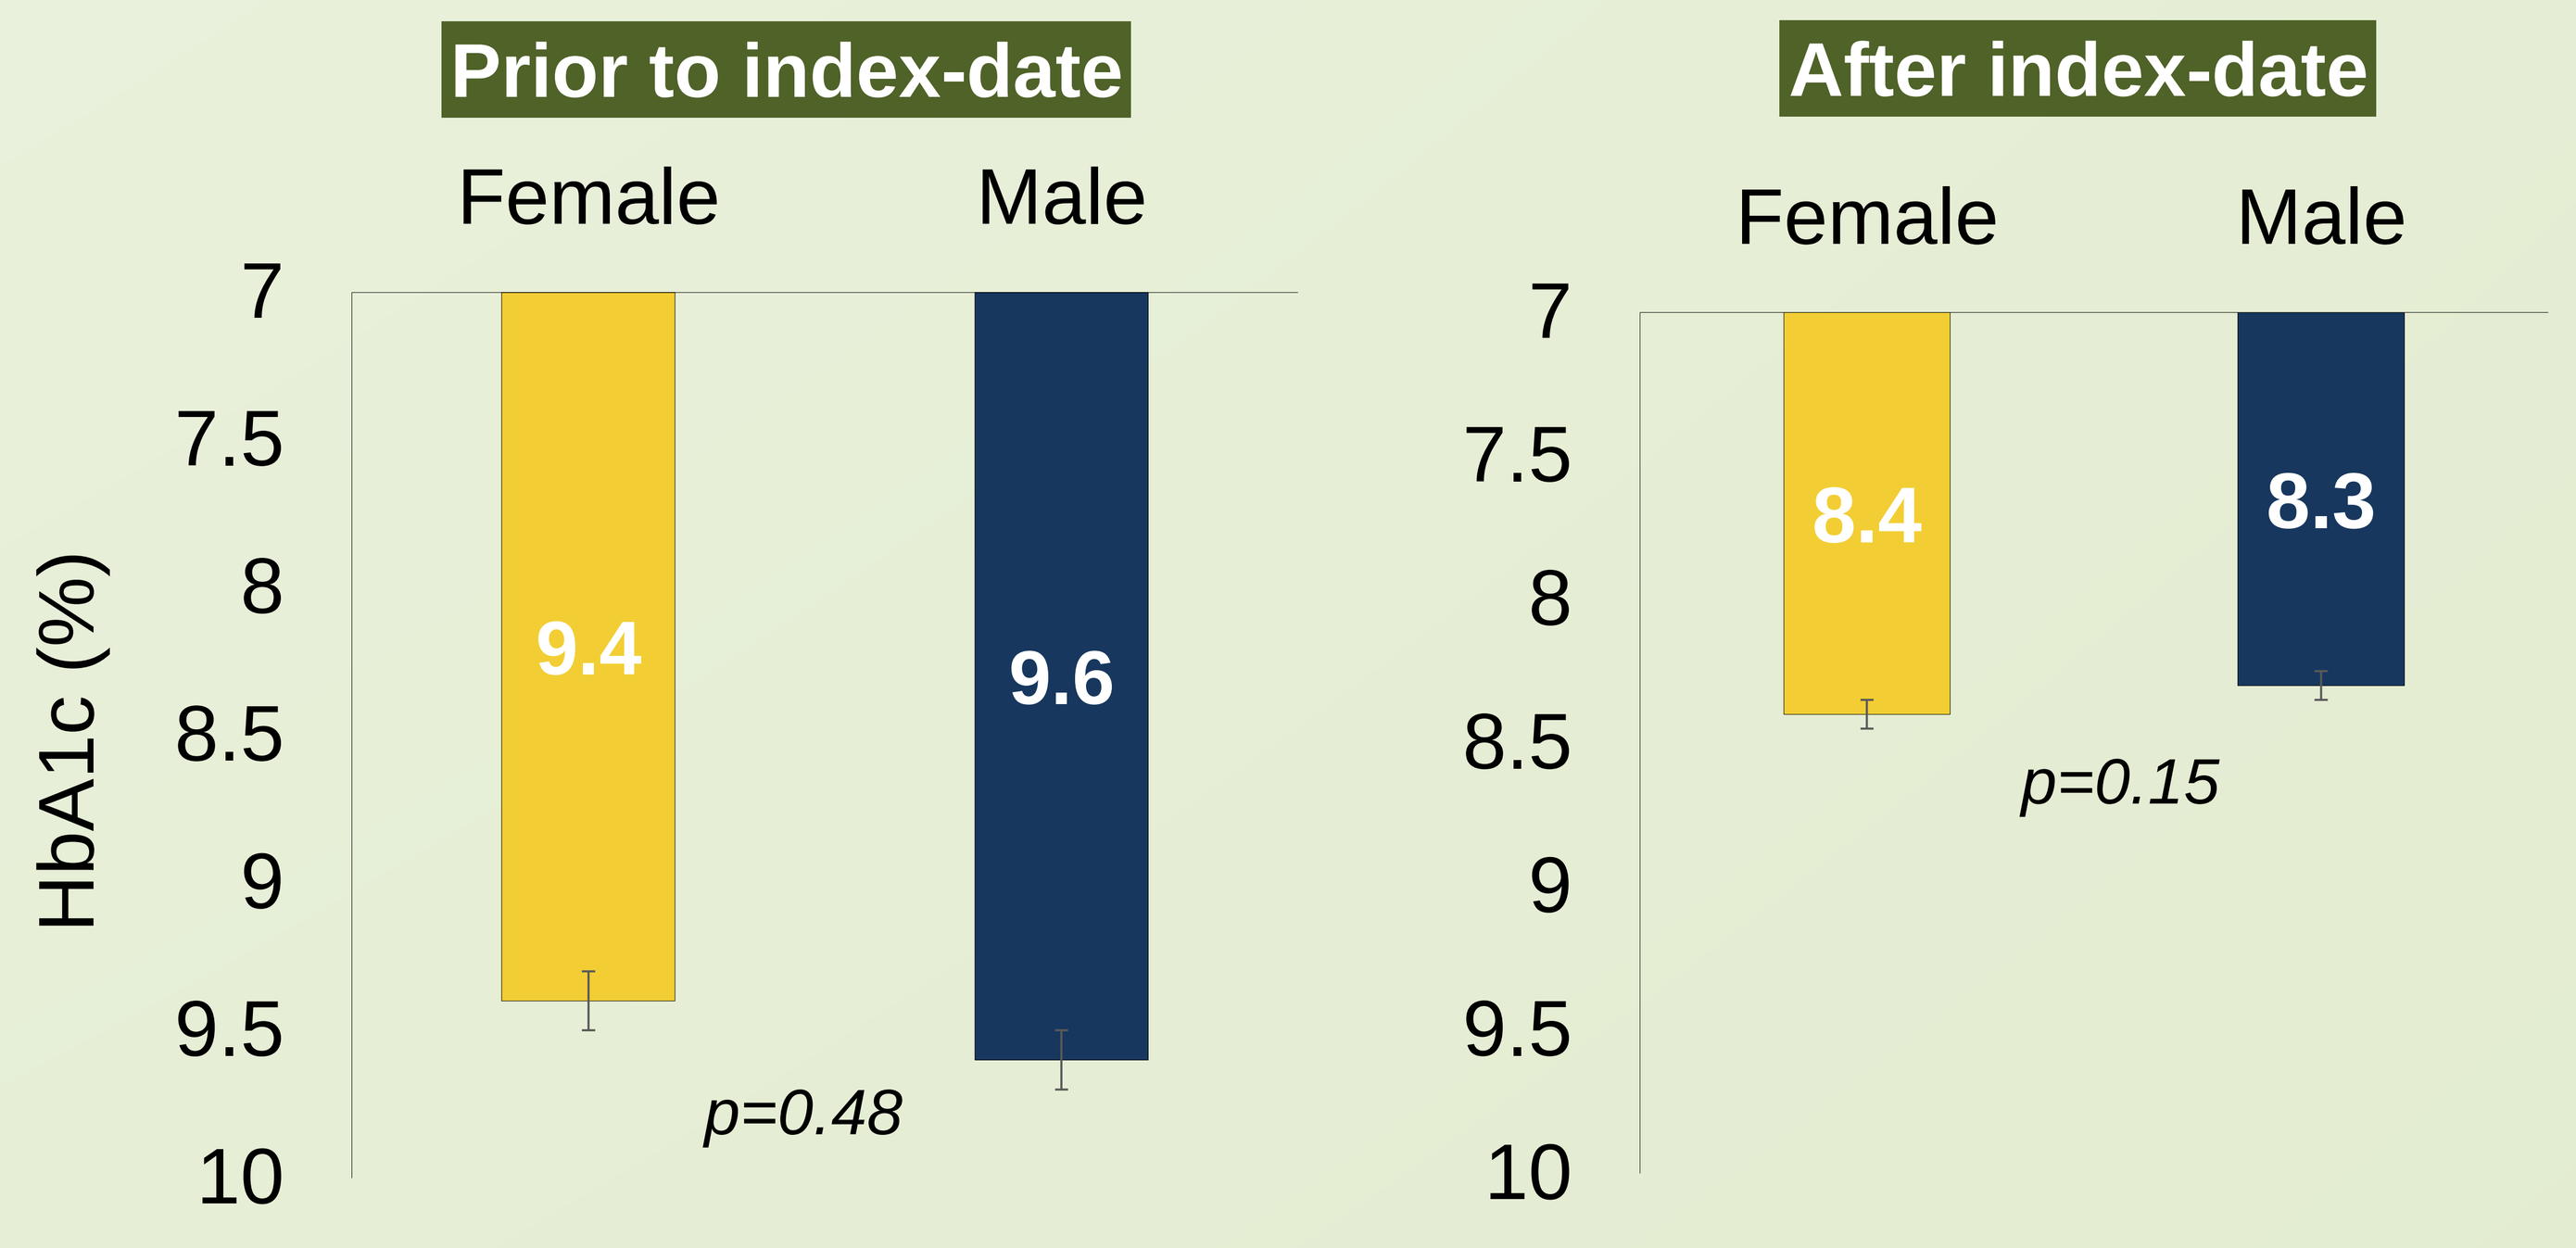
Prior to index-date
Female
Male
7
7.5
8
8.5
9
9.5
10
HbA1c (%)
9.4
9.6
p=0.48
After index-date
Female
Male
7
7.5
8
8.5
9
9.5
10
8.4
8.3
p=0.15

Conclusion

T2DM patients newly initiated insulin were mostly males and mid-older in age

- Users had a number of diabetes related complications
- Poor glycaemic control was observed prior to and following insulin therapy

Consequently, appropriate clinical management of T2DM is important in achieving glycaemic goals and these findings identify a specific cohort of patients with long-standing, complex, comorbidities who may require closer clinical management.

Conflict of interest

This research study has been funded by Sanofi.

UNIVERSITY OF LEICESTER University Hospitals of Leicester NHS Trust
www.leicesterdiabetescentre.org.uk
@LDC_Tweets
facebook.com/leicesterdiabetescentre