Long-Term Impact of Bedaquiline-containing Regimens on the Cost Burden of Drug-Resistant Tuberculosis in India

A. M. Agnarson,[1] R. Potluri,[2] H. Bhandari,[3] A. Dhir,[3] A. Kumar,[3] V. Gupta,[4] L. Raute,[4] C. Kambili,[5] L. Metz[1]

[1]Johnson & Johnson Global Public Health, New Brunswick, NJ, USA [2]SmartAnalyst Inc., New York, NY, USA [3]SmartAnalyst India Pvt. Ltd., Gurugram, India [4]Janssen India, Pharmaceuticals Division of Johnson and Johnson Pvt. Ltd., India [5]Johnson and Johnson Global Public Health, Raritan, NJ, USA

INTRODUCTION

- India is beset with a significant public health burden on account of drug-resistant tuberculosis (DR-TB), accounting for 24% of the estimated 558,000 global cases in 2017.[1]
- A TB patient potentially bears medical and non-medical direct costs, as well as indirect costs resulting from loss of work due to physical infirmity and time spent on seeking medical care.[2] Such costs force families of TB patients to spend from their savings, borrow money or sell assets, divert resources from other needs, and reduce food consumption.[2]
- Overall costs associated with treatment of DR-TB in India are about 25 times greater than those associated with treatment of drug-susceptible TB; this is attributed to the longer duration of treatment, higher costs of drugs, and greater need for diagnostic tests and management of adverse events.[3, 4]
- The occurrence of DR-TB is thus likely to impose catastrophic costs on families (WHO defines this to be when total costs incurred by a household exceed 20% of its annual income). A study showed 68% of DR-TB patients treated in a private tertiary care setting in India in 2016 to be facing catastrophic costs.[4]
- Bedaquiline, an oral agent approved in 2012 for DR-TB treatment, has demonstrated superior effectiveness in diverse patient populations;[5-7] treatment success rates of between 61% and 93% have been reported. WHO's latest DR-TB guidelines have categorized bedaquiline as a core first choice drug.[8]

The current study aims to evaluate the impact of bedaquiline-containing regimens on healthcare costs and catastrophic costs to families of patients with DR-TB in India.

OBJECTIVE

This study aims to

1. assess the long-term impact of an increase in the use of bedaquiline-based regimens in DR-TB patients in India on the direct costs to the healthcare system and catastrophic costs to patients associated with DR-TB
2. evaluate two scenarios with different expectations of the treatment success rate of bedaquiline-containing regimens

METHODS

- A dynamic state-transition model was used to estimate annual epidemiology of DR-TB in India. Details of the model structure have been published elsewhere.[9] The model builds up from the year 2000 to its current state and extends to the year 2040 based on estimations of relevant diagnostic and treatment-related parameters in the future, assuming the continued use of current standard-of-care treatments. To this baseline DR-TB forecast, the impact of adoption of bedaquiline-containing regimens on incidence, prevalence and mortality was evaluated under two scenarios that differed in the treatment success rates, 61%[10] in scenario 1 and 80%[7] in scenario 2 (Table 1), as detailed in a previous study.[11]
- The model was then employed to estimate the economic burden of DR-TB both at the societal as well as individual patient-household level. Costs considered for the analysis included direct medical and non-medical costs for the treated patients (Table 2).
- The proportion of patients catastrophically affected were separately estimated for 4 pools of patients corresponding to the stage in the disease cascade that they would exit from – a) those cured in LOT1, b) those that die after LOT1 failure but before starting LOT2, c) those cured in LOT2, and d) those that die after LOT2 failure. This exercise was separately carried out for the patient population treated in private and public care hospitals for each year of the forecast for patients starting first line DR-TB therapy.

Table 1: Inputs considered for scenario analysis: bedaquiline success rates and market penetration / adoption

Parameter		Scenario 1	Scenario 2
BDQ first line DR-TB treatment success rate		61%	80%
Peak utilization of bedaquiline-containing regimens in DR-TB treated patients	First line of DR-TB treatment	95%	
	DR-TB second line of treatment for patients failing front line[a]	7%	12%

BDQ, bedaquiline; DR-TB, drug resistant tuberculosis; LOT, line of treatment;
[a]For second line DR-TB treatment, it is assumed that patients who failed on front line treatment with bedaquiline are ineligible for repeat prescription of bedaquiline and that among second line patients not treated with bedaquiline in front line, 95% will be treated with bedaquiline in second line in both scenarios.

Table 2: Unit costs per treated patient, as input into the model

Costs in 2018	Cost per patient per DR-TB treatment course (in US Dollar 2018)					Source/ Remarks
	SOC LOT1	BDQ LOT1	XDR-TB : DR-TB Cost Factor	SOC LOT2 (XDR-TB)	BDQ LOT 2 (XDR-TB)	
Drug costs	$2,374	$1,654	3	$3,800	$2,356	Ionescu AM, et al.;[1] See Footnote A
Hospitalization costs	$1,478	$673	3	$6,299	$2,783	
Injection-based AEs	$1,818	$0	1.54	$3,798	$0	
Other costs	$1,957	$972	2.34	$5,296	$2,515	
Total costs (excluding productivity loss)	$7,627	$3,300	NA	$19,192	$7,654	

- **Footnote A:**
 - Per patient costs are calculated using per successful patient cost of SCR and LCR of the SOC and BDQ, multiplied by respective success rates, and weighted for 50-50 SCR-LCR volumes of regimens in India for DR-TB; for XDR-TB, only LCR use is assumed
 - XDR-TB costs are calculated from DR-TB costs per patient by multiplying by the XDR-TB cost factor. For injection-based AE and other costs, the factor is weighted for subgroups these consist of
 - Productivity loss is not considered in this calculation

1. Ionescu AM, et al. Bedaquiline- versus injectable-containing drug-resistant tuberculosis regimens: a cost-effectiveness analysis. Expert Rev Pharmacoecon Outcomes Res. 2018;18(6):677-689.

- Only patient-incurred costs, adjusted for inflation, were considered. To evaluate indirect costs, the probability of loss of productivity in each disease state was estimated.
- Variance in household income across the entire TB affected population was accounted for by simulating the nominal income distribution at the individual household level using a lognormal distribution of income among the population, with.
 - mean of the distribution as the average income of patients – privately treated patients were taken to earn annually the national per capita GDP, and those treated in public hospitals were taken to earn a third of the GDP[12]
 - standard deviation of the distribution as the GINI index of the country

RESULTS

- In the two scenarios involving use of bedaquiline, the reductions in the economic burden over the 20-year horizon through 2040, relative to the baseline scenario, are estimated to be $7.6B (47% of estimated baseline burden of $16.0B) and $9.3B (58%), respectively (Fig. 1). During this period, the average cost per successfully-treated patient was projected to decline, relative to the baseline scenario without use of bedaquiline, by 54% (or $12.3k) under scenario 1, and by 67% (or $15.4k) under scenario 2 (Fig. 2).

Figure 1: Estimated costs associated with DR-TB in India cumulatively in 2021-2040 under baseline (no bedaquiline use) and two bedaquiline use scenarios

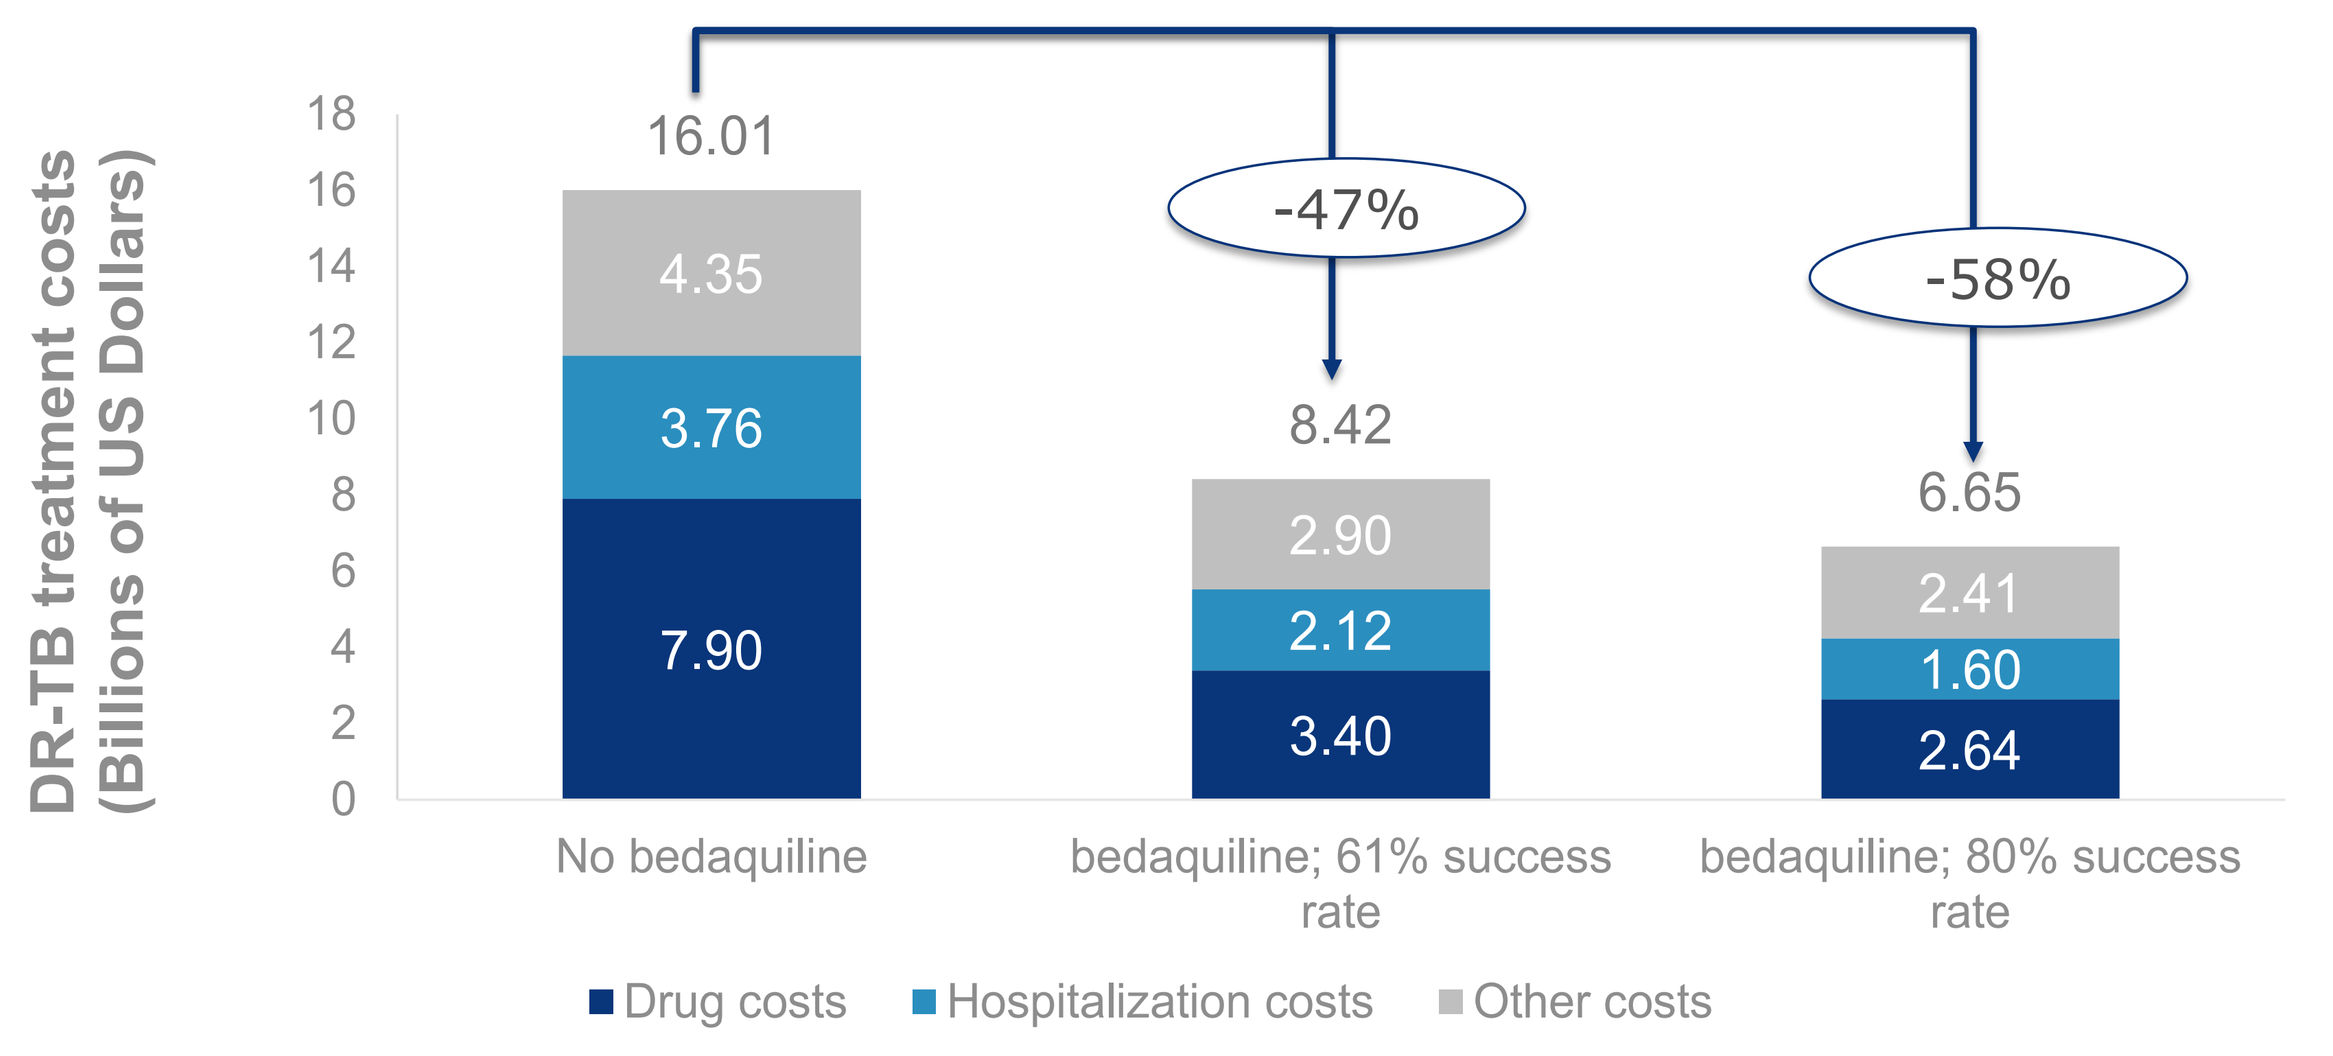
DR-TB treatment costs (Billions of US Dollars)
0
2
4
6
8
10
12
14
16
18
16.01
4.35
3.76
7.90
-47%
8.42
2.90
2.12
3.40
-58%
6.65
2.41
1.60
2.64
No bedaquiline
bedaquiline; 61% success rate
bedaquiline; 80% success rate
Drug costs
Hospitalization costs
Other costs

Figure 2: Estimated cost per successfully treated patient of DR-TB in India cumulatively in 2021-2040 under baseline (no bedaquiline use) and two bedaquiline use scenarios

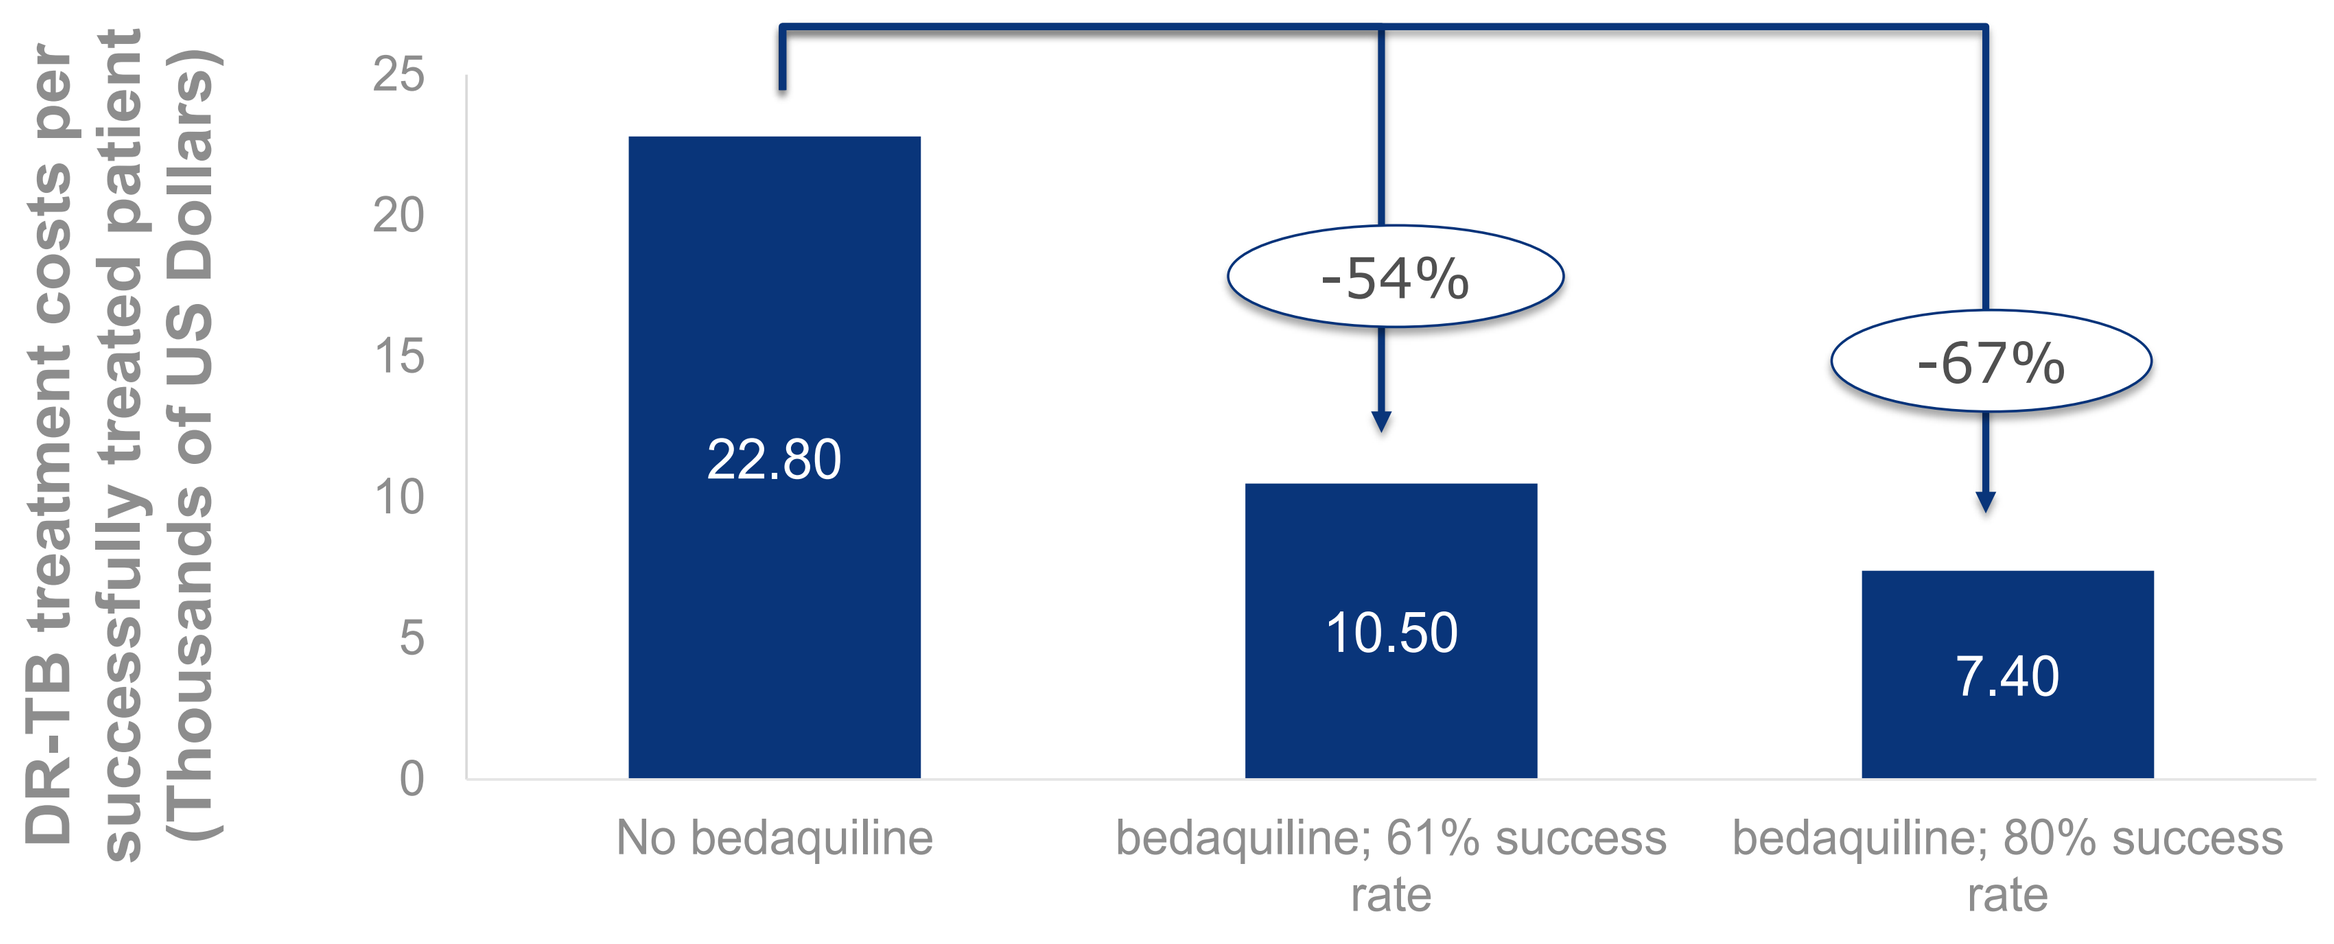
DR-TB treatment costs per successfully treated patient (Thousands of US Dollars)
0
5
10
15
20
25
22.80
-54%
10.50
-67%
7.40
No bedaquiline
bedaquiline; 61% success rate
bedaquiline; 80% success rate

- Increased use of bedaquiline is likely to bring about a reduction in the number of households affected catastrophically due to DR-TB. Under the two scenarios, in 2040, findings of this study reveal that the number of such families affected catadtrophically due to DR-TB will decrease from 43k in the baseline to 32k (−26%) in scenario 1 and 24k (−44%) in scenario 2 (Fig. 3).

Figure 3: Estimated number of catastrophically affected households in India in 2020-2040 under baseline (no bedaquiline use) and 2 bedaquiline use scenarios, YoY

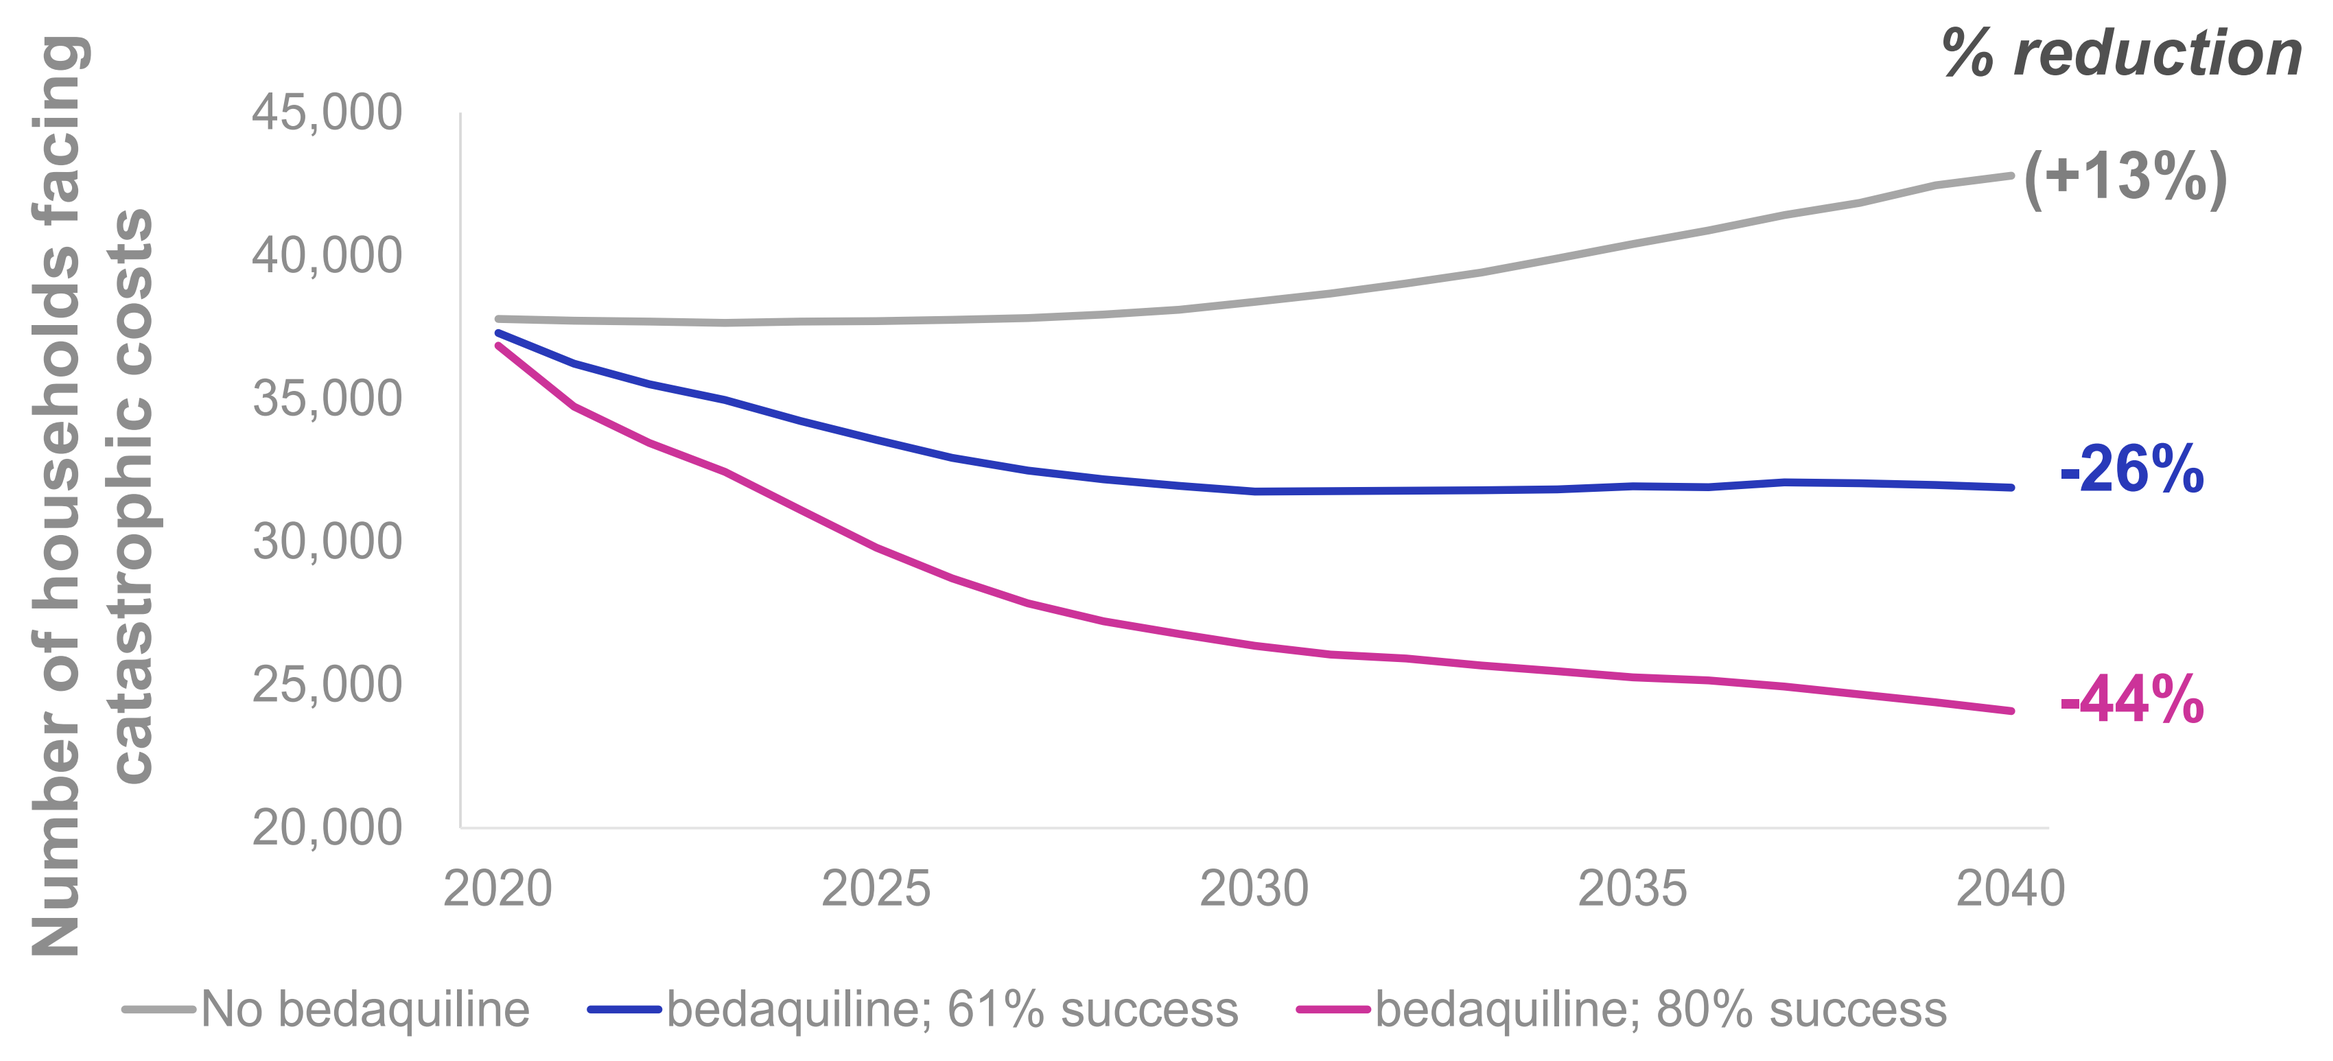
Number of households facing catastrophic costs
% reduction
45,000
40,000
35,000
30,000
25,000
20,000
2020
2025
2030
2035
2040
(+13%)
-26%
-44%
No bedaquiline
bedaquiline; 61% success
bedaquiline; 80% success

DISCUSSION

- Bedaquiline-containing regimens are expected to support realize 20-year direct cost savings of $7.6B and $9.3B for the two scenarios with treatment success rates of 61% and 80%, respectively, assuming a 95% adoption among first line DR-TB patients.
- In contrast, the documented lower success rates and high adverse event incidence rates associated with currently used injectable therapies translate into high rates of treatment discontinuation in India,[13] with the potential for continued disease transmission and perpetuation of the disease cycle,[14] resulting in missed opportunities to derive the documented economic benefits of effective tuberculosis control.
- The convenient oral dosing and proven effectiveness of bedaquiline can promote adherence and improve treatment outcomes, helping curb further disease transmission.
- At the patient level, lower costs are expected to be incurred by DR-TB patients treated with bedaquiline-containing regimens due to lower total drug costs, fewer adverse events, and fewer hospitalizations. At the societal level, benefits accrue from savings in direct drug costs as well as from reduced epidemiological burden. Together, these translate into significant economic benefits at the societal level over the 20-year horizon till 2040.
- The findings of this analysis have multiple public health, economic, and social implications, and support the long-term adoption of bedaquilne use as a first line treatment of DR-TB patients in India.
- The significantly reduced economic burden as a result of introduction of bedaquiline-containing regimens to treat DR-TB patients can free up resources for other measures to diagnose, test, treat, and manage DR-TB and DS-TB.

CONCLUSION

More widespread bedaquiline-based regimen adoption for the treatment of DR-TB in India will decrease the proportion of patients and families experiencing catastrophic costs over the current standard of care, and have a positive overall economic impact at both the individual and societal levels.

REFERENCES

1. World Health Organization. Global Tuberculosis Report 2018. Geneva, Switzerland: World Health Organization; 2018. Available from: https://apps.who.int/iris/bitstream/handle/10665/274453/9789241565646-eng.pdf?ua=1.
2. Barter DM, Agboola SO, Murray MB, Barnighausen T. Tuberculosis and poverty: the contribution of patient costs in sub-Saharan Africa--a systematic review. BMC Public Health. 2012;12:980.
3. Vassall A. Copenhagen Consensus Post-2015 Development Agenda: India Perspectives - Tuberculosis. Tewksbury, Massachusetts: Copenhagen Consensus Center; 2019 [Available from: https://www.copenhagenconsensus.com/sites/default/files/india_tuberculosis_resouce_packet.pdf.
4. Mullerpattan JB, Udwadia ZZ, Banka RA, Ganatra SR, Udwadia ZF. Catastrophic costs of treating drug resistant TB patients in a tertiary care hospital in India. Indian J Tuberc. 2019;66(1):87-91.
5. Diacon AH, Pym A, Grobusch MP, de los Rios JM, Gotuzzo E, Vasilyeva I, et al. Multidrug-resistant tuberculosis and culture conversion with bedaquiline. The New England journal of medicine. 2014;371(8):723-32.
6. Skrahina A, Hurevich H, Falzon D, Zhilevich L, Rusovich V, Dara M, et al. Bedaquiline in the multidrug-resistant tuberculosis treatment: Belarus experience. International journal of mycobacteriology. 2016;5 Suppl 1:S62-s3.
7. Guglielmetti L, Jaspard M, Le Du D, Lachatre M, Marigot-Outtandy D, Bernard C, et al. Long-term outcome and safety of prolonged bedaquiline treatment for multidrug-resistant tuberculosis. The European respiratory journal. 2017;49(3).
8. World Health Organization. WHO treatment guidelines for multidrug- and rifampicin-resistant tuberculosis: 2018 update (Pre-final text)2018 January 26, 2019. Available from: https://www.who.int/tb/publications/2018/WHO.2018.MDR-TB.Rx.Guidelines.prefinal.text.pdf.
9. Mehra M, Cossrow N, Kambili C, Underwood R, Makkar R, Potluri R. Assessment of tuberculosis burden in China using a dynamic disease simulation model. Int J Tuberc Lung Dis. 2013;17(9):1186-94.
10. Janssen Therapeutics. Sirturo® - Clinical Trials Titusville, New Jersey: Janssen Therapeutics; 2017 [Available from: https://www.sirturo.com/sirturo-clinical-trials.html.
11. Agnarson et al: Assessment of Long-Term Impact of Bedaquiline-Containing Regimens on Burden of Drug-Resistant Tuberculosis in India, a State Transition Model Analysis. Conference: International Society for Medical Decision Making, Abstract #12548
12. Prasanna T, et al. Catastrophic costs of tuberculosis care: a mixed methods study from Puducherry, India. Glob Health Action. 2018;11(1):1477493. doi:10.1080/16549716.2018.1477493.
13. Sarin R, Behera D, Khanna A, Singh V, Narang P, Deepak TS. Second-line injectable induced ototoxicity in drug resistant tuberculosis: A systematic review of Indian studies. Indian J Tuberc. 2019;66(2):279-87.
14. Dheda K, Gumbo T, Maartens G, Dooley KE, McNerney R, Murray M, et al. The epidemiology, pathogenesis, transmission, diagnosis, and management of multidrug-resistant, extensively drug-resistant, and incurable tuberculosis. Lancet Respir Med. 2017.

ACKNOWLEDGMENTS

This study was funded by Johnson & Johnson Global Public Health. Assistance in the creation of this poster was provided by Amandeep, an employee of SmartAnalyst India Pvt. Ltd.

ISPOR Europe 2019 | 2-6 November 2019 | Copenhagen, Denmark