A Multilevel Access Evidence Framework for Innovative Technologies Specific to Haematological Malignancies

Harrison KL[1], O'Rourke D[1], Chevrou-Severac H[2], Gaudig M[3], Garcia S[4], Collados C[4], Wirtz D[5], Conge M[6], Lee A[7], Jonsson P[1]

[1]NICE UK, [2]Celgene International [3]Janssen-Cilag Germany, [4]AEMPS Spain, [5]BfArM Germany, [6]AbbVie , [7]Novartis UK

Objective – Making innovative treatments available across the European Union is a complex process due to differing evidence requirements, assessment methods and value judgements by HTA agencies. The HARMONY Alliance is a big data for better outcomes project of the Innovative Medicines Initiative (IMI). HARMONY is a public-private partnership with over 90 organisations from 22 European countries with varying expertise in evidence development strategies to support new treatments and indications.
We aimed to produce a guide for the evidence required for market access and coverage/ reimbursement ("access evidence") focussing on challenges facing innovative haematological malignancies (HM) drugs and the jurisdictional differences across Europe.

Methods and results – We developed a multilevel framework based on methodology guidelines, interviews and previous submission reports and documentation from key selected countries: France, Spain, Norway, Sweden, England, Ireland, Germany, Italy, Poland, Estonia and Slovenia and EU wide (EMA, EUnetHTA) and umbrella payer organisations.

Level 1: The Market Access Landscape Cyclopaedia

Interviews, response rates were 70% (19/27) with HTA agencies (n=13), regulators (n=6) and payers (n=2) focusing on the themes detailed in diagram 1 along with methodology guidance and pragmatic literature searches forming the basis of the cyclopaedia.
This level is designed to act as a signposting tool to raise awareness of the regulatory/HTA and payer landscape and to present terminology, highlighting HM specific outcome requirements where available. Additionally this level has country specific information pages /dash boards with links to methodology and guidance documents at national level HTA, payers, regulatory bodies, service provision and patient co-pays.

Level 2: HM Outcome and Evidence Inventory

Consists of a series of easy access dashboards incorporating domains and topics from the HTA Core Model® including safety, clinical effectiveness, cost and economic evaluation and ethical and organisational aspects.
This level is populated from data extracted from 77 previous reimbursement decisions on 12 HM products across 9 HTAs. The framework highlights key differences/requirements and evidence use by both HTA and disease indication, and how these may impact on recommendations for reimbursement. Each series of dashboards includes summary text placing the findings in context with current literature and HTA method guidelines.

Practical implications – The freely available access evidence framework is anticipated to have several benefits for HARMONY and HM researchers. Developed as a unique resource for this clinical area it provides a concise point of reference with key learnings and requirements for HM evidence generation across the EU. It allows a direct comparison across differing agencies on the acceptability and use of evidence for key markets and could potentially increase the efficiency in generating and preparing relevant access evidence but also of predictability of HTA outcomes.

Resources and data underpinning the framework

Diagram 1: Survey, interviews and web based searches with HTA/regulators and payers

- Guidance on preferred outcomes
- Outcome preferences (including tools and acceptance of surrogacy
- Outcomes use (economic evaluation, perspective, modelling)
- Selection of outcomes
- Acceptability of real-world evidence or big data
- HM specific questions

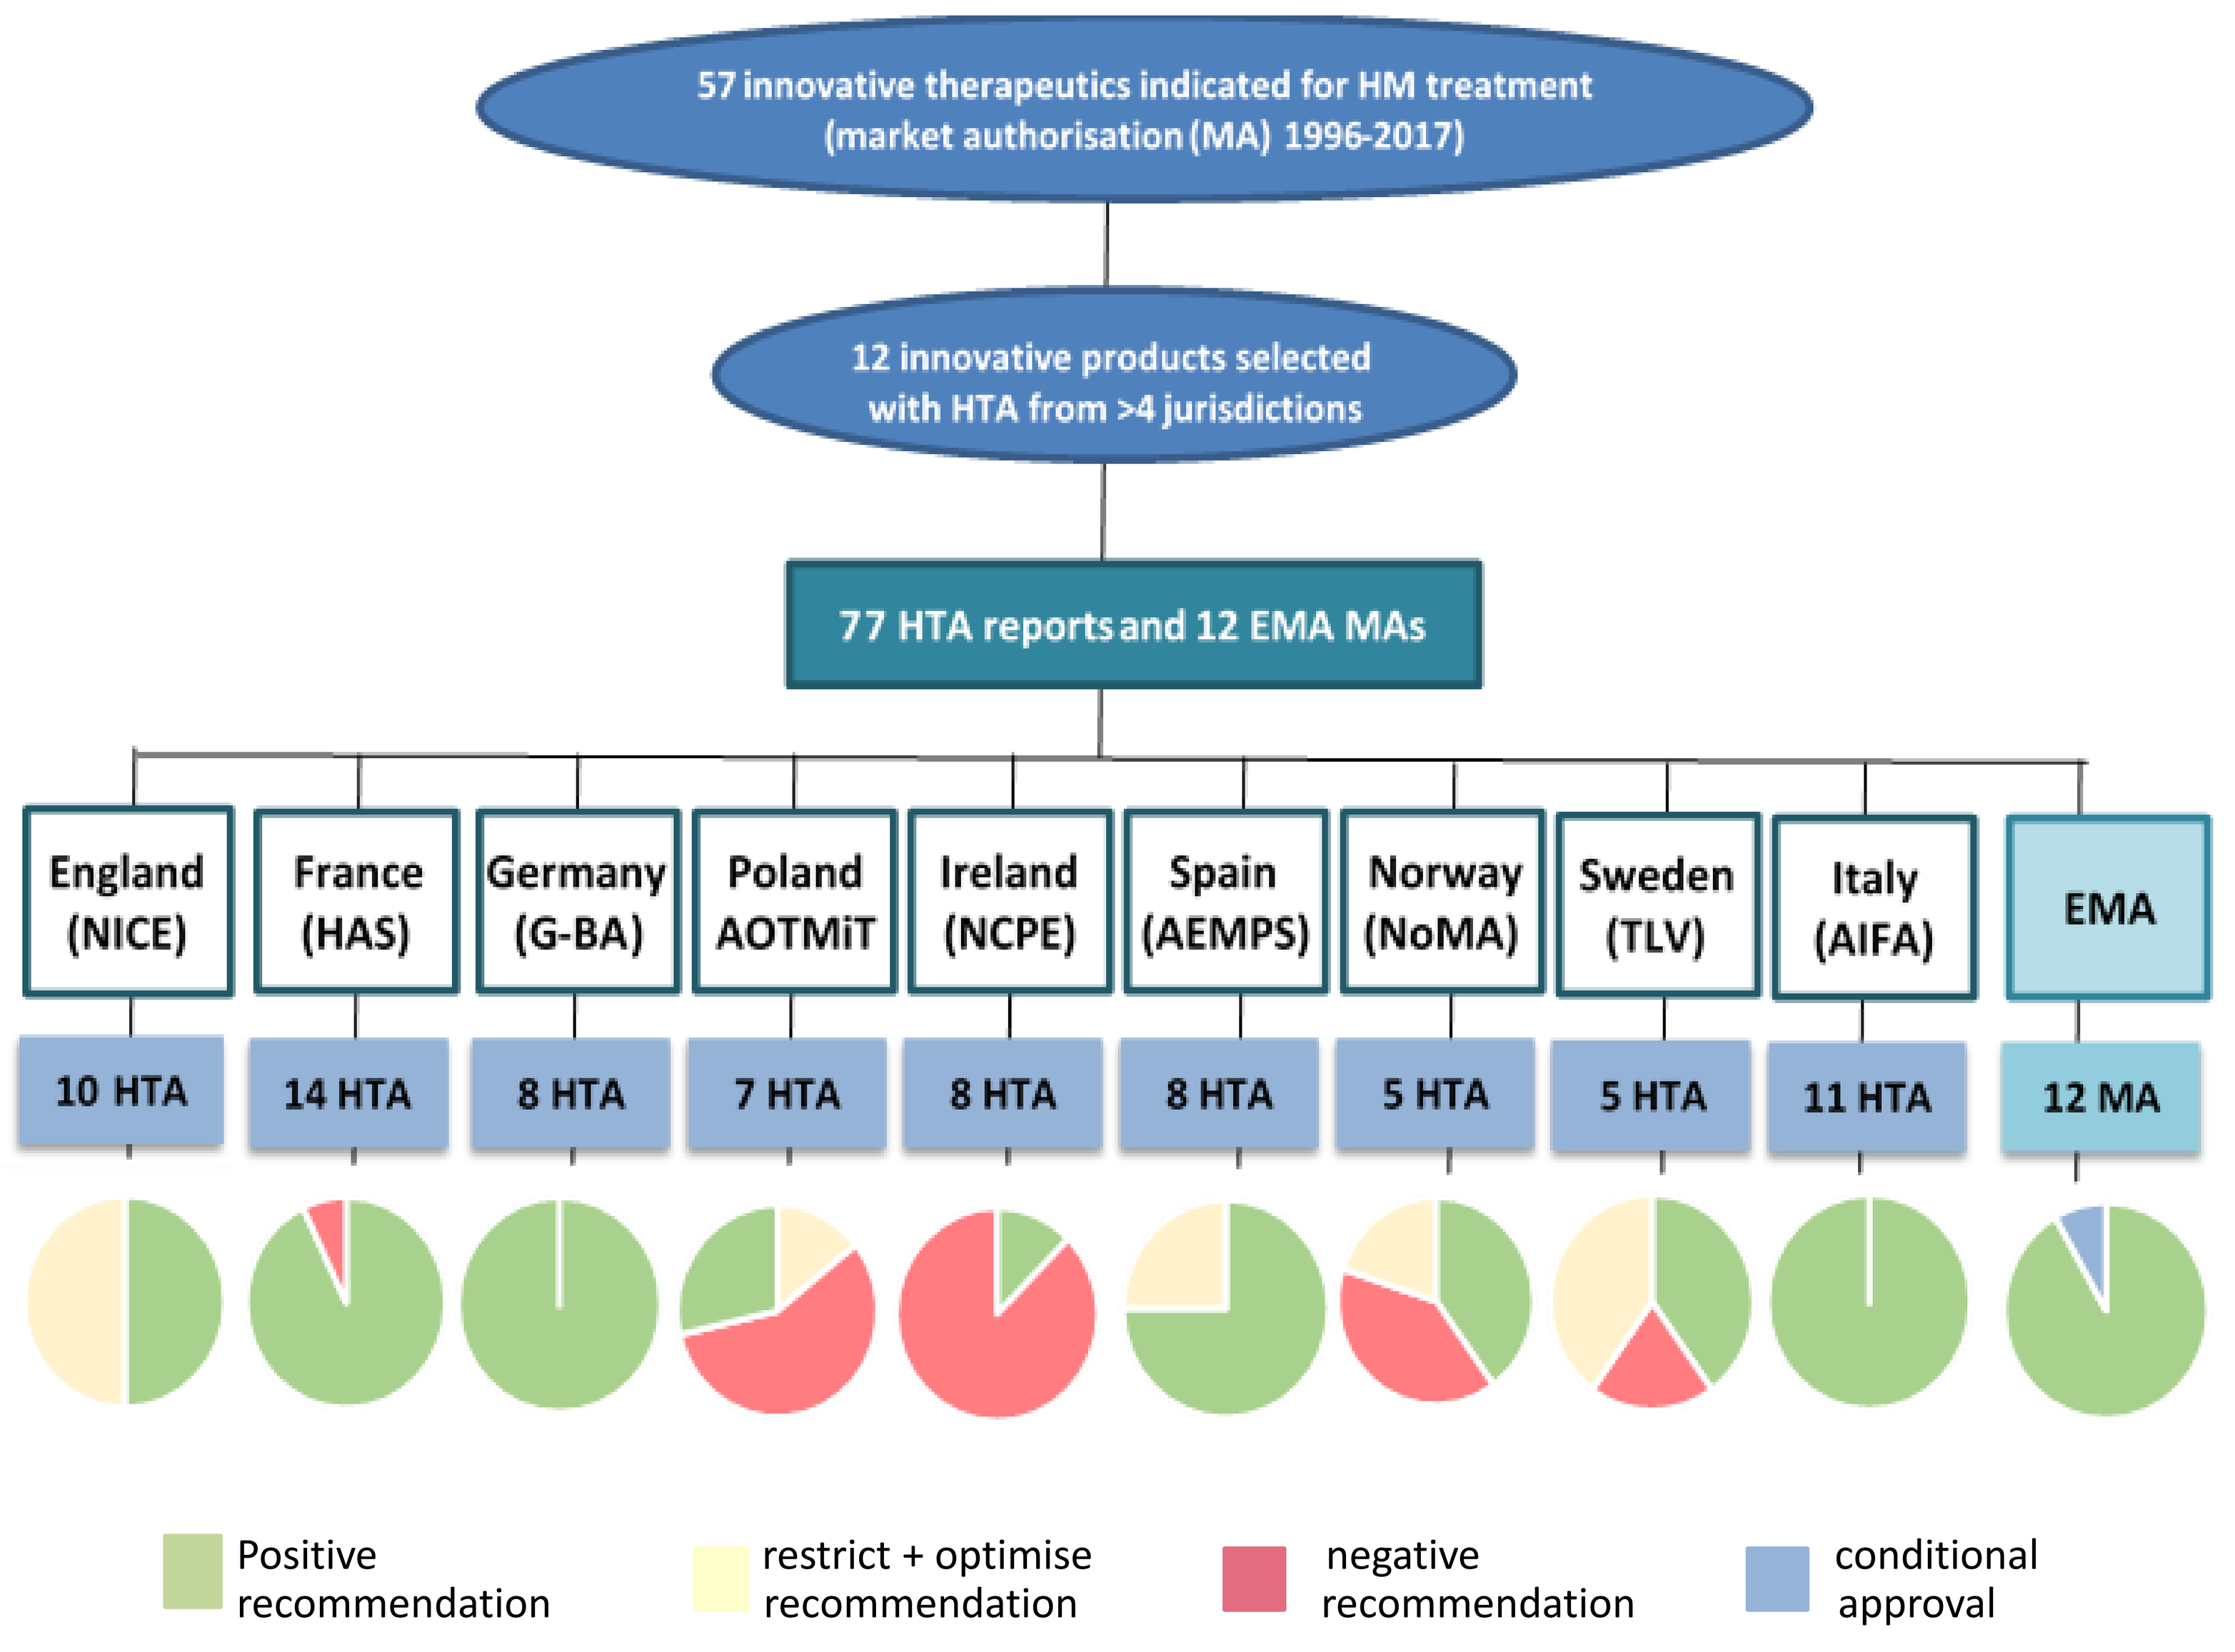
57 innovative therapeutics indicated for HM treatment (market authorisation (MA) 1996-2017)
12 innovative products selected with HTA from >4 jurisdictions
77 HTA reports and 12 EMA MAs
England (NICE)
France (HAS)
Germany (G-BA)
Poland AOTMiT
Ireland (NCPE)
Spain (AEMPS)
Norway (NoMA)
Sweden (TLV)
Italy (AIFA)
EMA
10 HTA
14 HTA
8 HTA
7 HTA
8 HTA
8 HTA
5 HTA
5 HTA
11 HTA
12 MA
Positive recommendation
restrict + optimise recommendation
negative recommendation
conditional approval

HM Access Evidence Framework & illustrative dashboards

Level 1: The Market Access Landscape Cyclopaedia

Overall EU pharmaceutical market access process

Regulator	HTA
National / EU	National / EU
Process and Procedures	Methods
Methods	Ontologies
Outcome preference: Clinical, PROs	Outcome preference: Clinical, PROs

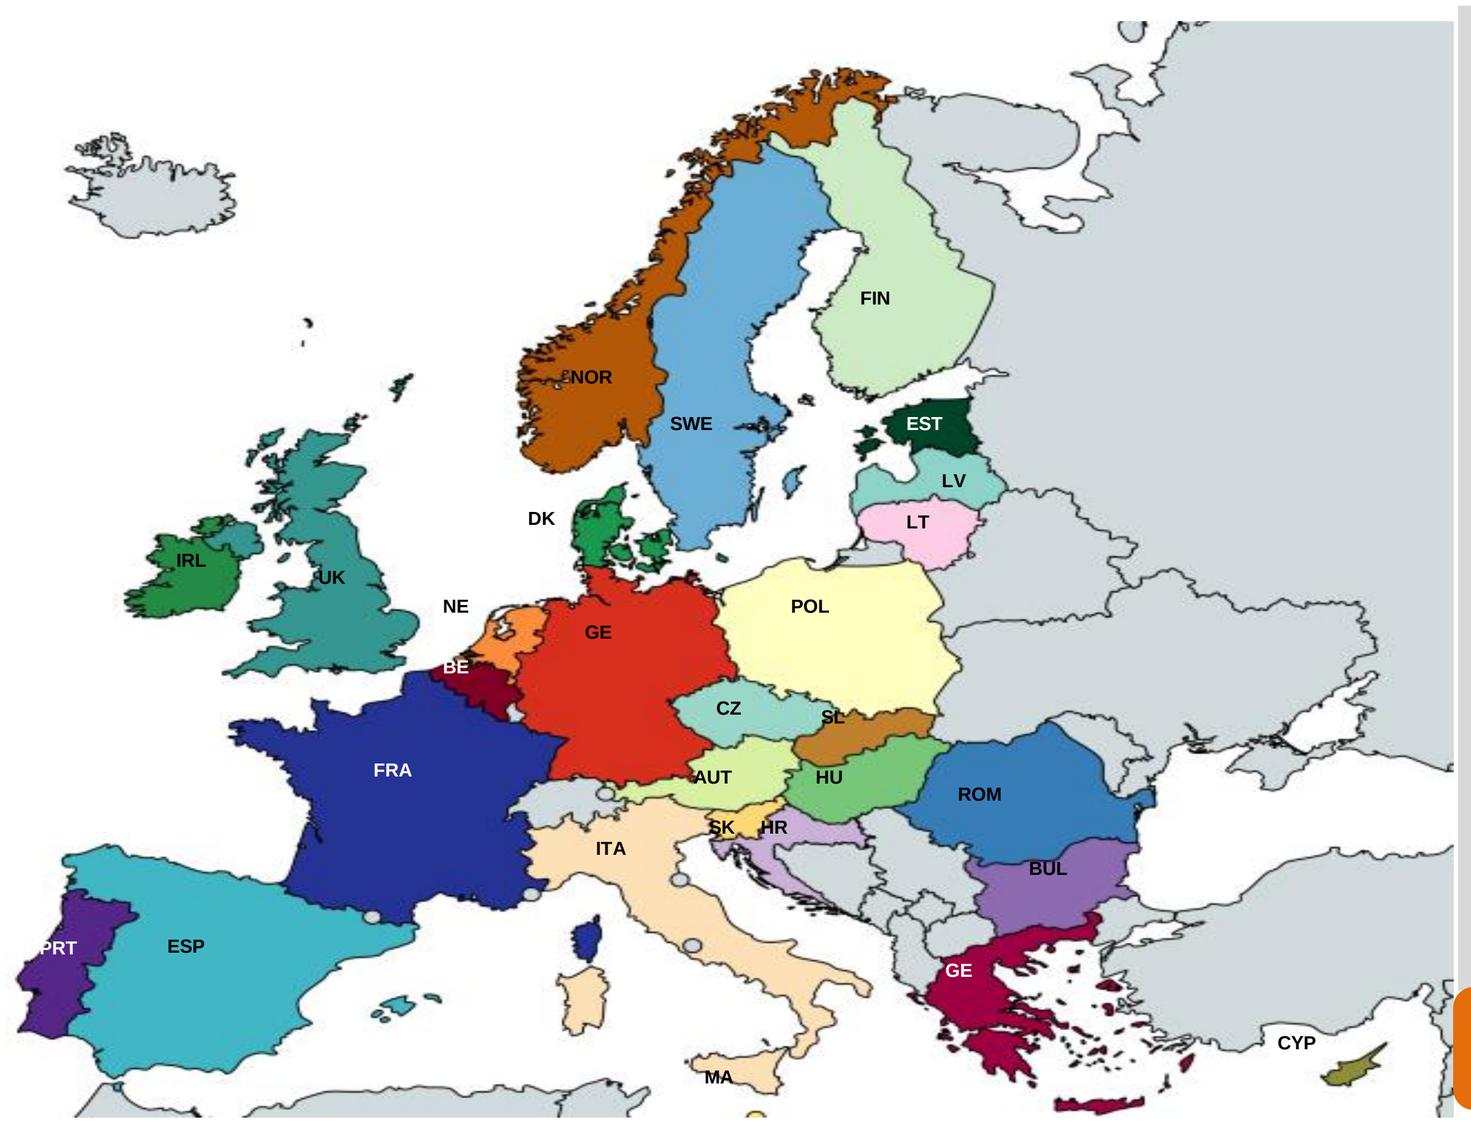
FIN
NOR
SWE
EST
LV
LT
DK
IRL
UK
NE
BE
GE
POL
CZ
FRA
AUT
HU
SK
HR
ROM
BUL
ITA
ESP
PRT
GE
MA
CYP

Austria, Belgium, Bulgaria, Croatia, Cyprus, Denmark, Estonia, Finland, France, Germany, Greece, Hungary, Ireland, Italy, Czech Republic, Latvia, Lithuania, Luxembourg, Malta, Netherlands, Portugal, Romania, Slovakia, Slovenia, Spain, Sweden, Norway, UK, Poland

Glossary

Level 2: HM Outcome and Evidence Inventory

Evidence for clinical effectiveness/ efficacy — **Alignment of EMA and HTA**

Outcome Selection
- Clinical effectiveness outcomes — *Key questions*
- Patient reported outcomes — *Key questions*
- Adverse events/safety — *Key questions*

Study Design
- Clinical trials phase — *Key questions*
- Trial Comparator — *Key questions*
- Use of real-world evidence — *Key questions*

Analysis and other considerations

Analytical Options
- Indirect treatment comparisons — *Key questions*
- Subgroup analysis — *Key questions*

HTA Approaches
- Ethical/social considerations — *Key questions*
- Cost and economic evaluation — *Key questions*

Methods & limitations | **Guidance Documents**

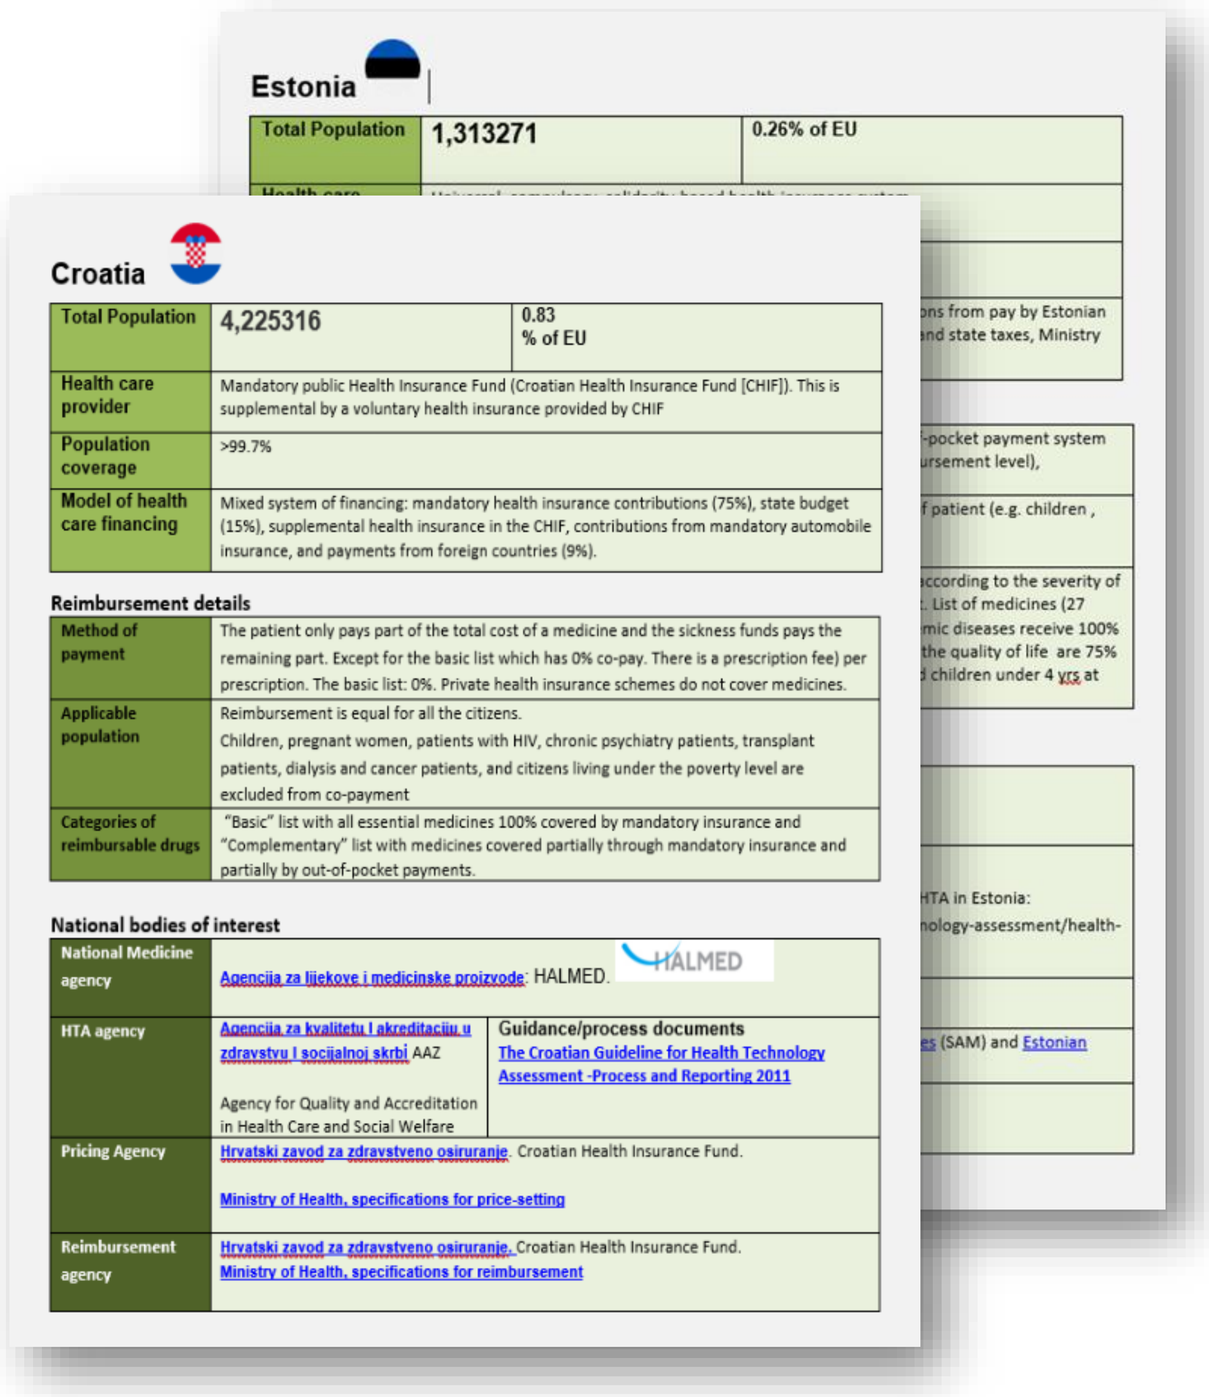
Estonia
Total Population
1,313271
0.26% of EU
Croatia
Total Population
4,225316
0.83 % of EU
Health care provider
Population coverage
>99.7%
Model of health care financing
Reimbursement details
Method of payment
Applicable population
Categories of reimbursable drugs
National bodies of interest
National Medicine agency
HTA agency
Pricing Agency
Reimbursement agency

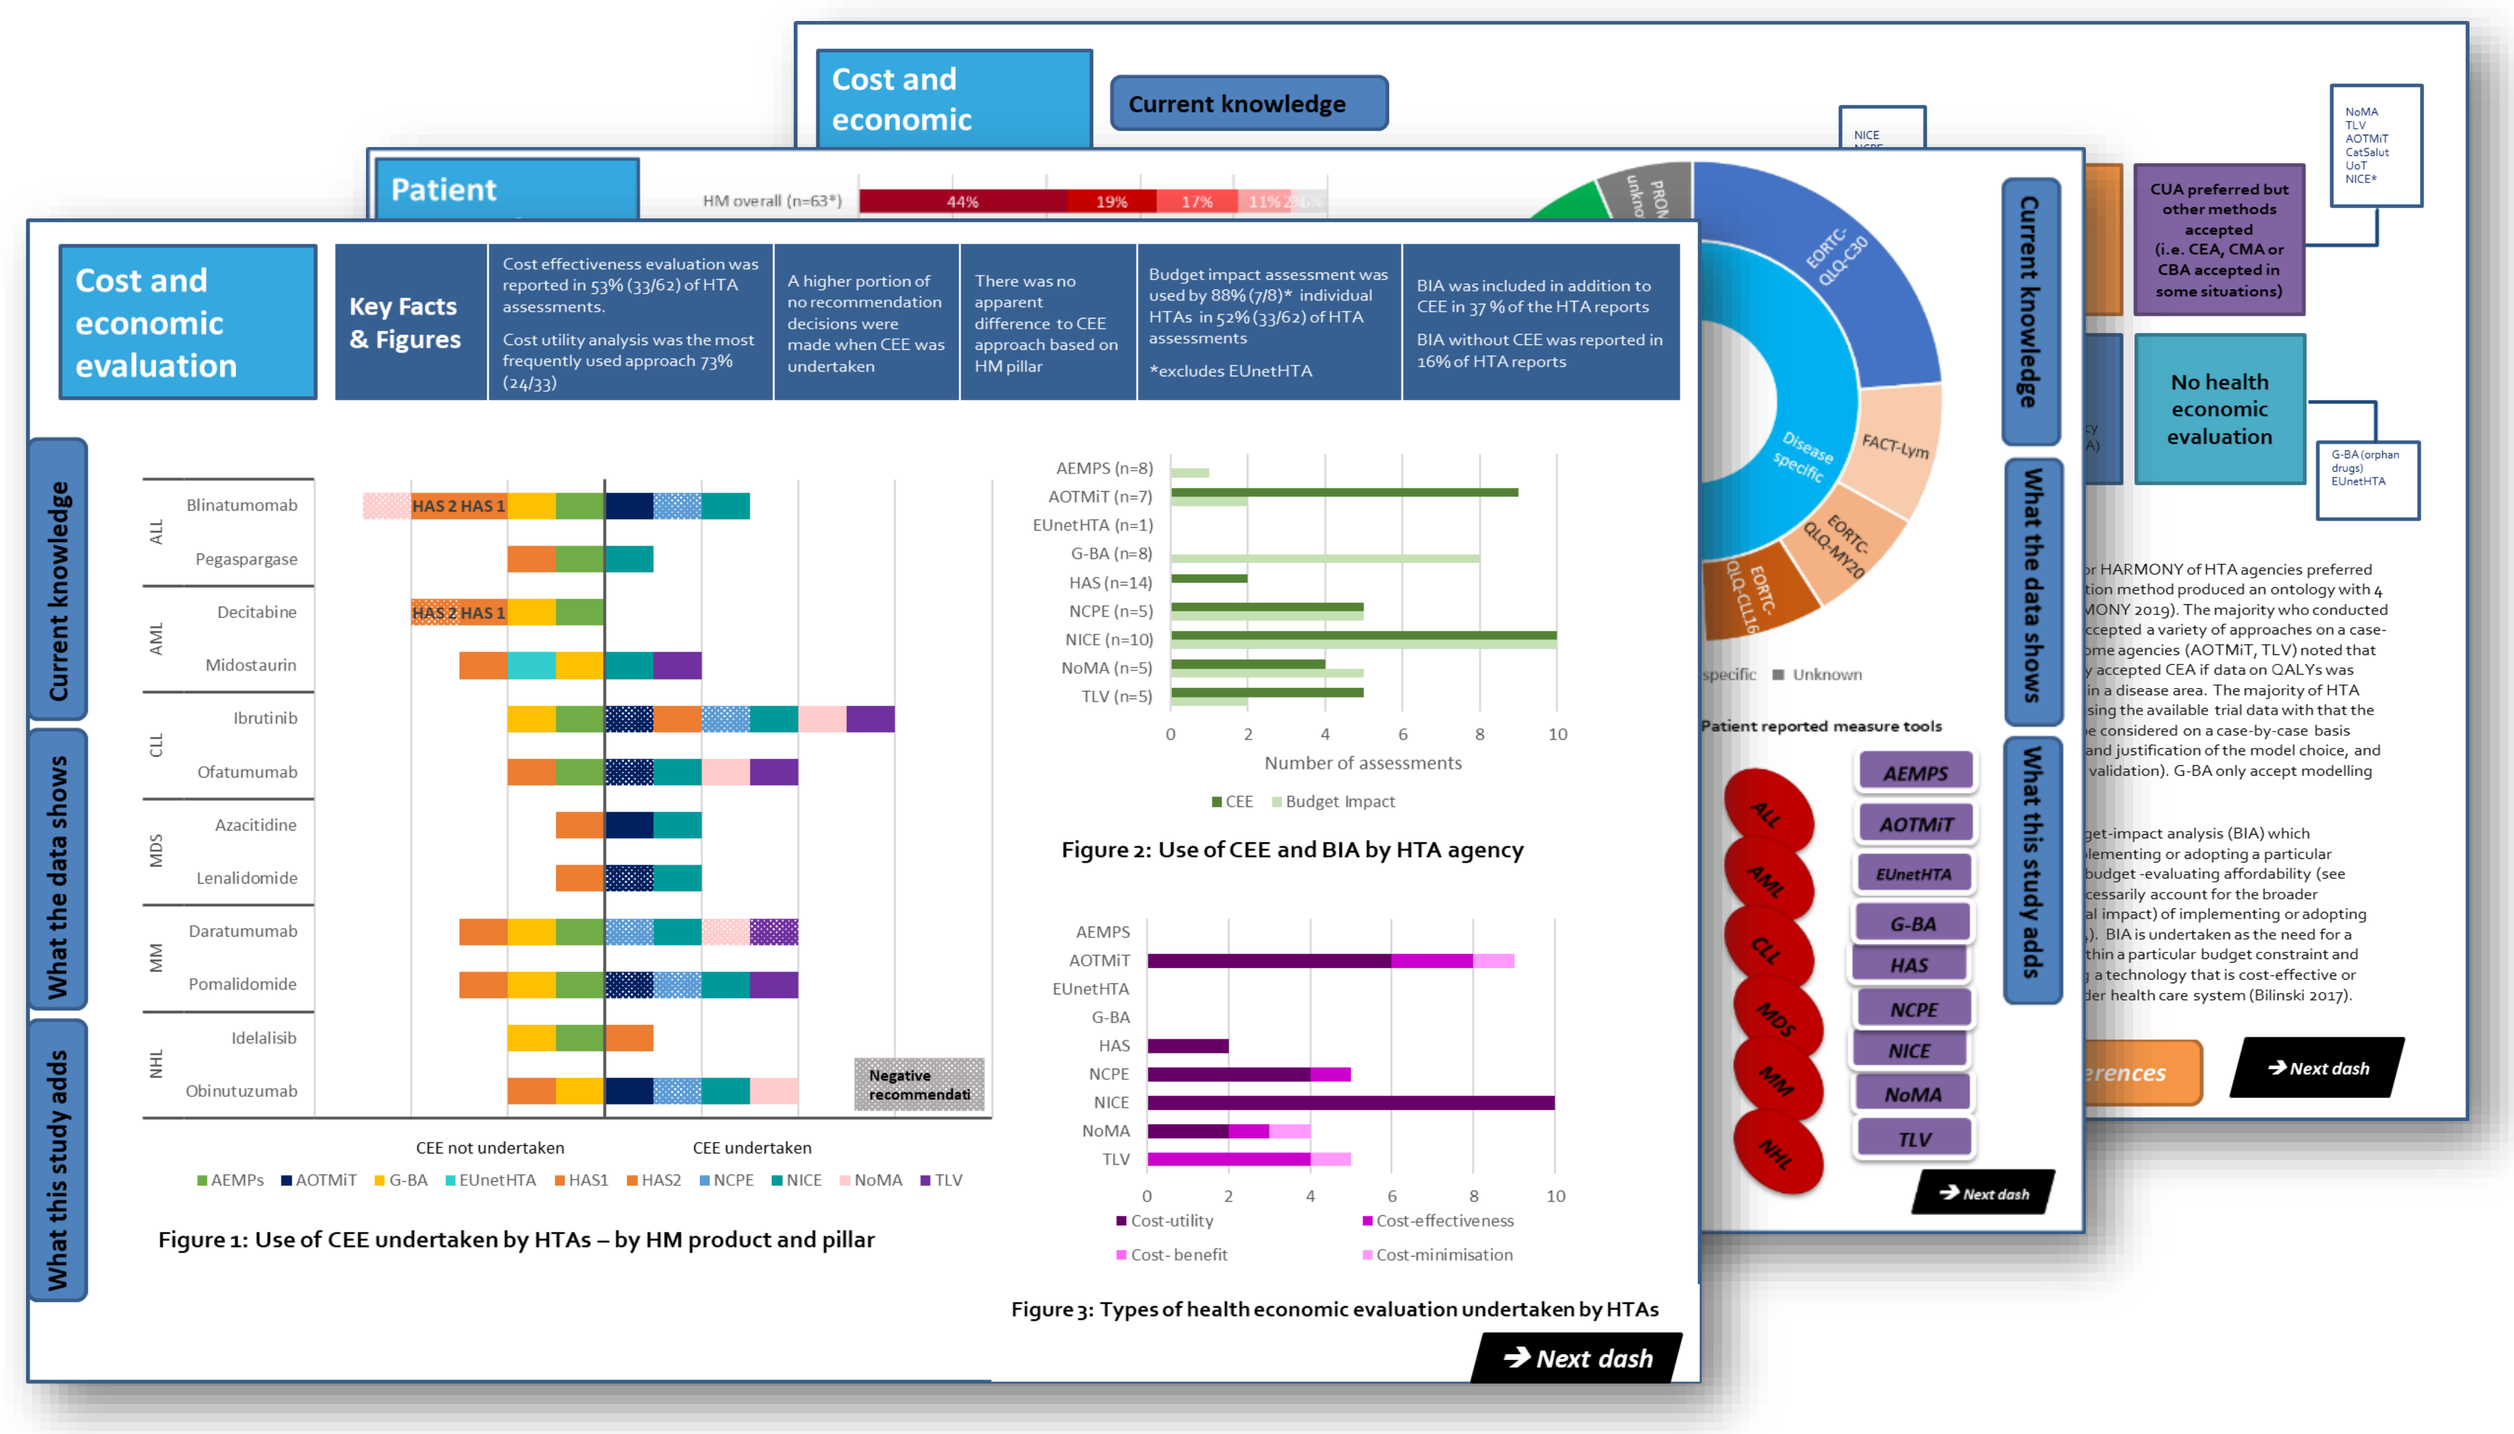
Cost and economic evaluation
Key Facts & Figures
Current knowledge
What the data shows
What this study adds
Figure 1: Use of CEE undertaken by HTAs – by HM product and pillar
Figure 2: Use of CEE and BIA by HTA agency
Figure 3: Types of health economic evaluation undertaken by HTAs
Next dash
Patient
No health economic evaluation

The HARMONY Alliance is funded through the Innovative Medicines Initiative (IMI), Europe's largest public-private initiative aiming to speed up the development of better and safer medicines for patients. HARMONY has received funding from IMI 2 Joint Undertaking and is listed under grant agreement No. 116026. This Joint Undertaking receives support from the European Union's Horizon 2020 Research and Innovation Programme and the European Federation of Pharmaceutical Industries and Associations (EFPIA). **www.harmony-alliance.eu** | HARMONY ©October 2019