SYSTEMATIC REVIEW AND NETWORK META-ANALYSIS COMPARING NETAKIMAB WITH OTHER BIOLOGIC AGENTS USED TO TREAT ADULT PATIENTS WITH MODERATE-TO-SEVERE PLAQUE PSORIASIS IN RUSSIA

D. G. Tolkacheva[1], V. D. Sokolova[2], V. V. Mladov[3]

[1]University of Oxford, BIOCAD, St. Petersburg, Russia;
[2]BIOCAD, St. Petersburg, Russia; [3]BIOCAD, St. Petersburg, Russia

INTRODUCTION

- Psoriasis is a chronic, non-communicable, inflammatory skin disease with an extensive emotional and psychological impact on patients.
- While multiple treatment options are available, biological therapy is the treatment of choice for moderate-to-severe plaque psoriasis when not responding to other systemic therapies.
- Multiple randomised controlled trials (RCTs) have compared the efficacy of biologics with placebo. However, the relative effectiveness of these treatments is not straightforward, due to the lack of head-to-head evidence.

OBJECTIVE

To compare the efficacy and safety of a new biologic — netakimab and biologic agents, approved and available in Russia (secukinumab, ustekinumab, infliximab, adalimumab, etanercept, certolizumab pegol, and ixekizumab) in adult patients with moderate-to-severe plaque psoriasis.

MATERIALS & METHODS

We performed the literature search in PubMed database, Cochrane library (Embase section only) and e-library, as well as in reference lists of original articles to identify publications of phase II, III, and IV RCTs, evaluating the efficacy and safety of biologics for the treatment of moderate-to-severe psoriasis in adult patients. We assessed eligible RCTs for the risk of bias and possible sources of heterogeneity. Finally, we conducted network meta-analysis using fixed effects model to synthesize evidence obtained from selected studies.

PRISMA 2009 Flow Diagram

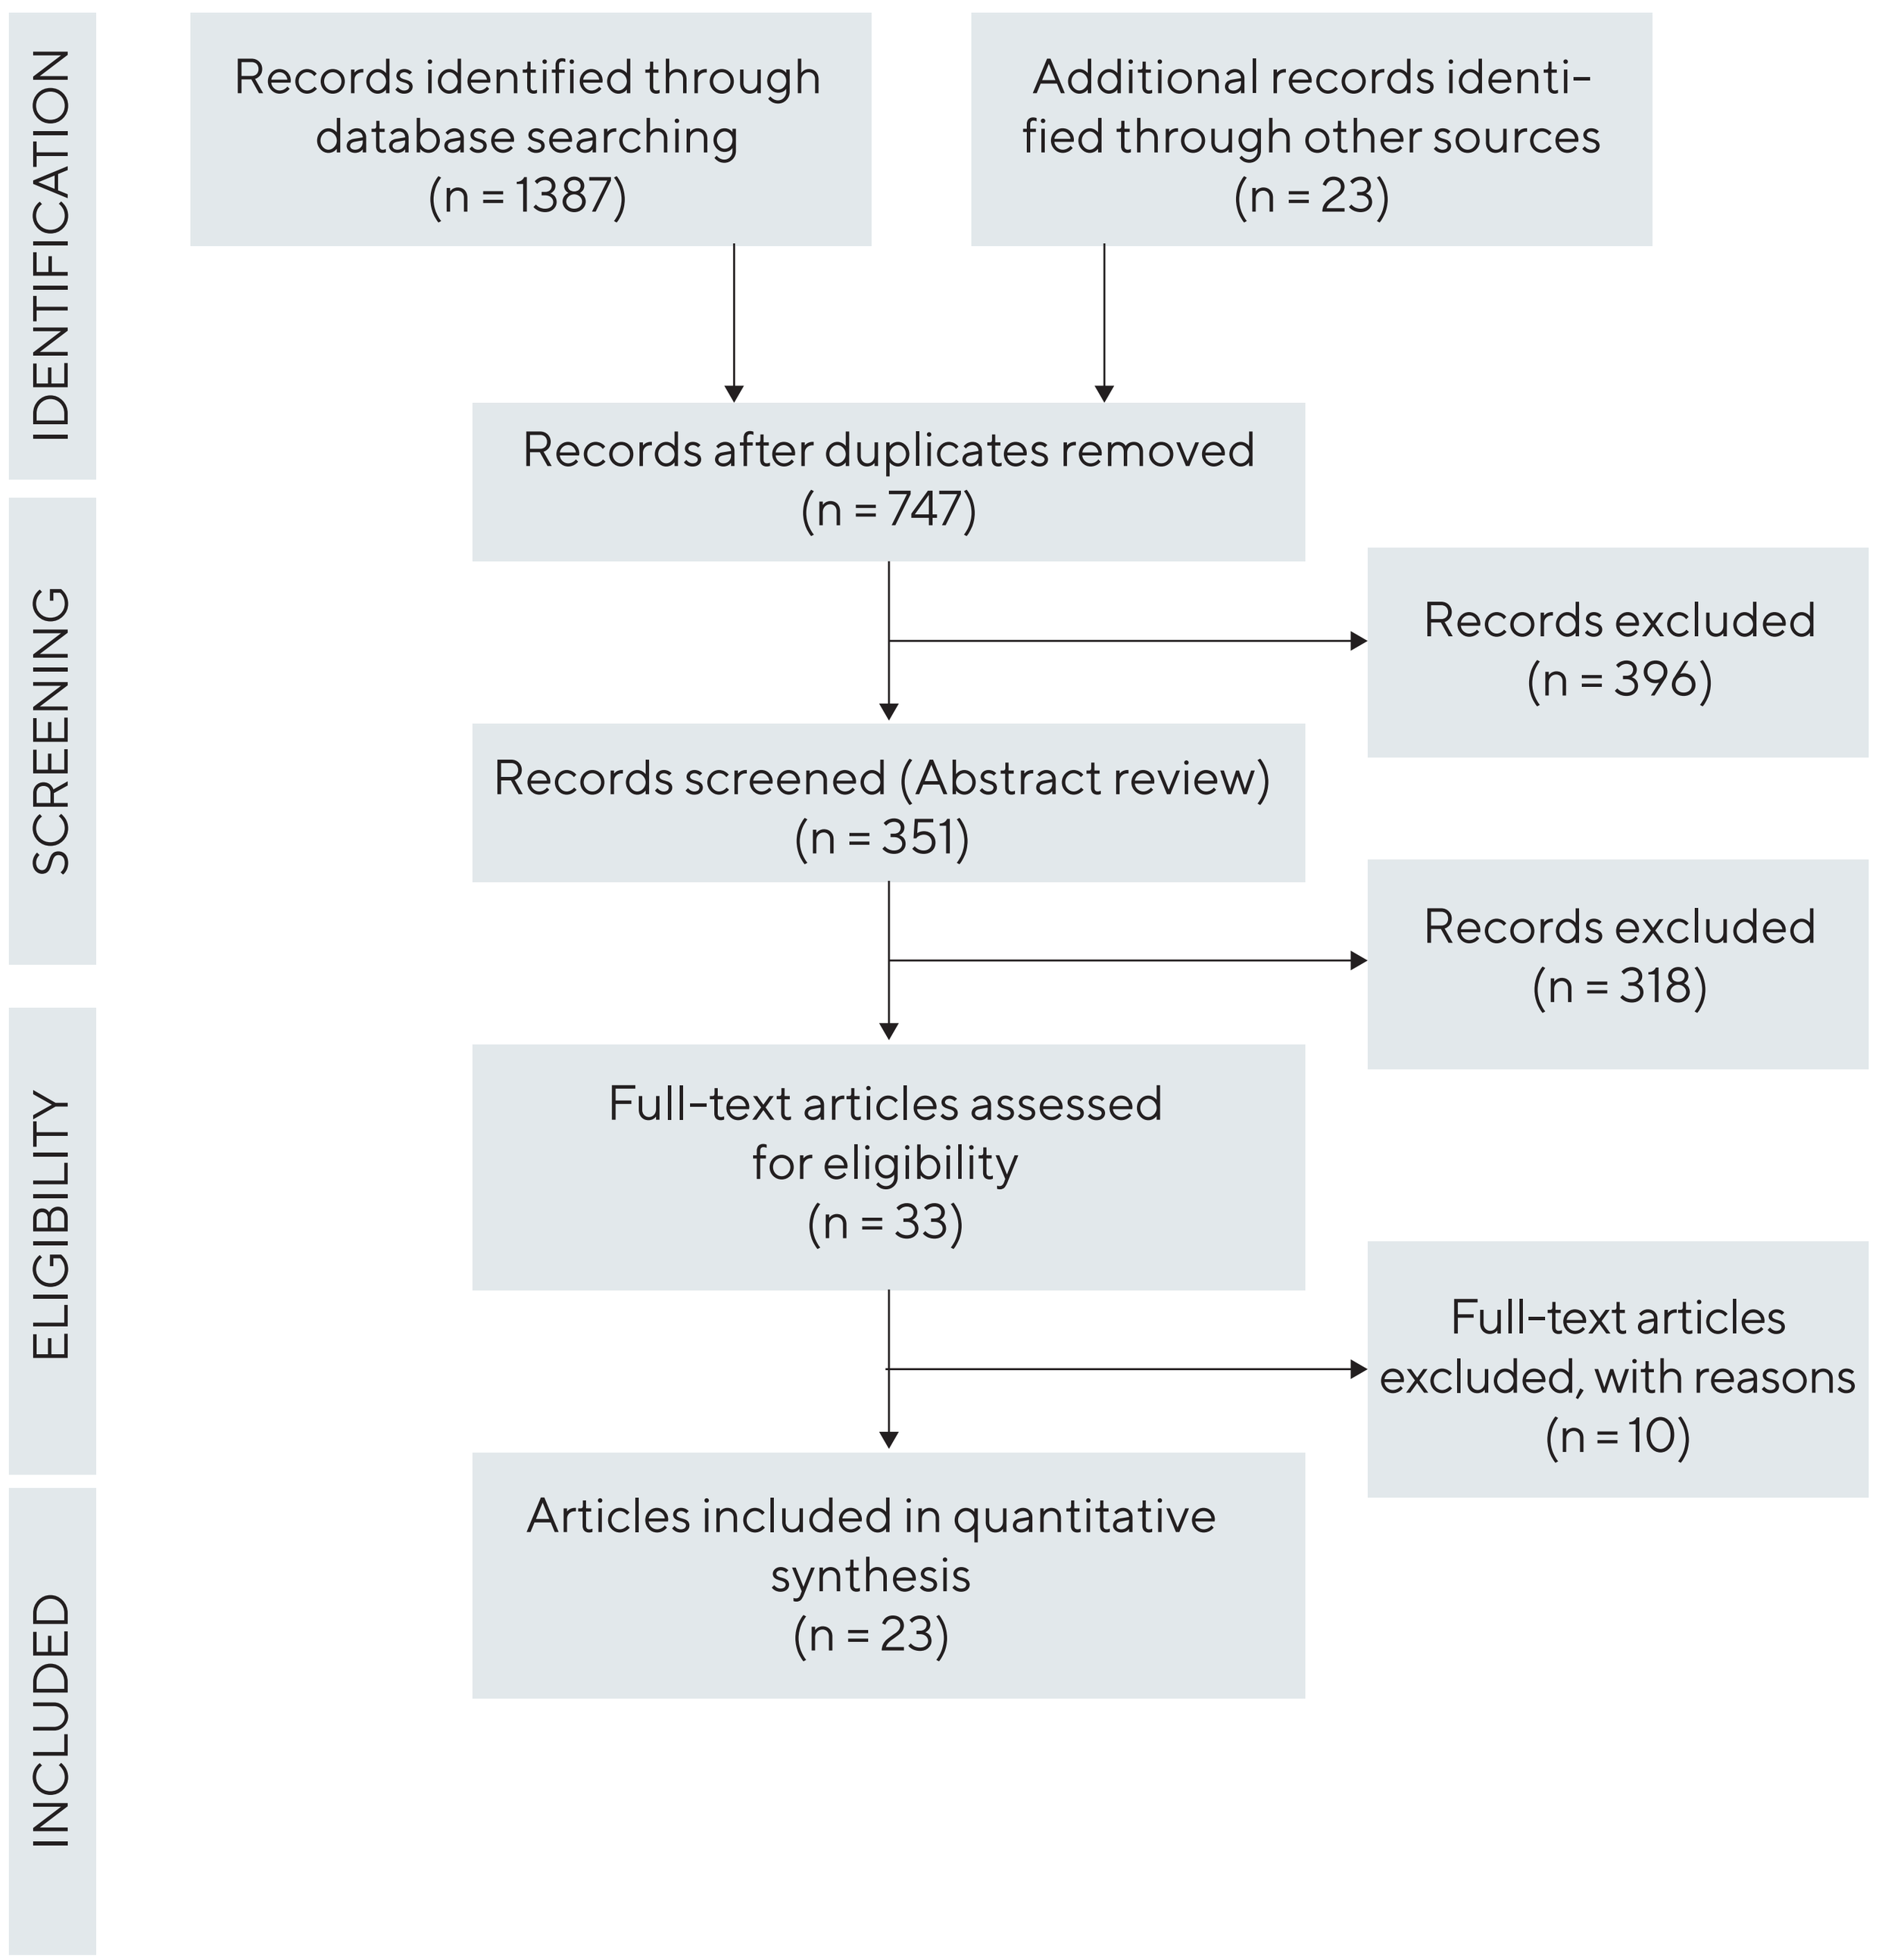

IDENTIFICATION
SCREENING
ELIGIBILITY
INCLUDED
Records identified through database searching (n = 1387)
Additional records identified through other sources (n = 23)
Records after duplicates removed (n = 747)
Records excluded (n = 396)
Records screened (Abstract review) (n = 351)
Records excluded (n = 318)
Full-text articles assessed for eligibility (n = 33)
Full-text articles excluded, with reasons (n = 10)
Articles included in quantitative synthesis (n = 23)

From: Moher D, Liberati A, Tetzlaff J, Altman DG. The PRISMA Group (2009). Preferred Reporting Items for Systematic Reviews and MetaAnalyses: The PRISMA Statement. PLoS Med 6(6): e1000097. doi: 10.1371/journal.pmed1000097

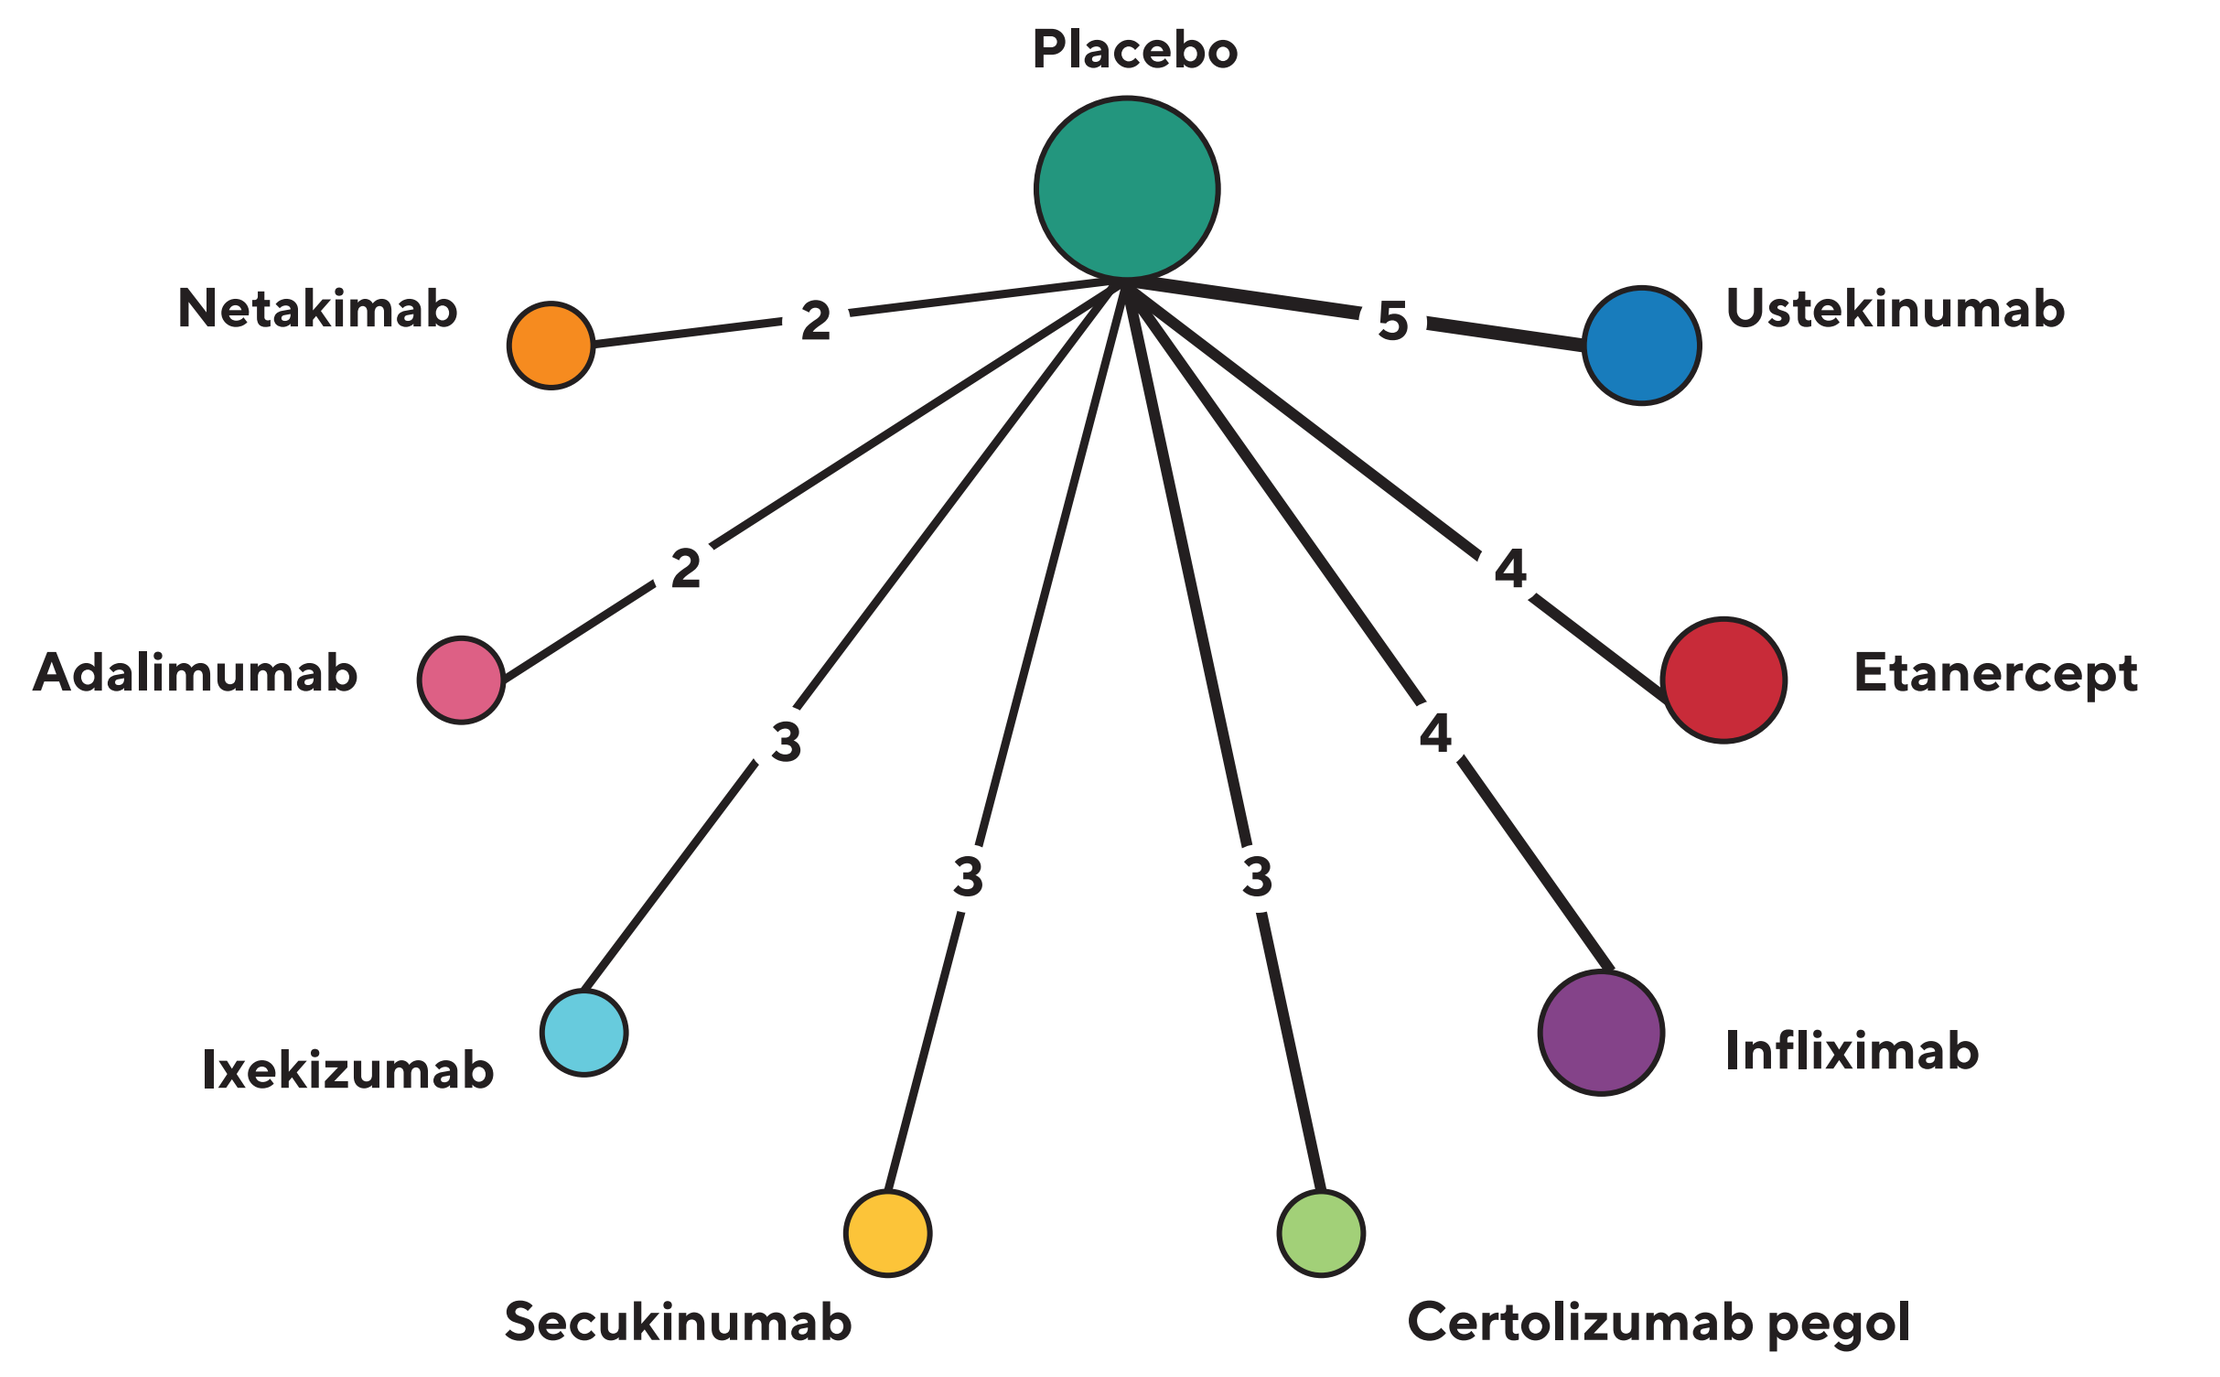

Placebo
Netakimab
2
Ustekinumab
5
Adalimumab
2
Etanercept
4
3
4
3
3
Ixekizumab
Infliximab
Secukinumab
Certolizumab pegol

RESULTS

Overall, 23 articles were selected for quantitative and qualitative synthesis. Network meta-analysis showed that netakimab is significantly more effective than adalimumab, etanercept, ustekinumab, and certolizumab, but there was no significant difference in effectiveness between netakimab, secukinumab, infliximab and ixekizumab. In terms of safety, we found no statistically significant difference between all the interventions, except for etanercept, which was associated with lower safety profile.

EFFECTIVENESS ANALYSIS

- Effectiveness was determined by the proportion of patients achieving PASI75 at week 12 of the treatment.
- Netakimab is statistically significantly more effective than adalimumab (odds ratio (OR): 7.19, 95% credible interval (CrI): 1.56-59.22), etanercept (OR: 10.78, 95% CrI: 2.65-83.04), ustekinumab (OR: 4.17, 95% CrI: 1.03-32.12) and certolizumab (OR: 5.63, 95% CrI: 1.21-46, 01).
- There are no statistically significant differences in effectiveness of netakimab compared to infliximab (OR: 1.14, 95% CrI: 0.22-9.49), secukinumab (OR: 2.40, 95% CrI: 0.57 -18.62) and ixekizumab (OR: 1.21, 95% CrI: 0.30-9.36).

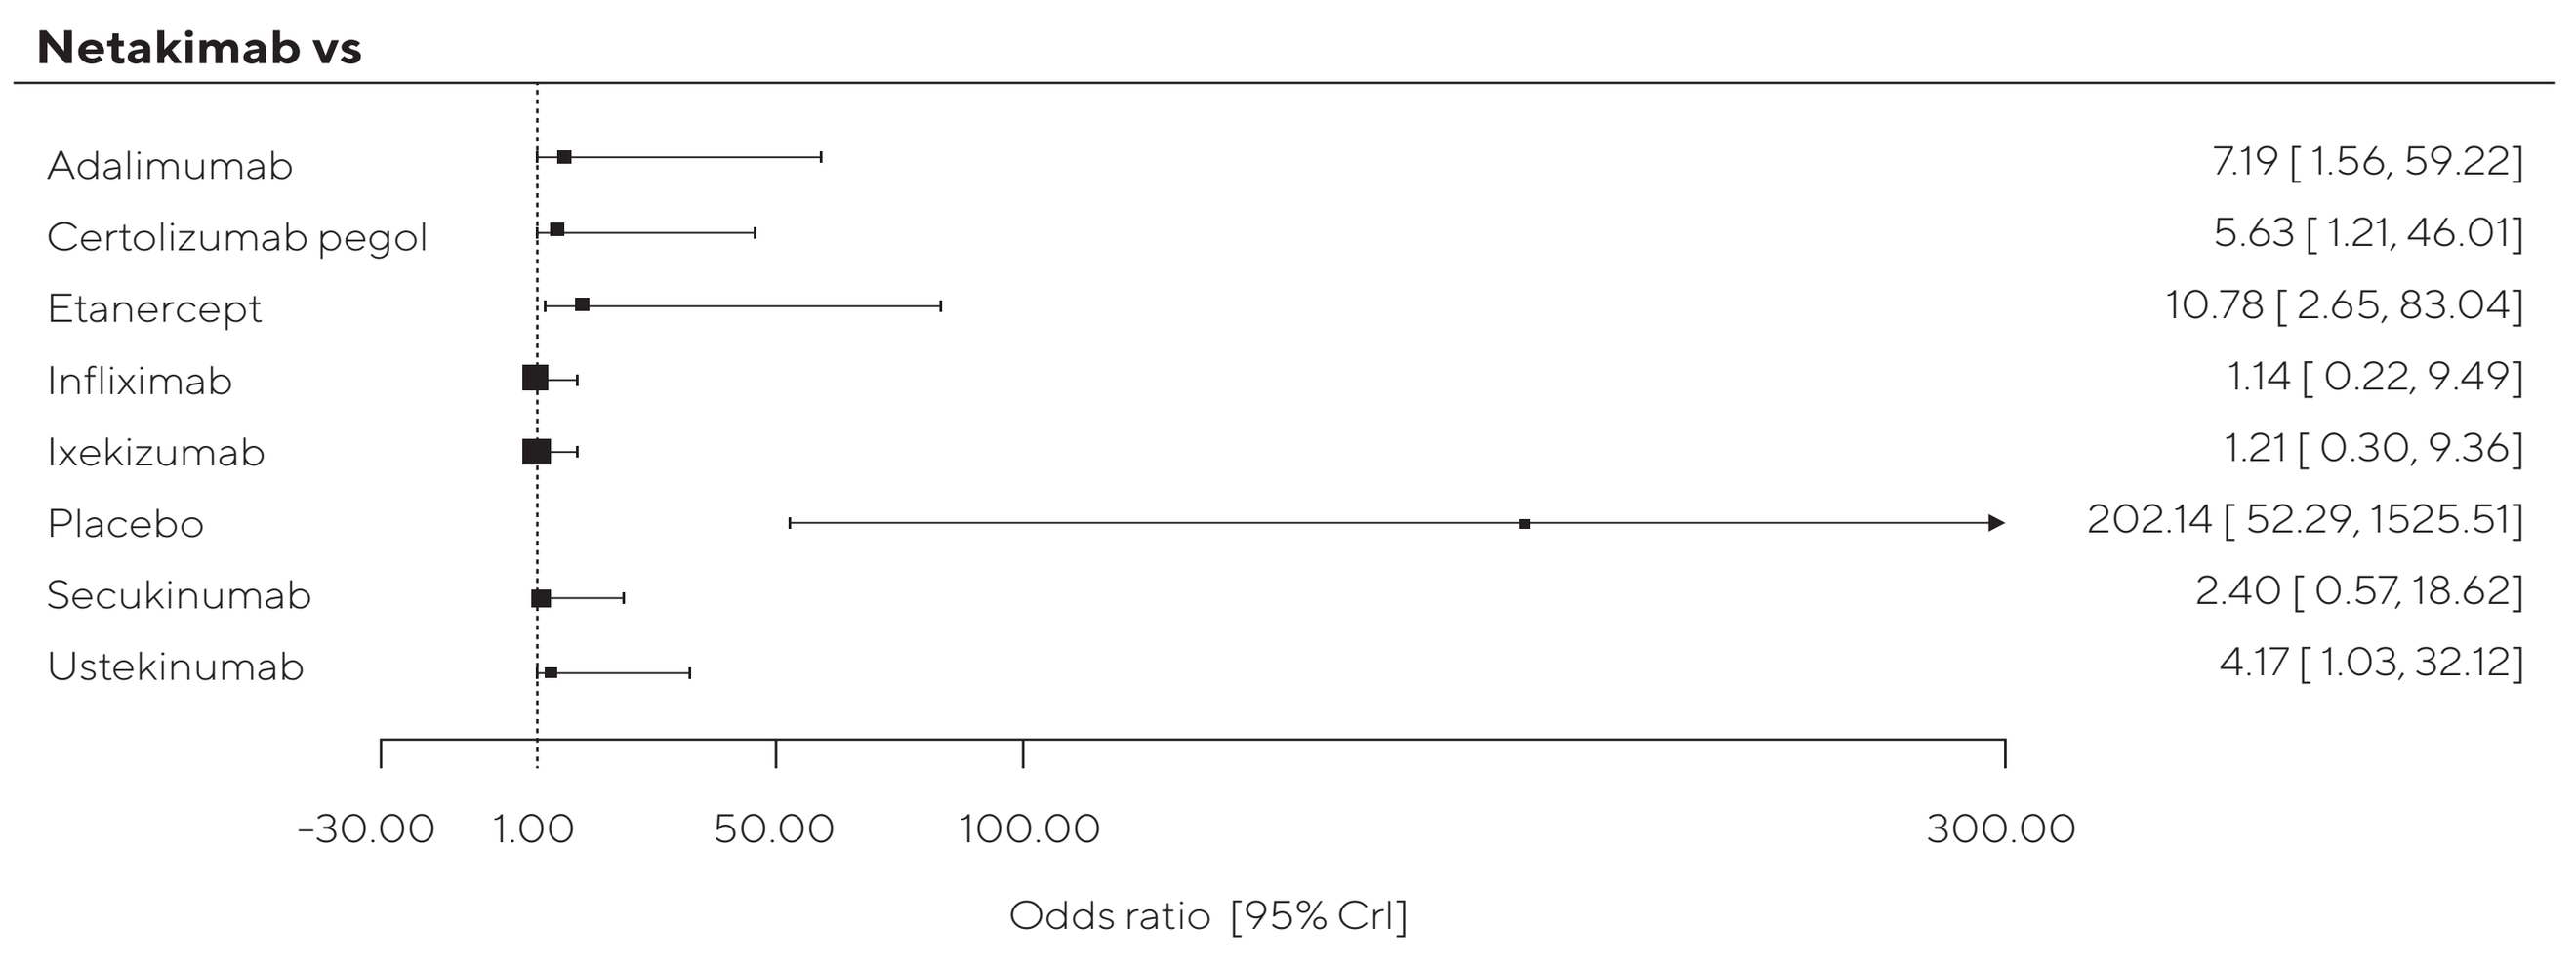

Netakimab vs
Adalimumab 7.19 [1.56, 59.22]
Certolizumab pegol 5.63 [1.21, 46.01]
Etanercept 10.78 [2.65, 83.04]
Infliximab 1.14 [0.22, 9.49]
Ixekizumab 1.21 [0.30, 9.36]
Placebo 202.14 [52.29, 1525.51]
Secukinumab 2.40 [0.57, 18.62]
Ustekinumab 4.17 [1.03, 32.12]
-30.00
1.00
50.00
100.00
300.00
Odds ratio [95% CrI]

SAFETY ANALYSIS

- Safety was defined as the proportion of patients suffering at least one adverse event (AE) during the first 12 weeks of the treatment.
- The use of netakimab has a statistically significantly more favorable safety profile compared to etanercept (OR: 0.30, 95% CrI: 0.10-0.77).
- There are no statistically significant differences in the safety profile of netakimab compared to adalimumab (OR: 0.58 , 95% CrI: 0.20-1.74), ustekinumab (OR: 0.68, 95% CrI: 0.26-1.75), secukinumab (OR: 0.47, 95% CrI: 0.17 -1.24), infliximab (OR: 0.41, 95% CrI: 0.15-1.12), certolizumab (OR: 0.74, 95% CrI: 0.26-2.08) and ixekizumab (OR: 0.42, 95% CrI: 0.14-1.25).

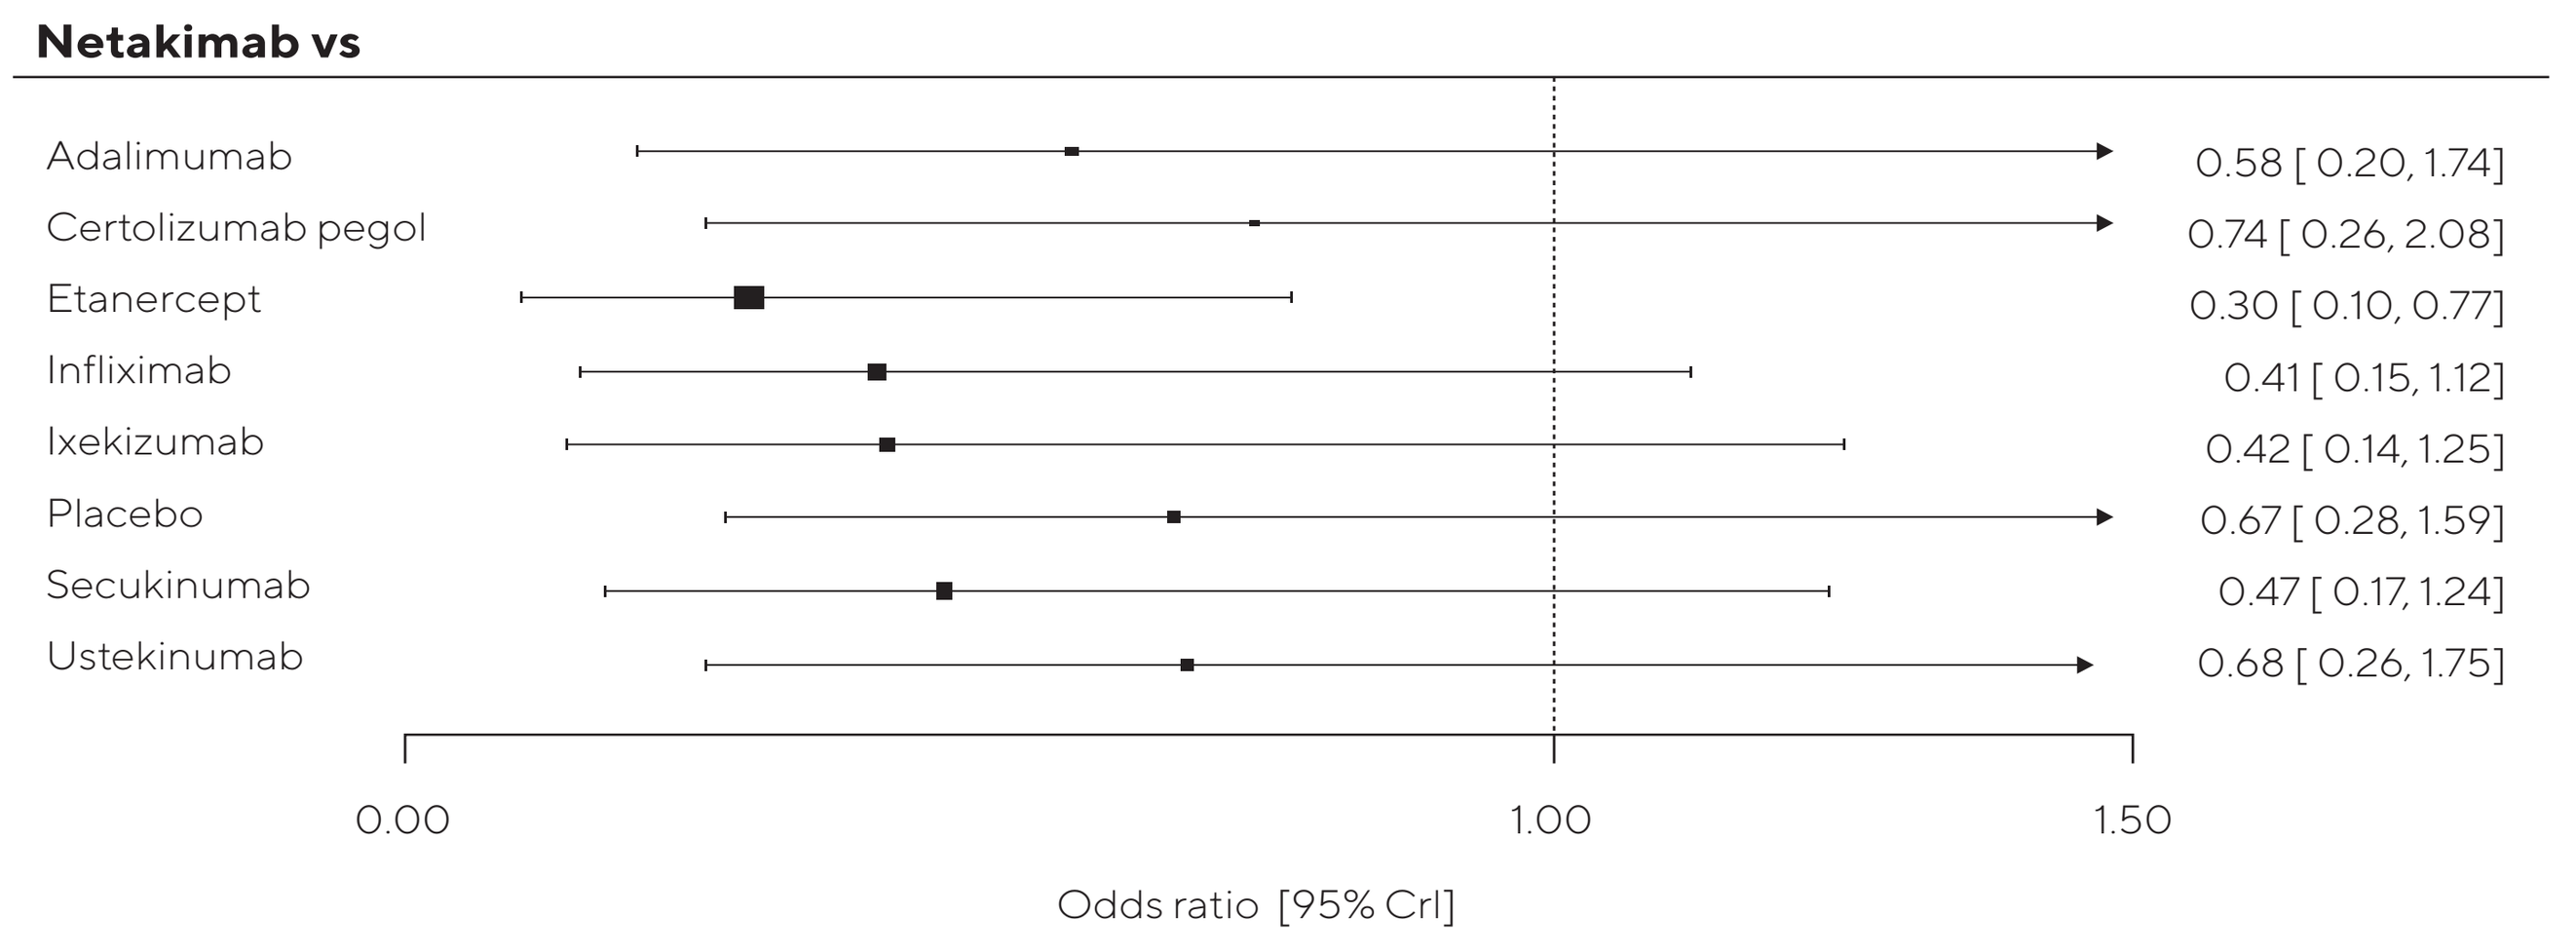

Netakimab vs
Adalimumab 0.58 [0.20, 1.74]
Certolizumab pegol 0.74 [0.26, 2.08]
Etanercept 0.30 [0.10, 0.77]
Infliximab 0.41 [0.15, 1.12]
Ixekizumab 0.42 [0.14, 1.25]
Placebo 0.67 [0.28, 1.59]
Secukinumab 0.47 [0.17, 1.24]
Ustekinumab 0.68 [0.26, 1.75]
0.00
1.00
1.50
Odds ratio [95% CrI]

CONCLUSION

Results suggest that netakimab has effectiveness superior to or comparable with all other biologics and a similar safety profile.

BIOCAD
Biotechnology Company