Progression, overall survival, and healthcare resource utilization in patients with prostate cancer on androgen deprivation therapy: a Swedish register-based study

PCN512

Johanna Svensson,[1] Oskar Gauffin,[2] Ingela Franck Lissbrant,[3] Suzanne Kilany,[1] Pär Stattin,[2] Karin Fagerlund[1]

[1]Astellas Pharma, Kastrup, Denmark; [2]Department of Surgical Sciences, Uppsala University Hospital, Uppsala, Sweden; [3]Department of Oncology, University of Gothenburg, Gothenburg, Sweden

BACKGROUND

- Diagnostic work-up and primary treatment of prostate cancer in Sweden is registered in the National Prostate Cancer Register (NPCR).[1] However, there are no data on progression from nonmetastatic (nm) or metastatic (m) hormone-sensitive prostate cancer (HSPC) to castration-resistant prostate cancer (CRPC) in men on androgen deprivation therapy (ADT) in the NPCR. The vast majority of patients who die from prostate cancer have made a transition through CRPC.[1] The incidence of transition to CRPC (nmCRPC and mCRPC) has been estimated at slightly more than 5500 per year in Nordic countries. It is important to note that some patients die from causes other than prostate cancer during the disease trajectory.[1]

OBJECTIVES

- The aim of this study was to assess time from disease-state entry to progression or death from nmHSPC or mHSPC through nmCRPC or mCRPC, as well as co-medications, comorbidities, and healthcare resource use using real-world data. The primary objective is to describe time in each prostate cancer stage to progression or death. See **Figure 1** for possible transitions between disease stages. The secondary objectives are to describe: 1) co-medications at each disease stage; 2) comorbidities at each disease stage; and 3) the healthcare resource use and costs associated with each disease stage.

Figure 1. Pathways for patient progression through prostate cancer stages and death

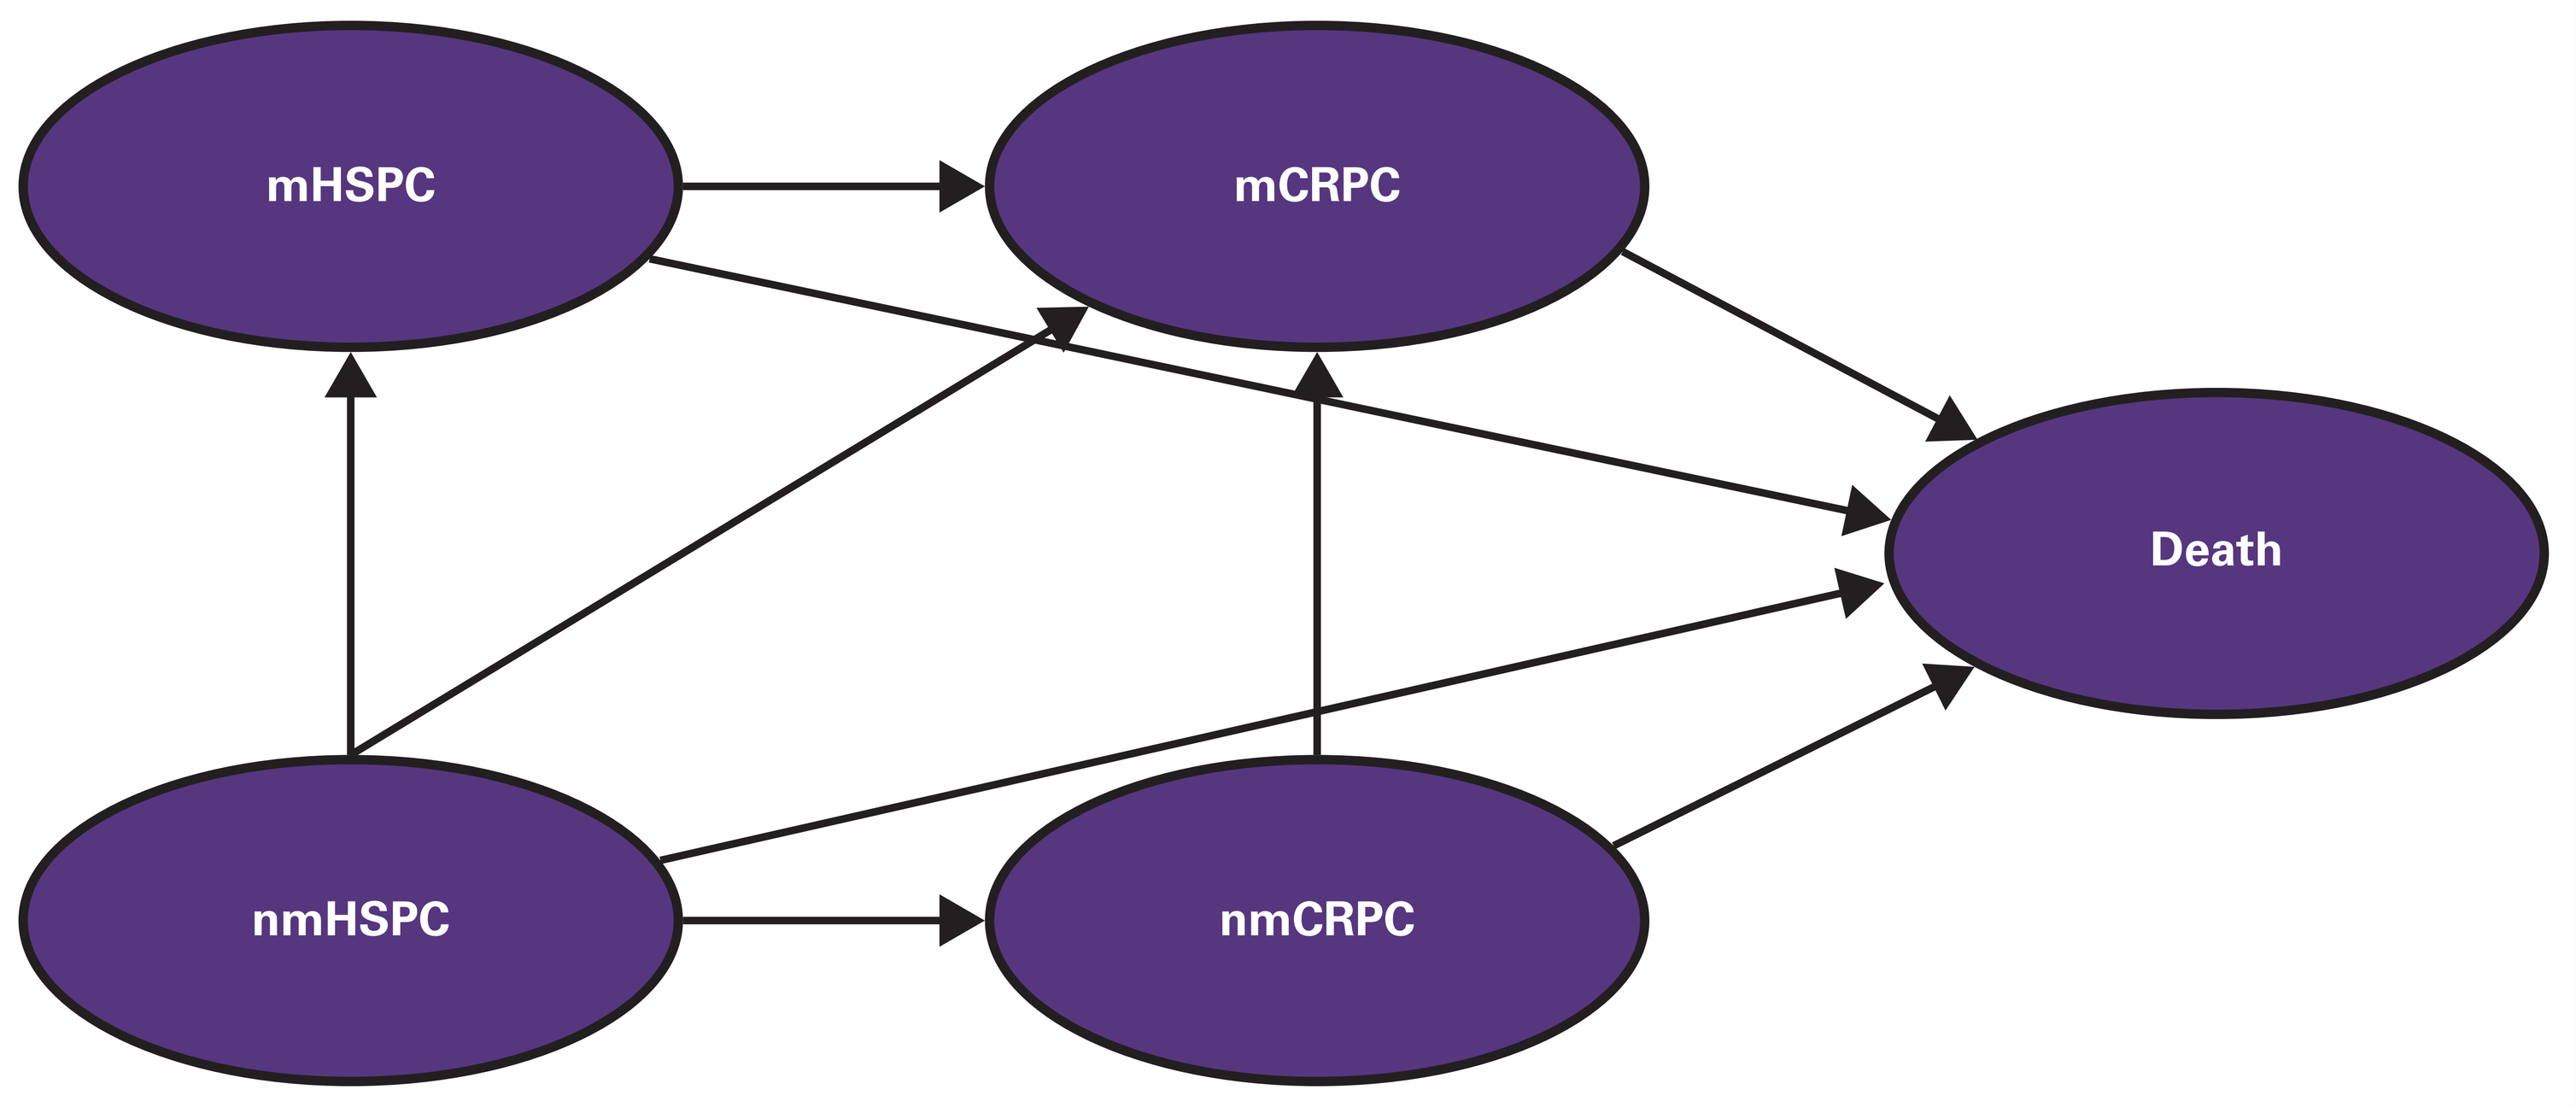
mHSPC
mCRPC
Death
nmHSPC
nmCRPC

CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic.

METHODS

- Real-world data were extracted from Prostate Cancer Database Sweden (PCBaSe), a registry linking data from Patient-overview Prostate Cancer (PPC),[2] national healthcare registries, and demographic databases from 2006–2016. This noninterventional, retrospective register study was conducted at Uppsala University Hospital and Regional Cancer Centre in Uppsala, Sweden. PPC began including patients on life-long ADT treatment in 2014 to follow new hormonal therapies. The study population comprised men with prostate cancer registered in PPC between January 1, 2014, and December 31, 2016. Patient history was collected by retrospective chart review going back no further than 2006. Data were extracted from PPC for the different prostate cancer disease stages from date of registry entry (in the period 2014–2016), prospectively (until death or December 31, 2016), and retrospectively, going back no further than 2006. Inclusion criteria: patients registered in PPC until December 31, 2016. Exclusion criteria: in the event of discrepancies in the dates of HSPC-stage entry reported in PCBaSe and PPC, patients were excluded because of complications of setting entry dates to HSPC. In addition, patients were excluded if date of ADT initiation was missing or the ADT initiation date occurred before 2006.
- Overall survival (OS) and time to progression (TTP) were estimated using Kaplan-Meier and cumulative incidence functions. Unit prices from official sources were used for cost calculations. Costs are presented as annual cost per patient.

RESULTS

- 1869 men were included in the study data set. Thirty-two percent of the study population had distant metastases at initial diagnosis. Mean age ranged from 70.1 years for mHSPC to 74.8 years for nmCRPC. **Table 1** shows patient characteristics by disease stage at time of entry into disease stage. Note that one patient can be represented in more than one disease stage due to transitioning between stages.

Table 1. Patient characteristics by disease stage

	nmHSPC	mHSPC	nmCRPC	mCRPC
Patients, n	1034	884	356	1172
Age, years				
Mean (SD)	72.18 (7.42)	70.1 (8.15)	74.78 (7.11)	72.71 (7.9)
Median	72.37	69.78	74.61	72.78
Years since prostate cancer diagnosis				
Mean (SD)	2.56 (3.35)	1.22 (2.85)	5.3 (3.82)	4.47 (4.01)
Median	0.65	0.11	4.73	3.28
Risk group at diagnosis,[a] n (%)				
Low	80 (8)	30 (3)	12 (4)	54 (5)
Intermediate	289 (28)	56 (6)	85 (24)	188 (16)
High	399 (38)	93 (11)	139 (39)	258 (22)
Regionally metastatic	144 (14)	67 (8)	61 (17)	123 (10)
Distant metastases	113 (11)	635 (72)	55 (15)	541 (46)
Missing	9 (1)	3 (0)	4 (1)	8 (1)

[a]A modified version of the National Comprehensive Cancer Network risk categorization was used (low risk: T1–2, Gleason score ≤6, and PSA <10 ng/l; intermediate risk: T1–2, Gleason score 7, and/or PSA >10 to <20 ng/l; localized high risk: T1–2, Gleason score 8–10, and/or PSA >20 to <50 ng/l; regionally metastatic: T4 and/or N1, and/or PSA >50 to <100 ng/l, and no distant metastases; distant metastases: M1 and/or PSA >100; missing: missing information for categorization).
CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic; PSA=prostate-specific antigen; SD=standard deviation.

- Median OS and median TTP for men with nmHSPC (n=1034) were 11 and 4 years, respectively. The corresponding numbers for mHSPC (n=884) were 7 and 1.3 years and 8.5 and 1.4 years for nmCRPC (n=356). Median OS for mCRPC (n=1172) was 3.9 years (**Table 2**, **Figure 2**, and **Figure 3**).

Table 2. Median time to event from each disease stage (years)

	Events, no.	Median (95% CI)
nmHSPC		
Total (N)	1034	
Time to progression[a]	674	4.0 (3.8, 4.4)
Time to death[b]	188	10.9 (10.1, NA)
mHSPC		
Total (N)	884	
Time to progression[a]	642	1.3 (1.2, 1.5)
Time to death[b]	202	7.1 (6.7, 7.9)
nmCRPC		
Total (N)	356	
Time to progression[a]	261	1.4 (1.2, 1.6)
Time to death[b]	80	8.5 (6.2, NA)
mCRPC		
Total (N)	1172	
Time to death[b]	341	3.9 (3.5, 4.4)

[a]Cumulative incidence function estimate; [b]Kaplan-Meier estimate.
CI=confidence interval; CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; NA=insufficient number of events; nm=nonmetastatic.

Figure 2. Stacked cumulative incidence of time from disease-stage entry to progression and death (prostate cancer and nonprostate cancer related reported separately) by each possible exit stage: A) nmHSPC; B) mHSPC; C) nmCRPC; and D) mCRPC

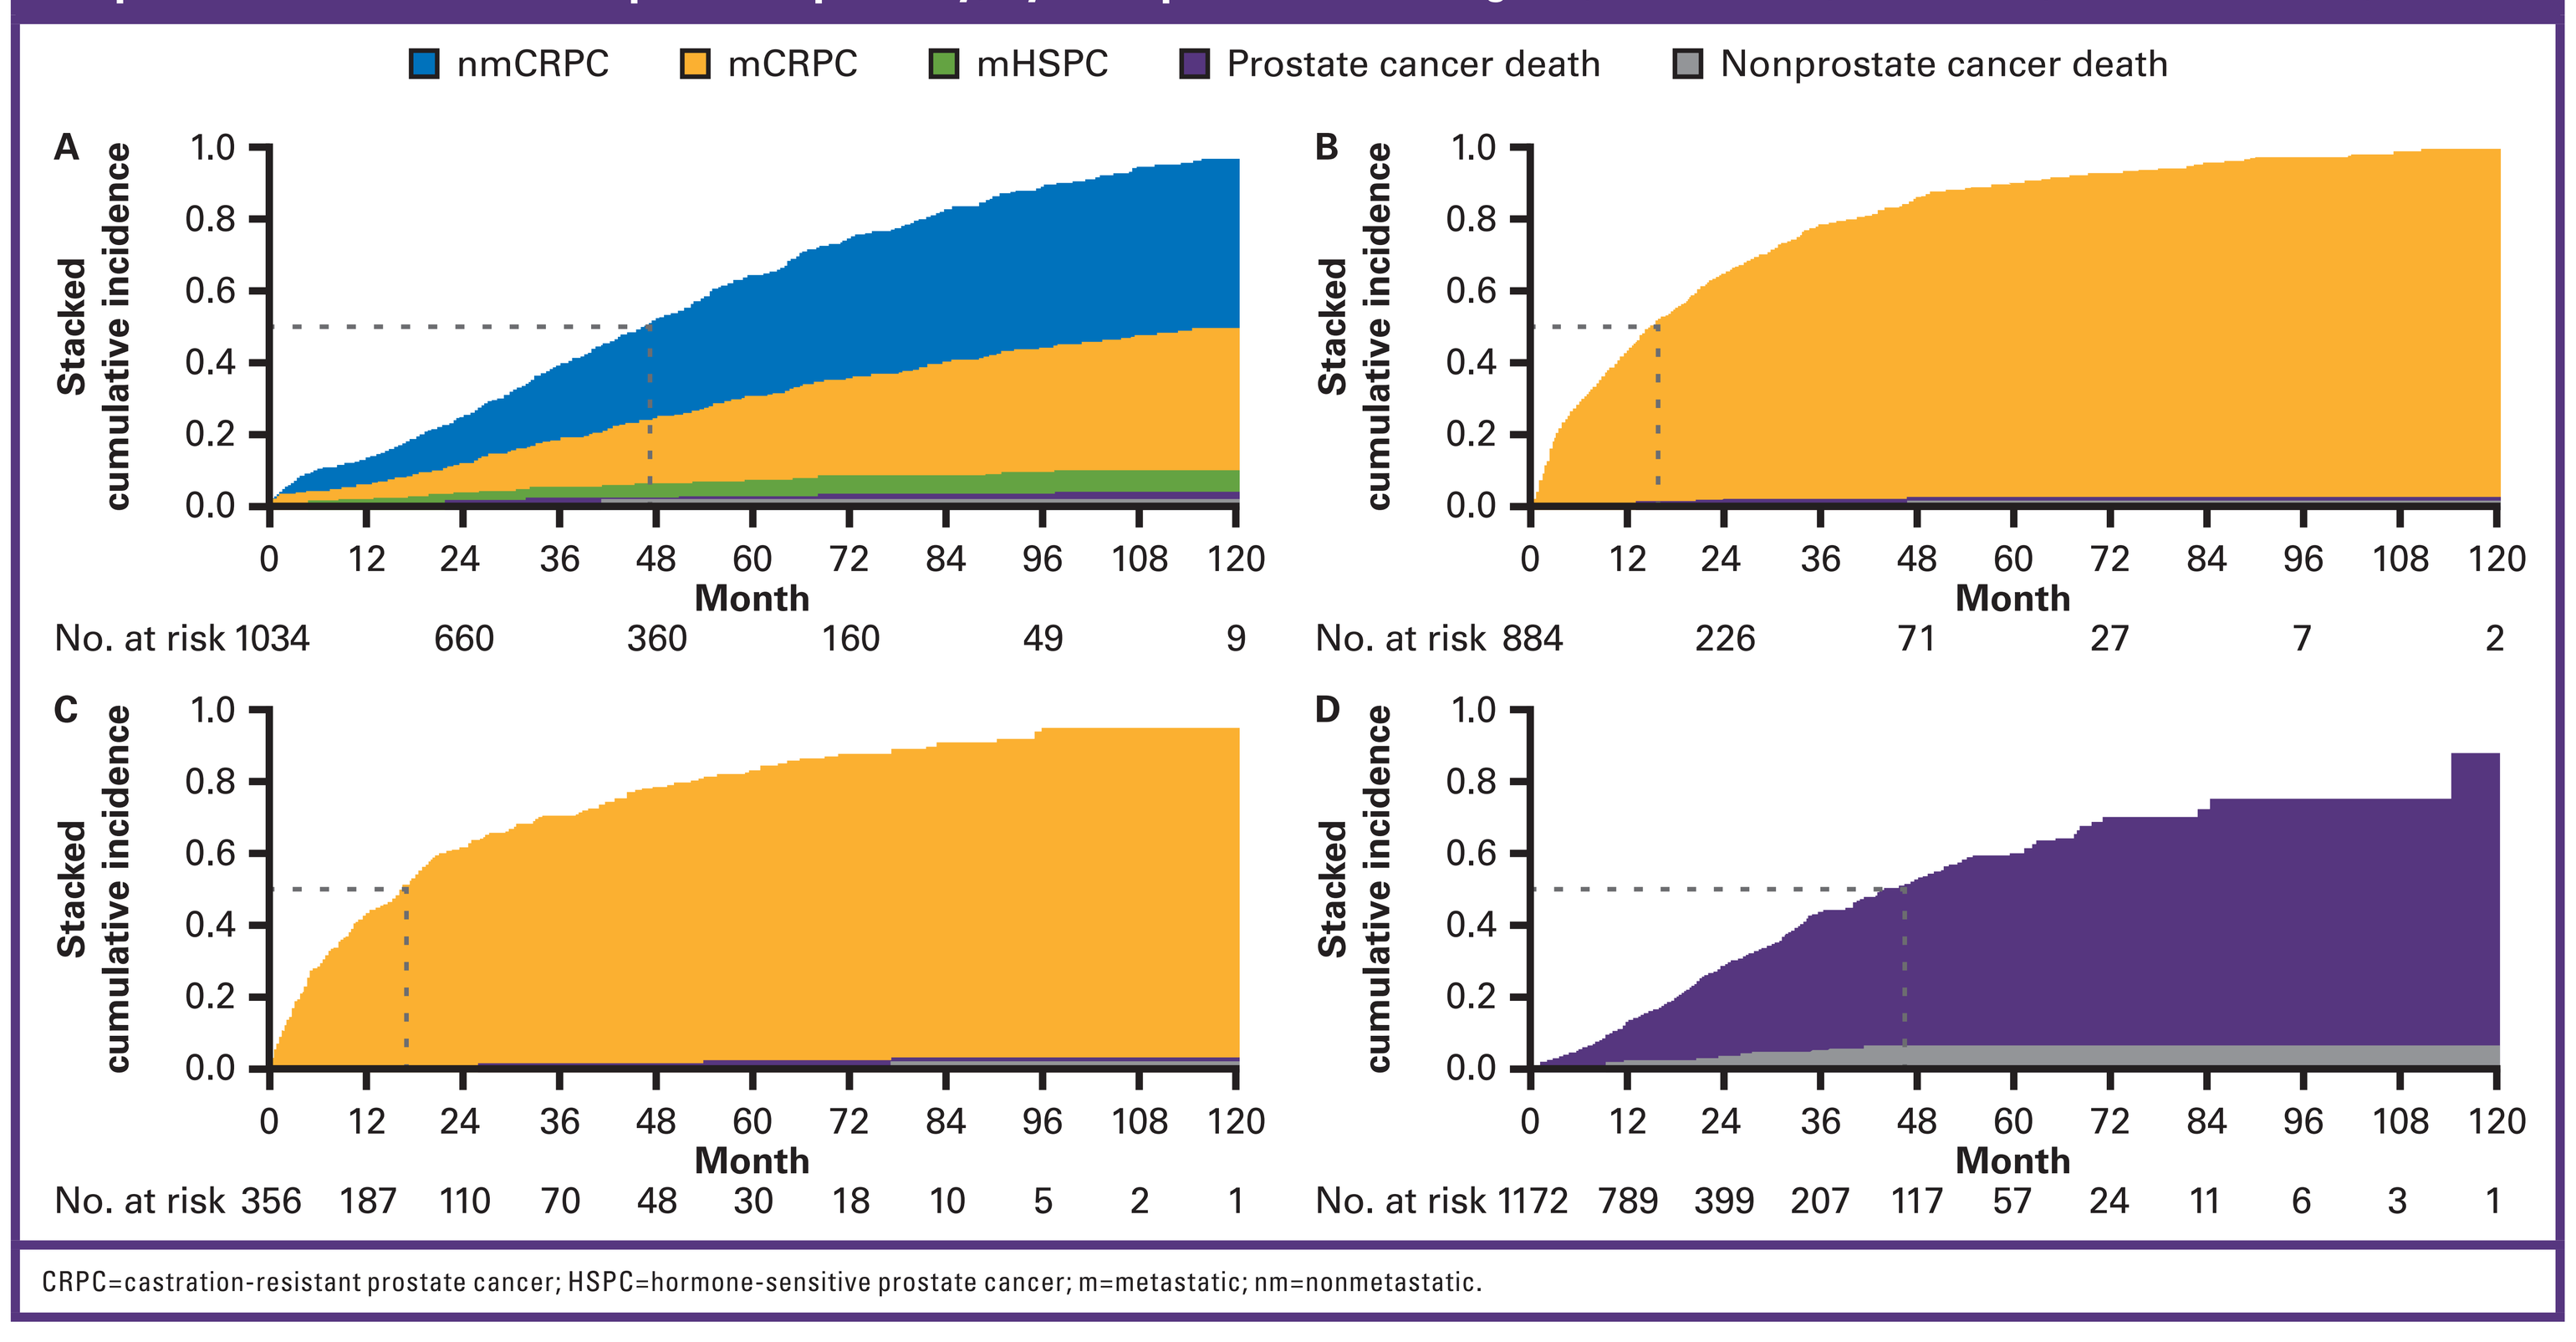
nmCRPC
mCRPC
mHSPC
Prostate cancer death
Nonprostate cancer death
A
B
C
D
Stacked cumulative incidence
Month
No. at risk 1034 660 360 160 49 9
No. at risk 884 226 71 27 7 2
No. at risk 356 187 110 70 48 30 18 10 5 2 1
No. at risk 1172 789 399 207 117 57 24 11 6 3 1

CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic.

Figure 3. Kaplan-Meier graph of time from disease-stage entry to all-cause death: A) nmHSPC; B) mHSPC; C) nmCRPC; and D) mCRPC

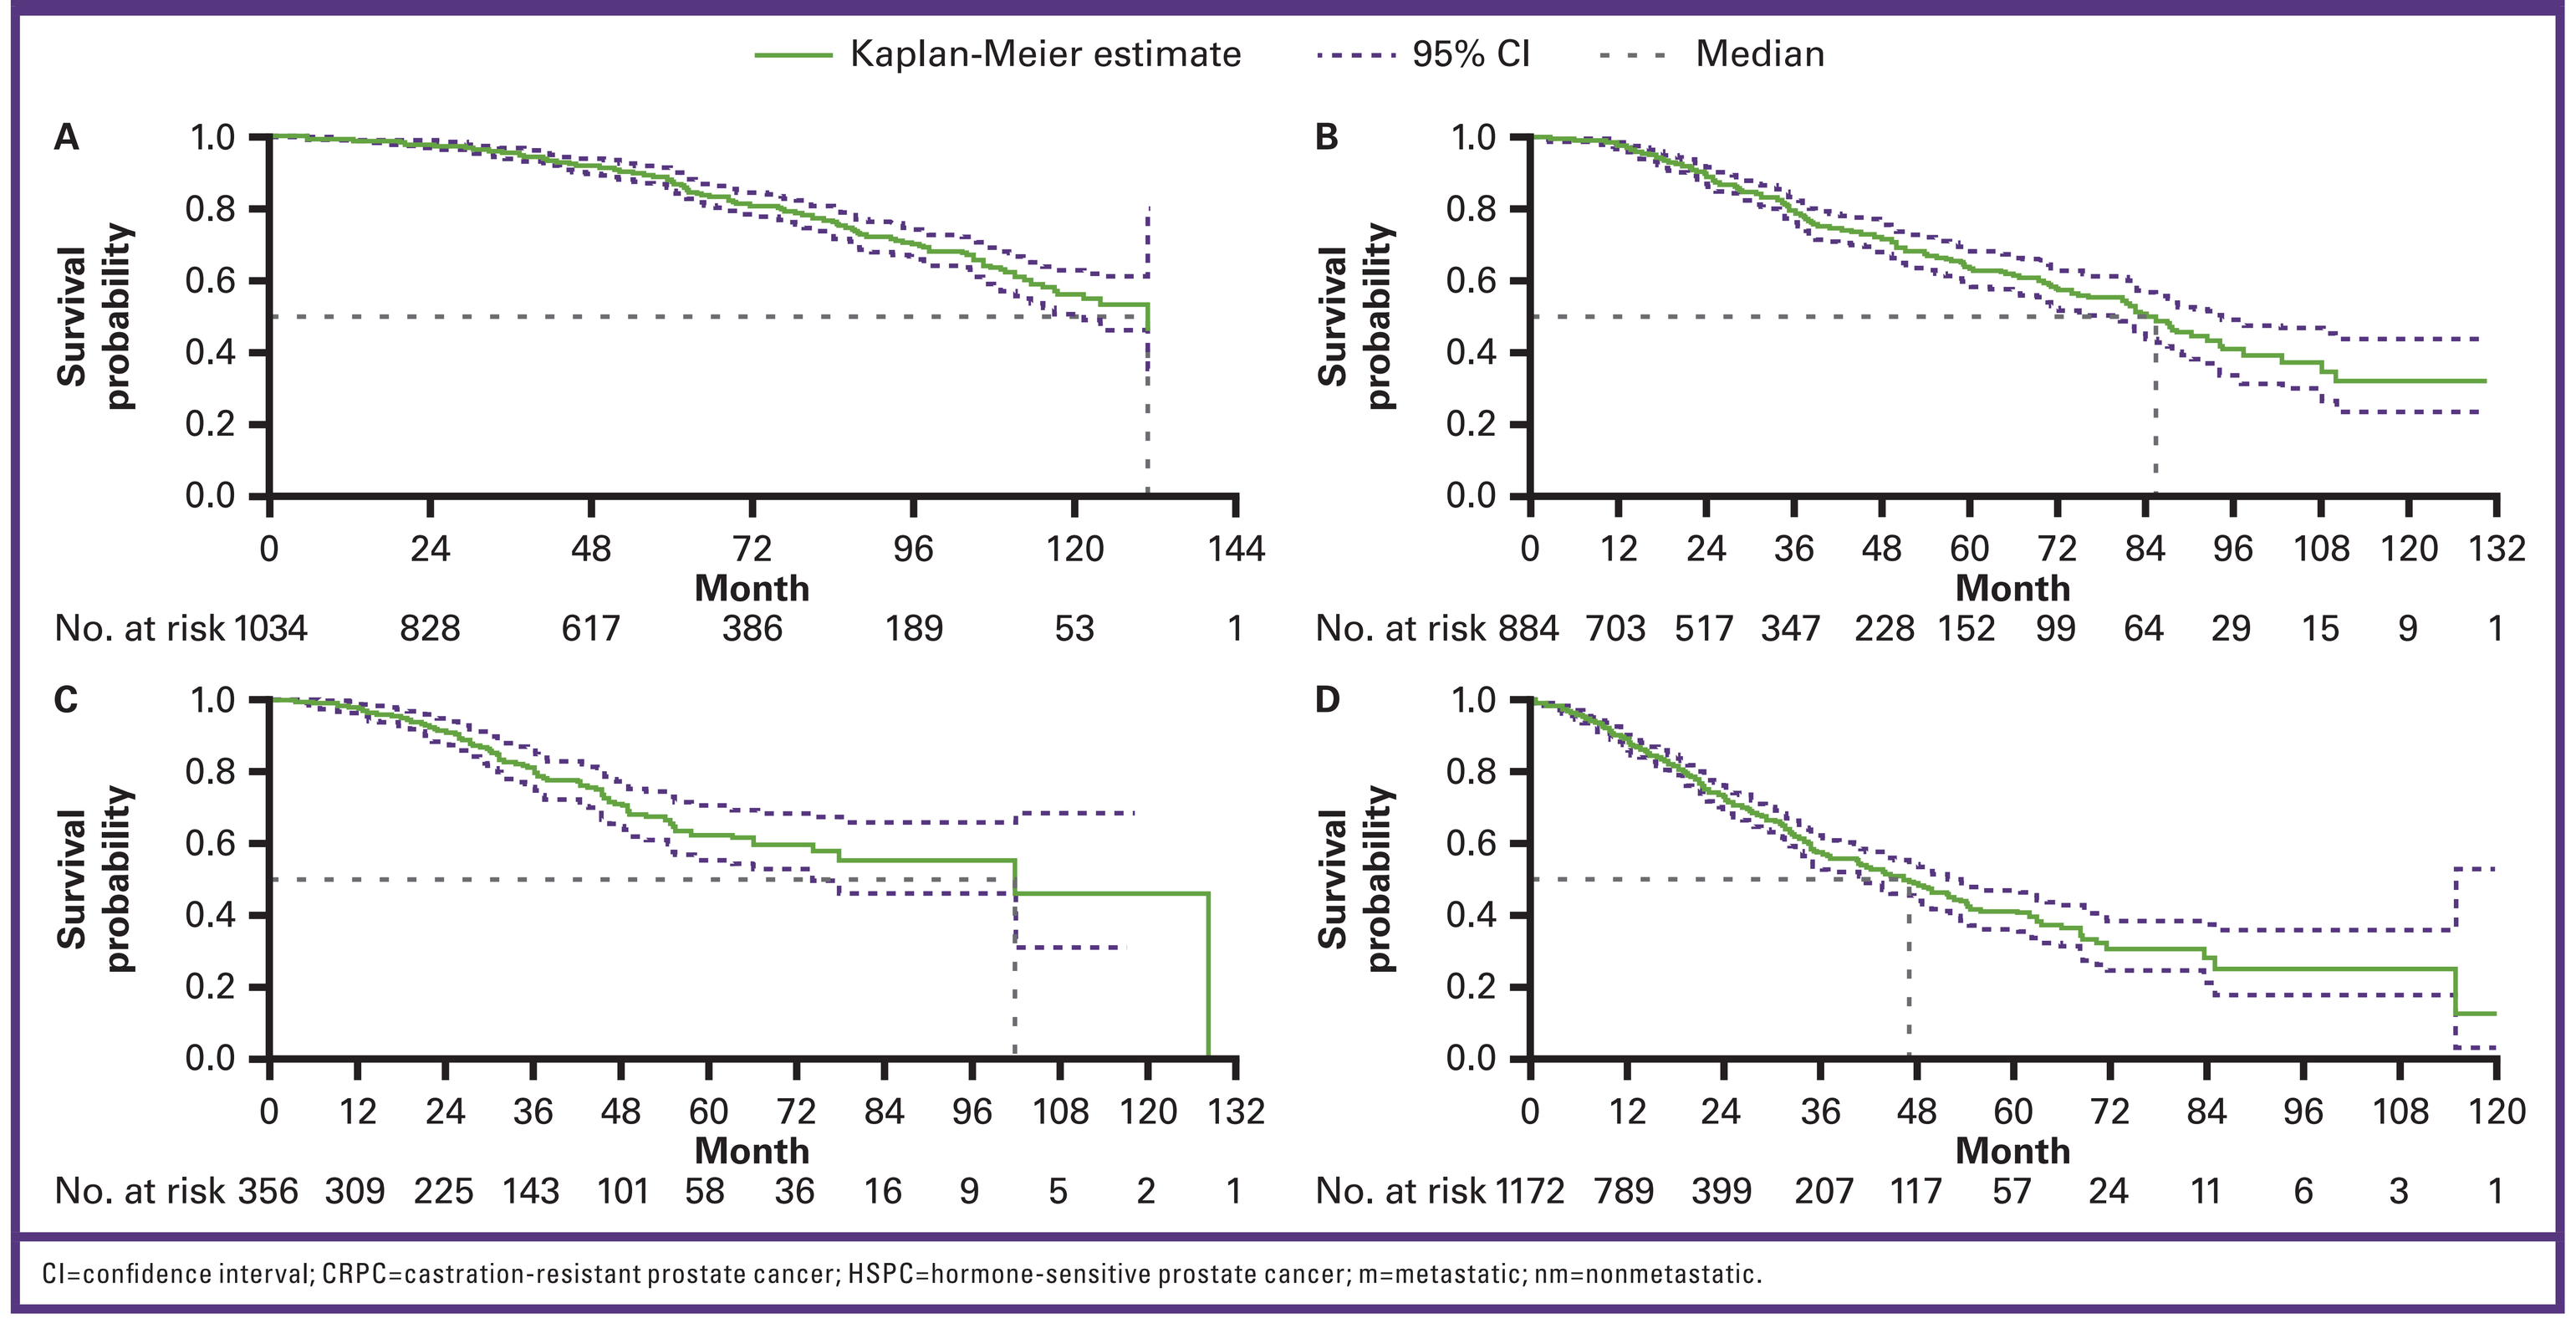
Kaplan-Meier estimate
95% CI
Median
A
B
C
D
Survival probability
Month
No. at risk 1034 828 617 386 189 53 1
No. at risk 884 703 517 347 228 152 99 64 29 15 9 1
No. at risk 356 309 225 143 101 58 36 16 9 5 2 1
No. at risk 1172 789 399 207 117 57 24 11 6 3 1

CI=confidence interval; CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic.

- The most common co-medications included analgesics and antipyretics. The most common comorbidities were cardiovascular (myocardial infarction and congestive heart disease) and diabetes, both of which affected 5–9%. **Figure 4** shows the prostate cancer treatments that were used in each disease stage. In earlier stages of the disease, different types of ADT (antiandrogens and gonadotropin-releasing hormones) dominated. Patients with mCRPC were treated with androgen receptor-targeted therapies (enzalutamide/abiraterone acetate [68%]), docetaxel (41%), and radiation therapy (19%), in addition to ADT.

Figure 4. Prostate cancer treatments by disease stage from stage entry to exit

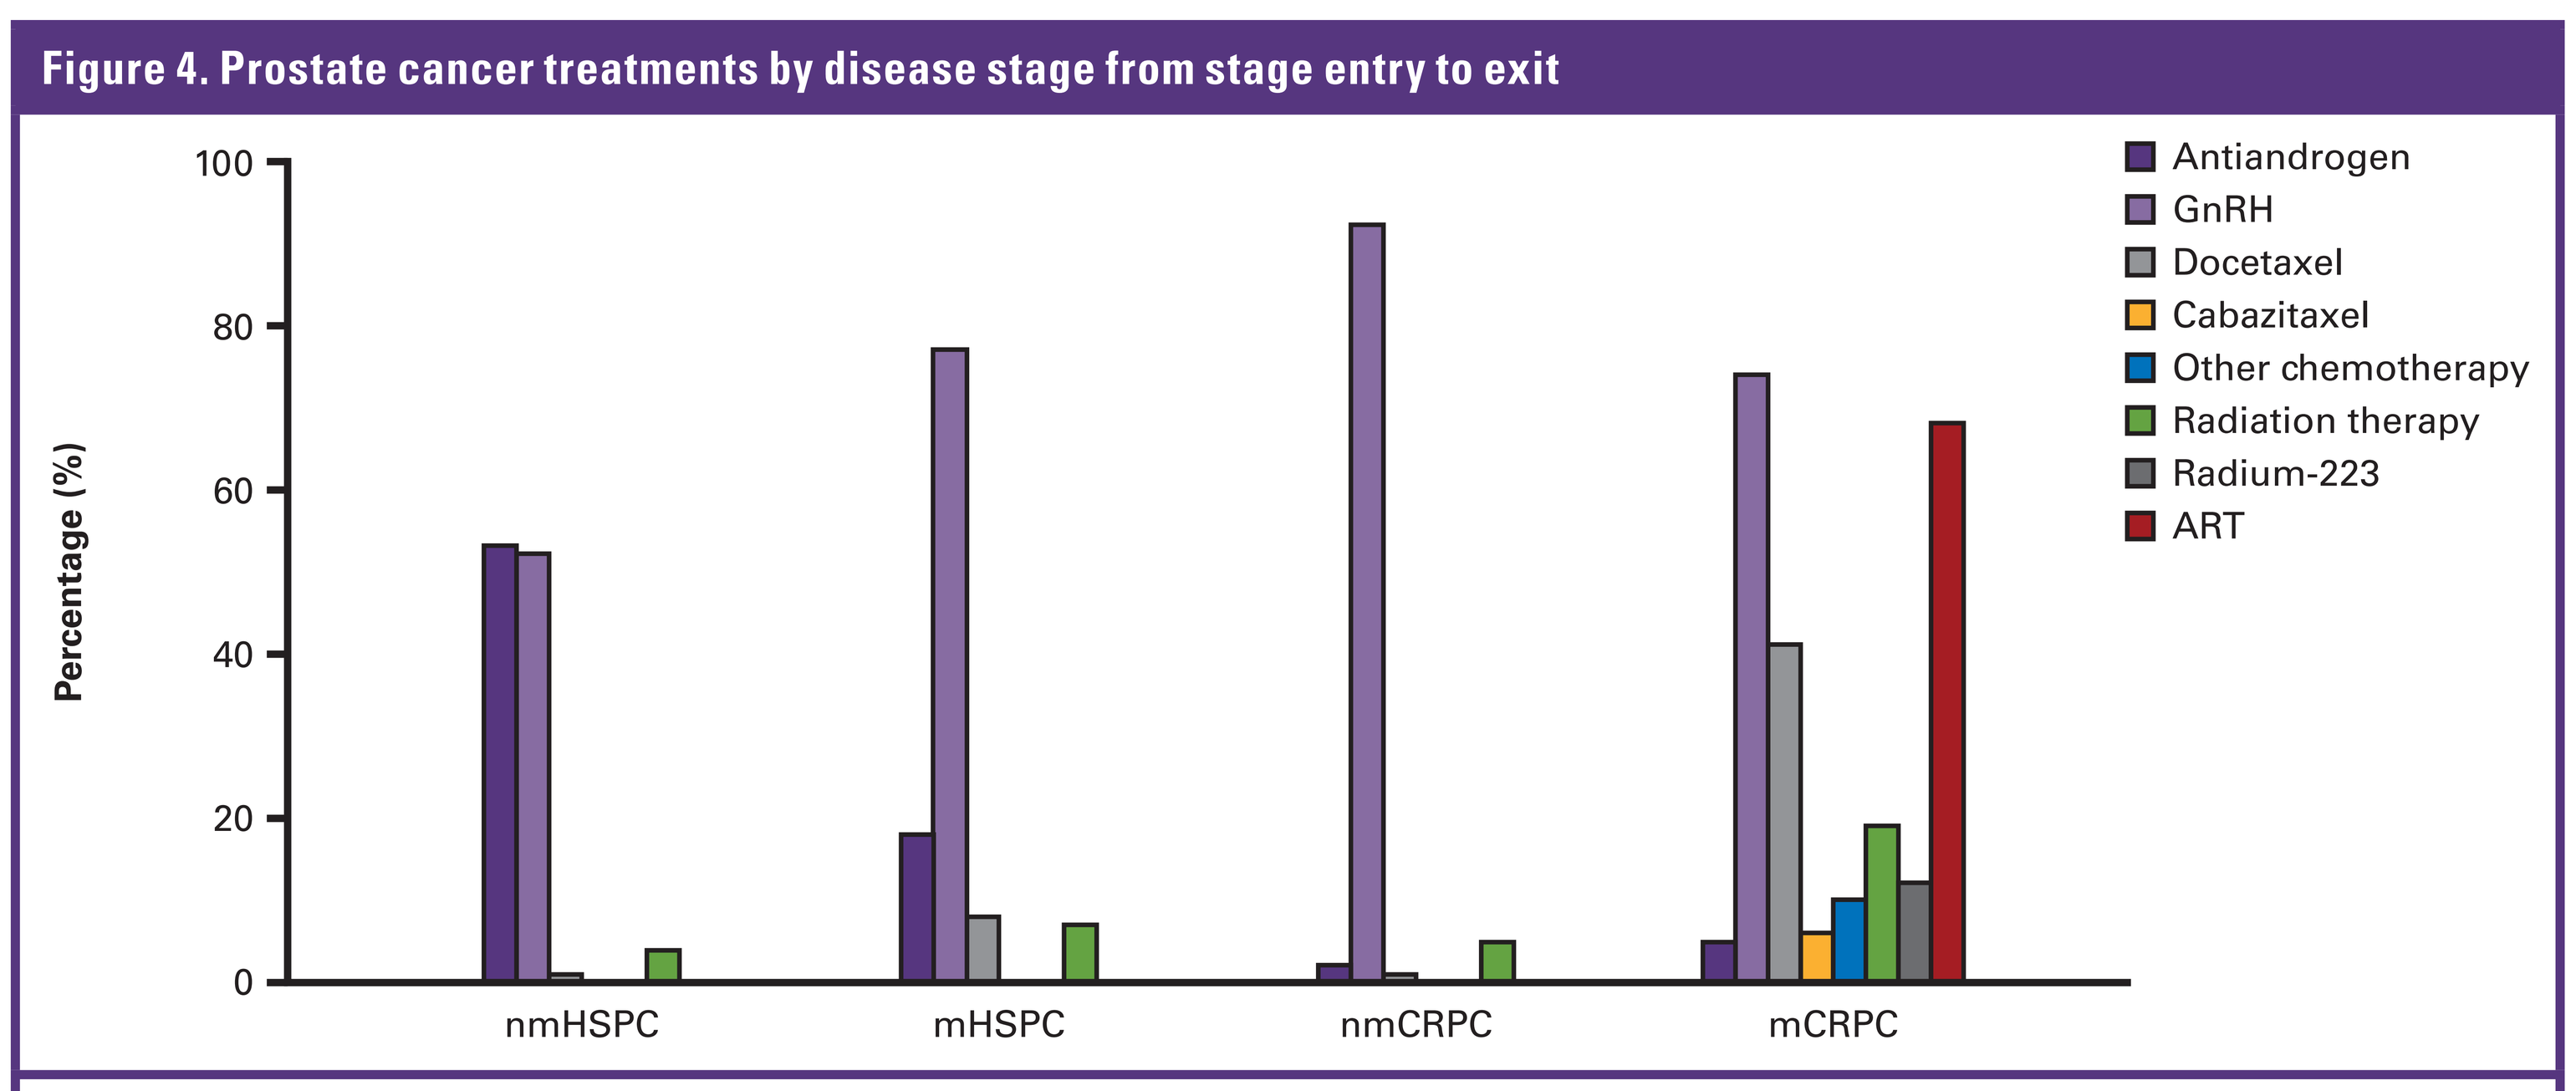
Percentage (%)
nmHSPC
mHSPC
nmCRPC
mCRPC
Antiandrogen
GnRH
Docetaxel
Cabazitaxel
Other chemotherapy
Radiation therapy
Radium-223
ART

ART=androgen receptor-targeted therapies (enzalutamide/abiraterone acetate); CRPC=castration-resistant prostate cancer; GnRH=gonadotropin-releasing hormone; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic.

- Costs for hospitalizations, outpatient visits, and prostate cancer treatments increased with increasing disease severity. Mean annual healthcare-related costs ranged from 36,000 Swedish krona (SEK) [€3300] for nmHSPC to SEK 274,000 (€25,400) for mCRPC (**Figure 5**).

Figure 5. Annual healthcare-related costs per patient by disease stage (SEK 2018)

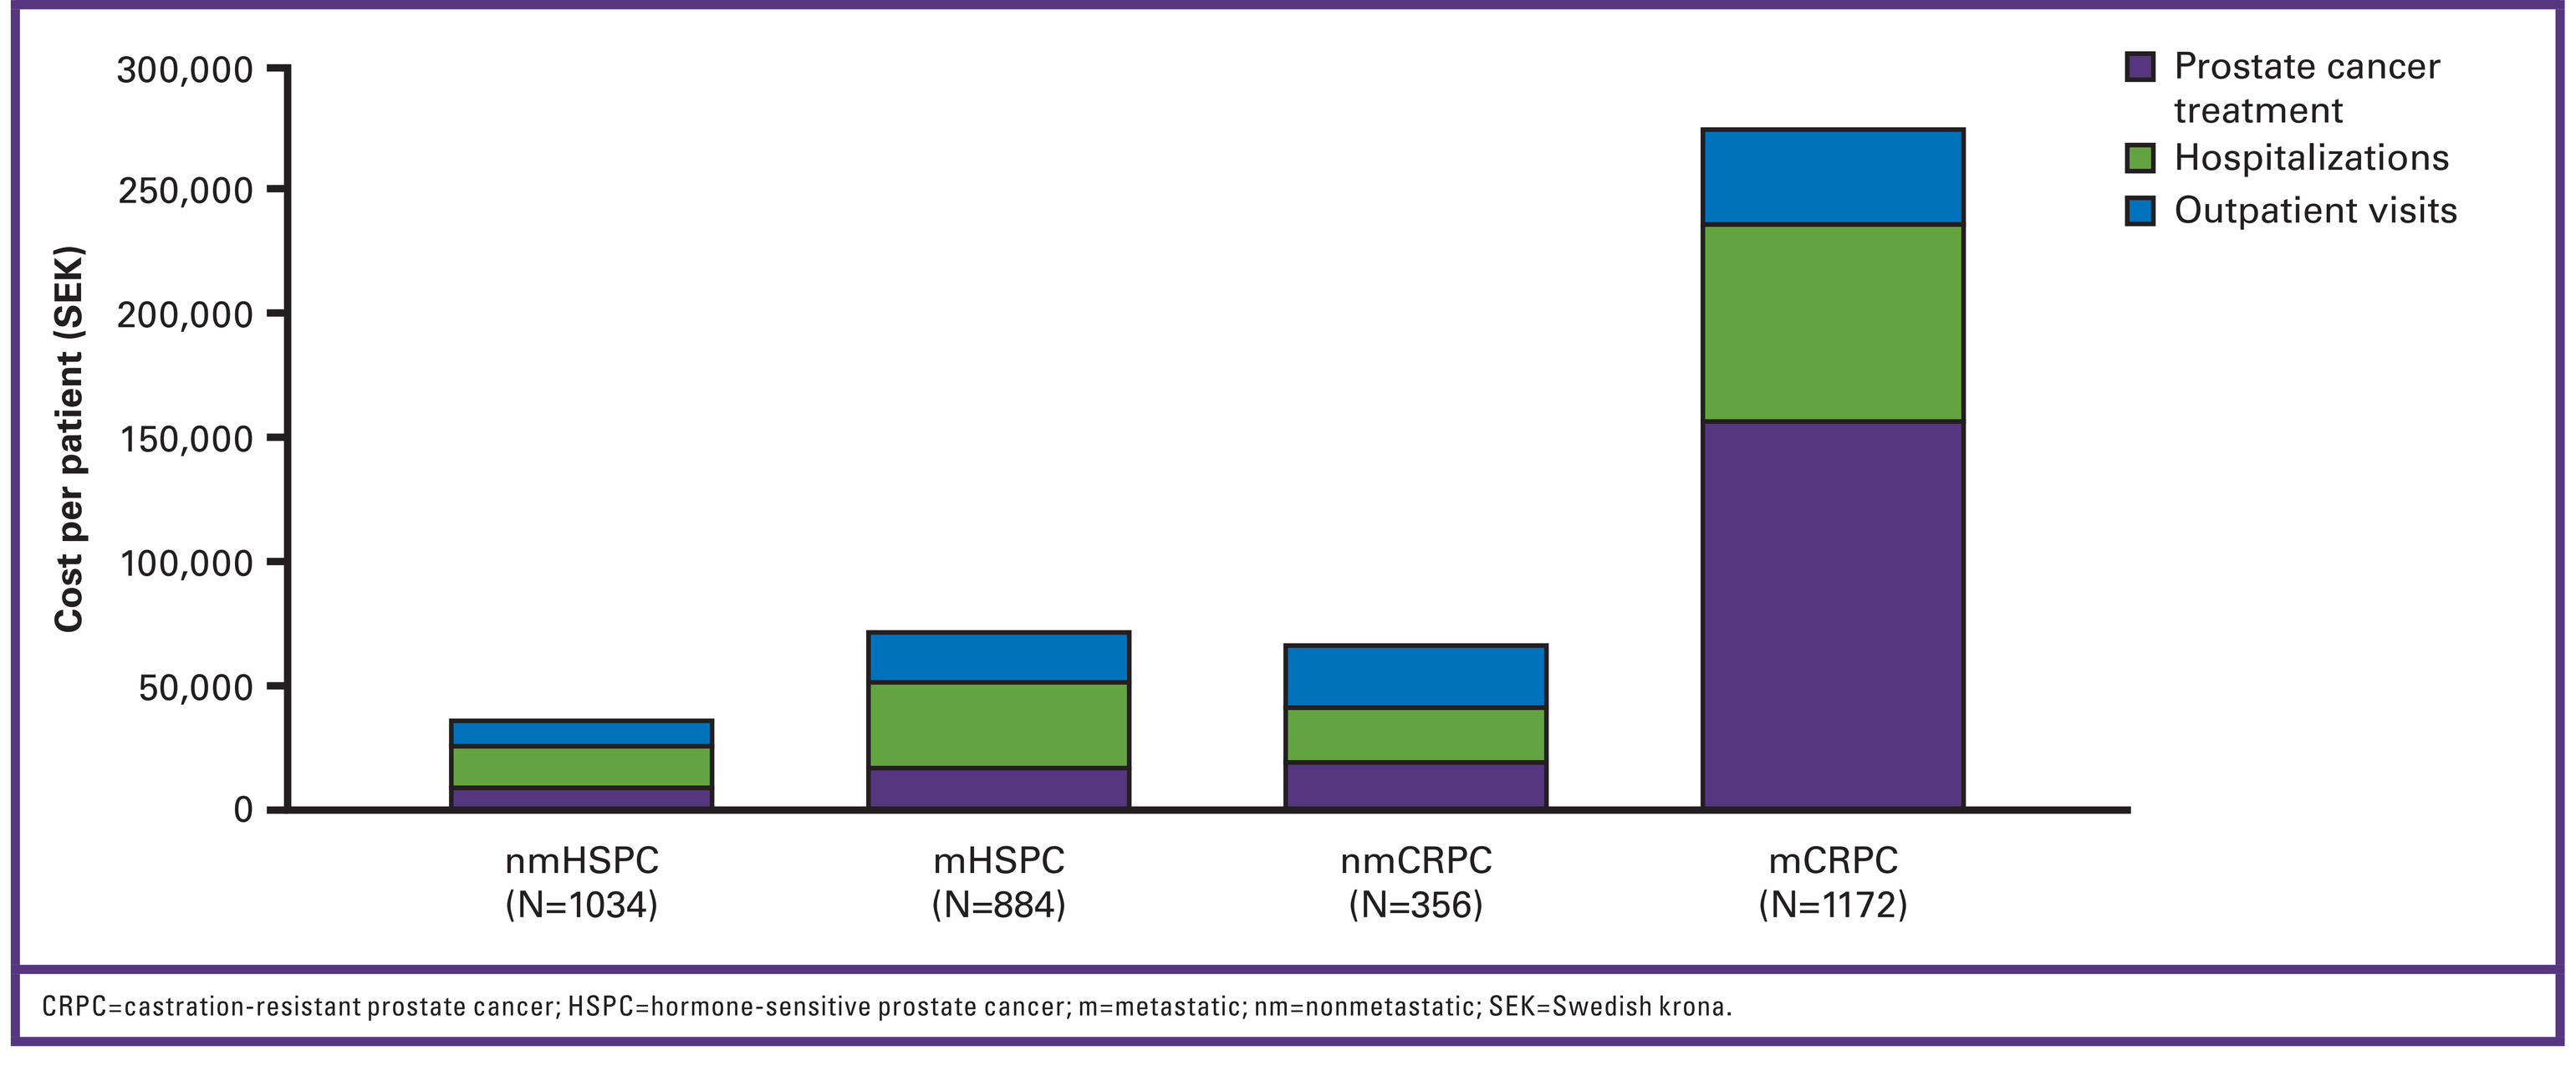
Cost per patient (SEK)
nmHSPC (N=1034)
mHSPC (N=884)
nmCRPC (N=356)
mCRPC (N=1172)
Prostate cancer treatment
Hospitalizations
Outpatient visits

CRPC=castration-resistant prostate cancer; HSPC=hormone-sensitive prostate cancer; m=metastatic; nm=nonmetastatic; SEK=Swedish krona.

CONCLUSIONS

- Time to progression in mHSPC and nmCRPC was relatively short, and men soon progressed to mCRPC, where they spent substantially longer time. The disease trajectory for men with advanced prostate cancer needs to be further elucidated.

ACKNOWLEDGMENTS

This study was funded by Astellas Pharma, Kastrup, Denmark. Editorial assistance was provided by Folabomi Oladosu, PhD, and Jane Beck from Complete HealthVizion, funded by Astellas Pharma Inc.

REFERENCES

1. Lissbrant IF et al. Acta Oncol 2013; 52: 1593–1601.
2. Stattin P et al. Tijdschr Urol 2017; 7: 50–59.

POSTER
Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

ISPOR 22nd Annual European Congress, November 2–6, 2019, Copenhagen, Denmark