PCN187

BUDGET IMPACT OF SEQUENTIAL TREATMENT WITH AFATINIB FOLLOWED BY OSIMERTINIB VERSUS FIRST-LINE OSIMERTINIB IN NON-SMALL-CELL LUNG CANCER PATIENTS WITH COMMON EGFR MUTATIONS IN THE NETHERLANDS

Westerink L[1,2], Nicolai J[3], Samuelsen CH[4], Griebsch I[5], Postma M[1,6]

[1]University Medical Center Groningen, Groningen, the Netherlands; [2]Asc Academics, Groningen, the Netherlands
[3]Boehringer Ingelheim International GmbH, Alkmaar, the Netherlands,
[4]Outcomes Analytica AS, Oslo, Norway, former employee of Boehringer of Ingelheim International GmbH
[5]Boehringer Ingelheim International GmbH, Ingelheim, Germany; [6]University of Groningen, Groningen, The Netherlands

INTRODUCTION

- Lung cancer is the leading cause of cancer mortality worldwide, with 40% of patients having subtype adenocarcinoma in non-small-cell lung cancer (NSCLC).[1,2] At diagnosis the majority of patients have advanced or metastatic disease. Mutations in the epidermal growth factor receptor (EGFR) gene are common in NSCLC and the mutation status improves predictions of the behaviour and characterisation of tumours.[3]
- 10–20% Of Caucasian patients with non-resectable lung adenocarcinoma have EGFR mutations, leading to approximately 294 new patients annually in the Netherlands.[4]
- The therapeutic landscape for these patients has changed radically with EGFR-tyrosine kinase inhibitors (TKIs) becoming the new first-line standard treatment. Eventually the majority (50-70%) progress on first-line TKI treatment by developing secondary mutations such as the T790M point mutation.
- **A budget impact analysis was performed to assess the value of sequential treatment for NSCLC patients with common EGFR mutations with first-line afatinib versus first-line osimertinib, followed by subsequent treatments in the Netherlands.**

METHODS

- A decision analytic model was constructed in Microsoft Excel with a Dutch healthcare perspective over a time horizon of 5 years in accordance with the ISPOR's good practices for budget impact analysis.[5] See table 1 for an overview of model parameters.
- Comparison of 2 treatment strategies:
 - ★First-line afatinib, followed by second-line osimertinib (when T790 mutation+) or by chemotherapy (when T790 mutation-), followed by third-line chemotherapy.
 - ★First-line osimertinib, followed by second-line chemotherapy.
- Out of 9175 patients with NSCLC in 2017 in the Netherlands, approximately 294 patients have EGFR deletion 19 or L858R mutations initiating first-line treatment every year.[2,4] Leading to an assumed monthly mean of 24.5 new patients entering the model per month.
- A survival model with reconstructed individual patient time-to-event data and a Weibull distribution was fitted using digitized progression free survival (PFS) curves with data from: FLAURA, AURA3 and IMPRESS studies for osimertinib and chemotherapy.[6,7,8] Patient level data from the LUX-Lung 3 clinical trial were used to fit the PFS curves for afatinib.[9] A network meta-analysis (NMA) was performed and the modelled PFS values were used as proxy to estimate time on treatment.[10]
- Only direct medical costs were taken into account. The mean cost for having grade 3 adverse events was calculated per patient per treatment step.[21]
- Scenario analyses and one-way sensitivity analysis tested the model robustness.

Table 1: Overview of model parameters for the budget impact model for sequential treatment with afatinib versus first-line osimertinib in NSCLC patients with common EGFR mutations

		Mean	Afatinib	Osimertinib		Chemotherapy	BSC/ Death	Source & Assumptions
				First-line	Second-line			
Transition probabilities – Afatinib treatment arm								
T790M mutation-positive	First-line		63%			--	--	[11]
	Second-line	--		88%		--	12%	Assumption
	Third-line	--	--			41%	59%	[7]
	Fourth-line	--	--			--	100%	Assumption
T790M mutation-negative	First-line		37%			--	--	[11]
	Second-line	--	--			88%	12%	[7, 12]
	Third-line	--	--			--	100%	Assumption
Transition probabilities – Osimertinib treatment arm								
First-line		--	--	100%	--	--	--	
Second-line		--	--	--	--	58%	42%	[6]
Third-line		--	--			--	100%	Assumption
Health states								
First-line		0.710	0.688	0.707		0.672	--	[6-9,12-14]
Second-line		0.730	0.708	0.727		0.692	--	
Third-line		0.620	0.598	0.617		0.582	--	
Simulated mean PFS from survival model								
First-line		--	16.99	20.80		--	--	[6,7,9]
Second-line		--	--	15.23		6.71	--	
Third-line		--	--	--		6.71	--	[8]
Costs								
Treatment costs/month		--	€2 440.15	€6 181.33		€3 679.04	--	[15-17]
T790M mutation testing		€157.02	--	--		--	--	[18]
Total cost per treatment for adverse events		--	€371.11	€62.46	€60.54	€554.33	--	[17,19-22]

RESULTS

The total budget impact over five years was €110.4 million for the sequence starting with afatinib and €158.6 million for the sequence starting with osimertinib, leading to a total incremental cost of €48.15 million over 5 years. The incremental cost savings were €6.06M, €12.25M, €12.26M, €9.87M and €7.72M for years 1 to 5 respectively, see figure 1.

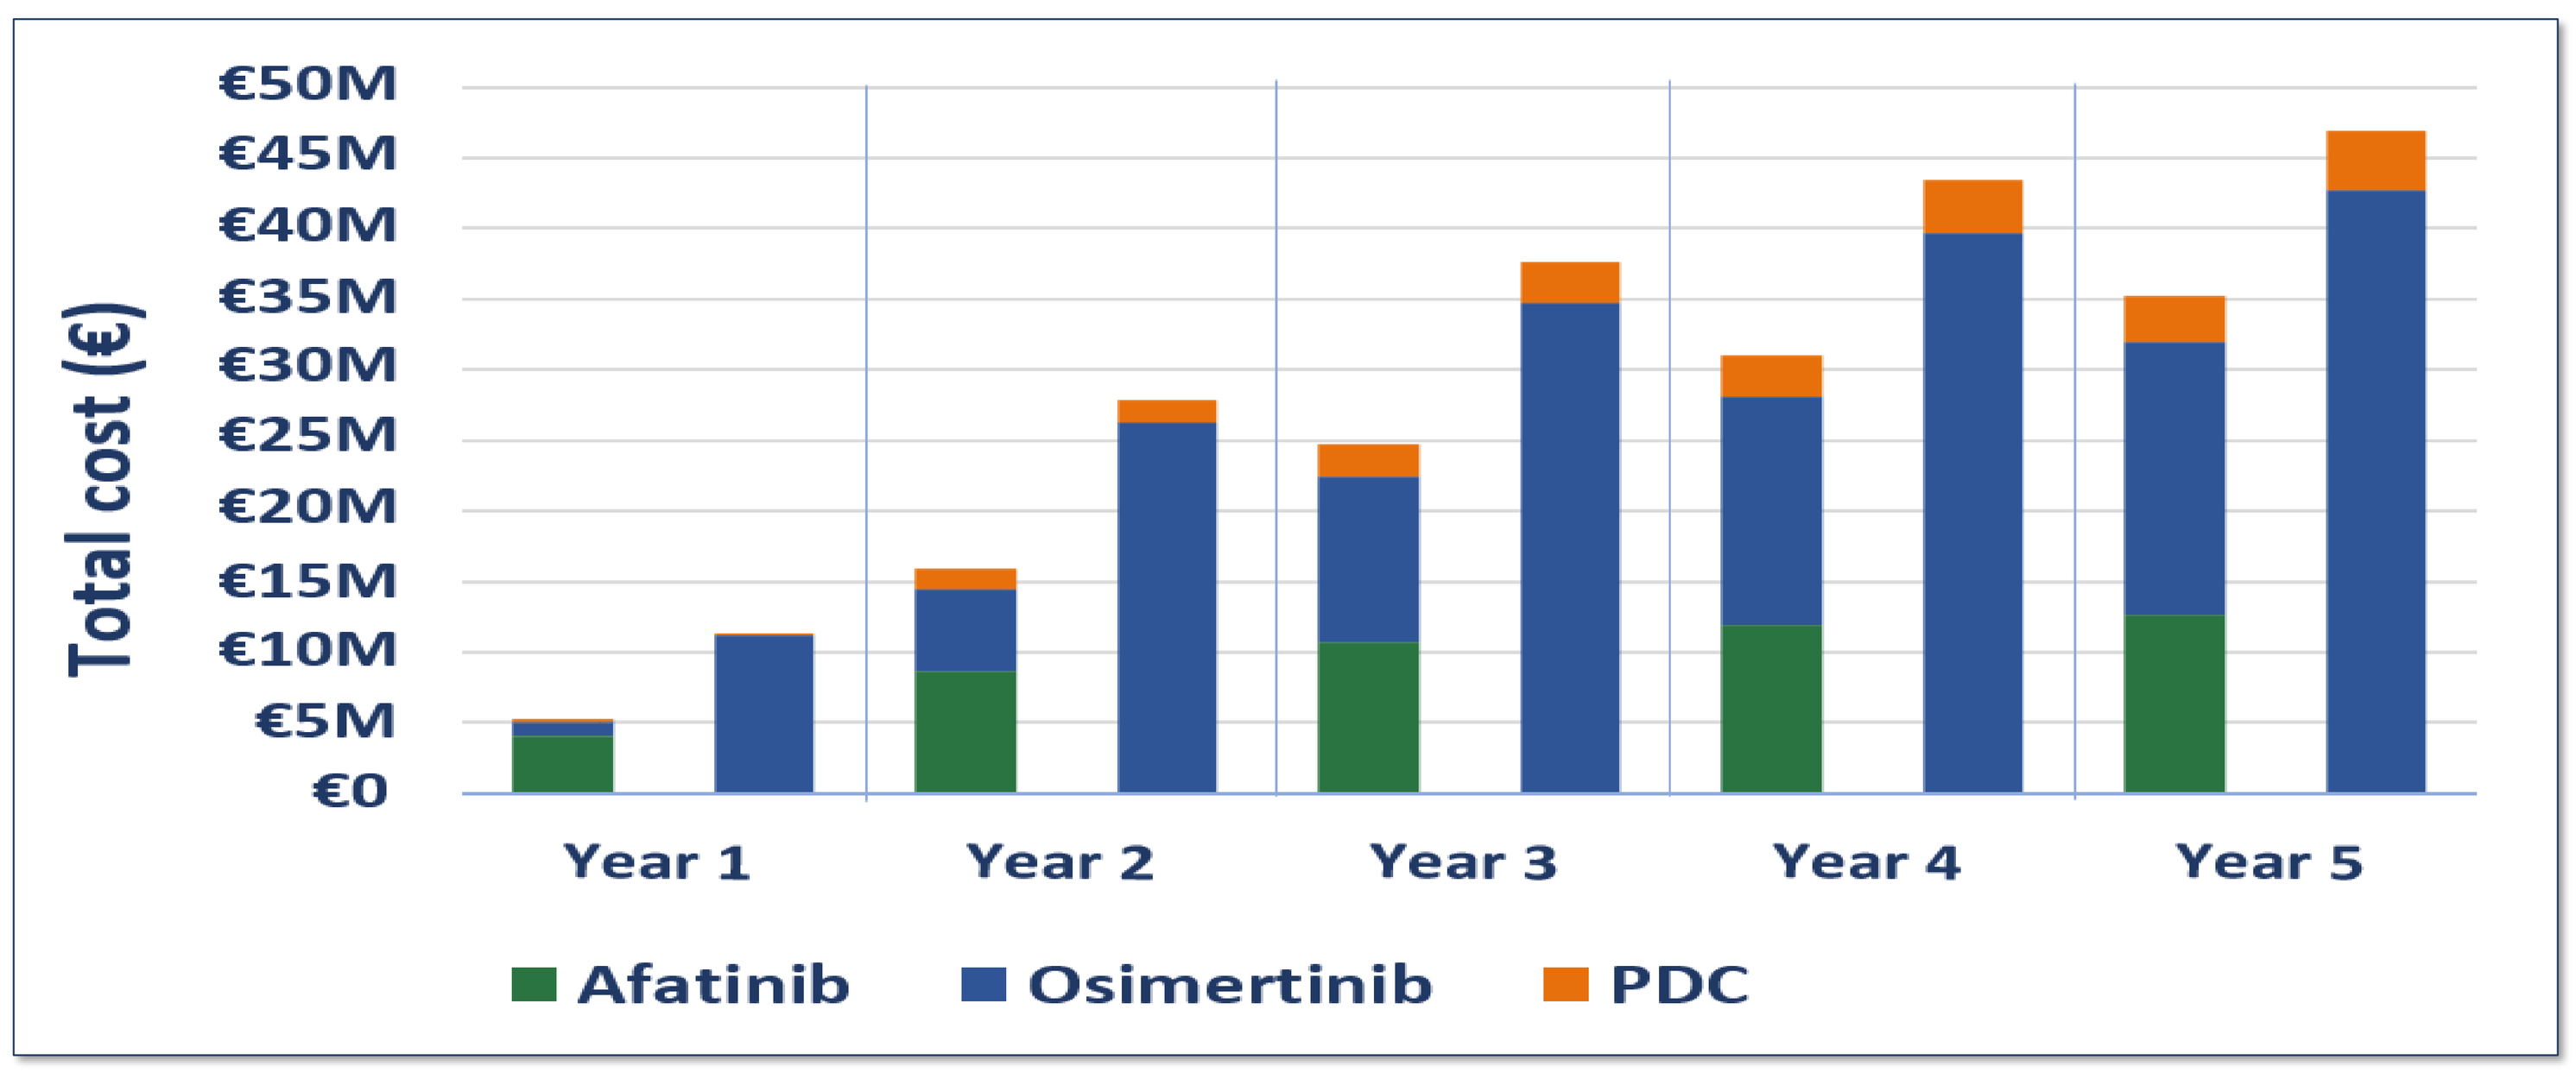
€50M
€45M
€40M
€35M
€30M
€25M
€20M
€15M
€10M
€5M
€0
Total cost (€)
Year 1
Year 2
Year 3
Year 4
Year 5
Afatinib
Osimertinib
PDC

Figure 1: Total cost (€) per year for first-line treatment strategies afatinib versus osimertinib

Sequential treatment with first-line afatinib showed mean total time on treatment of 29.1 months, quality-adjusted life months (QALMs) of 20.2 with mean cost of €108,166 per patient versus first-line osimertinib with mean total time on treatment of 24.7 months, QALM of 17.4 and mean cost per patient of €143,251.

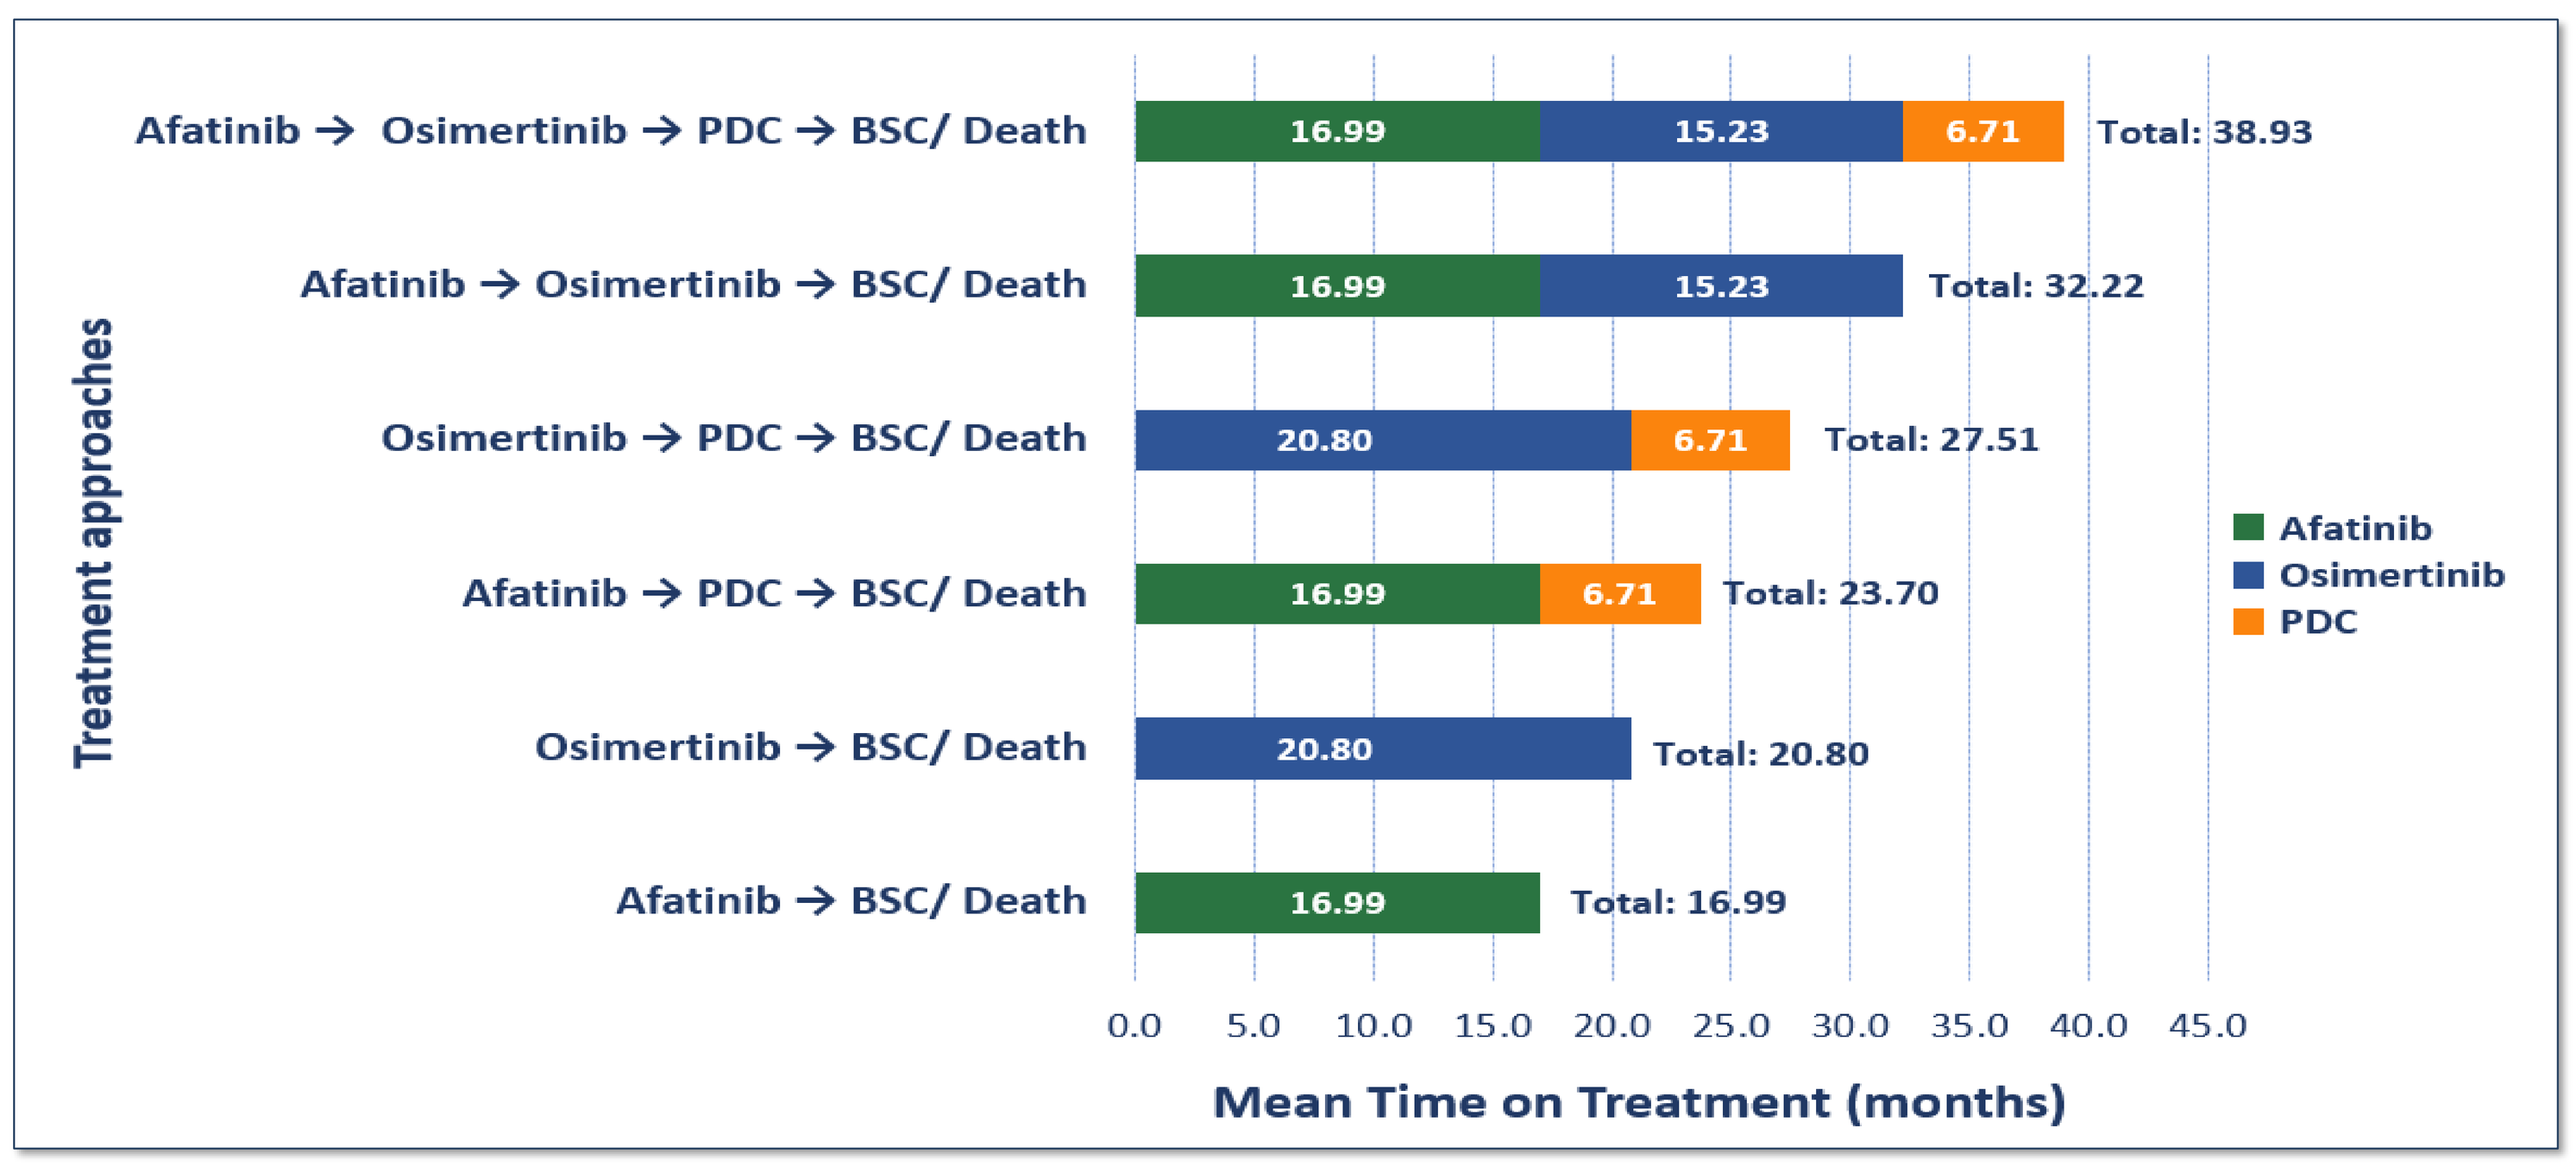
Treatment approaches
Afatinib → Osimertinib → PDC → BSC/ Death
16.99
15.23
6.71
Total: 38.93
Afatinib → Osimertinib → BSC/ Death
16.99
15.23
Total: 32.22
Osimertinib → PDC → BSC/ Death
20.80
6.71
Total: 27.51
Afatinib → PDC → BSC/ Death
16.99
6.71
Total: 23.70
Osimertinib → BSC/ Death
20.80
Total: 20.80
Afatinib → BSC/ Death
16.99
Total: 16.99
0.0 5.0 10.0 15.0 20.0 25.0 30.0 35.0 40.0 45.0
Mean Time on Treatment (months)
Afatinib
Osimertinib
PDC

Figure 2: Outcomes survival analyses with Weibull-distribution in decision analytic model: total mean Time on Treatment for patients by treatment paradigms

Sensitivity analyses showed that the results were most sensitive to the price of osimertinib and afatinib, shown in figure 3. Results were also sensitive to the percentage of patients with T790M mutation+ disease receiving subsequent treatment, the percentage of patients testing positive for T790M mutations and the percentage of subsequent treatment after first-line therapy with osimertinib.

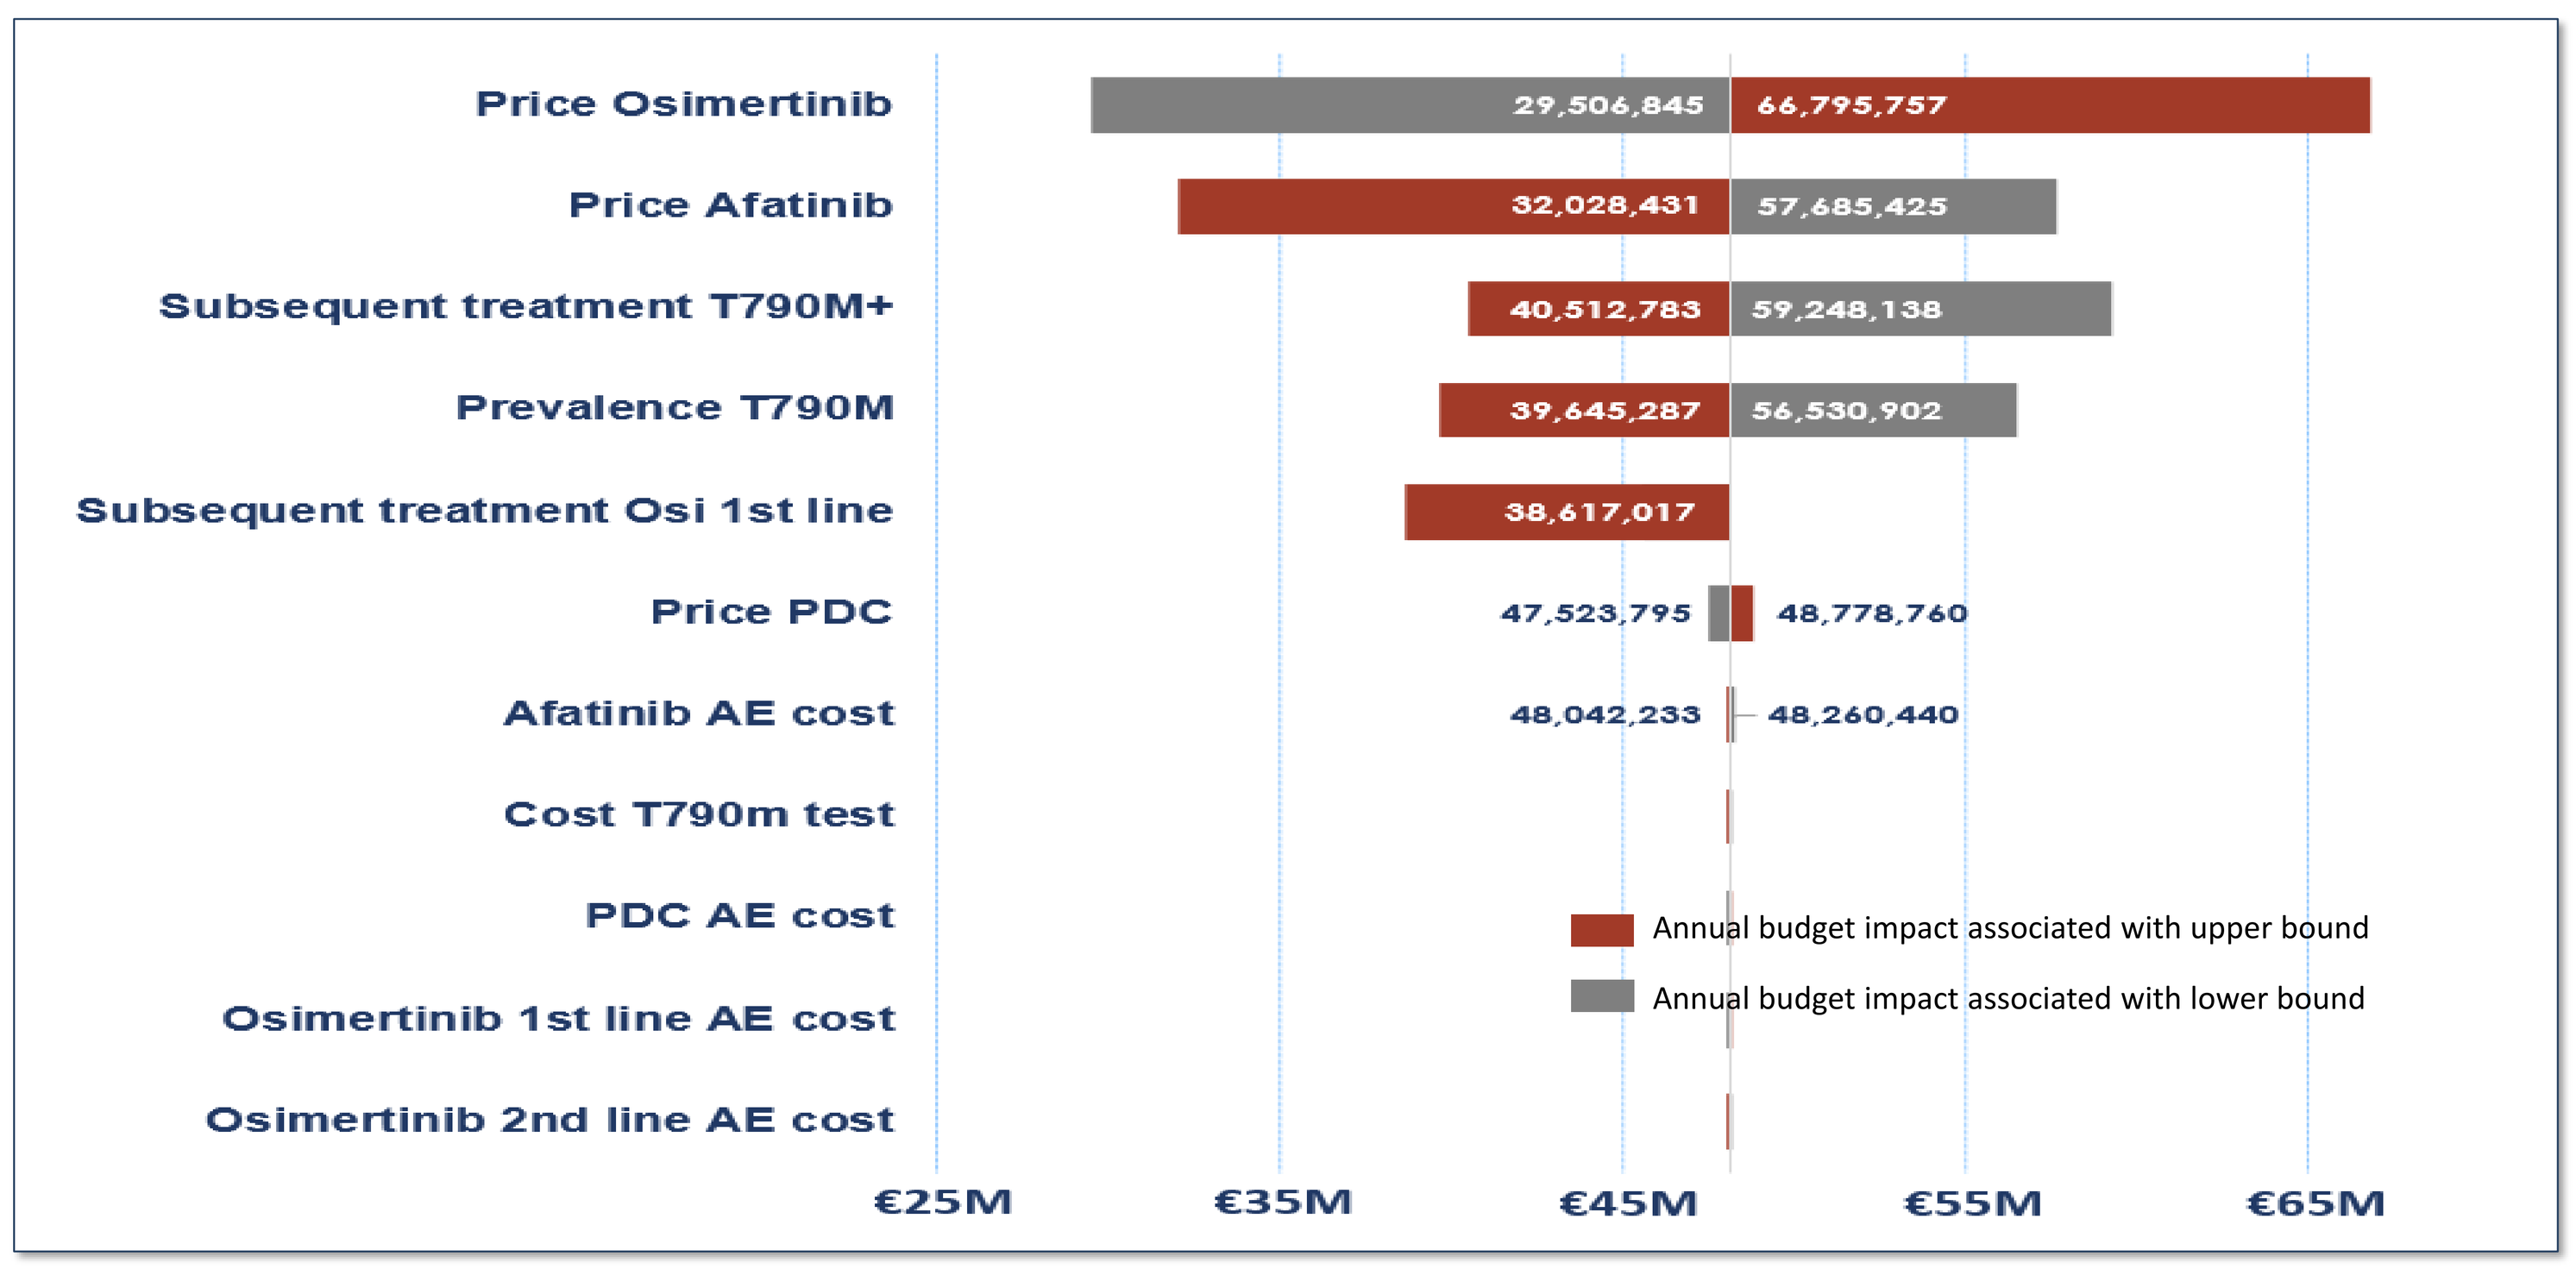
Price Osimertinib
29,506,845
66,795,757
Price Afatinib
32,028,431
57,685,425
Subsequent treatment T790M+
40,512,783
59,248,138
Prevalence T790M
39,645,287
56,530,902
Subsequent treatment Osi 1st line
38,617,017
Price PDC
47,523,795
48,778,760
Afatinib AE cost
48,042,233
48,260,440
Cost T790m test
PDC AE cost
Osimertinib 1st line AE cost
Osimertinib 2nd line AE cost
Annual budget impact associated with upper bound
Annual budget impact associated with lower bound
€25M
€35M
€45M
€55M
€65M

Figure 3: One-way sensitivity analysis: impact of changes of input parameters on the change in total annual budget in year 5

CONCLUSION

Over the last decade various treatment options showed improved PFS data in clinical trials and will support improvement of the burden of disease for lung cancer. The availability of these treatment options has an economic impact.

Sequential treatment with afatinib as a first-line treatment approach compared to a sequential treatment approach starting with osimertinib could be of high value since incremental cost savings were estimated at €48.15 million over 5 years, together with an incremental Time on Treatment of 4.5 months and an incremental QALM of 2.8 in patients with EGFR mutation positive NSCLC.

References: 1. Bray F, Ferlay J, Soerjomataram I, et al. Global cancer statistics 2018: GLOBOCAN estimates of incidence and mortality worldwide for 36 cancers in 185 countries. CA Cancer J Clin. 2018; 68: 394-424. **2.** Integrated Cancer center the Netherlands (IKNL), 2017, Dutch Cancer registration, Available: https://www.cijfersoverkanker.nl/selecties/Dataset_1/img5af2af2a2b91e, October 2,2018. **3.** Midha et al. EGFR mutation incidence in non-small-cell lung cancer of adenocarcinoma histology: a systematic review and global map by ethnicity (mutMapII); Am J Cancer Res; 2015; 5(9): 2892-2911. **4.** Planchard D, Popat S, Kerr K, et al. Metastatic non-small cell lung cancer: ESMO Clinical Practice Guidelines for diagnosis, treatment and follow-up. Ann Oncol. 2018; 29: iv192-iv237. **5.** Sullivan SD, Mauskopf JA, Augustovski F, et al. Budget impact analysis-principles of good practice: report of the ISPOR 2012 Budget Impact Analysis Good Practice II Task Force. Value Health. 2014; 17: 5-14. **6.** Soria JC, Ohe Y, Vansteenkiste J, et al. Osimertinib in Untreated EGFR-Mutated Advanced Non-Small-Cell Lung Cancer. N Engl J Med. 2018; 378: 113-25. **7.** Mok TS, Wu YL, Ahn MJ, et al. Osimertinib or Platinum-Pemetrexed in EGFR T790M-Positive Lung Cancer. N Engl J Med. 2017; 376: 629-40. **8.** Soria JC, Wu YL, Nakagawa K, et al. Gefitinib plus chemotherapy versus placebo plus chemotherapy in EGFR-mutation-positive non-small-cell lung cancer after progression on first-line gefitinib (IMPRESS): a phase 3 randomised trial. Lancet Oncol. 2015; 16: 990-8. **9.** Sequist LV, Yang JC, Yamamoto N, et al. Phase III study of afatinib or cisplatin plus pemetrexed in patients with metastatic lung adenocarcinoma with EGFR mutations. J Clin Oncol. 2013: Jul 1 [Epub ahead of print]. 10. Guyot P, Ades A, Ouwens MJ, Welton NJ. Enhanced secondary analysis of survival data: reconstructing the data from published Kaplan-Meier survival curves. BMC Medical Research Methodology. 2012; 12: 9. 11. Yu HA etal. Analysis of tumor specimens at the time of acquired resistance to EGFR-TKI therapy in 155 patients with EGFR-mutant lung cancers. Clin Cancer Res. 2013; 19: 2240-7. 12. Park K et al. Afatinib versus gefitinib as first-line treatment of patients with EGFR mutation-positive non-small-cell lung cancer (LUX-Lung 7): a phase 2B, open-label, randomised controlled trial. Lancet Oncol. 2016; 17: 577-89. 13. Nafees B et al. Health state utilities for non small cell lung cancer. Health Qual Life Outcomes. 2008; 6: 84. 14. Chouaid C et al. Health-related quality of life and utility in patients with advanced non-small-cell lung cancer: a prospective cross-sectional patient survey in a real-world setting. J Thorac Oncol. 2013; 8: 997-1003. 15. Wijma S., 2018, Healthcare Insitute the Netherlands; Package advice osimertinib (Tagrisso) for first line treatment of paients with advanced or metastatic non-small-cell lung cancer (NSCLC) with activating EGFR-mutations, Dutch Osimertinib dossier for Ministry of Health, Ref.nr: 2018051432. 16. Healthcare Institute the Netherlands, 2018, The price for a 30-day prescription of afatinib, Available https://www.medicijnkosten.nl/databank?zoekterm=afatinib&toedieningsvorm=TABLETTEN%20EN%20CAPSULES&ddd_per_artikel=0&hoeveelheid=30,00, December 11, 2018. 17. G-Standard, Dutch treatment database (Z-index), October 2018. 18. National Institute for Health and Care Excellence. Plasma EGFR mutation tests for adults with locally advanced or metastatic non-small-cell lung cancer (MIB137). https://wwwniceorguk/advice/mib137. 2018. 19. Uyl-de Groot CA AIMJ, Zaim R. Nivolumab (Opdivo) bij Gevorderd Plaveiselcelcarcinoom van de Long. Instituut Beleid & Management Gezondheidszorg. 2015. 20. Wehler E et al. Economic burden of toxicities associated with treating metastatic melanoma in eight countries. Eur J Health Econ. 2017; 18: 49-58. 21. Mickisch G et al. Costs of managing adverse events in the treatment of first-line metastatic renal cell carcinoma: bevacizumab in combination with interferon-alpha2a compared with sunitinib. Br J Cancer. 2010; 102: 80-6. 22. Hakkaart-van Roijen L et al. Cost manual, Methodlogy for cost research and reference pricing for economic evaluations in healthcare. 2015, paragraph 3.1.2.8; Outpatient setting.

ISPOR 22nd Annual European Congress, Copenhagen, Denmark (November 2-6, 2019)