REAL-WORLD GOLIMUMAB EFFECTIVENESS AND IMPACT ON PATIENT-REPORTED OUTCOMES IN PATIENTS WITH RHEUMATOID ARTHRITIS, PSORIATIC ARTHRITIS OR AXIAL SPONDYLARTHRITIS AND A TREATMENT FAILURE TO PREVIOUS TNFα INHIBITOR THERAPY

Authors: Georgiadis A[1], Theodoridou A[2], Vounotrypidis P[3], Zisopoulos D[4], Kallitsakis I[5], Kandyli A[6], Migkos M[7], Bournazos I[8], Sidiropoulos P[9], Efstathiou M[10], Athanassiou P[11], Haratsis Y[12], Petrikkou E[12], Vassilopoulos D[13]

Affiliations: 1. Private practice, Ioannina, Greece; 2. Private practice, Thessaloniki, Greece; 3. Private practice, Thessaloniki, Greece; 4. Private practice, Thessaloniki, Greece; 5. Private practice, Chania, Greece; 6. Private practice, Athens, Greece; 7. Private practice, Florina, Greece; 8. Private practice, Athens, Greece; 9. Department of Rheumatology, Clinical Immunology and Allergy, University of Crete, Heraklion, Greece; 10. Private practice, Thessaloniki, Greece; 11. Department of Rheumatology, St. Paul's General Hospital, Thessaloniki, Greece; 12. Merck Sharp & Dohme Pharmaceutical, Industrial and Commercial S.A, Athens, Medical Affairs, Greece; 13. 2nd Department of Internal Medicine and Laboratory, Hippokration General Hospital, Athens, Greece

OBJECTIVES

Primary objective:
To evaluate the **real-world effectiveness** of golimumab after 6 months in patients with active RA, PsA, or axSpA, by:
- **RA patients:** proportion of patients who achieved **Low Disease Activity (LDA)**, according to DAS28-CRP at the end of month 6 after initiation of golimumab.
- **PsA patients:** proportion of patients who achieved a **Minimal Disease Activity (MDA)** at the end of month 6 after initiation of golimumab.
- **axSpA patients:** proportion of patients who achieved **Moderate Disease Activity** according to the Bath Ankylosing Spondylitis Disease Activity Index [BASDAI] and **inactive disease** by Ankylosing Spondylitis Disease Activity Score [ASDAS] at the end of month 6 after initiation of golimumab.

Other objectives include:
the assessment of work productivity/activity impairment (by Work Productivity and Activity Impairment Questionnaire for RA) and quality of life (QoL; by EQ-5D-3L), at 3/6/12/18 months and changes from baseline.

RA = Rheumatoid arthritis ; PsA = Psoriatic arthritis ; axSpA = Axial spondyloarthritis

METHODS

GO-BEYOND: a non-interventional prospective study in patients with active RA, PsA or axSpA and an inadequate response/intolerability to 1 TNFα inhibitor, who are considered for golimumab treatment.
This interim analysis presents results for patients who participated in the study for 6 months (cut-off date: 28/02/2019).
Per-indication statistical comparisons were not performed due to small sample size at the time of this interim analysis.

Distribution of patients per indication (N=36)

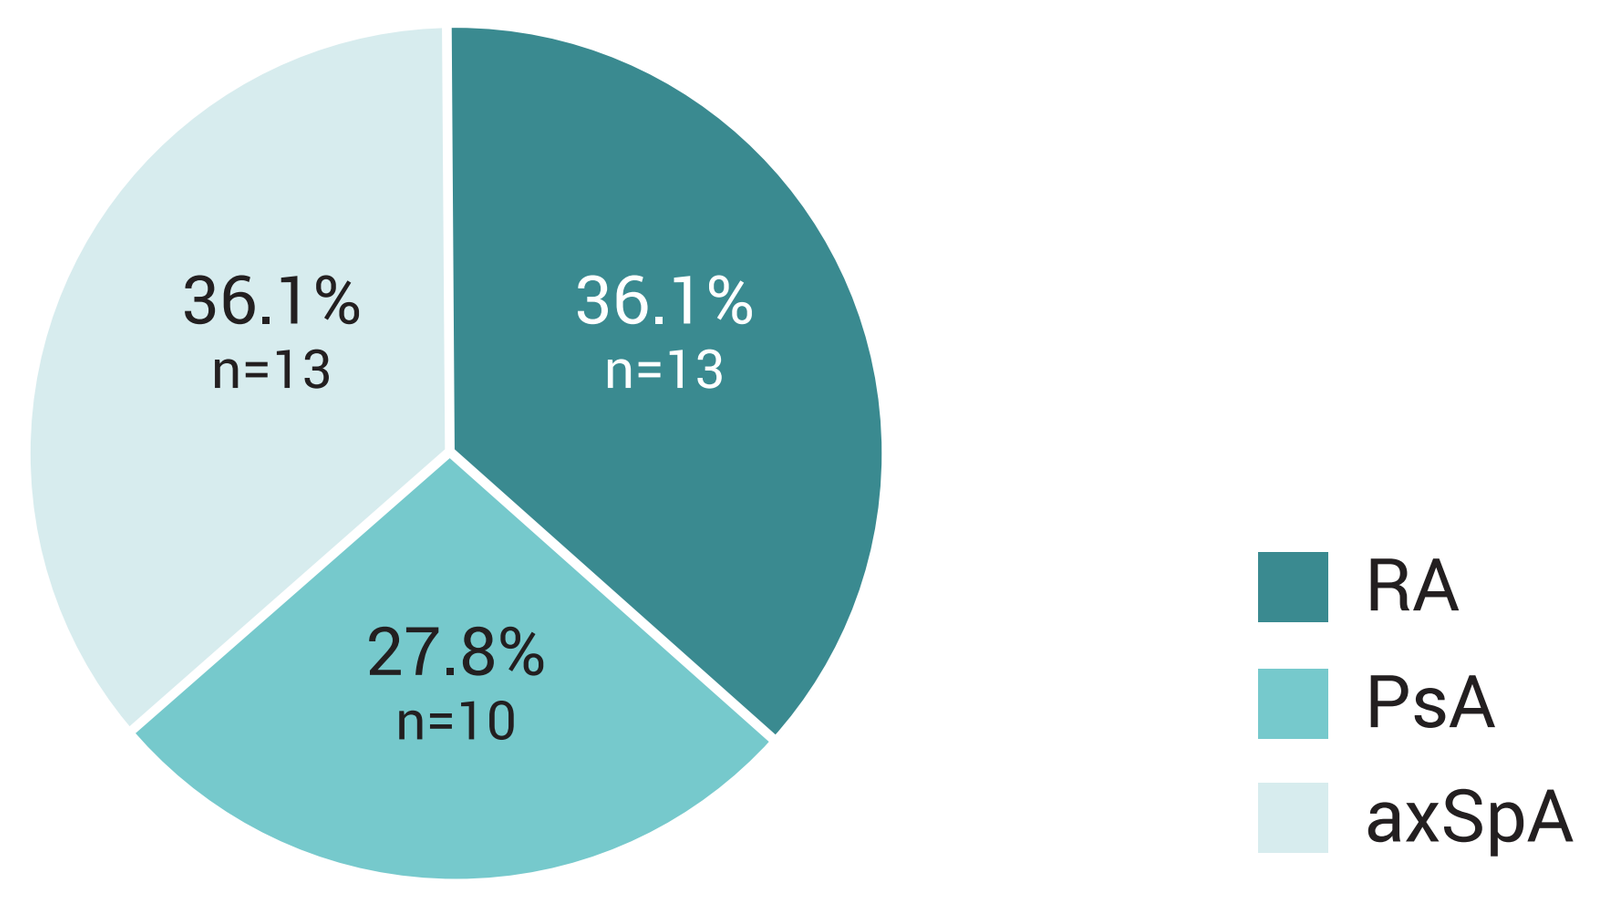
36.1%
n=13
36.1%
n=13
27.8%
n=10
RA
PsA
axSpA

Majority of RA patients achieved Low Disease Activity (LDA; DAS28-CRP<3.2) after 6 months of treatment (Baseline: N=13, 6m: N=11)

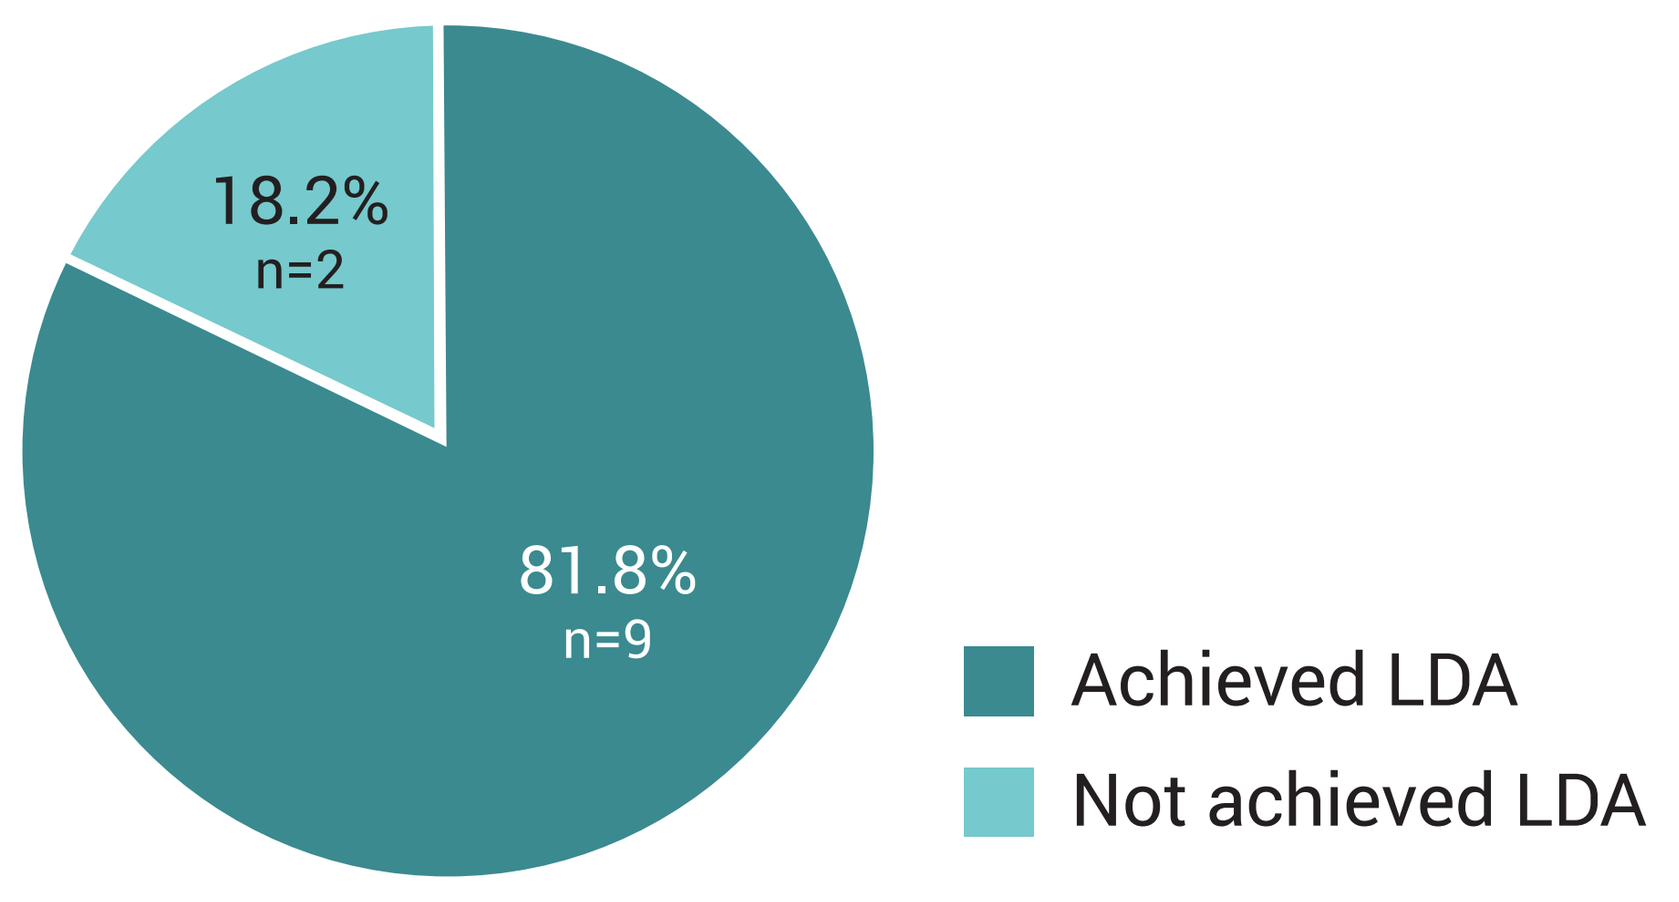
18.2%
n=2
81.8%
n=9
Achieved LDA
Not achieved LDA

More than 10% of PsA patients achieved Minimal Disease Activity (MDA; as per MDA criteria) after 6 months of treatment (Baseline: N=10, 6m: N=9)

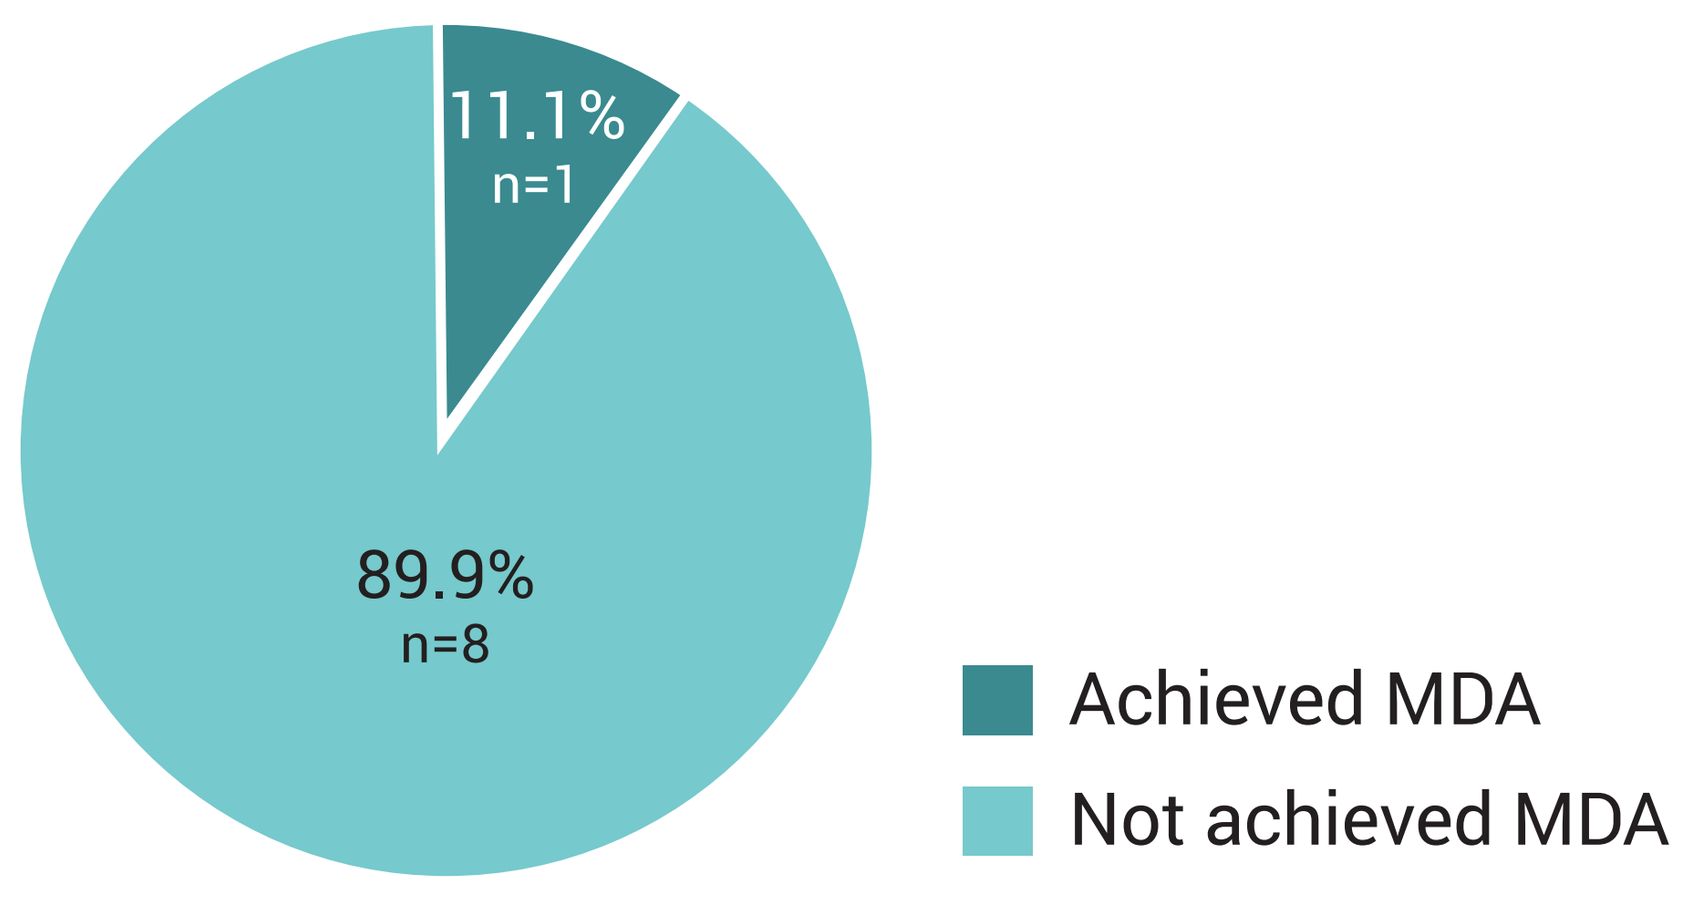
11.1%
n=1
89.9%
n=8
Achieved MDA
Not achieved MDA

More than 1/3 axSpA patients achieved moderate disease activity by BASDAI and inactive disease by ASDAS after 6 months of treatment (Baseline, 6m: N=13)

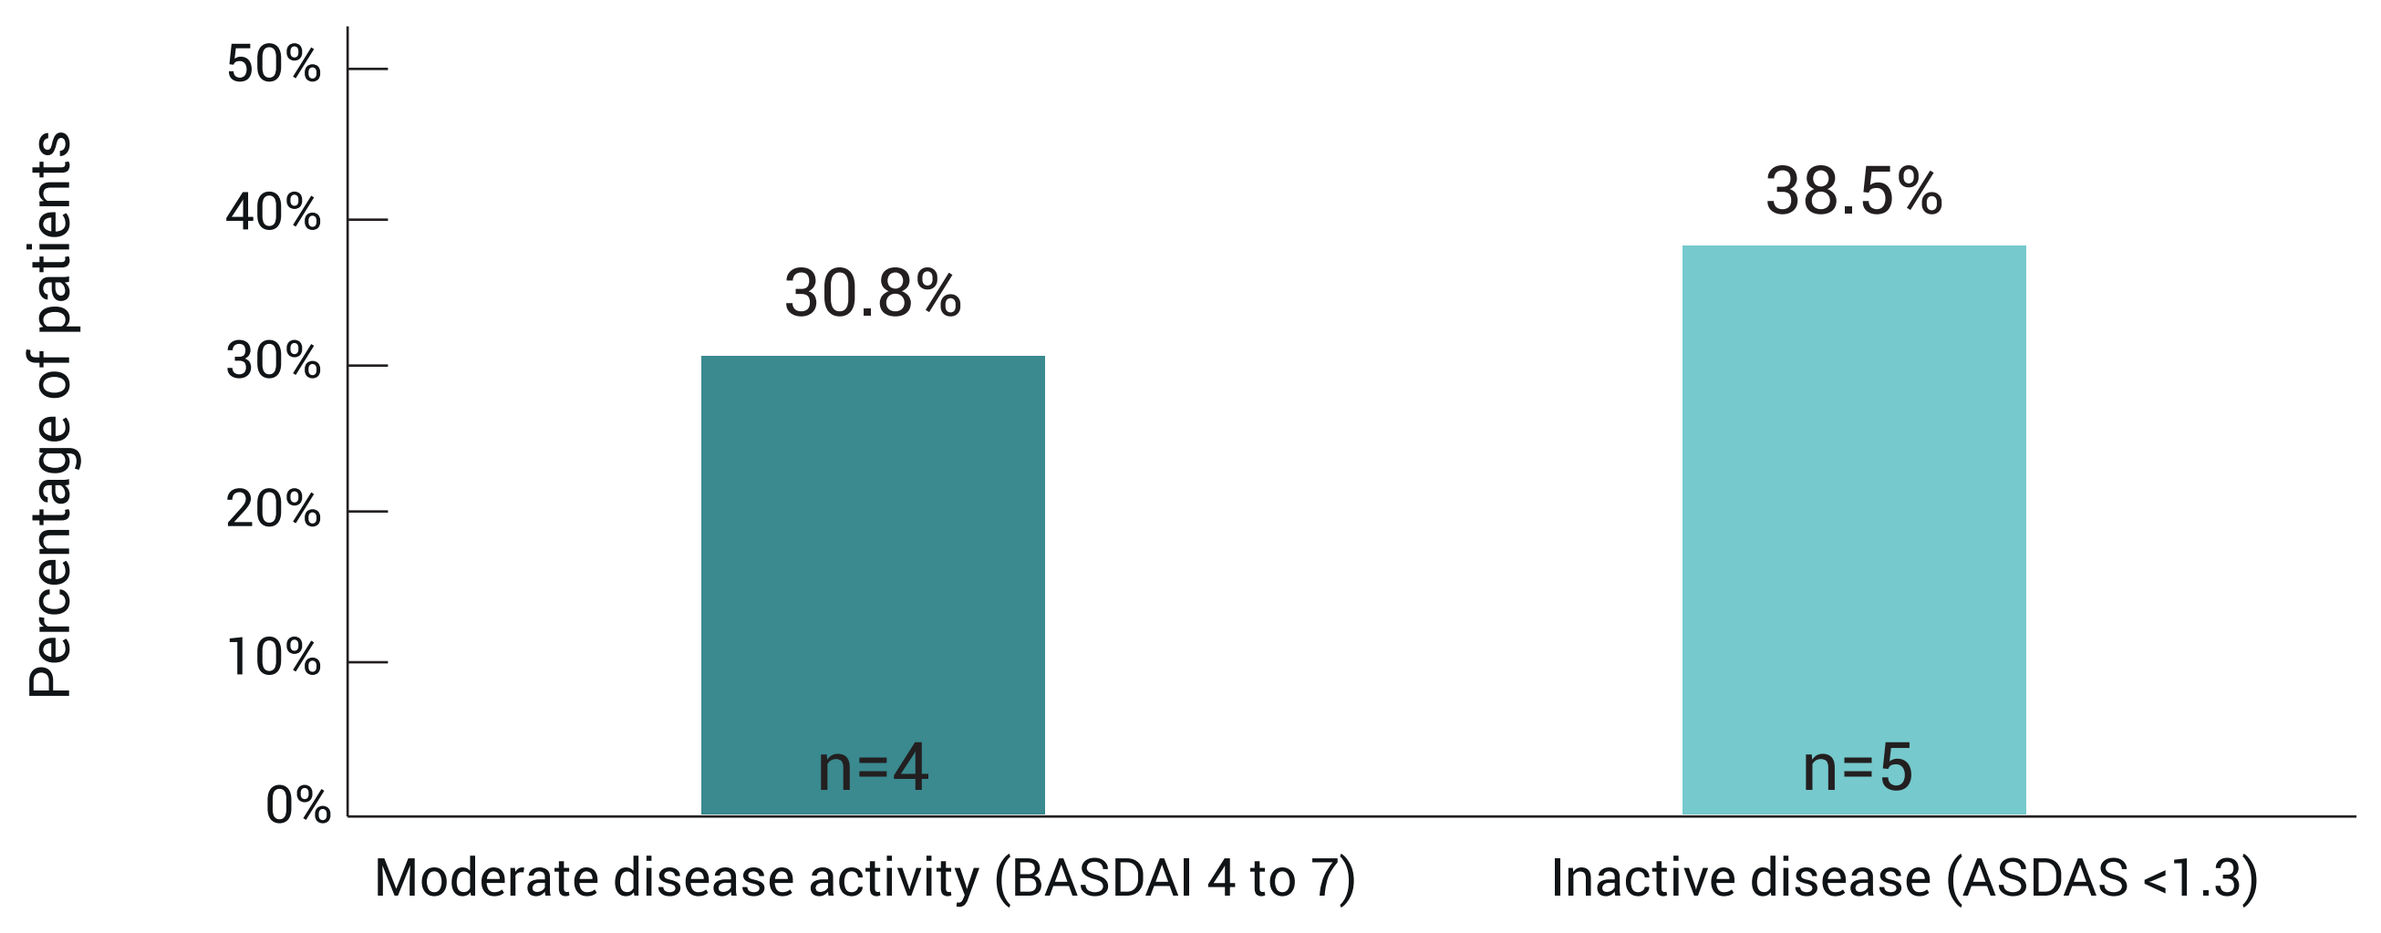
Percentage of patients
50%
40%
30%
20%
10%
0%
30.8%
n=4
38.5%
n=5
Moderate disease activity (BASDAI 4 to 7)
Inactive disease (ASDAS <1.3)

Notable reduction in work productivity after 3 and 6 months of treatment compared to baseline

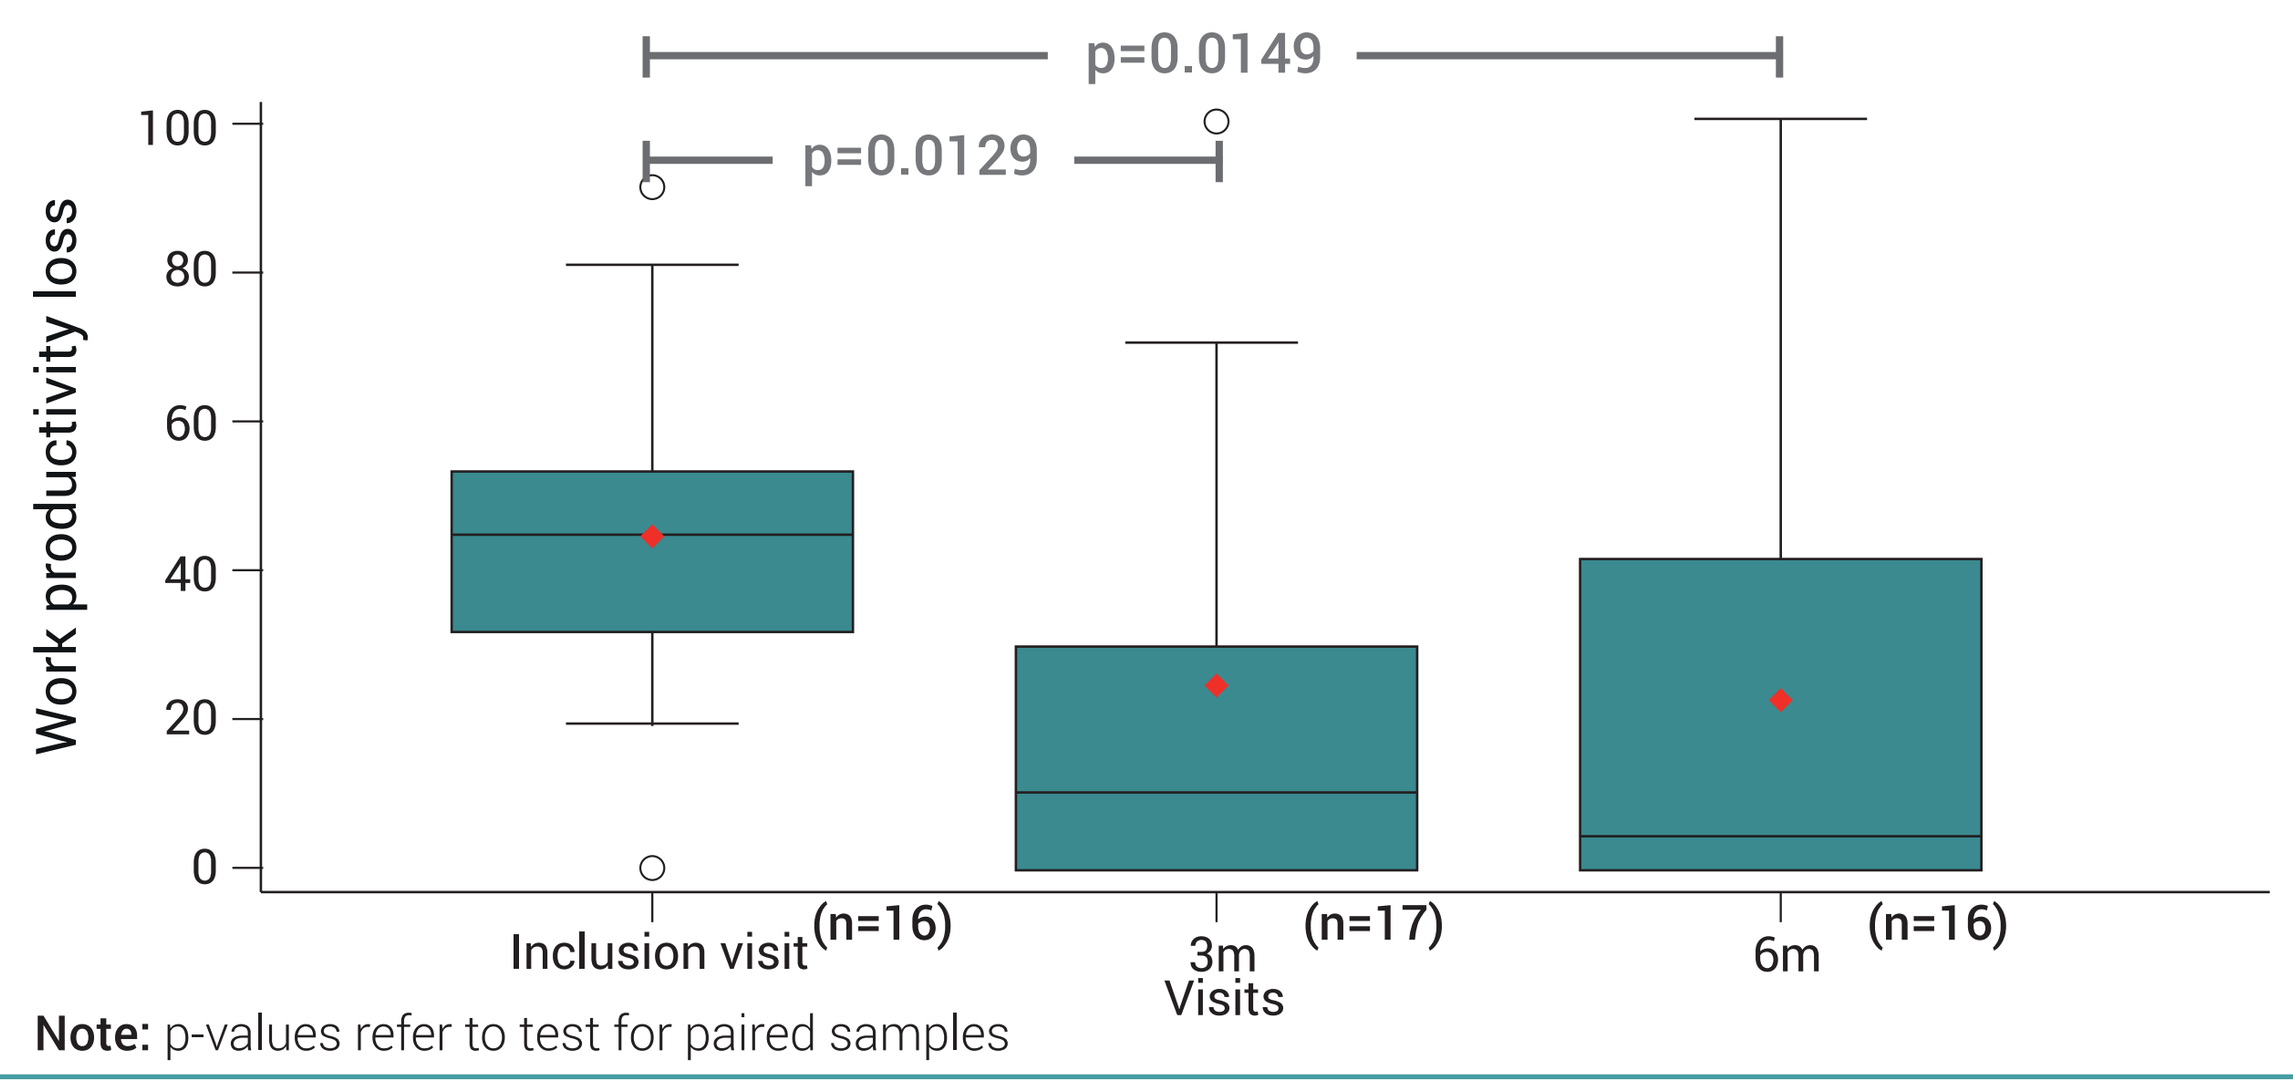
p=0.0149
p=0.0129
Work productivity loss
100
80
60
40
20
0
Inclusion visit (n=16)
3m (n=17)
6m (n=16)
Visits

Note: p-values refer to test for paired samples

Notable reduction in activity impairment after 3 and 6 months of treatment compared to baseline

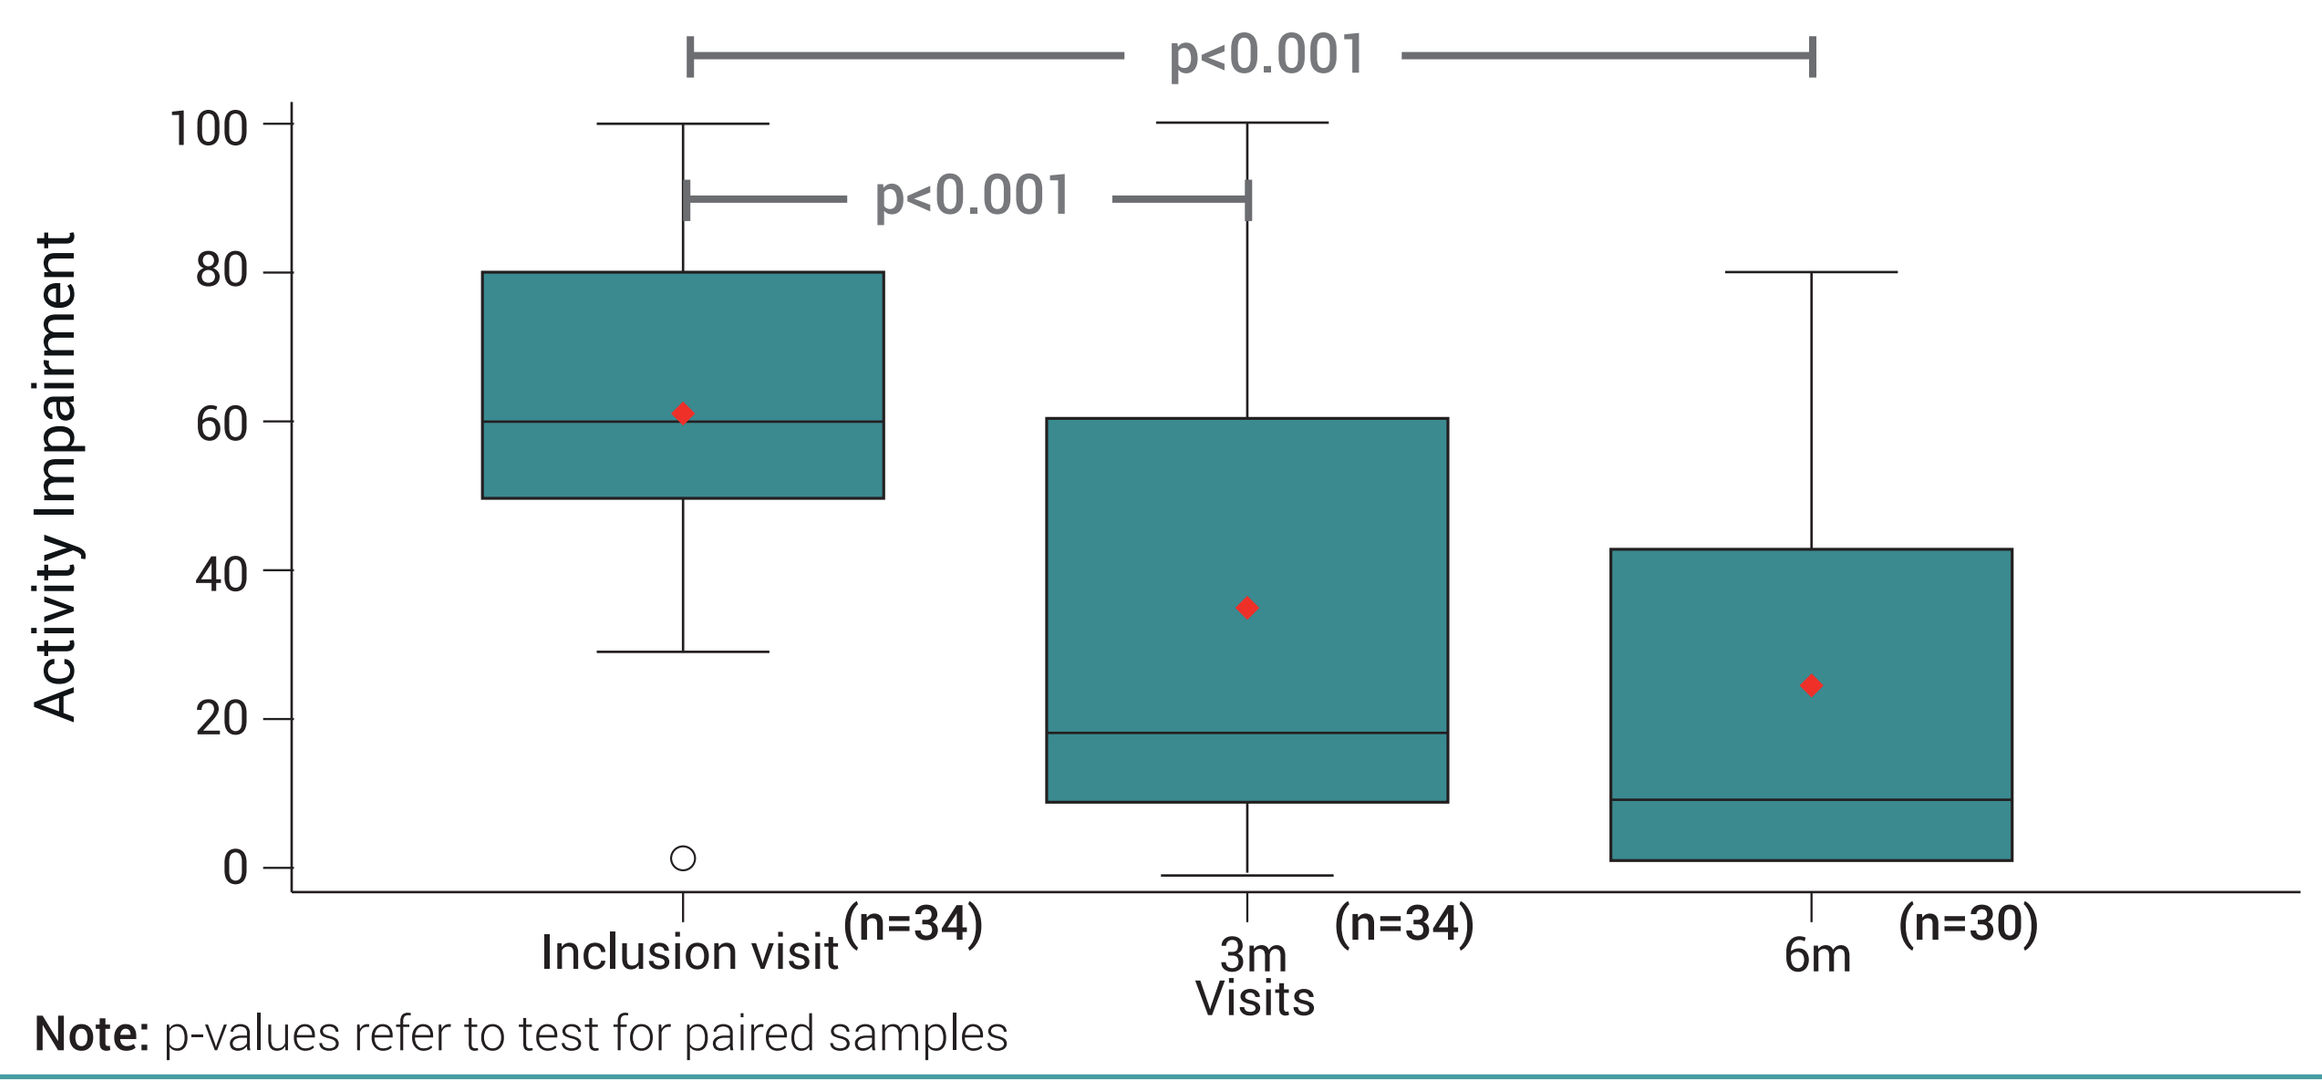
p<0.001
p<0.001
Activity Impairment
100
80
60
40
20
0
Inclusion visit (n=34)
3m (n=34)
6m (n=30)
Visits

Note: p-values refer to test for paired samples

Notable improvements in Quality of life by EQ-5D-3L UK index scores after 3 and 6 months of treatment compared to baseline

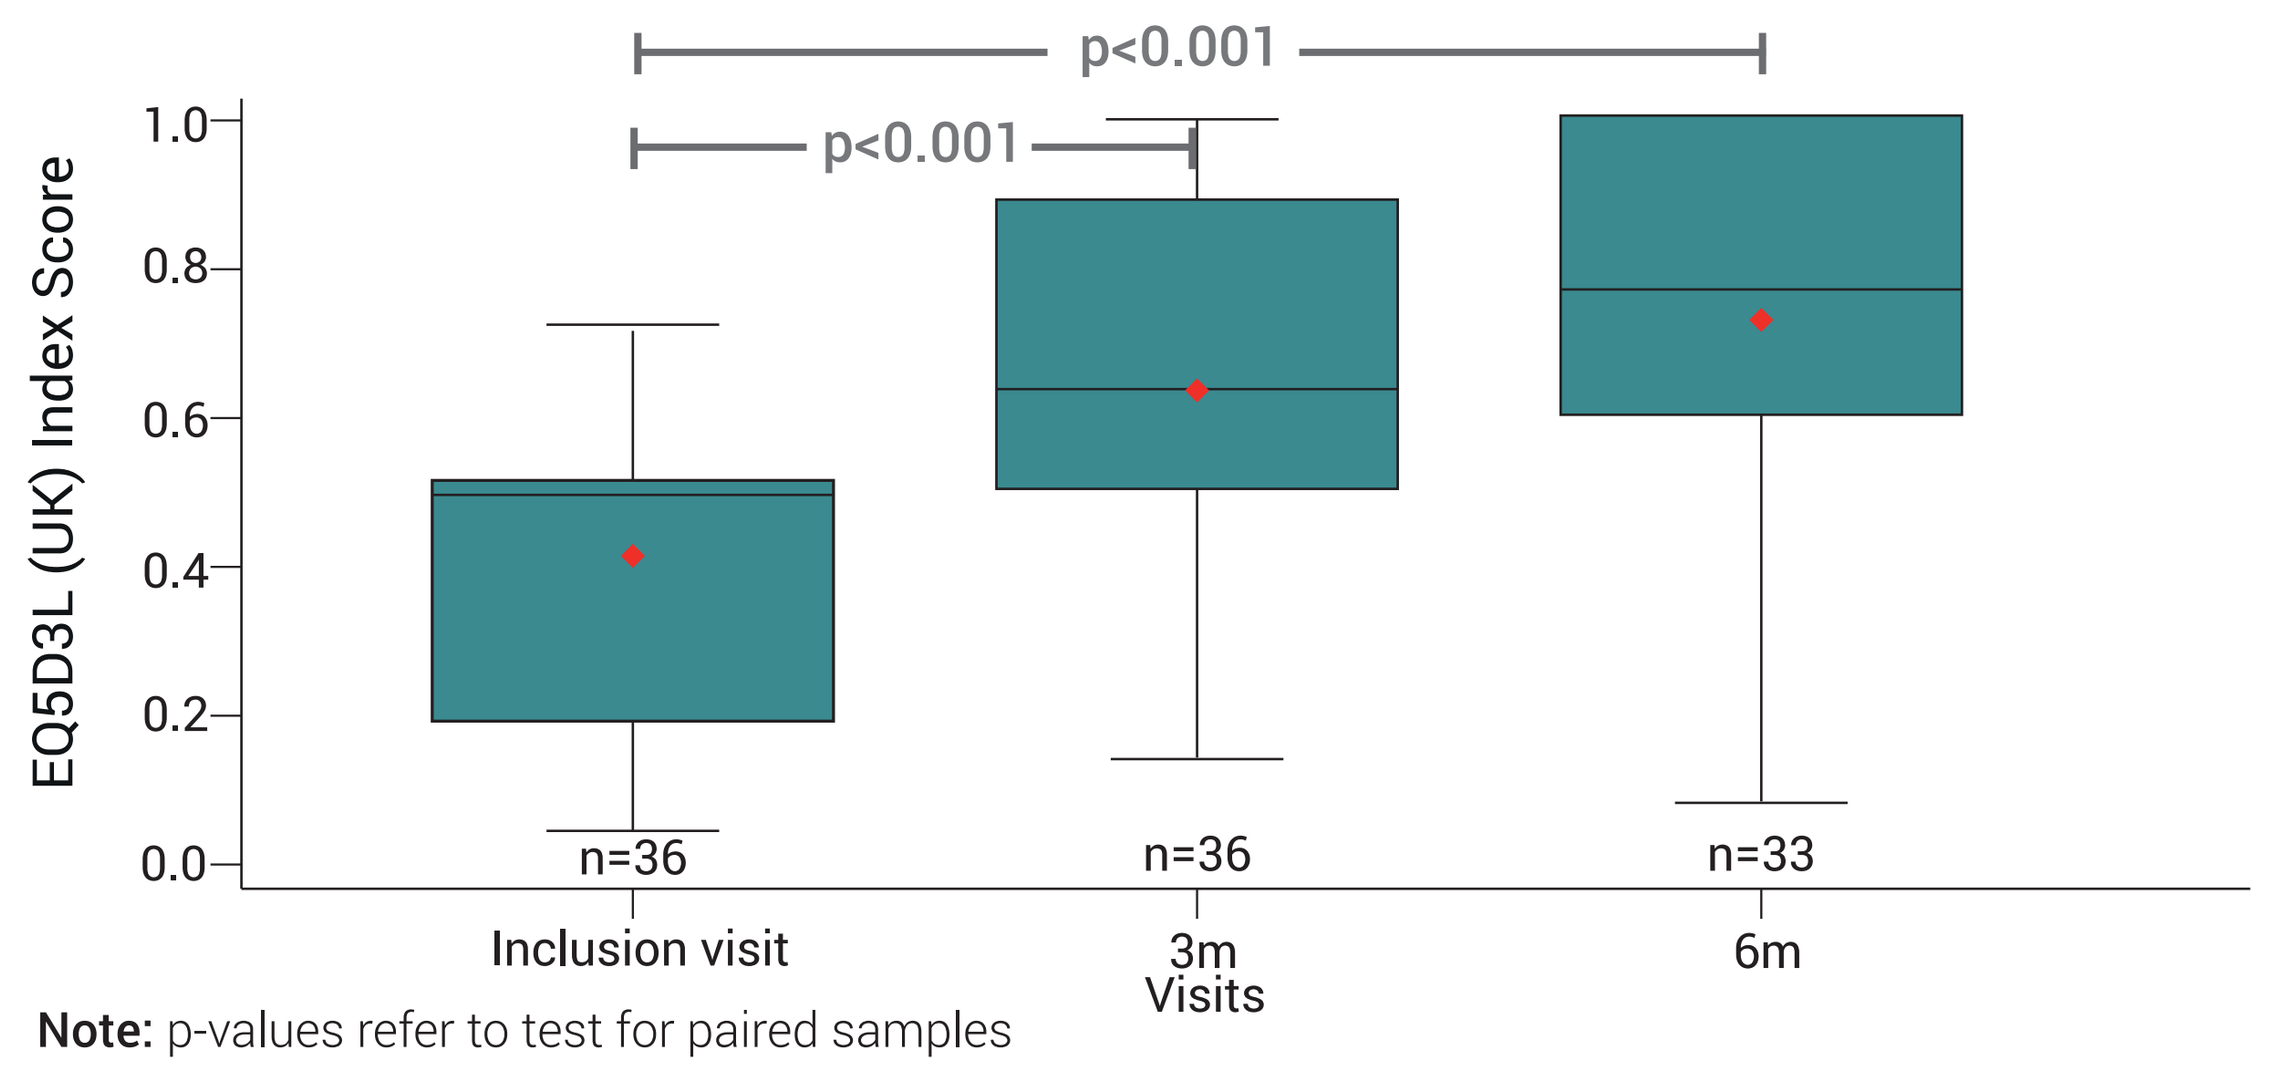
p<0.001
p<0.001
EQ5D3L (UK) Index Score
1.0
0.8
0.6
0.4
0.2
0.0
n=36
n=36
n=33
Inclusion visit
3m
6m
Visits

Note: p-values refer to test for paired samples

CONCLUSIONS

In this interim analysis, patients with active RA, PsA or axSpA showed improvements in disease activity, work productivity, activity impairment and quality of life **after 6 months of golimumab treatment.**

- ~82% of RA patients achieved low disease activity
- ~11% of PsA patients achieved moderate disease activity
- ~31% and ~39% of axSpA patients achieved moderate disease activity by BASDAI and inactive disease by ASDAS, respectively

These findings need to be considered within the limitations of the present analysis, mainly, the small sample size.

This study was sponsored by Merck Sharp & Dohme (MSD), Greece.