CHANGES IN PAIN AND STIFFNESS EXPERIENCED BY PATIENTS WITH TENOSYNOVIAL GIANT CELL TUMOR (TGCT) IN THE MANEUVER PHASE 3 TRIAL: INSIGHTS FROM EXIT INTERVIEWS

PCR18

Vinod Ravi,[1] Paul Kamudoni,[2] Andrea Phillips Beyer,[2] Niki Karachaliou,[2] Amy Clark,[3] Agkreta Leventi,[3] Savita Bakhshi Anand,[3] Carla Dias-Barbosa,[4] Hans Gelderblom[5]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]The Healthcare business of Merck KGaA, Darmstadt, Germany; [3]PPD Evidera Patient-Centered Research, Thermo Fisher Scientific, London, United Kingdom; [4]PPD Evidera Patient-Centered Research, Thermo Fisher Scientific, Ivry-sur-Seine, France; [5]Leiden University Medical Center, Leiden, The Netherlands

CONCLUSION

- **Similar to the findings of the clinical trial data, pain and stiffness were highlighted as important aspects of the patient experience**
- **This study highlights the baseline impact of pain and stiffness on patients with TGCT, with most patients experiencing symptoms of pain and stiffness at baseline**
- **During exit interviews, most patients treated with pimicotinib reported improvements in pain and stiffness at the end of Part 1, consistent with patient-reported outcome (PRO) data in the overall MANEUVER population[1]**
- **Patients with TGCT reported that they understood the Worst Pain and Worst Stiffness numeric rating scale (NRS) and found it relevant to their condition**
- **Results support the content validity of the Worst Pain and Worst Stiffness NRS and identified a patient-informed threshold for interpreting change on the NRS scale, with most patients reporting a 2-point change as a clinically meaningful improvement**

PLAIN LANGUAGE SUMMARY

- **TGCT is a rare disease where an abnormal growth of cells (tumor) forms in and around a joint. If not properly treated, it could cause lasting joint dysfunction, difficulty moving, and reduced quality of life**
- **The MANEUVER trial compared pimicotinib with a placebo ('dummy' treatment) for the treatment of TGCT**
- **Health authorities are interested in understanding patients' experiences with treatments. To capture this, some patients in the MANEUVER trial took part in interviews after 24 weeks in the trial. These interviews explored how patients' pain and stiffness changed after taking pimicotinib, and what level of improvement they considered meaningful**
- **During interviews, all patients said that they experienced pain or stiffness in their joint affected by TGCT before starting the trial. After 24 weeks, most patients treated with pimicotinib experienced improvements in their symptoms of pain and stiffness, as reported during the trial and at exit interview[1]**
- **Patients scored the level of their worst pain and worst stiffness between 0–10 (0=no pain or stiffness, 10=worst pain or stiffness imaginable). During interviews, most patients said that a 1- or 2-point improvement would be a meaningful change in their condition**

INTRODUCTION

- TGCT is a rare and locally aggressive soft-tissue tumor that affects the synovium, bursae, and tendon sheaths, and is associated with significant morbidity and reduced quality of life[2–4]
- TGCT can cause joint destruction, pain, stiffness, and reduced physical function in an otherwise healthy adult population. Inadequate treatment can cause irreversible joint and bone damage[2–4]
- Pain and stiffness are among the most common symptoms of TGCT, and can significantly impact a person's ability to work, perform daily activities, and engage in social life[2,3]
- Current treatment options for TGCT include surgery (standard of care), systemic treatments, and active surveillance; however, surgery is limited by high recurrence rates and cumulative morbidity. Systemic therapies have emerged as an important treatment option; however, their long-term use requires consideration of both clinical benefit and tolerability over time[3–5]
- Pimicotinib is an oral, once-daily, potent, and highly selective small molecule colony-stimulating factor-1 receptor inhibitor[6]
- In the Phase 3 MANEUVER trial (NCT05804045), pimicotinib demonstrated early, robust, and durable tumor responses that significantly reduced tumor burden and improved TGCT-associated clinical outcome assessments (including range of motion, Worst Pain NRS, Worst Stiffness NRS, and Patient-Reported Outcomes Measurement Information System-Physical Function), with a tolerable and manageable safety profile[1,6]
- Worst Pain and Worst Stiffness NRS (0–10 scale; higher scores indicating worse pain or stiffness) are single-item measures designed to assess patient-reported pain and stiffness severity[7]
- Exit interviews explored patient experiences of pain and stiffness during the MANEUVER trial and assessed the content validity of Worst Pain and Worst Stiffness NRS as PRO instruments

OBJECTIVES

- To characterize patient-perceived changes in pain and stiffness among a subgroup of patients enrolled in the global Phase 3 MANEUVER trial
- To gather patient input to support the establishment of clinically meaningful thresholds for Worst Pain and Worst Stiffness NRS scores

METHODS

- Exit interviews were conducted as part of the MANEUVER trial among patients who completed the 24-week, double-blind, placebo-controlled phase (Part 1) (**Figure 1**)
- Interviews were conducted across 6 countries: Canada, China, Italy, Spain, The Netherlands, and the USA
- Saturation of concepts was assessed on symptoms and impacts experienced at baseline and was reached for all symptom and impact categories

Figure 1. Study design

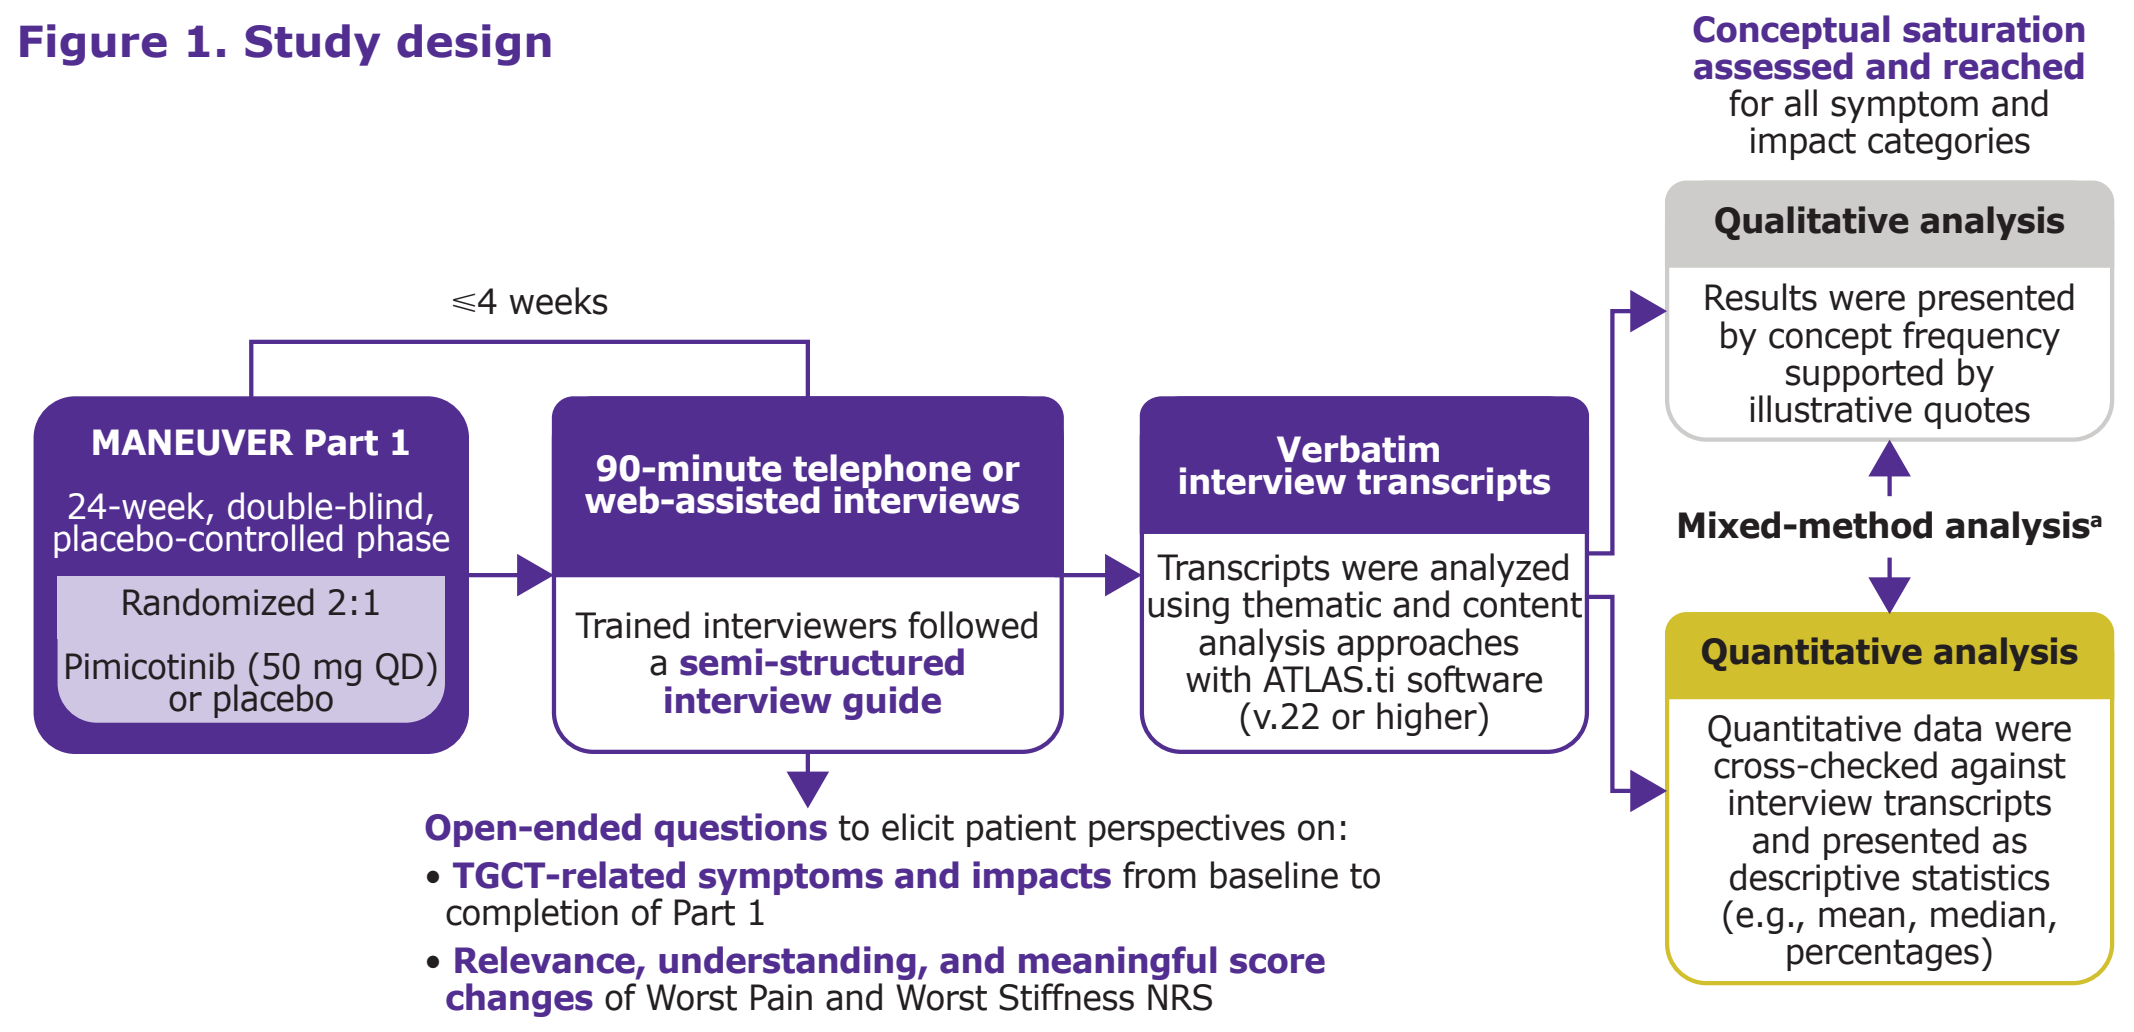
≤4 weeks
MANEUVER Part 1
24-week, double-blind, placebo-controlled phase
Randomized 2:1
Pimicotinib (50 mg QD) or placebo
90-minute telephone or web-assisted interviews
Trained interviewers followed a semi-structured interview guide
Verbatim interview transcripts
Transcripts were analyzed using thematic and content analysis approaches with ATLAS.ti software (v.22 or higher)
Conceptual saturation assessed and reached for all symptom and impact categories
Qualitative analysis
Results were presented by concept frequency supported by illustrative quotes
Mixed-method analysis[a]
Quantitative analysis
Quantitative data were cross-checked against interview transcripts and presented as descriptive statistics (e.g., mean, median, percentages)
Open-ended questions to elicit patient perspectives on:
• TGCT-related symptoms and impacts from baseline to completion of Part 1
• Relevance, understanding, and meaningful score changes of Worst Pain and Worst Stiffness NRS

[a]Some qualitative data were triangulated with quantitative data to support interpretation of findings by gaining a better understanding of what constituted a meaningful change and meaningful treatment benefit based on the PRO
QD, once daily

RESULTS

DEMOGRAPHICS AND CLINICAL CHARACTERISTICS

- In total, 20 patients were interviewed; 12 patients from the pimicotinib treatment group and 8 from the placebo group (**Table 1**)
- Most patients had a lower extremity (95.0%; n=19/20) affected by TGCT, and the knee was most commonly affected (68.4%; n=13/19)

Table 1. Demographics and clinical characteristics

	Total sample (N=20)
Age (years)	
Mean (SD)	43 (13.5)
Sex, n (%)	
Female	13 (65.0)
Male	7 (35.0)
Country, n (%)	
USA	5 (25.0)
The Netherlands	5 (25.0)
Italy	4 (20.0)
China	3 (15.0)
Canada	2 (10.0)
Spain	1 (5.0)
Joint affected by TGCT, n (%)	
Lower extremity	19 (95.0)
Upper extremity	1 (5.0)

SD, standard deviation

EXPERIENCE OF PAIN AND STIFFNESS IN PATIENTS WITH TGCT AT BASELINE AND AT THE END OF PART 1 OF MANEUVER

- PROs of pain and stiffness for all patients who completed Part 1 of the MANEUVER trial have previously been published[1]
- At baseline, 95.0% (n=19/20) of patients experienced pain and 90.0% (n=18/20) experienced stiffness/tightness of the affected joint
- At the end of Part 1, 68.4% (n=13/19) of patients perceived an improvement in pain and 76.5% (n=13/17) of patients perceived an improvement in stiffness in their affected joint compared with baseline
- There was a reduction in Worst Pain and Worst Stiffness NRS scores of −2.3 and −2.6, respectively. A greater reduction in Worst Pain and Worst Stiffness NRS scores was reported by the pimicotinib treatment group compared with the placebo group (**Figure 2**)
 - *"Since I started medication, the bad pains did not come back. So, I did no longer have to think where I woke up in the middle of night, at least. Like the bad surge in pain. It's only the constant pain that was still there. And the constant pain got less* […]*"*

WORST PAIN NRS

- Of patients who responded to questions about Worst Pain NRS, this measure was understood by 12/14 patients (85.7%) and was deemed relevant by 15/19 patients (78.9%) (**Figure 3**)
- Of patients responding to meaningful improvement thresholds for Worst Pain NRS, most found a 1- (42.1%; n=8/19) or 2-point (36.8%; n=7/19) improvement meaningful (**Table 2**)
- Of patients reporting on meaningful worsening, 10/14 patients (71.4%) found a 1-point worsening in Worst Pain NRS meaningful

WORST STIFFNESS NRS

- Worst Stiffness NRS was understood by 16/16 patients (100%) and was relevant to 15/18 patients (83.3%) who provided responses to these measures (**Figure 3**)
- Of patients responding to meaningful improvement thresholds for Worst Stiffness NRS, most found a 1- (31.6%; n=6/19) or 2-point (47.4%; n=9/19) improvement meaningful (**Table 3**)
- Of patients reporting on meaningful worsening for Worst Stiffness NRS, most (53.8%; n=7/13) found a 1-point worsening meaningful

STUDY LIMITATIONS

- Due to time constraints, some patients were not specifically asked or probed about every item, resulting in missing responses within the total sample
- Only one patient had an upper extremity affected by TGCT, thus limiting the generalizability of interpretations for this specific patient population
- Although concept saturation was achieved for both symptoms and impacts across the total sample, the small sample size may limit the generalizability to the broader TGCT population
- Patient-reported TGCT symptoms and impacts at baseline relied on retrospective recall during interviews and so were subject to recall and memory biases

Figure 2. Worst Pain and Worst Stiffness NRS rating at baseline and at the end of Part 1 of MANEUVER reported retrospectively in exit interviews

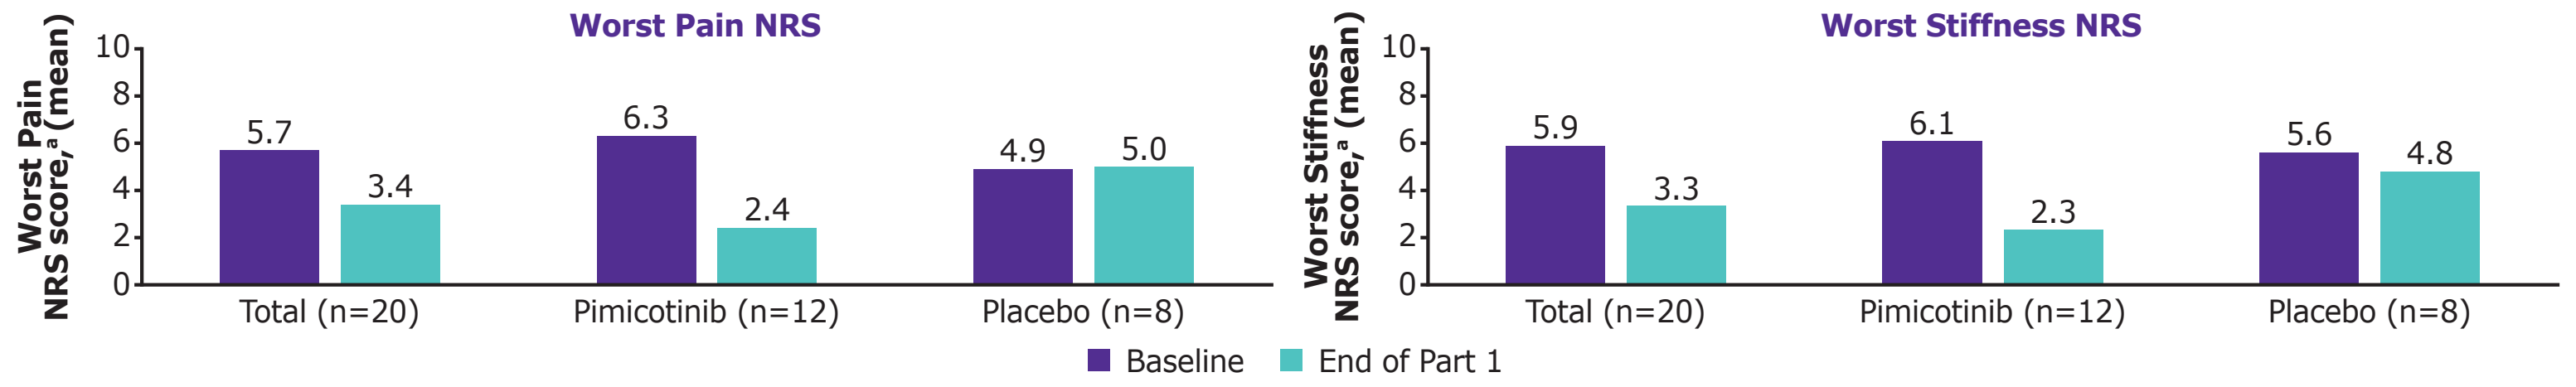
Worst Pain NRS
Worst Pain NRS score,[a] (mean)
10
8
6
4
2
0
5.7
3.4
Total (n=20)
6.3
2.4
Pimicotinib (n=12)
4.9
5.0
Placebo (n=8)
Worst Stiffness NRS
Worst Stiffness NRS score,[a] (mean)
5.9
3.3
Total (n=20)
6.1
2.3
Pimicotinib (n=12)
5.6
4.8
Placebo (n=8)
Baseline
End of Part 1

[a]Rating on a 0–10 scale. Interview guide question to establish Worst Pain and Worst Stiffness NRS: "Please rate your pain and stiffness by circling the one number that best describes your pain or stiffness at its worst in the last 24 hours". The response scale ranges from 0=no pain or stiffness to 10=worst pain or stiffness imaginable; PRO scores were derived from patients' responses provided during interviews. As these responses relied on patient recall, there is a potential for recall bias

Figure 3. Understanding and relevance[a] of Worst Pain and Worst Stiffness NRS[b] for patients with TGCT

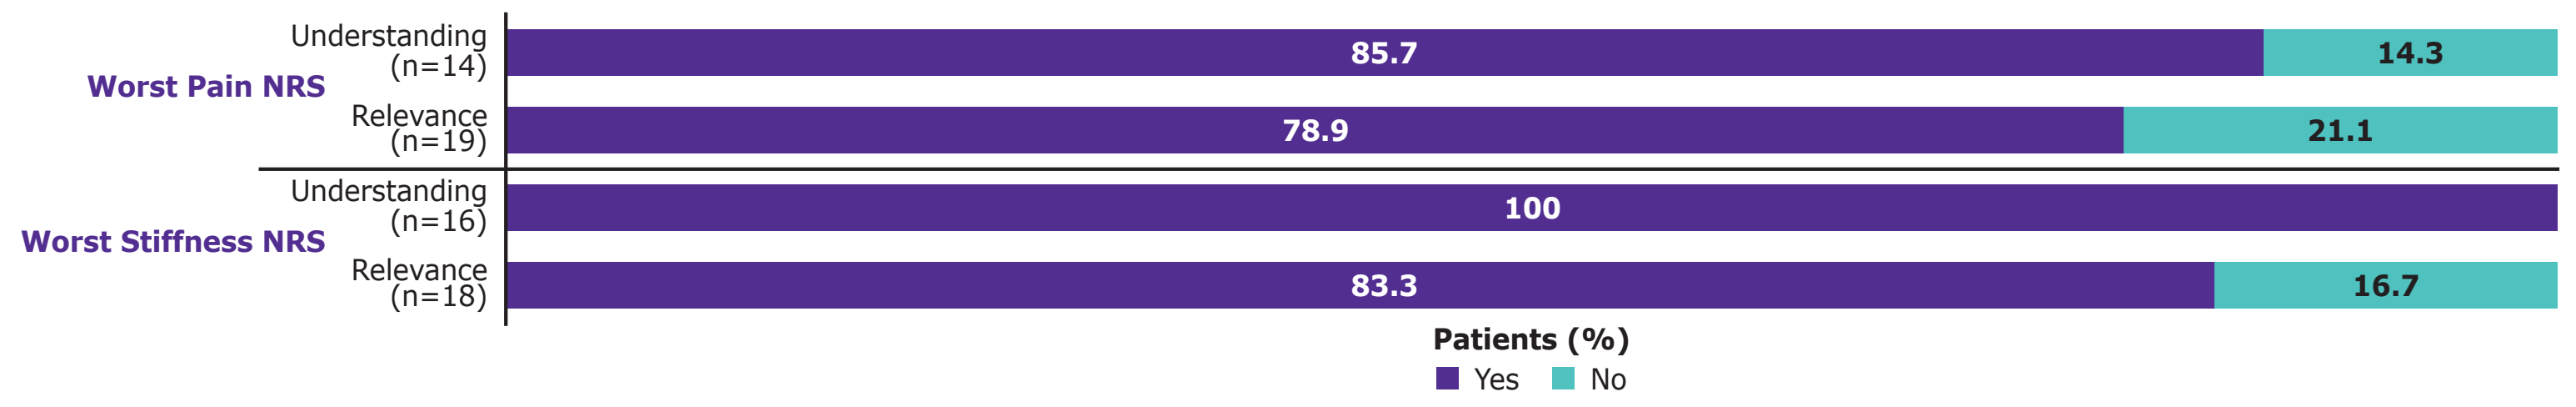
Worst Pain NRS
Understanding (n=14)
85.7
14.3
Relevance (n=19)
78.9
21.1
Worst Stiffness NRS
Understanding (n=16)
100
Relevance (n=18)
83.3
16.7
Patients (%)
Yes
No

[a]Interview guide question to establish understanding and relevance: "Is this question relevant to you?" and "Is the item easy to understand and interpret?"; [b]Interview guide question to establish Worst Pain and Worst Stiffness NRS: "Please rate your pain and stiffness by circling the one number that best describes your pain or stiffness at its worst in the last 24 hours". The response scale ranges from 0=no pain or stiffness to 10=worst pain or stiffness imaginable. Some patients were not specifically asked or probed about some items due to the natural flow of the interview and/or time constraints. Data are reported for the patients who answered questions on these items during exit interview; PRO scores were derived from patients' responses provided during interviews. As these responses relied on patient recall, there is a potential for recall bias

Table 2. Level of meaningful improvement in Worst Pain NRS scores in patients with TGCT

Level of improvement that would be meaningful	Total n (%)	Illustrative patient quotes
Worst Pain NRS		
Meaningful improvement	19/20 (95.0%)	
1-point improvement	8/19 (42.1%)	*"Yes. It's little, it's 1 point, but it's always positive."* – Patient with TGCT in the knee
2-point improvement	7/19 (36.8%)	Interviewer: *"How about a change from a 6 to a 4?"* *"You would start to notice that it's less. And I think that would be meaningful."* – Patient with TGCT in the knee
3-point improvement	1/19 (5.3%)	*"Yes, it would be a little bit more meaningful because it would signify that the improvement is better."* – Patient with TGCT in the foot
4-point improvement	2/19 (10.5%)	Interviewer: *"Okay. And then how about a 10 to a 6? Would that be meaningful to you?"* *"Yes. Again, more mobility. It's clear that something's improving, you know. Gives a bit more hope."* – Patient with TGCT in the knee
No change	1/19 (5.3%)	Interviewer: *"What I mean is the rating is 5 now, currently, and to which point it changed would make you feel satisfied, or you are satisfied now?"* *"I am satisfied. Yes."* – Patient with TGCT in the knee

Interview guide question: "From your score [share score pre-trial] before you started the study, what change would be meaningful or important to you in terms of improvement, resulting in a meaningful treatment benefit from your point of view? In other words, what number on the scale would you move to feel that is a noticeable and meaningful improvement to your worst pain?"

Table 3. Level of meaningful improvement in Worst Stiffness NRS scores in patients with TGCT

Level of improvement that would be meaningful	Total n (%)	Illustrative patient quotes
Worst Stiffness NRS		
Meaningful improvement	19/20 (95.0%)	
1-point improvement	6/19 (31.6%)	*"That's also meaningful…because it means that I could do more things, I could do something more. I could do more movements. I could manage to do something more, something that at 7 I couldn't do. It might be even just a simple action as putting on my shoes on my own."* – Patient with TGCT in the knee
2-point improvement	9/19 (47.4%)	*"Maybe a bit…I mean, if the swelling and stiffness went down. Usually, that's more noticeable, I would say."* – Patient with TGCT in the knee
3-point improvement	2/19 (10.5%)	*"I guess it at least should be 3 points of change. For example, from 8 to 5."* – Patient with TGCT in the knee
4-point improvement	1/19 (5.3%)	Interviewer: *"And if we went from a 5 to a 1?"* *"That would be the best."* – Patient with TGCT in the knee
No change	1/19 (5.3%)	*"It stayed the same."* – Patient with TGCT in the knee

Interview guide question: "From your score [share score pre-trial] before you started the study, what change would be meaningful or important to you in terms of improvement, resulting in a meaningful treatment benefit from your point of view? In other words, what number on the scale would you move to feel that is a noticeable and meaningful improvement to your worst stiffness?"

REFERENCES

1. Xu H et al. Lancet 2026;407:1072–83
2. Bernthal NM et al. Orphanet J Rare Dis 2021;16:191
3. Stern S et al. Future Oncol 2025;21:1501–10
4. Stacchiotti S et al. Cancer Treat Rev 2023;112:102491
5. Spierenburg G et al. Expert Opin Ther Targets 2022;26:333–45
6. Niu X et al. Future Oncol 2026;22:507–14
7. Gelhorn HL et al. Clin Ther 2016;38:778–93

ACKNOWLEDGMENTS

Medical writing support, under the direction of the authors, was provided by Rebecca Mobley, PhD, of the Publications Division of Omnicom Health Medical Communications, funded by the Healthcare business of Merck KGaA, Darmstadt, Germany in accordance with Good Publication Practice (GPP 2022) guidelines

Abbisko Therapeutics designed and funded this study and collected and analyzed the primary data. In April 2025, the Healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID:10.13039/100009945) gained exclusive rights to commercialize pimicotinib worldwide

AUTHOR DISCLOSURES

Vinod Ravi has received consulting fees and honoraria from EMD Serono. Paul Kamudoni, Andrea Phillips Beyer, and Niki Karachaliou are employees of the Healthcare business of Merck KGaA, Darmstadt, Germany and its affiliates. Amy Clark, Agkreta Leventi, Savita Bakhshi Anand, and Carla Dias-Barbosa are employees of PPD Evidera Patient-Centered Research, Thermo Fisher Scientific, which was paid by the Healthcare business of Merck KGaA, Darmstadt, Germany, to conduct the study. Hans Gelderblom was a site Principal Investigator for Abbisko Therapeutics (fees paid to institution)

GET POSTER PDF

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Poster code PCR18. Presented at The Professional Society for Health Economics and Outcomes Research (ISPOR) 2026 Congress, 17–20 May 2026, Philadelphia, PA, USA