Burden of Symptoms in Transthyretin Amyloid Cardiomyopathy: A United States Physician Survey Describing Gender Perspectives

HSD16

Janice Chung[1], Ann Willet[1], Jade Garratt-Wheeldon[2], Cynthia Khanji[1], Feng Dai[1], Alisha Braid[2], Jack Wright[2]

[1]Pfizer Inc, New York, NY, USA; [2]Adelphi Real World, Bollington, UK

INTRODUCTION

- Transthyretin amyloid cardiomyopathy (ATTR-CM) is a progressive and fatal disease caused by the deposition of misfolded transthyretin (TTR) amyloid fibrils in the myocardium.[1,2]
- Diagnostic delay and misdiagnosis of ATTR-CM are common partly due to clinical symptoms overlapping with other common diseases.[3]
 - Early diagnosis of ATTR-CM and timely initiation of disease-modifying treatment improve clinical outcomes.[2,3]
- Despite the fact that symptoms of ATTR-CM, including cardiovascular, neurologic, musculoskeletal, and gastrointestinal manifestations, place a substantial burden on patients with ATTR-CM and their caregivers, real-world data describing the most burdensome symptoms are limited.[4,5]

OBJECTIVE

- To identify the most burdensome symptoms of ATTR-CM from the perspectives of physicians in the United States using real-world data.

METHODS

- Secondary analyses were conducted using data from the Adelphi Real World ATTR Disease Specific Programme™ (DSP), a cross-sectional survey of physicians and patients with transthyretin amyloidosis (ATTR) in the United States between August 2024 and February 2025.
 - The DSP methodology has been previously published and validated.[6-8]
- Minimal inclusion criteria governed DSP data collection.
 - Eligible physicians included cardiologists and primary care physicians who had treated ≥2 patients with ATTR-CM at the time of the survey.
- Physicians completed a one-time online survey and electronic record forms for up to 10 consecutively consulting patients with ATTR-CM.
 - Physicians reported patient demographics, clinical characteristics, and symptom burden.
 - For each individual patient, physicians identified the five most burdensome symptoms.
 - Physician-reported burdensome symptoms were subsequently aggregated across all patients to identify the most burdensome symptoms overall and by sex.

RESULTS

Physician Clinical Practice Characteristics

- Overall, 34 physicians participated in the survey and provided data for 256 patients with ATTR-CM (**Table 1**).
 - Primary care physicians (44.1%) and general cardiologists (41.2%) comprised the majority of physicians.
- Most physicians (76.5%) had treated patients with ATTR for ≥5 years and 44.1% had been qualified for 15–25 years.
 - Across physicians' overall ATTR caseloads, a mean of 71.3% of patients were seen in an office setting.
- In the 12 months prior to the survey, physicians reported seeing a mean (SD) of 39.1 (47.0) patients with ATTR-CM.
- At the time of the survey, each physician had managed a mean (SD) of 44.8 (55.5) patients with ATTR-CM.

Figure: Physician-reported burdensome symptoms for patients with ATTR-CM

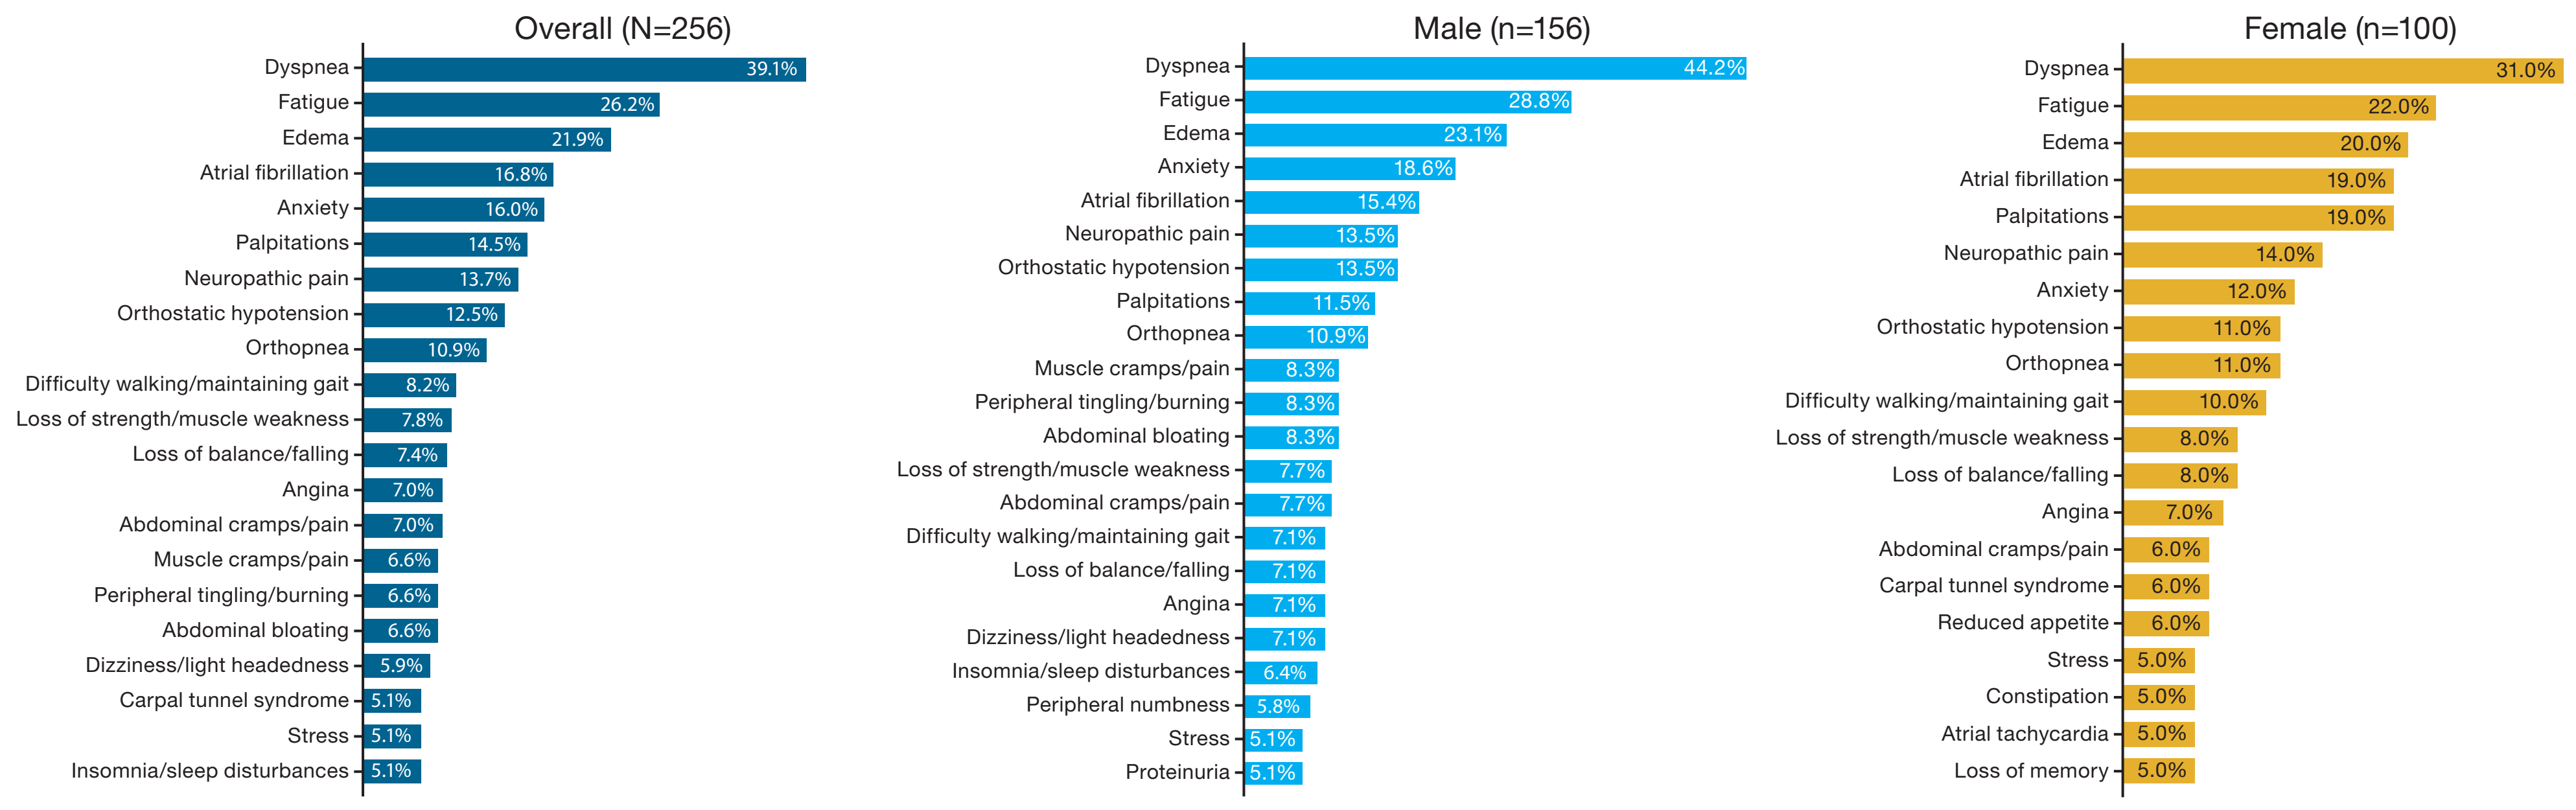
Overall (N=256)
Dyspnea 39.1%
Fatigue 26.2%
Edema 21.9%
Atrial fibrillation 16.8%
Anxiety 16.0%
Palpitations 14.5%
Neuropathic pain 13.7%
Orthostatic hypotension 12.5%
Orthopnea 10.9%
Difficulty walking/maintaining gait 8.2%
Loss of strength/muscle weakness 7.8%
Loss of balance/falling 7.4%
Angina 7.0%
Abdominal cramps/pain 7.0%
Muscle cramps/pain 6.6%
Peripheral tingling/burning 6.6%
Abdominal bloating 6.6%
Dizziness/light headedness 5.9%
Carpal tunnel syndrome 5.1%
Stress 5.1%
Insomnia/sleep disturbances 5.1%
Male (n=156)
Dyspnea 44.2%
Fatigue 28.8%
Edema 23.1%
Anxiety 18.6%
Atrial fibrillation 15.4%
Neuropathic pain 13.5%
Orthostatic hypotension 13.5%
Palpitations 11.5%
Orthopnea 10.9%
Muscle cramps/pain 8.3%
Peripheral tingling/burning 8.3%
Abdominal bloating 8.3%
Loss of strength/muscle weakness 7.7%
Abdominal cramps/pain 7.7%
Difficulty walking/maintaining gait 7.1%
Loss of balance/falling 7.1%
Angina 7.1%
Dizziness/light headedness 7.1%
Insomnia/sleep disturbances 6.4%
Peripheral numbness 5.8%
Stress 5.1%
Proteinuria 5.1%
Female (n=100)
Dyspnea 31.0%
Fatigue 22.0%
Edema 20.0%
Atrial fibrillation 19.0%
Palpitations 19.0%
Neuropathic pain 14.0%
Anxiety 12.0%
Orthostatic hypotension 11.0%
Orthopnea 11.0%
Difficulty walking/maintaining gait 10.0%
Loss of strength/muscle weakness 8.0%
Loss of balance/falling 8.0%
Angina 7.0%
Abdominal cramps/pain 6.0%
Carpal tunnel syndrome 6.0%
Reduced appetite 6.0%
Stress 5.0%
Constipation 5.0%
Atrial tachycardia 5.0%
Loss of memory 5.0%

Burdensome symptoms reported in ≥5% of patients overall and by sex are shown.
ATTR-CM=transthyretin amyloid cardiomyopathy

Table 1: Physician clinical practice characteristics

	Physicians n=34
Length of time physicians had been treating patients with ATTR, n (%)	
1–2 years	1 (2.9)
2–3 years	2 (5.9)
3–4 years	5 (14.7)
≥5 years	26 (76.5)
Physician specialty, n (%)	
Primary care physician	15 (44.1)
General cardiologist	14 (41.2)
Heart failure specialist	5 (14.7)
Physician care setting, mean (SD), %[a]	
Office	71.3 (34.0)
Academic hospital	21.6 (33.4)
Community hospital	7.1 (14.0)
Length of time physician had been qualified, n (%)	
≤5 years	2 (5.9)
5–15 years	11 (32.4)
15–25 years	15 (44.1)
25–35 years	3 (8.8)
>35 years	3 (8.8)
Number of patients with ATTR-CM seen by physician in the 12 months prior to the survey, mean (SD)	39.1 (47.0)
Number of patients with mixed phenotype seen by physician in the 12 months prior to the survey, mean (SD)[b]	26.5 (32.4)
Percentage of time spent managing patients with ATTR in the 12 months prior to the survey, mean (SD), %[c]	25.3 (30.0)
Number of patients with ATTR-CM managed by physician at the time of the survey, mean (SD), n	44.8 (55.5)
Number of patients with mixed phenotype managed by physician at the time of the survey, mean (SD), n[b]	28.8 (34.3)
Percentage of patients with ATTR seen or treated at a specialist amyloidosis center, mean (SD), %	32.4 (39.5)

[a] Percentage of patients among physicians' overall ATTR caseloads seen in each setting.
[b] Mixed phenotype refers to patients with both ATTR-CM and ATTR-PN.
[c] Physicians were asked to consider consultations, research, and administrative tasks when estimating time spent.
ATTR=transthyretin amyloidosis; ATTR-CM=transthyretin amyloid cardiomyopathy; ATTR-PN=transthyretin amyloid polyneuropathy; SD=standard deviation

Physician-Reported Patient Demographics and Clinical Characteristics

- Among the 256 patients with ATTR-CM, 156 (60.9%) were male and 100 (39.1%) were female; the mean age was 63.9 years (**Table 2**).

Table 2: Physician-reported patient demographics and clinical characteristics

	Overall N=256	Male n=156	Female n=100
Age, mean (SD), years	63.9 (13.2)	65.0 (11.2)	62.2 (15.8)
Ethnicity, n (%)			
White	143 (55.9)[a]	86 (55.1)[a]	57 (57.0)
Black African, African American or Caribbean	81 (31.6)	48 (30.8)	33 (33.0)
South or Central American Native	13 (5.1)	8 (5.1)	5 (5.0)
East or Southeast Asian	9 (3.5)	8 (5.1)	1 (1.0)
Middle Eastern or North African	6 (2.3)	4 (2.6)	2 (2.0)
South Asian (Indian subcontinent)	3 (1.2)	1 (0.6)	2 (2.0)
American Indian, Indigenous American, or Alaska Native	1 (0.4)	1 (0.6)	0
Other	1 (0.4)	1 (0.6)	0
ATTR phenotype, n (%)			
ATTR-CM	204 (79.7)	123 (78.8)	81 (81.0)
Mixed[b]	52 (20.3)	33 (21.2)	19 (19.0)
BMI, mean (SD), kg/m²	25.6 (4.8)	26.4 (5.1)	24.4 (4.0)
NYHA functional class at the time of the survey, n (%)			
I	56 (21.9)	31 (19.9)	25 (25.0)
II	139 (54.3)	87 (55.8)	52 (52.0)
III	51 (19.9)	31 (19.9)	20 (20.0)
IV	10 (3.9)	7 (4.5)	3 (3.0)
Number of diagnosed comorbidities, mean (SD), n	1.9 (1.7)	2.1 (1.7)	1.5 (1.5)
Comorbidities, n (%)[c]	n=202	n=130	n=72
Hypertension	97 (48.0)	72 (55.4)	25 (34.7)
Congestive heart failure	73 (36.1)	50 (38.5)	23 (31.9)
Anxiety	47 (23.3)	31 (23.8)	16 (22.2)
Depression	40 (19.8)	23 (17.7)	17 (23.6)

[a] One patient selected both White and East or Southeast Asian ethnicities.
[b] Mixed phenotype refers to patients with both ATTR-CM and ATTR-PN.
[c] Comorbidities reported in ≥20% of patients overall or in male and female subgroups are presented.
ATTR=transthyretin amyloidosis; ATTR-CM=transthyretin amyloid cardiomyopathy; ATTR-PN=transthyretin amyloid polyneuropathy; BMI=body mass index; NYHA=New York Heart Association; SD=standard deviation

- Most patients were White (55.9% overall: 55.1% male, 57.0% female) and were classified as New York Heart Association class II (54.3% overall: 55.8% male, 52.0% female) at the time of the survey.
- The two most commonly reported comorbidities in patients with ATTR-CM were hypertension (48.0% overall: 55.4% male, 34.7% female) and congestive heart failure (36.1% overall: 38.5% male, 31.9% female).

Physician-Reported Most Burdensome Symptoms for Patients With ATTR-CM

- Across all patients, the three most burdensome symptoms were dyspnea (39.1% overall: 44.2% male, 31.0% female), fatigue (26.2% overall: 28.8% male, 22.0% female), and edema (21.9% overall: 23.1% male, 20.0% female) (**Figure**).
 - Atrial fibrillation ranked among the top five burdensome symptoms overall (16.8%) and in male (15.4%) and female (19.0%) patients.
 - Anxiety was among the five most burdensome symptoms overall (16.0%) and in male patients (18.6%), whereas palpitations ranked among the five most burdensome symptoms in female patients (19.0%).

LIMITATIONS

- Physicians from the ATTR DSP constitute a pragmatic, rather than a true random sample. Physician participation may have been influenced by their willingness to complete the record forms.

CONCLUSIONS

- The most burdensome symptoms for patients with ATTR-CM, as reported by US physicians, were similar between male and female patients.
- The symptoms identified as most burdensome are commonly observed in patients with cardiovascular diseases.
- Effective treatments for ATTR-CM should address both these key symptoms and the underlying causes of disease.

REFERENCES

1. Ruberg FL, et al. J Am Coll Cardiol 2019;73:2872-91. **2.** Porcari A, et al. Cardiovasc Res 2023;118:3517-35. **3.** Rozenbaum MH, et al. Cardiol Ther 2021;10:141-59. **4.** Ponti L, et al. Front Cardiovasc Med 2023;10:1238843. **5.** Cappelli F, et al. Front Cardiovasc Med 2025;12:1595797. **6.** Babineaux SM, et al. BMJ Open 2016;6:e010352. **7.** Anderson P, et al. Curr Med Res Opin 2008;24:3063-72. **8.** Anderson P, et al. Curr Med Res Opin 2023;39:1707-15.

DISCLOSURES

This study used data from the Adelphi Real World ATTR DSP, an independently conducted survey. The DSP is a wholly owned Adelphi Real World product. Pfizer was one of multiple subscribers to the DSP and funded the analysis described here.
JG-W, AB, JW: Employees of Adelphi Real World, which was a paid contractor to Pfizer in connection with this study and the development of this poster. **JC, AW, CK, FD:** Employees of and hold stock/options in Pfizer.

ACKNOWLEDGMENTS

Medical writing support was provided by April Cabang of Engage Scientific Solutions and was funded by Pfizer.

Electronic Poster
Copies of this poster, which can be obtained through this QR code, are for your personal use only and may not be reproduced without permission from the authors. Scan to download a copy of this poster.
If you don't have a smartphone, access the poster via the internet at:
https://scientificpubs.congressposter.com/p/n9l5s1e3qxuzcuyp
Copyright © 2026. All rights reserved.

Presented at the 2026 International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Conference, May 17–20, 2026, Philadelphia, PA, USA