Poster 2- C079

Clinical and Humanistic Burden of Adult Patients With Neurofibromatosis Type 1 With Plexiform Neurofibroma in the Real-World Setting Across Four European Countries: Results From a Real-World Survey

Blanco I[1], Moreno N[2], Male N[2], Gatta F[3], Baldock D[4], Adeyemi A[5]

[1]*Clinical Genetics Department, Germans Trias i Pujol University Hospital, Barcelona, Spain;* [2]*Alexion, AstraZeneca Rare Disease, Barcelona, Spain;* [3]*Alexion, AstraZeneca Rare Disease, Zurich, Switzerland;* [4]*Adelphi Real World, Bollington, UK;* [5]*Alexion, AstraZeneca Rare Disease, Boston, MA, USA;*

OBJECTIVES

- To describe the clinical and humanistic burden of the NF1-PN disease in adult patients in Germany, Italy, Spain, and the UK.

CONCLUSIONS

- Adults with NF1-PN in selected European countries experienced a high burden of disease that extends well beyond clinical symptoms, affecting daily functioning and work.
- Over a third of the patients report functional impairment, often affecting core activities of their daily life.
- Half of the patients experienced pain as a symptom, associated with a persistent symptomatic burden.
- Half of patients experienced an impact on employment, with a third reducing work hours, contributing to economic burden through lost productivity.
- These findings highlight the humanistic, clinical and economic burden for adult patients with NF1-PN , underscoring the importance of comprehensive care and an optimal treatment management of these patients.

PLAIN LANGUAGE SUMMARY

Why did we perform this research?
Neurofibromatosis type 1 (NF1) is a rare condition that can be inherited or occur spontaneously due to mutations in the NF1 tumour suppression gene. Plexiform neurofibromas (PN) can form in a small group of patients and often presents with burdensome symptoms such as pain and mobility issues. Although NF1-PN typically develops in childhood, PN can continue to grow into adulthood. This study aimed to describe the impact of NF1-PN on adult patients in selected European countries.

How did we perform this research?
Data were drawn from the Adelphi Real World Disease Specific Programme™, in which doctors completed electronic record forms after consulting their adult patients with NF1-PN in Germany, Italy, Spain, and the UK. Surveys were completed between June-November 2025 and doctors provided information on patients' demographics, clinical characteristics, and healthcare resource utilisation.

What were the findings of this research and what are the implications?
Most patients experienced some level of burden due to their NF1-PN, with more than half experiencing pain, one-third having impairment to activities of their daily life, and more than half requiring surgical intervention for PN. These findings highlight the impact of NF1-PN on patients' quality of life and the need for effective treatment and care.

Where can I access more information?
For more information on the DSP, please contact Denise Baldock (Denise.Baldock@OMC.com)

Poster

Plain language summary

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link below.
https://rarediseaseresource.com/2026/ISPOR05-17/ADELPHI_EU_Adult_BoD_poster

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Poster presented at ISPOR 2026, Philadelphia, PA, USA, May 17-20, 2026
Corresponding author email address Ayo.Adeyemi@Alexion.com

INTRODUCTION

- Neurofibromatosis type 1 (NF1) is a rare condition, that can be inherited or occur spontaneously owing to a mutation in the *NF1* tumour suppression gene[1].
- Plexiform neurofibromas (PN) can form in a subset of patients with NF1 and are often associated with burdensome symptoms such as pain and impact to motor function[2].
- NF1-PN typically develops during childhood, although PN can continue to grow into adulthood[3].
- With a scarcity of real-world evidence to quantify the unmet needs among the adult NF1-PN population, it is important to assess the impact the disease may have on adult patients.

METHODS

- Data were drawn from the Adelphi Real World Disease Specific Programme™, a cross-sectional survey with retrospective data collection of physicians and their adult patients diagnosed with NF1-PN in Germany, Italy, Spain, and the UK between June-November 2025.
- The DSP methodology has been previously described[4,5], validated[6], and proven consistent over time[7].
- Participating physicians reported data for their next consecutive consulting adult patients with NF1-PN.
- Physicians completed electronic record forms capturing patient demographics, clinical characteristics (including ECOG score), perceived severity, and PN specifics (including surgical interventions).
- Descriptive analyses were conducted for the overall cohort, and by country. Frequencies (%) were calculated for categorical or ordinal variables and mean (standard deviation [SD]) or medians (interquartile ranges [IQR]) were calculated for continuous variables.
- All data were reported by physicians

RESULTS AND INTERPRETATION

Demographics

Physician and Patient overview

- This study included 74 physicians who provided data for 330 patients (Patient distribution: Germany, 29%; Italy, 31%, Spain, 24% and the UK, 16%).
- Physicians were mostly neurologists (34%), dermatologists (31%), and oncologists (14%).
- Less than half (44%) of patients had familial NF1, mean (SD) age was 35.0 (13.7) years, 56% were male, and 95% were white.
- In total, 45% of patients were employed, of whom physicians reported 50% had work that was unaffected by NF1-PN. Among those whose work was affected, the most common impact was a reduction in work hours (30%).
- Among employed patients, productivity loss was evident: mean work absence was 1.7 days per month overall, with country means ranging from about 1.1 days in Italy to 2.1 days in Germany.
- Approximately one-third of patients experienced impairment in basic activities of daily living and over half experienced impairment in instrumental activities of daily living.
- Patient demographics and clinical characteristics can be seen split by country in **Table 1**.

NF1-PN Severity

- At survey date, physicians reported that 65% of patients had scores of 1-4 in the Eastern Cooperative Oncology Group (ECOG) scale, meaning almost 2/3 of the sample experienced some level of restriction.
- Physicians perceived NF1-PN to be moderate-very severe in 37% of cases as seen in Table 1.

Table 1. Characteristics of NF1-PN patients

	Total (n=330)	Germany (n=95)	Italy (n=101)	Spain (n=80)	UK (n=54)
Age (years), mean (SD)	35.0 (13.7)	34.2 (11.6)	34.3 (13.8)	40.0 (16.3)	30.2 (10.2)
Sex, Male; n (%)	184 (56%)	51 (54%)	54 (53%)	47 (59%)	32 (59%)
Racial Identity, n (%)					
White	313 (95%)	93 (98%)	100 (99%)	78 (98%)	42 (78%)
Other	18 (5%)	2 (2%)	1 (1%)	2 (3%)	13 (24%)
Time since diagnosis (years), median (IQR)	13.0 (4.0-23.0)	13.0 (6.0-21.0)	8.0 (2.0-18.5)	14.0 (8.0-26.8)	16.0 (1.8-23.5)
Employment status, n (%)	(n=325)	(n=95)	(n=99)	(n=78)	(n=53)
Employed	**146 (45%)**	**57 (60%)**	**43 (43%)**	**27 (35%)**	**19 (36%)**
Student	69 (21%)	13 (14%)	34 (34%)	12 (15%)	10 (19%)
Unemployed	32 (10%)	5 (5%)	9 (9%)	5 (6%)	13 (25%)
Other	78 (24%)	20 (21%)	13 (13%)	34 (44%)	11 (21%)
Days absent from work per month*,	(n=119)	(n=48)	(n=33)	(n=24)	(n=14)
mean (SD)	1.7 (3.0)	2.1 (3.7)	1.1 (1.3)	1.8 (3.5)	1.5 (2.1)
Effect of NF1-PN on employment, n (%)	(n=141)	(n=54)	(n=43)	(n=25)	(n=19)
Work unaffected	71 (50%)	26 (48%)	27 (63%)	12 (48%)	6 (32%)
Reduced hours of work	43 (30%)	22 (41%)	9 (21%)	6 (24%)	6 (32%)
Other	43 (30%)	19 (35%)	10 (23%)	7 (28%)	7 (37%)
Impairment to basic activities of daily living, n (%)	(n=323)	(n=93)	(n=96)	(n=80)	(n=54)
	117 (36%)	38 (41%)	30 (31%)	26 (33%)	23 (43%)
Impairment to instrumental activities of daily living, n (%)	(n=321)	(n=92)	(n=97)	(n=79)	(n=53)
	183 (57%)	53 (58%)	49 (51%)	44 (56%)	37 (70%)
ECOG score distribution at time of survey, n (%)					
0	115 (35%)	32 (34%)	46 (46%)	25 (31%)	12 (22%)
1	138 (42%)	42 (44%)	35 (35%)	34 (43%)	27 (50%)
2	64 (19%)	15 (16%)	15 (15%)	19 (24%)	15 (28%)
3	11 (3%)	6 (6%)	3 (3%)	2 (3%)	0 (0%)
4	1 (0%)	0 (0%)	1 (1%)	0 (0%)	0 (0%)

*Of patients employed at time of survey; Basic activities of daily living: ambulating, feeding, dressing, personal hygiene, toileting; Instrumental activities of daily living: transportation, managing finances, shopping and meal preparation, housecleaning and home maintenance, managing communication with others, managing medication; ECOG, Eastern Cooperative Oncology Group; IQR, Interquartile Range; N, Sample Size; SD, Standard Deviation

Symptomatology

- Pain (50%) and skin-related disorders (40%) were the most common NF1/NF1-PN-related symptoms as reported by physicians across the study population.
- In Spain, 41% of patients experienced anxiety, compared to 26% in Germany, 19% in Italy, and 33% in the UK. A large proportion of patients experiencing motor dysfunction was reported in Germany (39%).
- A symptom breakdown is shown in **Figure 1.**

Figure 1. Physician Reported Symptoms experienced by NF1-PN patients at time of survey (top 10)

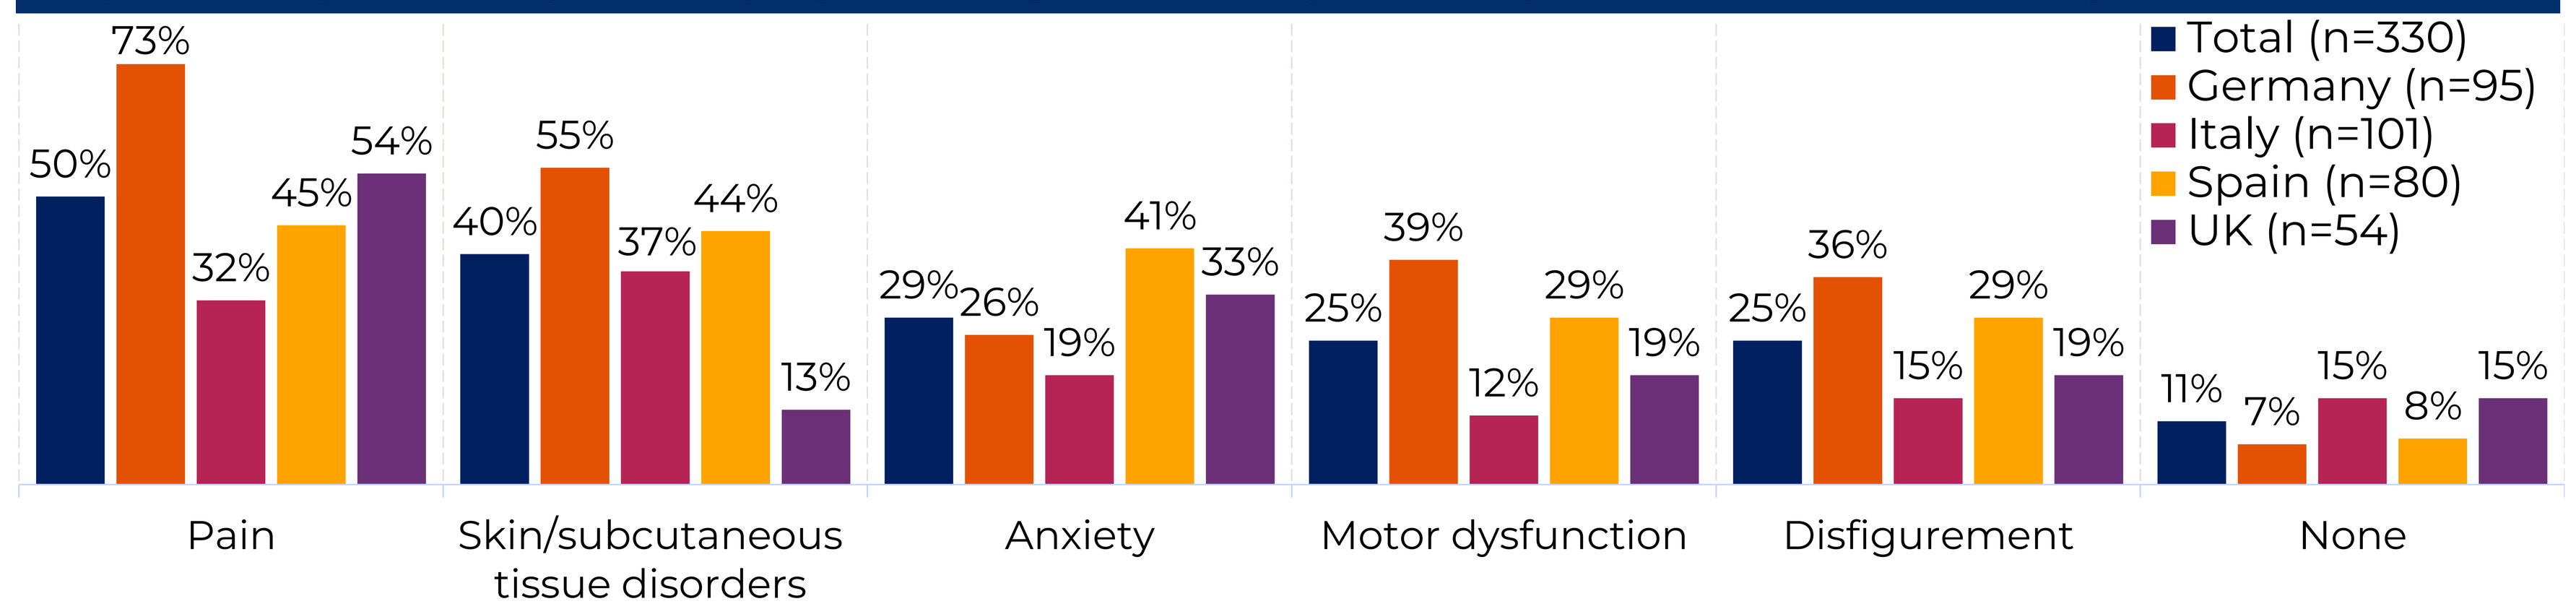
Total (n=330)
Germany (n=95)
Italy (n=101)
Spain (n=80)
UK (n=54)
Pain: 50% 73% 32% 45% 54%
Skin/subcutaneous tissue disorders: 40% 55% 37% 44% 13%
Anxiety: 29% 26% 19% 41% 33%
Motor dysfunction: 25% 39% 12% 29% 19%
Disfigurement: 25% 36% 15% 29% 19%
None: 11% 7% 15% 8% 15%

- Physicians reported that patients most commonly had ≥1 PN in the legs (46%), with a mean (SD) of 3.3 (3.6) PN in the legs for these patients (n=132). Of the patients with ≥1 PN in the spine (24%), the largest PN was internal in 84% of cases compared to 30% being internal for the legs.
- The most clinically relevant PN were also in the legs (26%); reasons for classifying these PN was due to their location (45%), size (41%) and symptoms such as pain (40%).
- Further details on PN locations are summarised in **Figure 2**.

Figure 2. PN profile of NF1-PN patients (n=329)

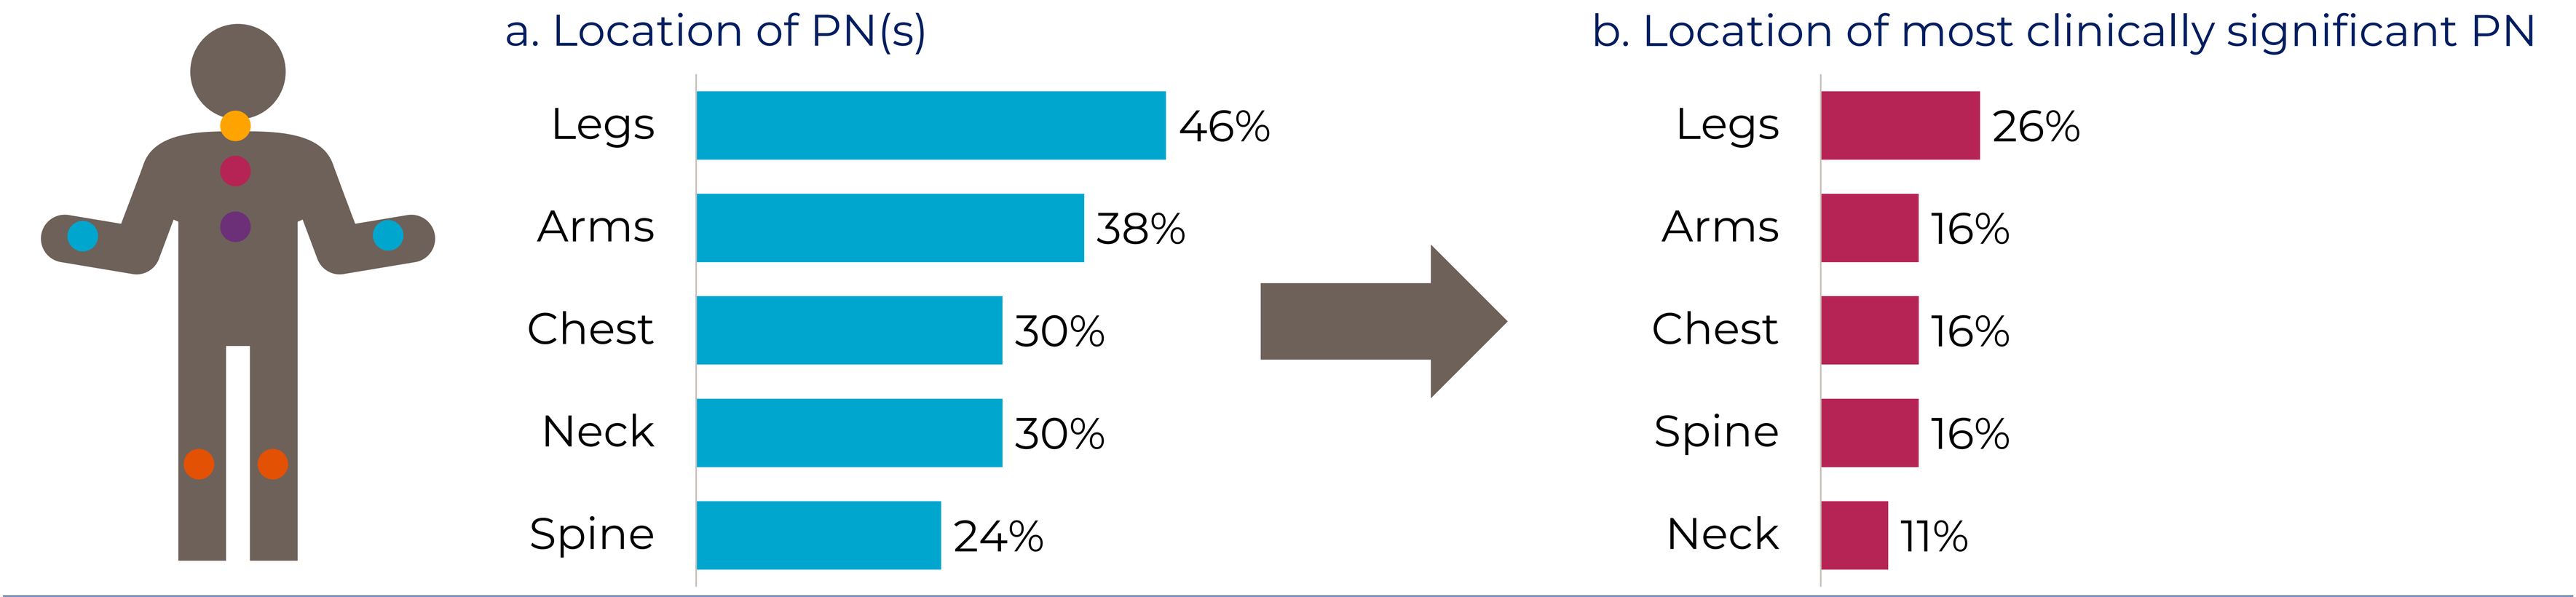
a. Location of PN(s)
Legs 46%
Arms 38%
Chest 30%
Neck 30%
Spine 24%
b. Location of most clinically significant PN
Legs 26%
Arms 16%
Chest 16%
Spine 16%
Neck 11%

Disease Burden

Monitoring

- Various tests and assessments were used to monitor patients in the year prior to data collection. The most common was whole-body MRI, performed in 23% of the study population overall and 41% of the German cohort.

Healthcare Resource Utilisation

- Over half (54%) of patients had undergone NF1-PN-related surgery with a mean (SD) of 2.9 (3.2) surgical interventions. Reasons for NF1-PN related surgery are summarised in **Figure 3**.
- Almost a third (31%) of patients had clinically significant PN, which had not been operated on, most commonly located in the spine (28%), arms (25%), or neck (25%)
- Where reported, 15% of patients experienced ≥1 hospitalization in the year prior to survey; the most common reasons were extreme pain (40%) and treatment of an NF1-PN (27%)

Figure 3. PN-related surgery performed for NF1-PN patients

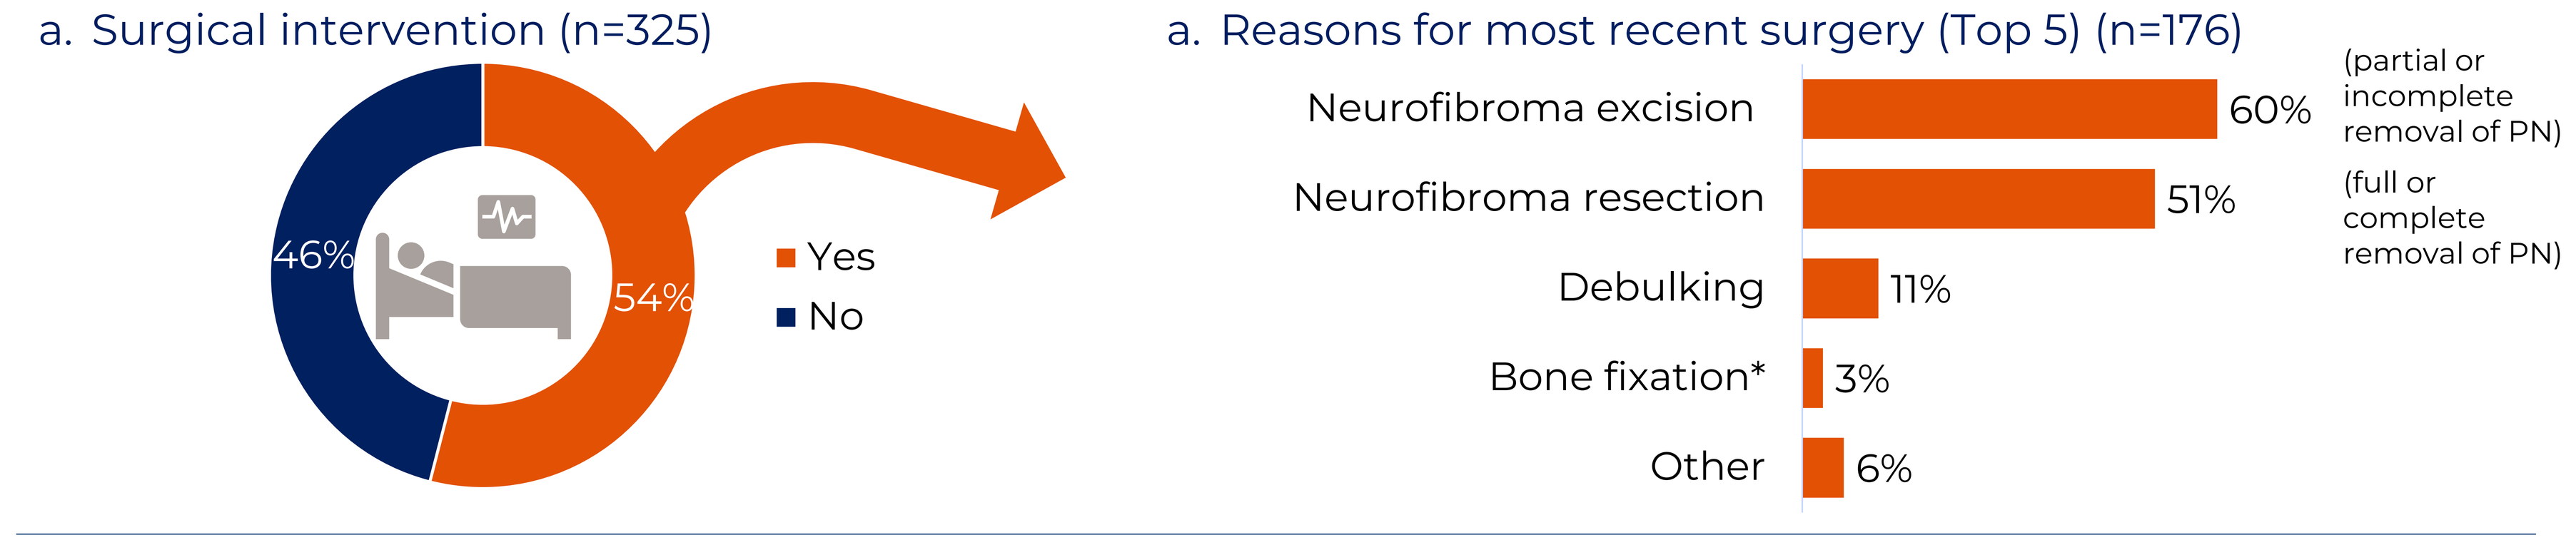
a. Surgical intervention (n=325)
46%
54%
Yes
No
a. Reasons for most recent surgery (Top 5) (n=176)
Neurofibroma excision 60%
(partial or incomplete removal of PN)
Neurofibroma resection 51%
(full or complete removal of PN)
Debulking 11%
Bone fixation* 3%
Other 6%

a. Surgical intervention on PN; b. Reason for most recent surgery on PN; * due to PN.

Limitations

·This real-world study was based on a pragmatic rather than true random sample; in addition to the inclusion criteria, while minimal, physician and patient participation was influenced by willingness to complete the questionnaires; furthermore, several variables related to the impact on daily life, were physician reported, which may not fully capture patients' lived experience.

Funding source

·The NF1-PN Wave I DSP is a wholly owned Adelphi Real World product. Alexion, AstraZeneca Rare Disease and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, was a subscriber to the DSP and funded the analysis described here.

Acknowledgments

Editorial assistance was provided by Brianne Kerr- Adelphi Real World

References

1. Gutmann DH et al., Nat Rev Dis Primers. 2017;3:17004
2. Fisher MJ et al., Neuro-onc. 2022;24(11):1827-1844.
3. Yoo HK et al., BMC neurol. 2023;23(1):419
4. Hirbe H et al., The Lancet Neuro. 2014;13(8):834-843
5. Anderson P et al., Curr Med Res Opin. 2008;24(11):3063-72.
6. Anderson P et al., Curr Med Res Opin. 2023;39(12):1707-15.
7. Babineaux SM et al., BMJ Open. 2016;6(8):e010352.
8. Higgins V et al., Diabetes Metab Syndr Obes. 2016; 9:371-80.