EE443

Evaluating the economic value of first-line treatment strategies for locally advanced or metastatic urothelial carcinoma: Insights from the Portuguese healthcare system

Nuno Silverio[1], Thitima Kongnakorn[2], Solene De Boisvilliers[3], Eszter Tichy[4], Ahmed Elsada[5]

[1]Merck S.A., Alges, Portugal, an affiliate of Merck KGaA, Darmstadt, Germany, [2]PPD Evidera Health Economics & Market Access, Bangkok, Thailand, [3]PPD Evidera Health Economics & Market Access, Thermofisher Scientific Paris, France, [4]PPD Evidera Health Economics & Market Access, Budapest, Hungary, [5]Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany.

CONCLUSIONS

- **Our cost of care model showed that the healthcare costs associated with treating locally advanced/metastatic urothelial cancer (la/mUC) in Portugal are significant**
- **In the base case, the total direct healthcare cost per patient treated with induction chemotherapy with or without maintenance therapy (chemo ± maintenance) was estimated at €81,778 over a 3-year time horizon. This was 56% lower than that with enfortumab vedotin in combination with pembrolizumab (EV+P) (€185,721)**
- **Adjusting for clinical outcomes, the costs per month in progression-free survival (PFS) and overall survival (OS) were lower with chemo ± maintenance (€4,409 and €3,670, respectively) than with EV+P (€9,919 and €7,342, respectively)**
- **The incremental cost per additional survivor after 3 years for EV+P vs chemo ± maintenance was estimated at €715,551 over 3 years**
- **Within a fixed hypothetical healthcare budget of €10 million, choosing a treatment strategy of chemo ± maintenance would enable treatment of more patients and result in more patients alive after 3 years than EV+P**
- **This study highlights the financial impact of therapy for la/mUC, emphasizing the need for the economic assessment of alternative therapeutic options for la/mUC to ensure value for money and sustain healthcare delivery**

PLAIN LANGUAGE SUMMARY

- In this study, researchers estimated the costs of care over 3 years for two different treatments for patients with advanced urothelial cancer in Portugal
 - The treatments looked at in this study were chemotherapy with or without maintenance therapy (chemo ± maintenance) and enfortumab vedotin plus pembrolizumab (EV+P)
 - The costs of care considered in this study included buying the drugs, giving the drugs to patients, managing side effects, monitoring the disease, and other drugs received after disease progression
- Total costs per patient were 56% lower with chemo ± maintenance than with EV+P, mainly due to lower costs of buying the drugs
- The cost per month of survival was 50% lower with chemo ± maintenance than with EV+P
- The cost of keeping one additional patient alive after 3 years as a result of using EV+P instead of chemo ± maintenance was over €238,000 per year
- Within a fixed budget to treat patients with this disease, the healthcare system could treat more patients, and more patients would remain alive after 3 years if it used chemo ± maintenance rather than EV+P
- These findings highlight the need to balance clinical benefits with economic sustainability when selecting treatments for patients with advanced urothelial cancer

BACKGROUND

- Bladder cancer is one of the most common cancers in Portugal, with an estimated 50 and 61 new cases per 100,000 men and women, respectively, each year[1]. It represents about 4% of all new cancers and nearly 4% of cancer deaths nationwide[2]
- Recent regulatory updates in Portugal have expanded first-line (1L) treatment options for platinum-eligible patients with la/mUC. In July 2024, the EMA's CHMP issued a positive opinion for EV+P[3], followed by European Commission's approval in August 2024[4]
- In 2023, the Portuguese National Authority for Medicines and Health Products (INFARMED) approved the public financing of avelumab (AVE) for la/mUC without disease progression on or after 1L platinum-based induction chemotherapy[5] (based on the JAVELIN Bladder 100 trial[6])
- Given the financial burden associated with la/mUC, this study sought to estimate the economic impact of 1L la/mUC treatments. While EV+P has not yet been approved in Portugal for public financing, it is included in this analysis given the potential for near-term changes in the local treatment landscape

METHODS

- A health economic model compared costs and clinical outcomes between two treatment strategies for 1L over a 3-year period: chemotherapy with or without maintenance therapy (chemo ± maintenance) and EV+P. For chemo ± maintenance, the model included all patients from 1L **(Figure 1)**

Figure 1. Comparative health economic analysis of 1L treatment strategies

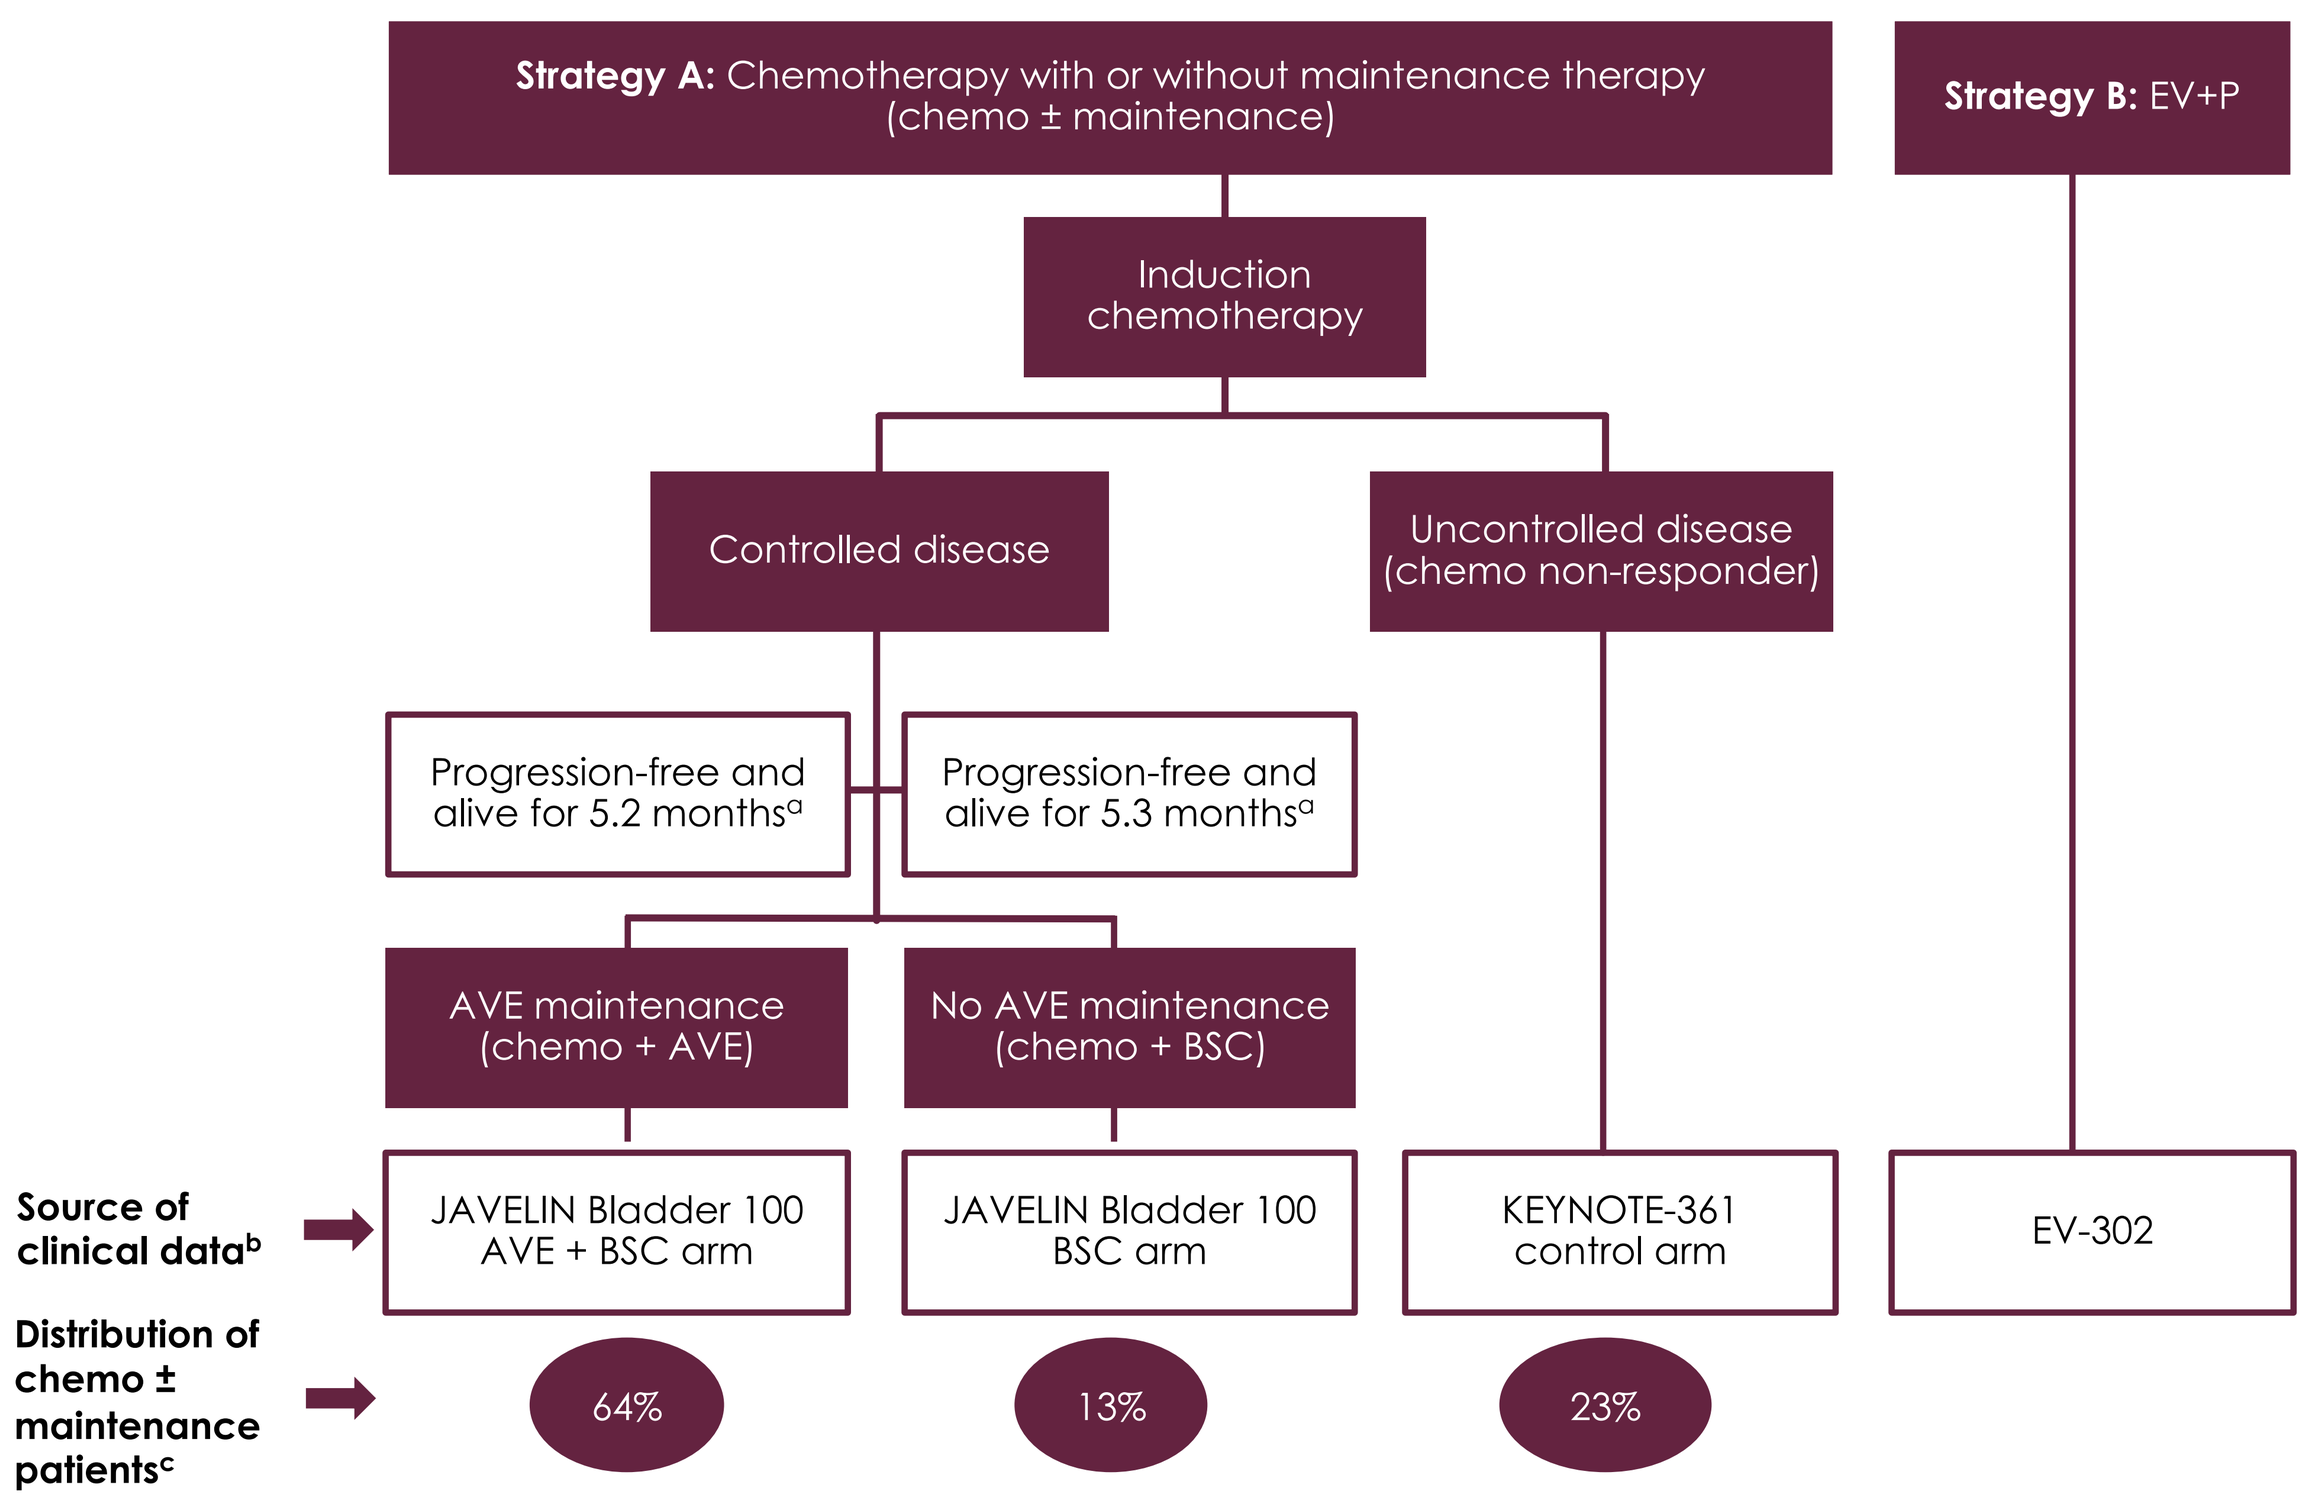
Strategy A: Chemotherapy with or without maintenance therapy (chemo ± maintenance)
Strategy B: EV+P
Induction chemotherapy
Controlled disease
Uncontrolled disease (chemo non-responder)
Progression-free and alive for 5.2 months[a]
Progression-free and alive for 5.3 months[a]
AVE maintenance (chemo + AVE)
No AVE maintenance (chemo + BSC)
Source of clinical data[b]
JAVELIN Bladder 100 AVE + BSC arm
JAVELIN Bladder 100 BSC arm
KEYNOTE-361 control arm
EV-302
Distribution of chemo ± maintenance patients[c]
64%
13%
23%

[a]In JAVELIN Bladder 100, the mean time from induction chemotherapy to randomization was 5.2 months in the AVE + BSC arm and 5.3 months in the BSC arm. Accordingly, the model assumed that chemo + AVE and chemo + BSC patients were progression-free and alive for 5.2 months and 5.3 months, respectively. Subsequently, PFS and OS reflected JAVELIN Bladder 100[6]. [b]Mean treatment duration, PFS and OS were estimated using restricted mean survival over a 3-year period (i.e., area under the curve up to 3 years), derived from parametric survival analyses. For patients with uncontrolled disease, PFS and OS were simulated based on the control arm of KEYNOTE-361[7]; treatment duration was assumed to be equivalent to PFS. [c]Based on the opinion of three local medical oncologists, 77% of patients receiving chemotherapy induction do not have disease progression and are eligible for maintenance treatment. Among those, 83% receive AVE maintenance. Therefore, among all 1L patients, 64% receive chemo + AVE, 13% receive chemo + BSC, and 23% receive chemotherapy and their disease does not respond

1L, first-line; **AVE,** avelumab; **BSC,** best supportive care; **chemo,** chemotherapy; **EV,** enfortumab vedotin; **OS,** overall survival; **P,** pembrolizumab; **PFS,** progression free survival.

- To estimate results for the chemo ± maintenance strategy, the model averaged the costs and outcomes for the 3 constituent groups, weighted by the proportion of patients in each group **(Figure 2)**
- The latest available costs (2025 EUR)—including drug acquisition and administration, disease management (which does not include hospitalization costs), adverse event (AE) management, and subsequent therapy (second-line or later line) costs—were calculated based on treatment duration, PFS, OS, and AE incidence from key clinical trials (JAVELIN Bladder 100, EV-302, KEYNOTE-361)[6-9]
- Treatment dosing schedules were obtained from Portuguese product prescribing information, and costs were obtained from public Portuguese databases and published literature[10-12]. Based on feedback from local clinicians, BSC consisted of a basket of treatments, including non-opioid and opioid analgesics, anticonvulsants, corticosteroids, benzodiazepines, antidepressants, and proton pump inhibitors
- Subsequent treatment distribution included pembrolizumab, EV, and standard chemotherapy care, based on clinical trials and expert opinion
- Main inputs are presented in **Table 1**
- The model outcomes comprise:
 - 3-year total cost of care per patient
 - Cost per month in PFS and OS: total costs incurred during PFS and OS were divided by the 3-year restricted mean PFS and mean OS, respectively
 - Incremental cost per additional survivor after 3 years: the difference in total per-patient cost between strategies was divided by the 3-year absolute reduction in the risk of death
 - Treatable patients and potential survivors within a fixed budget: The number of treatable patients was the total budget over 3 years divided by the per-patient cost for each strategy; the potential survivors after 3 years was the number of treatable patients multiplied by the 3-year OS rate for that strategy

Figure 2. PFS and OS for each treatment

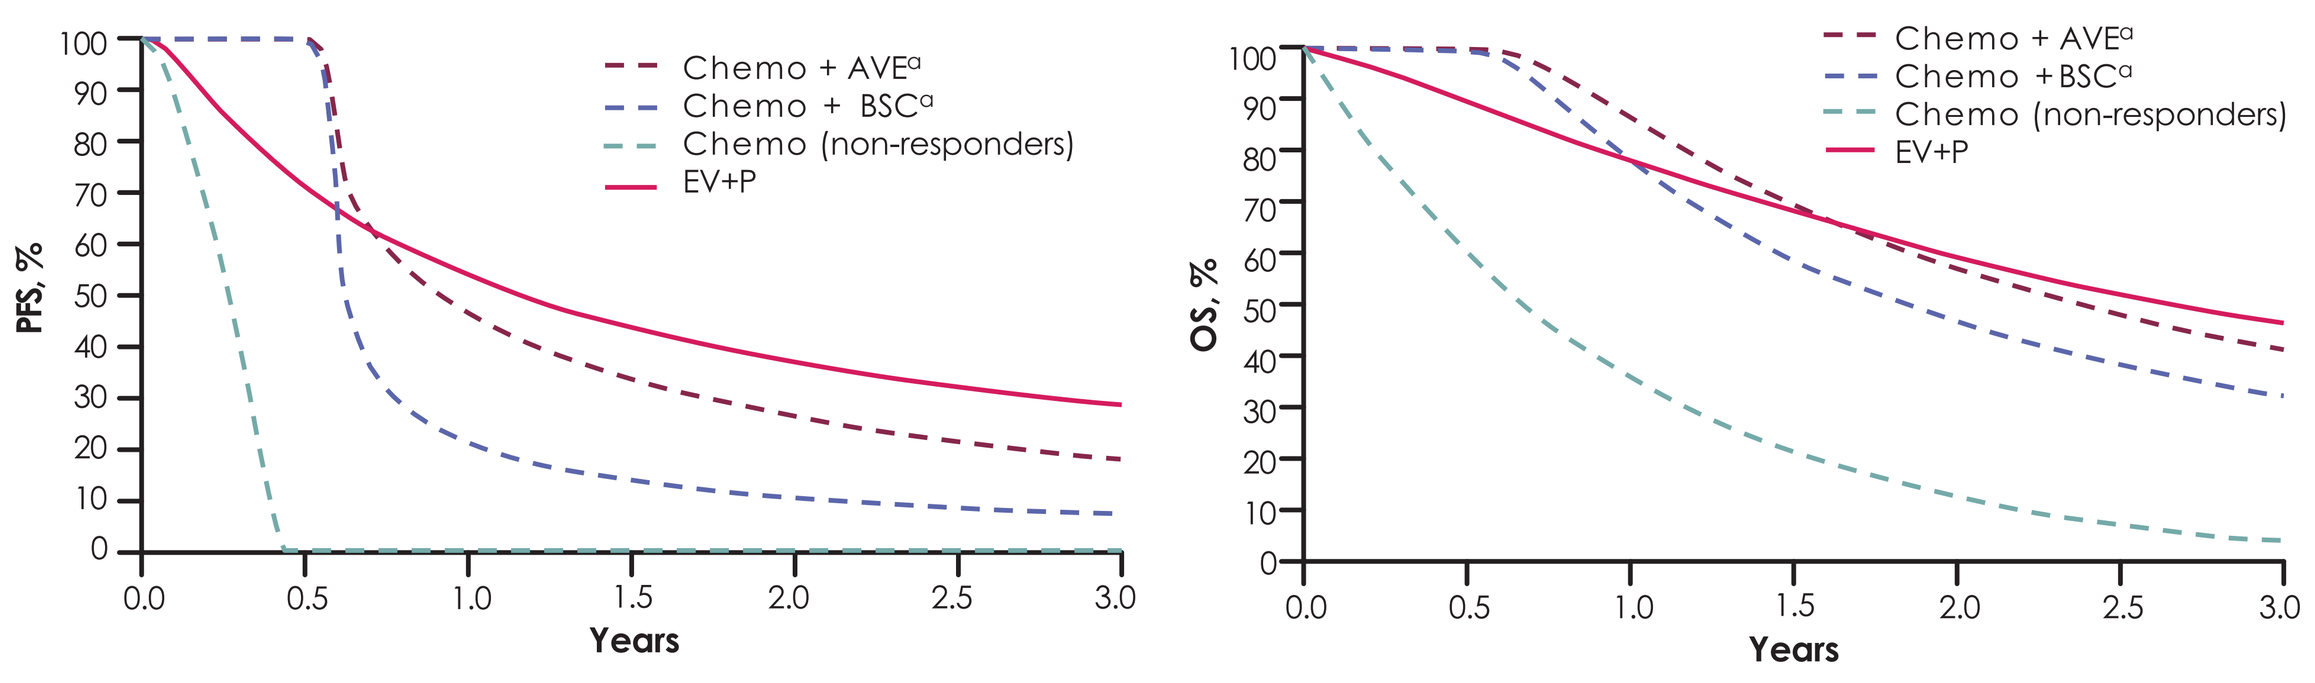
Chemo + AVE[a]
Chemo + BSC[a]
Chemo (non-responders)
EV+P
PFS, %
OS, %
Years
0.0
0.5
1.0
1.5
2.0
2.5
3.0
0
10
20
30
40
50
60
70
80
90
100

[a]Analysis includes patients who were progression-free following chemotherapy; patients who had progression or died prior to initiation of AVE or BSC were not included.
Note: The PFS and OS curves for chemo ± maintenance, from 1L, are derived by taking the weighted average of the chemo + AVE, chemo + BSC and chemo (non-responders) curves.
AVE, avelumab; **BSC,** best supportive care; **EV,** enfortumab vedotin; **OS,** overall survival; **P,** pembrolizumab; **PFS,** progression free survival.

Table 1. Key model inputs

	Chemo ± maintenance			EV+P
	Chemo + AVE	Chemo + BSC	Chemo (non-responder)	
Efficacy, months[a]				
Restricted mean duration of treatment	Chemo: 3.4[b] AVE: 11.5[6]	Chemo: 3.4[b] BSC: 6.2[c]	3.2[d]	EV: 9.9[8] PEM: 10.7[e8]
Restricted mean PFS[f]	16.8[6]	11.5[6]	3.2[7]	18.2[9]
Restricted mean OS[f]	26.1[6]	23.4[6]	11.3[7]	25.3[9]
Distribution of chemotherapies (induction therapy)[g]				
Carboplatin	42.7%			
Cisplatin	54.0%			
Gemcitabine	94.3%			
Doxorubicin, methotrexate, vinblastin	4.0 % (each)			
Rate of subsequent therapy (among patients whose disease progressed during 3 years)	88.5%	91.6%	65.0%	83.7%
Distribution of subsequent treatments (duration in months)[h]				
EV	69.3% (5.00)	35.0% (5.00)	27.5% (5.00)	0.0% (5.00)
Pembrolizumab	1.8% (4.49)	65.0% (5.71)	77.5% (4.57)	0.0% (4.49)
Carboplatin	13.6% (3.68)	12.5% (3.82)	0.0% (3.06)	55.0% (3.68)
Cisplatin	8.9% (3.68)	7.5% (3.82)	0.0% (3.06)	30.0% (3.68)
Gemcitabine	19.3% (3.68)	12.5% (3.82)	0.0% (3.06)	65.0% (3.68)
Paclitaxel	17.0% (3.68)	12.5% (3.82)	8.3% (3.06)	15.0% (3.68)
Cost per month, 2025 Euro				
Drug and administration cost	Induction: €1,491 Maintenance: €6,481[i]	Induction: €1,491 Maintenance: €9	€1,491	EV: €9,955[i] P: €7,075[i]
Disease management costs (per month unless noted)[j]				
Disease management - Pre-progression/ On-treatment	Induction: €332 Maintenance: €269	Induction: €332 Maintenance: €312	€332	€332
Disease management - Pre-progression/ Off-treatment	€312			
One-time cost of progression	€580			
Disease management – Post-progression/on subsequent active treatment	€390			
Disease management – Post-progression/off-treatment	€527			
Monthly cost of subsequent treatments	€3,870	€6,237	€4,443	€314
AEs management cost[k] (one-off)	Induction: €883 Maintenance: €115	Induction: €883 Maintenance: €63	€820	€299

[a]Estimated using restricted mean survival over a 3-year period, derived from parametric survival analyses. [b]Induction phase duration assumed to be 15 weeks based on JAVELIN Bladder trial. [c]Assumed BSC was given until disease progression. Treatment duration was estimated as PFS (11.5 months) - time to start maintenance (5.3 months) = 6.2 months. [d]Assumed to match PFS (treated to progression). [e]Maximum treatment duration of 24 months for pembrolizumab was taken into account in this estimate. [f]From the initiation of chemotherapy. [g]Distribution was based on average clinician input. [h]Distribution was based on average clinician input; durations were from trials[13-15]. For chemo (non-responder), duration of pembrolizumab and chemotherapies was assumed 20% shorter than those for chemo + BSC, based on clinician input. [i]AVE cost is based on a 200-mg vial price of €621.25[11]. EV cost based on a 20-mg vial price of €626.00[11]. P cost is based on a 100-mg vial price of €2,440.72[11]. [j]Calculated from monthly frequency, percentages of patients using each resource provided by local clinicians, and unit costs. [k]Included grade 3+ AEs occurring in ≥2% of any of the treatment options. Calculated based on frequency of AEs from trial data[6,8] and unit costs sourced from published literature and databases.

AEs, adverse events; **AVE,** avelumab; **BSC,** best supportive care; **chemo,** chemotherapy; **EV,** enfortumab vedotin; **OS,** overall survival; **P,** pembrolizumab; **PFS,** progression free survival.

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ISPOR and the corresponding author of this poster.

Correspondence: **Nuno Silverio,** nuno.silverio@merckgroup.com

RESULTS

Base case results

- As shown in **Figure 3**, the estimated total cost of care per patient over 3 years was 56% lower with chemo ± maintenance than with EV+P, with a cost of €81,778 compared to €185,721, respectively. This was driven by higher drug costs for EV+P, despite chemo ± maintenance incurring higher AE management and subsequent therapy costs
- Monthly costs in PFS and OS with chemo ± maintenance are 56% and 50% lower than those of EV+P, respectively (**Table 2**)
- The incremental cost per additional survivor after 3 years for EV+P vs chemo ± maintenance was estimated at €715,551 over 3 years (**Table 3**)
- Within a fixed hypothetical budget of €10,000,000 over 3 years, 122 patients could be treated with chemo ± maintenance vs 54 patients with EV+P, and 39 vs 25 patients, respectively, could be alive at year 3 (**Figure 4**)

Figure 3. Base case results

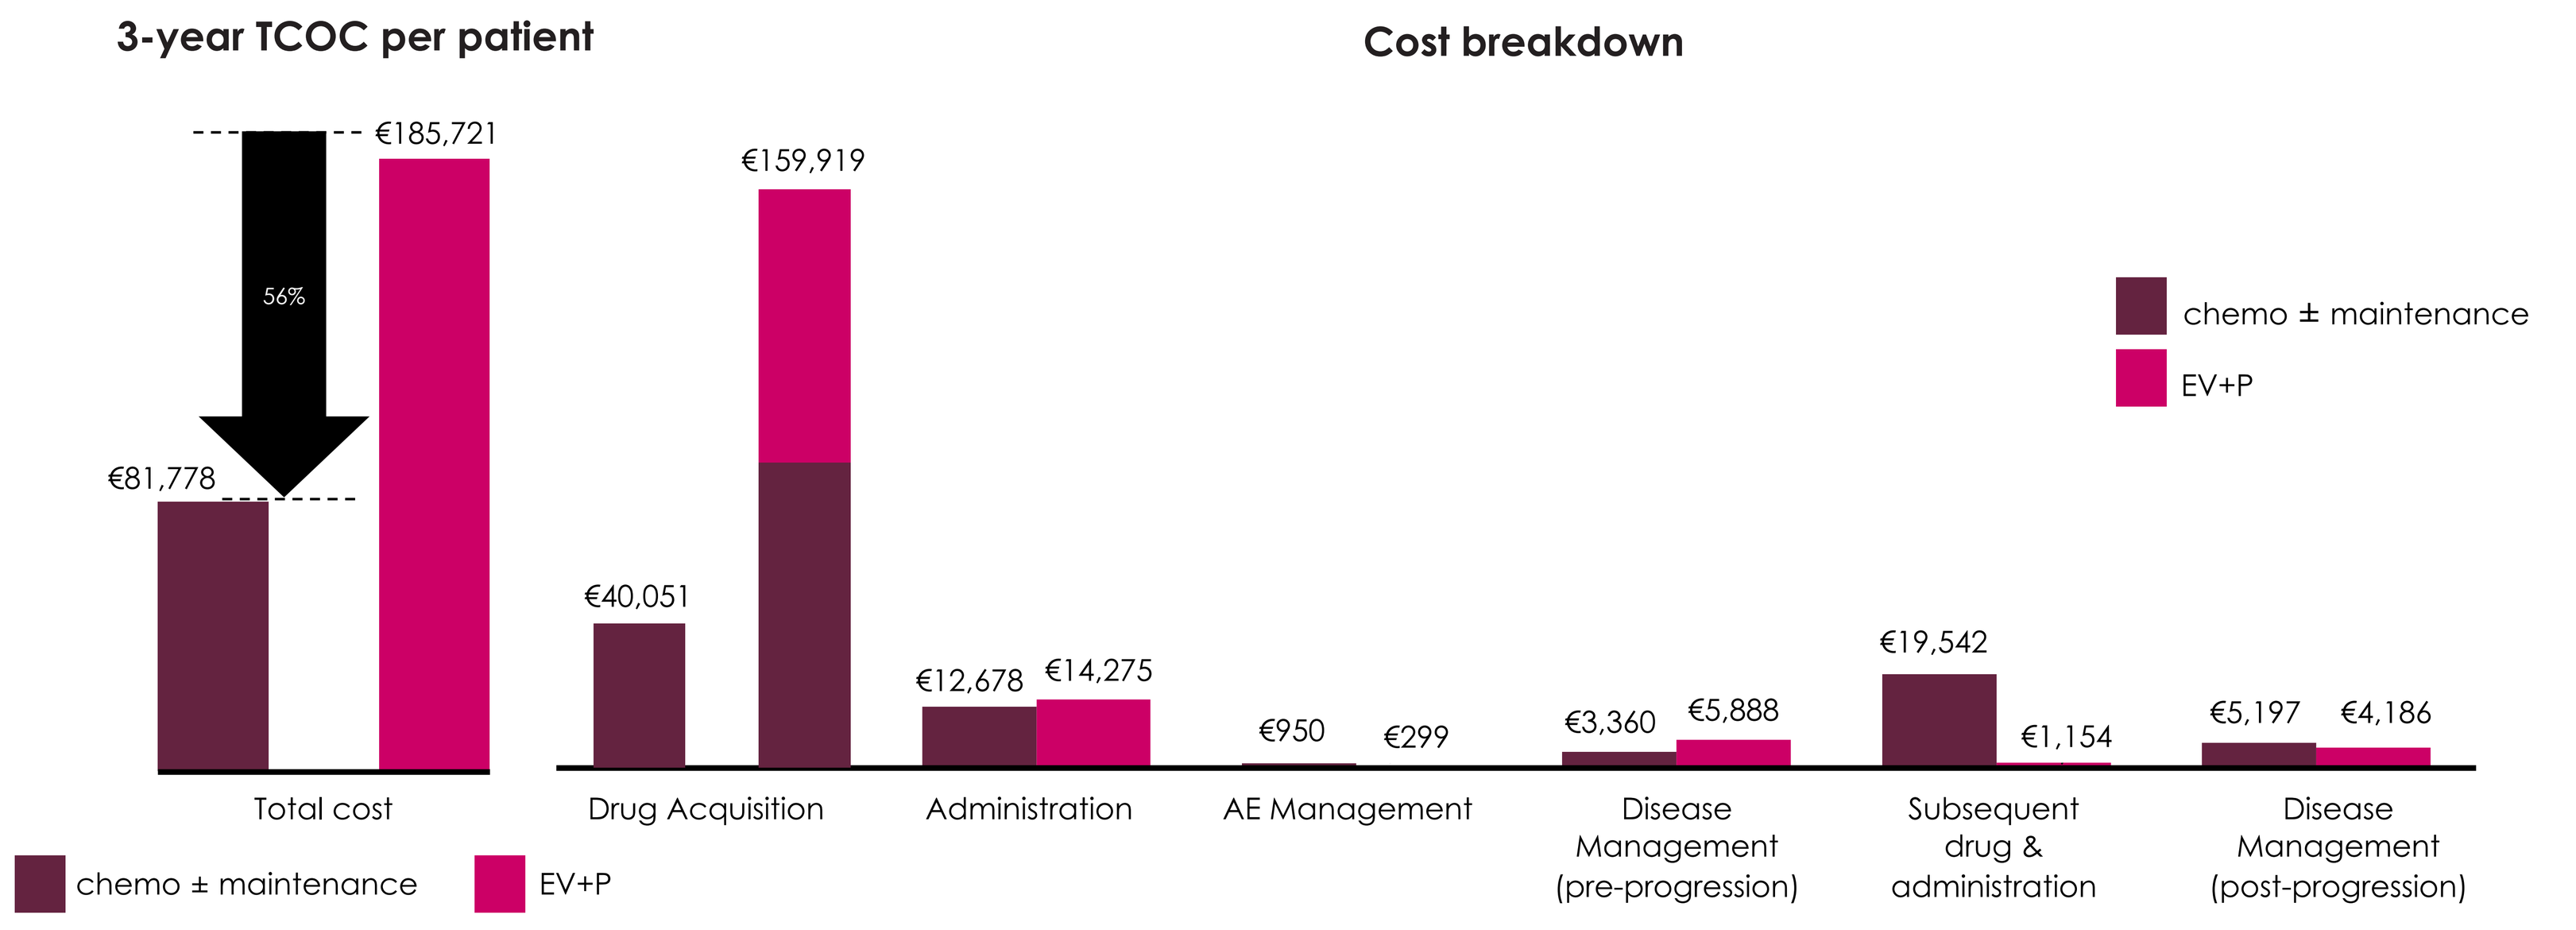
3-year TCOC per patient
Cost breakdown
€185,721
56%
€81,778
Total cost
€159,919
€40,051
Drug Acquisition
€12,678
€14,275
Administration
€950
€299
AE Management
€3,360
€5,888
Disease Management (pre-progression)
€19,542
€1,154
Subsequent drug & administration
€5,197
€4,186
Disease Management (post-progression)
chemo ± maintenance
EV+P

1L, first line; **AE,** adverse event; **AVE,** avelumab; **EV,** enfortumab vedotin; **P,** pembrolizumab; **TCOC,** total cost of care.

Table 2. Cost per month in PFS and OS

	Total per-patient cost during PFS	Total per-patient cost during OS	Mean PFS (months)	Mean OS (months)	Cost per month in PFS	Cost per month in OS
Chemo ± maintenance	€57,039	€81,778	12.9	22.3	€4,409	€3,670
EV+P	€180,380	€185,721	18.2	25.3	€9,919	€7,342
% difference (chemo ± maintenance vs EV+P)					−55.55%	−50.01%

Chemo, chemotherapy; **EV,** enfortumab vedotin; **OS,** overall survival; **P,** pembrolizumab; **PFS,** progression free survival.

Table 3. Cost per additional survivor

	3-year TCOC	OS rate at 3 years	Incremental 3-year TCOC	Absolute risk reduction	Cost of keeping one additional patient alive after 3 years
Chemo ± maintenance	€81,778	32.0%	–	–	–
EV+P	€185,721	46.5%	€103,943	14.5%	€715,551 (€238,517 annually)

Chemo, chemotherapy; **EV,** enfortumab vedotin; **OS,** overall survival; **P,** pembrolizumab; **TCOC,** total cost of care.

Figure 4. Treatable patients and potential survivors within a fixed budget

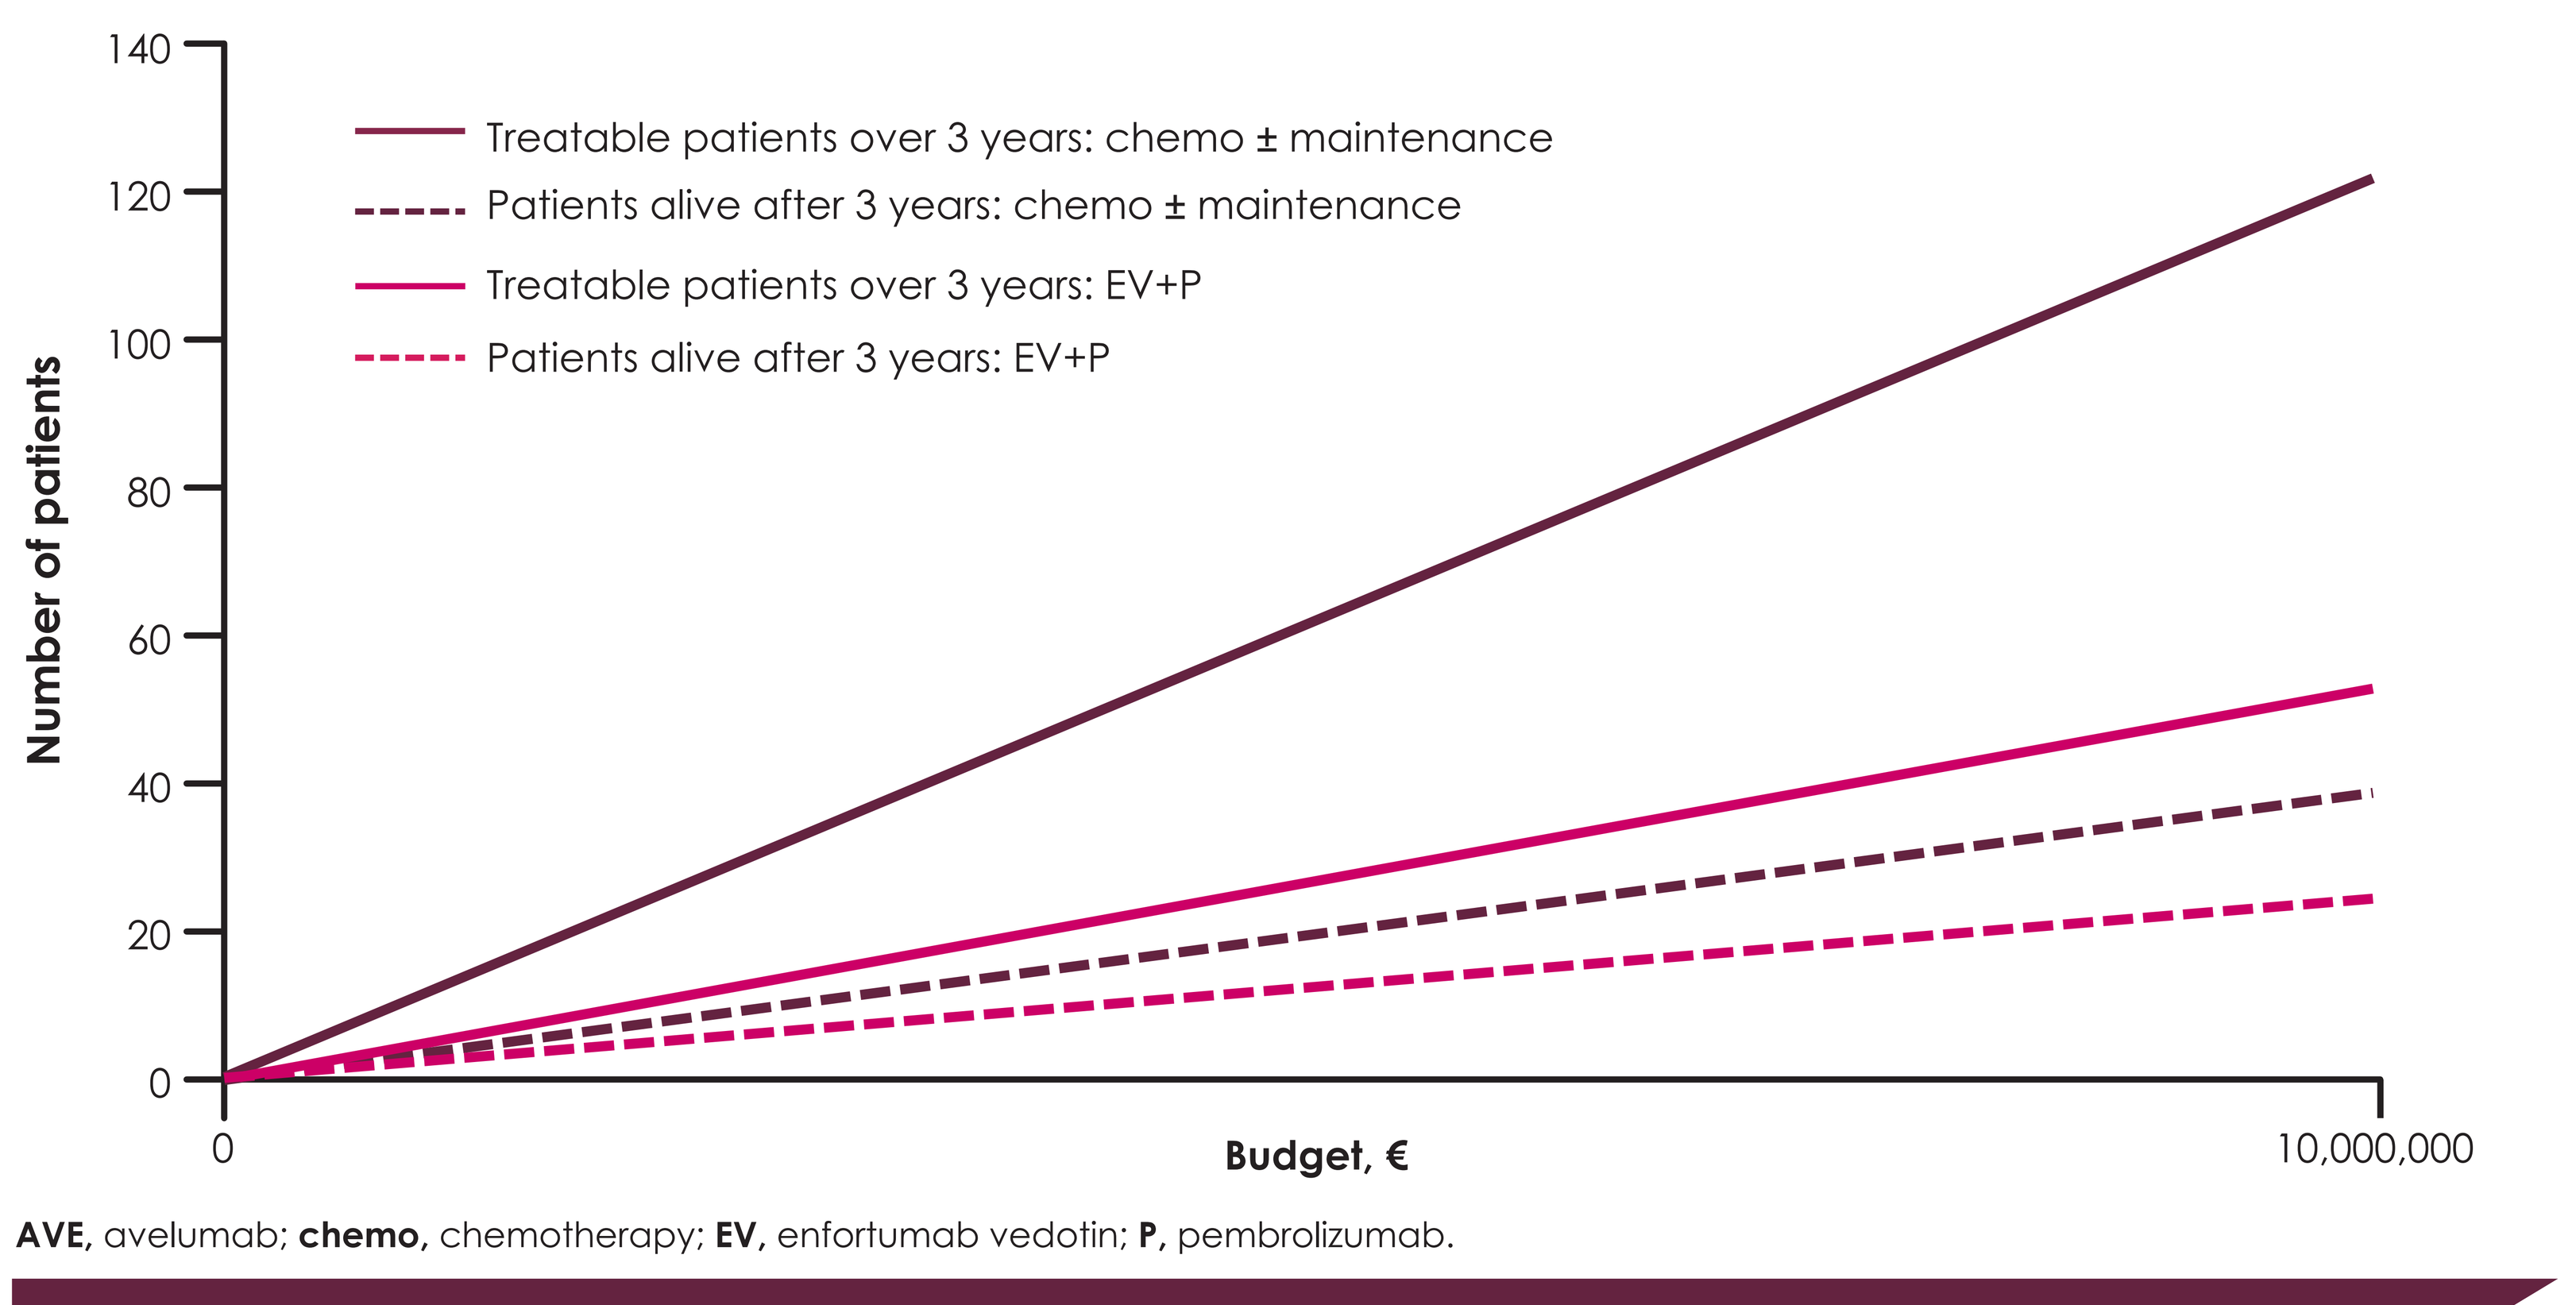
Treatable patients over 3 years: chemo ± maintenance
Patients alive after 3 years: chemo ± maintenance
Treatable patients over 3 years: EV+P
Patients alive after 3 years: EV+P
Number of patients
0
20
40
60
80
100
120
140
Budget, €
0
10,000,000

AVE, avelumab; **chemo,** chemotherapy; **EV,** enfortumab vedotin; **P,** pembrolizumab.

LIMITATIONS

- Clinical inputs, such as treatment duration, PFS, OS, and AE profiles, were based on clinical trials and assumed to represent real-world practice in Portugal
- Treatments have been compared naively, as baseline characteristics at the start of 1L therapy were not available for all treatments. Consequently, some selection bias due to imbalances in certain baseline characteristics between treatments may be present
- Costs and outcomes are disconnected, as clinical inputs were derived from published trials and data, while other inputs, such as disease management, were based on published literature and input from local clinicians
- To minimize uncertainty about long-term outcomes and costs, the model used a 3-year time horizon; therefore, the clinical and economic effects beyond 3 years are not captured
- The model assumed 100% compliance with the recommended treatment dosages, which in a real-world scenario may not be the case as patients may interrupt treatment or adjust the dose
- AEs associated with subsequent treatments were not considered in the model
- Efficacy of subsequent treatments were assumed to be captured in the OS obtained from the clinical trials. Given the evolving treatment landscape for la/mUC, subsequent treatments used in the clinical trials may not represent those currently used.

REFERENCES 1. OECD - Organisation for Economic Co-operation and Development. Perfis Do Cancro Por País - Portugal, 2025. Accessed 30 September 2025. www.oecd.org/content/dam/oecd/pt/publications/reports/2025/02/eu-country-cancer-profile-portugal-2025_699bde59/ffdcd7a9-pt.pdf. **2.** World Health Organization. Cancer Country Profile 2020: Portugal. prt-2020.pdf. Accessed 30 October 2025 **3.** OncLive. Pembrolizumab plus enfortumab vedotin earns CHMP recommendation in frontline metastatic urothelial carcinoma, 2024. Accessed 29 Jul 2025. www.onclive.com/view/pembrolizumab-plus-enfortumab-vedotin-earns-chmp-recommendation-in-frontline-metastatic-urothelial-carcinoma. **4.** Astellas. Accessed 29 Jul 2025. https://newsroom.astellas.us/. **5.** National Authority for Medicines and Health Products (INFARMED). Accessed 30 Sept. 2025. **6.** Powles T, et al. J Clin Oncol. 2023;41(19):3486-3492. **7.** Powles T, et al. Lancet Oncol. 2021;22(7):931-945. **8.** Powles T, et al. N Engl J Med. 2024;390(10):875-888. **9**. Powles T, et al. J Clin Oncol. 2025,43(5);664. **10.** Republica Portuguesa, SNS. CIRCULAR NORMATIVA. 4 Feb. 2025. https://www.acss.min-saude.pt/wp-content/uploads/2016/11/Circular_Normativa_2025.pdf. **11**. SPMS- Catálogo de Aprovisionamento Público Da Saúde. Accessed June 2025. http://www.catalogo.min-saude.pt/. **12.** Decree-Order number 207/2017, published July 11, 2017. **13.** Merck Data on file. **14.** Powles T, et al. N Engl J Med. 2021;384(12):1125-1135. **15.** Loriot Y, et al. N Engl J Med. 2023;389(21):1961-1971.

DISCLOSURES N. Silverio and **A. Elsada** are employees of Merck. **T. Kongnakorn, S. de Boisvilliers,** and **E. Tichy** are employees of Evidera, a company that received financial support from Merck for the development of this study and the abstract. **ACKNOWLEDGMENTS** This study was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Nucleus Global and was funded by Merck. Editorial support was provided by Pranali Mehulbhai Bhatt (Merck Specialities Private Limited) and was funded by Merck. Authors would like to express their thanks to the clinical medical panel supporting the validation of the data included in the model, namely: Maria Rosinha, MD, Sandra Custódio, MD and Raquel Brás, MD.

Poster No. EE443. Presented at ISPOR Europe, 9-12 November 2025; Glasgow, Scotland, UK.