Anggie Wiyani | anggie.wiyani@novartis.com

HTA 136

Evaluating the Value Elements Considered in Health Technology Assessments of Paroxysmal Nocturnal Haemoglobinuria Treatments: A Targeted Review

Anggie Wiyani,[1] Misaki Okuyama,[2] Eve McArthur,[2] Maria-Magdalena Balp[3]

[1]Novartis Pharmaceuticals UK Ltd., London, UK; [2]Costello Medical, London, UK; [3]Novartis Pharma AG, Basel, Switzerland

KEY FINDINGS & CONCLUSIONS

- This review of the use of value elements for treatments in PNH identified that several elements beyond QALYs and Net Costs were included in CSs and CRs.
- The impact of the novel value elements on HTA decision-making remains unclear; however, some were acknowledged by few HTA bodies, but the individual impact of each element was not quantified.
- Standardized definitions and quantifiable metrics might be valuable and needed to support the implementation of the value elements in the submission dossiers and better consideration by HTA bodies.
- Joint efforts by all stakeholders are essential to facilitate adoption of novel value elements in HTA decision-making.

This study is funded by Novartis Pharma AG
Poster presented at the ISPOR Europe, held on 9–12 November 2025

INTRODUCTION

- The ISPOR value flower includes 10 value elements beyond Quality-Adjusted Life Years (QALYs) and Net Cost that warrant consideration in health technology assessments (HTAs): Productivity, Family and Scientific Spillover, Equity, Disease Severity, Insurance, Fear of Disease, Value of Knowing, Value of Hope and Real-Option Value.[1]
- HTA bodies' perspectives on these values and their adoption remains unclear, particularly in rare diseases including paroxysmal nocturnal hemoglobinuria (PNH).

OBJECTIVE

- The objective of this analysis was to assess the inclusion of the value flower elements in Company HTA Submissions for PNH therapies and how the HTA bodies assessed and considered them in their final decisions. The review focused on the impact of novel value elements, excluding QALY and Net Costs, which are already well recognized by HTA bodies.

METHODS

- A targeted review was conducted in June 2025 for PNH treatments approved since 2020 (iptacopan, pegcetacoplan, danicopan, crovalimab) across five HTA body websites; National Institute for Health and Care Excellence (NICE; United Kingdom) ,Gemeinsamer Bundesausschuss (GBA; Germany), Haute Autorité de Santé (HAS; France), Tandvårds- och läkemedelsförmånsverket (TLV; Sweden), Medicinrådet (Denmark).
- Value elements and their impact on HTA bodies decisions were extracted from each available company submission (CSs) and HTA Committee reports (CRs) into a structured grid based on the interpretation of the reviewers in relation to ISPOR's definition of value elements.[1]
- Extracted data contained:
 - The value elements reported by CSs, including the format in which they were presented:
 - Qualitative: Value elements presented with narrative description
 - Quantitative: Value elements presented with numerical values / specific scales
 - Mixed-methods: Quantitative and qualitative
 - HTA assessment and decision on the submitted value elements as present in the CRs

RESULTS

- A total of 13 CSs were reviewed: NICE (n=4), GBA (n=6), and Medicinrådet (n=3). CSs to HAS (n=4) and TLV (n=2) were not publicly available. All CSs to NICE and Medicinrådet received favorable recommendation. Out of 6 CSs to GBA, 5 received 'hint for a non-quantifiable benefit', while 1 received 'no additional benefit'.
- Eight value elements were identified in CSs: QALY, Net Cost, Productivity, Real Option-Value, Equity, Disease Severity, Family Spillover and Value of Knowing.
- A total of 19 HTA CRs were reviewed: NICE (n=4), GBA (n=6), Medicinrådet (n=3), HAS (n=4), and TLV (n=2). Among 4 HAS CRs, 3 concluded 'favorable opinion', while 1 concluded 'unfavorable opinion'. For TLV, both CRs concluded with 'no benefit'.
- Six value elements were identified in CRs: QALY, Net Cost, Productivity, Real Option-Value, Equity and Disease Severity.
- Fear of Contagion, Insurance Value, Value of Hope and Scientific Spillover were not identified in any of the CSs and CRs **(Figure 1)**.

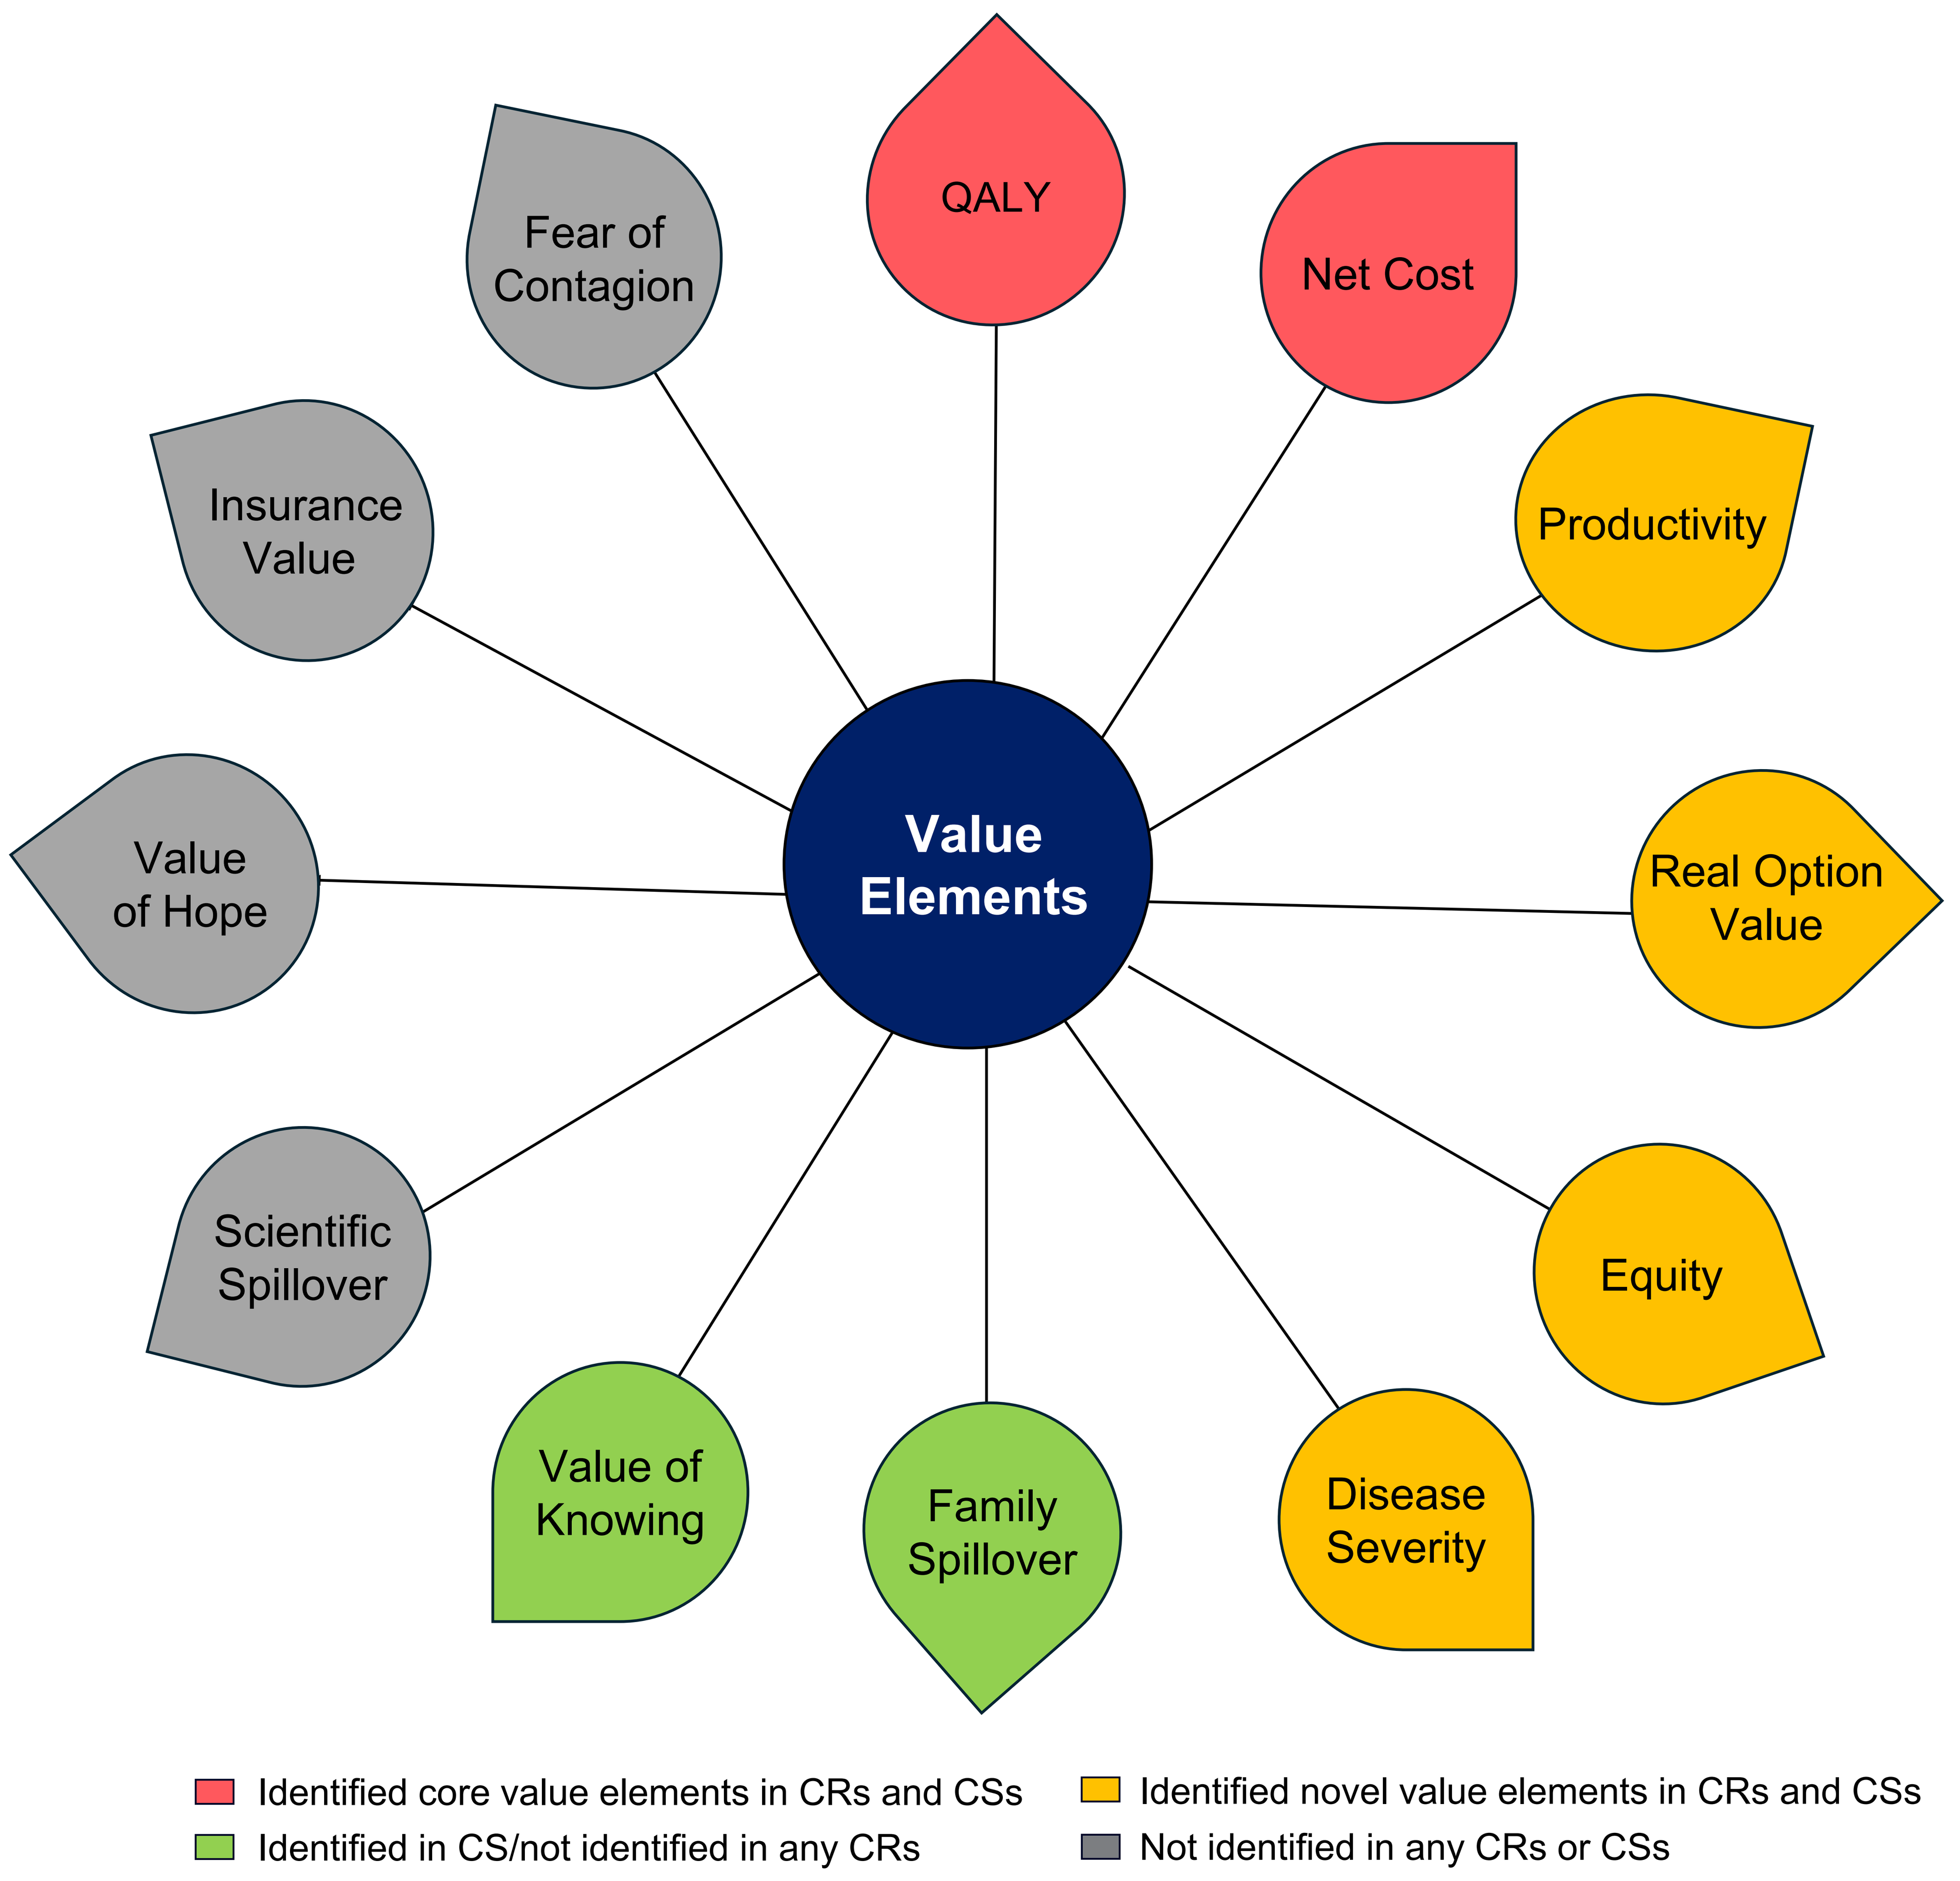
QALY
Net Cost
Fear of Contagion
Insurance Value
Productivity
Value of Hope
Value Elements
Real Option Value
Scientific Spillover
Equity
Value of Knowing
Family Spillover
Disease Severity
Identified core value elements in CRs and CSs
Identified novel value elements in CRs and CSs
Identified in CS/not identified in any CRs
Not identified in any CRs or CSs

Figure 1: Value elements in PNH HTAs

Value Elements Reported in Company Submissions (CSs)

- QALY (n=7) and Net Cost (n=13) were the most frequently reported value elements.
- Among the novel value elements, Productivity (n=9) and Real-Option Value (n=9) were the most frequently identified.
- The value elements were presented mostly qualitatively (n=23) and few with mixed-method (n=3) or quantitative data (n=1) **(Table 1)**.

Table 1: Novel Value Elements Captured Across Company Submissions in PNH

Value elements	Total CSs (n)	HTA body			Reporting method		
		NICE (n)	GBA (n)	Medicinrådet (n)	Quantitative (n)	Qualitative (n)	Mixed (n)
Productivity	**9**	4	4	1	1	6	2
Real-Option Value	**9**	4	2	3	NA	9	NA
Disease Severity	**1**	NA	NA	1	NA	1	NA
Equity	**4**	4	NA	NA	NA	4	NA
Family Spillover	**3**	2	1	NA	NA	2	1
Value of Knowing	**1**	1	NA	NA	NA	1	NA

NA: Not Applicable

Value Elements Considered in HTA Committee Reports (CRs)

- Medicinrådet and TLV did not consider any of the novel value elements mentioned in the CSs within their published CRs.
- Although Productivity was the most identified value element in CSs, only 2 CRs considered it (NICE acknowledged and GBA dismissed).
- Real-Option Value was also commonly reported in CSs; however, only 2 NICE CRs considered it - one acknowledged and the other dismissed it.
- Disease Severity was identified in three CRs published by HAS. However, as the corresponding CSs to HAS were not publicly available, the authors cannot confirm how this element was submitted in those CSs.
- Equity was identified in three CRs as follows: acknowledged in 1 HAS and 1 NICE CRs and dismissed in another NICE report **(Table 2)**.

Table 2: Novel Value Elements Identified Across HTA Committee Reports in PNH and HTA Bodies' Considerations

Value elements	Total CRs (n)	HTA body consideration		
		NICE (A/D)	HAS (A/D)	GBA (A/D)
Productivity	**2**	1/NA	NA	NA/1
Real-option value	**2**	1/1	NA	NA
Disease severity	**3**	NA	2/1	NA
Equity	**3**	1/1	1/NA	NA

A: Acknowledged; D: Dismissed; NA: Not Applicable

References

1.Darius N et al.Value in health.2018;21;Issue 2:131-139.
2.NICE.[TA 778];2022.
3.NICE.[TA 1000];2024 4.NICE.[TA 1010];2024.
5.NICE.[TA 1019];2024.
6.HAS. [CT20829];2024.
7.HAS.[CT20839];2024.
8.HAS.[CT20930];2024.
9.HAS.[CT2021273];2025.
10.Medicinrådet. [181784];2023.
11.Medicinrådet.[197691];2024.
12.Medicinrådet.[210764];2025.
13.GBA;2022.
14.GBA;2024.
15.TLV;2022. 16.TLV;2025

Acknowledgements:

The authors acknowledge Jebas Arun Singh from Novartis Healthcare Private Limited, India for medical writing assistance. The final responsibility of the content lies with the authors.

Disclosures:

AW: Employee of Novartis Pharmaceuticals UK Ltd.; EM, MO: Employees of Costello Medical, which received payment from Novartis Pharma AG for analytical services for this study; M-MB: Employee of Novartis Pharma AG

Scan to obtain:
- Poster

https://medicalcongressposters.com/Default.aspx?doc=472ba
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.