EE250

Helen Trenz | helen.trenz@novartis.com

Quantifying Spillover Impacts: Effect of Novel Therapies for IgA Nephropathy on Patients Awaiting Kidney Transplant

Jaehong Kim,[1] Jacob Fajnor,[1] Jason Shafrin,[1,2] Briana Ndife,[3] Helen Trenz,[3] Titte R. Srinivas,[3] Ajay Israni[4]

[1]Center for Healthcare Economics and Policy, FTI Consulting, Los Angeles, California, USA;
[2]Alfred E. Mann School of Pharmacy and Pharmaceutical Sciences, University of Southern California, Los Angeles, California, USA;
[3]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA;
[4]Department of Internal Medicine, University of Texas Medical Branch, Galveston, Texas, USA

KEY FINDINGS & CONCLUSIONS

- Novel IgAN interventions saved an average of 669 kidneys per year, which reduced overall kidney transplant waitlist size, reduced average transplant wait time by 25 days per waitlist candidate, and generated an additional 0.087 QALYs (monetized benefit: $13,116) per waitlist candidate
- Within 5 years of model start, IgAN interventions reduced the overall size of the kidney transplant waitlist by 2138 candidates
- Spillover benefits (0.410 QALYs and $61,485 per patient with IgAN) comprised 23.1% of total IgAN treatment value
- To capture the full societal value of novel IgAN treatments, the spillover benefits of reduced kidney transplant demand should be considered

Scan to obtain:
- Poster
- Supplementary methods
- Supplementary figure and tables

https://novartis.medicalcongressposters.com/Default.aspx?doc=6e929

Copies of this poster obtained through the Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study was sponsored by Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA.

Poster presented at: International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2025, May 13–16, 2025, Montréal, Québec, Canada.

INTRODUCTION

- IgAN is the most common primary glomerulonephritis worldwide and a leading cause of CKD; up to 50% of patients experience kidney failure within 10 to 20 years of diagnosis[1-4]
- In the US, the OPTN kidney transplant waitlist includes >90,000 candidates with a median transplant wait time of ~4 years[5,6]
- In recent years, the treatment landscape for IgAN has rapidly evolved.[7] Novel IgAN therapeutics may reduce kidney transplant demand among patients with IgAN and thereby increase organ availability for other transplant candidates
- This study quantified not only the direct health economic benefits of novel interventions for patients with IgAN but also highlights their substantial impact ("spillover benefits") on the US kidney transplant system by reducing the number of transplant candidates on the kidney transplant waitlist

METHODS

- This study implemented two health economic modeling frameworks:
 - First, a Markov model estimated health benefits accrued among adults with IgAN receiving novel therapies
 - Second, Markov model results were inputted into a double queuing model to quantify the additional value ("spillover benefits") accrued by all candidates in need of a living or deceased donor kidney transplant in the US ("waitlist")

Spillover Model

- The overall structure of the spillover model (comprising the Markov model and the double queuing model) is shown in **Figure 1**
- **Markov model:** The progression-based simulation predicted reduced entry of patients with IgAN to the transplant waitlist due to interventions, using CKD staging and assuming a uniform uptake rate for all interventions (**Supplementary Methods, Supplementary Figure 1**)
- **Double queuing model:** Accounting for the dynamic interplay between patient demand and organ supply, the relative reduction in patients with IgAN entering the kidney transplant waitlist from the Markov model was applied to the OPTN-reported annual arrival rate of patients with IgAN and incorporated into the double queuing model
- PICOTS criteria for the spillover model are summarized in **Table 1**
- Key inputs for the spillover model are summarized in **Table 2**; the full list of inputs is provided in **Supplementary Table 1**

Figure 1. Spillover Model Structure

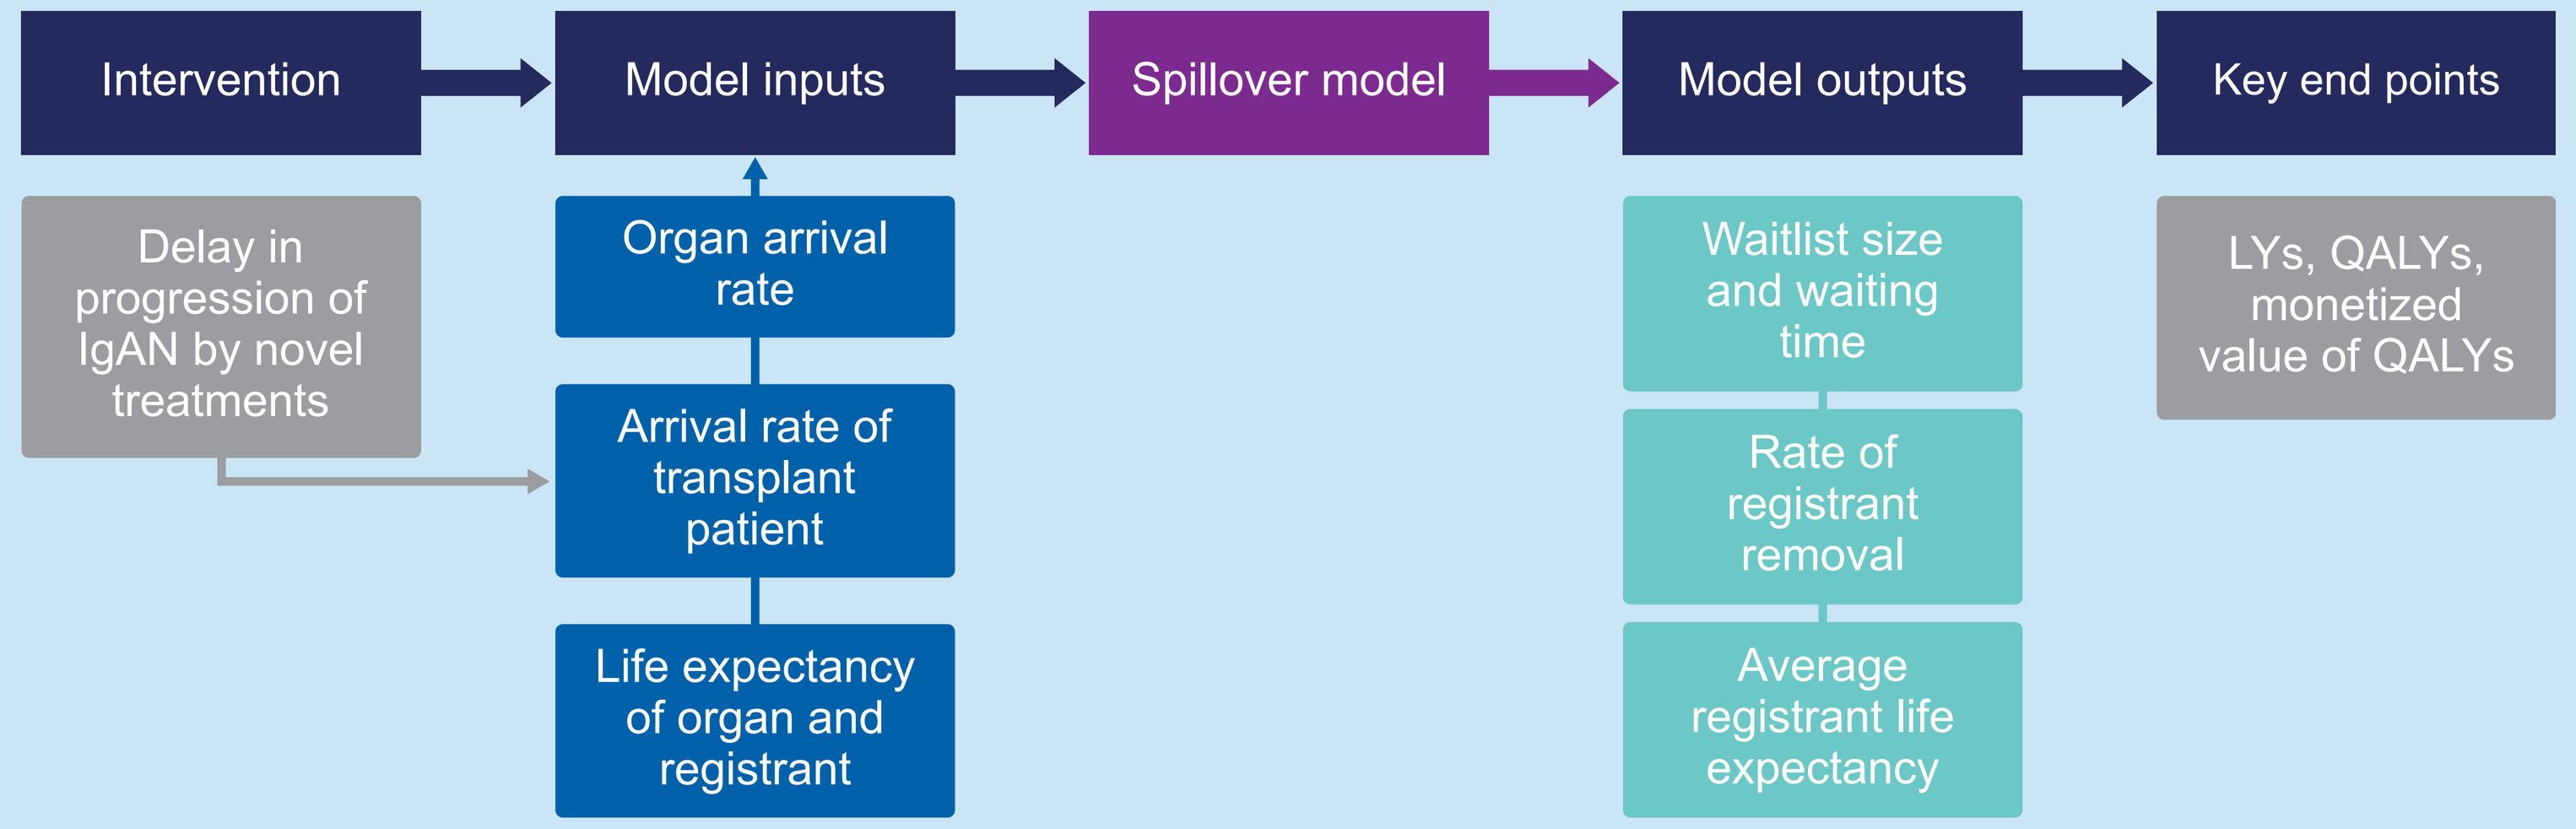
Intervention
Model inputs
Spillover model
Model outputs
Key end points
Delay in progression of IgAN by novel treatments
Organ arrival rate
Arrival rate of transplant patient
Life expectancy of organ and registrant
Waitlist size and waiting time
Rate of registrant removal
Average registrant life expectancy
LYs, QALYs, monetized value of QALYs

IgAN, immunoglobulin A nephropathy; LY, life-year; QALY, quality-adjusted life-year.

Table 1. PICOTS Criteria for the Spillover Model

Criteria	Description
Population	Individuals in the US of any age awaiting kidney transplant
Intervention	FDA-approved therapeutics that reduce proteinuria (iptacopan, atrasentan)[8,9] or slow eGFR decline (sparsentan, delayed-release budesonide, dapagliflozin)[10-12] in patients with IgAN, in addition to supportive care and systemic corticosteroids
Comparator	Supportive care (ACEi/ARB, blood pressure management, lifestyle changes, CVD management) and systemic corticosteroids. Patients with kidney failure were assumed to receive dialysis
Outcomes	**Clinical outcomes:** Reduction in kidney demand (average number of kidneys saved annually); arrival rate of treated patients with IgAN to the waitlist; average number of patients with CKD on the waitlist; average transplant wait time; LYs **Health economic outcomes:** QALYs; monetized value of additional health gains; share of treatment value attributable to spillover effects
Timing	Lifetime horizon of 70 years; 3-month cycle length
Setting	Societal perspective

ACEi, angiotensin-converting enzyme inhibitor; ARB, angiotensin II receptor blocker; CKD, chronic kidney disease; CVD, cardiovascular disease; eGFR, estimated glomerular filtration rate; FDA, US Food and Drug Administration; IgAN, immunoglobulin A nephropathy; LY, life-year; PICOTS, population, intervention, comparator, outcomes, timing, and setting; QALY, quality-adjusted life-year; US, United States.

Table 2. Key Spillover Model Inputs

Parameter	Value
No. of organ arrivals per year	27,332
No. of total waitlist arrivals per year	44,561
No. of patients with IgAN arriving to waitlist per year	2004
Time to candidate removal (years)	5.239
US IgAN prevalence (%)	0.033
US IgAN incidence (%)	0.003
Treatment efficacy (HR)[a]	0.526
Quality of life	
CKD 1, CKD 2, CKD 3a, CKD 3b	0.670
CKD4	0.550
ESKD	0.540
LYs[b]	
With transplant	14.66
Without transplant	7.96
QALYs[b]	
With transplant	11.39
Willingness to pay for a QALY (US$)	150,000

[a]The Markov model used a mean disease progression delay calculated from observed reductions in eGFR decline or meta-regression estimates of eGFR decline from available proteinuria reduction data.[8-12] [b]Literature values discounted at a rate of 3.5%. Values were undiscounted assuming a uniform distribution of LYs and constant universal discounting method.

CKD, chronic kidney disease; eGFR, estimated glomerular filtration rate; ESKD, end-stage kidney disease; HR, hazard ratio; IgAN, immunoglobulin A nephropathy; LY, life-year; QALY, quality-adjusted life-year; US, United States.

Scenario Analysis

- The study considered several scenarios of interest, including variations in (i) treatment utilization; (ii) cost-effectiveness threshold; (iii) waitlist size; (iv) average candidate time on waitlist; and (v) annual waitlist candidate arrival rate

RESULTS

- Novel IgAN interventions reduced annual kidney transplant demand by 33.4%, saving an average of 669 kidneys per year (**Figure 2**)
- The reduction in kidney transplant demand among treated patients with IgAN reduced the overall waitlist size by 2138 candidates within 5 years of model start and by 3586 candidates in the steady-state equilibrium (**Figure 3**)
- The spillover benefits of IgAN interventions for patients on the waitlist are summarized in **Table 3**
 - Reductions in waitlist size decreased the average transplant wait time by 25 days (0.070 years) per waitlist candidate
 - Reductions in wait times generated 0.087 QALYs (monetized benefit US$13,116) per waitlist candidate
- Per treated patient with IgAN, novel interventions generated 1.772 QALYs, 23.1% (0.410 QALYs) of which were attributable to spillover benefit (**Figure 4**)
- Spillover benefits per patient with IgAN were mildly sensitive to treatment utilization (100% dapagliflozin treatment: 21.7% of total value) and modestly sensitive to changes in the annual rate of candidate arrivals to the waitlist (+20%: 19.7% of total value, −20%: 28.0% of total value) (**Supplementary Table 2**)

Figure 2. Annual Demand for Kidney Transplants by Patients With IgAN

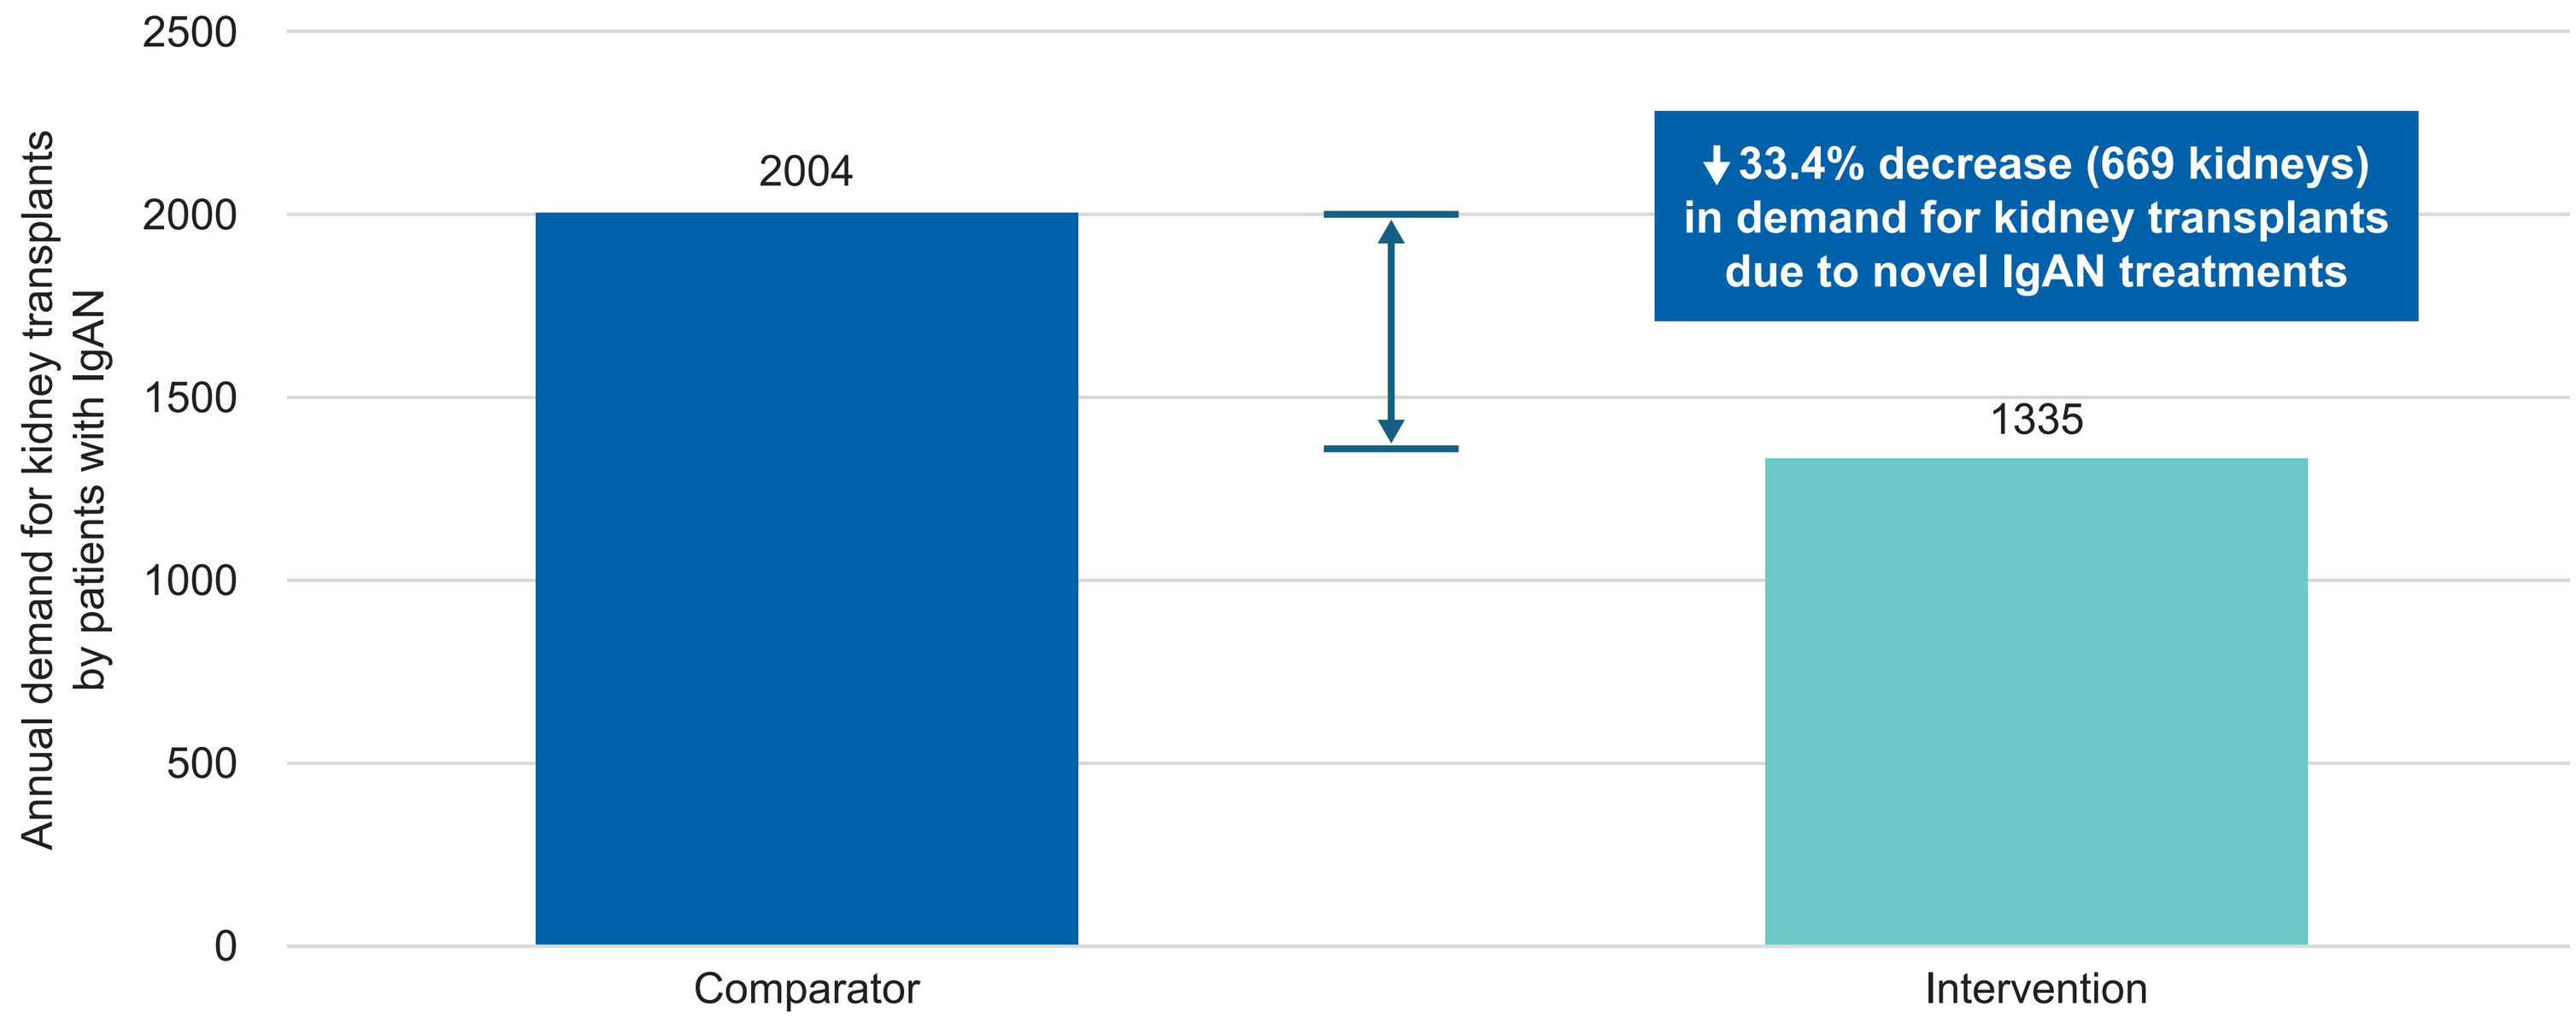
2500
2000
1500
1000
500
0
Annual demand for kidney transplants by patients with IgAN
2004
1335
↓33.4% decrease (669 kidneys) in demand for kidney transplants due to novel IgAN treatments
Comparator
Intervention

IgAN, immunoglobulin A nephropathy.

Figure 3. US Kidney Transplant Waitlist Size Over Time[a]

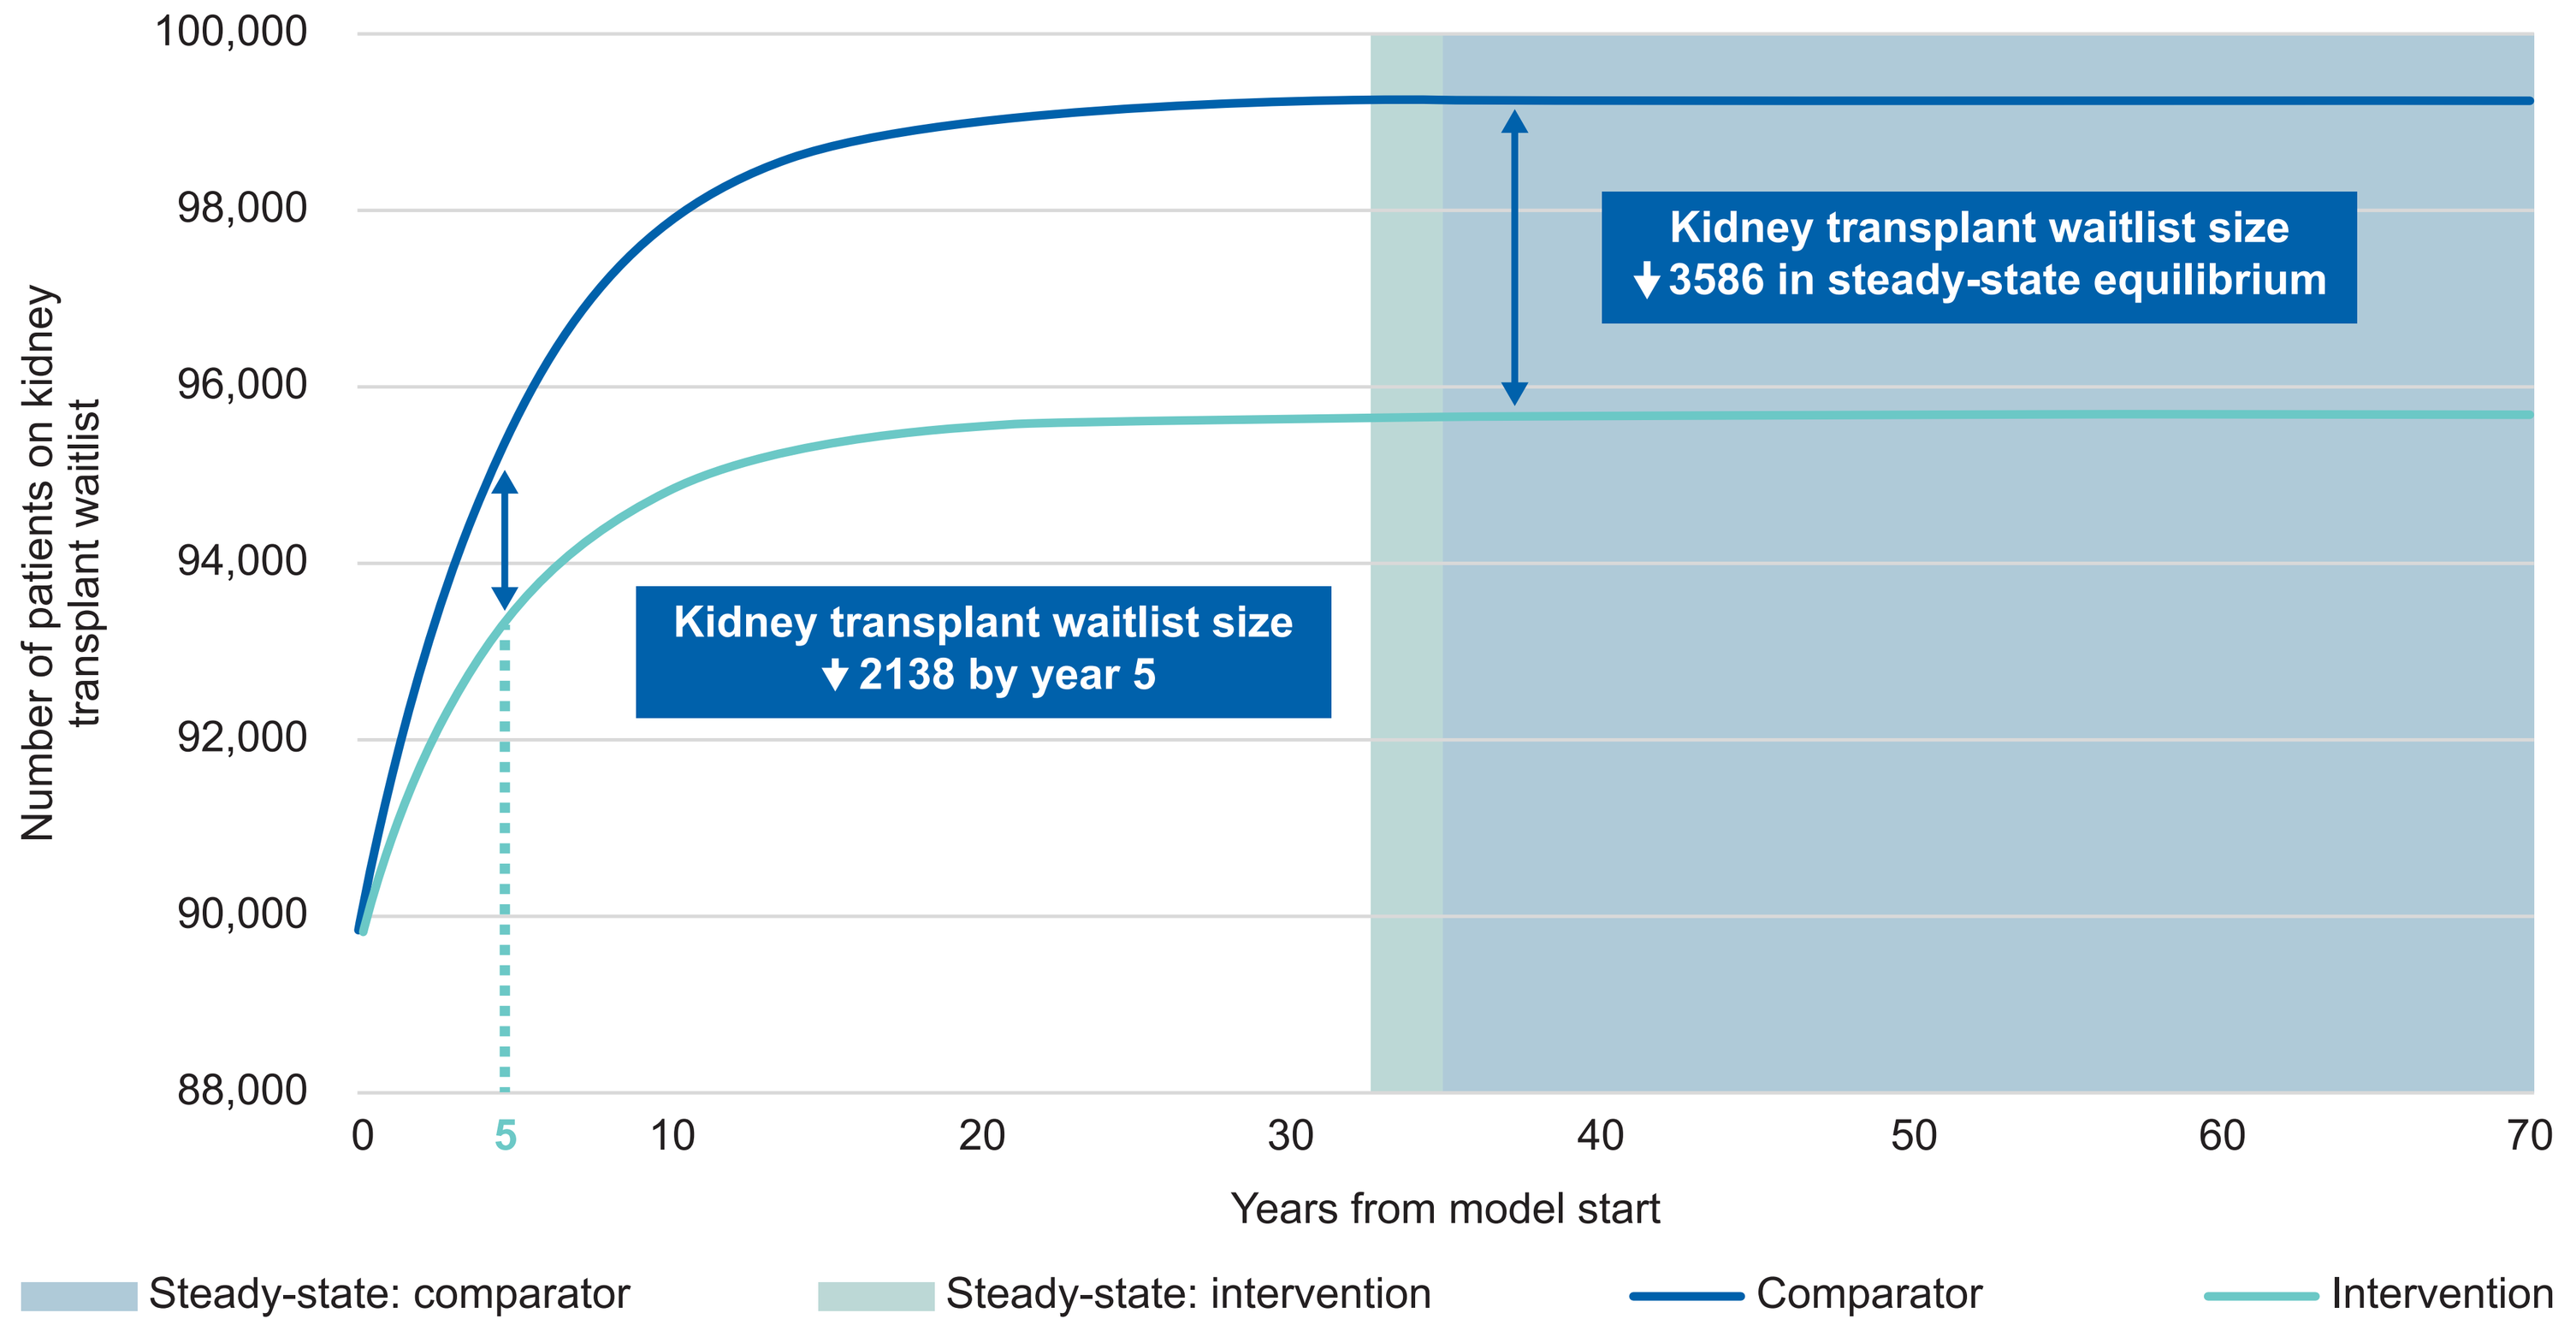
100,000
98,000
96,000
94,000
92,000
90,000
88,000
Number of patients on kidney transplant waitlist
0
5
10
20
30
40
50
60
70
Years from model start
Kidney transplant waitlist size ↓3586 in steady-state equilibrium
Kidney transplant waitlist size ↓2138 by year 5
Steady-state: comparator
Steady-state: intervention
Comparator
Intervention

[a]The transplant waitlist size reached steady-state equilibrium 27 months earlier in the intervention arm vs the comparator arm (first quarter of year 33 vs second quarter of year 35, respectively). Steady-state equilibrium was defined as the point when quarterly change in waitlist size relative to both (i) the previous quarter and (ii) the average waitlist size over the remaining period (until year 70) was less than 0.01%.

US, United States.

Table 3. Spillover Benefits per Kidney Transplant Candidate

Outcomes	Intervention [A]	Comparator [B]	Difference [A−B]
Wait time for kidney transplant (years)	2.423	2.493	−0.070
LYs	12.134	12.072	0.063
QALYs	7.786	7.698	0.087
Monetized QALYs (US$)	1,167,847	1,154,730	13,116

LY, life-year; QALY, quality-adjusted life-year; US, United States

Figure 4. Share of Treatment Value Attributable to Spillover Benefits per Patient With IgAN

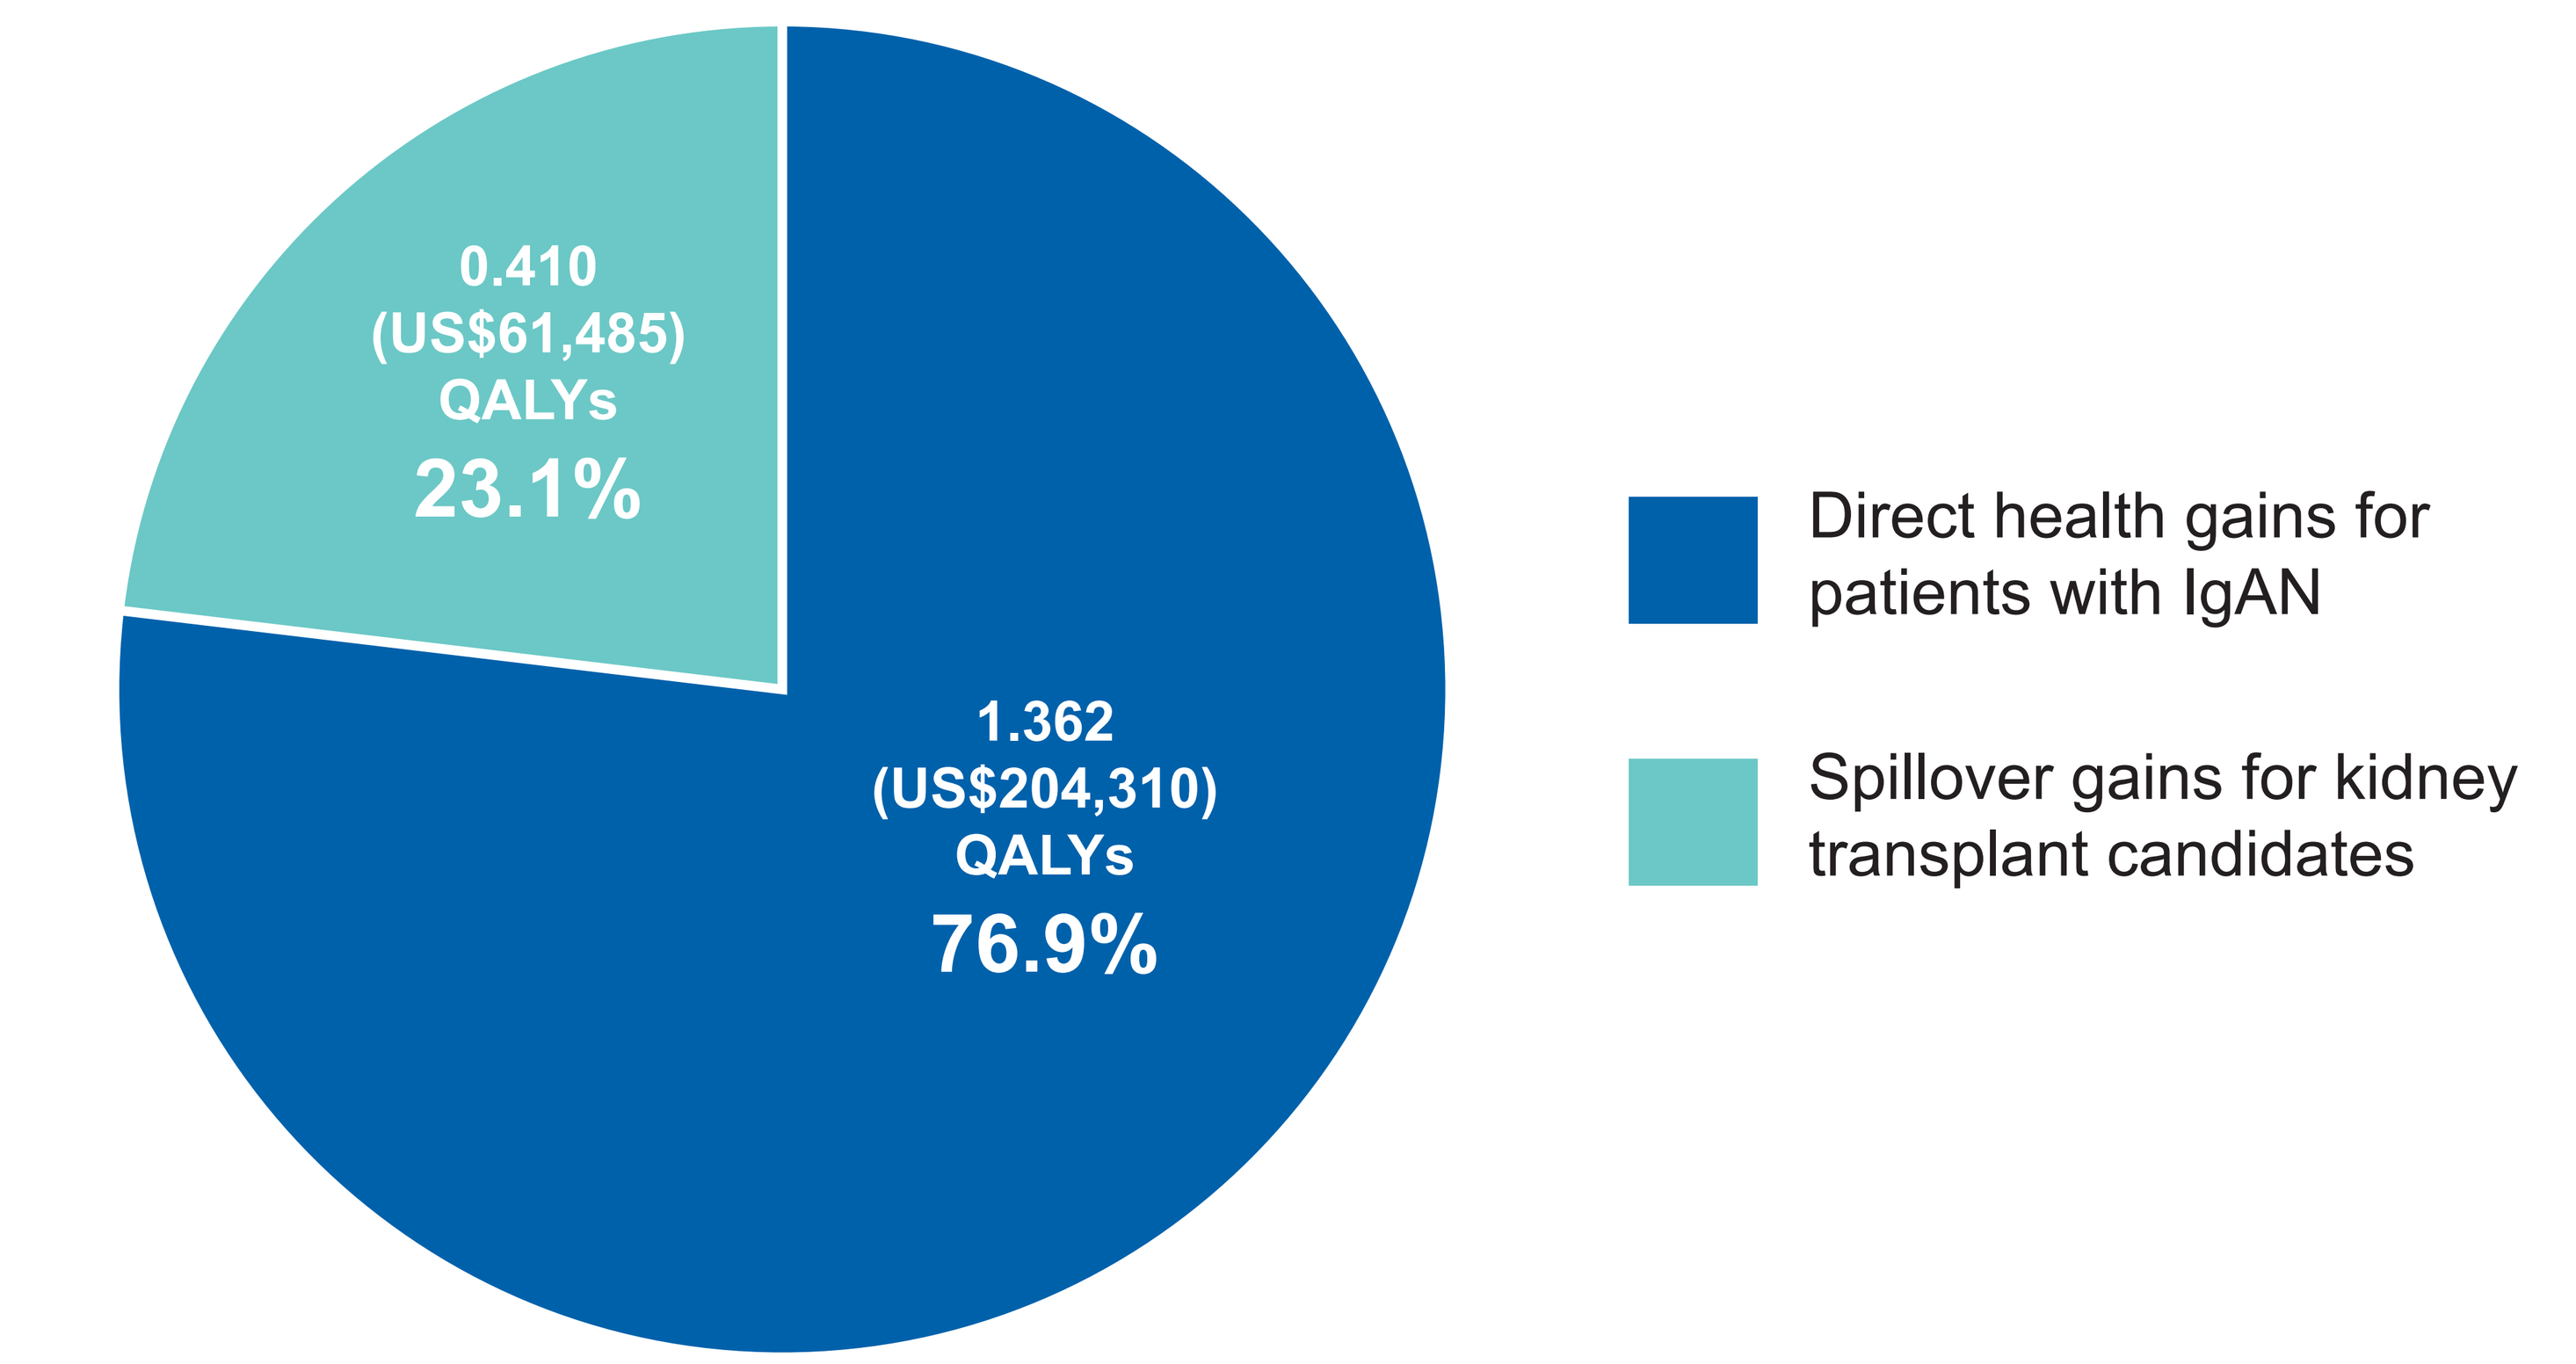
0.410 (US$61,485) QALYs
23.1%
1.362 (US$204,310) QALYs
76.9%
Direct health gains for patients with IgAN
Spillover gains for kidney transplant candidates

IgAN, immunoglobulin A nephropathy; QALY, quality-adjusted life-year; US, United States.

Limitations

- Modeled efficacy was not specific to individual treatments
- The model did not consider that patients with IgAN may experience additional health improvements from novel interventions due to reduced kidney transplant wait times in the IgAN population; therefore, the measured spillover effect is a conservative estimate
- Waitlist survival outcomes were based on the median patient

References

1. KDIGO Glomerular Diseases Work Group. *Kidney Int.* 2021;100(4S):S1–S276.
2. Lai KN, et al. *Nat Rev Dis Primers.* 2016;2:16001.
3. Kwon CS, et al. *J Health Econ Outcomes Res.* 2021;8(2):36–45.
4. Pitcher D, et al. *Clin J Am Soc Nephrol.* 2023;18(6):727–738.
5. OPTN. National data. 2023. Accessed March 6, 2025. https://optn.transplant.hrsa.gov/data/view-data-reports/national-data/
6. Stewart D, et al. *Am J Transplant.* 2023;23(2):223–231.
7. Lim RS, et al. *J Clin Med.* 2024;13(4):947.
8. Perkovic V, et al. *N Engl J Med.* 2025;392(6):531–543.
9. Heerspink HJL, et al. *N Engl J Med.* 2025;392(6):544–554.
10. Rovin BH, et al. *Lancet.* 2023;402(10417):2077–2090.
11. Keskinis C, et al. Nephrol Dial Transplant. 2024;39(Supplement_1):gfae069-1297-1165.
12. Wheeler DC, et al. *Kidney Int.* 2021;100(1):215–224.

Abbreviations

ACEi, angiotensin-converting enzyme inhibitor; ARB, angiotensin II receptor blocker; CKD, chronic kidney disease; CVD, cardiovascular disease; eGFR, estimated glomerular filtration rate; ESKD, end-stage kidney disease; FDA, US Food and Drug Administration; HR, hazard ratio; IgAN, immunoglobulin A nephropathy; KDIGO, Kidney Disease: Improving Global Outcomes; LY, life-year; OPTN, Organ Procurement and Transplantation Network; PICOTS, population, intervention, comparator, outcomes, timing, and setting; QALY, quality-adjusted life-year; US, United States.

Disclosures

JK, **JF**, and **JS** are employees of FTI Consulting, Los Angeles, California, USA. **BN**, **HT**, and **TRS** are employees of Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA. **AI** is an employee of University of Texas Medical Branch, Galveston, Texas, USA, and has received consulting fees from Novartis and Vera Pharmaceuticals.

Acknowledgments

Medical writing support and editorial support were provided by Elizabeth Murray, PhD (BOLDSCIENCE Ltd. UK) and were funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP) guidelines. The authors had full control of the content and made the final decision on all aspects of this publication.